AANEM Poster **241** | MGFA Scientific Session Poster **97**

Characterization of patients with generalized myasthenia gravis on rozanolixizumab treatment in European Managed Access Programmes

Chioma Ezenduka[1], Thaïs Tarancón[2], Paolo Emilio Alboini[3], Silvia Falso[4], Rita Frangiamore[5], Matteo Gastaldi[6], Melania Guida[7], Solange Kapetanovic Garcia[8,9], Nicasio Rini[10], Carmelo Rodolico[11], Elena Rossini[12], Sabrina Sacconi[13], Guilhem Solé[14], Karin Annoni[15], Kerina Bonar[16], Grégory Chollet[17], Cristina Lopez[18], Patrik Öhagen[19], Cassandra Slader[20]

[1]UCB, Raleigh, NC, USA; [2]UCB, Madrid, Spain; [3]Neurology Unit, Fondazione IRCCS Casa Sollievo della Sofferenza, San Giovanni Rotondo, Italy; [4]Department of Neuroscience, Catholic University of the Sacred Heart, Rome, Italy; [5]Neuroimmunology and Muscle Pathology Unit, IRCCS Carlo Besta Neurological Institute, Milan, Italy; [6]IRCCS Mondino Foundation, Neuroimmunology Research Unit, Pavia, Italy; [7]Neurology Unit, Department of Neurosciences, Azienda Ospedaliero Universitaria Pisana, Pisa, Italy; [8]ALS and Neuromuscular Unit, Hospital Universitario Basurto, Bilbao, Spain; [9]NAT-RD Research Group, IIS Biobizkaia, Barakaldo, Spain; [10]Department of Biomedicine, Neuroscience and Advanced Diagnostics (BiND), University of Palermo, Palermo, Italy; [11]Department of Clinical and Experimental Medicine (DIMED), University of Messina, Sicily, Italy; [12]Department of Neurology, Mental Health and Sensory Organs (NESMOS), Faculty of Medicine and Psychology, Sapienza University of Rome, Rome, Italy; [13]Université Côte d'Azur, Peripheral Nervous System & Muscle Department, Pasteur 2 Hospital, Centre Hospitalier Universitaire de Nice, Nice, France; [14]Centre de Référence des Maladies Neuromusculaires AOC CHU de Bordeaux, Bordeaux, France; [15]UCB, Milan, Italy; [16]UCB, Slough, UK; [17]UCB, Colombes, France; [18]UCB, Barcelona, Spain; [19]UCB, Uppsala, Sweden; [20]UCB, Brussels, Belgium

AANEM & MGFA Scientific Session 2026, Orlando, FL, USA; September 29–October 2, 2026

Introduction

- gMG is a rare, chronic, autoimmune disease, characterized by fluctuating muscle weakness[1]
- In the Phase 3 MycarinG study (NCT03971422), rozanolixizumab – a neonatal Fc receptor blocker – demonstrated improvement across multiple MG-specific outcomes in patients with gMG, with this improvement maintained over repeated 6-week treatment cycles during the OLE studies MG0004 (NCT04124965) and MG0007 (NCT04650854)[2,3]
- MAPs enable treatments to be administered before they are authorized for patients with a severe/debilitating/life-threatening condition, where available treatments are deemed inadequate by patients and physicians and there is no available clinical trial
- At the time of this analysis, data were available for France, Italy and Spain (**Figure 1**)
- **The objective of this study was to describe patient characteristics and rozanolixizumab treatment outcomes in MAPs from three countries: France, Italy and Spain**

Methods

- This was an observational, retrospective, longitudinal cohort study of adults with gMG who initiated rozanolixizumab treatment in MAPs between November 2022 and May 2025, and who consented for their data to be collected in the real-world database (**Figure 1**)
- All analyses were descriptive

Results

- 21 patients from France and 44 from Italy were included
- Data were available for <5 patients from Spain, who had a mean (SD) age of 61.5 (16.5) years; 50.0% were female
 - Given the small number of patients from Spain, further results are not presented
- Patients had a mean (SD) baseline MG-ADL score of 9.0 (6.7) in France and 7.4 (2.6) in Italy (**Table 1**)
- In Italy, 30/38 (78.9%) patients had an MG exacerbation in the prior 2 years (**Figure 2**)
- In Italy, 44 (100%) patients received corticosteroids before entering the MAP, 41 (93.2%) received immunosuppressants, 25 (56.8%) received IVIg, 38 (86.4%) received PLEX or plasmapheresis, 31 (70.5%) received cholinesterase inhibitors, 13 (29.5%) received rituximab, one (2.3%) received tacrolimus and two (4.5%) received efgartigimod
 - These data were not available for France
- The majority of patients in Italy received concomitant long-term immunotherapy; fewer patients in France were reported to be on long-term immunotherapy (**Table 2**)
- Mean (SD) treatment duration was 6.8 (5.0) months in France and 9.6 (5.5) months in Italy
- Patients in France received a mean (range) of 2.6 (1–7) treatment cycles during their first year of treatment; in Italy, 2.9 (1–6) cycles were received
- Treatment-free intervals varied between patients in both France and Italy (**Figure 3**)
- Patient numbers for treatment outcomes data were too low to draw any meaningful conclusions, so these data are not presented*
- In Italy, 20/37 (54.1%) patients receiving corticosteroids reduced their corticosteroid dose during follow-up and 2/37 (5.4%) increased their dose; the 17 patients who were receiving immunosuppressants remained on the same dose
 - These data were not available for patients in France
- One (4.8%) patient in France required IVIg rescue medication during follow-up, with none requiring this in Italy
- 4/21 (19.0%) patients discontinued rozanolixizumab treatment in France and 4/20 (20.0%) discontinued in Italy
- 7/22 (31.8%) patients had a TEAE in France, with 19/71 (26.8%) patients having a TEAE in Italy (**Table 3**)
 - One (4.8%) death occurred in France, attributed to inhalation due to epistaxis in a high-risk cardiovascular setting, unrelated to rozanolixizumab

