Impact of the CIMplicity® Patient Support Program on Persistence to Certolizumab Pegol Treatment: A Retrospective Cohort Analysis of Claims Data in the United States

Aimee Near,[1] Srihari Jaganathan,[2] Chao Song,[2] Edward Lee[2]

Presented by: Robert Low[2]

Presented at CCR East 2023 | May 4–7 | Destin, FL

Objective

The objective of this study was to assess the impact of the CIMplicity® patient support program (PSP) on persistence to certolizumab pegol (CZP) among patients enrolled in the PSP compared with patients not enrolled in the PSP.

Background

- CZP is a tumor necrosis factor (TNF) inhibitor used to treat chronic diseases such as rheumatoid arthritis (RA), psoriatic arthritis (PsA), ankylosing spondylitis (AS), Crohn's disease, psoriasis, and non-radiographic axial spondyloarthritis.[1]
- CZP is administered subcutaneously either by patients or by a healthcare provider.[2,3]
- CIMplicity® is a PSP sponsored by UCB Pharma, providing live telehealth nurse support and is available to all eligible patients treated with CZP.[2]

Methods

- This study was a retrospective claims database analysis comparing persistence to CZP for patients enrolled in CIMplicity® PSP with those not enrolled (**Figure 1**).
 - Data were linked between UCB CIMplicity® and IQVIA longitudinal prescription (LRx) and medical (Dx) claims databases (September 10, 2015–June 30, 2020).
 - Patients were ≥18 years of age with RA, PsA, or AS, newly treated with CZP (≥1 claim), and were identified during the study selection period. The study index date was the first claim for CZP and the follow-up period was ≥90 days post-index.
- Two 1:1 propensity score-matched cohorts based on 6-month baseline patient characteristics were created: the PSP cohort (enrolled in PSP with welcome call and ≥2 follow-up calls) and the non-PSP cohort (not enrolled in PSP during study period).
 - Persistence to CZP was measured as time to treatment discontinuation (TTD). Kaplan-Meier analyses of TTD were performed to assess persistence between cohorts. Subgroup analyses of patients who self-administered CZP and those with RA were performed.

Results

- This study consisted of 753 PSP and 7,176 non-PSP patients prior to matching; after matching, 616 patients remained in both (PSP and non-PSP) RA, PsA, AS combined cohorts; 471 remained in both RA cohorts; 337 patients remained in both RA, PsA, or AS self-administered cohorts; 211 patients remained in both RA self-administered cohorts (**Figure 2**).
- There were no significant differences in baseline characteristics following propensity matching of cohorts (**Table 1**).
- The PSP cohort had significantly greater persistence (TTD) over follow-up compared to the non-PSP cohort (median days [95% confidence interval {CI}]: 194 [175, 215] vs 166 [142, 188]; p=0.001; **Table 2** and **Figure 3**).
- Among the subgroup of patients that self-administered CZP (n=337), the PSP cohort had greater TTD over follow-up compared to the non-PSP cohort (median days [95% CI]: 189 [169, 224] vs 151 [130, 182]; p=0.003; **Table 2** and **Figure 4**).
- Similar results were observed in patients with RA (n=471), among whom the PSP cohort had greater TTD over follow-up compared to the non-PSP cohort (median days [95% CI]: 188 [170, 209] vs 168 [137, 196]; p=0.03; **Table 2** and **Figure 5**).
- In patients with RA who self-administered CZP (n=211), the PSP cohort had greater TTD over follow-up compared to the non-PSP cohort (median days [95% CI]: 180 [152, 212] vs 129 [102, 163]; p=0.01; **Table 2** and **Figure 6**).

Limitations

- Misreporting of outcomes may have occurred due to dependency on the accuracy of the underlying LRx and Dx coding | LRx and Dx claims data were not collected for research purposes | Severity of disease was not collected | Few patients had follow-up times that far exceeded minimum requirement | Continuous enrollment, continuity of care, and continuous treatment cannot be verified by available data | Included patients had heterogenous disease indications | Patients were recruited over a wide selection period (approximately 5 years) | An immortality bias may have been introduced by requiring a minimum length of follow-up (approximately 3 months) | Patients with multiple concomitant diagnoses of interest were excluded.

Summary

This analysis examined persistence to CZP for RA, PsA, and AS patients who were enrolled in the CIMplicity® PSP compared with propensity matched patients that were not

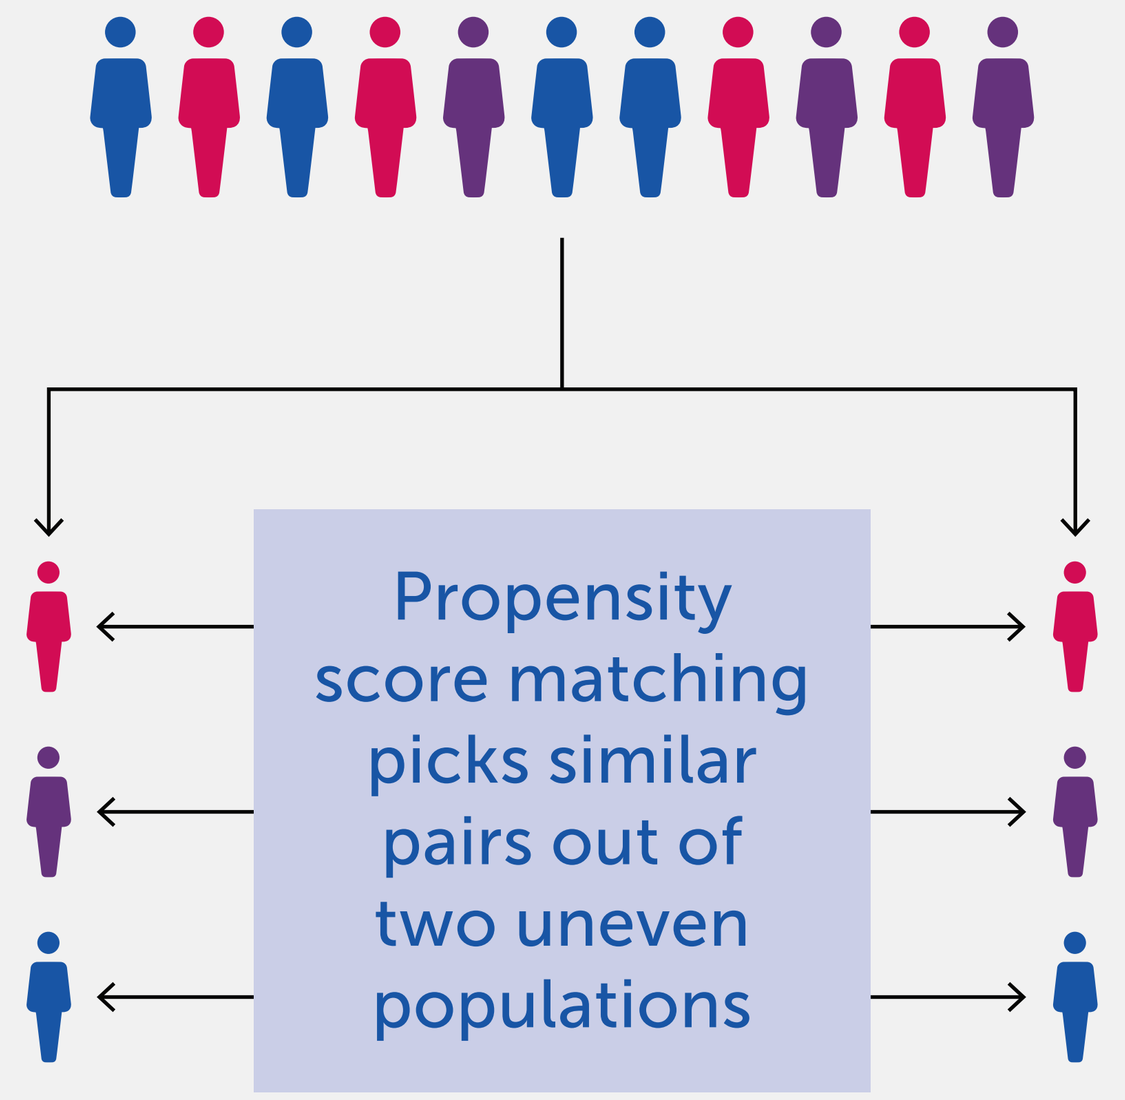
Propensity score matching picks similar pairs out of two uneven populations

