Streamlining the Process of Caregiving for a Loved One With Dravet or Lennox-Gastaut Syndrome

Tanja S. Kellermann[1]
Nikita Meer[2]
Colin Lake[1]

1. UCB, Smyrna, GA, USA
2. UCB, Smyrna, GA, USA*
*During the present work.

Poster P1-010

Introduction

- Caregiving for people living with Dravet syndrome (DS) or Lennox-Gastaut syndrome (LGS) is often complex and emotionally demanding.
- Caregivers are responsible for coordinating multiple aspects of care, including medical treatments, daily routines, and communication with healthcare teams.
- Many caregivers rely on fragmented systems—such as handwritten notes, phone reminders, or spreadsheets—to manage care-related information.
- This lack of centralization creates significant challenges, particularly when onboarding new care team members, introducing a new treatment, or ensuring continuity of care if the primary caregiver is unavailable.
- Maintaining these disparate sources adds an additional burden to caregivers who are already managing high-stress responsibilities.
- This fragmentation can compromise clarity, consistency, and confidence in care delivery, ultimately impacting both caregivers and the individuals they support.
- The CareCompass™ platform was designed to address these challenges by providing a secure, centralized digital platform that organizes essential caregiving resources in one place.
- The goal is to simplify daily tasks, improve communication, and reduce stress for caregivers navigating complex care needs.

Objective

- This poster presents findings from a pilot study conducted among caregivers of individuals living with DS and LGS.
- We aimed to assess the usability, acceptability, and early impact of CareCompass™, a digital platform designed to support daily caregiving:
 1. Demonstrate practical application
 - Show how the platform can be used as a real-life tool to support caregivers of individuals with DS and LGS managing daily care responsibilities.
 2. Present pilot phase insights
 - Share data and observations from the pilot phase to illustrate initial caregiver engagement, usability, and perceived value of the platform.
 3. Highlight key features and benefits
 - Explain the core functionalities and how these features aim to simplify caregiving tasks.

Methods

Figure 1. From a fragmented to a centralized caregiver experience

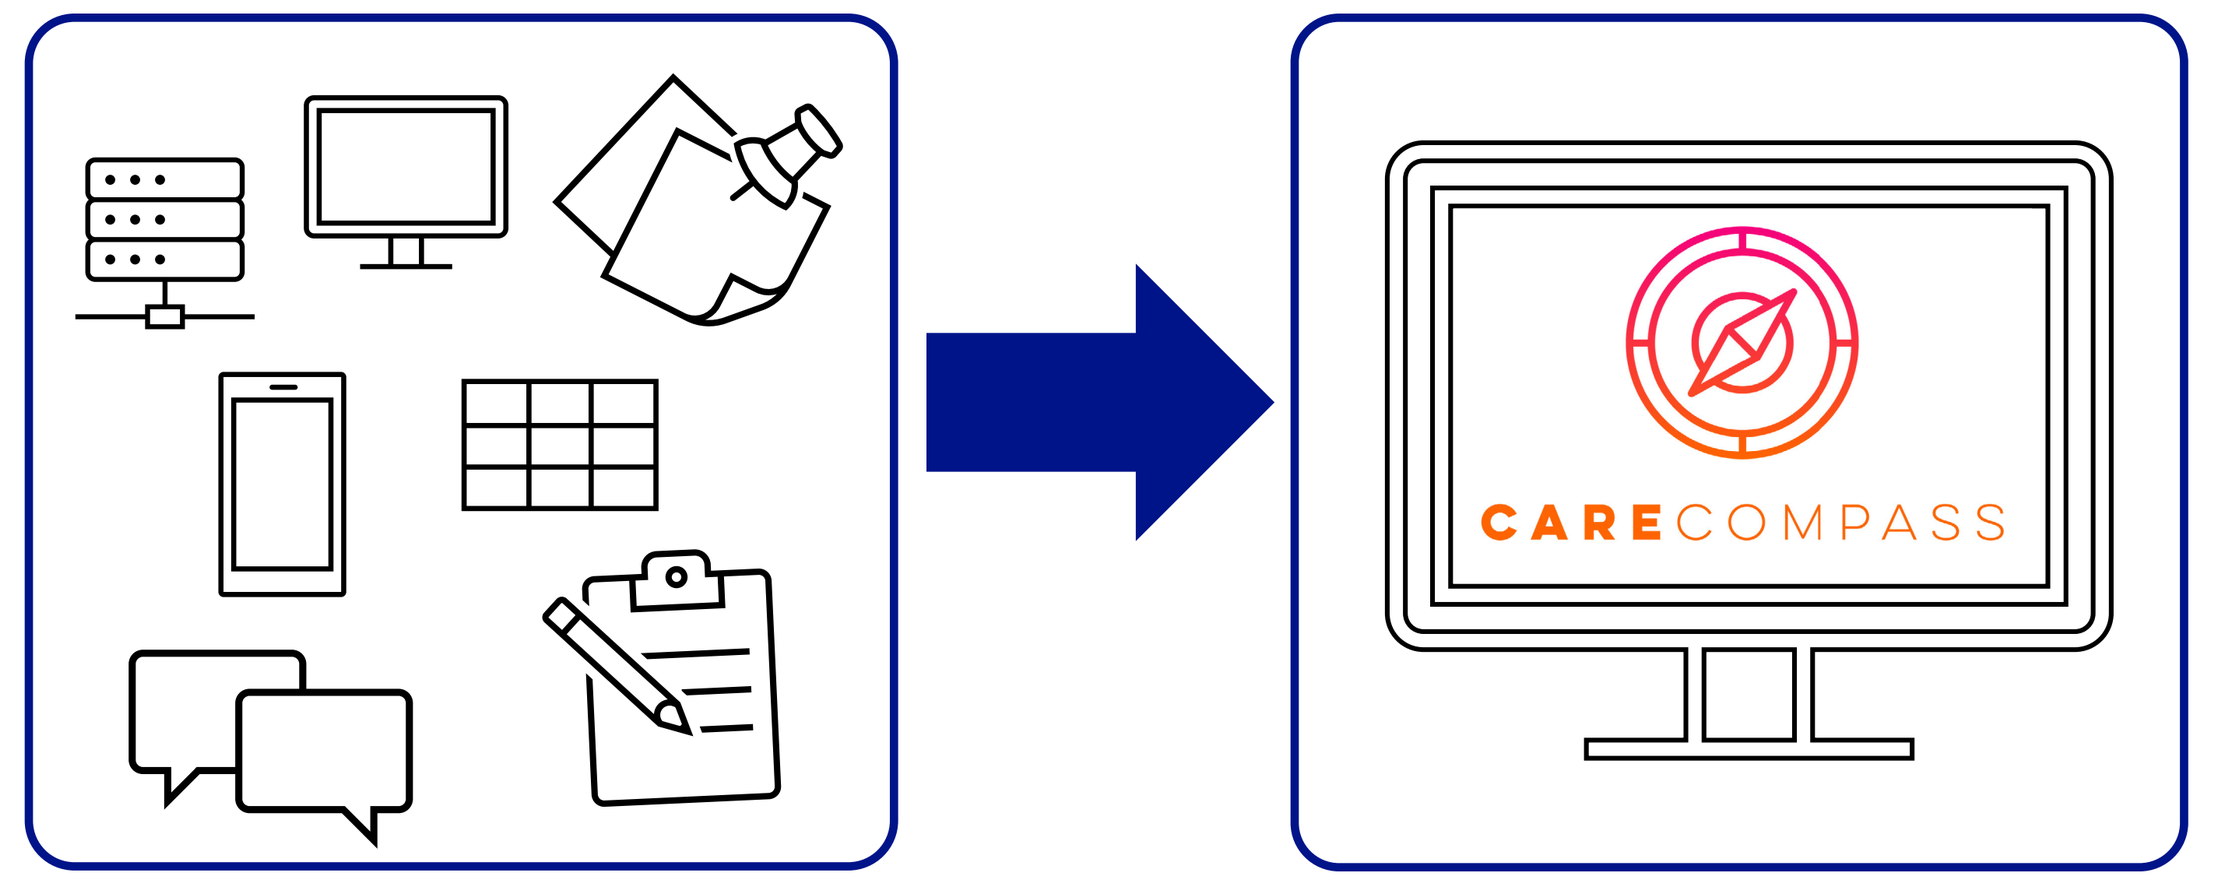

CARECOMPASS

Overview

QUESTION

How can we reduce the emotional and logistical burden on caregivers of loved ones living with DS or LGS?

