Low Uveitis Rates in Patients with Axial Spondyloarthritis or Psoriatic Arthritis Treated with Bimekizumab: Long-Term Phase 2b/3 Results

Irene E. van der Horst-Bruinsma,[1] Matthew A. Brown,[2] Floris A. van Gaalen,[3] Nigil Haroon,[4] Lianne S. Gensler,[5] Alexander Marten,[6] Myriam Manente,[7] George Stojan,[8] Thomas Vaux,[9] Katy White,[9] Atul Deodhar,[10] Martin Rudwaleit[11]

[1]Department of Rheumatology, Radboud University Medical Centre, Nijmegen, The Netherlands; [2]Genomics England, London, UK; [3]Department of Rheumatology, Leiden University Medical Center, Leiden, The Netherlands; [4]Department of Medicine/Rheumatology, University Health Network, Schroeder Arthritis Institute, University of Toronto, Toronto, ON, Canada; [5]Department of Medicine/Rheumatology, University of California, San Francisco, CA, USA; [6]UCB, Monheim am Rhein, Germany; [7]UCB, Braine-l'Alleud, Belgium; [8]UCB, Atlanta, GA, USA; [9]UCB, Slough, UK; [10]Division of Arthritis & Rheumatic Diseases, Oregon Health & Science University, Portland, OR, USA; [11]Klinikum Bielefeld, University of Bielefeld, Bielefeld, Germany.

Presenting on behalf of the authors: Anna Parievsky

Objective

To assess the long-term incidence of uveitis in patients with axial spondyloarthritis (axSpA) or psoriatic arthritis (PsA) treated with bimekizumab (BKZ).

Introduction

- Acute anterior uveitis ('uveitis') is a common extra-musculoskeletal manifestation in patients with spondyloarthritis (SpA), and incidence varies with SpA type and disease duration.[1–4]
 - The prevalence of uveitis has been estimated as approximately 18–26% in patients with radiographic (r-)axSpA, and 12–16% in non-radiographic (nr-)axSpA.[2–6] The prevalence of uveitis in PsA has been estimated at around 5%.[3]
- Interleukin (IL)-17 has been implicated in the pathogenesis of uveitis; however, previous data have not demonstrated efficacy for IL-17A inhibition alone in managing the condition.[7,8]
- BKZ is a monoclonal IgG1 antibody that selectively inhibits IL-17F in addition to IL-17A.
 - As previously reported, the exposure-adjusted incidence rate (EAIR) per 100 patient-years (PY) of uveitis was lower in patients with axSpA randomized to BKZ (1.8/100 PY) vs placebo (15.4/100 PY) after 16 weeks.[9]

Methods

- Safety data are reported for two pools, each comprising three phase 2b/3 studies and their open-label extensions, in patients with active nr-axSpA and r-axSpA, and active PsA, respectively (**Figure 1**).
- Uveitis events were identified using the preferred terms "Autoimmune uveitis", "Iridocyclitis", "Iritis", and "Uveitis", coded according to MedDRA version 19.0; note that "Acute anterior uveitis" was not a specific preferred term available in MedDRA version 19.0 and events reported as "Acute anterior uveitis" were coded as "Iridocyclitis".
- Uveitis rates and EAIRs/100 PY for patients who received at least one BKZ 160 mg dose are reported over median durations of approximately 2.8 years (axSpA) and 2.7 years (PsA); the data cut-off for both patient populations was set at July 2023.
 - As uveitis is generally more prevalent in patients with axSpA than PsA,[2–6] results here have a greater focus on patients with axSpA.
 - To explore the relationship between time on BKZ treatment and incidence of uveitis, a Kaplan-Meier analysis of time to first uveitis event was performed in patients with axSpA.

Results

- The baseline characteristics of patients in the phase 2b/3 axSpA trial pool (N=848) and PsA trial pool (N=1,409) are shown in **Table 1**.
 - Of patients with axSpA, 130 (15.3%) had reported a history of uveitis; 21 (1.5%) patients with PsA had reported a history of uveitis.
 - Most patients with axSpA were HLA-B27 positive (717/848 [84.6%]).
- The baseline characteristics of patients with axSpA who had at least one uveitis event in the phase 2b/3 axSpA trials, and patients who did not, are presented in **Table 2**.
- In patients with axSpA across the pooled phase 2b/3 axSpA trial data, BKZ 160 mg exposure was 2,514 PY. Uveitis occurred in 31/848 (3.7%) patients overall (EAIR [95% CI]: 1.3/100 PY [0.9, 1.8]; **Figure 2**).
 - In those with a history of uveitis, 18/130 (13.8%) patients had uveitis events (4.8/100 PY [2.8, 7.6]).
 - In patients without a history of uveitis, 13/718 (1.8%) had uveitis events (0.6/100 PY [0.3, 1.1]).
 - All events were mild/moderate, one led to treatment discontinuation.
- Kaplan-Meier analyses suggest no clear trend between the time of the first BKZ administration and the first incidence of uveitis in patients with axSpA (**Figure 3**).
- In patients with PsA across the pooled phase 2b/3 PsA trial data, BKZ 160 mg exposure was 3,656 PY. Overall incidence of uveitis was low, occurring in three patients (0.2%; EAIR [95% CI]: 0.1/100 PY [0.0, 0.2]; **Figure 4**).
 - In patients with a history of uveitis, 1/21 (4.8%) had a uveitis event (2.4/100 PY [0.1, 13.5]).
 - In patients without a history of uveitis, 2/1,388 (0.1%) had uveitis events (0.1/100 PY [0.0, 0.2]).
 - All events were mild/moderate, no uveitis events led to treatment discontinuation.
- Insufficient uveitis events were reported to justify conducting Kaplan-Meier analyses in patients with PsA.

Conclusions

Previous reported data have demonstrated that the incidence of uveitis is lower in patients with axSpA randomized to bimekizumab vs placebo after 16 weeks of treatment. Across 2,514 PY in patients with axSpA and 3,656 PY in patients with PsA, the long-term data suggest that the incidence of uveitis in patients treated with bimekizumab remains low. This suggests that bimekizumab may confer protective effects for uveitis in patients with SpA.

Summary

Previous reports have indicated that the EAIR of uveitis was lower in patients with axSpA randomized to bimekizumab (1.8/100 PY) vs placebo (15.4/100 PY) after 16 weeks.

