Dapirolizumab Pegol Demonstrated Significant Improvement in Systemic Lupus Erythematosus Disease Activity: Efficacy and Safety Results of a Phase 3 Trial

Megan E. B. Clowse,[1] David A. Isenberg,[2] Joan T. Merrill,[3] Thomas Dörner,[4] Michelle Petri,[5] Edward Vital,[6,7] Eric F. Morand,[8] Teri Jimenez,[9] Stephen Brookes,[10] Janine Gaiha-Rohrbach,[11] Christophe Martin,[12] Annette Nelde,[13] Christian Stach[14]

Presenter: Scott Case[15]

[1]Division of Rheumatology and Immunology, Duke University, Durham, NC, USA; [2]Department of Ageing, Rheumatology and Regenerative Medicine, Division of Medicine, University College London, London, UK; [3]Oklahoma Medical Research Foundation, Oklahoma City, OK, USA; [4]Department of Medicine/Rheumatology and Clinical Immunology, Charité Universitätsmedizin Berlin, Berlin, Germany; [5]Johns Hopkins University School of Medicine, Baltimore, MD, USA; [6]Leeds Institute of Rheumatic and Musculoskeletal Medicine, University of Leeds, Leeds, UK; [7]NIHR Leeds Biomedical Research Centre, Leeds Teaching Hospitals NHS Trust, Leeds, UK; [8]Centre for Inflammatory Diseases, Monash University, Melbourne, Australia; [9]UCB, Raleigh, NC, USA; [10]Biogen, Maidenhead, UK; [11]Biogen, Cambridge, MA, USA; [12]UCB, Slough, UK; [13]Biogen, Baar, Switzerland; [14]UCB, Monheim am Rhein, Germany; [15]Biogen, Malvern, PA, USA

To receive a copy of this poster, scan the QR code
Link expiration: 20 December 2025

Objective

- **To report results of the phase 3 PHOENYCS GO trial (NCT04294667) which evaluated the efficacy and safety of dapirolizumab pegol (DZP) in patients with systemic lupus erythematosus (SLE).**

Background

- DZP is a novel CD40L inhibitor with broad modulatory effects on SLE immunopathology;[1,2] it consists of a polyethylene glycol (PEG)-conjugated antigen-binding fragment (Fab'), which lacks an Fc domain.

Methods

- PHOENYCS GO was a 48-week, global, randomized, double-blind, placebo (PBO)-controlled trial. After the treatment period, patients could enter an open-label extension (OLE) or complete a 6-week safety follow-up (SFU; **Figure 1**).
 - Patients aged ≥16 years with moderate-to-severe, active SLE characterized by persistently active or frequently flaring/relapsing-remitting disease activity despite stable standard of care (SOC) medication (antimalarials, glucocorticoids, and/or immunosuppressants) were included.
- The primary endpoint was BICLA response at Week 48.

Figure 1 PHOENYCS GO study design

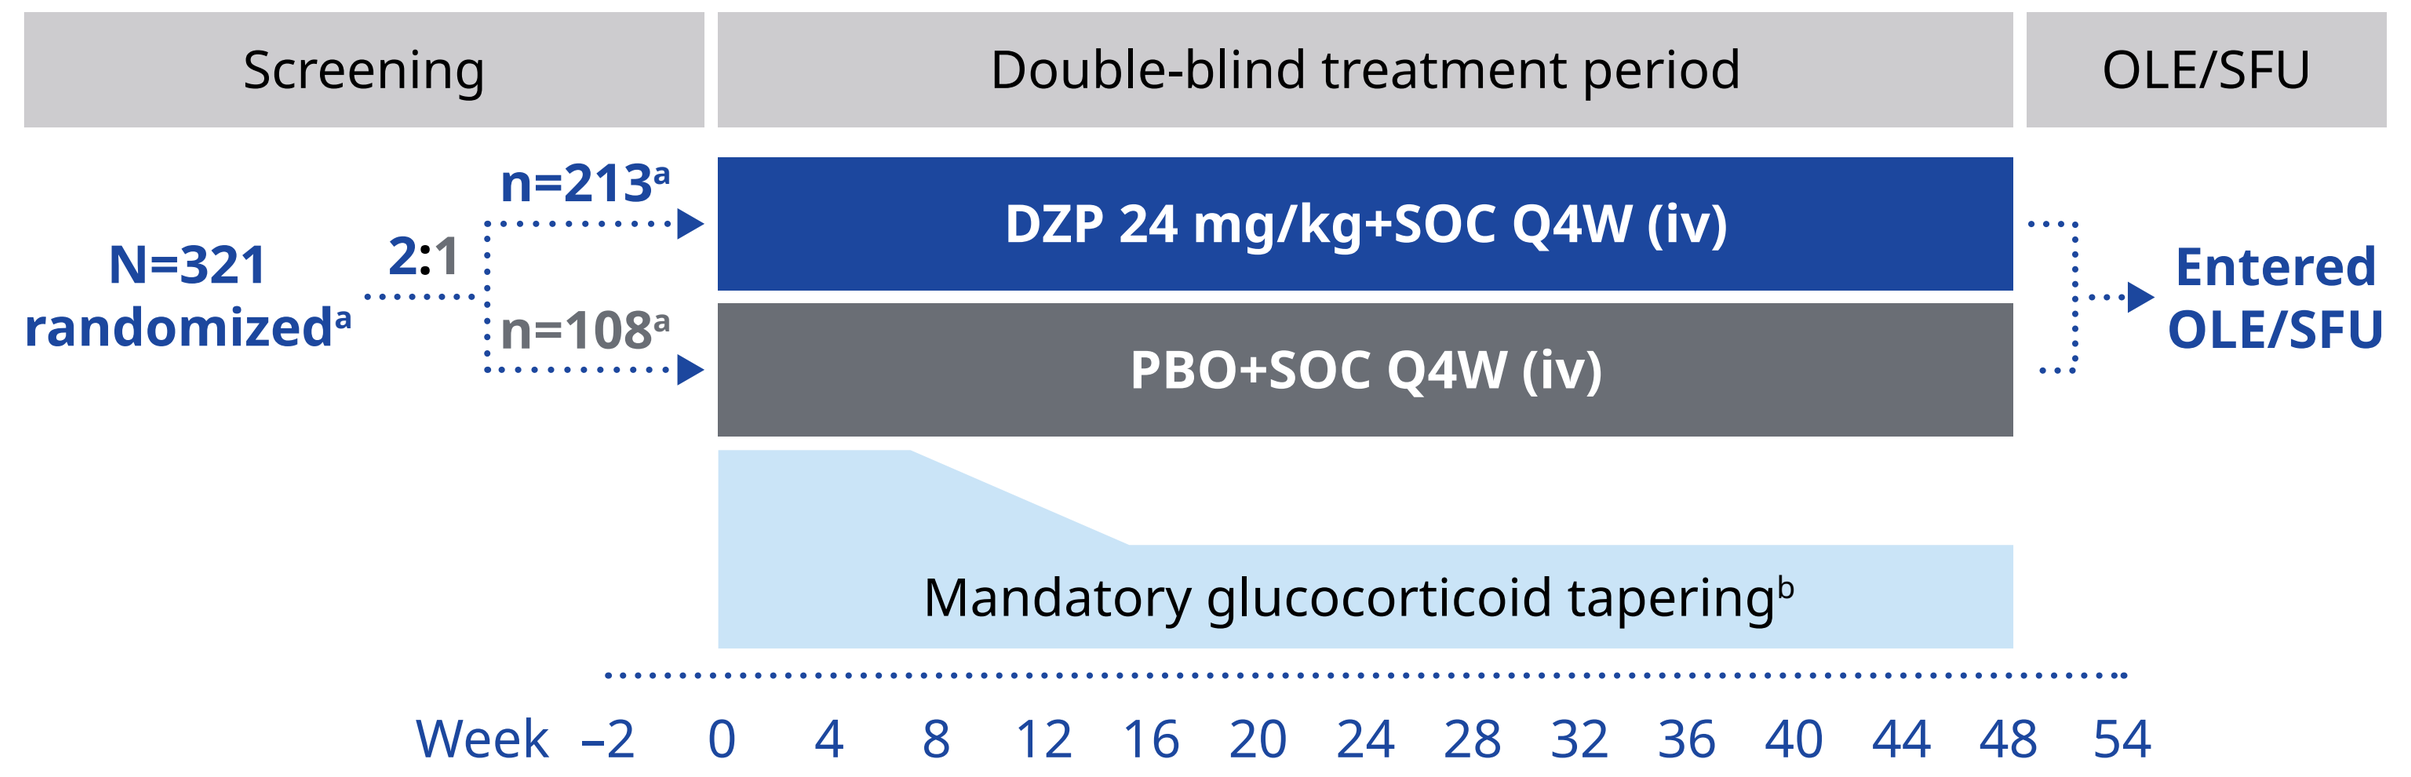
Screening
Double-blind treatment period
OLE/SFU
N=321 randomized[a]
2:1
n=213[a]
n=108[a]
DZP 24 mg/kg+SOC Q4W (iv)
PBO+SOC Q4W (iv)
Entered OLE/SFU
Mandatory glucocorticoid tapering[b]
Week –2 0 4 8 12 16 20 24 28 32 36 40 44 48 54