INVESTIGATION

- A human-centered design approach was used to co-develop a digital solution with caregivers.
- Insights were gathered through focus groups and interviews to identify unmet needs and inform the design of a prototype.
- The resulting tool, CareCompass™, a secure, free-to-use, and mobile-friendly online platform that helps caregivers organize care information, track well-being, and create personalized care guides, is currently available for US residents.
- A 2- to 4-week pilot study with 23 caregivers was conducted to evaluate usability, acceptability, and early impact, using weekly surveys and regular check-ins.
 - To accommodate demanding schedules, caregivers could choose to participate for either 2 or 4 weeks; 21 chose the 4-week option, while 2 caregivers participated for 2 weeks.

CONCLUSIONS

The CareCompass™ platform shows strong potential as a digital support tool for caregivers of individuals with rare epilepsies (DS and LGS). It simplifies care coordination, addresses emotional and informational gaps, and may improve quality of life for both caregivers and their loved ones. A dedicated mobile app could further enhance caregiver satisfaction and accessibility.

Abbreviations: DS, Dravet syndrome; LGS, Lennox-Gastaut syndrome; NPS, Net Promoter Score.

RESULTS

- NPS of 62 during the pilot phase
- Caregivers suggested that a **dedicated mobile app** could further enhance their experience
- Reported benefits: centralized care information, peace of mind, improved communication, and valued journaling feature

For a copy of the poster, scan:

Overview of the essentials of CareCompass™

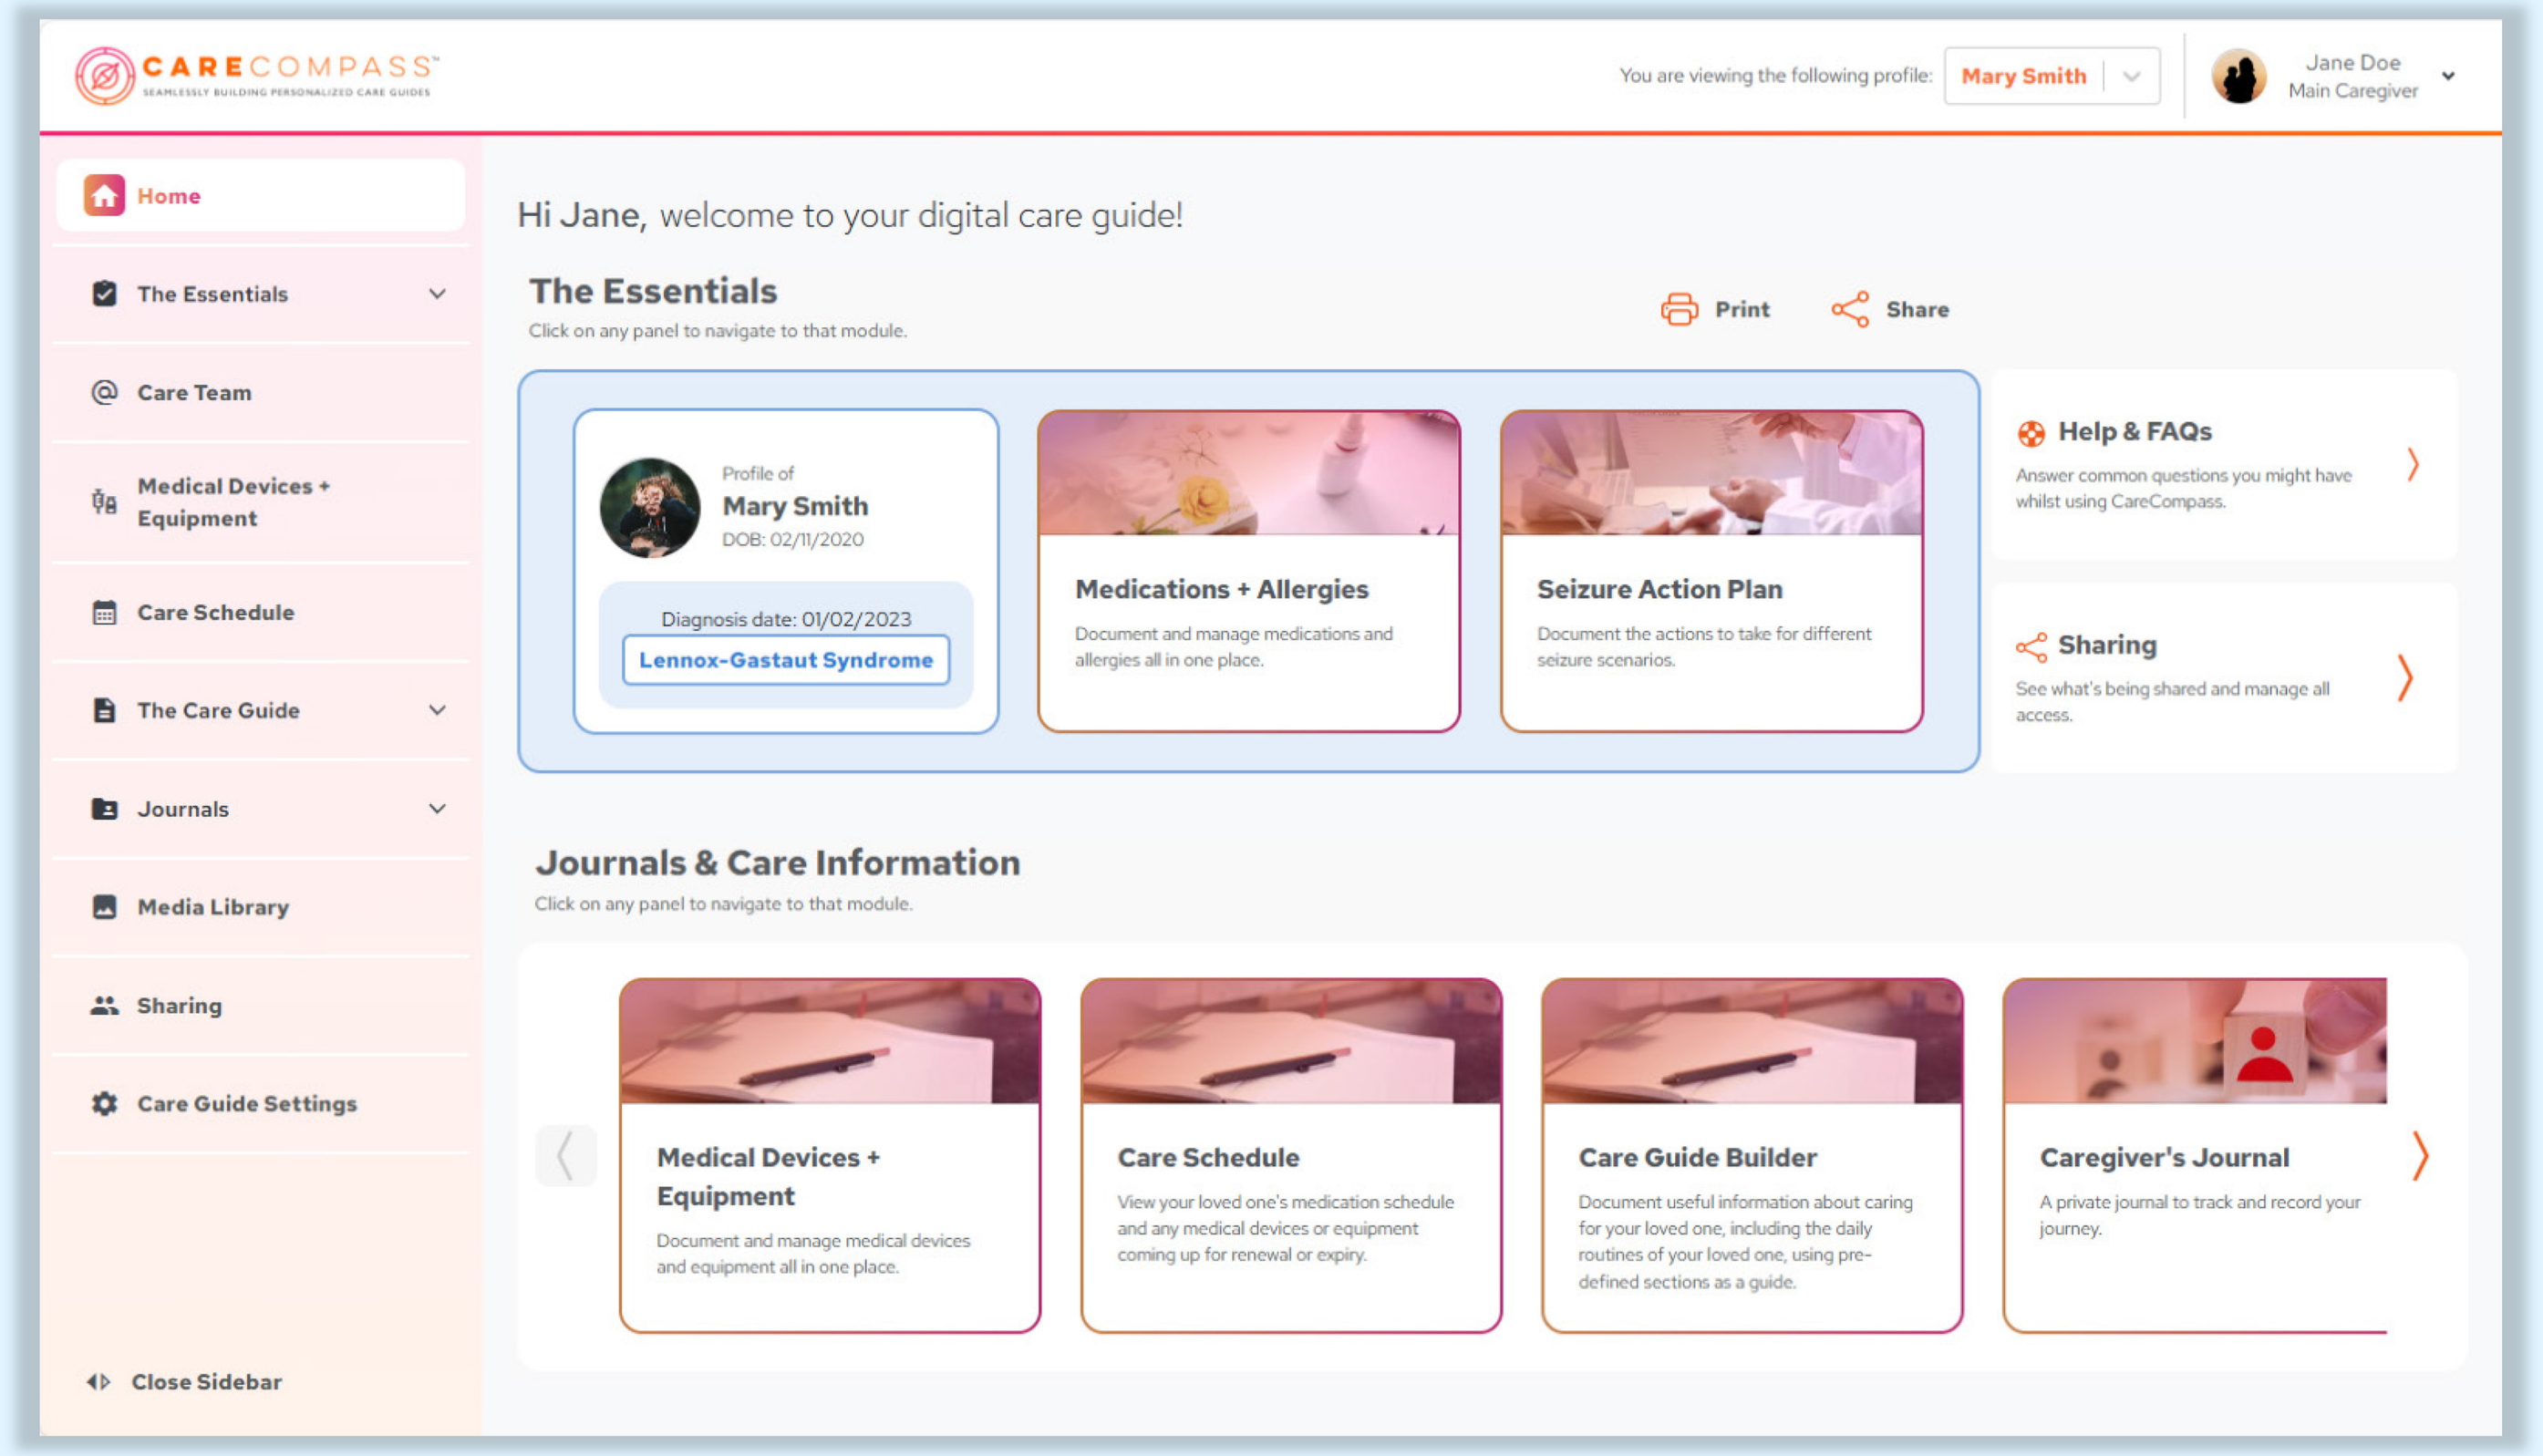

CARECOMPASS
You are viewing the following profile: Mary Smith
Jane Doe
Main Caregiver
Home
The Essentials
Care Team
Medical Devices + Equipment
Care Schedule
The Care Guide
Journals
Media Library
Sharing
Care Guide Settings
Close Sidebar
Hi Jane, welcome to your digital care guide!
The Essentials
Click on any panel to navigate to that module.
Print
Share
Profile of
Mary Smith
DOB: 02/11/2020
Diagnosis date: 01/02/2023
Lennox-Gastaut Syndrome
Medications + Allergies
Document and manage medications and allergies all in one place.
Seizure Action Plan
Document the actions to take for different seizure scenarios.
Help & FAQs
Answer common questions you might have whilst using CareCompass.
Sharing
See what's being shared and manage all access.
Journals & Care Information
Click on any panel to navigate to that module.
Medical Devices + Equipment
Document and manage medical devices and equipment all in one place.
Care Schedule
View your loved one's medication schedule and any medical devices or equipment coming up for renewal or expiry.
Care Guide Builder
Document useful information about caring for your loved one, including the daily routines of your loved one, using pre-defined sections as a guide.
Caregiver's Journal
A private journal to track and record your journey.