This analysis evaluated the **long-term incidence of uveitis in patients with axSpA and PsA treated with bimekizumab** across pooled phase 2b/3 studies.

- Long-term data from **2,514 PY** in axSpA, and **3,656 PY** in PsA were analyzed
- Overall, the **incidence of uveitis** was low in **both bimekizumab-treated patient populations**, with rates of 1.3/100 PY in patients with axSpA and 0.1/100 PY in patients with PsA
- Kaplan-Meier analysis showed **no clear trend between the time of the first bimekizumab administration** and the **onset of uveitis** in patients with axSpA
- These findings, together with previous reports, suggest that **bimekizumab may confer protective effects** against uveitis in patients with SpA

Figure 1 The two safety pools (axSpA, PsA) of patients treated with BKZ 160 mg Q4W across six phase 2b/3 studies and their OLEs

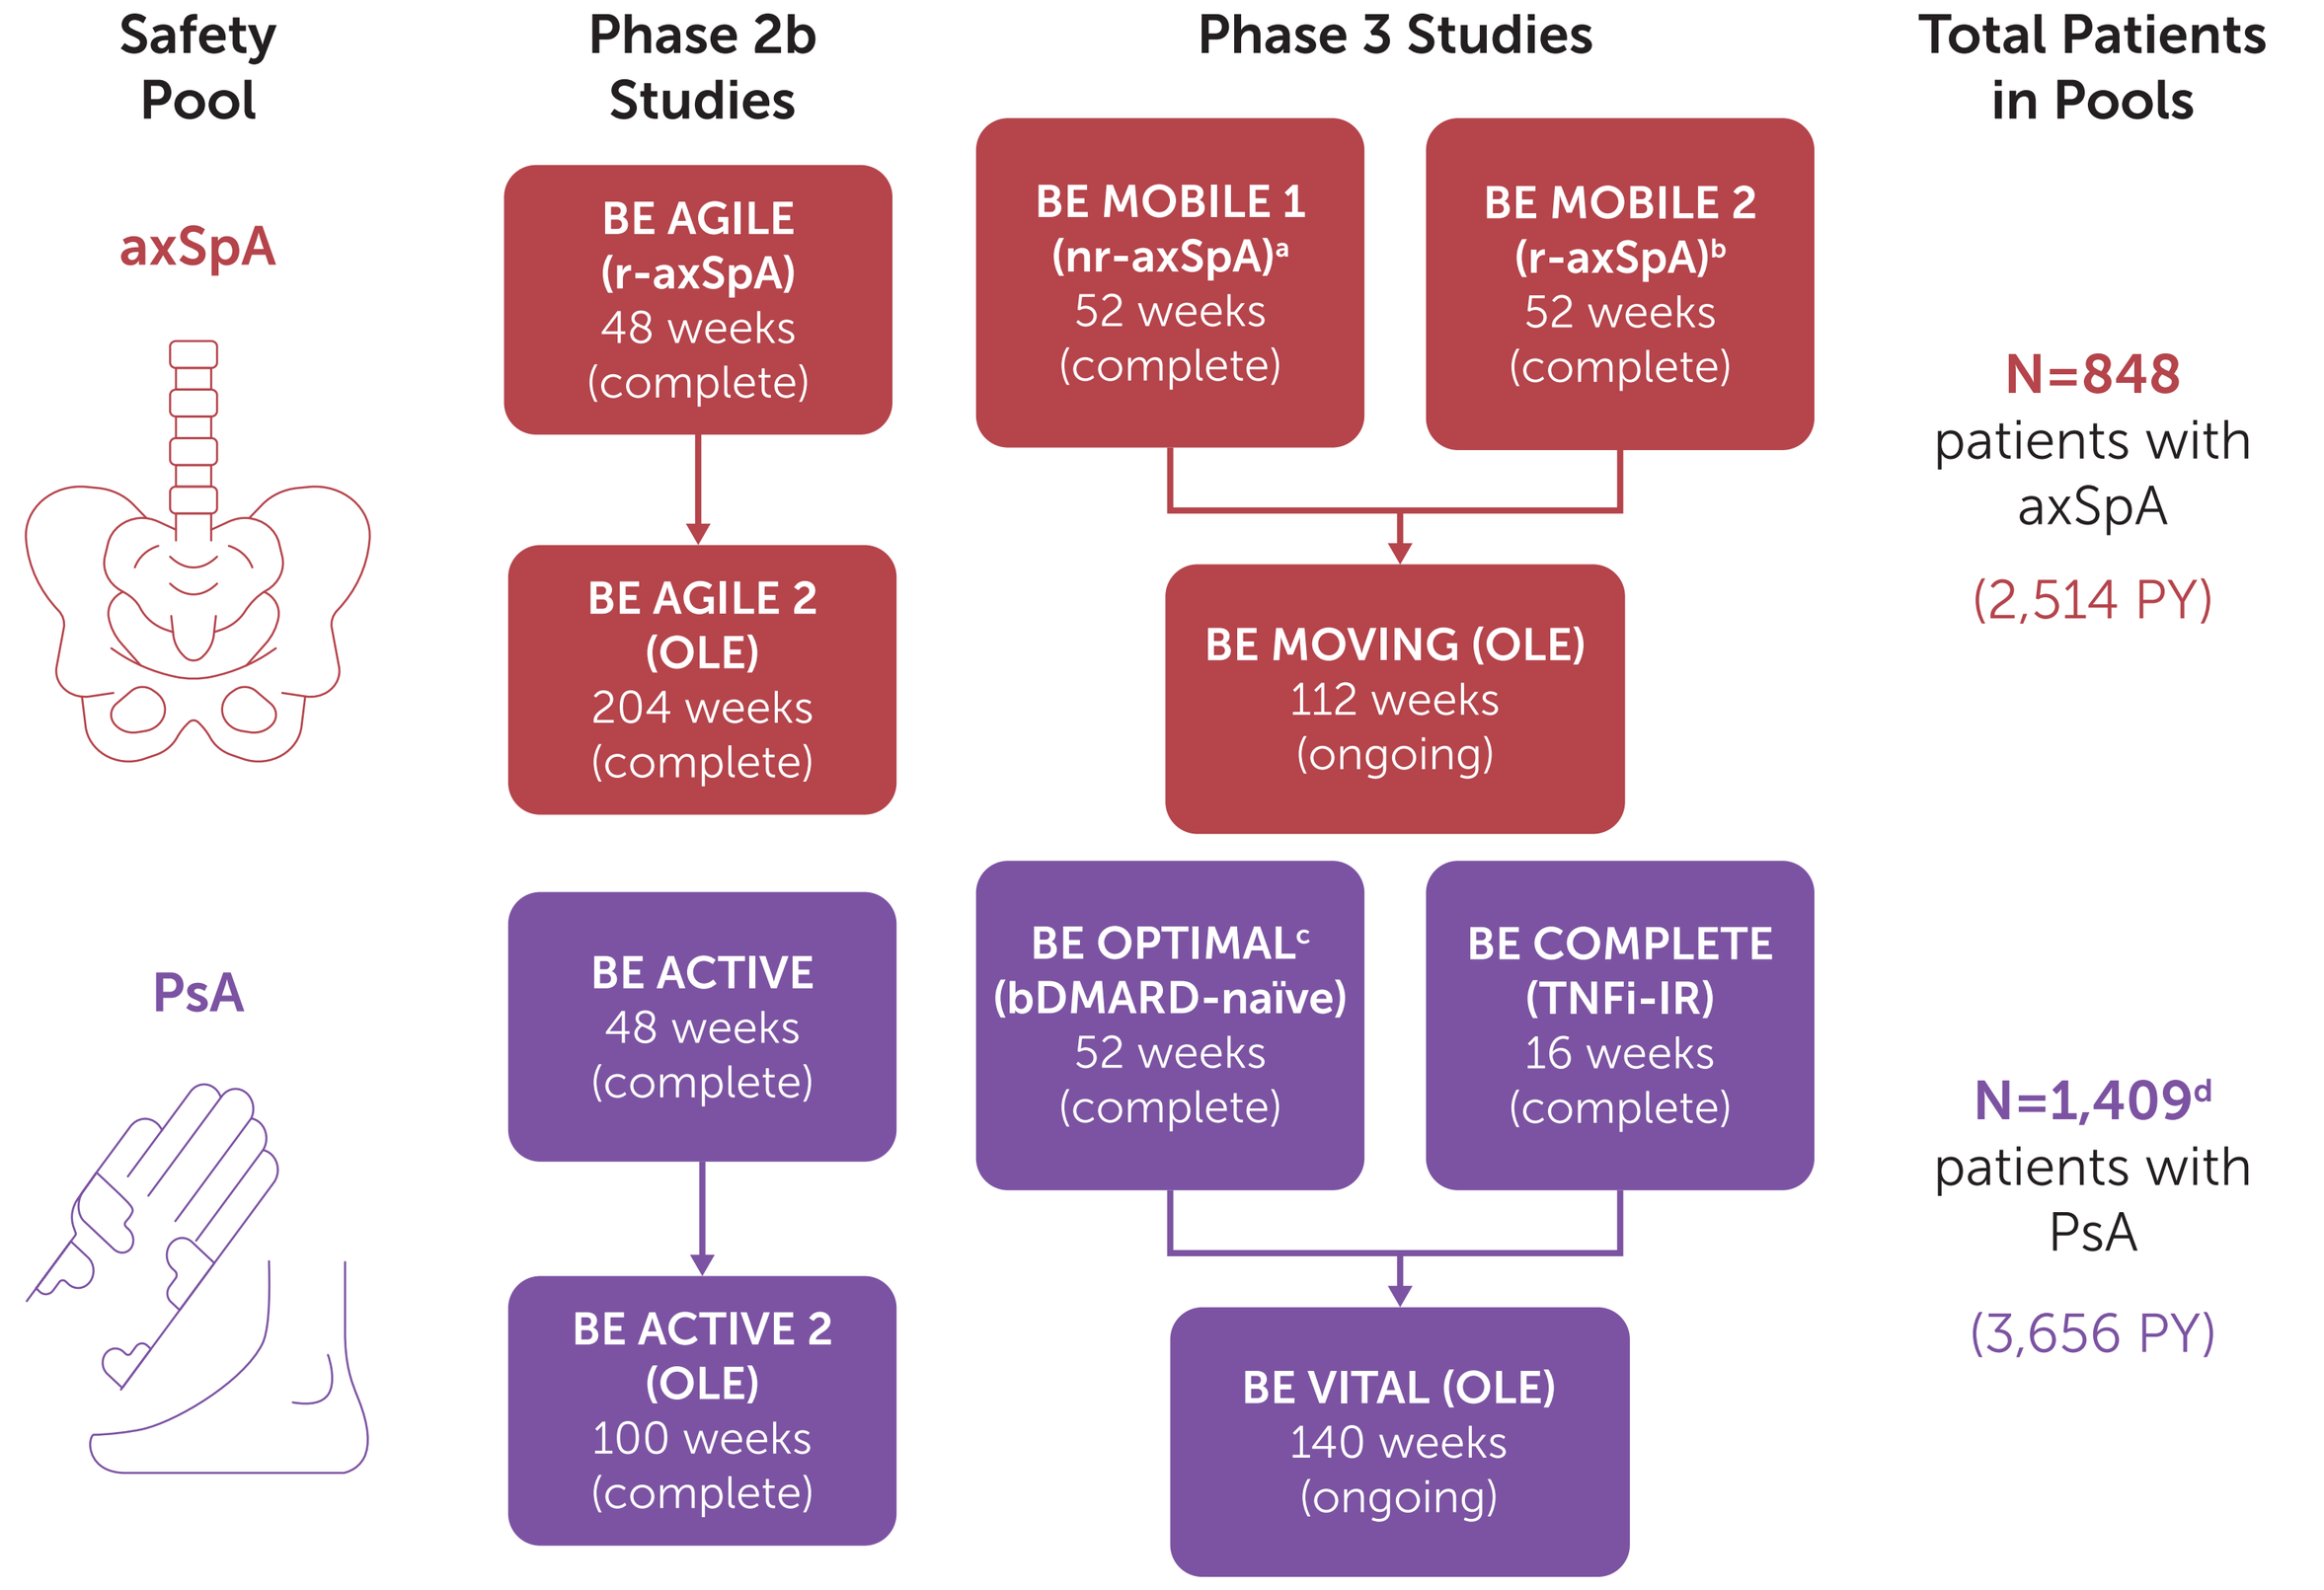
Safety Pool
Phase 2b Studies
Phase 3 Studies
Total Patients in Pools
axSpA
BE AGILE (r-axSpA) 48 weeks (complete)
BE AGILE 2 (OLE) 204 weeks (complete)
BE MOBILE 1 (nr-axSpA)a 52 weeks (complete)
BE MOBILE 2 (r-axSpA)b 52 weeks (complete)
BE MOVING (OLE) 112 weeks (ongoing)
N=848 patients with axSpA
(2,514 PY)
PsA
BE ACTIVE 48 weeks (complete)
BE ACTIVE 2 (OLE) 100 weeks (complete)
BE OPTIMALc (bDMARD-naïve) 52 weeks (complete)
BE COMPLETE (TNFi-IR) 16 weeks (complete)
BE VITAL (OLE) 140 weeks (ongoing)
N=1,409d patients with PsA
(3,656 PY)

