Bimekizumab Treatment Resulted in Sustained Improvements to 3.7 Years in Routine Assessment of Patient Index Data 3 in Psoriatic Arthritis

Arthur Kavanaugh,[1] William Tillett,[2,3] Proton Rahman,[4] Patrick Healy,[5] Georgina Apostoli,[6] Jérémy Lambert,[7] M. Elaine Husni[8]

Presenter: Mac McLaughlin[9]

[1]Division of Rheumatology, Autoimmunity and Inflammation, UC San Diego School of Medicine, La Jolla, CA, USA; [2]Royal National Hospital of Rheumatic Diseases, Bath, UK; [3]Department of Life Sciences, Centre for Therapeutic Innovation, University of Bath, Bath, UK; [4]Division of Rheumatology, Craig L. Dobbin Genetics Research Centre, Discipline of Medicine, Memorial University of Newfoundland, St John's, NL, Canada; [5]UCB, Morrisville, NC, USA; [6]UCB, Slough, UK; [7]UCB, Courbevoie, France; [8]Department of Rheumatic and Immunologic Diseases, Cleveland Clinic, Cleveland, OH, USA; [9]UCB, Smyrna, GA, USA.

Objective

To report Routine Assessment of Patient Index Data 3 (RAPID3) responses in bimekizumab (BKZ)-treated patients with active psoriatic arthritis (PsA) from the phase 3 BE OPTIMAL trial to Week 192 (3.7 years; end of trial).

Background

- RAPID3 is a composite outcome measure of 3 patient-reported outcomes used to assess PsA disease activity.[1]
- The feasibility of use in routine practice and ability to capture the multidimensional patient experience of PsA make RAPID3 a clinically relevant measure of treatment response.[1]
- Treatment with bimekizumab (BKZ), a monoclonal IgG1 antibody that selectively inhibits interleukin (IL)-17F in addition to IL-17A, has demonstrated improved RAPID3 responses to 3 years.[2]

Methods

- BE OPTIMAL (NCT03895203) assessed subcutaneous BKZ 160 mg every 4 weeks (Q4W) in patients with active PsA who were biologic disease-modifying antirheumatic drug (bDMARD)-naïve.
- BE OPTIMAL was placebo (PBO)-controlled to Week 16, when PBO-randomized patients switched to BKZ (PBO/BKZ). Patients who completed Week 52 were eligible to enter BE VITAL (NCT04009499; open-label extension), in which all patients received BKZ 160 mg Q4W.
- RAPID3 was calculated using the Health Assessment Questionnaire-Disability Index, Patient's Assessment of Arthritis Pain Visual Analog Scale, and Patient's Global Assessment of Psoriatic Arthritis. Each measure was rescaled to 0–10 and combined for a total score of 0–30 (higher scores indicate higher disease activity).[1]
- RAPID3 outcomes reported to Week 192 include change from baseline (CfB), minimal clinically important difference (MCID; ≥3.8-point decrease from baseline in patient with baseline score ≥3.8),[3] and disease activity state (high [>12], moderate [>6–≤12], low [LDA; >3–≤6], or remission [REM; ≤3]).[1]
- Data are reported for PBO/BKZ and BKZ-randomized patients from baseline to Week 52, and for BKZ Total (PBO/BKZ and BKZ-randomized) patients from Week 52 to Week 192.
- Data are reported as observed case and using modified non-responder or multiple imputation (mNRI/MI). mNRI considered all visits following discontinuation due to adverse events or lack of efficacy as non-response; all other missing data imputed with MI and response derived from imputed values.

Results

- Overall, 533/712 (74.9%) BKZ Total patients completed Week 192.
- Rapid improvements in RAPID3 were observed in BKZ-treated patients at Week 16; improvements in RAPID3 in BKZ Total patients were sustained from Week 52 to Week 192 (**Figure 1A**).
- A high proportion of BKZ-treated patients achieved RAPID3 MCID at Week 16. Among BKZ Total patients, a high proportion achieved RAPID3 MCID at Week 52 which was sustained to Week 192 (**Figure 1B**).
- A similar trend was observed for RAPID3 disease states, where the proportion of BKZ Total patients achieving REM or LDA was sustained from Week 52 (58.1%) to Week 192 (56.7%; MI; **Figure 2**).

Conclusions

End-of-trial results from BE OPTIMAL showed that a high proportion of bDMARD-naïve patients with active PsA treated with bimekizumab achieved RAPID3 low disease activity or remission as early as Week 16, and this was sustained to Week 192 (3.7 years). These results highlight the deep and sustained improvement in patient-reported disease activity and symptom burden with the dual inhibition of IL-17A and IL-17F.

Summary

This post hoc analysis examined responses of **RAPID3**, a composite patient-reported outcome, in patients with active psoriatic arthritis treated with bimekizumab to the **end of the phase 3 BE OPTIMAL trial** (3.7 years).

RAPID3	Physical function (HAQ-DI)	Pain	Patient's global assessment

74.9% BKZ Total patients **completed Week 192**.