Figure 2. Human-centered method and timeline

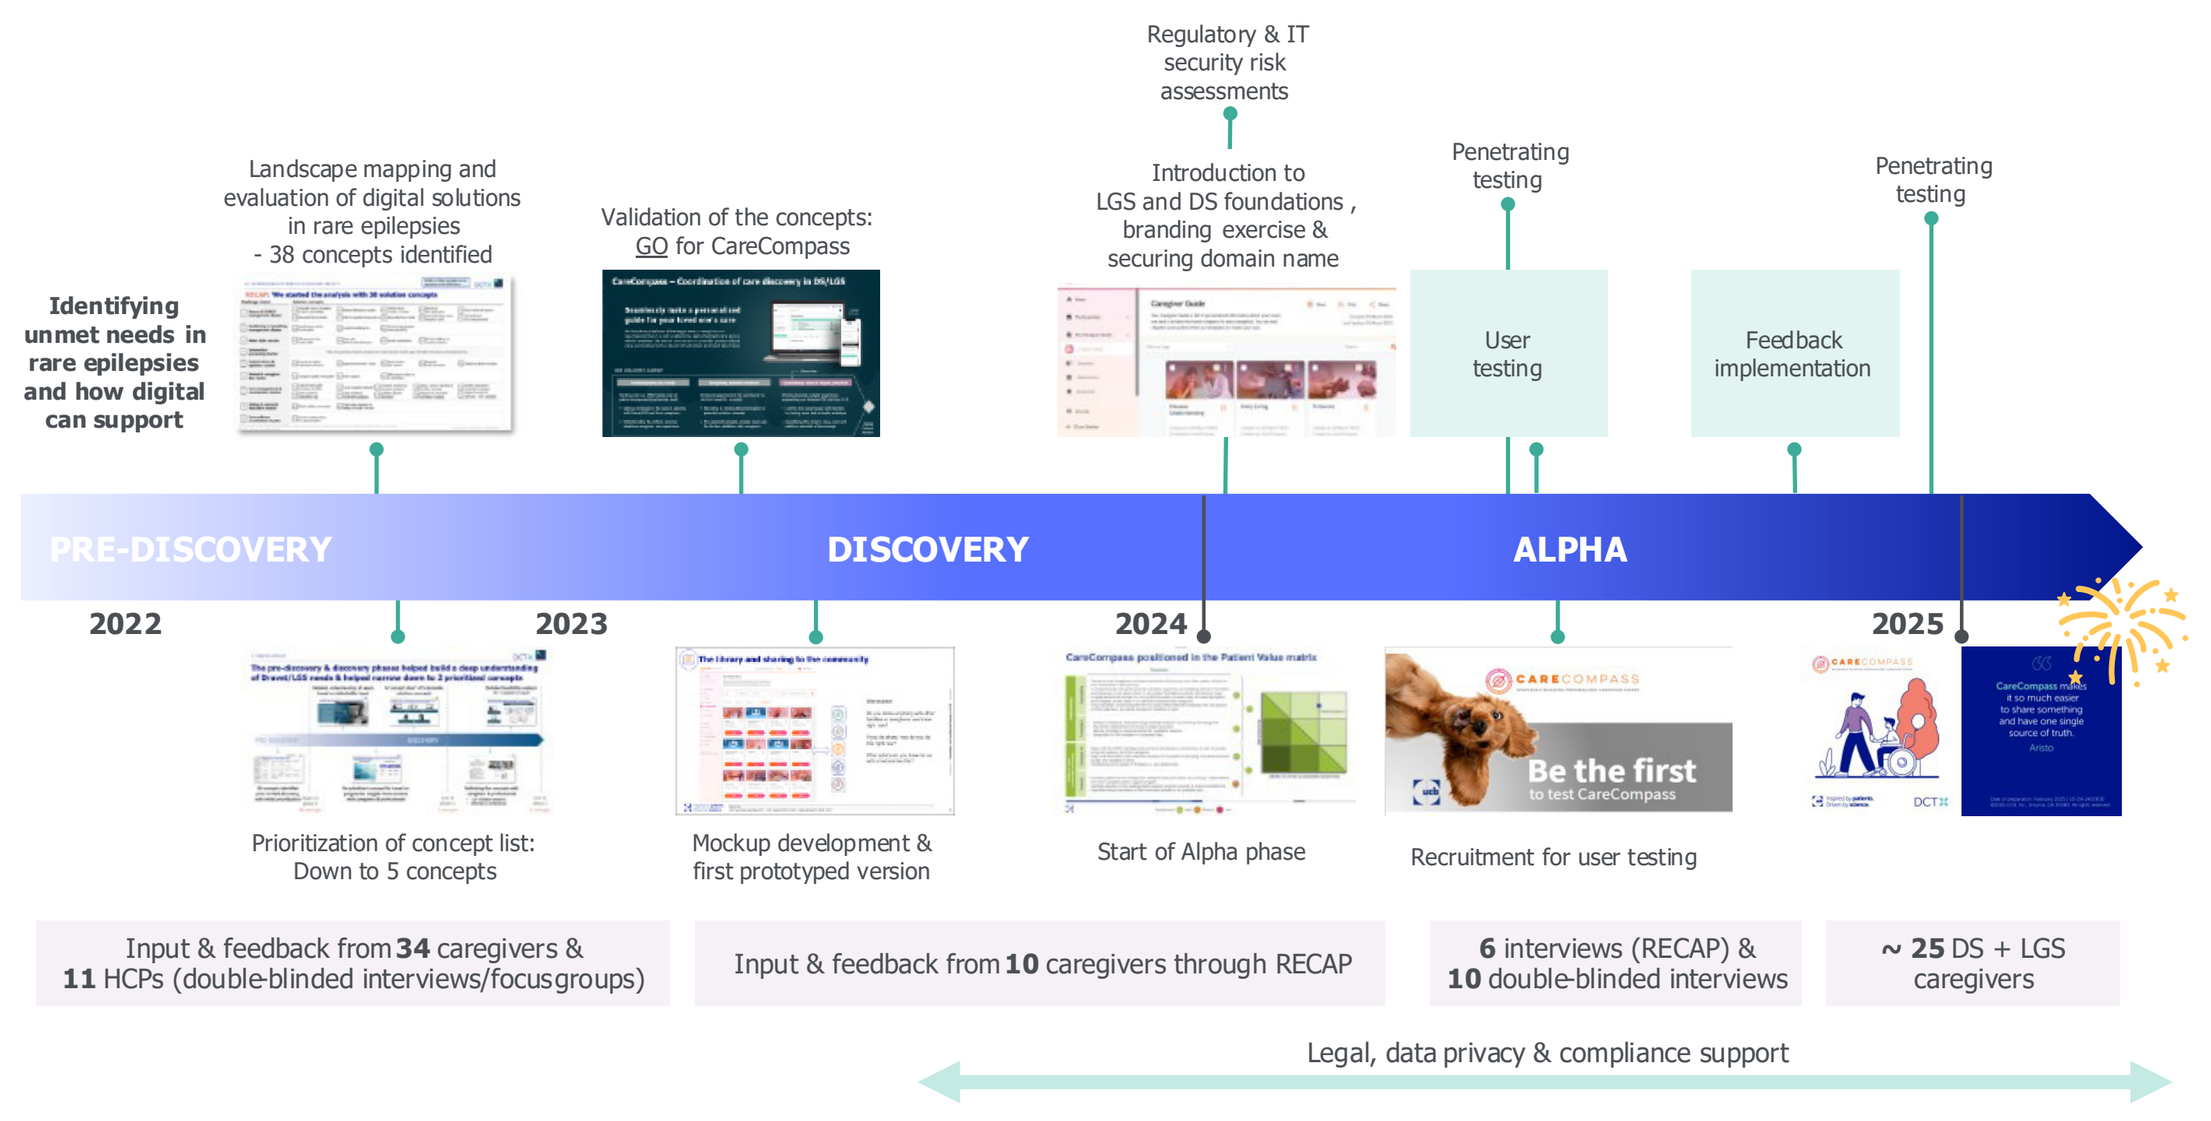

Identifying unmet needs in rare epilepsies and how digital can support
Landscape mapping and evaluation of digital solutions in rare epilepsies - 38 concepts identified
Validation of the concepts: GO for CareCompass
Regulatory & IT security risk assessments
Introduction to LGS and DS foundations, branding exercise & securing domain name
Penetrating testing
User testing
Feedback implementation
Penetrating testing
PRE-DISCOVERY
DISCOVERY
ALPHA
2022
2023
2024
2025
Prioritization of concept list: Down to 5 concepts
Mockup development & first prototyped version
Start of Alpha phase
Recruitment for user testing
Be the first
Input & feedback from 34 caregivers & 11 HCPs (double-blinded interviews/focusgroups)
Input & feedback from 10 caregivers through RECAP
6 interviews (RECAP) & 10 double-blinded interviews
~ 25 DS + LGS caregivers
Legal, data privacy & compliance support

DS, Dravet syndrome; HCP, healthcare professional; IT, Information Technology; LGS, Lennox-Gastaut syndrome; RECAP, Rare Epilepsy Caregiver Advisory Panel.

- The **CareCompass™** platform was developed in collaboration with caregivers to provide practical support for those managing the daily responsibilities of caring for individuals with DS and LGS (**Figure 1, Figure 2**).
- The platform includes practical templates that assist with organizing and completing routine caregiving tasks.
- It also offers resources aimed at providing emotional support to caregivers.