Data from the July 2023 data-cut shown, including all patients who received ≥1 dose of BKZ 160 mg Q4W in the phase 2b/3 studies. Duration of overall treatment period shown; BKZ treatment duration by data-cut varied between patients, depending on study duration and initial randomization in the feeder studies. **[a]** Patients with nr-axSpA met Assessment of SpondyloArthritis international Society (ASAS) classification criteria. Patients with radiographic sacroiliitis were excluded; **[b]** Patients with r-axSpA met modified New York criteria and fulfilled ASAS classification criteria; **[c]** BE OPTIMAL also included an adalimumab treatment arm. Data from patients treated with adalimumab were not included in the PsA safety pool prior to switching to BKZ, but were included following the switch; **[d]** Since the previous data-cut (July 2022),[11] two additional patients started receiving BKZ in BE VITAL (originally randomized to adalimumab).

BE AGILE: NCT02963506; BE AGILE 2: NCT03355573; BE MOBILE 1: NCT03928704; BE MOBILE 2: NCT03928743; BE MOVING: NCT04436640; BE ACTIVE: NCT02969525; BE ACTIVE 2: NCT03347110; BE OPTIMAL: NCT03895203; BE COMPLETE: NCT03896581; BE VITAL: NCT04009499.

Table 1 Baseline demographics and characteristics in the pooled axSpA and PsA phase 2b/3 trials

	axSpA	PsA
	BKZ 160 mg Q4W (N=848)	**BKZ 160 mg Q4W (N=1,409)**
Age, years, mean (SD)	40.3 (11.9)	49.3 (12.4)
Male, n (%)	606 (71.5)	672 (47.7)
r-axSpA, n (%)	604 (71.2)	N/A
BMI, kg/m², mean (SD)	27.0 (5.5)	29.4 (6.3)
HLA-B27 positive, n (%)	717 (84.6)[a]	Not available
Geographic region, n (%)		
Asia	88 (10.4)	99 (7.0)
Eastern Europe	543 (64.0)	908 (64.4)
North America	37 (4.4)	238 (16.9)
Western Europe	180 (21.2)	164 (11.6)
Time since first symptoms, years, mean (SD)	12.4 (9.9)	Not available
Time since first diagnosis, years, mean (SD)	6.1 (7.8)	7.0 (8.0)
History of uveitis, n (%)	130 (15.3)	21 (1.5)
Prior TNFi exposure, n (%)	108 (12.7)	425 (30.2)
Baseline concomitant medications, n (%)		
csDMARDs	197 (23.2)	909 (64.5)
NSAIDs	689 (81.3)	834 (59.2)
Corticosteroids	70 (8.3)	219 (15.5)
CRP, mg/L, geometric mean (geometric CV, %)	7.1 (238.1)	4.2 (254.1)
CRP > or ≥5 mg/L, n (%)[b]	545 (64.3)	645 (45.8)

Pooled axSpA and PsA safety sets. **[a]** Six patients had missing HLA-B27 status; **[b]** CRP >5 mg/L recorded for patients with axSpA and ≥5 mg/L recorded for patients with PsA.

Table 2 Baseline demographics and characteristics split by patients with and without a uveitis event in the pooled axSpA phase 2b/3 trials

	axSpA	
	Patients with a uveitis event (N=31)	**Patients without a uveitis event (N=817)**
Age, years, mean (SD)	39.4 (11.9)	40.3 (11.9)
Male, n (%)	20 (64.5)	586 (71.7)
r-axSpA, n (%)	26 (83.9)	578 (70.7)
BMI, kg/m², mean (SD)	26.9 (5.3)	27.0 (5.5)
HLA-B27 positive, n (%)	29 (93.5)[a]	688 (84.2)[b]
Geographic region, n (%)		
Asia	4 (12.9)	84 (10.3)
Eastern Europe	22 (71.0)	521 (63.8)
North America	2 (6.5)	35 (4.3)
Western Europe	3 (9.7)	177 (21.7)
Time since first symptoms, years, mean (SD)	14.8 (10.6)	12.3 (9.9)
Time since first diagnosis, years, mean (SD)	7.2 (9.3)	6.0 (7.7)
Prior TNFi exposure, n (%)	5 (16.1)	103 (12.6)
Baseline concomitant medications, n (%)		
csDMARDs	7 (22.6)	190 (23.3)
NSAIDs	27 (87.1)	662 (81.0)
Corticosteroids	4 (12.9)	66 (8.1)
CRP, mg/L, geometric mean (geometric CV, %)	9.4 (208.7)	7.1 (239.2)
CRP >5 mg/L, n (%)	21 (67.7)	524 (64.1)

Pooled axSpA safety set. **[a]** One patient had missing HLA-B27 status; **[b]** Five patients had missing HLA-B27 status.