[a] Randomized set; **[b]** Investigators were required to initiate glucocorticoid tapering for patients with a dose >7.5 mg/day prednisone equivalent at baseline with the goal of reaching ≤7.5 mg/day, in line with EULAR 2019 treatment guidelines,[3] with tapering starting no later than Week 8. Guidance was provided on tapering, but the exact tapering regimen was at the discretion of the investigator and adapted to the individual patient's disease state. Tapering between Week 44 and 48 was avoided.

Results

- In total, 85.4% (182/213) of randomized patients receiving DZP+SOC and 79.6% (86/108) receiving PBO+SOC completed the study to Week 48 on treatment.
- Baseline characteristics in both treatment groups were typical for a moderate-to-severe SLE population (full analysis set; DZP+SOC: n=208; PBO+SOC: n=107; **Table 1**).
- **The primary endpoint was met; significantly more patients receiving DZP+SOC had BICLA response at Week 48 versus PBO+SOC** (p=0.0110; **Figure 2**).
 - The first secondary endpoint, BICLA response at Week 24, was not met.
- A nominally significant difference in glucocorticoid tapering to ≤7.5 mg/day from baseline dose >7.5 mg/day between DZP+SOC and PBO+SOC was seen at Week 48, with differences observed as early as Week 8 (**Figure 3**).
- SRI-4 response at Weeks 16–48 was achieved by a greater proportion of patients receiving DZP+SOC versus PBO+SOC (**Figure 4a**).
- At Weeks 16–48, a greater improvement from baseline in SLEDAI-2K was seen in patients receiving DZP+SOC versus PBO+SOC (**Figure 4b**).
- Through Week 48, fewer patients receiving DZP+SOC versus PBO+SOC had severe BILAG flares (**Figure 4c**).
- At Weeks 32–48, more patients receiving DZP+SOC versus PBO+SOC achieved LLDAS (**Figure 4d**).
 - Through 48 weeks, 23.6% (49/208) of patients receiving DZP+SOC achieved LLDAS in ≥50% of visits, compared with 15.9% (17/107) of patients receiving PBO+SOC (nominal p=0.1042).
- DZP was generally well tolerated; the safety profile was generally consistent with previous DZP studies (**Table 2**).[4–6]

Conclusions

PHOENYCS GO met its primary endpoint of BICLA response at Week 48.

Treatment with DZP, a novel CD40L inhibitor, was associated with consistent improvements across multiple measures of disease activity, alongside successful glucocorticoid tapering, in patients with SLE.

DZP was generally well tolerated.

A second phase 3 trial (PHOENYCS FLY; NCT06617325) has been initiated and is currently enrolling.

Table 1 Baseline demographics and disease characteristics

	DZP+SOC n=208	PBO+SOC n=107
Age, years, mean (SD)	43.5 (12.3)	41.5 (12.4)
Female, n (%)	193 (92.8)	100 (93.5)
Time since first diagnosis of SLE, years, mean (SD)	10.1 (7.9)	9.8 (8.5)
SLEDAI-2K total score, mean (SD)	10.7 (3.5)	11.2 (3.4)
<10, n (%)	68 (32.7)	28 (26.2)
≥10, n (%)	140 (67.3)	79 (73.8)
≥1 BILAG 2004 Grade A, n (%)	89 (42.8)	48 (44.9)
anti-dsDNA (EliA) >10 IU, n (%)	91 (43.8)	62 (57.9)
C3 <LLN, n (%)	66 (31.7)	43 (40.2)
C4 <LLN, n (%)	112 (53.8)	55 (51.4)
Any aPLs,[a] n (%)	129 (62.0)	64 (59.8)
LAC ratio >ULN, n (%)	16 (7.7)	13 (12.1)
Concomitant SLE medications at baseline, n (%)	208 (100.0)	107 (100.0)
Antimalarials, n (%)	166 (79.8)	91 (85.0)
Systemic glucocorticoids, n (%)	171 (82.2)	87 (81.3)
Immunosuppressants, n (%)	129 (62.0)	70 (65.4)
Triple therapy, n (%)	86 (41.3)	50 (46.7)
Systemic glucocorticoid dose >7.5 mg/day, n (%)	105 (50.5)	51 (47.7)

Full analysis set. **[a]** aPLs include anti-phosphatidylserine and anti-prothrombin.