Rapid improvements in RAPID3 were observed by **Week 16**, which were sustained to **Week 192 (3.7 years)**:

Mean **change from baseline** (MI)

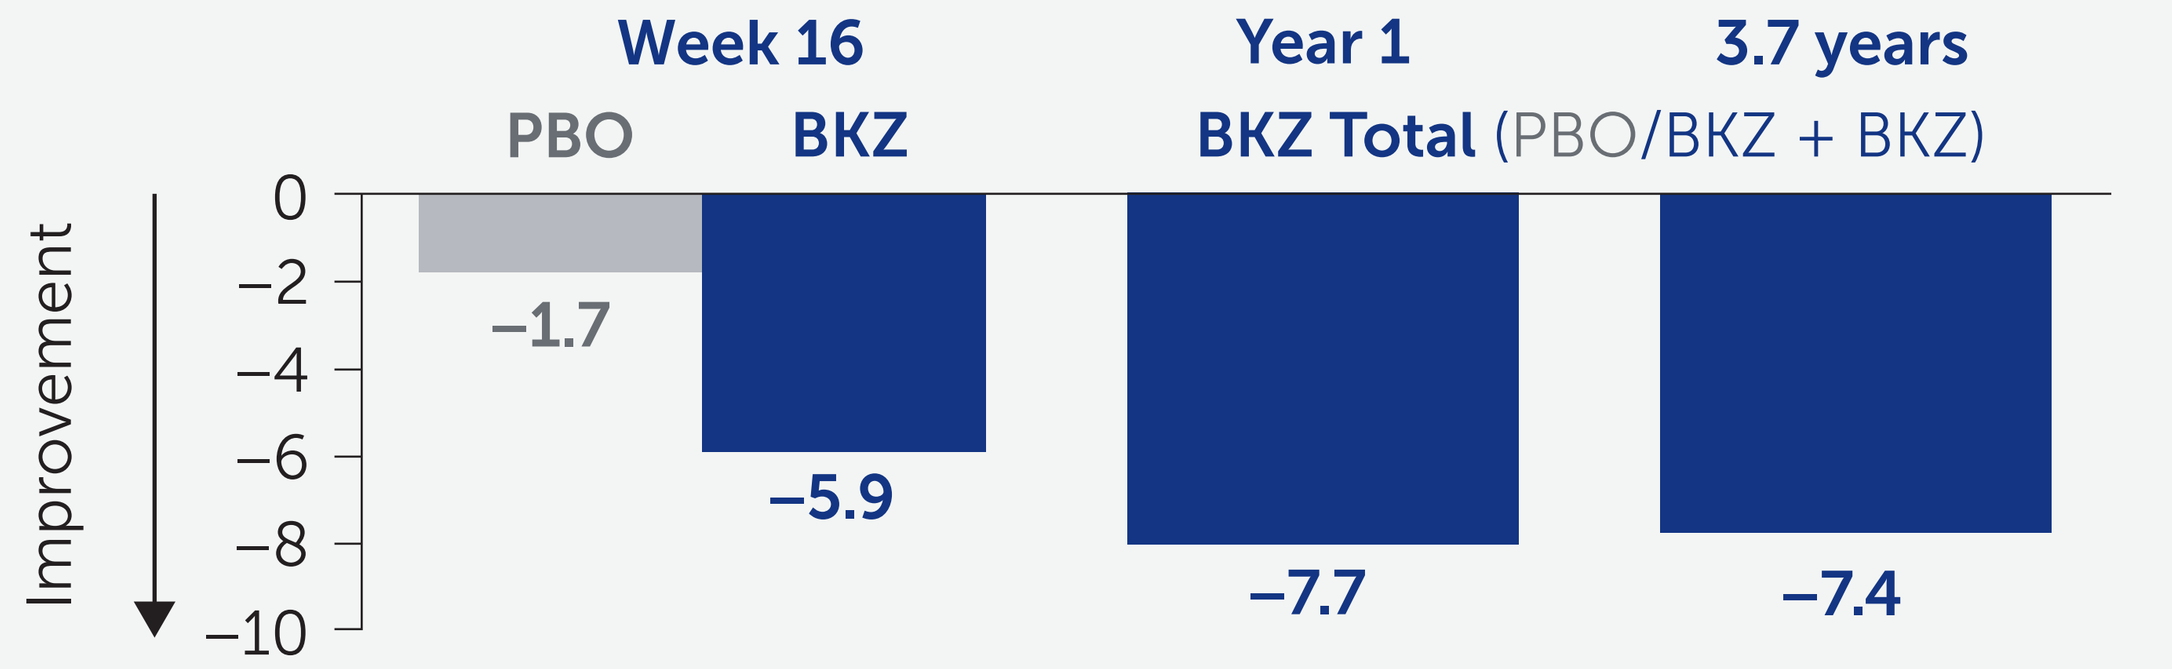
Week 16
PBO
BKZ
Year 1
3.7 years
BKZ Total (PBO/BKZ + BKZ)
Improvement
0
−2
−4
−6
−8
−10
−1.7
−5.9
−7.7
−7.4

Proportion of patients achieving **MCID** (mNRI)