Limitations

- The primary purpose of a MAP is to provide treatments for patients who are otherwise unable to access them, not data collection
- Patients in the MAP could optionally provide consent for additional data collection. It was not mandatory for clinicians to complete additional data entry. As a result, data are limited beyond the baseline assessment
- The small sample size meant that only descriptive analyses were conducted
- The time on treatment during the MAP depended on when the patient joined the program and the timing of national reimbursement in the participating country. In some instances, this was a limited duration

Summary and conclusions

This observational, retrospective, longitudinal cohort study assessed adults with gMG who initiated rozanolixizumab treatment in MAPs in France, Italy and Spain

Previous treatments before entering the MAP included long-term immunotherapy, rescue treatments and cholinesterase inhibitors, with more patients previously on corticosteroids in Italy than in France

Over half of patients in Italy reduced their corticosteroid dose during follow-up after receiving rozanolixizumab

These data characterize patients receiving rozanolixizumab in a MAP in France and Italy who otherwise would not have been able to access the treatment

Figure 1 Rozanolixizumab has been provided in MAPs across the world

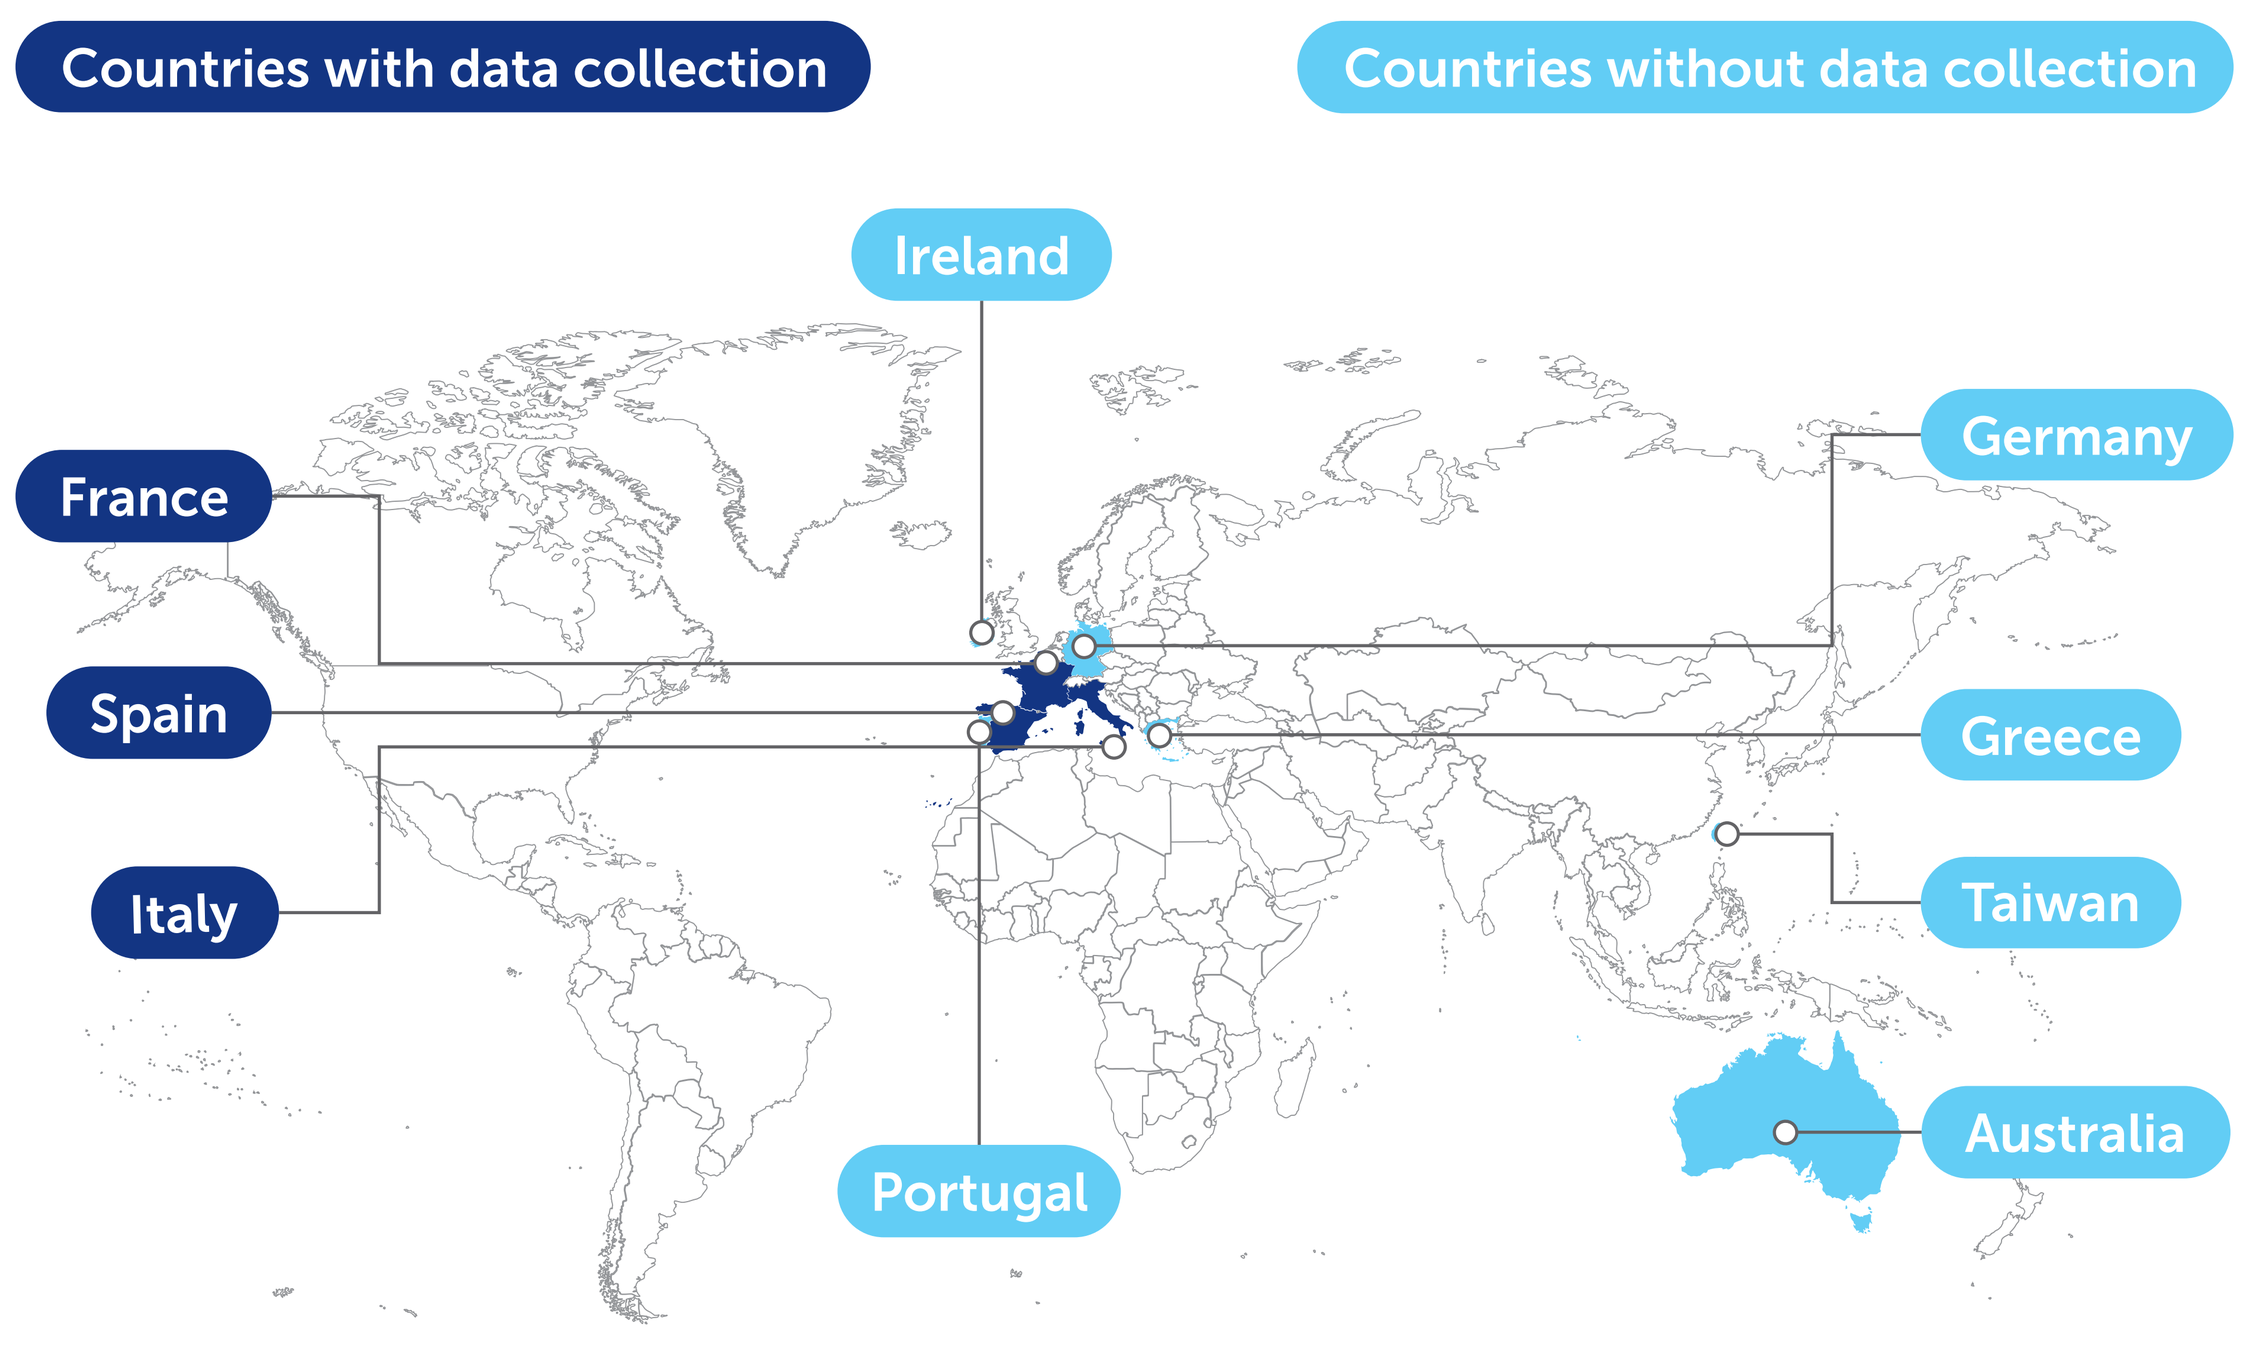
Countries with data collection
Countries without data collection
Ireland
France
Germany
Spain
Greece
Italy
Taiwan
Portugal
Australia