Figure 2 Primary and first secondary endpoint: BICLA response

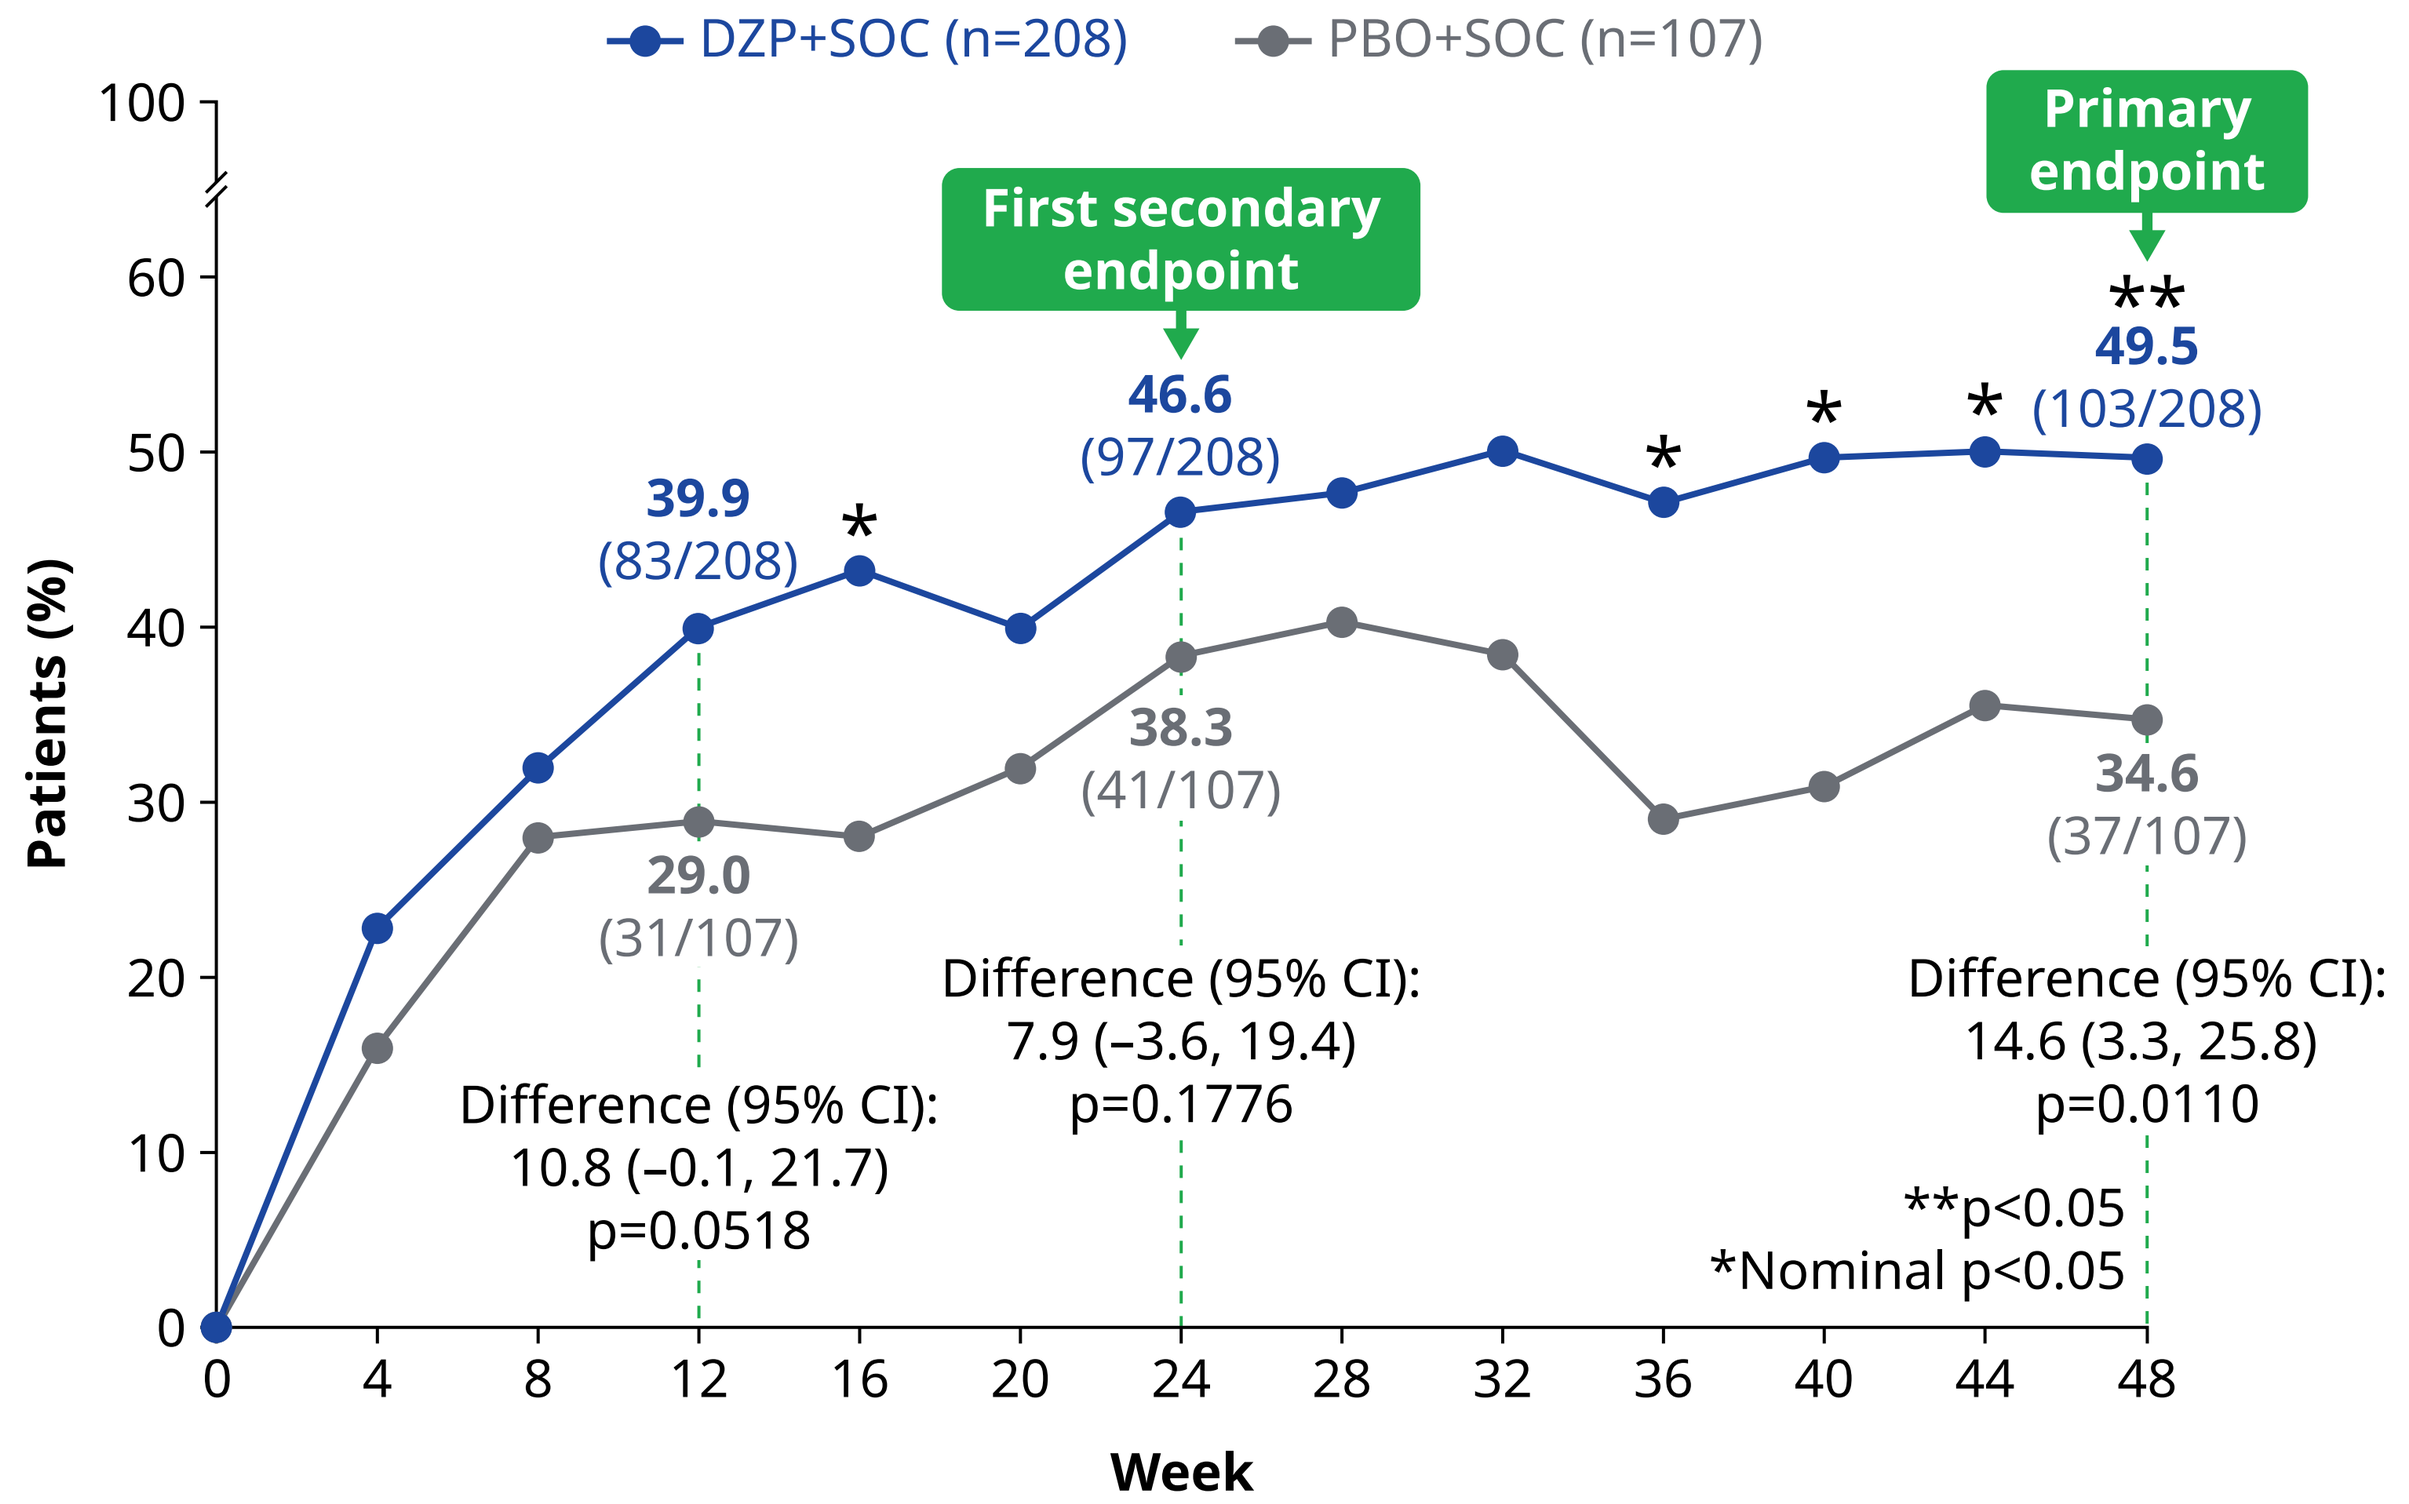
DZP+SOC (n=208)
PBO+SOC (n=107)
Patients (%)
Week
First secondary endpoint
Primary endpoint
39.9 (83/208)
29.0 (31/107)
46.6 (97/208)
38.3 (41/107)
** 49.5 (103/208)
34.6 (37/107)
Difference (95% CI): 10.8 (–0.1, 21.7) p=0.0518
Difference (95% CI): 7.9 (–3.6, 19.4) p=0.1776
Difference (95% CI): 14.6 (3.3, 25.8) p=0.0110
**p<0.05
*Nominal p<0.05

