AANEM Poster **198** | MGFA Scientific Session Poster **96**

Rozanolixizumab is not detectable in breast milk after a single dose in healthy, lactating women: Results from an open-label, Phase 1 study

Babak Boroojerdi[1], Marion Boehnlein[1], François Bonfitto[2], Timothy Mutsvari[3], Andreea Lavrov[1], Jagdev Sidhu[4]

[1]UCB, Monheim, Germany; [2]UCB, Brussels, Belgium; [3]UCB, Braine-L'alleud, Belgium; [4]UCB, Melbourne, Australia

AANEM & MGFA Scientific Session 2026, Orlando, FL, USA; September 29–October 2, 2026

Introduction

- Rozanolixizumab is a humanized IgG4 monoclonal antibody that binds with high affinity to FcRn and is approved for the treatment of adults with gMG[1,2]
- The incidence of gMG in women increases between the ages of 20 and 39 and thus impacts women of childbearing age[3]
- Information about the transfer of rozanolixizumab into breast milk would be useful for women with IgG autoantibody-mediated conditions (e.g., gMG) who are considering breastfeeding, and their physicians
- **The aim of the UP0141 study (NCT06720714) was to assess the concentration of rozanolixizumab in the mature breast milk of healthy, lactating women, including the estimated daily infant dosage and relative infant dose exposure**

Methods

- This study included healthy women who were ≥6 weeks postpartum and who had decided, independently, to stop breastfeeding before details of the study were shared with them (**Figure 1**)
- Participants received a single, weight-tiered dose of rozanolixizumab, equivalent to the approved label in the USA[2]
 - All milk expressed over 7 days post-dose was collected
 - Participants agreed not to resume breastfeeding and to discard any milk collected during the 8-week follow-up period
- The primary endpoint was the concentration of rozanolixizumab in breast milk over 7 days
- The secondary PK endpoints were the estimated daily infant dose and the relative infant dose of rozanolixizumab from breast milk
- Maternal plasma rozanolixizumab PK parameters were also measured: $AUC_{(0-7d)}$, C_{max} and t_{max}
- The secondary safety endpoint was incidence of TEAEs from Day 1 through the safety follow-up visit

Results

- Fifteen participants received a single dose of rozanolixizumab and completed the study
 - Mean (SD) age was 30.5 (4.6) years
 - Mean (SD) BMI on Day 1 was 26.7 (5.1) kg/m^2
- Among all participants at all time points, rozanolixizumab was not detectable in breast milk using a sensitive, validated assay (LLOQ of 0.05 µg/mL) (**Figure 2**)
 - Therefore, the relative infant dose was not estimable
- The PK profile for rozanolixizumab in maternal plasma is shown in **Table 1**
 - Median t_{max} was 71.9 hours
 - PK findings were consistent with previous healthy volunteer studies[4]
- Overall, 11/15 (73.3%) participants reported TEAEs, all of which were non-serious (**Table 2**)
 - The most common TEAE was headache (9/15 [60.0%] participants)

Summary and conclusions

This study assessed the concentration of rozanolixizumab in the breast milk of healthy, lactating women

In healthy, lactating women, rozanolixizumab was not excreted into breast milk in measurable quantities over 7 days after a single dose

The plasma concentrations and safety profile of rozanolixizumab were consistent with previously reported data

Although based on a single-dose study, the undetectable levels of rozanolixizumab in breast milk suggest that the potential for infant drug exposure during nursing is considered low

Figure 1 UP0141 was a single-dose, post-marketing, open-label, Phase 1 study of rozanolixizumab in healthy, lactating women