Figure 2 Incidence of uveitis (EAIR/100 PY [95% CI]) in patients with axSpA stratified by history of uveitis

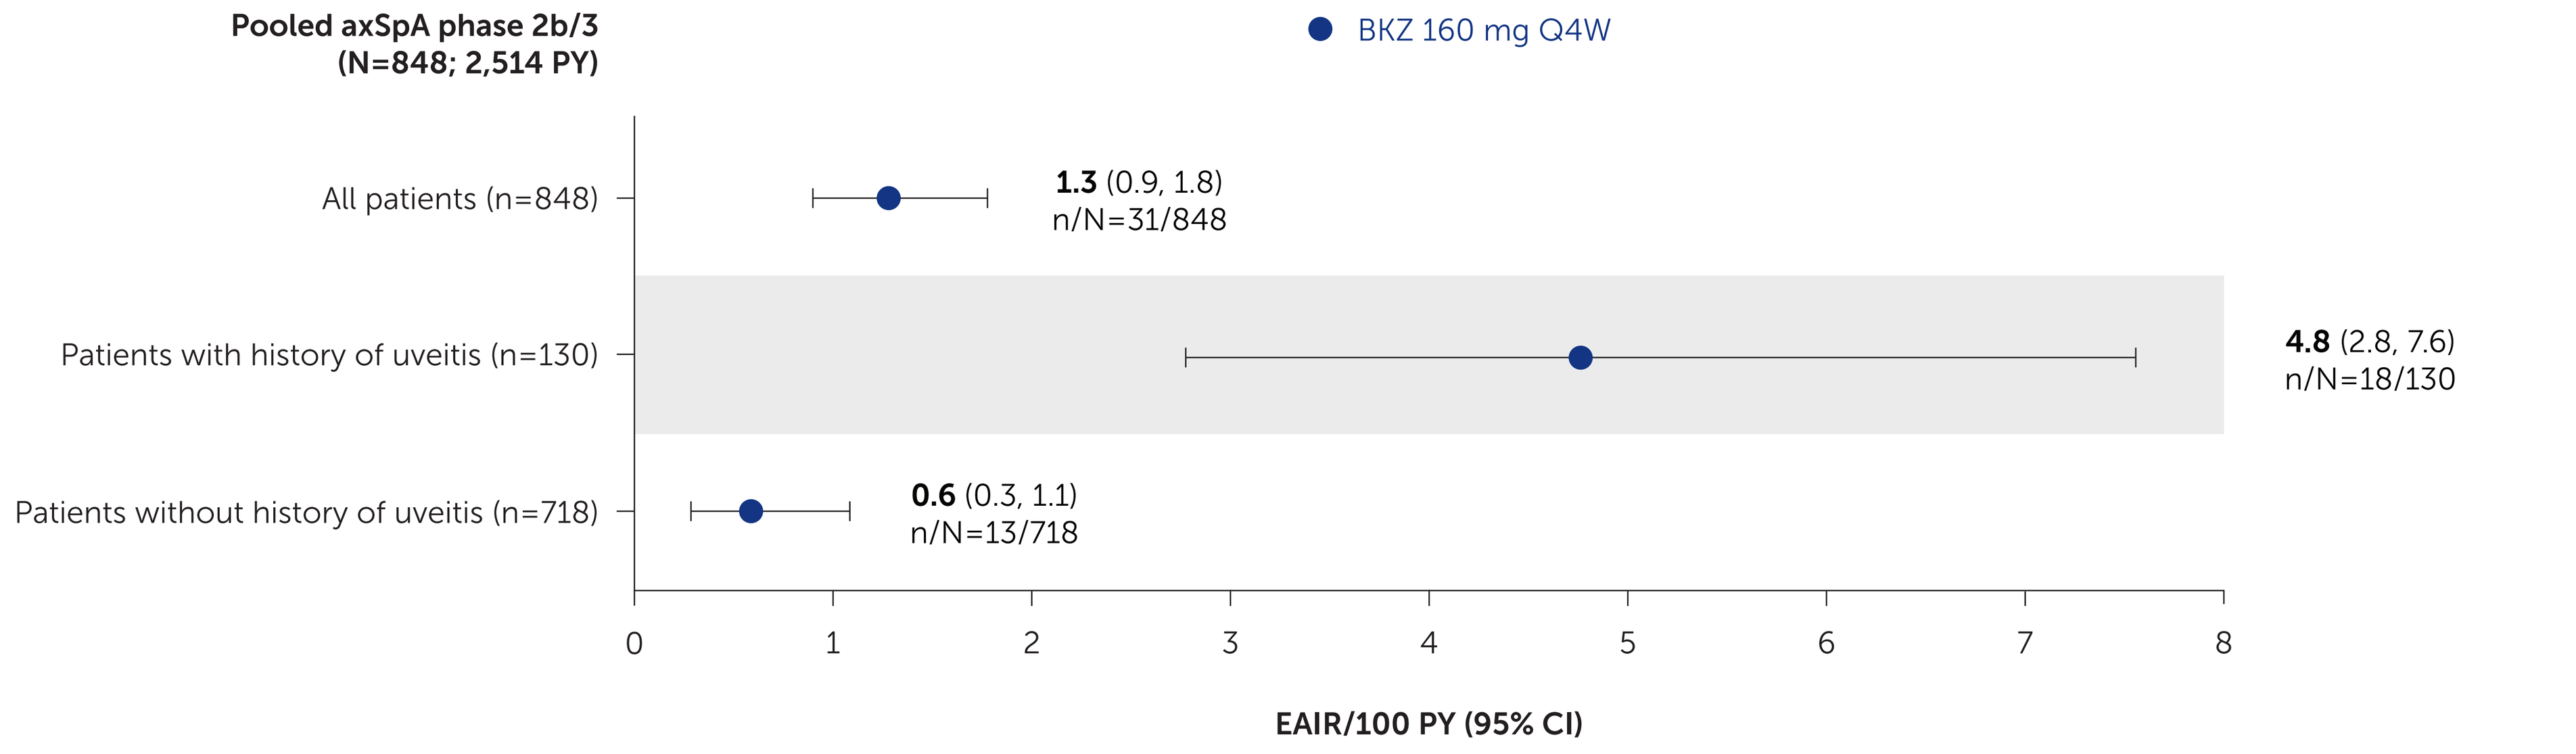
Pooled axSpA phase 2b/3 (N=848; 2,514 PY)
BKZ 160 mg Q4W
All patients (n=848)
1.3 (0.9, 1.8)
n/N=31/848
Patients with history of uveitis (n=130)
4.8 (2.8, 7.6)
n/N=18/130
Patients without history of uveitis (n=718)
0.6 (0.3, 1.1)
n/N=13/718
0
1
2
3
4
5
6
7
8
EAIR/100 PY (95% CI)

Pooled axSpA safety set. n/N is the number of patients who had at least one uveitis TEAE/total number of patients.

