Poster **91**

Switching to rozanolixizumab from efgartigimod: Real-world outcomes in patients with myasthenia gravis in the United States

Chioma Ezenduka[1], Kristina Bardenheuer[2], Thaïs Tarancón[3], Rosa Falotico[4], Jianing Yi[5], Minjee Park[6], Diogo Pata[7]

[1]UCB, Raleigh, NC, USA; [2]UCB, Monheim, Germany; [3]UCB, Madrid, Spain; [4]Alira Health, Milan, Italy; [5]Alira Health, Framingham, MA, USA; [6]Alira Health, Basel, Switzerland; [7]UCB, Brussels, Belgium

MGFA Scientific Session at AANEM 2026, Orlando, FL, USA; September 29, 2026

Introduction

- Rozanolixizumab is a humanized IgG4 mAb that targets and blocks FcRn with high affinity, approved for treating AChR and MuSK Ab+ gMG[1,2]
- The molecular characteristics of rozanolixizumab and efgartigimod (another FcRn inhibitor) differ, which could warrant patients with gMG switching from efgartigimod to rozanolixizumab
 - An *in vitro* modeling study showed that rozanolixizumab achieved high intracellular drug levels more quickly than efgartigimod due to its higher affinity and pH-independent binding to FcRn, with higher IgG reductions[2]
 - Based on Phase 3 clinical trial data, AChR autoantibody reduction tracks more closely with total IgG clearance with rozanolixizumab than efgartigimod[3]
- **The aim of this study was to assess real-world demographics, clinical characteristics, treatment patterns and HCRU among patients with MG initiating rozanolixizumab who switched from efgartigimod**

Methods

- This was a retrospective non-interventional cohort study of adult patients (≥18 years) with MG treated in the US, using de-identified, patient-level data from medical and pharmacy claims from the Komodo Healthcare Map®
- Data were included from patients who received ≥1 infusion of rozanolixizumab between July 21, 2023, and February 28, 2025, after receiving ≥1 infusion of efgartigimod in the previous 12 months and had continuous plan coverage for 12 months pre- and post-index date (first recorded date for receiving rozanolixizumab)
- The baseline period was 12 months before the index date and the follow-up period was 12 months after and including the index date

Results

Patient characteristics

- In total, 70 patients switched from efgartigimod to rozanolixizumab and were included in the analysis
- During baseline, patients had a mean (SD) age of 63.6 (15.6) years and 50% (n=35/70) were female
 - The largest proportion of the cohort (42.9%; n=30/70) was from the South, and more than half (55.7%; n=39/70) were covered by Medicare
 - Most patients were between the ages of 51 and 80 years and the most common comorbidities were hypertension, dyslipidemia and infections (**Figure 1**)

Treatment patterns

- Patients experienced a reduction in annualized treatment cycles (mean 4.8 vs 3.0) and increased duration of treatment-free intervals (mean 5.6 vs 6.8) when switching from efgartigimod to rozanolixizumab (**Table 1**)

Oral corticosteroid and NSIST use

- In total, 61.4% (n=43/70) of patients received oral corticosteroids during baseline and 57.1% (n=40/70) received them during follow-up
 - The mean (SD) daily dose was 7.4 mg (6.5; n=39) during baseline and 9.0 mg (12.1; n=34) during follow-up
 - Of patients receiving steroids during baseline, 29.7% (n=11/37) did not require steroids after initiating rozanolixizumab (**Figure 2**)
- In total, 51.4% (n=36/70) of patients received NSISTs during baseline; of these, 50.0% (n=18/36) decreased the number of NSISTs used during follow-up, 41.7% (n=15/36) exhibited no change in their NSIST use and 8.3% (n=3/36) increased the number of NSISTs used
- Most HCRU events were less frequent during follow-up than baseline (**Figure 3**)

Summary and conclusions

This retrospective, non-interventional cohort study assessed real-world demographics, clinical characteristics, treatment patterns and HCRU among patients with MG in the US who switched from efgartigimod to rozanolixizumab

In the 12 months following initiation of rozanolixizumab, the mean annualized number of cycles decreased and the mean treatment-free interval increased versus baseline (efgartigimod)

Most patients either discontinued, decreased or maintained their corticosteroid dose during follow-up, after initiating rozanolixizumab

In general, HCRU decreased during follow-up, after initiating rozanolixizumab

These real-world findings demonstrate that switching from efgartigimod to rozanolixizumab is a clinically viable treatment strategy and was associated with improved treatment outcomes and reduced healthcare burden, which could occur due to differences in molecular characteristics

Figure 1 Most patients were between 51 and 80 years of age at initiation of rozanolixizumab (a) and the most common comorbidities during baseline were hypertension, dyslipidemia and infections (b)

(a) Age at initiation of RLZ (N=70)

(b) Comorbidities (N=70)