Full analysis set; NRI. BICLA response at Weeks 48 and 24 was controlled for multiplicity; response at all other weeks was not controlled for multiplicity and p-values are nominal. Difference in proportion responding between DZP+SOC and PBO+SOC, 95% CIs and p-values were estimated and tested using the CMH risk difference estimate controlling for the randomization stratification factors.

Figure 3 Glucocorticoid tapering to ≤7.5 mg/day in patients with baseline dose >7.5 mg/day

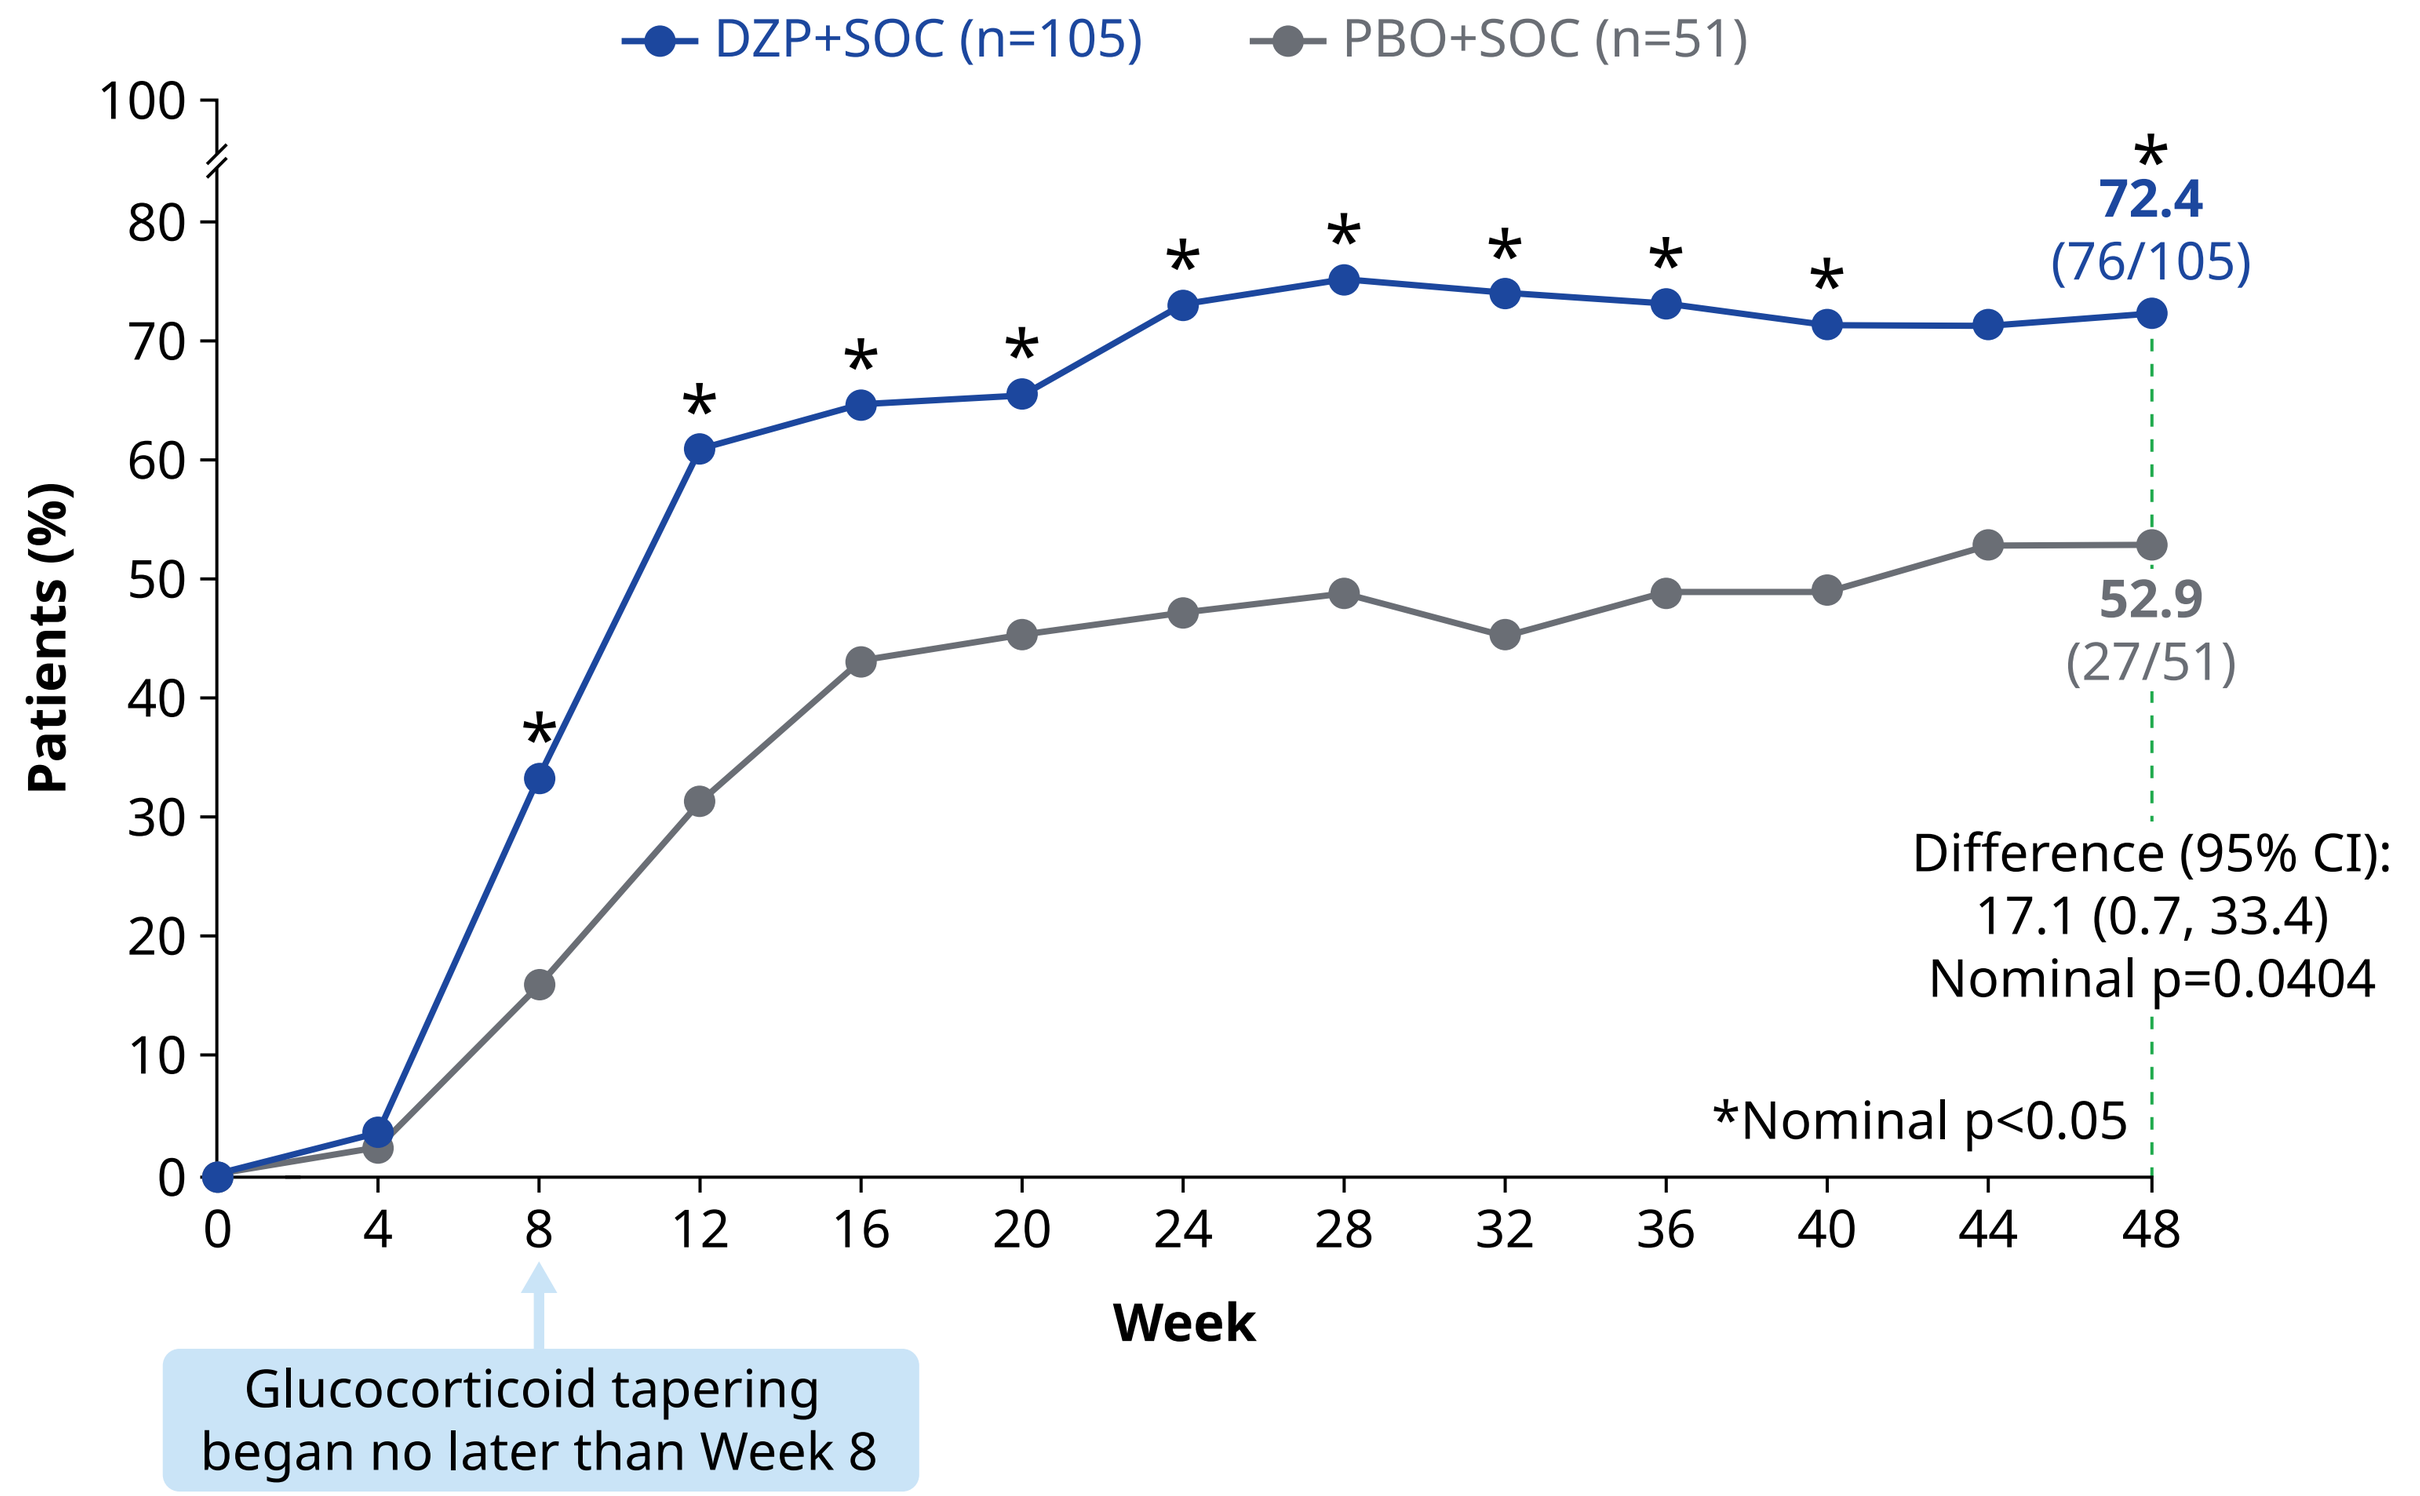
DZP+SOC (n=105)
PBO+SOC (n=51)
Patients (%)
Week
72.4 (76/105)
52.9 (27/51)
Difference (95% CI): 17.1 (0.7, 33.4) Nominal p=0.0404
*Nominal p<0.05
Glucocorticoid tapering began no later than Week 8