Figure 3 Kaplan-Meier estimates for time to first uveitis events in patients with axSpA

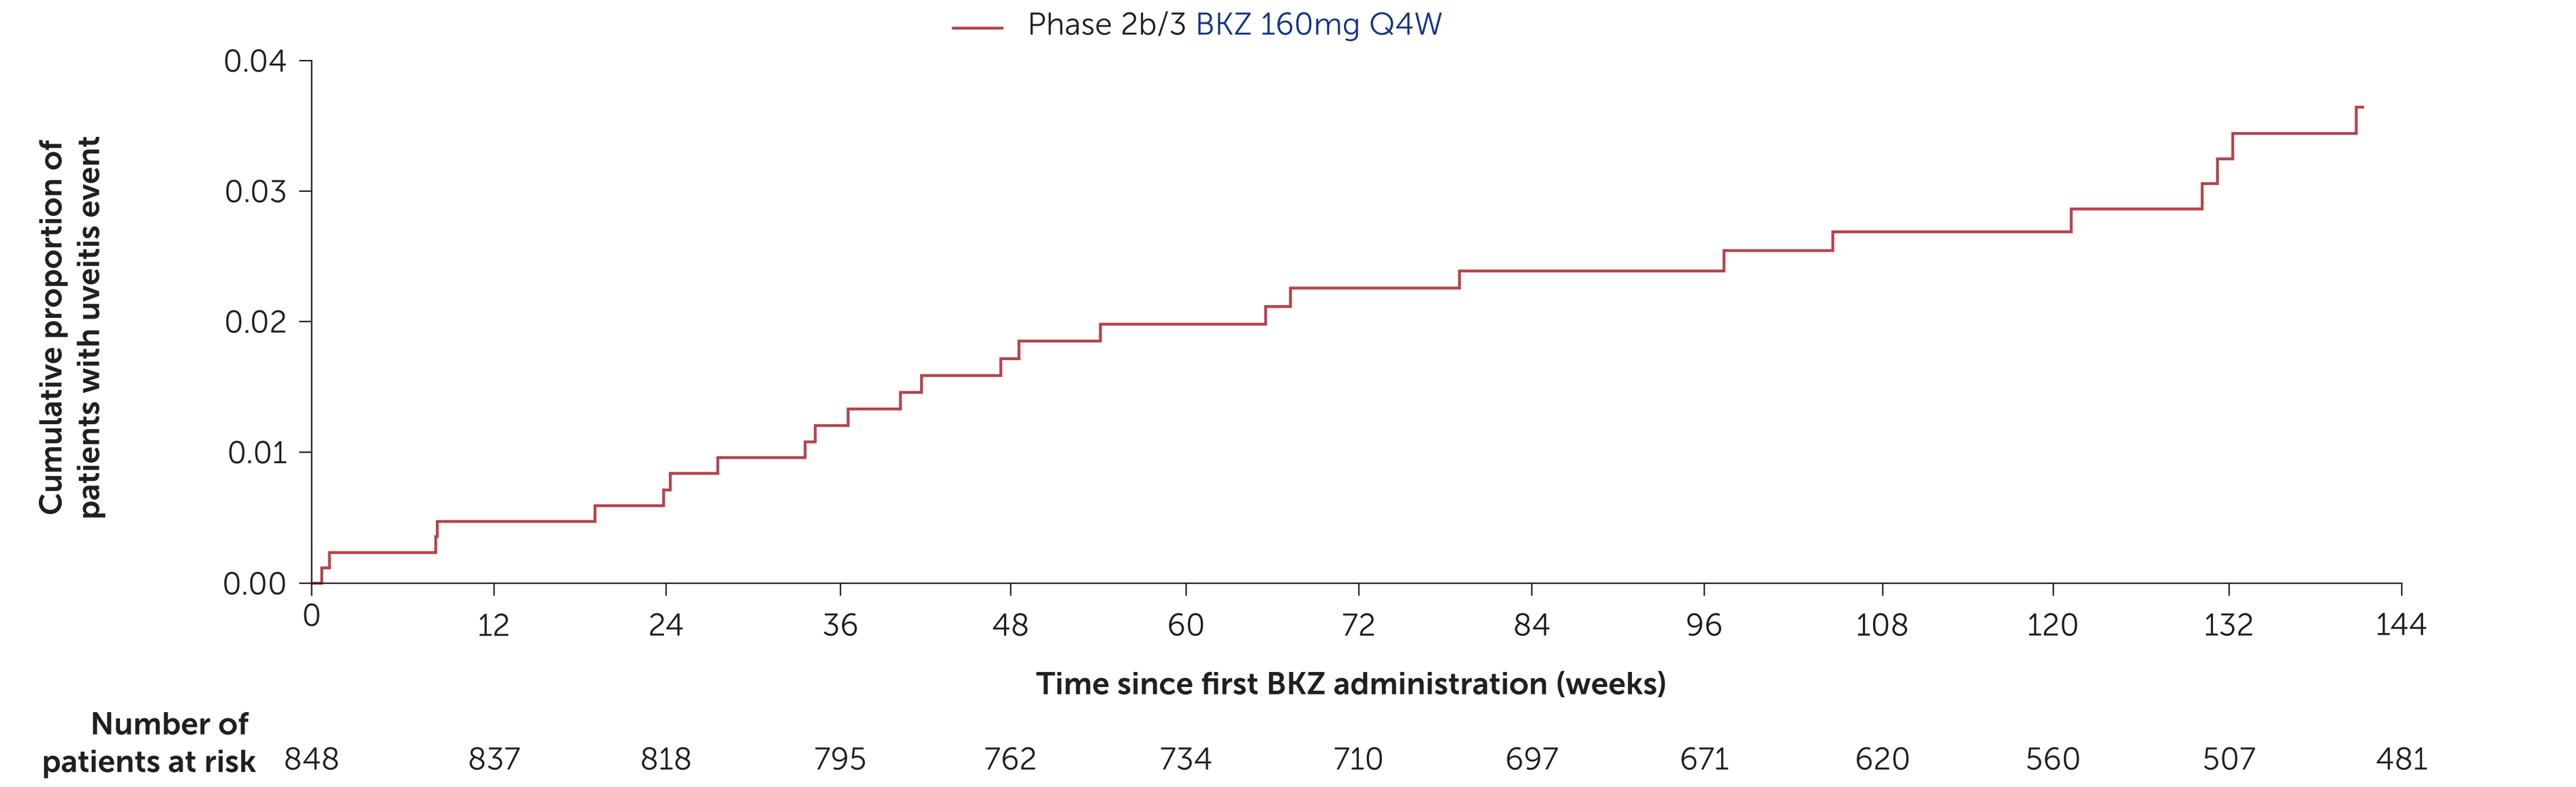
Phase 2b/3 BKZ 160mg Q4W
Cumulative proportion of patients with uveitis event
0.04
0.03
0.02
0.01
0.00
0
12
24
36
48
60
72
84
96
108
120
132
144
Time since first BKZ administration (weeks)
Number of patients at risk
848
837
818
795
762
734
710
697
671
620
560
507
481

Pooled axSpA safety set. Includes all patients who received BKZ 160 mg Q4W. Time to first occurrence of uveitis (weeks) was calculated as (date of first onset of uveitis − date of first BKZ 160 mg administration + 1)/7.