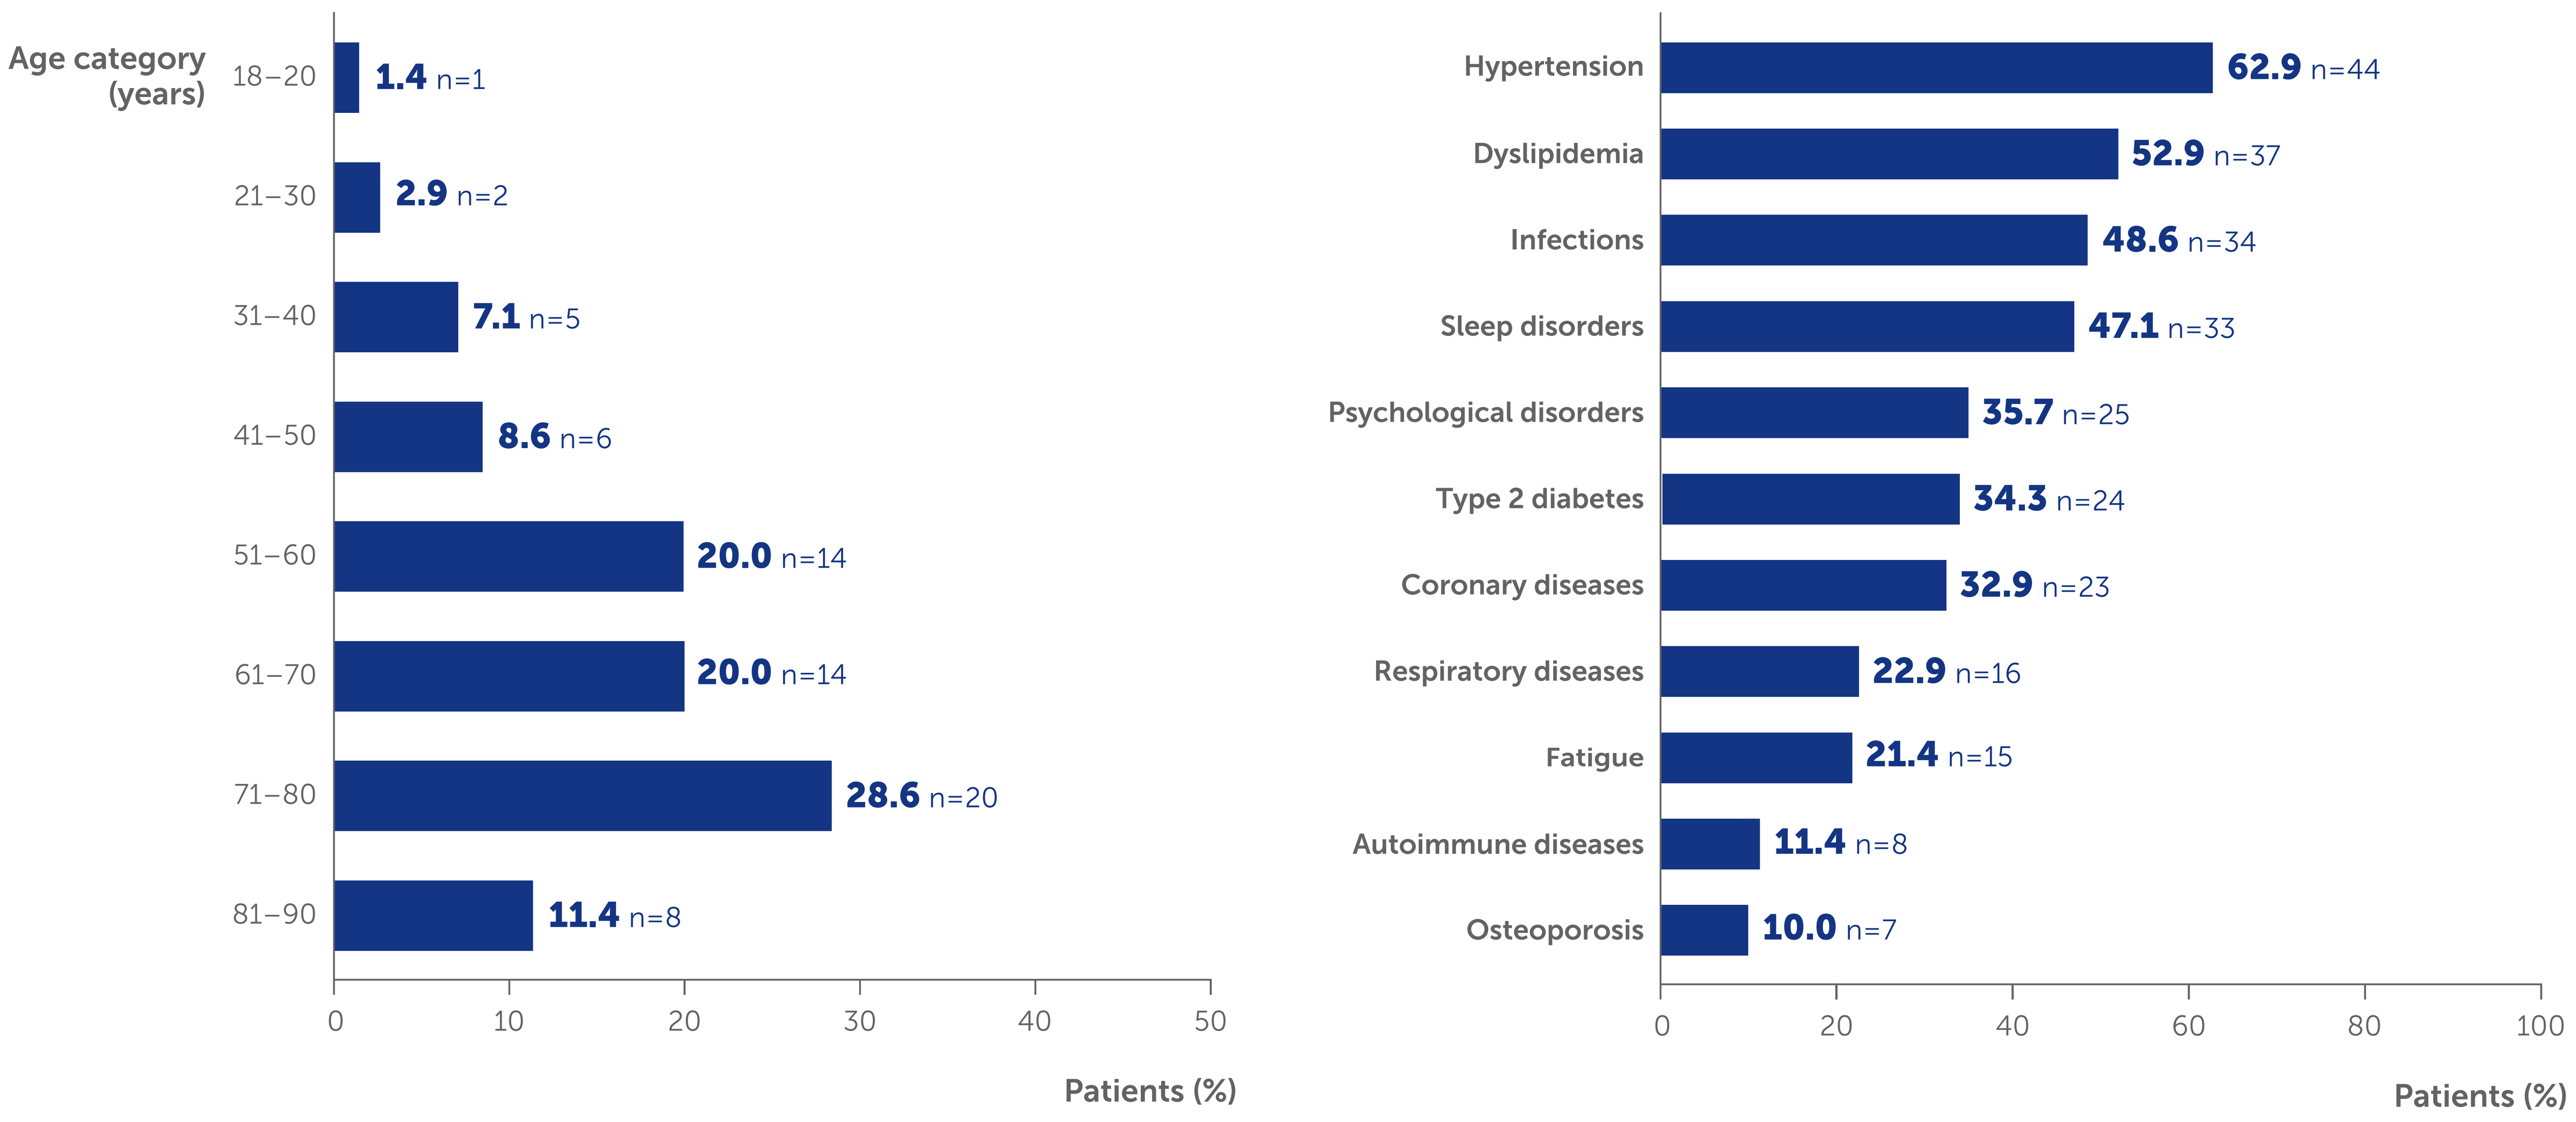
Age category (years)
18–20 1.4 n=1
21–30 2.9 n=2
31–40 7.1 n=5
41–50 8.6 n=6
51–60 20.0 n=14
61–70 20.0 n=14
71–80 28.6 n=20
81–90 11.4 n=8
0 10 20 30 40 50
Patients (%)
Hypertension 62.9 n=44
Dyslipidemia 52.9 n=37
Infections 48.6 n=34
Sleep disorders 47.1 n=33
Psychological disorders 35.7 n=25
Type 2 diabetes 34.3 n=24
Coronary diseases 32.9 n=23
Respiratory diseases 22.9 n=16
Fatigue 21.4 n=15
Autoimmune diseases 11.4 n=8
Osteoporosis 10.0 n=7
0 20 40 60 80 100
Patients (%)