Full analysis set; NRI. All p-values are nominal and were not controlled for multiplicity. Difference in proportion responding between DZP+SOC and PBO+SOC, 95% CIs and p-values were estimated and tested using the CMH risk difference estimate controlling for the randomization stratification factors.

Figure 4 Other endpoints

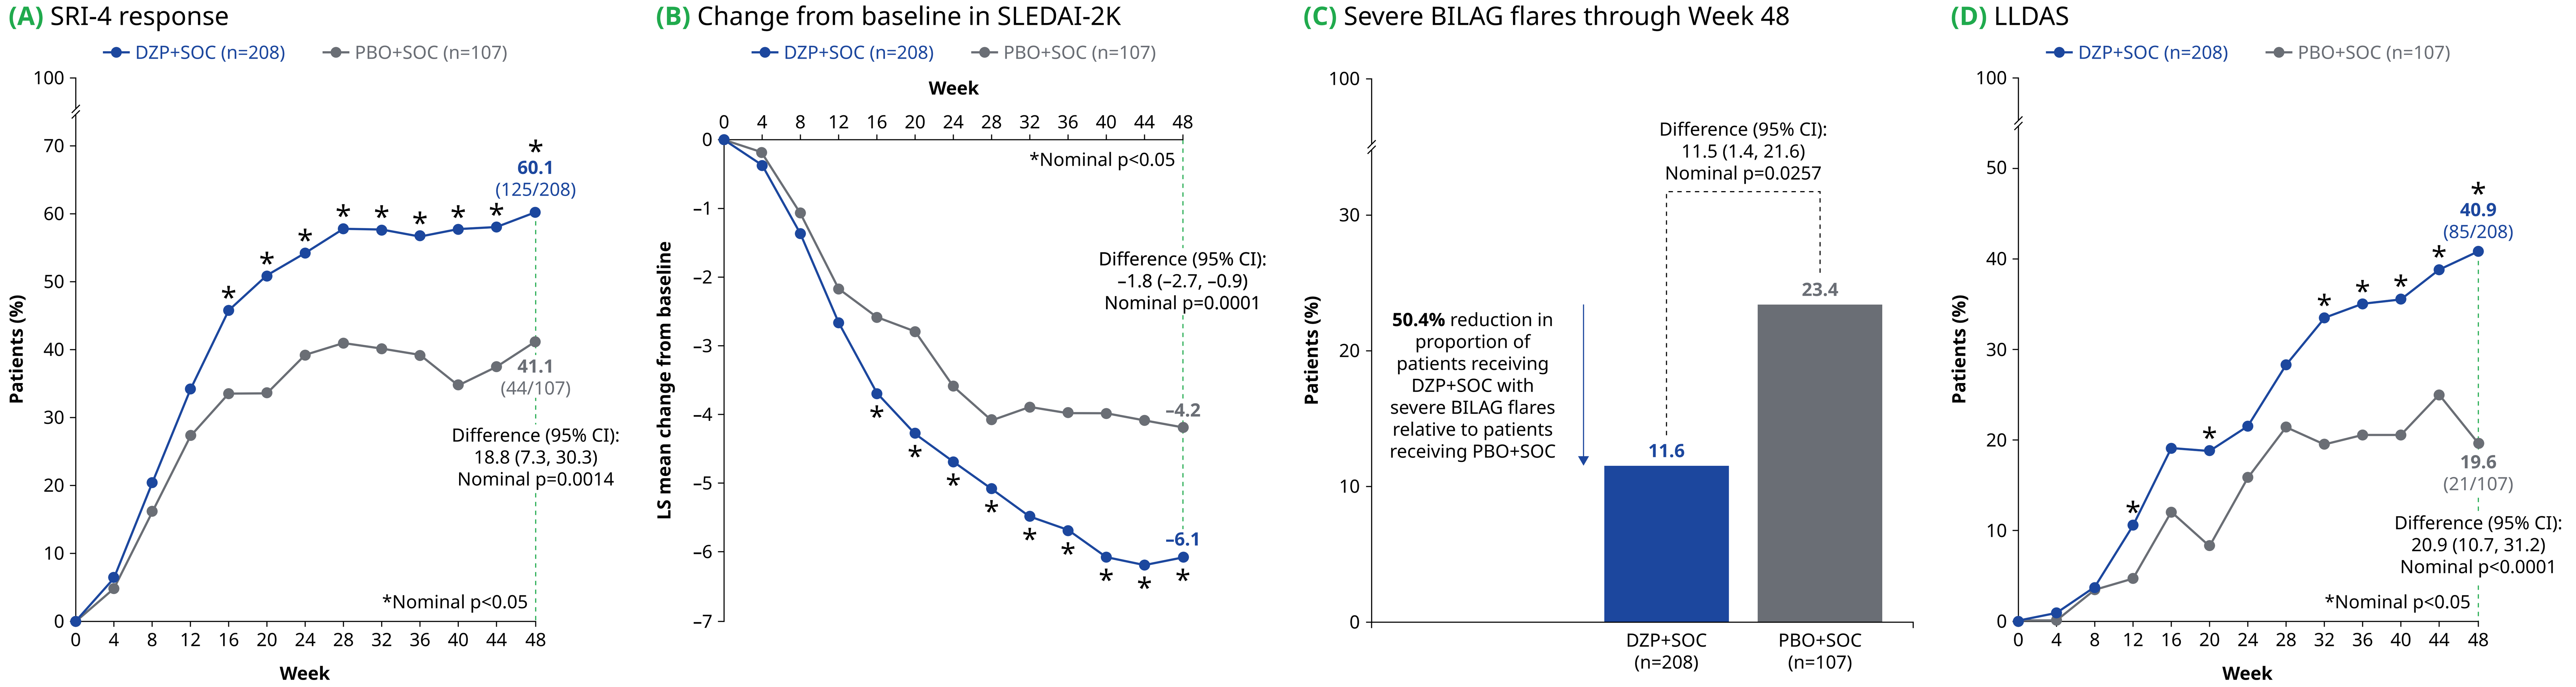
(A) SRI-4 response
DZP+SOC (n=208)
PBO+SOC (n=107)
Patients (%)
Week
60.1 (125/208)
41.1 (44/107)
Difference (95% CI): 18.8 (7.3, 30.3) Nominal p=0.0014
*Nominal p<0.05
(B) Change from baseline in SLEDAI-2K
DZP+SOC (n=208)
PBO+SOC (n=107)
Week
LS mean change from baseline
*Nominal p<0.05
Difference (95% CI): –1.8 (–2.7, –0.9) Nominal p=0.0001
–4.2
–6.1
(C) Severe BILAG flares through Week 48
Patients (%)
Difference (95% CI): 11.5 (1.4, 21.6) Nominal p=0.0257
50.4% reduction in proportion of patients receiving DZP+SOC with severe BILAG flares relative to patients receiving PBO+SOC
11.6
23.4
DZP+SOC (n=208)
PBO+SOC (n=107)
(D) LLDAS
DZP+SOC (n=208)
PBO+SOC (n=107)
Patients (%)
Week
40.9 (85/208)
19.6 (21/107)
Difference (95% CI): 20.9 (10.7, 31.2) Nominal p<0.0001
*Nominal p<0.05

Full analysis set; (A, D) NRI; (B) MMRM; (C) MI-MAR. All p-values are nominal and were not controlled for multiplicity. (A, C, D) Difference in proportion responding between DZP+SOC and PBO+SOC, 95% CIs and p-values were estimated and tested using the CMH risk difference estimate controlling for the randomization stratification factors. (B) The LS mean, the difference between DZP+SOC and PBO+SOC, 95% CIs and p-value were computed from a MMRM.