Patients were further stratified into those who self-administered CZP and patients that received CZP in-office from a healthcare provider

A subgroup analysis was also performed on those patients with RA only

	Non-PSP	PSP
At home (self-administered)	RA or RA, PsA, AS	RA or RA, PsA, AS
In-Office	RA or RA, PsA, AS	RA or RA, PsA, AS

Persistence to CZP (RA, PsA, and AS)

Time to treatment discontinuation (TTD)

Non-PSP: **166** days → p=0.001 → PSP: **194** days

Clinical Implications

Physicians should consider increasing awareness of CIMplicity® PSP for those who may benefit

Figure 1 Study design

Index Date: First CZP Claim (PSP[a] and non-PSP)
Selection Period: September 10, 2015 to June 30, 2020

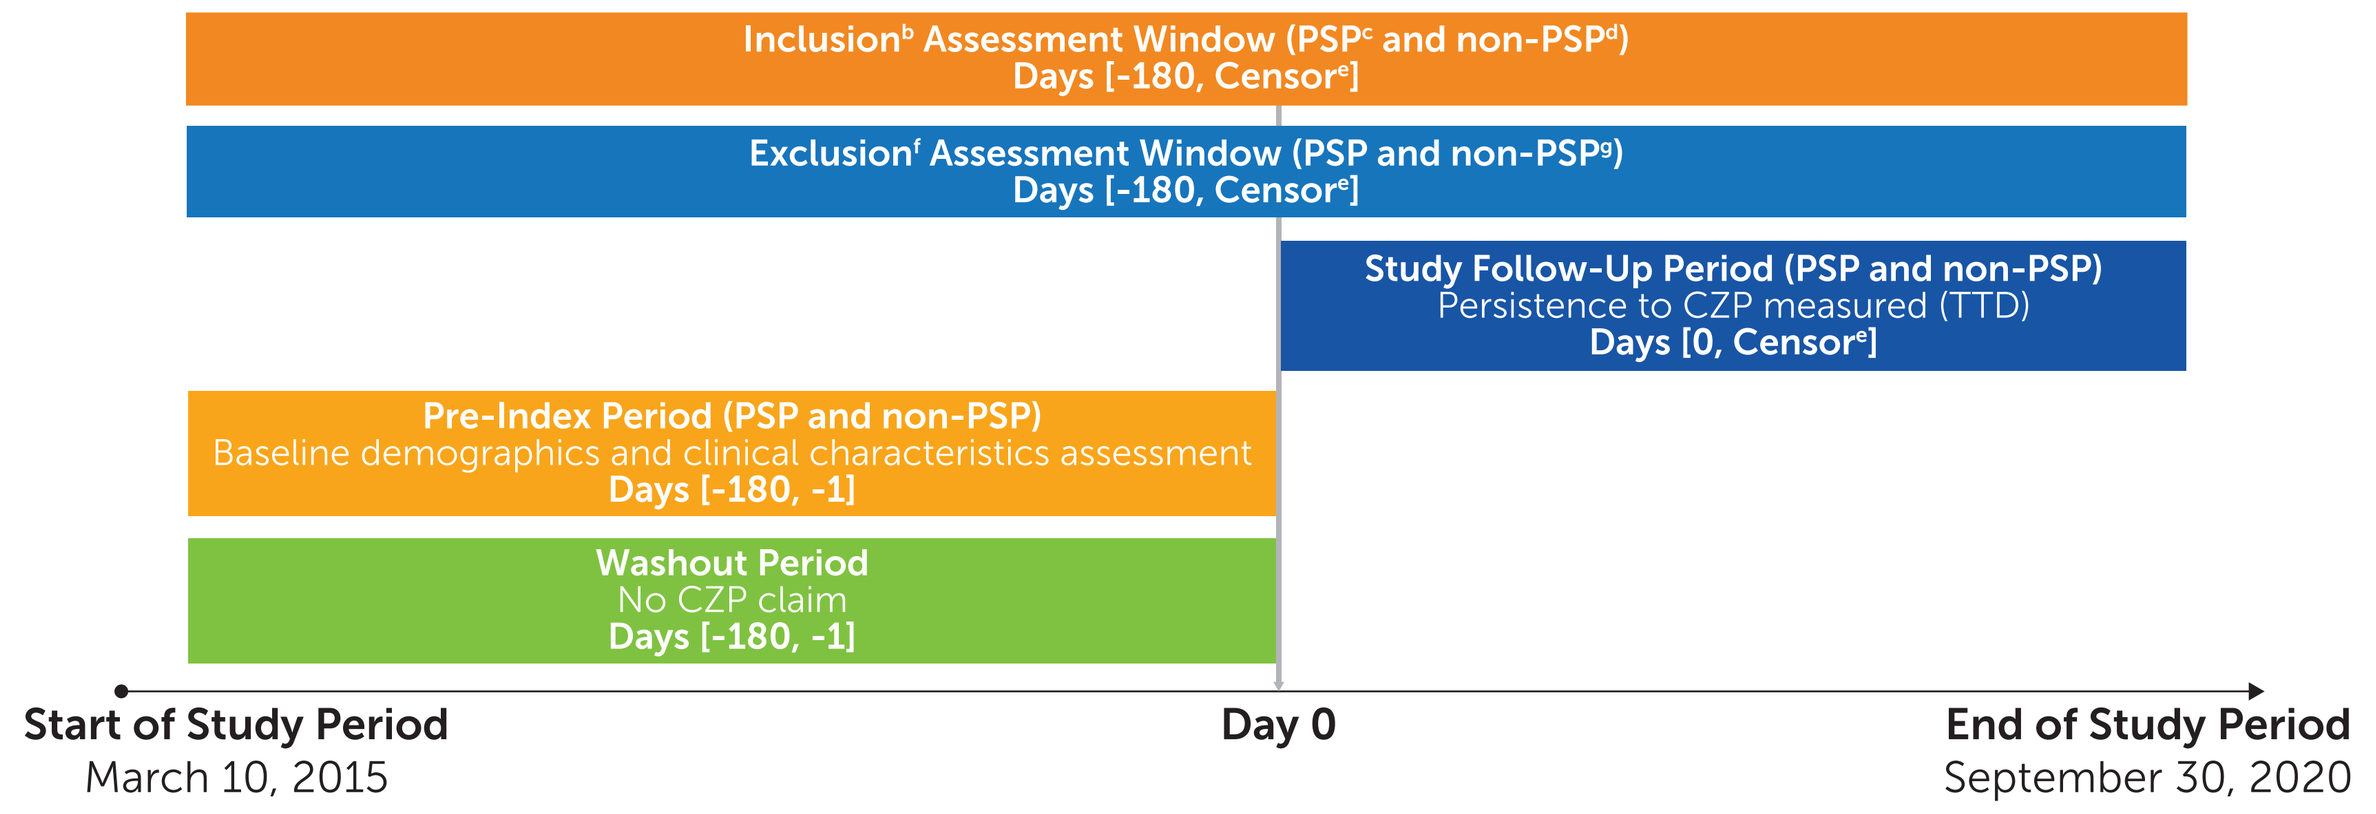
Inclusion[b] Assessment Window (PSP[c] and non-PSP[d])
Days [-180, Censor[e]]
Exclusion[f] Assessment Window (PSP and non-PSP[g])
Days [-180, Censor[e]]
Study Follow-Up Period (PSP and non-PSP)
Persistence to CZP measured (TTD)
Days [0, Censor[e]]
Pre-Index Period (PSP and non-PSP)
Baseline demographics and clinical characteristics assessment
Days [-180, -1]
Washout Period
No CZP claim
Days [-180, -1]
Start of Study Period
March 10, 2015
Day 0
End of Study Period
September 30, 2020

[a]CZP claim in LRx or Dx databases ±60 days from enrollment in CIMplicity® PSP; [b]≥1 Dx claim and ≥1 LRx claim, for patients indexing with Dx claim ≥1 Dx claim in follow-up period, for patients indexing with LRx claim ≥1 LRx claim in follow-up period, and ≥18 years of age in pre-index period; [c]One CIMplicity® PSP welcome call and ≥2 follow-up calls and, linkable UCB CIMplicity® and IQVIA LRx and Dx data; [d]≥2 outpatient Dx claims with diagnosis of AS, PsA, or RA 30 days apart during pre-index period; [e]Patients were censored at discontinuation (gap of ≥56 days or start of new biologic), disenrollment, or end of study period (September 30, 2020), whichever occurred first; [f]Missing age, gender, or any data quality issue, evidence of CZP use in pre-index period, <90 days of follow-up data due to censoring, diagnosis of diseases other than disease assigned in UCB CIMplicity® database (i.e. PsA patients cannot have a diagnosis of AS or RA); gEnrollment in CIMplicity® PSP.