Figure 2 More than three-quarters of patients in Italy had an MG exacerbation in the prior 2 years

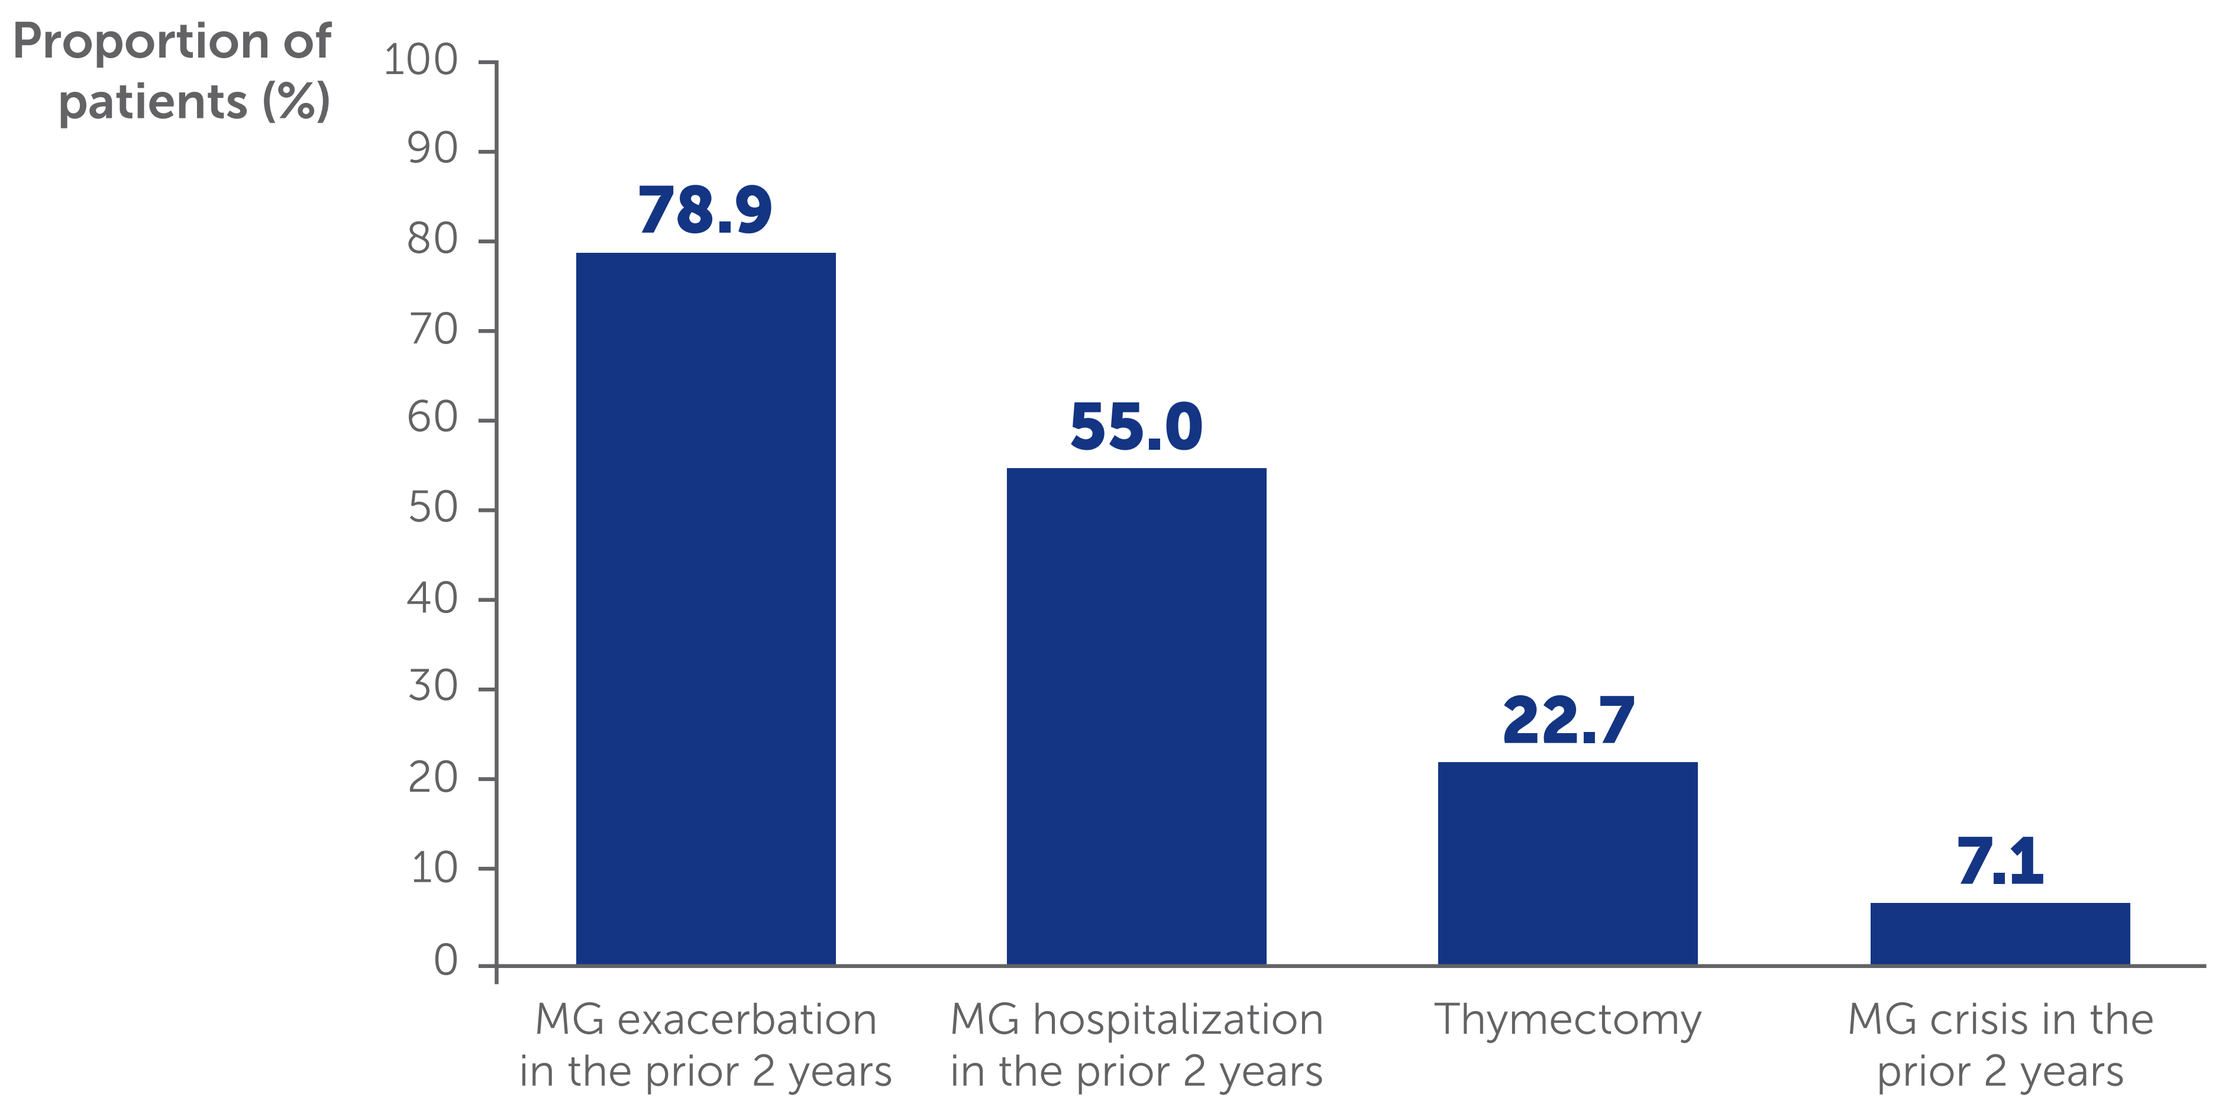
Proportion of patients (%)
100
90
80
70
60
50
40
30
20
10
0
78.9
55.0
22.7
7.1
MG exacerbation in the prior 2 years
MG hospitalization in the prior 2 years
Thymectomy
MG crisis in the prior 2 years