Table 2 Safety

n, (%)	DZP+SOC n=213	PBO+SOC n=108
Any TEAE	176 (82.6)	81 (75.0)
Serious TEAEs	21 (9.9)	16 (14.8)
Permanent discontinuation of drug or study discontinuation due to TEAEs	10 (4.7)	4 (3.7)
Hypersensitivity TEAEs starting on the day of or the day after an infusion	6 (2.8)	0 (0.0)
Infections and infestations	131 (61.5)	56 (51.9)
Mild	95 (44.6)	35 (32.4)
Moderate	66 (31.0)	36 (33.3)
Severe	3 (1.4)	4 (3.7)
Serious	8 (3.8)	6 (5.6)
Herpes viral infections	13 (6.1)	14 (13.0)
Herpes zoster	4 (1.9)	7 (6.5)
Ophthalmic herpes zoster	2 (0.9)[a]	0 (0.0)
Herpes ophthalmic	1 (0.5)[b]	0 (0.0)
Thromboembolic TEAEs confirmed by an adjudication committee	1 (0.5)	0 (0.0)
Acute myocardial infarction	1 (0.5)	0 (0.0)
Deaths	1 (0.5)	0 (0.0)
Gangrene-related sepsis	1 (0.5)	0 (0.0)

Safety set. MedDRA v24.0. **[a]** The two events were reported as "herpes zoster over left eyelid and forehead, V1" and "left herpes zoster ophthlamicus (dermatome V1/V2)"; **[b]** Reported as "herpetic queratitis".

aPL: antiphospholipid antibody; **anti-dsDNA:** anti-double stranded DNA; **BICLA:** BILAG-based Combined Lupus Assessment; **BILAG:** British Isles Lupus Assessment Group; **C3:** complement C3; **C4:** complement C4; **CI:** confidence interval; **CMH:** Cochran-Mantel Haenszel; **DZP:** dapirolizumab pegol; **EULAR:** European League Against Rheumatism; **Fab':** antigen-binding fragment; **IU:** international unit; **iv:** intravenous; **LAC:** lupus anticoagulant; **LLDAS:** Lupus Low Disease Activity State; **LLN:** lower limit of normal; **LS:** least square; **MedDRA:** Medical Dictionary for Regulatory Activities; **MI-MAR:** multiple imputation assuming missing at random; **MMRM:** mixed model for repeated measurements; **NRI:** non-responder imputation; **OLE:** open-label extension; **PBO:** placebo; **PEG:** polyethylene glycol; **Q4W:** every 4 weeks; **SD:** standard deviation; **SFU:** safety follow-up; **SLE:** systemic lupus erythematosus; **SLEDAI-2K:** SLE Disease Activity Index-2K; **SOC:** standard of care; **SRI-4:** SLE Responder Index 4; **TEAE:** treatment-emergent adverse event; **ULN:** upper limit of normal.

References: [1]Cutcutache I. Arthritis Rheumatol 2023;75 (suppl 9); [2]Powlesland AS. Annals Rheum Dis 2024;83 (suppl 1):261; [3]Fanouriakis A. Ann Rheum Dis 2019;78:736–45; [4]Furie RA. Rheumatology (Oxford) 2021;60:5397–407; [5]Chamberlain C. Ann Rheum Dis 2017;76:1837–44; [6]Tocoian A. Lupus 2015;24:1045–56. **Author Contributions:** Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **MEBC**, **DAI**, **JTM**, **TD**, **MP**, **EV**, **EFM**, **TJ**, **SB**, **JGR**, **CM**, **AN**, **CS**; Drafting of the publication, or reviewing it critically for important intellectual content: **MEBC**, **DAI**, **JTM**, **TD**, **MP**, **EV**, **EFM**, **TJ**, **SB**, **JGR**, **CM**, **AN**, **CS**; Final approval of the publication: **MEBC**, **DAI**, **JTM**, **TD**, **MP**, **EV**, **EFM**, **TJ**, **SB**, **JGR**, **CM**, **AN**, **CS**. **Author Disclosures: MEBC:** Consultant for GSK and UCB; received grant/research support from GSK and UCB; **DAI:** Consultant for AstraZeneca, Eli Lilly, GSK, Merck Serono, Novartis, Servier, and UCB; honoraria passed to local arthritis charity; **JTM:** Consultant for AbbVie, Alexion, Almiral, Alumis, Amgen, AstraZeneca, Aurinia, Biogen, BMS, Eli Lilly, EMD Serono, Equillium, Genentech, Gilead, GSK, Kezar, Merck, Novartis, Ono, Remegen, Sanofi, Takeda, Tenet, UCB, Veloxis, and Zenas; received grant/research support from AstraZeneca, BMS, and GSK; **TD:** Consultant for AbbVie, Eli Lilly, Janssen, Roche/GNE, and UCB; speaker's bureau for Novartis; **MP:** Consultant for Amgen, AnaptysBio, Annexon Bio, AstraZeneca, Atara Biosciences, Aurinia, Autolus, Bain Capital, Baobab Therapeutics, Biocryst, Biogen, Boxer Capital, Cabaletta Bio, Caribou Biosciences, CTI Clinical Trial and Consulting Services, CVS Health, DualityBio, Eli Lilly, EMD Serono, Emergent Biosolutions, Escient Pharmaceuticals, Exo Therapeutics, Gentibio, GSK, iCell Gene Therapeutics, Innovaderm Research, IQVIA, Kezar Life Sciences, Kira Pharmaceuticals, Nexstone Immunology, Nimbus Lakshmi, Novartis, Ono Pharma, PPD Development, Proviant, Regeneron, Seismic Therapeutic, Senti Biosciences, Sinomab Biosciences, Steritas, Takeda, Tenet Medicines, TG Therapeutics, UCB, Variant Bio, Worldwide Clinical Trials, and Zydus; speaker's bureau for Arthros-FocusMedEd, AstraZeneca, and Aurinia; received grant/research support from AstraZeneca, Aurinia, Eli Lilly, Exagen, GSK, and Janssen; **EV:** Consultant for AbbVie, Alpine, AstraZeneca, Aurinia, BMS, Eli Lilly, Merck, Novartis, Otsuka, Pfizer, Roche, and UCB; paid instructor for Novartis; speaker's bureau for AstraZeneca, Novartis, and Otsuka; received grant/research support from AstraZeneca and Sandoz; **EFM:** Consultant for AbbVie, AstraZeneca, Biogen, BMS, Cabaletta, Cullinan, Dragonfly, Eli Lilly, EMD Serono, GSK, Novartis, Quell, Remegen, and UCB; speaker's bureau for AstraZeneca, BMS, and Novartis; received grant/research support from AbbVie, Amgen, AstraZeneca, Biogen, BMS, Eli Lilly, EMD Serono, Genentech, GSK, Janssen, Novartis, Takeda, and UCB; **TJ**, **CM**, **CS:** Employees and shareholders of UCB; **SB**, **JGR**, **AN:** Employees and shareholders of Biogen; **SC:** Employee of Biogen. **Acknowledgements:** We would like to thank the patients and their caregivers in addition to all the investigators and their teams who contributed to this study. The authors acknowledge Heather Edens, PhD, UCB, Atlanta, GA, USA, and Valerie Zediak, PhD, Biogen, Pittsburgh, PA, USA, for publication coordination, Rebecca Light, BSc, and Patrick Cox, BSc, Costello Medical, UK, for medical writing and editorial assistance, and the Costello Medical Creative team for design support. Funded by UCB and Biogen. All costs associated with development of this presentation were funded by UCB and Biogen.

Presented at CCR West 2025 | September 18–21 | Huntington Beach, CA, USA

Previously presented at ACR Convergence 2024 and EULAR 2025