Table 1 Baseline characteristics for propensity matched PSP and non-PSP cohorts

Characteristic	RA, PsA, or AS[a] PSP (n=616)	Non-PSP (n=616)	P value	RA[b] PSP (n=471)	Non-PSP (n=471)	P value
Disease, n (%)			0.133			
AS	75 (12.2)	72 (11.7)	-			-
PsA	66 (10.7)	59 (9.6)	-			-
RA	475 (77.1)	485 (78.7)	-	471 (100)	471 (100)	-
Age, mean (SD)	55.5 (15.6)	56.1 (16.4)	0.200	57.7 (15.5)	57.8 (16.4)	0.867
Gender, n (%)						
Male	109 (17.7)	109 (17.7)	-	70 (14.9)	70 (14.9)	-
Female	507 (82.3)	507 (82.3)	-	401 (85.1)	401 (85.1)	-
Payer type, n (%)			0.260			0.103
Medicare	171 (27.8)	183 (29.7)	-	156 (33.1)	165 (35.0)	-
Commercial	437 (70.9)	424 (68.8)	-	307 (65.2)	302 (64.1)	-
Other	8 (1.3)	9 (1.5)	-	8 (1.7)	4 (0.9)	-
Geographic region, n (%)			0.864			0.869
Northeast	84 (13.6)	85 (13.8)	-	50 (10.6)	51 (10.8)	-
South	362 (58.8)	362 (58.8)	-	299 (63.5)	303 (64.3)	-
Midwest	101 (16.4)	97 (15.8)	-	72 (15.3)	64 (13.6)	-
West	69 (11.2)	72 (11.7)	-	50 (10.6)	53 (11.3)	-
Charlson Comorbidity Index (CCI), mean (SD)	1.1 (1.0)	1.1 (1.0)	0.889	1.4 (1.0)	1.3 (1.0)	0.540
Anxiety, n (%)	33 (5.4)	33 (5.4)	1.000	24 (5.1)	25 (5.3)	0.862
Depression, n (%)	39 (6.3)	30 (4.9)	0.180	28 (5.9)	27 (5.7)	0.847
Atherosclerotic cardiovascular disease, n (%)	77 (12.5)	72 (11.7)	0.484	71 (15.1)	65 (13.8)	0.553
Diabetes, n (%)	71 (11.5)	68 (11.0)	0.701	57 (12.1)	46 (9.8)	0.071
Hyperlipidemia, n (%)	97 (15.8)	104 (16.9)	0.419	86 (18.3)	77 (16.4)	0.225
Hypertension, n (%)	137 (22.2)	136 (22.1)	0.908	117 (24.8)	127 (27.0)	0.166
Prior biologic use,[c] n (%)						
0	350 (56.8)	338 (54.9)	0.475	286 (60.7)	292 (62.0)	0.491
1	228 (37.0)	239 (38.8)	0.513	160 (34.0)	156 (33.1)	0.555
2+	38 (6.2)	39 (6.3)	0.907	25 (5.3)	23 (4.9)	0.746
Place of administration, n (%)						
In-office	279 (45.3)	279 (45.3)	-	260 (55.2)	260 (55.2)	-
Self-administered	337 (54.7)	337 (54.7)	-	211 (44.8)	211 (44.8)	-
Treatment history, n (%)						
Injectable corticosteroids	132 (21.4)	132 (21.4)	1.000	119 (25.3)	114 (24.2)	0.435
TNF inhibitors	187 (30.4)	208 (33.8)	0.180	125 (26.5)	119 (25.3)	0.574

[a]Propensity matching for RA, PsA, and AS cohorts were sequentially matched based on: disease, age, gender, payer type, CCI, select comorbidities, history of injectable steroid use, place of administration, region, history of TNF inhibitor use; [b]Propensity matching for RA cohorts were sequentially matched based on: place of administration, gender, age, payer type, CCI, region, select comorbidities, treatment history, prior biologic use; [c]Assessed as claims for biologics other than CZP during pre-index period.

AS: ankylosing spondylitis; **CCI:** Charlson Comorbidity Index; **CI:** confidence interval; **CZP:** certolizumab pegol; **Dx:** medical; **LRx:** prescription; **PsA:** psoriatic arthritis; **PSP:** patient support program; **RA:** rheumatoid arthritis; **TNF:** tumor necrosis factor; **TTD:** time to treatment discontinuation.

