Poster **P9-9-004**

Response to rozanolixizumab across treatment cycles in patients with generalized myasthenia gravis: A *post hoc* analysis of final pooled Phase 3 data

AAN 2026, Chicago, IL, USA; April 18–22, 2026

Robert M. Pascuzzi[1], Ali A. Habib[2], Zabeen K. Mahuwala[3], Renato Mantegazza[4], Sabrina Sacconi[5], John Vissing[6], Tuan Vu[7], Joel Baldwin[8], Thaïs Tarancón[9], Vera Bril[10]

[1]Neurology Department, Indiana University School of Medicine, Indiana University Health, Indianapolis, IN, USA; [2]MDA ALS & Neuromuscular Center, Department of Neurology, University of California, Irvine, Orange, CA, USA; [3]Department of Neuromuscular Medicine, Epilepsy and Clinical Neurophysiology, University of Kentucky, Lexington, KY, USA; [4]Emeritus and Past Director, Department of Neuroimmunology and Neuromuscular Diseases, Fondazione IRCCS, Istituto Nazionale Neurologico Carlo Besta, Milan, Italy; [5]Université Côte d'Azur, Peripheral Nervous System & Muscle Department, Pasteur 2 Hospital, Centre Hospitalier Universitaire de Nice, Nice, France; [6]Copenhagen Neuromuscular Center, Department of Neurology, Rigshospitalet, University of Copenhagen, Copenhagen, Denmark; [7]Department of Neurology, University of South Florida Morsani College of Medicine, Tampa, FL, USA; [8]UCB, Slough, UK; [9]UCB, Madrid, Spain; [10]Ellen and Martin Prosserman Centre for Neuromuscular Diseases, Toronto General Hospital, University of Toronto, Toronto, Ontario, Canada

Introduction

- gMG is a rare, chronic, autoimmune neuromuscular disease characterized by fluctuating muscle weakness and fatigue[1–3]
- Rozanolixizumab is a humanized IgG4 mAb that targets and blocks FcRn with high affinity, and is approved for the treatment of adults with anti-AChR Ab+ or anti-MuSK Ab+ gMG[3,4]
- In the Phase 3 MycarinG study (NCT03971422), one 6-week cycle of rozanolixizumab led to statistically significant and clinically meaningful improvements in MG-specific outcomes versus placebo in patients with gMG[2]
 - Following MycarinG, patients could enroll in open-label extension studies MG0004 (NCT04124965) and MG0007 (NCT04650854), or MG0007 directly
- **This *post hoc* analysis assessed the response to rozanolixizumab over multiple treatment cycles in patients with gMG, based on their Cycle 1 response**

Methods

- Patients enrolled in MycarinG were aged ≥18 years with MGFA Disease Class II–IVa anti-AChR Ab+ or anti-MuSK Ab+ gMG with an MG-ADL score ≥3 (for non-ocular symptoms) and a QMG score ≥11[3]
 - Patients were randomized to receive one 6-week cycle of subcutaneous rozanolixizumab 7 mg/kg, 10 mg/kg or placebo[3]
- In MG0004, patients received chronic, once-weekly rozanolixizumab 7 mg/kg or 10 mg/kg for ≤52 weeks
- In MG0007, after an initial cycle of rozanolixizumab 7 mg/kg or 10 mg/kg, subsequent cycles were administered upon symptom worsening, at the investigator's discretion
- Final efficacy data were pooled for patients with ≥2 symptom-driven rozanolixizumab treatment cycles across MycarinG, MG0004 (first 6 weeks) and MG0007
- Response to rozanolixizumab was defined as an improvement from baseline of ≥2.0 points in MG-ADL score or an improvement from baseline of ≥3.0 points in QMG score at the end of each cycle (Day 43)
 - The proportion of patients achieving a response at Day 43 in Cycles 2–13, based on Cycle 1 response, was assessed

Results

- Overall, 129 patients received ≥2 symptom-driven cycles of rozanolixizumab
- Baseline demographics and disease characteristics were indicative of a broad population of patients with gMG

Baseline demographics and disease characteristics were indicative of a broad population of patients with gMG

		RLZ total (N=129)
	Age, years, mean (SD)	**50.9** (16.3)
	Sex, female, n (%)	**77** (59.7)
	Age at initial gMG diagnosis, years, mean (SD)	**43.1** (17.8)
	Duration of disease from diagnosis, years, mean (SD)	**8.1** (8.5)
MGFA Disease Class, n (%)	II	**52** (40.3)
	III	**72** (55.8)
	IV	**5** (3.9)
	Anti-AChR Ab+, n (%)	**117** (90.7)
	Anti-MuSK Ab+, n (%)	**12** (9.3)
	MG-ADL score, mean (SD)	**8.7** (3.4)
	QMG score, mean (SD)	**16.0** (3.8)
Baseline gMG medication, n (%)	Systemic CS use	**80** (62.0)
	Immunosuppressants	**66** (51.2)
	Parasympathomimetics	**113** (87.6)

Cycle 1 responders and non-responders to rozanolixizumab treatment and subsequent response rates

- In Cycle 1, 74.4% (96/129) and 68.2% (88/129) of patients were MG-ADL and QMG responders at Day 43, respectively

