Poster **183**

Repeated cycles of rozanolixizumab treatment in anti-muscle-specific tyrosine kinase antibody-positive generalized myasthenia gravis

Nate Cashion[1], Artur Drużdż[2], Ali A. Habib[3], Dale J. Lange[4], Hiroyuki Naito[5], Robert M. Pascuzzi[6], Sabrina Sacconi[7], Kimiaki Utsugisawa[8], Fiammetta Vanoli[9], John Vissing[10], Tuan Vu[11], Jiann-Horng Yeh[12–14], Joel Baldwin[15], Thaïs Tarancón[16], Vera Bril[17]*

[1]UCB, Phoenix, AZ, USA; [2]Department of Neurology, Municipal Hospital, Poznań, Poland; [3]MDA ALS & Neuromuscular Center, Department of Neurology, University of California, Irvine, Orange, CA, USA; [4]Lange Neurology, New York, NY, USA; [5]Department of Clinical Neuroscience and Therapeutics, Hiroshima University, Hiroshima, Japan; [6]Neurology Department, Indiana University School of Medicine, Indiana University Health, Indianapolis, IN, USA; [7]Université Côte d'Azur, Peripheral Nervous System & Muscle Department, Pasteur 2 Hospital, Centre Hospitalier Universitaire de Nice, Nice, France; [8]Department of Neurology, Hanamaki General Hospital, Hanamaki, Japan; [9]Department of Neuroimmunology and Neuromuscular Diseases, Fondazione IRCCS, Instituto Nazionale Neurologico Carlo Besta, Milan, Italy; [10]Copenhagen Neuromuscular Center, Department of Neurology, Rigshospitalet, University of Copenhagen, Copenhagen, Denmark; [11]Department of Neurology, University of South Florida Morsani College of Medicine, Tampa, FL, USA; [12]Department of Neurology, Shin Kong Wu Ho-Su Memorial Hospital, Taipei, Taiwan; [13]College of Medicine, Fu Jen Catholic University, New Taipei City, Taiwan; [14]Department of Neurology, Kaohsiung Medical University, Kaohsiung, Taiwan; [15]UCB, Slough, UK; [16]UCB, Madrid, Spain; [17]Ellen and Martin Prosserman Centre for Neuromuscular Diseases, Toronto General Hospital, University of Toronto, Toronto, Ontario, Canada

AANEM 2026, Orlando, FL, USA; September 29–October 2, 2026

Introduction

- Anti-MuSK Ab+ MG is a rare (5–8% of patients) and frequently severe subtype of MG[1,2]
- Rozanolixizumab, a humanized IgG mAb that targets and blocks FcRn with high affinity, is approved for the treatment of adults with anti-AChR Ab+ or anti-MuSK Ab+ gMG[3,4]
- In the Phase 3 MycarinG (NCT03971422) study, one 6-week cycle of rozanolixizumab (7 mg/kg or 10 mg/kg) led to clinically meaningful improvements in MG-specific outcomes in patients with gMG[5]
- Following MycarinG, patients could enroll in the OLE studies MG0004 (NCT04124965) then MG0007 (NCT04650854), or MG0007 directly[2]
- **This analysis evaluated the efficacy and safety of repeated rozanolixizumab cycles in patients with anti-MuSK Ab+ gMG**

Methods

- In MycarinG, patients were aged ≥18 years with MGFA Clinical Class II–IVa anti-AChR Ab+ or anti-MuSK Ab+ gMG, with an MG-ADL score ≥3 (for non-ocular symptoms) and a QMG score ≥11
- In MG0004, patients received once-weekly rozanolixizumab 7 mg/kg or 10 mg/kg for ≤52 weeks
- MG0007 comprised an initial 6-week rozanolixizumab 7 mg/kg or 10 mg/kg cycle, with further cycles administered upon symptom worsening at the investigator's discretion
- Final efficacy data were pooled across MycarinG, MG0004 (first 6 weeks) and MG0007 for patients with ≥2 symptom-driven rozanolixizumab treatment cycles
- Time to event data for the treatment-free interval were pooled across patients who had received rozanolixizumab, and had either initiated or were awaiting their first symptom-driven rozanolixizumab treatment cycle
- For MG-ADL and QMG, across Cycles 1–9, the following outcomes were assessed *post hoc* (for each cycle and as weighted means across 9 cycles):
 - Mean total scores at baseline and Day 43
 - CFB at Day 43
 - Responder rates at Day 43
- Final safety data were pooled across MycarinG and MG0007 for patients with ≥1 rozanolixizumab treatment cycle, with a treatment-free follow-up period; the incidence of TEAEs was assessed