Figure 2 Study attrition

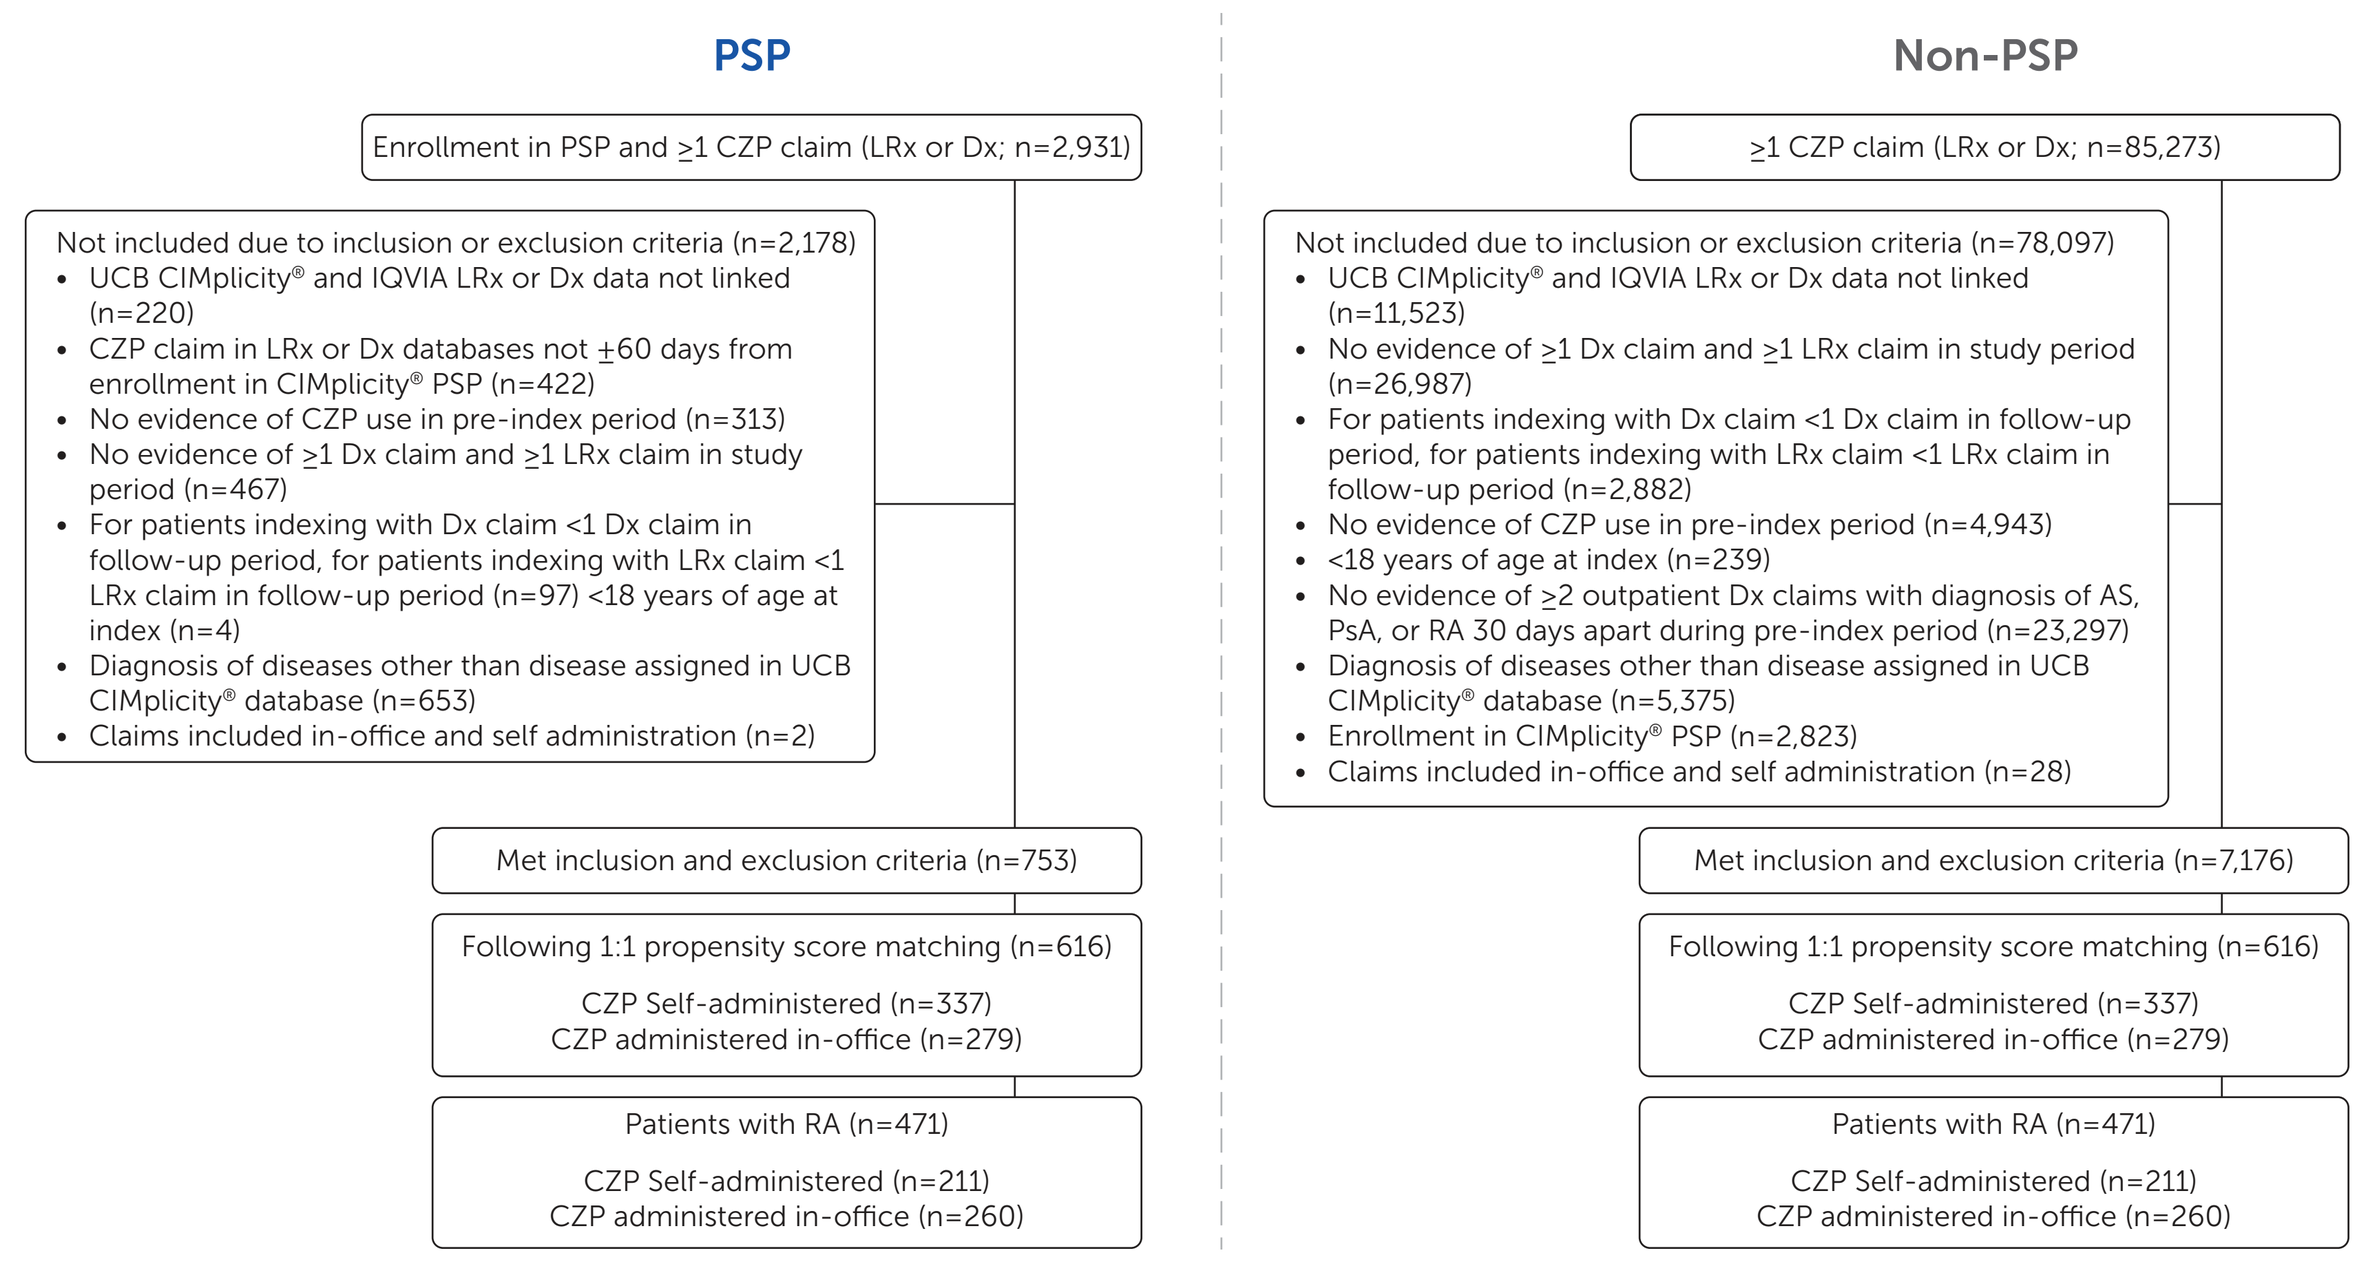
PSP
Enrollment in PSP and ≥1 CZP claim (LRx or Dx; n=2,931)
Not included due to inclusion or exclusion criteria (n=2,178)
• UCB CIMplicity® and IQVIA LRx or Dx data not linked (n=220)
• CZP claim in LRx or Dx databases not ±60 days from enrollment in CIMplicity® PSP (n=422)
• No evidence of CZP use in pre-index period (n=313)
• No evidence of ≥1 Dx claim and ≥1 LRx claim in study period (n=467)
• For patients indexing with Dx claim <1 Dx claim in follow-up period, for patients indexing with LRx claim <1 LRx claim in follow-up period (n=97) <18 years of age at index (n=4)
• Diagnosis of diseases other than disease assigned in UCB CIMplicity® database (n=653)
• Claims included in-office and self administration (n=2)
Met inclusion and exclusion criteria (n=753)
Following 1:1 propensity score matching (n=616)
CZP Self-administered (n=337)
CZP administered in-office (n=279)
Patients with RA (n=471)
CZP Self-administered (n=211)
CZP administered in-office (n=260)
Non-PSP
≥1 CZP claim (LRx or Dx; n=85,273)
Not included due to inclusion or exclusion criteria (n=78,097)
• UCB CIMplicity® and IQVIA LRx or Dx data not linked (n=11,523)
• No evidence of ≥1 Dx claim and ≥1 LRx claim in study period (n=26,987)
• For patients indexing with Dx claim <1 Dx claim in follow-up period, for patients indexing with LRx claim <1 LRx claim in follow-up period (n=2,882)
• No evidence of CZP use in pre-index period (n=4,943)
• <18 years of age at index (n=239)
• No evidence of ≥2 outpatient Dx claims with diagnosis of AS, PsA, or RA 30 days apart during pre-index period (n=23,297)
• Diagnosis of diseases other than disease assigned in UCB CIMplicity® database (n=5,375)
• Enrollment in CIMplicity® PSP (n=2,823)
• Claims included in-office and self administration (n=28)
Met inclusion and exclusion criteria (n=7,176)
Following 1:1 propensity score matching (n=616)
CZP Self-administered (n=337)
CZP administered in-office (n=279)
Patients with RA (n=471)
CZP Self-administered (n=211)
CZP administered in-office (n=260)

Figure 3 Probability of remaining on CZP (Kaplan-Meier analysis) for PSP compared with non-PSP (RA, PsA, and AS patients)