Cycle 1 responders:
MG-ADL: 74.4%
QMG: 68.2%

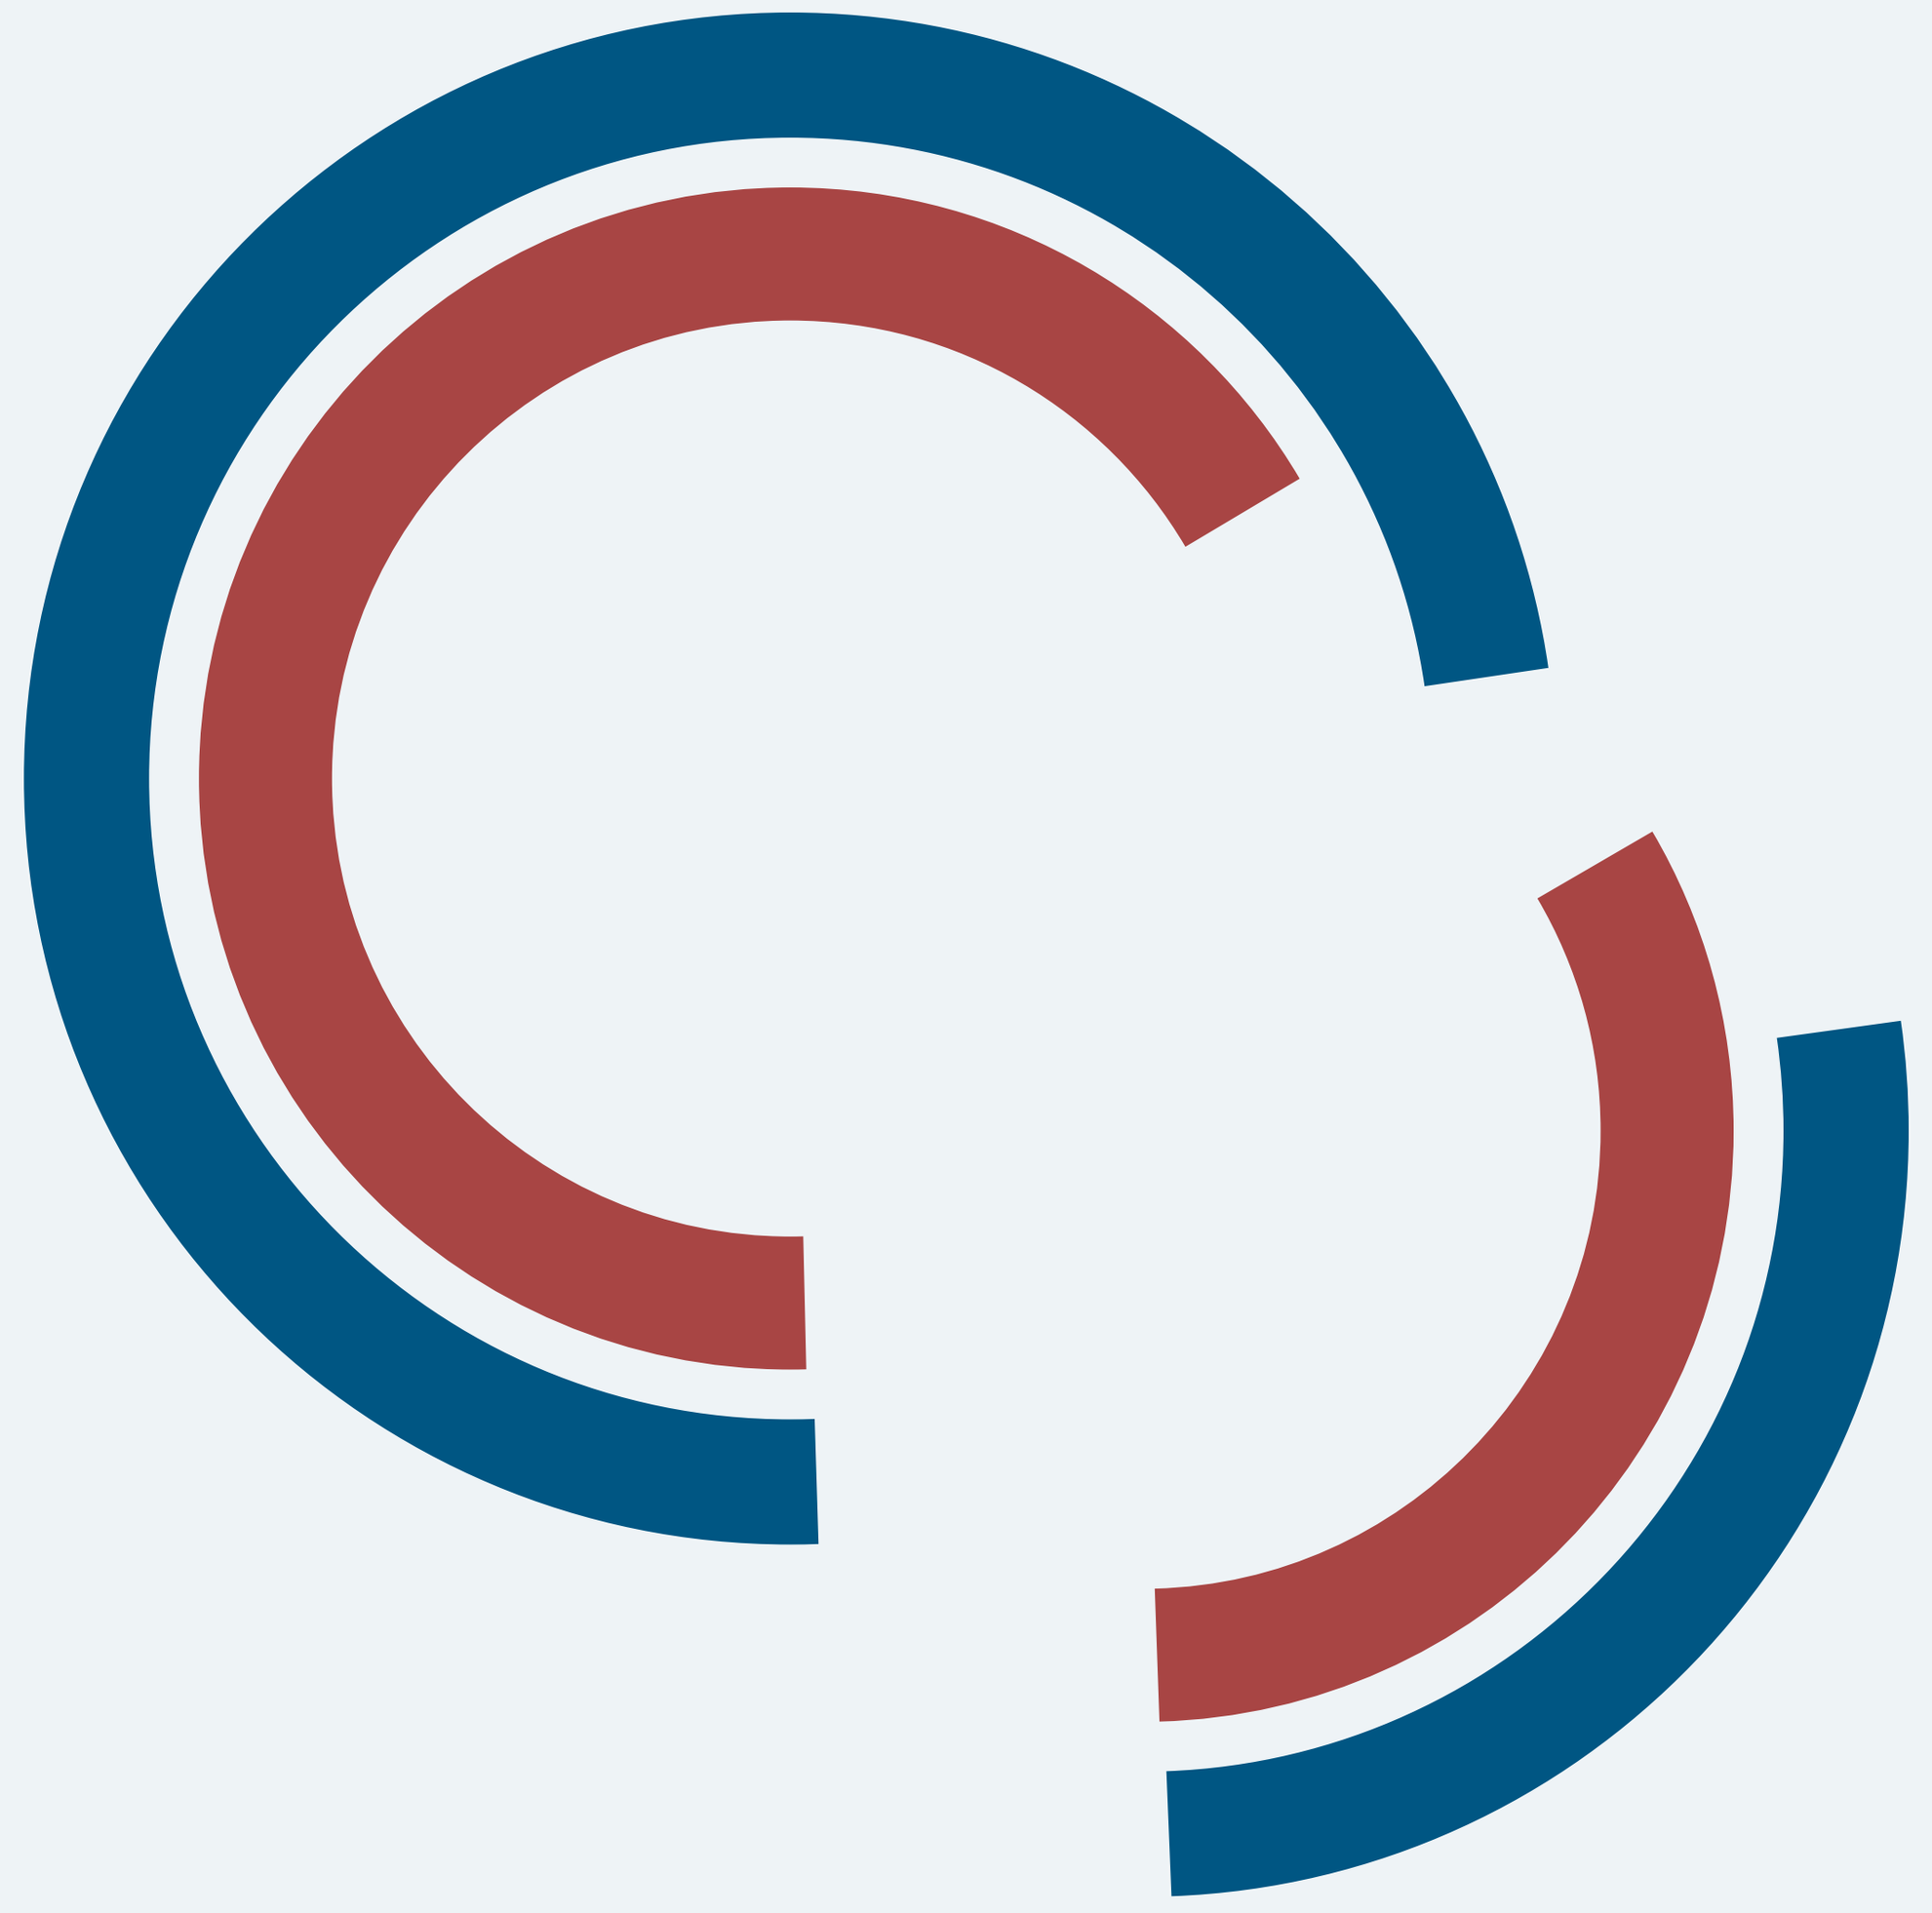

- Among responders at Cycle 1, response rates remained consistently high over subsequent cycles
- Among patients who were MG-ADL responders in Cycle 1, the weighted mean (accounting for varying sample size per cycle) response rate across Cycles 2–13 was 80.1%
- Similarly, among patients who were QMG responders in Cycle 1, the weighted mean response rate was 71.9% across Cycles 2–13