Results

- Of 129 patients who received ≥2 symptom-driven rozanolixizumab cycles, 12 (9.3%) had anti-MuSK Ab+ gMG (**Table 1**)
- Consistent improvements in mean MG-ADL scores from baseline to Day 43 were observed in patients with anti-MuSK Ab+ gMG:
 - Across Cycles 1–9, mean CFB in MG-ADL scores ranged from −3.0 (n=8; Cycle 5) to −7.0 (n=12; Cycle 1) (**Figure 1a**)
 - Across Cycles 1–9, the overall weighted-mean MG-ADL score was 10.37 at baseline and 5.14 at Day 43, corresponding to a weighted-mean reduction of 5.23 points (**Figure 1a**)
- Similar improvements were observed in mean QMG scores:
 - Across Cycles 1–9, mean CFB in QMG scores ranged from −5.2 (n=5; Cycle 8) to −10.6 (n=8; Cycle 3) (**Figure 1b**)
 - Across Cycles 1–9, the overall weighted-mean QMG score was 16.76 at baseline and 9.23 at Day 43, corresponding to a weighted-mean reduction of 7.54 points (**Figure 1b**)
- Across Cycles 1–9, responder rates in patients with anti-MuSK Ab+ gMG ranged from 75.0% to 100% for MG-ADL (weighted mean: 87.3%) and 60.0% to 100% (weighted mean: 84.5%) for QMG (**Figure 2**)
- The median (Q1–Q3) treatment-free interval to the first symptom-driven cycle, estimated using Kaplan–Meier analysis, was 10.7 (5.1–29.9) weeks in patients with anti-MuSK Ab+ gMG
- Of 188 patients included in the safety pool, 18 (9.6%) had anti-MuSK Ab+ gMG
 - TEAEs were reported in 14/18 (77.8%) patients, of whom 11/14 (78.6%) reported TEAEs of mild or moderate intensity only (**Table 2**)

Summary and conclusions

- This pooled analysis of data from the MycarinG study and its OLEs assessed the efficacy and safety of repeated rozanolixizumab cycles in patients with anti-MuSK Ab+ gMG, a rare and frequently severe subtype of MG[1,2]
- Patients with anti-MuSK Ab+ gMG who received rozanolixizumab treatment consistently had improvements from baseline in MG-ADL and QMG scores across cycles
- Across Cycles 1–9, high average responder rates of 87.3% for MG-ADL and 84.5% for QMG were achieved in patients with anti-MuSK Ab+ gMG
- Rozanolixizumab was generally well tolerated, with an acceptable safety profile, in this subgroup of patients
- These data support the long-term use of rozanolixizumab as a treatment option for patients with anti-MuSK Ab+ gMG

Figure 1 Reductions in mean MG-ADL and QMG scores were observed in patients with anti-MuSK Ab+ gMG and maintained over repeated treatment cycles