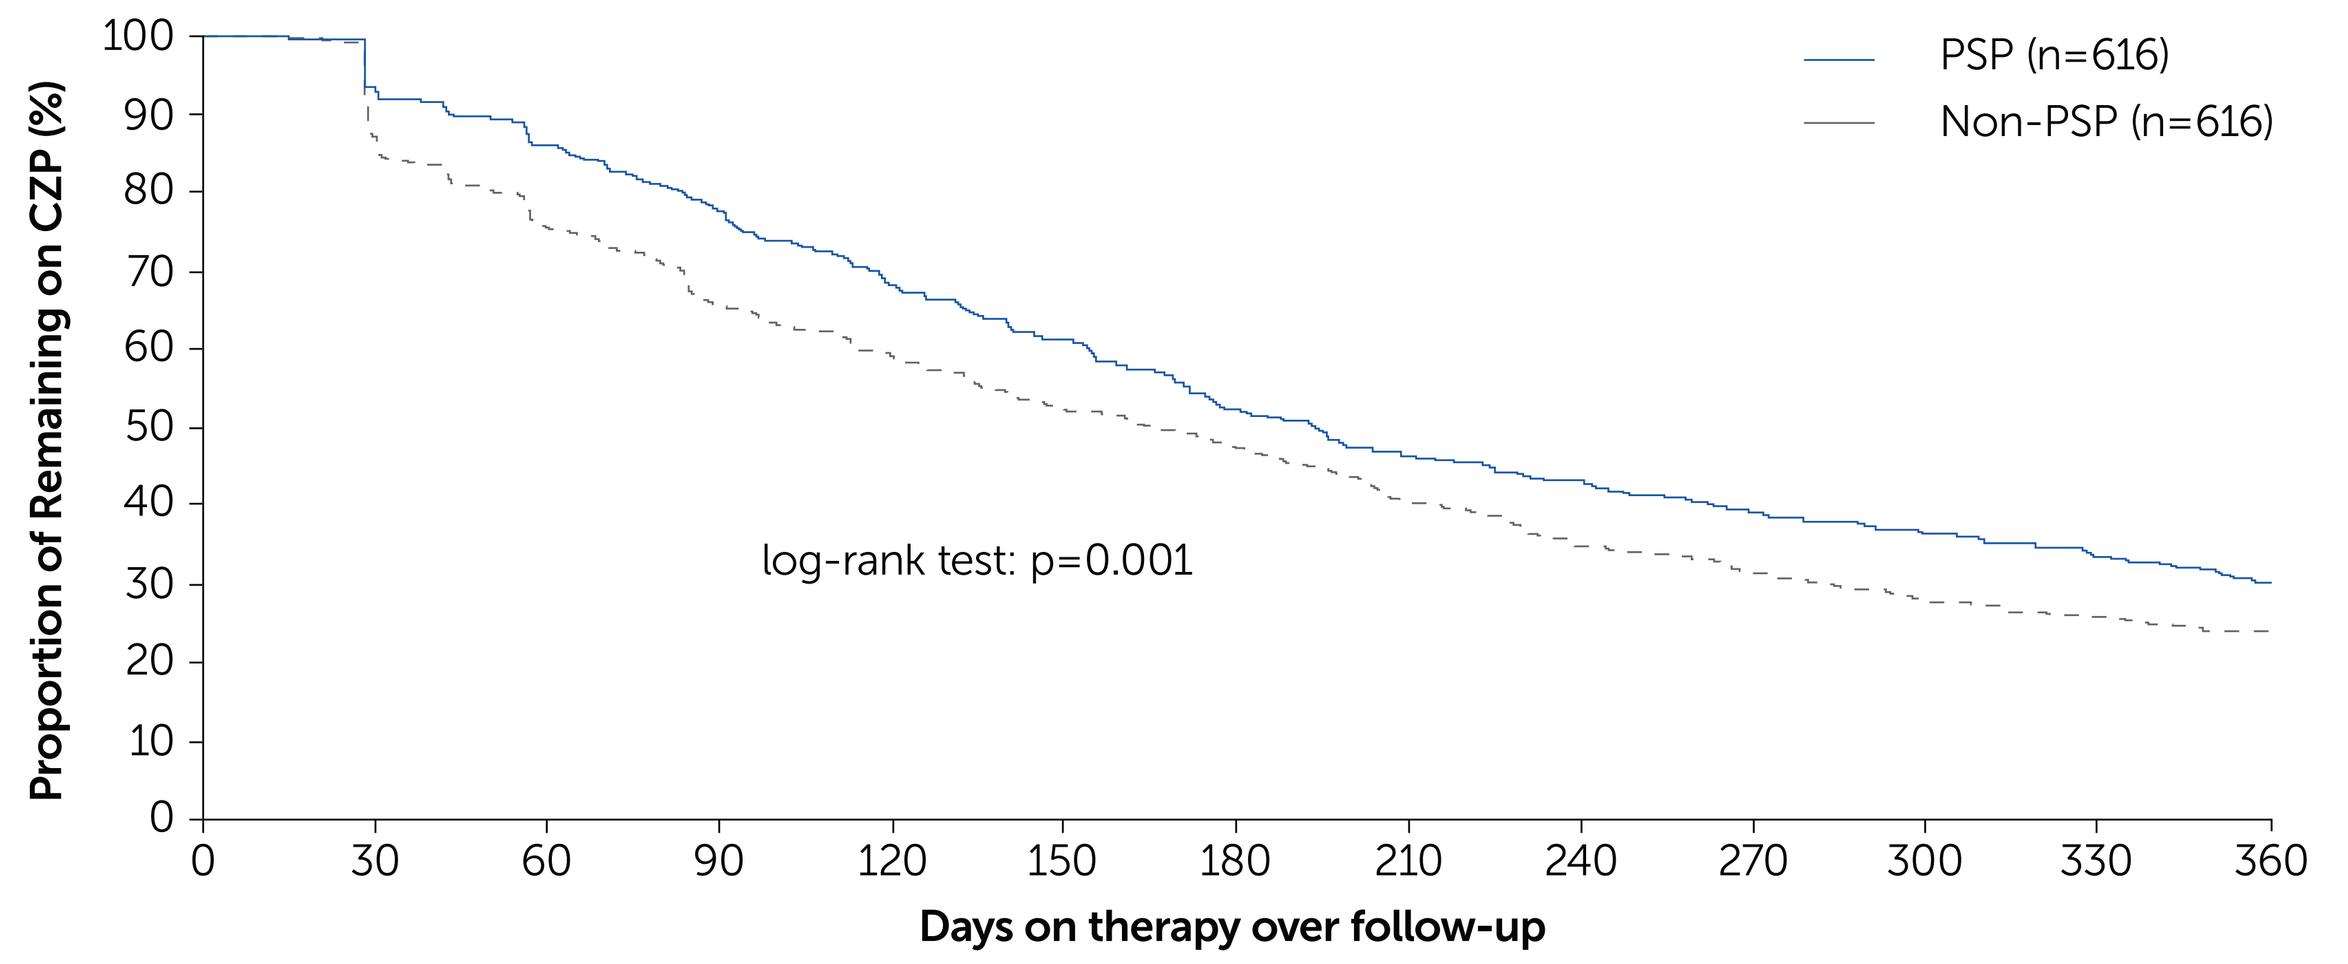
PSP (n=616)
Non-PSP (n=616)
log-rank test: p=0.001
Proportion of Remaining on CZP (%)
Days on therapy over follow-up

Propensity matching for RA, PsA, and AS cohorts were sequentially matched based on: disease, age, gender, payer type, CCI, select comorbidities, history of injectable steroid use, place of administration, region, history of TNF inhibitor use.