Figure 4 Incidence of uveitis (EAIR/100 PY [95% CI]) in patients with PsA stratified by history of uveitis

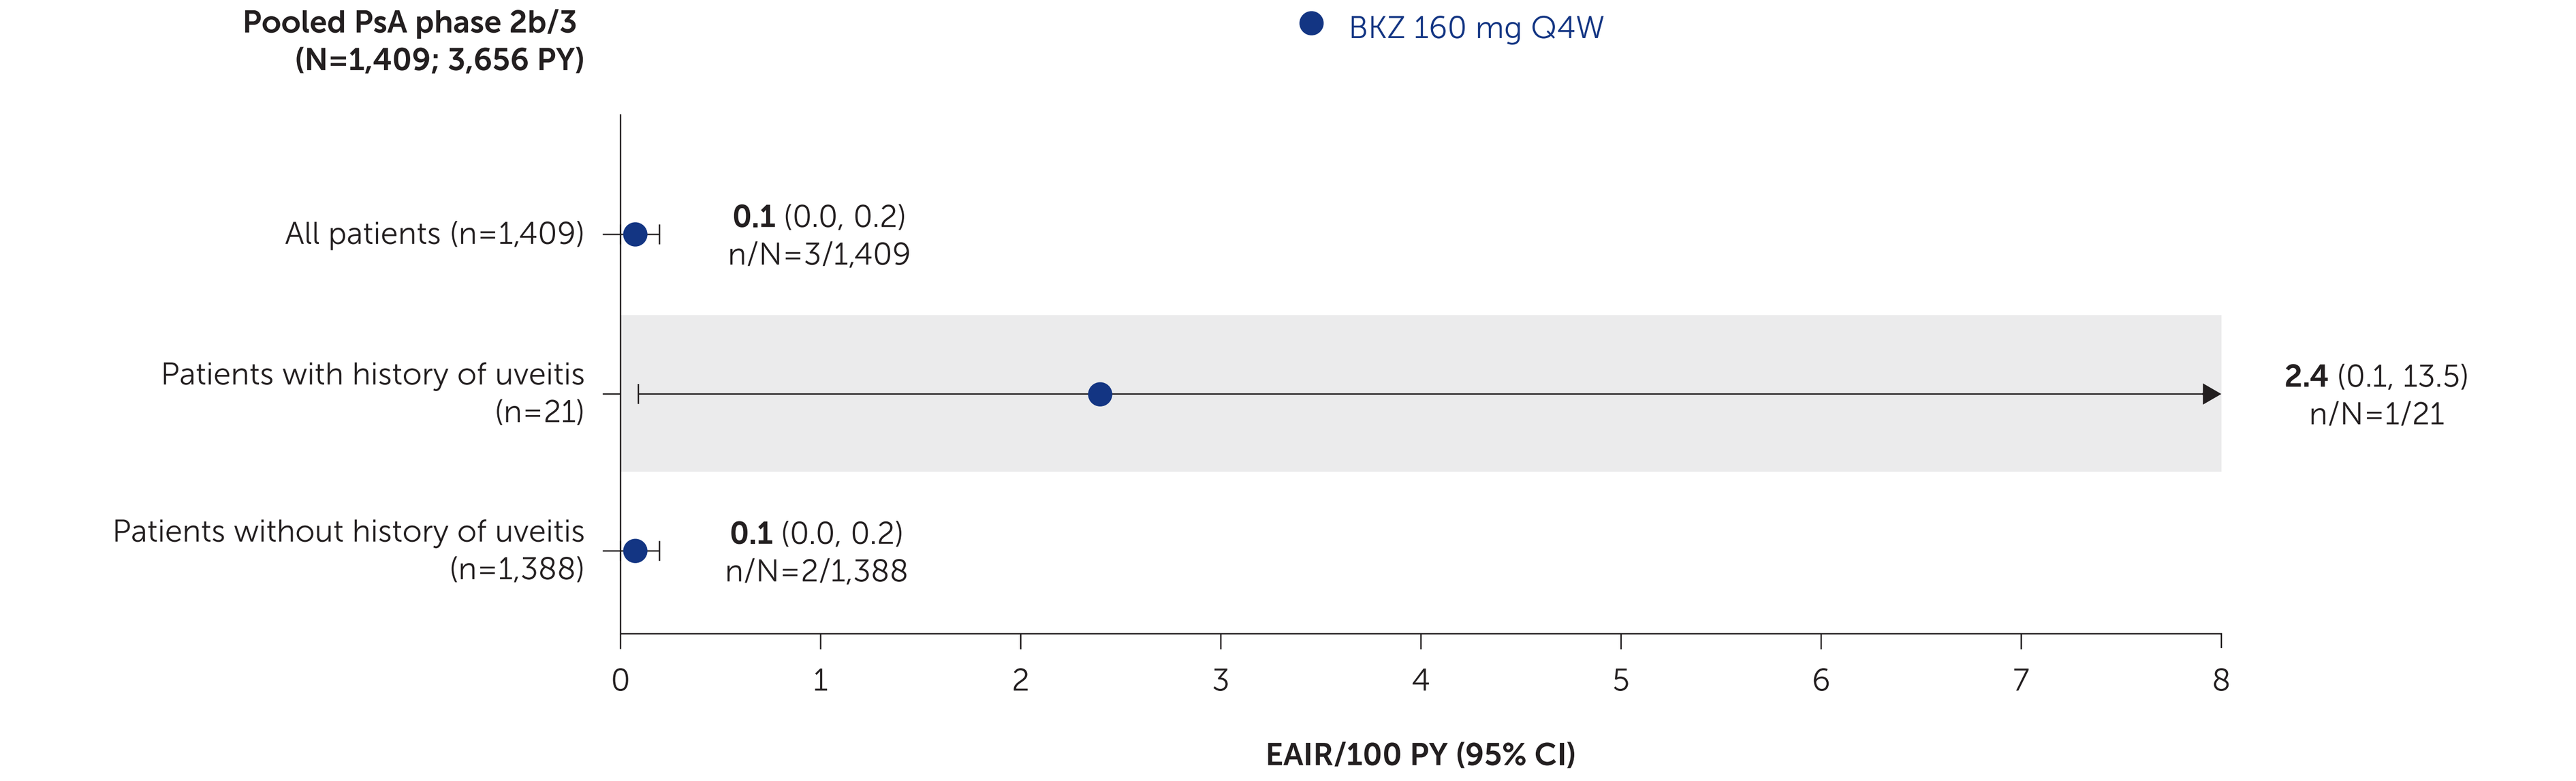
Pooled PsA phase 2b/3 (N=1,409; 3,656 PY)
BKZ 160 mg Q4W
All patients (n=1,409)
0.1 (0.0, 0.2)
n/N=3/1,409
Patients with history of uveitis (n=21)
2.4 (0.1, 13.5)
n/N=1/21
Patients without history of uveitis (n=1,388)
0.1 (0.0, 0.2)
n/N=2/1,388
0
1
2
3
4
5
6
7
8
EAIR/100 PY (95% CI)