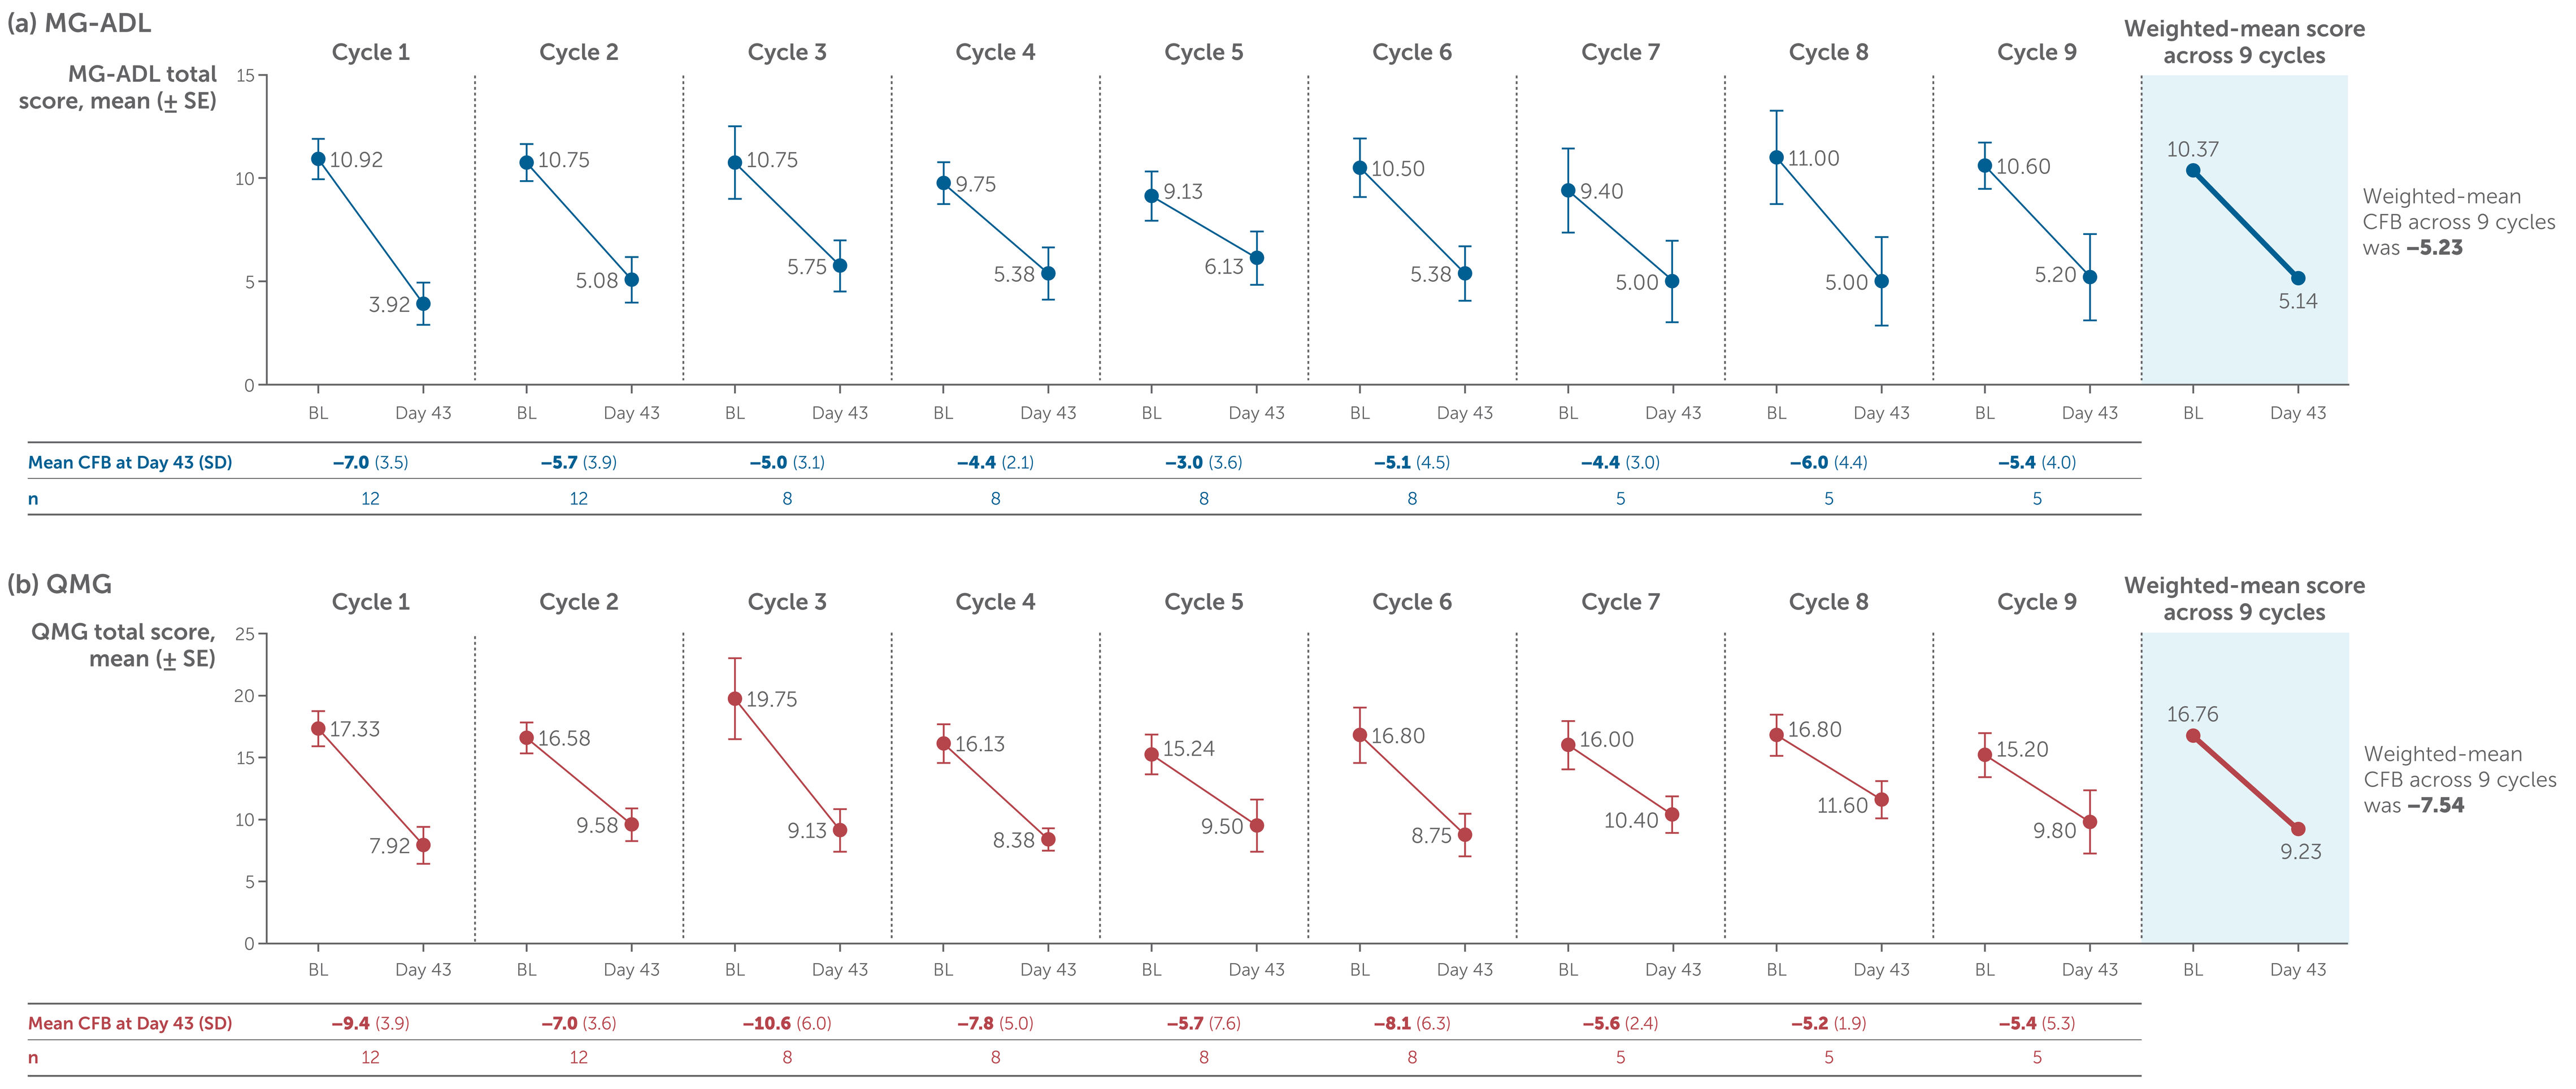
(a) MG-ADL
MG-ADL total score, mean (± SE)
Cycle 1: 10.92 → 3.92; Cycle 2: 10.75 → 5.08; Cycle 3: 10.75 → 5.75; Cycle 4: 9.75 → 5.38; Cycle 5: 9.13 → 6.13; Cycle 6: 10.50 → 5.38; Cycle 7: 9.40 → 5.00; Cycle 8: 11.00 → 5.00; Cycle 9: 10.60 → 5.20
Weighted-mean score across 9 cycles: 10.37 → 5.14
Weighted-mean CFB across 9 cycles was −5.23
BL Day 43
Mean CFB at Day 43 (SD): −7.0 (3.5); −5.7 (3.9); −5.0 (3.1); −4.4 (2.1); −3.0 (3.6); −5.1 (4.5); −4.4 (3.0); −6.0 (4.4); −5.4 (4.0)
n: 12; 12; 8; 8; 8; 8; 5; 5; 5
(b) QMG
QMG total score, mean (± SE)
Cycle 1: 17.33 → 7.92; Cycle 2: 16.58 → 9.58; Cycle 3: 19.75 → 9.13; Cycle 4: 16.13 → 8.38; Cycle 5: 15.24 → 9.50; Cycle 6: 16.80 → 8.75; Cycle 7: 16.00 → 10.40; Cycle 8: 16.80 → 11.60; Cycle 9: 15.20 → 9.80
Weighted-mean score across 9 cycles: 16.76 → 9.23
Weighted-mean CFB across 9 cycles was −7.54
Mean CFB at Day 43 (SD): −9.4 (3.9); −7.0 (3.6); −10.6 (6.0); −7.8 (5.0); −5.7 (7.6); −8.1 (6.3); −5.6 (2.4); −5.2 (1.9); −5.4 (5.3)
n: 12; 12; 8; 8; 8; 8; 5; 5; 5