- In addition, it provides essential tools designed to help simplify daily activities and reduce stress (**Figure 1**).
- All caregiving resources are stored in one centralized location, making them easy to access and securely share.
- This structure is intended to help caregivers feel supported, maintain confidence, and stay connected throughout their care journey.

Results

- **Consistent use:**
 Overall, 21 out of 23 caregivers (91%) actively used the CareCompass™ platform throughout a 4-week pilot.
- **User satisfaction:**
 📊 These caregivers responded to the question:
 'After another week of usage, how likely are you to recommend CareCompass™ to a friend or colleague who also cares for someone with DS/LGS?'
- **Net Promoter Score (NPS): 62**
 - *14 Promoters* (67%)
 - *6 Passives* (28%)
 - *1 Detractor* (5%)

This score suggests **strong satisfaction** and enthusiasm for the solution (**Figure 3**).

- **Feature feedback:**
 💡 Caregivers suggested that a **dedicated mobile app** could further enhance their experience.
- **Perceived usefulness:**
 📝 At the end of the pilot, caregivers rated several statements about the usefulness of the CareCompass™ platform; results were **encouragingly positive** (**Figure 4**).

Figure 3. Definition of Net Promotor Score

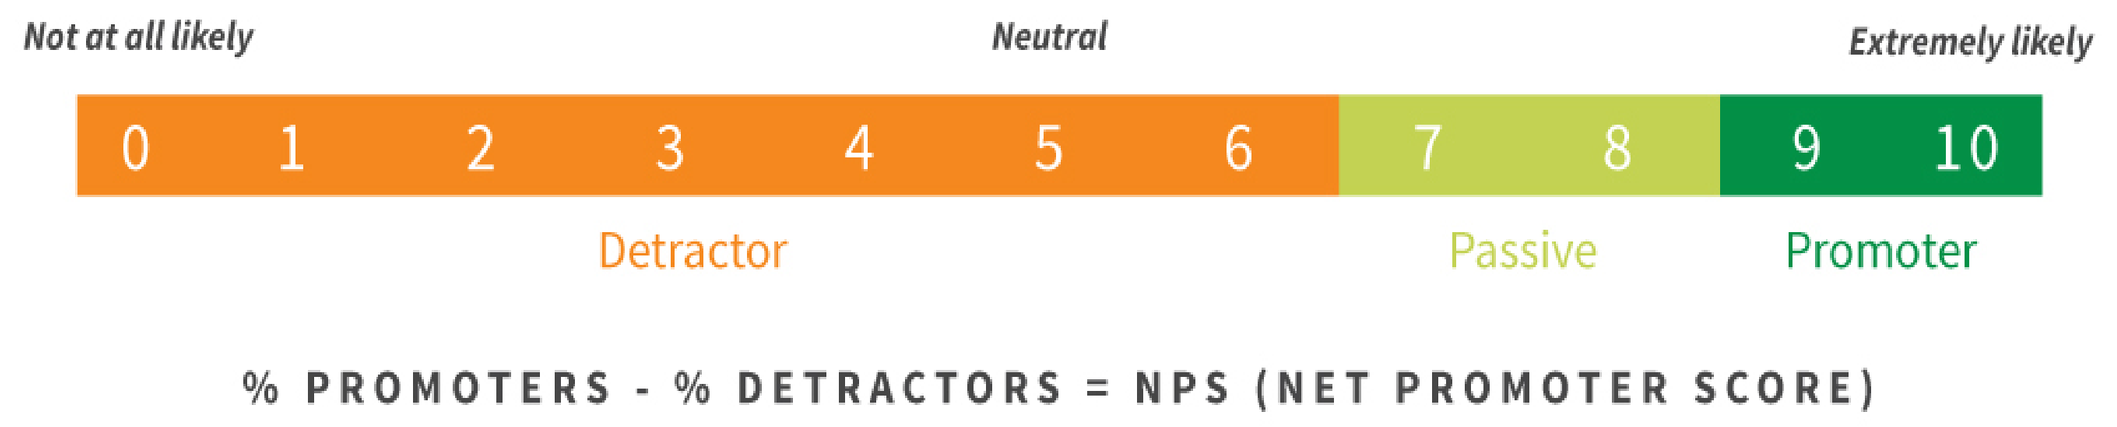

Not at all likely
Neutral
Extremely likely
0 1 2 3 4 5 6 7 8 9 10
Detractor
Passive
Promoter
% PROMOTERS - % DETRACTORS = NPS (NET PROMOTER SCORE)

Figure 4. What 4 weeks revealed about the CareCompass™ platform value for 21 caregivers