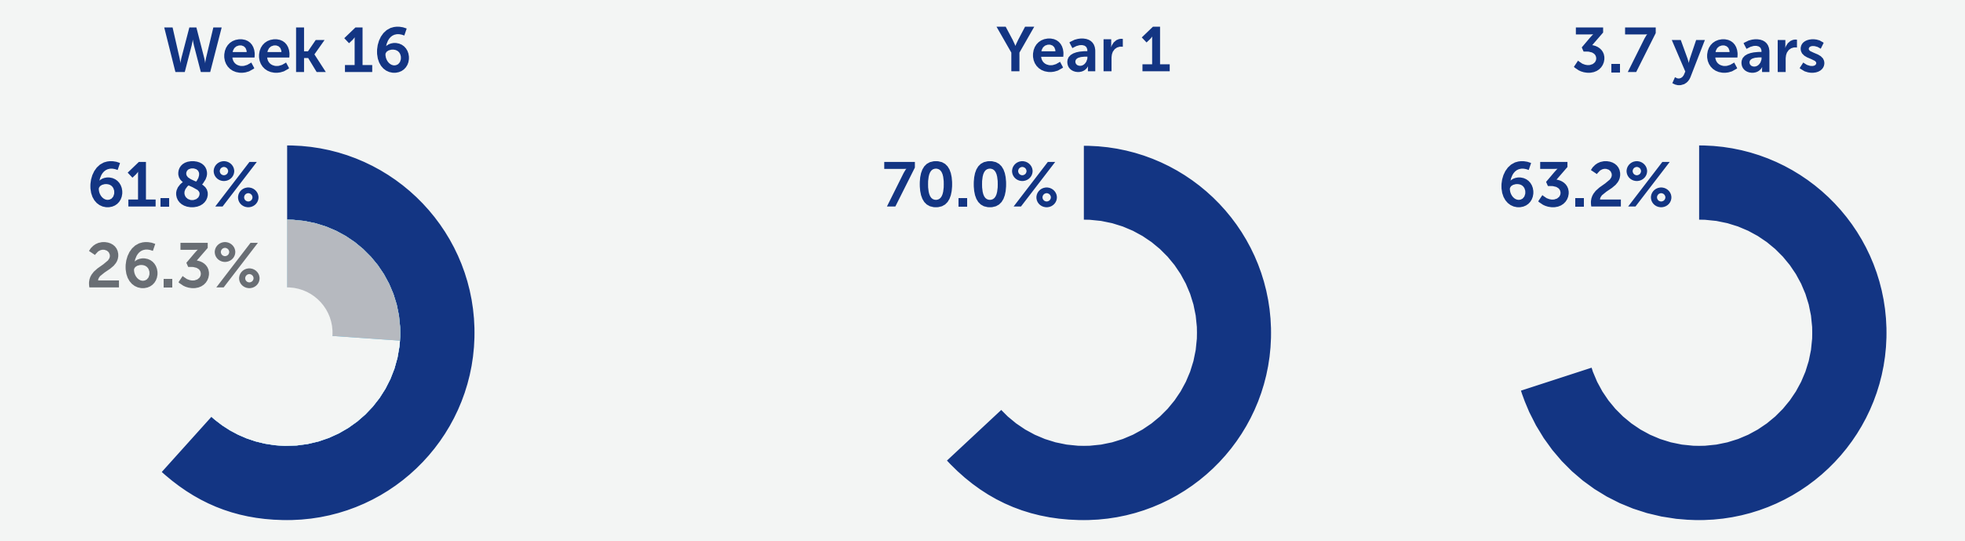
Week 16
Year 1
3.7 years
61.8%
26.3%
70.0%
63.2%

Proportion of patients achieving **remission or low disease acitivity state** (MI)

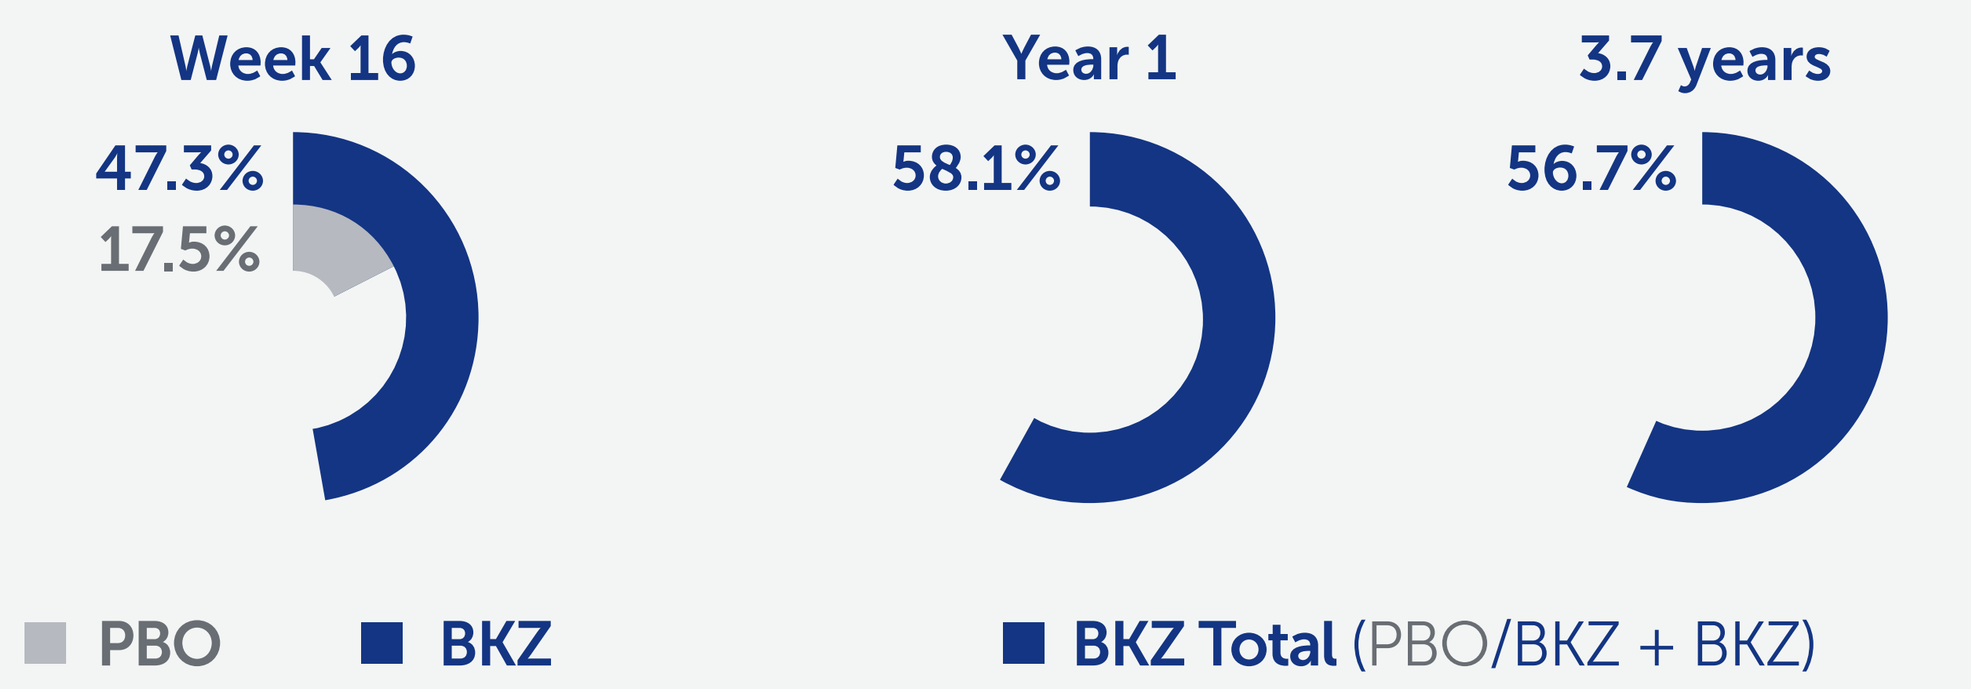
Week 16
Year 1
3.7 years
47.3%
17.5%
58.1%
56.7%
PBO
BKZ
BKZ Total (PBO/BKZ + BKZ)