Table 1 Observed treatment patterns indicated a reduced treatment burden when patients switched from efgartigimod to rozanolixizumab

	Efgartigimod	Rozanolixizumab
Cycles initiated, mean (SD)	**3.5** (2.0)	**2.5** (1.6)
Treatment duration, weeks (SD)	**38.3** (13.6)	**47.2** (11.5)
Annualized cycles, mean (SD)	**4.8** (2.2)	**3.0** (2.2)
Treatment-free interval, weeks, (SD)	**5.6** (2.7)	**6.8** (4.1)

Per-patient annualized cycles=(number of cycles initiated in a year/treatment duration days)*365.

Figure 2 Most patients either discontinued or decreased their corticosteroid dose during follow-up (after initiating rozanolixizumab)

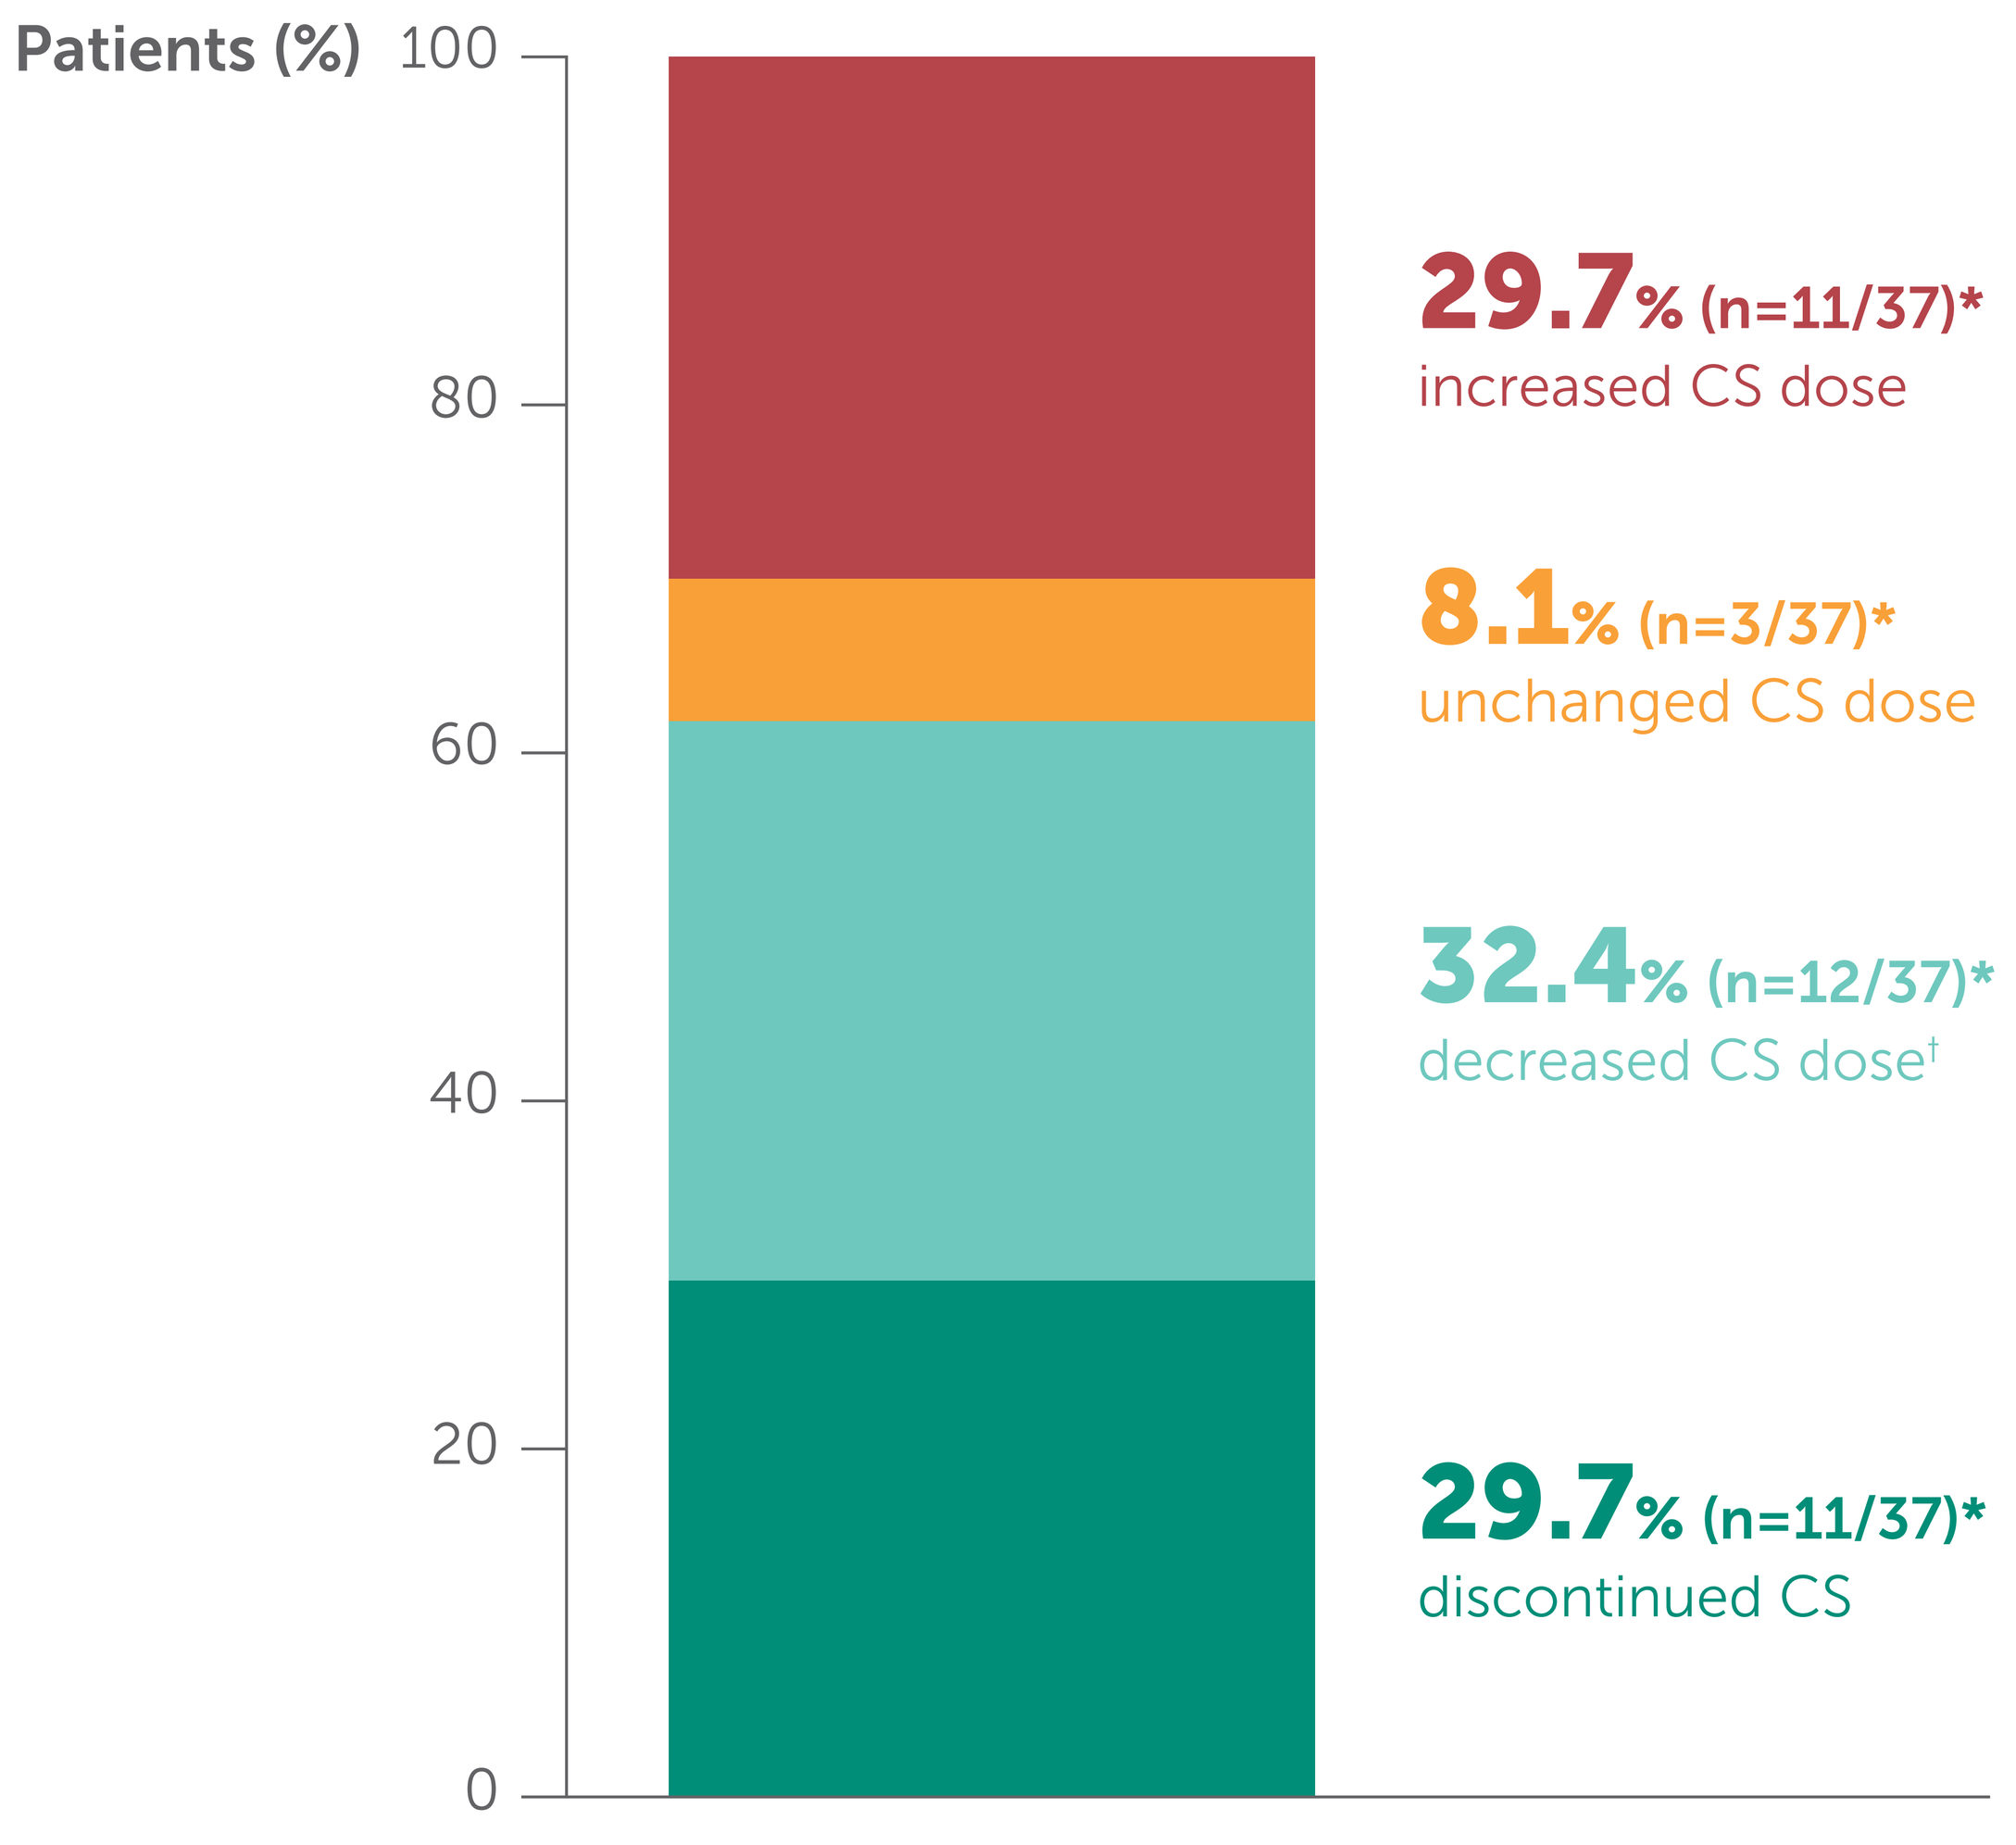
Patients (%)
100
80
60
40
20
0
29.7% (n=11/37)*
increased CS dose
8.1% (n=3/37)*
unchanged CS dose
32.4% (n=12/37)*
decreased CS dose†
29.7% (n=11/37)*
discontinued CS