These data were not available for France.

Figure 3 Treatment-free intervals varied between patients in (a) France and (b) Italy

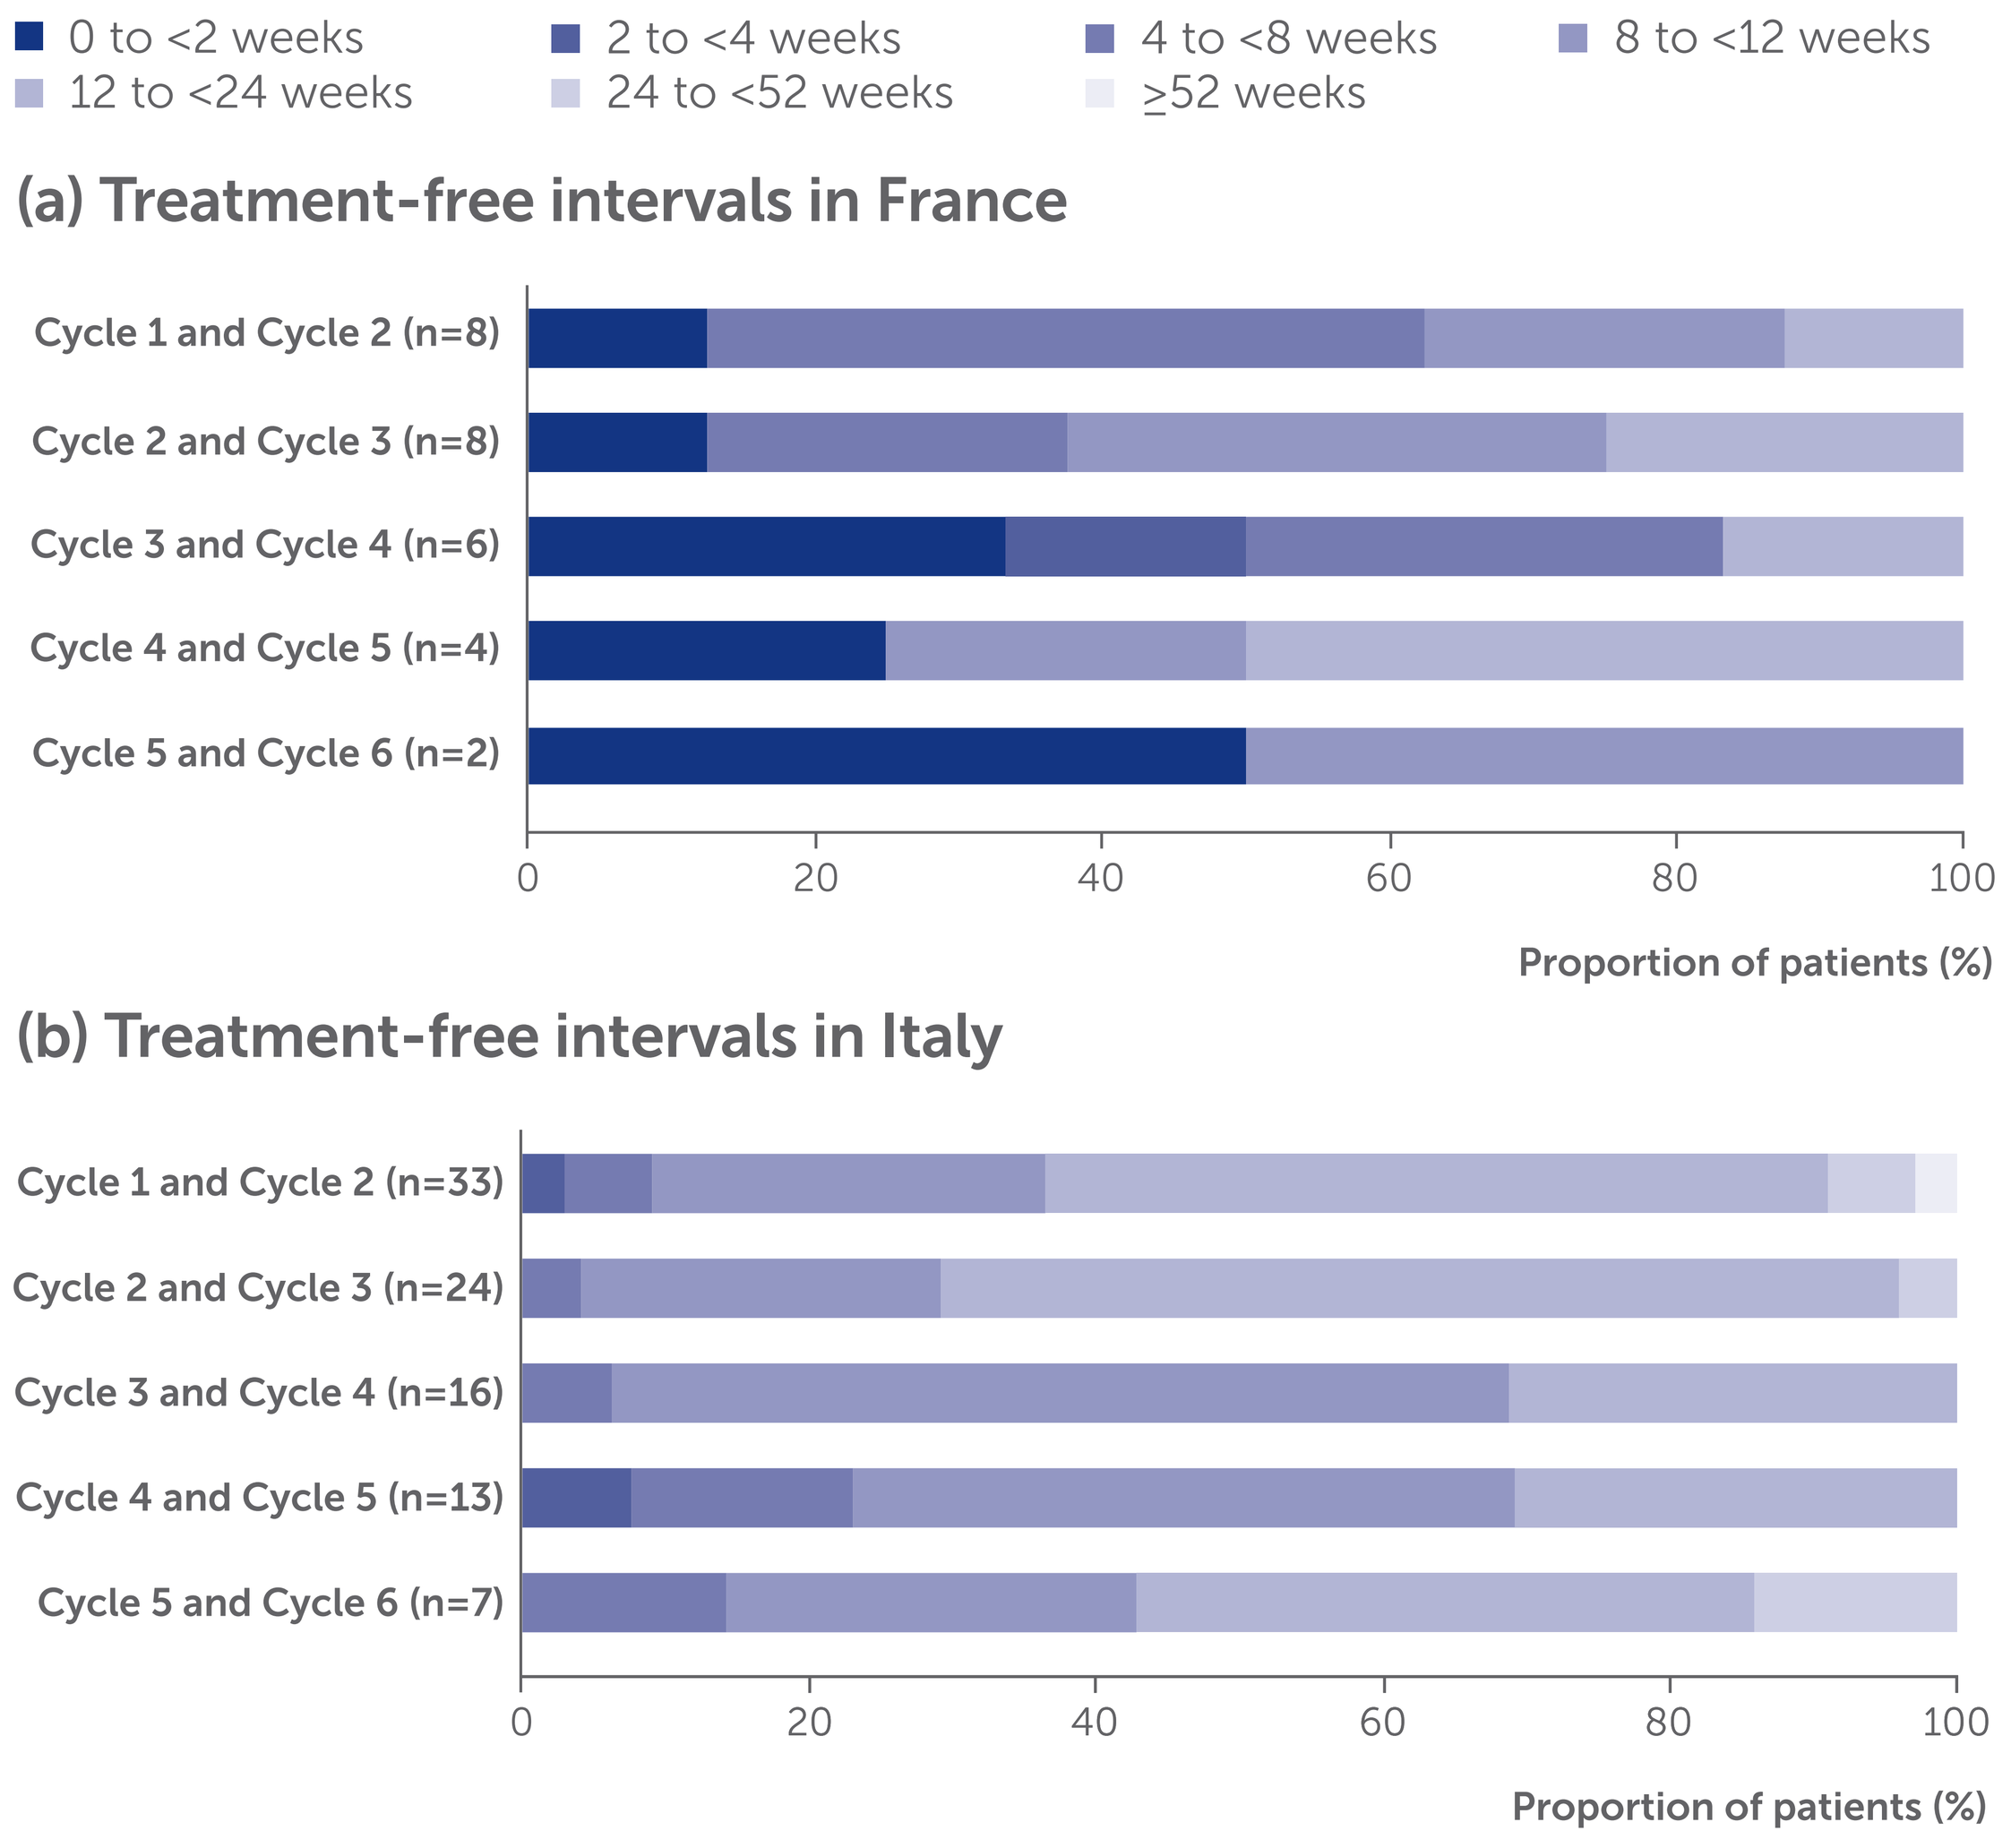
0 to <2 weeks
2 to <4 weeks
4 to <8 weeks
8 to <12 weeks
12 to <24 weeks
24 to <52 weeks
≥52 weeks
(a) Treatment-free intervals in France
Cycle 1 and Cycle 2 (n=8)
Cycle 2 and Cycle 3 (n=8)
Cycle 3 and Cycle 4 (n=6)
Cycle 4 and Cycle 5 (n=4)
Cycle 5 and Cycle 6 (n=2)
0
20
40
60
80
100
Proportion of patients (%)
(b) Treatment-free intervals in Italy
Cycle 1 and Cycle 2 (n=33)
Cycle 2 and Cycle 3 (n=24)
Cycle 3 and Cycle 4 (n=16)
Cycle 4 and Cycle 5 (n=13)
Cycle 5 and Cycle 6 (n=7)
0
20
40
60
80
100
Proportion of patients (%)