Bimekizumab treatment was associated with **sustained, clinically meaningful improvements** in patient-reported disease activity and symptom burden as measured by **RAPID3 to 3.7 years**.

Figure 1 (A) Mean change from baseline and (B) proportion of patients achieving MCID in RAPID3 to Week 192 (MI, mNRI, OC)

A) Mean change from baseline (MI, OC)

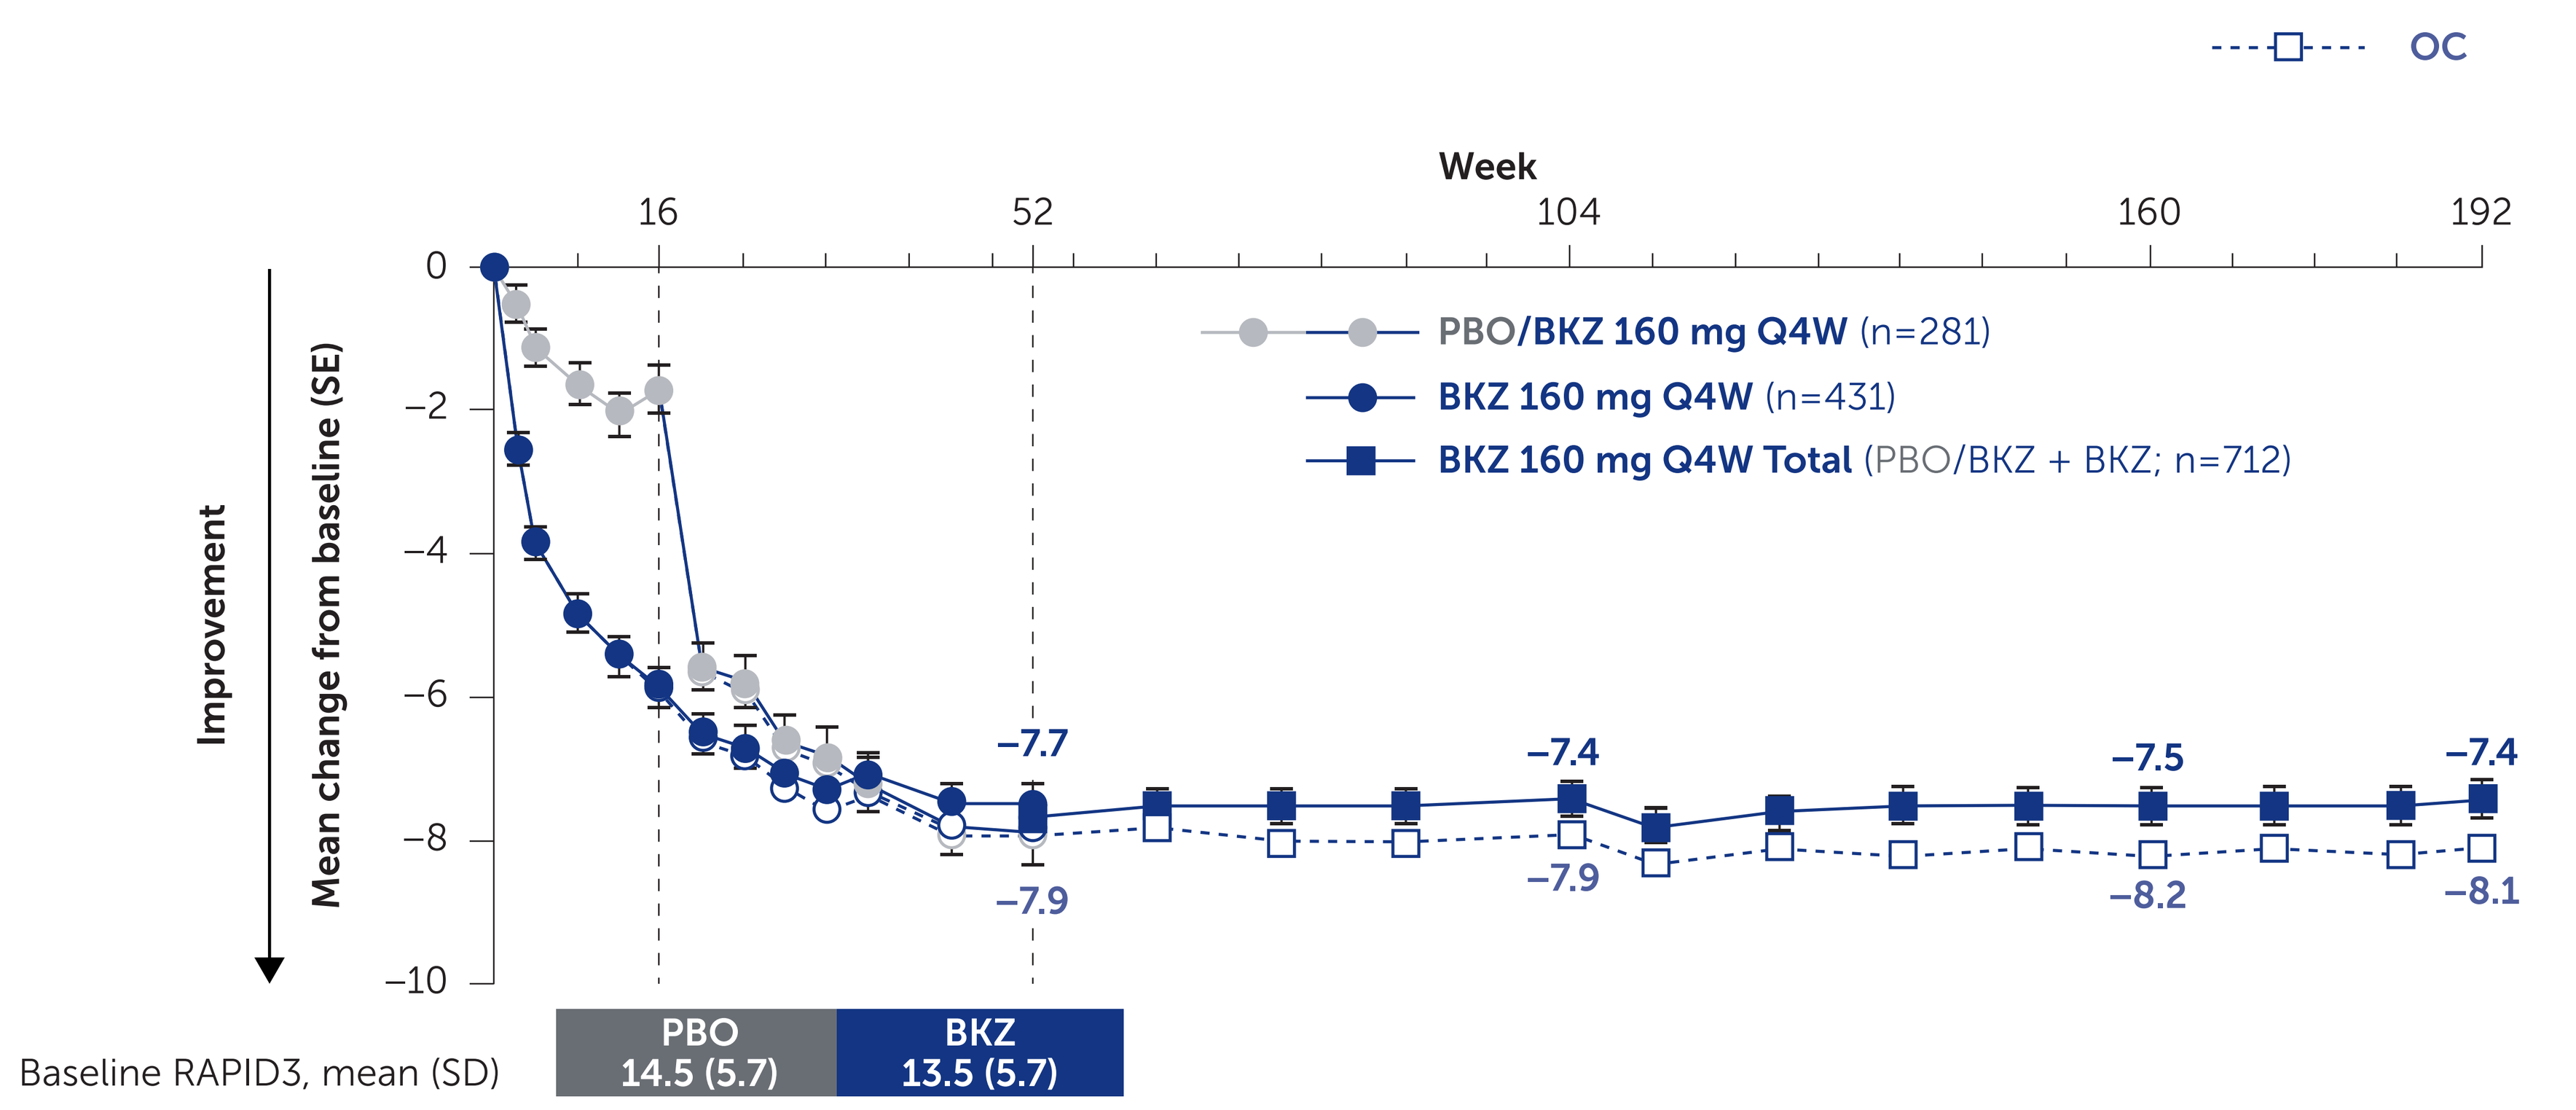
OC
MI/mNRI
Week
16
52
104
160
192
0
−2
−4
−6
−8
−10
Improvement
Mean change from baseline (SE)
PBO/BKZ 160 mg Q4W (n=281)
BKZ 160 mg Q4W (n=431)
BKZ 160 mg Q4W Total (PBO/BKZ + BKZ; n=712)
−7.7
−7.9
−7.4
−7.9
−7.5
−8.2
−7.4
−8.1
Baseline RAPID3, mean (SD)
PBO 14.5 (5.7)
BKZ 13.5 (5.7)

Week, n (OC)	Baseline	52	104	160	192
PBO/BKZ	281	254			
BKZ	430	381			
BKZ Total	711	635	581	530	515

B) MCID[a] (mNRI, OC)[b]