Cycle 1 responders

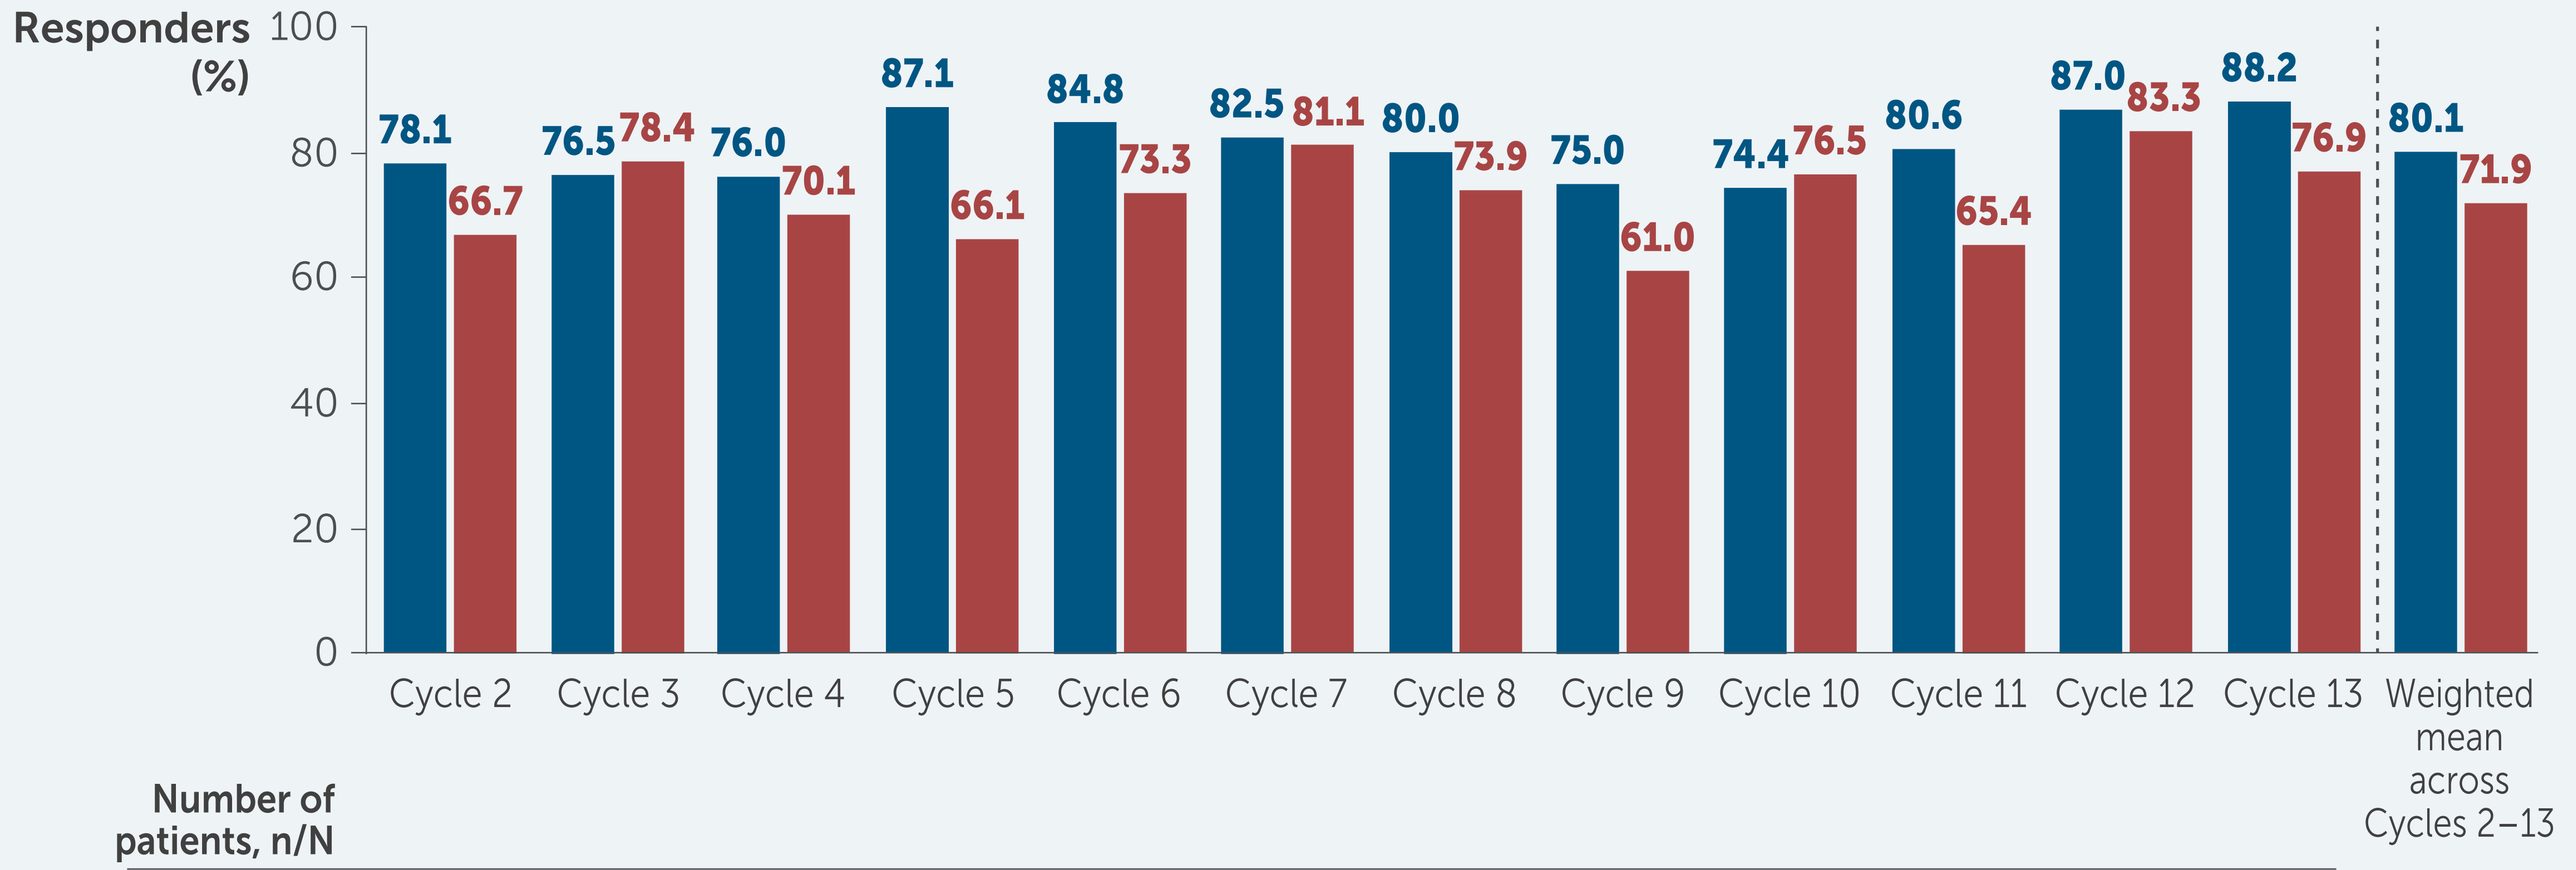
Responders (%)
100
80
60
40
20
0
78.1
66.7
76.5
78.4
76.0
70.1
87.1
66.1
84.8
73.3
82.5
81.1
80.0
73.9
75.0
61.0
74.4
76.5
80.6
65.4
87.0
83.3
88.2
76.9
80.1
71.9
Cycle 2
Cycle 3
Cycle 4
Cycle 5
Cycle 6
Cycle 7
Cycle 8
Cycle 9
Cycle 10
Cycle 11
Cycle 12
Cycle 13
Weighted mean across Cycles 2–13

Number of patients, n/N	Cycle 2	Cycle 3	Cycle 4	Cycle 5	Cycle 6	Cycle 7	Cycle 8	Cycle 9	Cycle 10	Cycle 11	Cycle 12	Cycle 13
MG-ADL	75/96	62/81	57/75	61/70	56/66	47/57	40/50	33/44	29/39	25/31	20/23	15/17
QMG	58/87	58/74	47/67	41/62	44/60	43/53	34/46	25/41	26/34	17/26	15/18	10/13