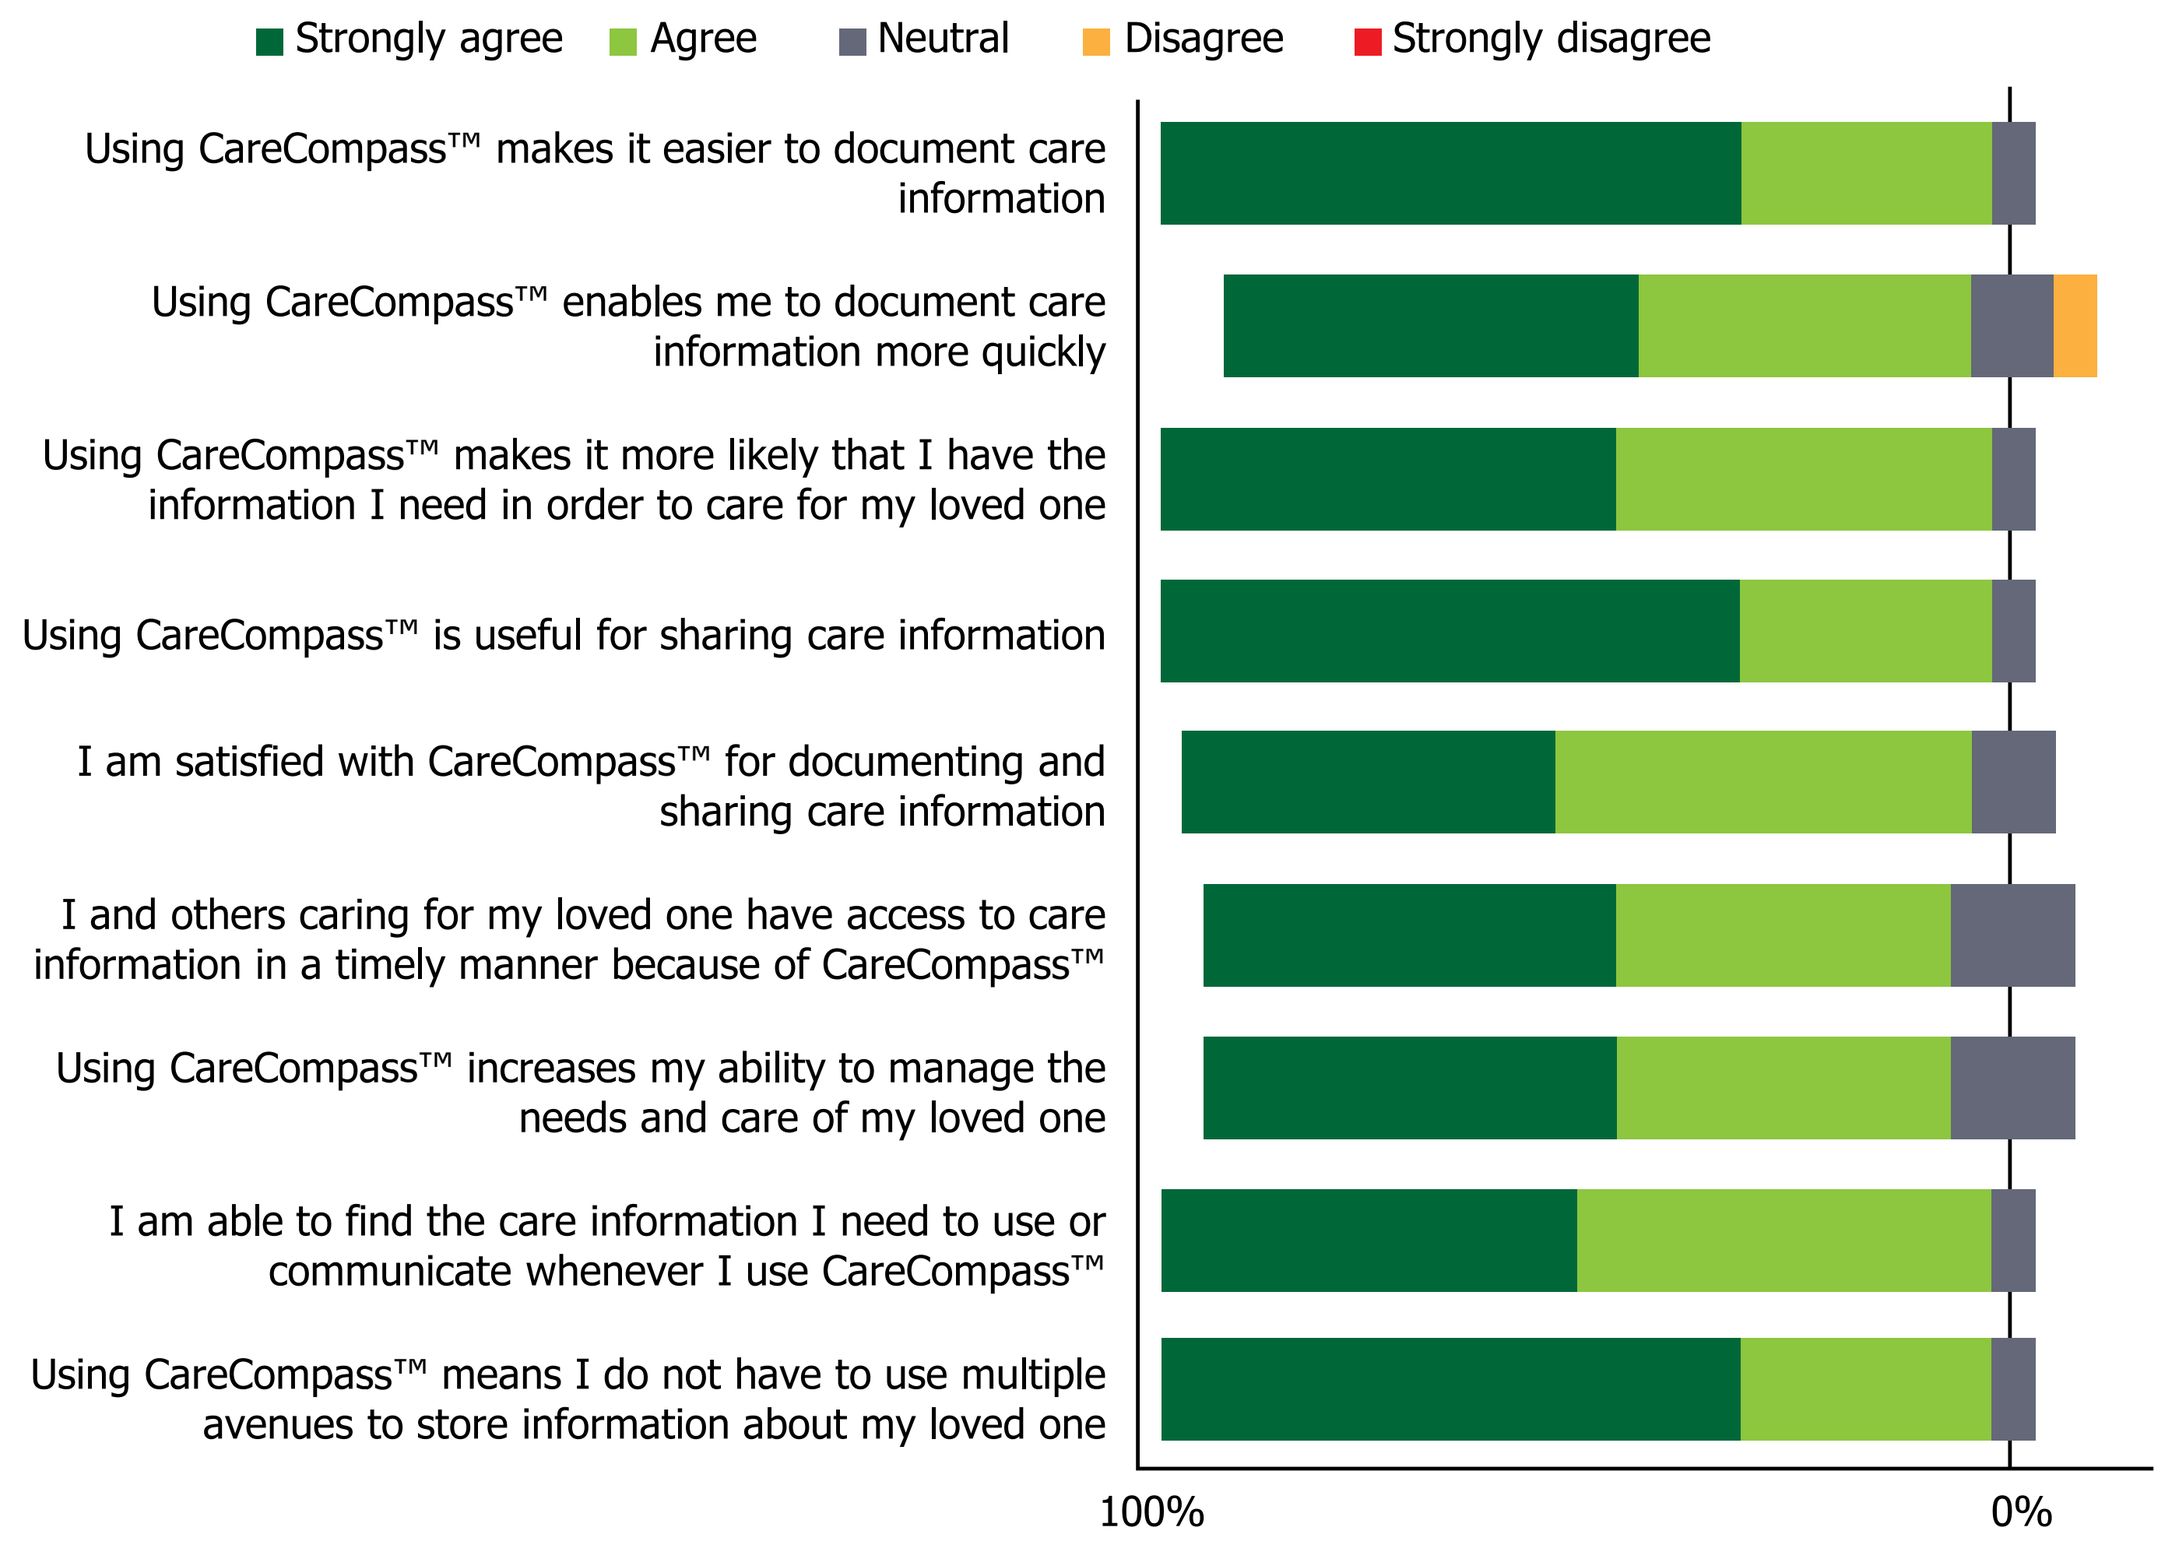

Strongly agree
Agree
Neutral
Disagree
Strongly disagree
Using CareCompass™ makes it easier to document care information
Using CareCompass™ enables me to document care information more quickly
Using CareCompass™ makes it more likely that I have the information I need in order to care for my loved one
Using CareCompass™ is useful for sharing care information
I am satisfied with CareCompass™ for documenting and sharing care information
I and others caring for my loved one have access to care information in a timely manner because of CareCompass™
Using CareCompass™ increases my ability to manage the needs and care of my loved one
I am able to find the care information I need to use or communicate whenever I use CareCompass™
Using CareCompass™ means I do not have to use multiple avenues to store information about my loved one
100%
0%

Figure 5. Voices from the pilot: what caregivers shared

'You don't want to think about getting sick and not being able to take care of your child (…), but if you have a tool like CareCompass™ you are going to be ok.' *(Melanie, caregiver of a loved one living with DS)*

'Our focus should be on our loved ones and what's best for them.' *(Kurt, caregiver of a loved one living with LGS)*

Figure 6. Popular features: 1) medication and allergies; 2) seizure action plan; 3) daily care templates; 4) loved one's journal