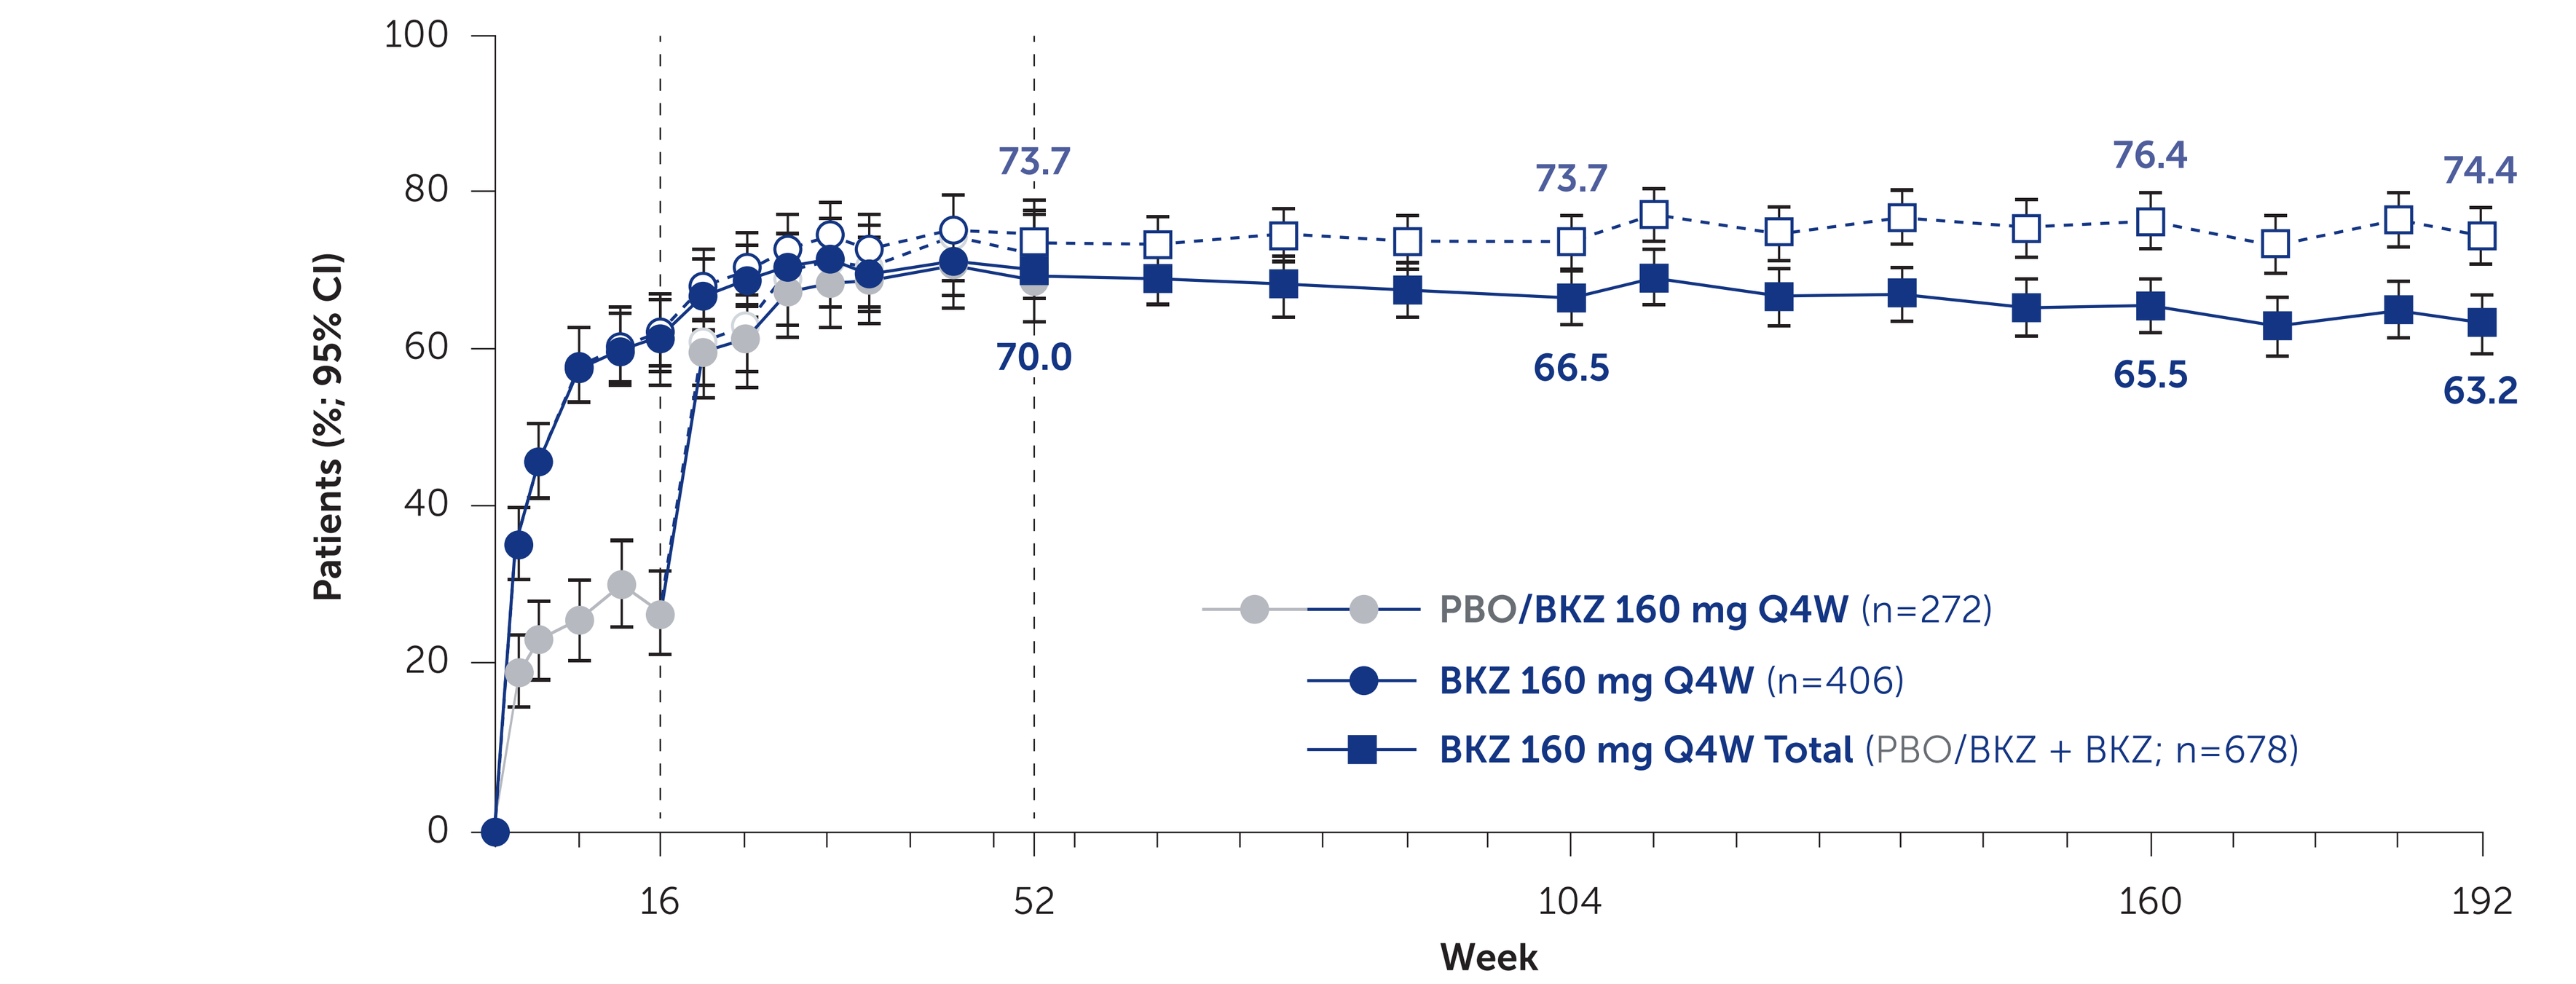
100
80
60
40
20
0
Patients (%; 95% CI)
73.7
70.0
73.7
66.5
76.4
65.5
74.4
63.2
PBO/BKZ 160 mg Q4W (n=272)
BKZ 160 mg Q4W (n=406)
BKZ 160 mg Q4W Total (PBO/BKZ + BKZ; n=678)
16
52
104
160
192
Week