Table 1 Patients were indicative of a population with moderate-to-severe gMG

		France (N=21)	Italy (N=44)
	Age, years, mean (SD)	**64.8** (18.2)	**54.0** (16.6)
	Male, n (%)	**6** (28.6)	**11** (25.0)
	Weight, kg, mean (SD)	**80.5** (23.1)	**70.4** (16.2)
	MG-ADL score, mean (SD) [n]	**9.0** (6.7) [13]	**7.4** (2.6) [41]
MGFA Clinical Class, n (%)	II	–	**25** (56.8)
	III	–	**19** (43.2)
	Garches score, mean (SD) [n]	**62.4** (30.6) [12]	–
Time since initial MG diagnosis	Mean (SD), years	–	**11.9** (12.1)
	<5 years, n (%)	–	**16** (36.4)
	≥5 years, n (%)	–	**28** (63.6)

– Indicates data not collected or missing.

Table 2 More patients were on concomitant corticosteroids in Italy than in France

		France (N=21)*	Italy (N=44)†
Long-term immunotherapy, n (%)	Corticosteroids	**7** (33.3)	**34** (77.3)
	Azathioprine	**4** (19.0)	**8** (18.2)
	MMF	**3** (14.3)	**4** (9.1)
	Ciclosporin	**0**	**2** (4.5)
	MTX	**0**	**1** (2.3)
	Total	**17** (81.0)	**–**
Cholinesterase inhibitors, n (%)	Pyridostigmine	**0**	**6** (13.6)
	Ambenonium chloride	**1** (4.8)	**0**
Rescue treatments, n (%)	IVIg	**1** (4.8)	**0**
	PLEX	**0**	**0**
	Plasmapheresis	**0**	**0**
Rozanolixizumab, n/N (%)	280 mg	**1/14** (7.1)	**3/44** (6.8)
	420 mg	**5/14** (35.7)	**18/44** (40.9)
	560 mg	**7/14** (50.0)	**21/44** (47.7)
	840 mg	**1/14** (7.1)	**2/44** (4.5)

– Indicates data not collected or missing.
*Data for long-term immunotherapy, cholinesterase inhibitors and rescue treatments were collected at baseline or during follow-up; data for rozanolixizumab dose were collected at baseline.
†Data for long-term immunotherapy, cholinesterase inhibitors and rozanolixizumab dose were collected at baseline; data for rescue treatments were collected during follow-up.

Table 3 The safety profile was consistent with the known safety profile of rozanolixizumab

	France (N=22)	Italy (N=71)
Any TEAE, n (%)*	**7** (31.8)	**19** (26.8)
Influenza	**0**	**4** (5.6)
Vomiting	**0**	**4** (5.6)
Diarrhoea	**3** (13.6)	**3** (4.2)
Headache	**1** (4.5)	**3** (4.2)
MG	**2** (9.1)	**3** (4.2)
Nausea	**0**	**3** (4.2)
Pyrexia	**0**	**3** (4.2)
Abdominal pain	**2** (9.1)	**1** (1.4)
Cardiac failure	**2** (9.1)	**0**
Fall	**2** (9.1)	**1** (1.4)
Asthenia	**0**	**2** (2.8)
COVID-19	**1** (4.5)	**2** (2.8)
Dyspnea	**0**	**2** (2.8)
Death, n/N (%)	**1/21** (4.8)†	**0**

Reporting on safety information was required for all patients in the MAP in addition to those who consented to RWE data collection.
*Individual TEAEs listed are those occurring in >1 patient in either France or Italy.
†Unrelated to rozanolixizumab.

Footnotes: *MG-ADL: In France, 14/18 timepoints had data for ≤4 patients (out of 21); in Italy, 20/30 timepoints had data for ≤8 patients (out of 44); Garches: In France, 14/18 timepoints had data for ≤4 patients (out of 21); no data were available for Italy; MG-QoL15r: In Italy, 20/30 timepoints had data for ≤4 patients (out of 44); no data were available for France.

Abbreviations: Fc, fragment crystallizable; gMG, generalized myasthenia gravis; IVIg, intravenous immunoglobulin; MAP, Managed Access Programme; MG, myasthenia gravis; MG-ADL, Myasthenia Gravis Activities of Daily Living; MGFA, Myasthenia Gravis Foundation of America; MG-QoL15r, Myasthenia Gravis Quality of Life 15-item revised; MMF, mycophenolate mofetil; MTX, methotrexate; OLE, open-label extension; PLEX, plasma exchange; RWE, real-world evidence; SD, standard deviation; TEAE, treatment-emergent adverse event.

Acknowledgments: This study was funded by UCB. The authors acknowledge Luke Worley of Ogilvy Health, London, UK, for editorial assistance, which was funded by UCB. The authors thank Veronica Porkess, PhD, of UCB for publication and editorial support. The authors thank the patients and their caregivers, in addition to the investigators and their teams who contributed to this study.

Author disclosures: Chioma Ezenduka, Thaïs Tarancón and Kerina Bonar are employees and shareholders of UCB. Paolo Emilio Alboini has received funding from Alexion Pharmaceuticals, Amgen, argenx and UCB. Silvia Falso has received speaker honoraria from Alexion Pharmaceuticals, argenx and UCB. Rita Frangiamore has received funding for travel, consulting, meeting attendance and advisory board participation from Alexion Pharmaceuticals, argenx and UCB. Matteo Gastaldi has received contributions for advisory board participation and speaker honoraria from Alexion Pharmaceuticals, Roche and UCB. Melania Guida has received funding for travel, speaker honoraria, meeting attendance and advisory board participation from Alexion Pharmaceuticals, Amgen, argenx, Johnson & Johnson and UCB. Solange Kapetanovic Garcia has received contributions for meeting attendance and advisory board participation and speaker honoraria from Alexion Pharmaceuticals, argenx and UCB. Nicasio Rini has received funding for travel and meeting attendance from Alexion Pharmaceuticals, argenx, Roche and UCB. Carmelo Rodolico has received funding for meeting attendance and advisory board participation from Alexion Pharmaceuticals, Amgen, argenx, Johnson & Johnson and UCB. Elena Rossini has received speaker honoraria from UCB. Sabrina Sacconi and Guilhem Solé have nothing to disclose. Karin Annoni is a former employee of UCB. Grégory Chollet and Cristina Lopez are employees of UCB. Patrik Öhagen is a former contractor for UCB. Cassandra Slader is an employee and shareholder of UCB, and is a shareholder of Novartis.

References: 1. Gilhus NE, et al. Nat Rev Dis Primers. 2019;5:30. 2. Bril V, et al. Lancet Neurol. 2023;22:383–394. 3. Bril V, et al. J Neuromuscul Dis. 2025;12:218–230.

Please use this QR code to download a PDF of the poster.

These data were previously presented at ICNMD 2026, Florence, Italy; July 7–11, 2026