Efficacy data presented for patients with ≥2 symptom-driven cycles. 'n' is the number of participants who have assessments recorded at both baseline and Day 43 of the cycle. Due to low patient numbers, results for Cycles 10–13 are not shown (Cycle 10, n=4; Cycles 11 and 12, n=3; Cycle 13, n=2). The small numerical discrepancy between the QMG weighted means is due to the rounding of data.

Figure 2 High average responder rates (across Cycles 1–9) of ≥84.5% for MG-ADL and QMG were observed in patients with anti-MuSK Ab+ gMG

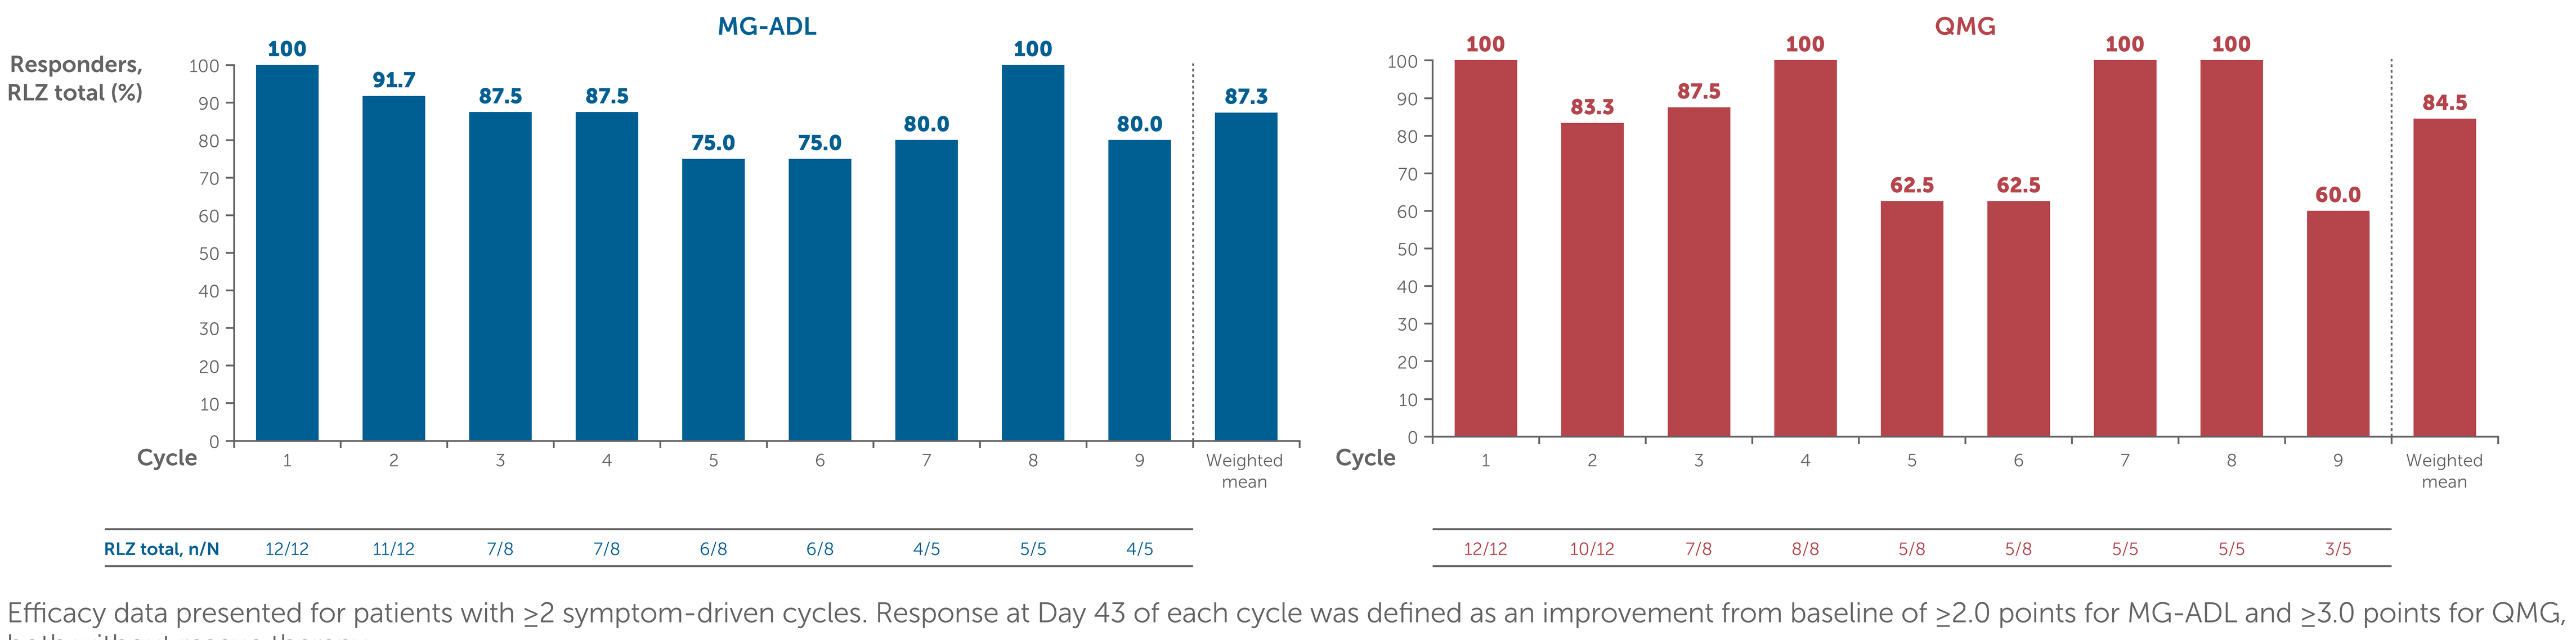
MG-ADL
Responders, RLZ total (%)
Cycle 1: 100; 2: 91.7; 3: 87.5; 4: 87.5; 5: 75.0; 6: 75.0; 7: 80.0; 8: 100; 9: 80.0; Weighted mean: 87.3
RLZ total, n/N: 12/12; 11/12; 7/8; 7/8; 6/8; 6/8; 4/5; 5/5; 4/5
QMG
Cycle 1: 100; 2: 83.3; 3: 87.5; 4: 100; 5: 62.5; 6: 62.5; 7: 100; 8: 100; 9: 60.0; Weighted mean: 84.5
12/12; 10/12; 7/8; 8/8; 5/8; 5/8; 5/5; 5/5; 3/5

Efficacy data presented for patients with ≥2 symptom-driven cycles. Response at Day 43 of each cycle was defined as an improvement from baseline of ≥2.0 points for MG-ADL and ≥3.0 points for QMG, both without rescue therapy.