Figure 4 Probability of remaining on self-administered CZP (Kaplan-Meier analysis) for PSP compared with non-PSP (RA, PsA, and AS patients)

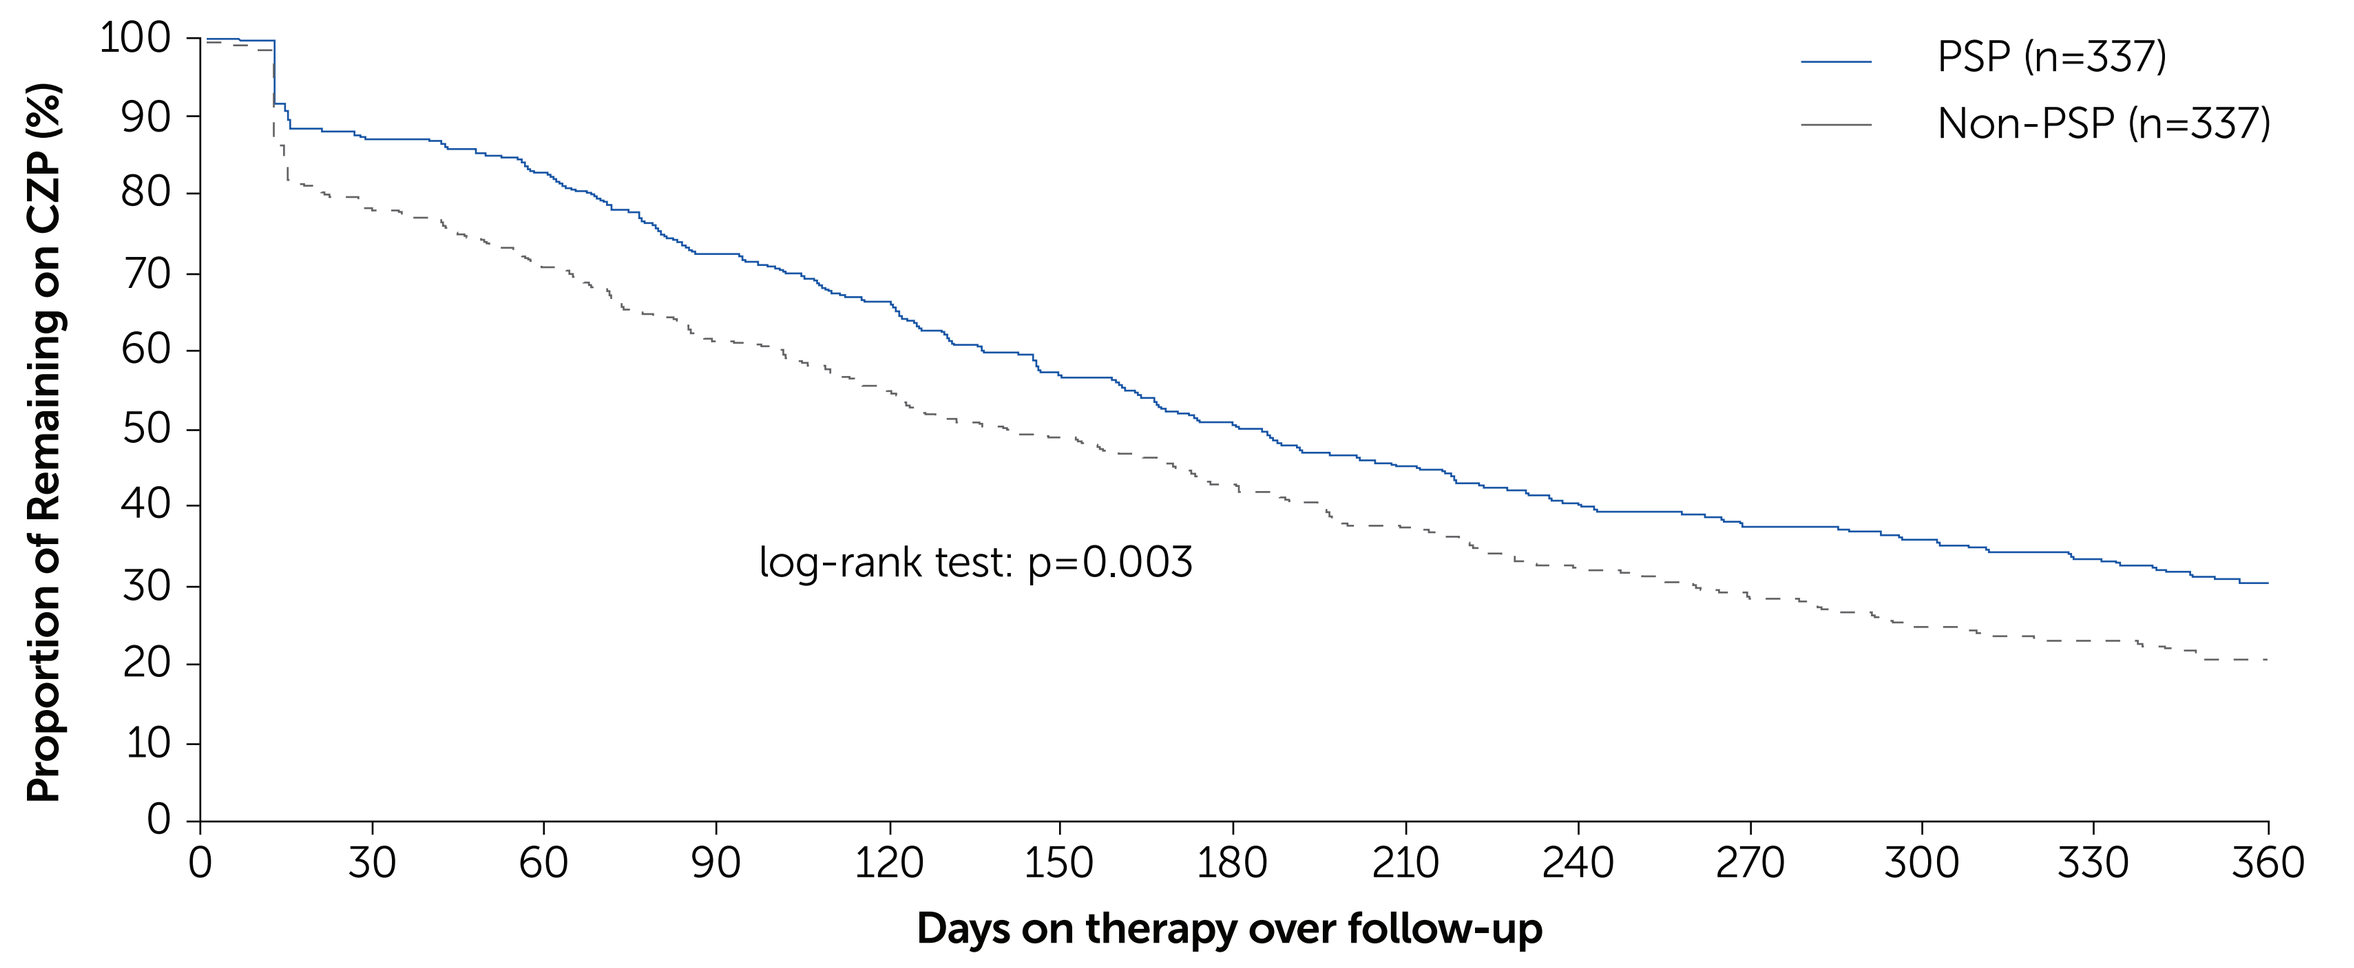
PSP (n=337)
Non-PSP (n=337)
log-rank test: p=0.003
Proportion of Remaining on CZP (%)
Days on therapy over follow-up

Propensity matching for RA, PsA, and AS cohorts were sequentially matched based on: disease, age, gender, payer type, CCI, select comorbidities, history of injectable steroid use, place of administration, region, history of TNF inhibitor use.

Table 2 Persistence to CZP for PSP compared to non-PSP

			Median days on therapy: Point Estimate	95% CI	P value
RA, PsA, AS[a]	Entire Cohort	PSP (n=616)	194	175, 215	0.001
		Non-PSP (n=616)	166	142, 188	-
	In-office	PSP (n=279)	196	171, 245	0.119
		Non-PSP (n=279)	187	147, 207	-
	Self-administered	PSP (n=337)	189	169, 224	0.003
		Non-PSP (n=337)	151	130, 182	-
RA[b]	Entire Cohort	PSP (n=471)	188	170, 209	0.027
		Non-PSP (n=471)	168	137, 196	-
	In-office	PSP (n=260)	196	171, 255	0.439
		Non-PSP (n=260)	200	166, 230	-
	Self-administered	PSP (n=211)	180	152, 212	0.014
		Non-PSP (n=211)	129	102, 163	-

[a]Propensity matching for RA cohorts were sequentially matched based on: place of administration, gender, age, payer type, CCI, region, select comorbidities, treatment history, prior biologic use; [b]Assessed as claims for biologics other than CZP during pre-index period.