Cycle 1 non-responders:
MG-ADL: 25.6%
QMG: 31.8%

- Among 33 (25.6%) MG-ADL non-responders at Cycle 1, 63.6% achieved a response at Cycle 2
- Among 41 (31.8%) QMG non-responders at Cycle 1, 50.0% were responders at Cycle 2

Cycle 1 non-responders

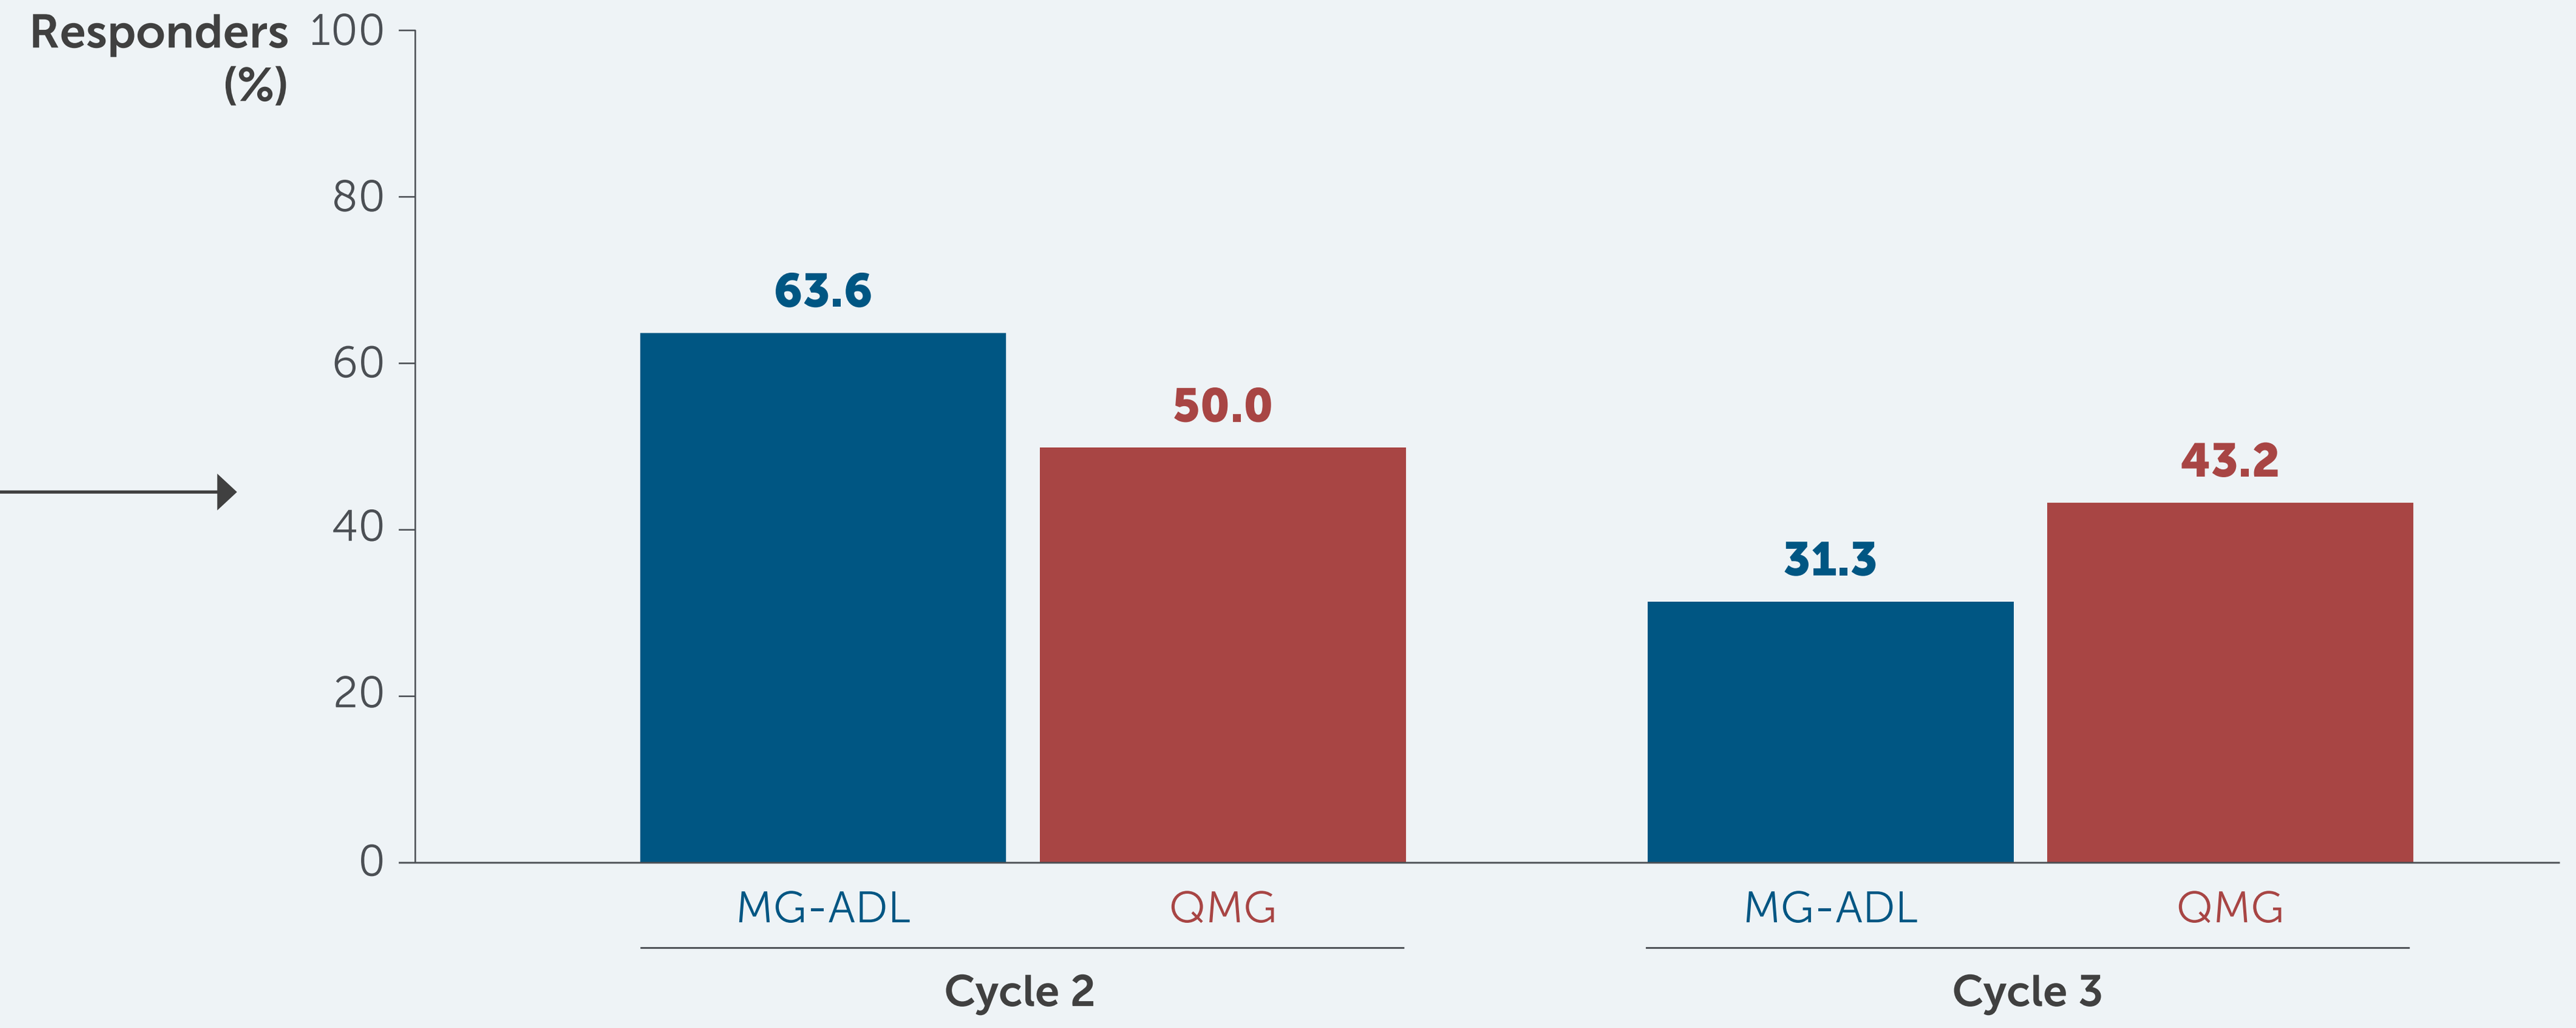
Responders (%)
100
80
60
40
20
0
63.6
50.0
31.3
43.2
MG-ADL
QMG
MG-ADL
QMG
Cycle 2
Cycle 3