Table 1 Baseline demographics and disease characteristics were broadly similar between the overall population and the anti-MuSK Ab+ subgroup

		RLZ total (N=129)	RLZ anti-MuSK Ab+ subgroup (N=12)
Age, years, mean (SD)		**50.9** (16.3)	**46.8** (10.8)
Sex, female, n (%)		**77** (59.7)	**9** (75.0)
Age at initial gMG diagnosis, years, mean (SD)		**43.1** (17.8)	**37.5** (10.1)
Duration of disease from diagnosis, years, mean (SD)		**8.1** (8.5)	**9.8** (8.0)
MGFA Clinical Class, n (%)	II	**52** (40.3)	**4** (33.3)
	III	**72** (55.8)	**7** (58.3)
	IV	**5** (3.9)	**1** (8.3)
MG-ADL score, mean (SD)		**8.7** (3.4)	**9.8** (3.5)
QMG score, mean (SD)		**16.0** (3.8)	**17.3** (4.5)
Thymectomy at baseline, n (%)		**55** (42.6)	**2** (16.7)
Baseline gMG medication, n (%)	Corticosteroids for systemic use	**80** (62.0)	**8** (66.7)
	Immunosuppressants	**66** (51.2)	**4** (33.3)
	Parasympathomimetics	**113** (87.6)	**7** (58.3)

Table 2 Rozanolixizumab was generally well tolerated in patients with anti-MuSK Ab+ gMG

	RLZ overall population (N=188), n (%)	RLZ anti-MuSK Ab+ subgroup (N=18), n (%)
Any TEAE	**175** (93.1)	**14** (77.8)
Serious TEAEs	**55** (29.3)	**3** (16.7)
Patient discontinuation due to TEAEs	**33** (17.6)	**3** (16.7)
Treatment-related TEAEs	**120** (63.8)	**11** (61.1)
Severe TEAEs	**62** (33.0)	**3** (16.7)
TEAEs leading to death†	**4** (2.1)	**0**

All cycles (encompassing cycles beyond Cycle 9). Safety data presented from data pooled from MycarinG and MG0007 for patients with ≥1 rozanolixizumab treatment cycle with a treatment-free follow-up period. 'n' is the number of patients reporting at least one TEAE within the category.
†All deaths were considered not related to rozanolixizumab by the investigator.

Abbreviations: Ab+, antibody positive; AChR, acetylcholine receptor; BL, baseline; CFB, change from baseline; FcRn, neonatal fragment crystallizable receptor; gMG, generalized myasthenia gravis; IgG4, immunoglobulin G subunit 4; mAb, monoclonal antibody; MG, myasthenia gravis; MG-ADL, Myasthenia Gravis Activities of Daily Living; MGFA, Myasthenia Gravis Foundation of America; MuSK, muscle-specific tyrosine kinase; OLE, open-label extension; QMG, Quantitative Myasthenia Gravis; RLZ, rozanolixizumab; SD, standard deviation; SE, standard error; TEAE, treatment-emergent adverse event.

Acknowledgments: *In memoriam of Prof. Vera Bril, whose invaluable contribution to this work is gratefully acknowledged. This study was funded by UCB. The authors acknowledge Qudsia Ehsan, BSc, of Ogilvy Health, London, UK, for editorial assistance, which was funded by UCB. The authors thank Veronica Porkess, PhD, CMPP, of UCB for publication and editorial support. The authors thank the patients and their caregivers, in addition to the investigators and their teams who contributed to this study.