Figure 5 Probability of remaining on CZP (Kaplan-Meier analysis) for PSP compared with non-PSP (RA patients)

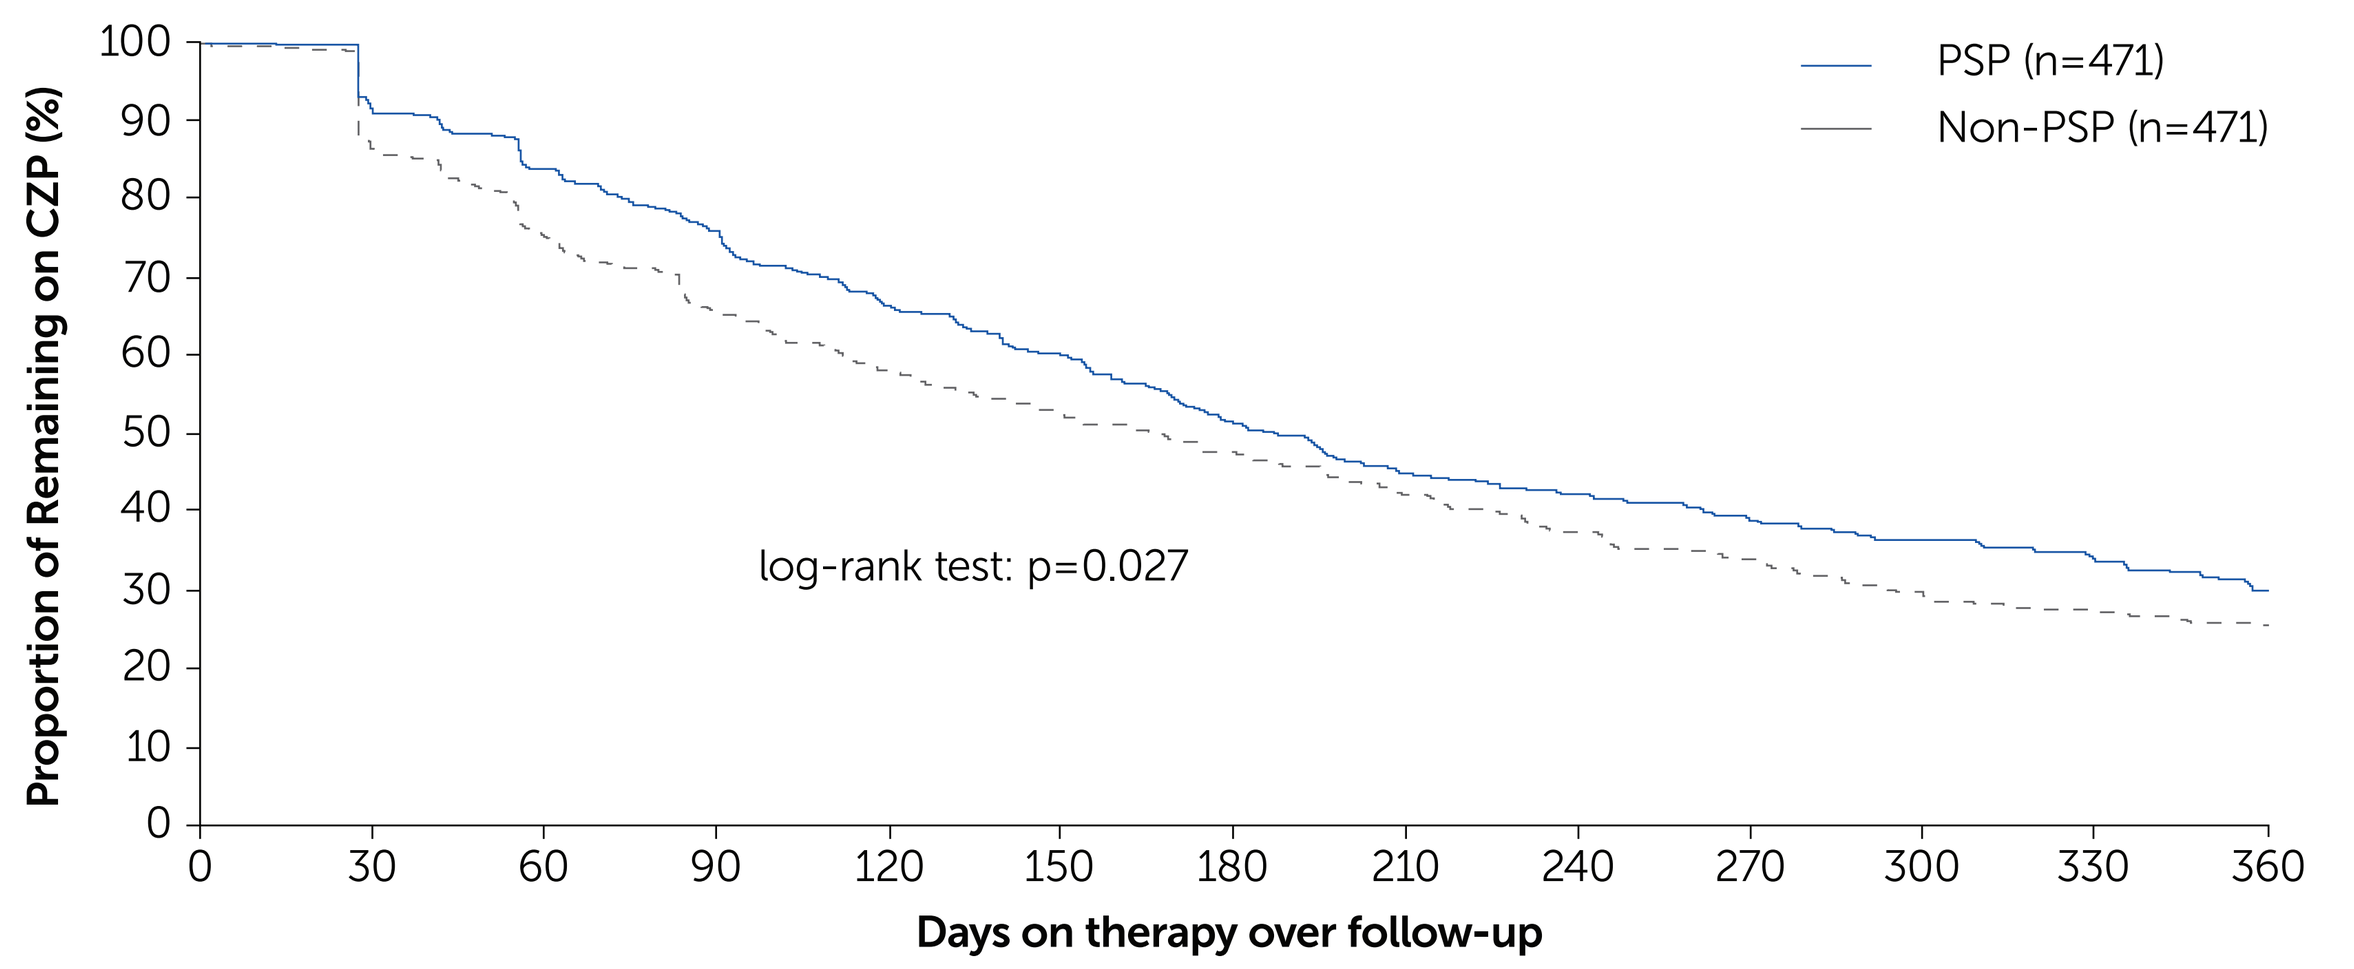
PSP (n=471)
Non-PSP (n=471)
log-rank test: p=0.027
Proportion of Remaining on CZP (%)
Days on therapy over follow-up

Propensity matching for RA cohorts were sequentially matched based on: place of administration, gender, age, payer type, CCI, region, select comorbidities, treatment history, prior biologic use.