Number of patients, n/N	Cycle 2	Cycle 3
MG-ADL	21/33	10/32
QMG	20/40	16/37

N is the number of patients who completed MG-ADL or QMG assessment at Day 43.

Safety

- Final safety data were pooled across MycarinG and MG0007 for patients with ≥1 treatment cycle
- Over a median follow-up of 2.0 years, 93.1% (175/188) of patients experienced a TEAE; most were mild or moderate
 - The most common TEAEs were headache (50.0%), diarrhea (33.5%), COVID-19 (21.8%) and pyrexia (20.7%)

Summary and conclusions

This pooled analysis of data from the Phase 3 MycarinG study and OLEs assessed the response across repeated cycles of rozanolixizumab treatment among Cycle 1 responders and non-responders

Among Cycle 1 responders, MG-ADL and QMG response rates remained consistently high across subsequent cycles

Among Cycle 1 MG-ADL non-responders who received another cycle, most achieved a response in Cycle 2

This *post hoc* analysis showed that Cycle 1 responders had generally consistent response rates in subsequent cycles of rozanolixizumab and indicated that initial non-responders may benefit from additional rozanolixizumab treatment cycles

Abbreviations: Ab+, antibody positive; AChR, acetylcholine receptor; COVID-19, coronavirus disease 2019; CS, corticosteroid; FcRn, neonatal fragment crystallizable receptor; gMG, generalized myasthenia gravis; IgG4, immunoglobulin G subunit 4; mAb, monoclonal antibody; MG, myasthenia gravis; MG-ADL, Myasthenia Gravis Activities of Daily Living; MGFA, Myasthenia Gravis Foundation of America; MuSK, muscle-specific tyrosine kinase; OLE, open-label extension; QMG, Quantitative Myasthenia Gravis; RLZ, rozanolixizumab; SD, standard deviation; TEAE, treatment-emergent adverse event.