Author disclosures: Nate Cashion is an employee of UCB. Artur Drużdż, Hiroyuki Naito and Sabrina Sacconi have nothing to disclose. Ali A. Habib has received research support and/or honoraria from Alexion/AstraZeneca Rare Disease, Amgen, Arcellx, argenx, Cabaletta Bio, Cartesian Therapeutics, COUR Pharmaceuticals, Genentech/Roche, Grifols, Immunovant, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Kyverna, Merck, MGNet, Nkarta, NMD Pharma, Novartis, Regeneron Pharmaceuticals and UCB. Dale J. Lange has received research support from Novartis Pharmaceuticals. He has received funding for advisory board participation from AbbVie, Inc., Alexion Pharmaceuticals and Calico Life Sciences. He has received funding for consultations from Summus Global. Robert M. Pascuzzi is Professor Emeritus of Neurology at Indiana University and receives compensation for his professional work from Indiana University Health. He has no financial relationship with any pharmaceutical company and receives no compensation from any pharmaceutical company (present or past). Robert M. Pascuzzi speaks at educational seminars on a broad variety of general neurology topics for primary care physicians through the organisation Medical Education Resources (an educational organisation with no links or ties to any pharmaceutical or healthcare business company). Therefore, Robert M. Pascuzzi has no conflicts of interest related to this research, manuscript, presentation or publication. Kimiaki Utsugisawa has served as a paid Consultant for argenx, Chugai Pharmaceutical, HanAll Biopharma, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Merck, Mitsubishi Tanabe Pharma, UCB and Viela Bio (now Amgen); he has received speaker honoraria from Alexion Pharmaceuticals, argenx, the Japan Blood Products Organization and UCB. Fiammetta Vanoli has received funding for travel, congress attendance, speaking and advisory boards from Alexion Pharmaceuticals, argenx, Biogen and UCB. John Vissing has been a Consultant on advisory boards for Amicus Therapeutics, Biogen, Edgewise Therapeutics, Fulcrum Therapeutics, Genethon, Horizon Therapeutics (now Amgen), Lupin, ML Biopharma, Novartis, Regeneron Pharmaceuticals, Roche, Sanofi Genzyme (now Sanofi), Sarepta Therapeutics and UCB. He has received research and travel support and/or speaker honoraria from Alexion Pharmaceuticals, argenx, Biogen, Edgewise Therapeutics, Fulcrum Therapeutics, Lupin, Sanofi Genzyme (now Sanofi) and UCB. He is a Principal Investigator in clinical trials for Alexion Pharmaceuticals, argenx, Atamyo Therapeutics, Genethon, Horizon Therapeutics (now Amgen), Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), ML Biopharma, Novartis, Regeneron Pharmaceuticals, Roche, Sanofi Genzyme (now Sanofi) and UCB. Tuan Vu is the USF Site Principal Investigator for MG clinical trials sponsored by Alexion/AstraZeneca Rare Disease, Amgen, argenx, Cartesian Therapeutics, COUR Pharmaceuticals, Dianthus Therapeutics, ImmunAbs, Immunovant, Johnson & Johnson, NMD Pharma, Regeneron Pharmaceuticals, UCB and Vor, and has served as a speaker for Alexion/AstraZeneca Rare Disease, Amgen, argenx and CSL Behring. He has performed consulting work for Alexion/AstraZeneca Rare Disease, argenx, Dianthus Therapeutics, NMD Pharma, Regeneron and Vor. Jiann-Horng Yeh serves as a Principal Investigator for clinical trials sponsored by Alexion/AstraZeneca Rare Disease, argenx, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Merck, Novartis, Regeneron Pharmaceuticals, UCB and Viela Bio (now Amgen). He has served as a Consultant on advisory boards for Alexion/AstraZeneca Rare Disease, CSL, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine) and Merck. Joel Baldwin is an employee of Cogent Skills Limited working for UCB. Thaïs Tarancón is an employee and shareholder of UCB. Vera Bril was a Consultant for Akcea, Alexion Pharmaceuticals, Alnylam, argenx, CSL, Grifols, Immunovant, Ionis, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Momenta (now Johnson & Johnson), Novo Nordisk, Octapharma, Pfizer, Powell Mansfield, Roche, Sanofi, Takeda Pharmaceuticals and UCB. She received research support from Akcea, Alexion Pharmaceuticals, argenx, CSL, Grifols, Immunovant, Ionis, Momenta (now Johnson & Johnson), Octapharma, Takeda Pharmaceuticals, UCB and Viela Bio (now Amgen).

References: 1. Vakrakou AG, et al. Front Immunol. 2023;14:1212757. 2. Bril V, et al. J Neuromuscul Dis. 2025;12(2):218–230. 3. Smith B, et al. MAbs. 2018;10(7):1111–1130. 4. RYSTIGGO® US PI. https://www.ucb-usa.com/RYSTIGGO-prescribing-information.pdf. Accessed August 2026. 5. Bril V, et al. Lancet Neurol. 2023;22(5):383–394.

Please use this QR code to download a PDF of the poster.

These data were previously presented at ICNMD 2026, Florence, Italy; July 7–11, 2026