Week, n/N (OC)	Baseline	52	104	160	192
PBO/BKZ	272	177/245			
BKZ	406	272/364			
BKZ Total	678	449/609	410/556	388/508	367/493

Randomized set. **[a]** RAPID3 MCID defined as ≥3.8-point decrease from baseline in patients with a RAPID3 score ≥3.8 at baseline;[3] **[b]** mNRI considered all visits following discontinuation due to adverse events or lack of efficacy as non-response; all other missing data were imputed with MI and the response derived from imputed values.

Figure 2 RAPID3 disease activity states to Week 192 (MI)

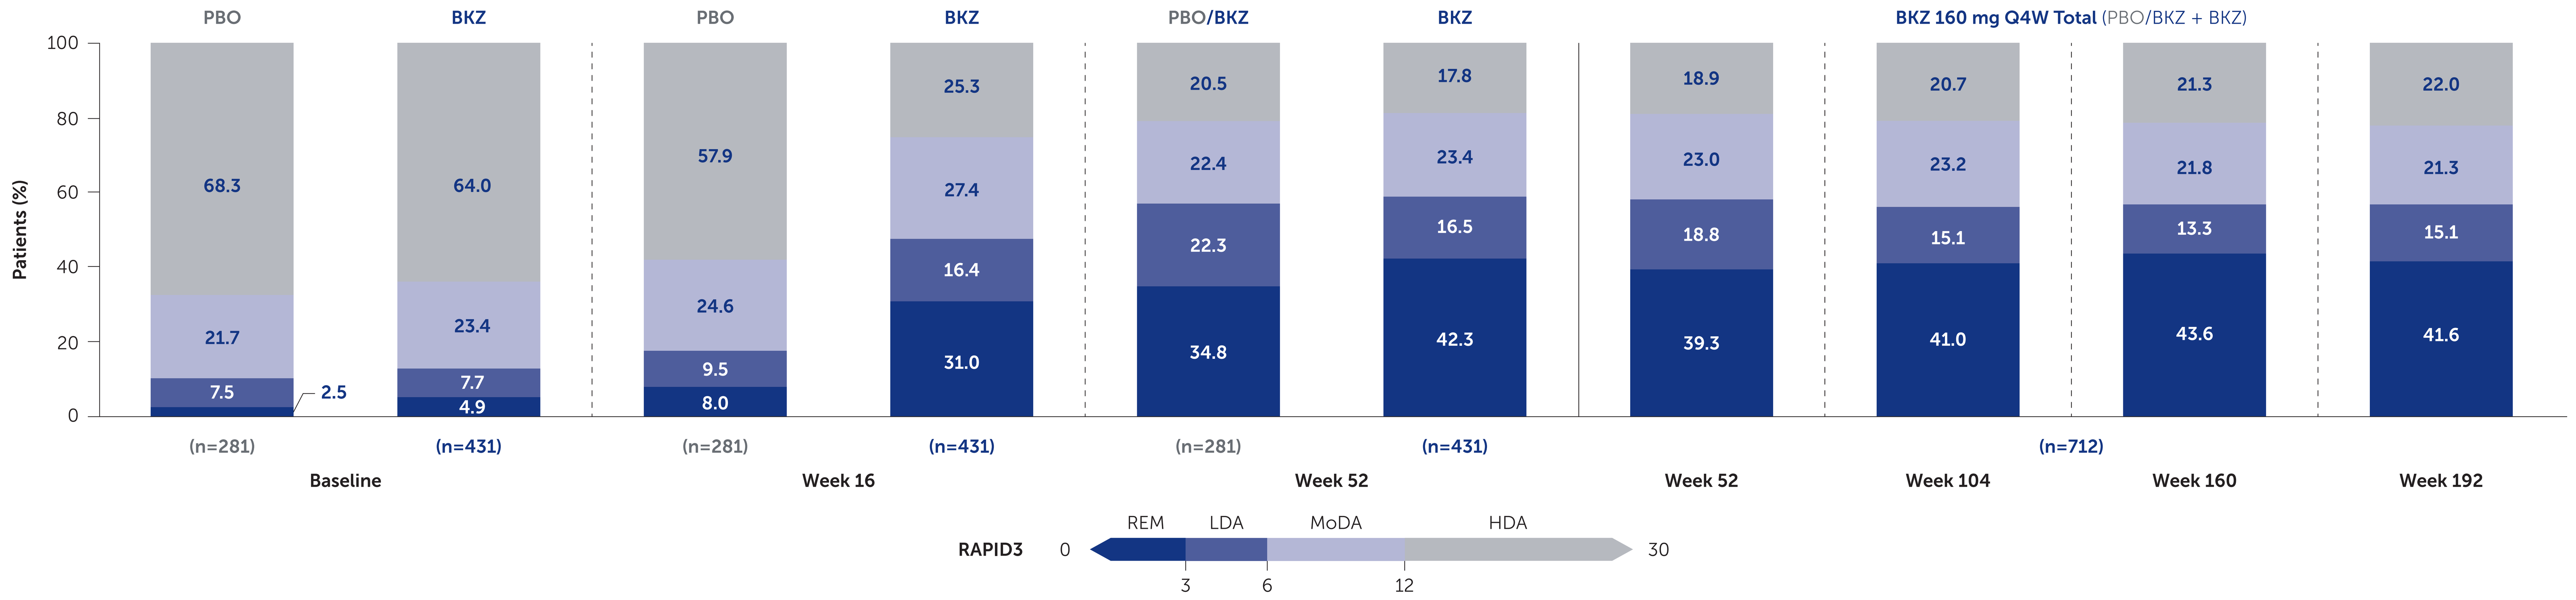
PBO
BKZ
PBO
BKZ
PBO/BKZ
BKZ
BKZ 160 mg Q4W Total (PBO/BKZ + BKZ)
100
80
60
40
20
0
Patients (%)
68.3
21.7
7.5
2.5
64.0
23.4
7.7
4.9
57.9
24.6
9.5
8.0
25.3
27.4
16.4
31.0
20.5
22.4
22.3
34.8
17.8
23.4
16.5
42.3
18.9
23.0
18.8
39.3
20.7
23.2
15.1
41.0
21.3
21.8
13.3
43.6
22.0
21.3
15.1
41.6
(n=281)
(n=431)
(n=281)
(n=431)
(n=281)
(n=431)
(n=712)
Baseline
Week 16
Week 52
Week 52
Week 104
Week 160
Week 192
REM
LDA
MoDA
HDA
RAPID3
0
3
6
12
30