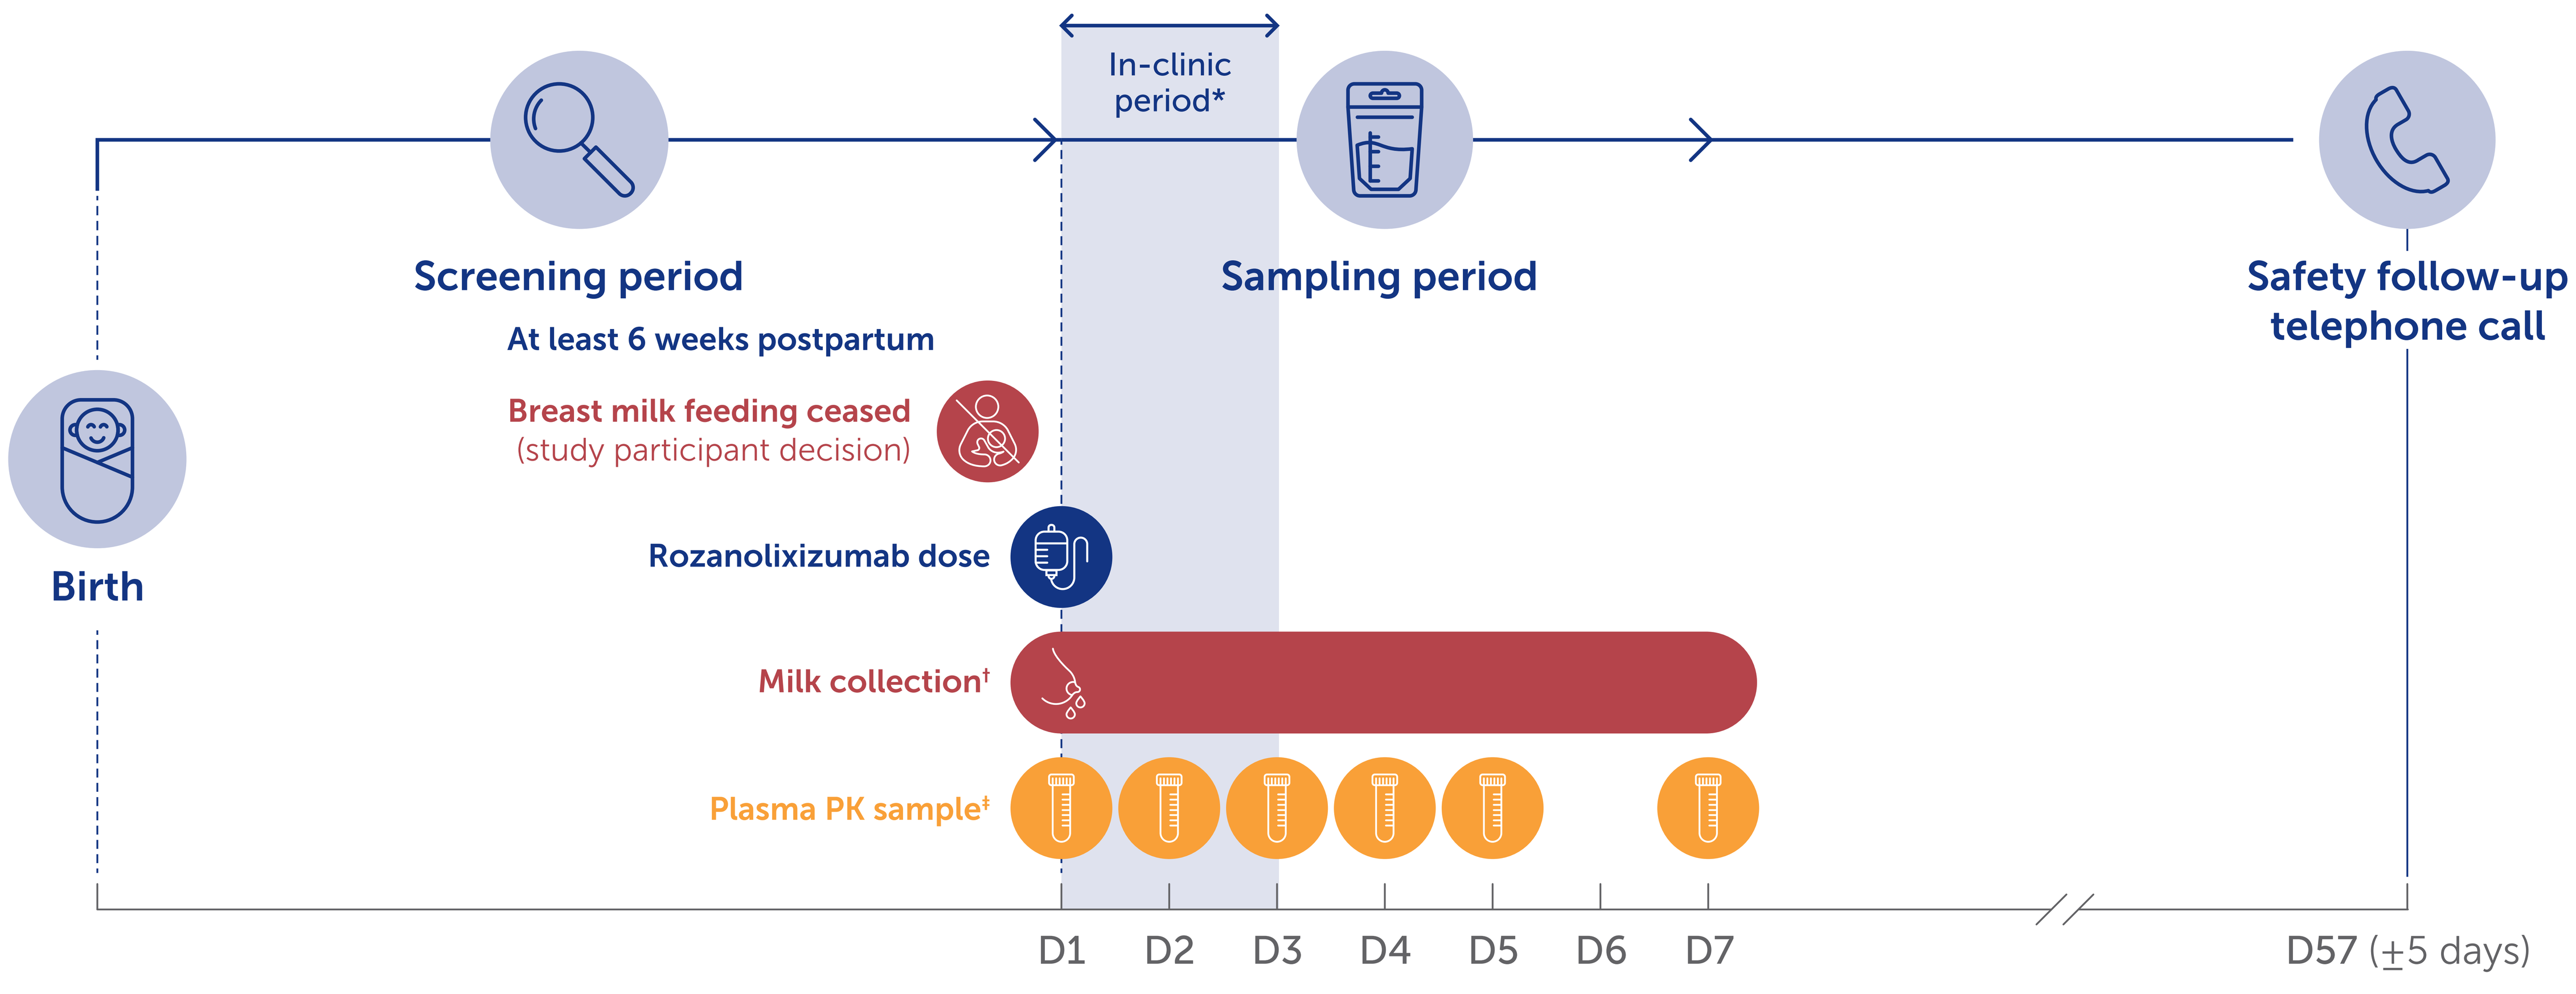
In-clinic period*
Screening period
At least 6 weeks postpartum
Sampling period
Safety follow-up telephone call
Birth
Breast milk feeding ceased
(study participant decision)
Rozanolixizumab dose
Milk collection†
Plasma PK sample‡
D1
D2
D3
D4
D5
D6
D7
D57 (±5 days)