Figure 6 Probability of remaining on self-administered CZP (Kaplan-Meier analysis) for PSP compared with non-PSP (RA patients)

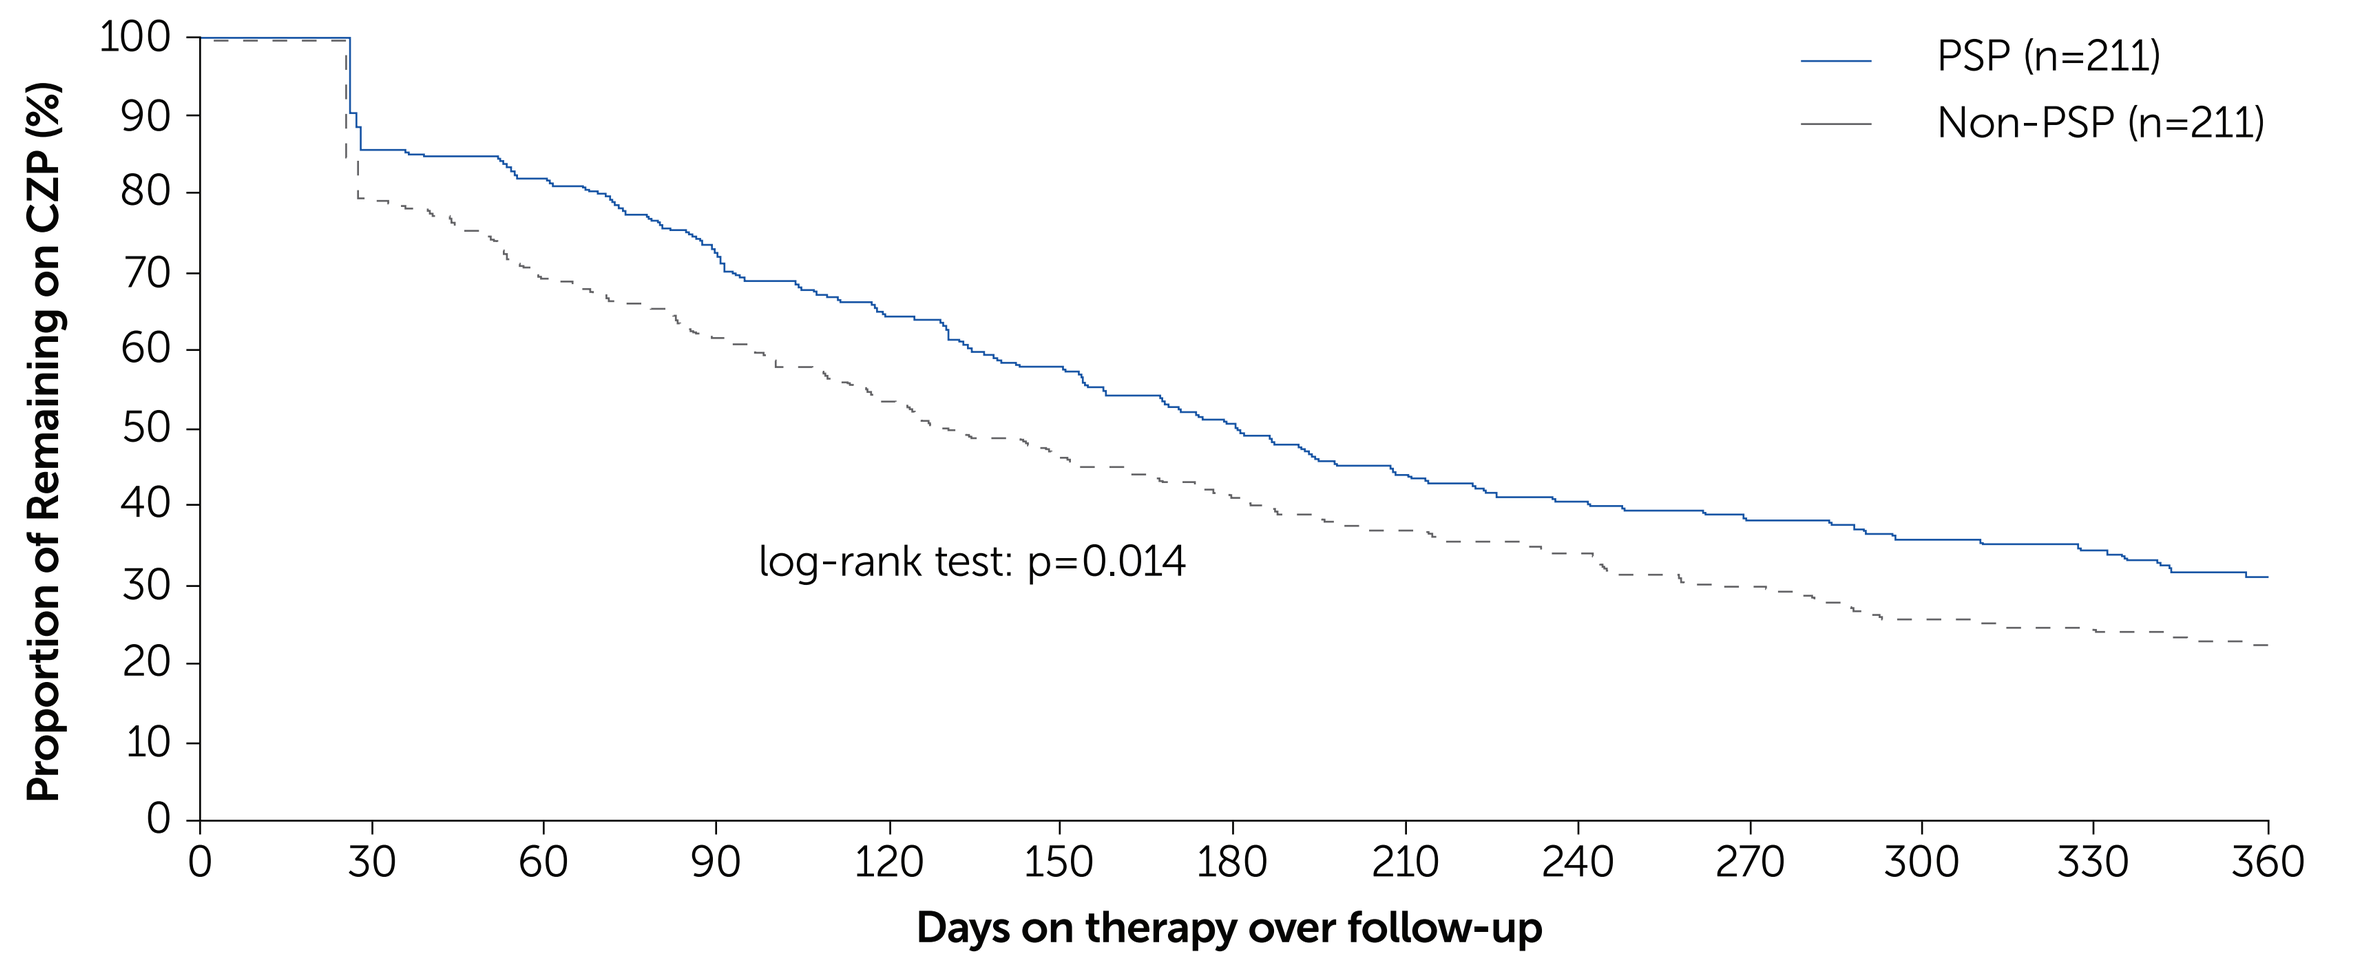
PSP (n=211)
Non-PSP (n=211)
log-rank test: p=0.014
Proportion of Remaining on CZP (%)
Days on therapy over follow-up

Propensity matching for RA cohorts were sequentially matched based on: place of administration, gender, age, payer type, CCI, region, select comorbidities, treatment history, prior biologic use.

Conclusions

This study suggests that CIMplicity® PSP, a live telehealth nurse support program, is associated with improved persistence to CZP. Further research should examine additional benefits that CIMplicity® may offer to patients with chronic rheumatic disease and the value of CIMplicity® in value-based care approaches.

Institutions: [1]IQVIA Inc., Durham, North Carolina, USA; [2]UCB Pharma, Smyrna, Georgia, USA.

References: [1]Chimenti MS. et al. Drug Des Devel Ther. 2013; 7:339–48; [2]Wolf DC. et al. Patient Prefer Adherence. 2018; 21:869–78. [3]Certolizumab pegol [package insert]. US FDA. 2019. **Author Contributions:** Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **AN, SJ, CS, EL;** drafting of the publication, or revising it critically for important intellectual content: **AN, SJ, CS, EL;** final approval of the publication: **AN, SJ, CS, EL. Disclosures: AN:** Employee of IQVIA; **SJ, CS, EL:** Employees and shareholders of UCB Pharma. **Acknowledgements:** This study was funded by UCB Pharma. We would like to thank the patients and their caregivers in addition to the investigators and their teams who contributed to this study. The authors acknowledge Patrick Reilly, BS, Costello Medical, Boston, MA, for medical writing, and the Design Team at Costello Medical for graphic design assistance. The authors would like to thank Robert Low, UCB Pharma, for presenting this research. All costs associated with development of this poster were funded by UCB Pharma.