Pooled PsA safety set. n/N is the number of patients who had at least one uveitis TEAE/total number of patients.

AS: ankylosing spondylitis; **ASAS:** Assessment of SpondyloArthritis International Society; **axSpA:** axial spondyloarthritis; **bDMARD:** biologic disease-modifying antirheumatic drug; **BKZ:** bimekizumab; **BMI:** body mass index; **CI:** confidence interval; **CRP:** C-reactive protein; **csDMARD:** conventional synthetic disease-modifying antirheumatic drug; **CV:** coefficient of variation; **EAIR:** exposure-adjusted incidence rate; **HLA:** human leukocyte antigen; **MedDRA:** Medical Dictionary for Regulatory Activities; **nr-axSpA:** non-radiographic axSpA; **NSAID:** nonsteroidal anti-inflammatory drug; **OLE:** open-label extension; **PsA:** psoriatic arthritis; **PY:** patient-year; **Q4W:** every 4 weeks; **r-axSpA:** radiographic axSpA; **SD:** standard deviation; **SpA:** spondyloarthritis; **TNFi:** tumor necrosis factor inhibitor; **TNFi-IR:** TNFi inadequate response.

References: [1]Robinson PC. Arthritis Rheumatol. 2015;67:140–51; [2]López-Medina C. RMD Open 2019;5:e001108; [3]Delmás A. RMD Open 2023;9:e002781; [4]Stolwijk C. Ann Rheum Dis. 2015;74:65–73; [5]de Winter JJ. Arthritis Res Ther. 2016;18:196; [6]Rudwaleit M. Arthritis Rheum 2009;60:717–27; [7]Dick AD. J. Ophthalmol. 2013;120:777–87; [8]Kwon OC. Rheumatol. 2024;keae003; [9]Brown MA. Ann Rheum Dis. 2024;1–9; [10]Baraliakos X. Ann Rheum Dis. 2024;83:199–213; [11]Mease PJ. Arthritis Rheumatol. 2023;75(suppl 9). Abstract 0511. **Author Contributions**: Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **IEvdHB**, **MAB**, **FAvG**, **NH**, **LSG**, **AM**, **MM**, **GS**, **TV**, **KW**, **AD**, **MR**; Drafting of the publication, or reviewing it critically for important intellectual content: **IEvdHB**, **MAB**, **FAvG**, **NH**, **LSG**, **AM**, **MM**, **GS**, **TV**, **KW**, **AD**, **MR**; Final approval of the publication: **IEvdHB**, **MAB**, **FAvG**, **NH**, **LSG**, **AM**, **MM**, **GS**, **TV**, **KW**, **AD**, **MR**. **Disclosures: IHB:** Consultant for AbbVie, Eli Lilly, MSD, Novartis, and UCB; unrestricted grants received for investigator-initiated studies from AbbVie, MSD, Pfizer, and UCB; fees received for lectures from AbbVie, BMS, MSD, and Pfizer. **MAB:** Grant/Research support from UCB; consultant for Altis Medicines, Clementia, Grey Wolf Therapeutics, Incyte, Ipsen, Regeneron, and Xinthera. **FAvG:** Grants from Jacobus Stichting, Novartis, Stichting ASAS, Stichting Vrienden van Sole Mio, and UCB; fees from Novartis; personal fees from AbbVie, BMS, Eli Lilly, and MSD. **NH:** Consultant for AbbVie, Eli Lilly, Novartis, and UCB. **LSG:** Consultant for Acelyrin, Eli Lilly, Janssen, Novartis, Pfizer, and UCB; grants from UCB paid to institution. **AM, AP, GS:** Employees of UCB. **MM, TV, KW:** Employees and shareholders of UCB. **AD:** Speaker for Eli Lilly, Janssen, Novartis, Pfizer, and UCB; consulting fees from BMS, Eli Lilly, Janssen, MoonLake Immunotherapeutics, Novartis, Pfizer, and UCB; grant/research support from BMS, Eli Lilly, Janssen, Novartis, Pfizer, and UCB. **MR:** Speakers bureau from AbbVie, Boehringer Ingelheim, Eli Lilly, Janssen, Novartis, Pfizer, and UCB; consultant of AbbVie, Eli Lilly, Janssen, Novartis, and UCB. **Acknowledgments:** We would like to thank the patients and their caregivers in addition to all the investigators and their teams who contributed to these studies. The authors acknowledge Celia Menckeberg, PhD, UCB, Breda, The Netherlands, and Heather Edens, PhD, UCB, Smyrna, GA, USA for publication coordination, Isabel Raynaud, MBBS iBSc, Costello Medical, Cambridge, UK, for medical writing and editorial assistance, and the Costello Medical Creative team for graphic design assistance. These studies were funded by UCB. All costs associated with development of this presentation were funded by UCB.

To receive a copy of this poster, scan the QR code.
Link expiration: October 5, 2025

Presented at CCR West 2025 | September 18–21 | Huntington Beach, CA

Previously presented at ACR 2024