Randomized set. Disease activity state categorized as high (>12), moderate (>6–≤12), low (>3–≤6), or remission (≤3).[1]

bDMARD: biologic disease-modifying antirheumatic drug; **BKZ:** bimekizumab; **CfB:** change from baseline; **CI:** confidence interval; **HDA:** high disease activity; **IL:** interleukin; **LDA:** low disease activity; **MCID:** minimal clinically important difference; **MI:** multiple imputation; **mNRI:** modified non-responder imputation; **MoDA:** moderate disease activity; **OC:** observed case; **PBO:** placebo; **PsA:** psoriatic arthritis; **Q4W:** every 4 weeks; **RAPID3:** Routine Assessment of Patient Index Data 3; **REM:** remission; **SD:** standard deviation; **SE:** standard error.

References: [1]Coates LC. Arthritis Care Res 2018;70:1198–205; [2]Kavanaugh A. CCR-East 2026. Presented 1 May 2026; [3]Ward MM. J Rheumatol 2019;46:27–30.
Author Contributions: Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **AK**, **WT**, **PR**, **PH**, **GA**, **JL**, **MEH**; Drafting of the publication, or reviewing it critically for important intellectual content: **AK**, **WT**, **PR**, **PH**, **GA**, **JL**, **MEH**; Final approval of the publication: **AK**, **WT**, **PR**, **PH**, **GA**, **JL**, **MEH**. **Author Disclosures: AK:** Research grants and/or consulting fees from Amgen, BMS, Eli Lilly and Company, Janssen, Moonlake, Pfizer, Takeda, and UCB; **WT:** Research grants, consulting fees, speaking fees and/or honoraria from AbbVie, Amgen, BMS, Celgene, Eli Lilly and Company, GSK, Janssen, MSD, Novartis, Ono Pharma, Pfizer, Takeda, and UCB; **PR:** Consulting fees from Abbott, AbbVie, Amgen, Bristol Myers Squibb, Celgene, Eli Lilly and Company, Janssen, Novartis, Pfizer, and UCB; received research grants from Janssen and Novartis; **PH**, **JL:** Employee and shareholder of UCB; **GA:** Employee of UCB; shareholder of UCB and Vertex Pharmaceuticals; **MEH:** Advisory board member and consultant for AbbVie, Amgen, BMS, Eli Lilly, Janssen, Novartis, Pfizer, and UCB; **MM:** Employee of UCB. **Acknowledgments:** The authors thank the patients, the investigators, and their teams who took part in these studies. The authors acknowledge Lyes Derouchie, PhD, of UCB, for editorial review during poster development and publication coordination, Becky Sainty, PhD, of Costello Medical, Cambridge, UK for medical writing and editorial assistance, and the Costello Medical Creative team for design support. These studies were funded by UCB. All costs associated with development of this presentation were funded by UCB.

To receive a copy of this poster, scan the QR code.
Link expiration:
19 December 2026

Presented at CCR West 2026 | 17–20 September | Huntington Beach, USA