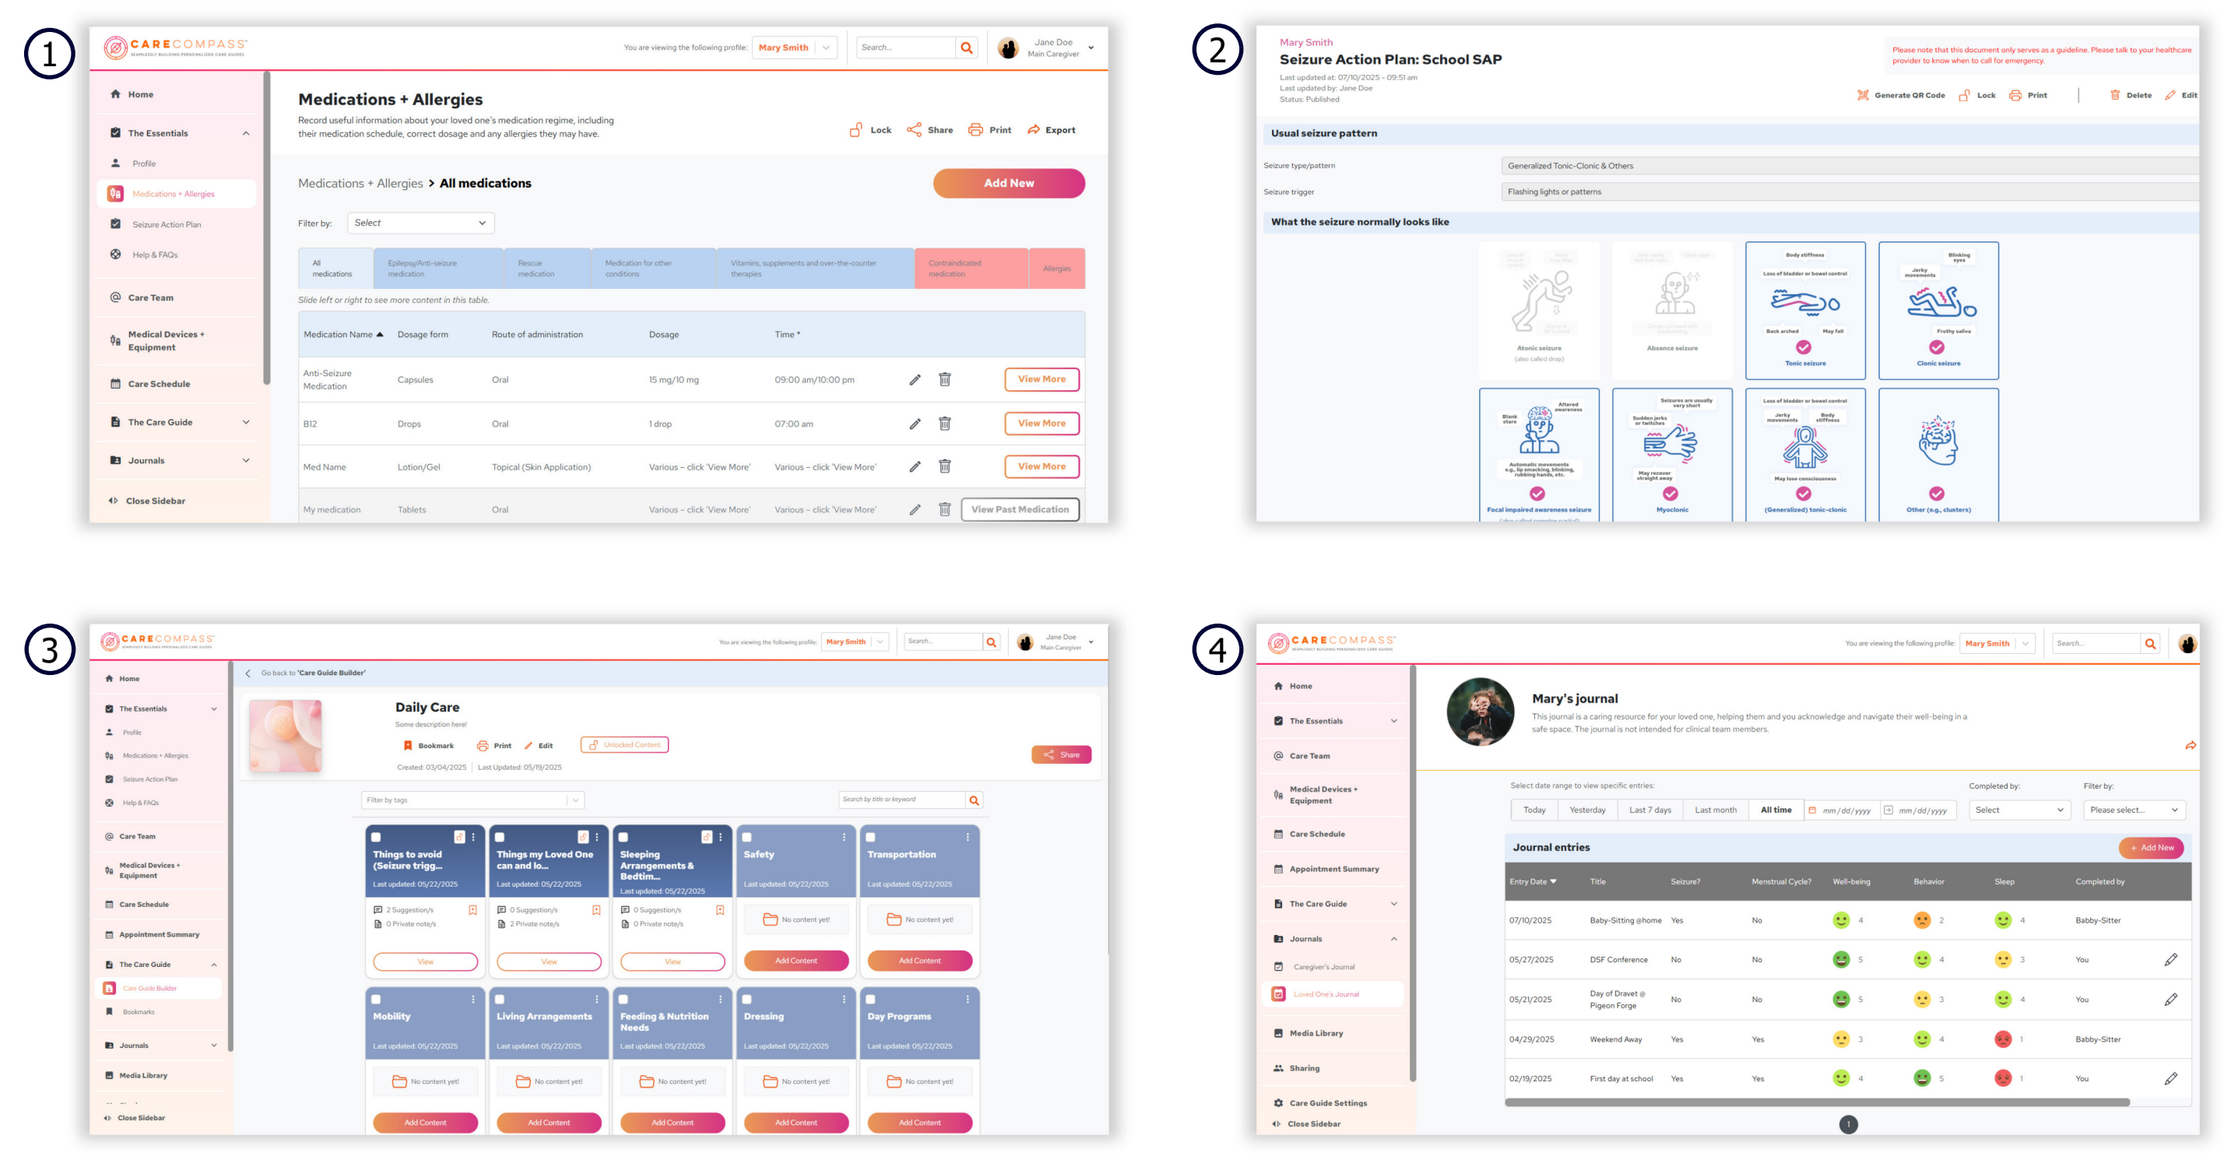

1
2
3
4
Medications + Allergies
Seizure Action Plan: School SAP
Daily Care
Mary's journal

Conclusions

- This pilot study suggests that the CareCompass™ platform may have strong satisfaction from the NPS and **potential as a supportive digital tool** for caregivers of individuals with rare epilepsies, particularly DS and LGS.
- **Co-designed** features were shown to directly address emotional and informational gaps experienced by caregivers.
- **Key benefits of the CareCompass™ platform were shown to include:**
 - Centralized care information to simplify communication and coordination.
 - Increased peace of mind and confidence in care continuity.
 - Enhanced quality of life for both caregivers and their loved ones.
- Native mobile app was preferred by caregivers over a mobile-friendly web for stronger adoption and usability.
- **Future research** should assess long-term impact on:
 - Caregiver burden.
 - Confidence in caregiving.
 - Engagement with the healthcare system.
- **Scalable impact:** The CareCompass™ platform is currently available for US residents; however, there is potential to expand the platform to support caregivers across other rare epilepsies, broader epilepsy populations, and diverse global regions worldwide.

References

1. CareCompass™. https://care-compass.io/. Accessed February 26, 2026.
2. Net Promoter Network. What is Net Promoter? https://www.netpromoter.com/know/. Accessed February 26, 2026.

UCB-sponsored. We gratefully acknowledge the DS and LGS Foundation for their support and all the caregivers of individuals living with DS and LGS whose insights, experiences, and dedication were essential in shaping and developing CareCompass™. Your voices made this work possible. The authors further acknowledge Stephanie Kennedy and her team from Partizan International for developing the solution based on caregivers' voices. The authors also acknowledge Sandra M Aguero, PharmD, BCPS, and Courtney Breuel, ELS, of the Sensified Division of Woven Health Collective, LLC, for editorial assistance, which was funded by UCB. Author disclosures: TS Kellermann, C Lake: Employee of UCB. N Meer: Employee of UCB at the time of preparation.

For a copy of this poster, use your smartphone to scan the QR code or contact UCBCares®
Phone: +1-844-599-CARE (2273)
Email: UCBCares@ucb.com

American Academy of Neurology 78th Annual Meeting
Chicago, IL, USA | April 18–22, 2026
Previously presented at American Epilepsy Society 79th Annual Meeting, Atlanta, GA, USA, December 5–9, 2025