*Participants were admitted on the evening of Day −1 (the day before D1) or during the early morning of D1; they were discharged after D3 assessments were completed. †Breast milk collection and sampling were performed within 30 minutes pre-dose and at the following intervals after infusion (hours): 0 to ≤3, >3 to ≤6, >6 to ≤9, >9 to ≤12, >12 to ≤24, >24 to ≤36, >36 to ≤48, >48 to ≤60, >60 to ≤72, >72 to ≤84, >84 to ≤96, >96 to ≤108, >108 to ≤120, >120 to ≤132, and >132 to ≤144. ‡Blood sampling was performed within 60 minutes pre-dose and at the following intervals after infusion (hours): 6, 9, 12, 24, 48, 72, 96, and 144.

Figure 2 Rozanolixizumab was undetectable in breast milk while maternal plasma concentrations peaked at a median of 71.9 hours

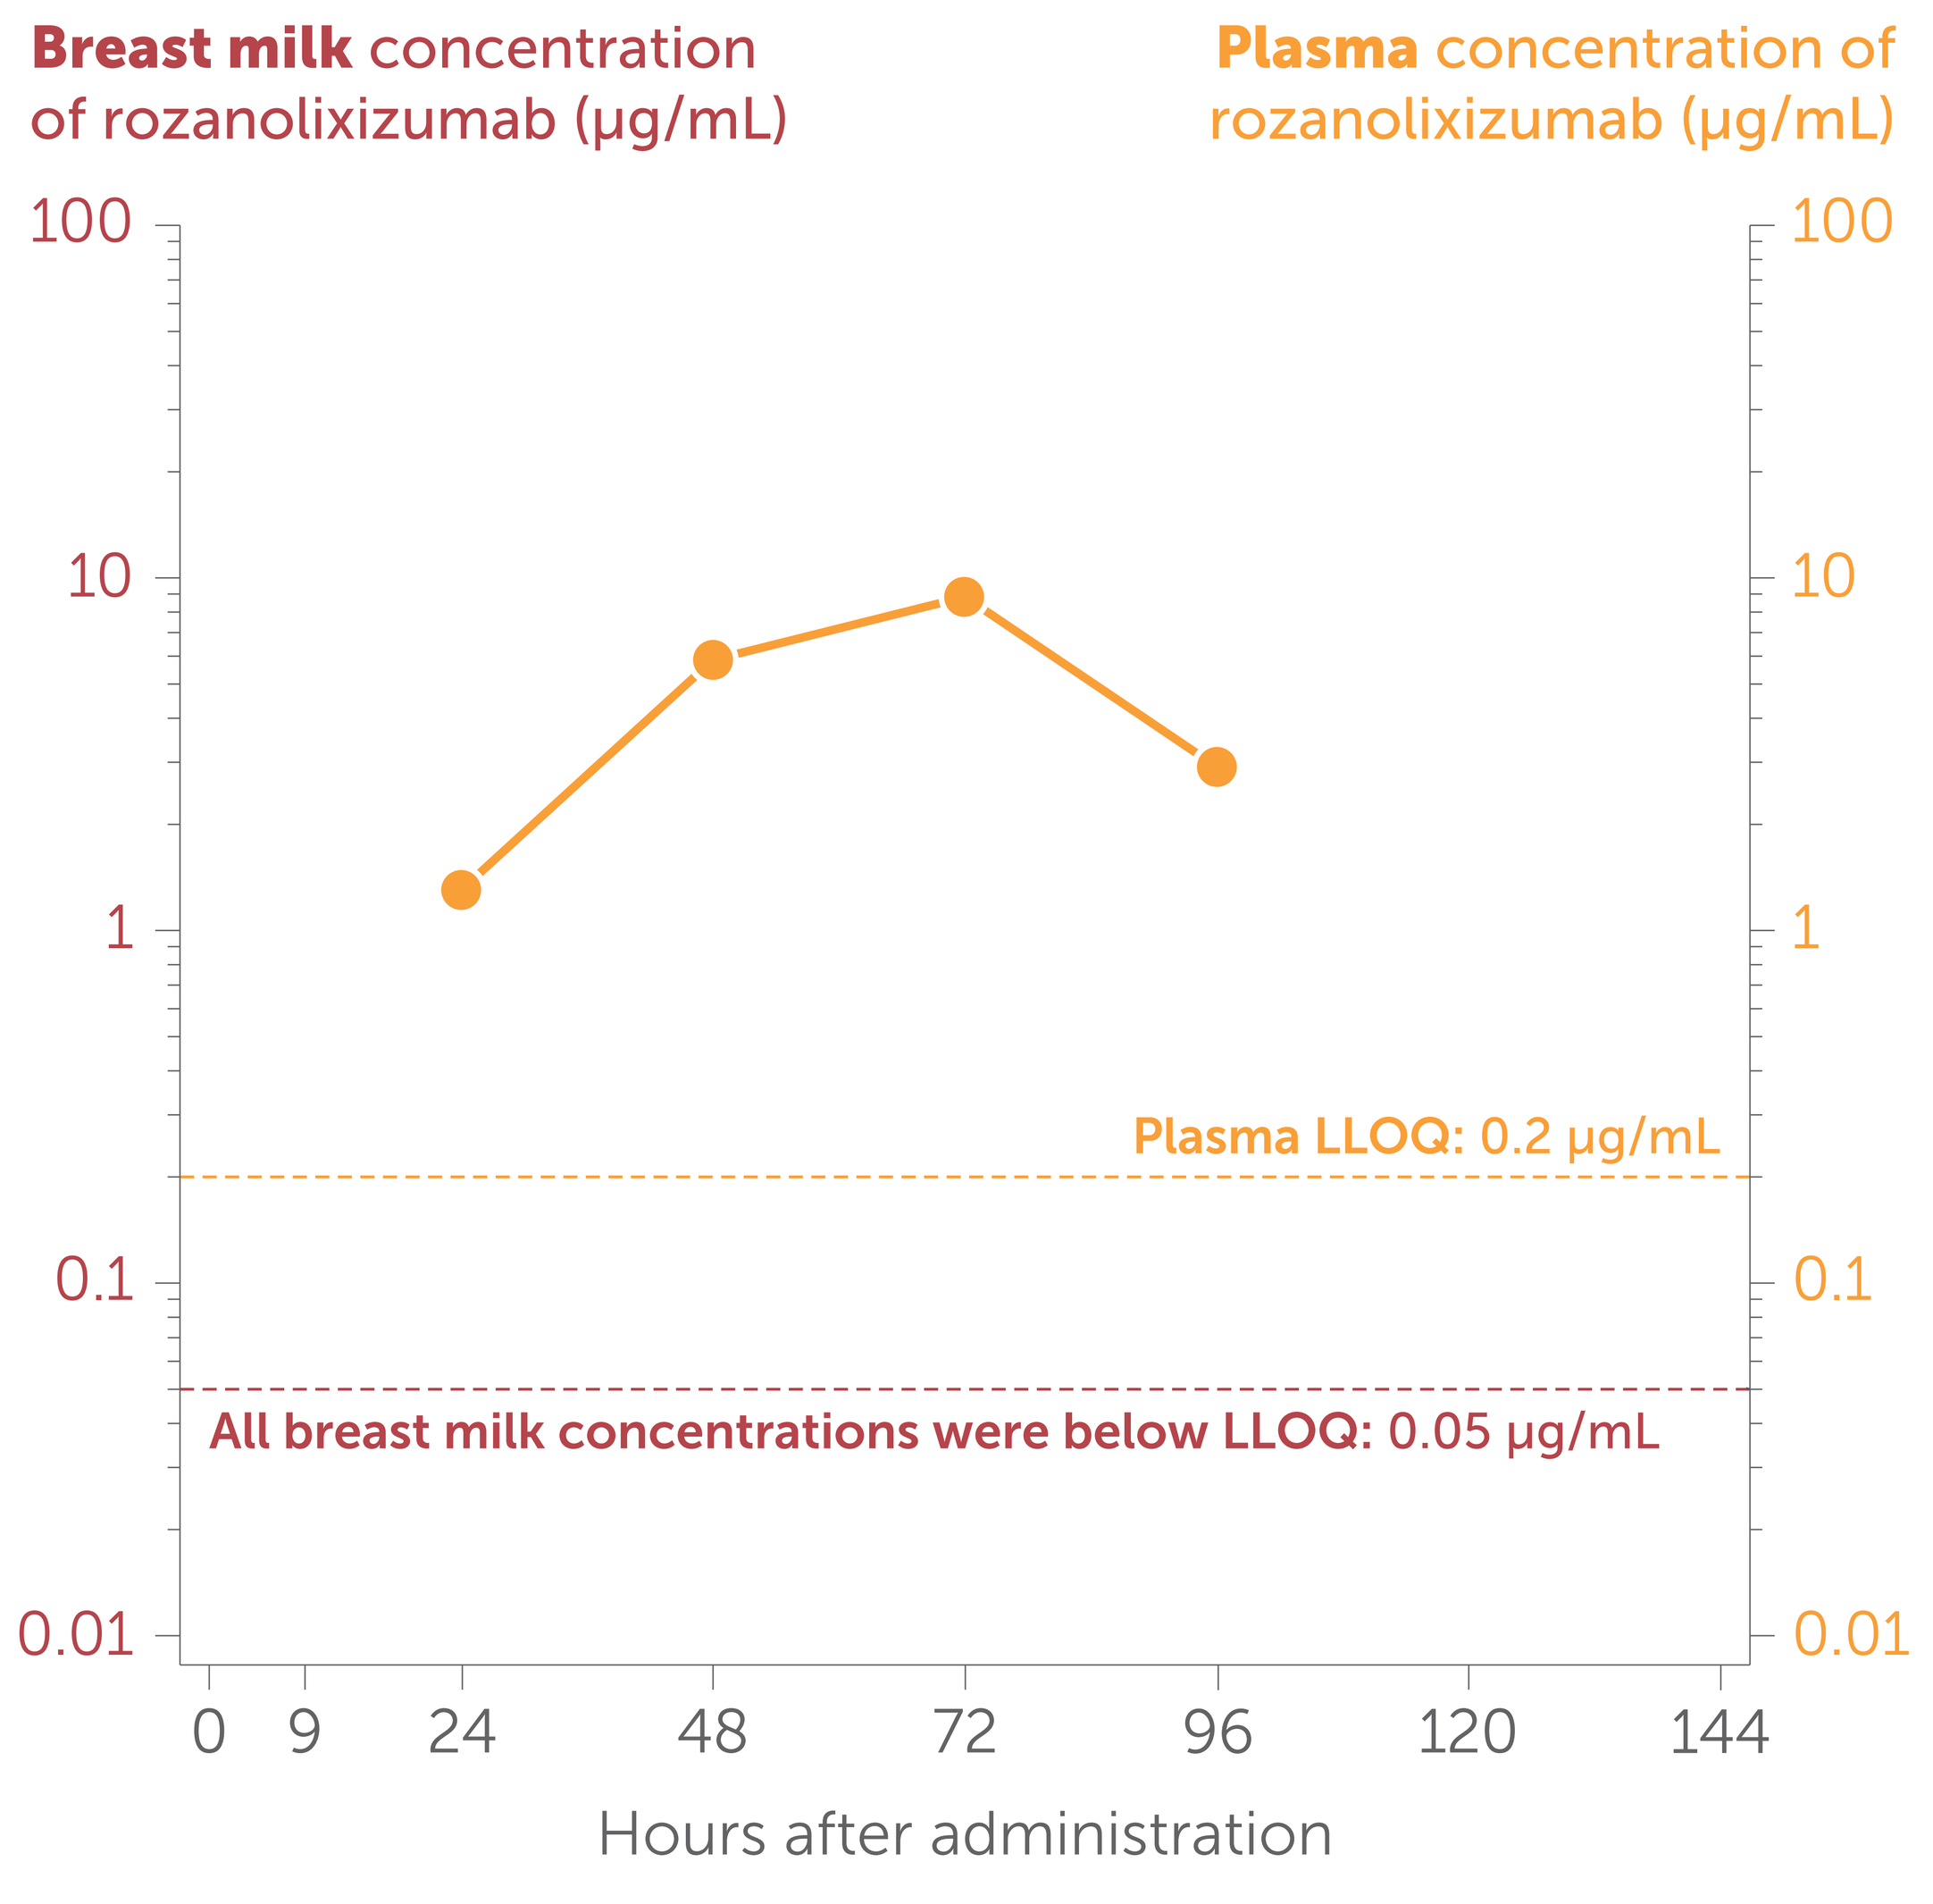
Breast milk concentration of rozanolixizumab (µg/mL)
Plasma concentration of rozanolixizumab (µg/mL)
100
10
1
0.1
0.01
Plasma LLOQ: 0.2 µg/mL
All breast milk concentrations were below LLOQ: 0.05 µg/mL
0
9
24
48
72
96
120
144
Hours after administration