Acknowledgments: This study was funded by UCB. The authors acknowledge Qudsia Ehsan, BSc, of Ogilvy Health, London, UK, for editorial assistance, which was funded by UCB. The authors thank Veronica Porkess, PhD, of UCB for publication and editorial support. The authors thank the patients and their caregivers, in addition to the investigators and their teams who contributed to this study.

Author disclosures: Robert M. Pascuzzi is Professor Emeritus of Neurology at Indiana University and receives compensation for his professional work from Indiana University Health. He has no financial relationship with any pharmaceutical company and receives no compensation from any pharmaceutical company (present or past). Robert M. Pascuzzi speaks at educational seminars on a broad variety of general neurology topics for primary care physicians through the organisation Medical Education Resources (an educational organisation with no links or ties to any pharmaceutical or healthcare business company). Therefore, Robert M. Pascuzzi has no conflicts of interest related to this research, manuscript, presentation or publication. Ali A. Habib has received research support and/or honoraria from Alexion/AstraZeneca Rare Disease, Amgen, Arcellx, argenx, Cabaletta Bio, Cartesian Therapeutics, COUR Pharmaceuticals, Genentech/Roche, Grifols, Immunovant, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Kyverna, Merck, MGNet, Nkarta, NMD Pharma, Novartis, Regeneron Pharmaceuticals and UCB. Zabeen K. Mahuwala has received compensation for advisory board participation from Alexion Pharmaceuticals and Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine) and has served as a panellist for Academic CME. She has received research funding for clinical trials paid to the University of Kentucky from Alexion Pharmaceuticals, argenx, Immunovant, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), RemeGen Biosciences and UCB. Renato Mantegazza has received funding for travel and meeting attendance or advisory board participation from Alexion Pharmaceuticals, argenx, BioMarin, Catalyst, Regeneron Pharmaceuticals, Sanofi and UCB. Sabrina Sacconi has nothing to disclose. John Vissing has been a Consultant on advisory boards for Amicus Therapeutics, Biogen, Edgewise Therapeutics, Fulcrum Therapeutics, Genethon, Horizon Therapeutics (now Amgen), Lupin, ML Biopharma, Novartis, Regeneron Pharmaceuticals, Roche, Sanofi Genzyme (now Sanofi), Sarepta Therapeutics and UCB. He has received research and travel support and/or speaker honoraria from Alexion Pharmaceuticals, argenx, Biogen, Edgewise Therapeutics, Fulcrum Therapeutics, Lupin, Sanofi Genzyme (now Sanofi) and UCB. He is a Principal Investigator in clinical trials for Alexion Pharmaceuticals, argenx, Atamyo Therapeutics, Genethon, Horizon Therapeutics (now Amgen), Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), ML Biopharma, Novartis, Regeneron Pharmaceuticals, Roche, Sanofi Genzyme (now Sanofi) and UCB. Tuan Vu is the USF Site Principal Investigator for MG clinical trials sponsored by Alexion/AstraZeneca Rare Disease, Amgen, argenx, Cartesian Therapeutics, COUR Pharmaceuticals, Dianthus Therapeutics, ImmunAbs, Immunovant, Johnson & Johnson, NMD Pharma, Regeneron Pharmaceuticals, UCB and Vor, and has served as a speaker for Alexion/AstraZeneca Rare Disease, Amgen, argenx and CSL Behring. He has performed consulting work for Alexion/AstraZeneca Rare Disease, argenx, Dianthus Therapeutics, NMD Pharma, Regeneron and Vor. Joel Baldwin is an employee of Cogent Skills Limited working for UCB. Thaïs Tarancón is an employee and shareholder of UCB. Vera Bril is a Consultant for Akcea, Alexion Pharmaceuticals, Alnylam, argenx, CSL, Grifols, Immunovant, Ionis, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Momenta (now Johnson & Johnson), Novo Nordisk, Octapharma, Pfizer, Powell Mansfield, Roche, Sanofi, Takeda Pharmaceuticals and UCB. She has received research support from Akcea, Alexion Pharmaceuticals, argenx, CSL, Grifols, Immunovant, Ionis, Momenta (now Johnson & Johnson), Octapharma, Takeda Pharmaceuticals, UCB and Viela Bio (now Amgen).

References: 1. Gilhus NE, et al. Nat Rev Dis Primers. 2019;5(1):30. 2. Bril V, et al. Lancet Neurol. 2023;22(5):383–394. 3. Bril V, et al. J Neuromuscul Dis. 2025;12(2):218–230. 4. RYSTIGGO® US PI. https://www.ucb-usa.com/RYSTIGGO-prescribing-information.pdf. Accessed March 2026.

Please use this QR code to download a PDF of the poster.