PK set included participants from safety set who received a full dose of rozanolixizumab and provided ≥1 valid measurement of rozanolixizumab concentration in maternal plasma or breast milk.

Table 1 Plasma PK parameters in healthy, lactating women after a single dose of rozanolixizumab were consistent with previous healthy volunteer studies[4]

	Maternal plasma		
	$AUC_{(0-7d)}$, h•µg/mL N=15	C_{max}, µg/mL N=15	t_{max}, hours N=15
GeoMean (95% CI)	**540.7** (357.6–817.6)	**10.9** (7.8–15.3)	**NE**
GeoCV %	**86.4**	**67.4**	**NE**
Median (min–max)	**537.4** (113.0–1580.0)	**11.4** (3.0–29.1)	**71.9** (48.0–72.4)

PK set.

Table 2 There were no serious or severe TEAEs during the 8-week safety follow-up

	Rozanolixizumab, N=15 % (n)
Any TEAE	**73.3** (11)
Serious TEAEs	**0**
Discontinuation due to TEAEs	**0**
Rozanolixizumab-related TEAEs	**66.7** (10)
Severe TEAEs	**0**
ADEs	**13.3** (2)
TEAEs leading to death	**0**

Safety set included participants who received a full or partial dose of rozanolixizumab.

Abbreviations: ADE, adverse device event; $AUC_{(0-7d)}$, area under the concentration–time curve from time 0 to 7 days; BMI, body mass index; CI, confidence interval; C_{max}, maximum plasma concentration; D, day; FcRn, neonatal fragment crystallizable receptor; GeoCV, geometric coefficient of variation; GeoMean, geometric mean; gMG, generalized myasthenia gravis; IgG, immunoglobulin G; LLOQ, lower limit of quantification; NE, not estimable; PK, pharmacokinetic; SD, standard deviation; TEAE, treatment-emergent adverse event; t_{max}, time to maximum plasma concentration.

Acknowledgments: This study was funded by UCB. The authors acknowledge Annick Moon, PhD, for Ogilvy Health, London, UK, for editorial assistance, which was funded by UCB. The authors thank Veronica Porkess, PhD, of UCB, Slough, UK, for publication and editorial support. The authors thank the participants in addition to the investigators and their teams who contributed to this study.

Disclosures: Babak Boroojerdi, Marion Boehnlein, François Bonfitto, Andreea Lavrov and Jagdev Sidhu are employees and shareholders of UCB. Timothy Mutsvari is an employee of UCB.

References: 1. Smith B, et al. MAbs. 2018;10(7):1111–1130. 2. RYSTIGGO® US PI. www.accessdata.fda.gov/drugsatfda_docs/label/2025/761286s007lbl.pdf. Accessed August 2026. 3. Sciancalepore F, et al. Neuroepidemiology. 2025;59(5):579–592. 4. El Baghdady A, et al. Clin Pharmacol Drug Dev. 2025;14(1):26–35.

Please use this QR code to download a PDF of the poster.