Denominator is patients with ≥1 CS claim at BL. Data are based on claims only, so may not reflect what the patient ultimately received as a dose.
*Six patients with baseline CS use had missing dosage information and were excluded from this analysis.
†Decreased, but did not discontinue CS.

Figure 3 Generally, HCRU decreased during follow-up

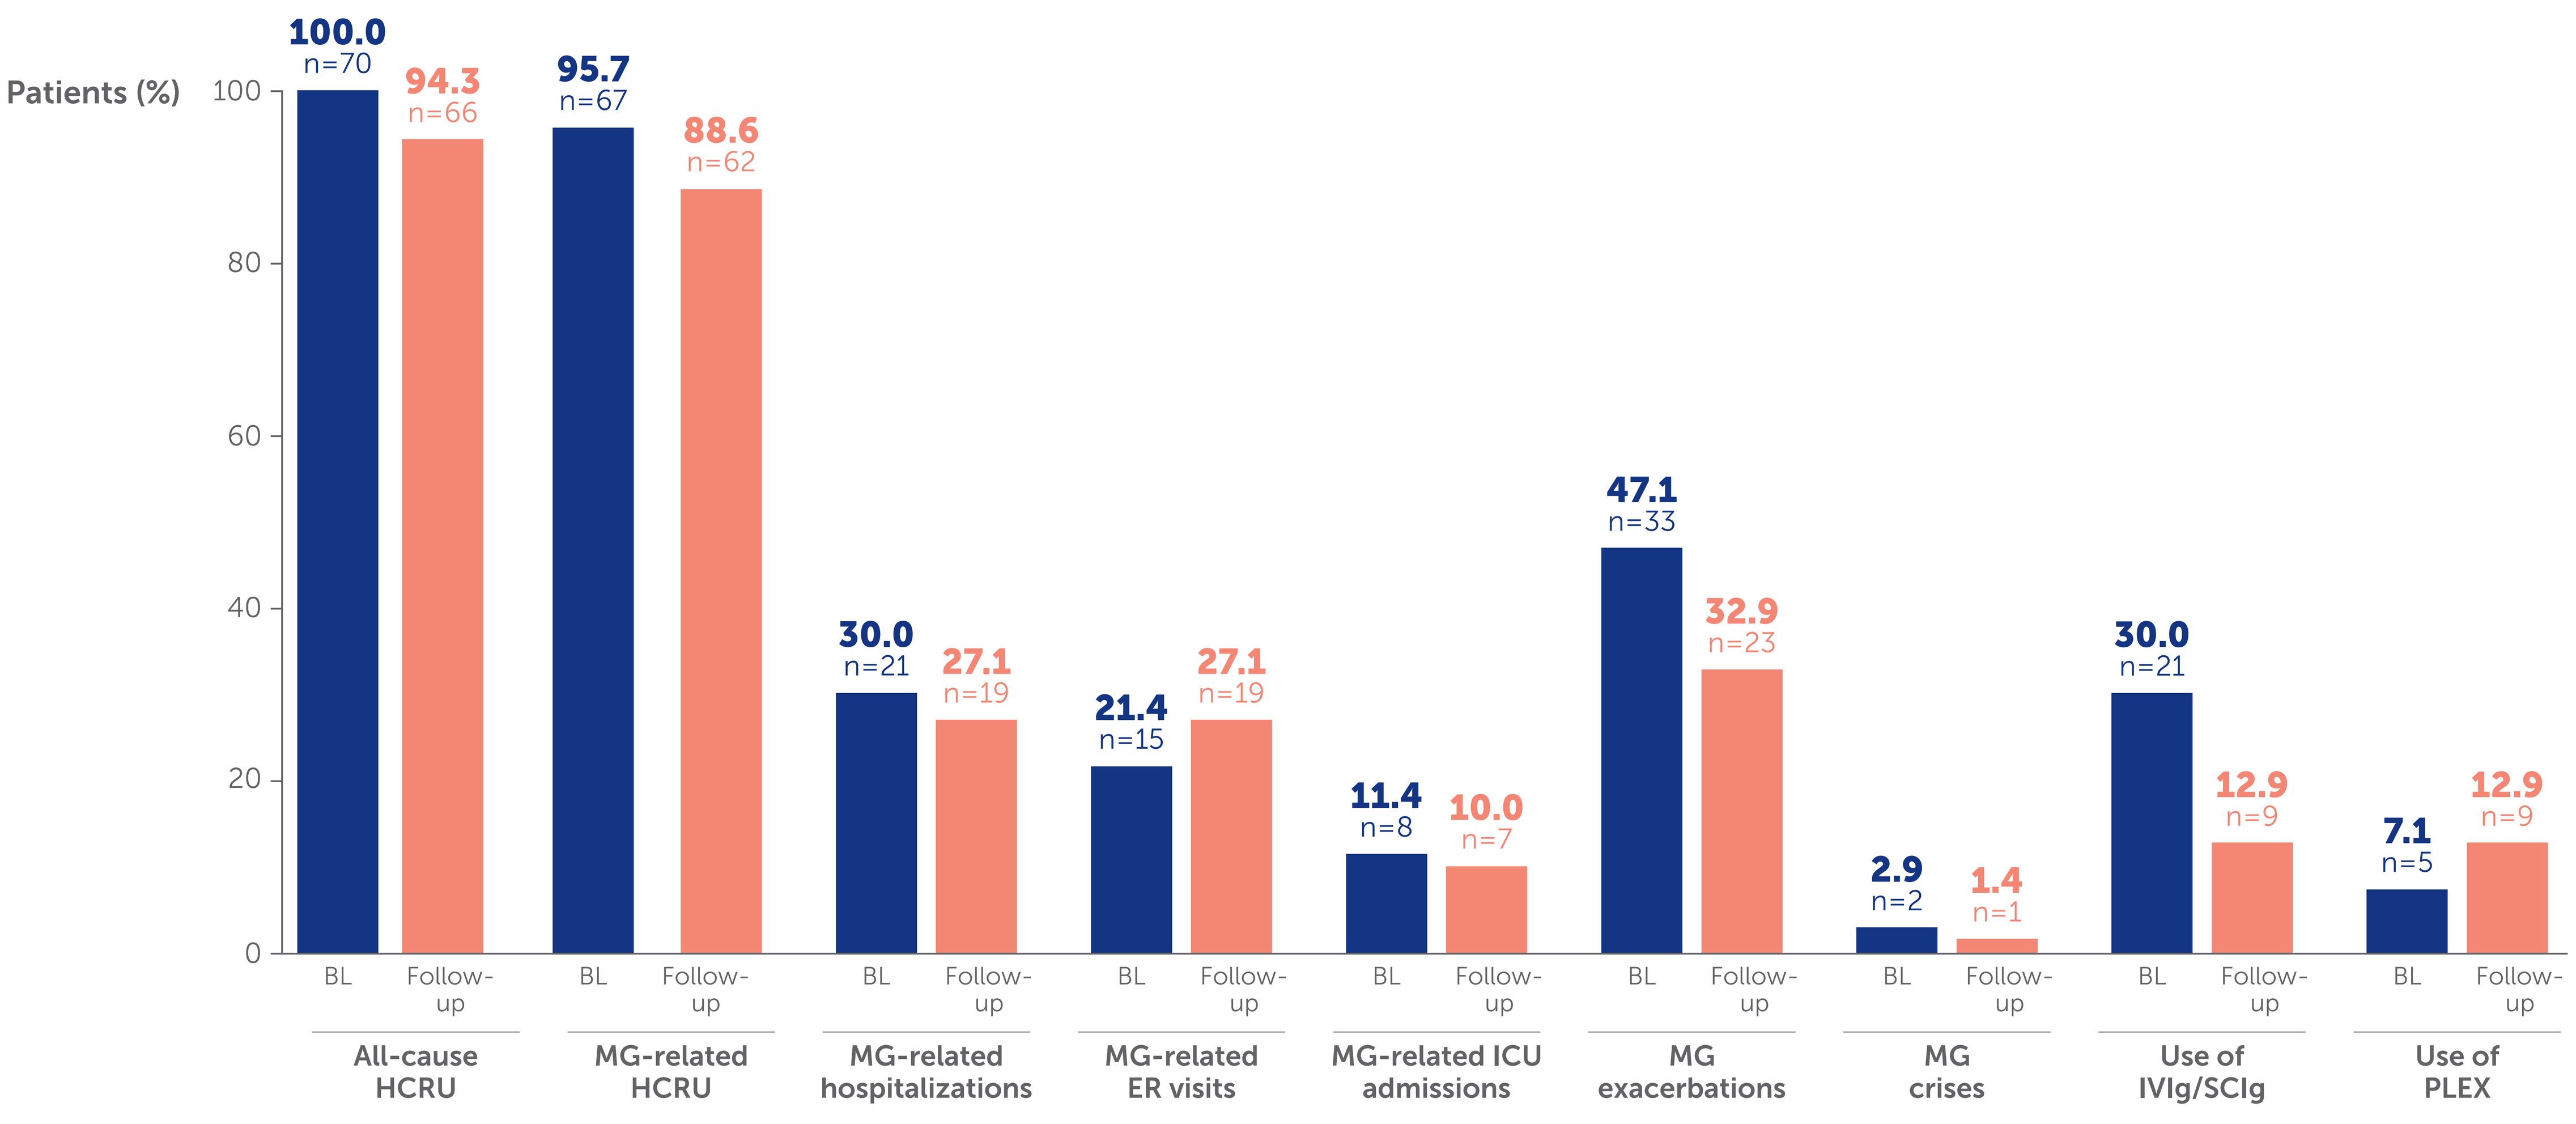
Patients (%)
0 20 40 60 80 100
All-cause HCRU: BL 100.0 n=70; Follow-up 94.3 n=66
MG-related HCRU: BL 95.7 n=67; Follow-up 88.6 n=62
MG-related hospitalizations: BL 30.0 n=21; Follow-up 27.1 n=19
MG-related ER visits: BL 21.4 n=15; Follow-up 27.1 n=19
MG-related ICU admissions: BL 11.4 n=8; Follow-up 10.0 n=7
MG exacerbations: BL 47.1 n=33; Follow-up 32.9 n=23
MG crises: BL 2.9 n=2; Follow-up 1.4 n=1
Use of IVIg/SCIg: BL 30.0 n=21; Follow-up 12.9 n=9
Use of PLEX: BL 7.1 n=5; Follow-up 12.9 n=9

Abbreviations: Ab+, antibody positive; AChR, acetylcholine receptor; BL, baseline; CS, corticosteroids; ER, emergency room; FcRn, neonatal fragment crystallizable receptor; gMG, generalized myasthenia gravis; HCRU, healthcare resource utilization; ICU, intensive care unit; IgG, immunoglobulin G; IVIg, intravenous immunoglobulin; MG, myasthenia gravis; MuSK, muscle-specific tyrosine kinase; NSIST, nonsteroidal immunosuppressant therapy; PLEX, plasma exchange; RLZ, rozanolixizumab; SCIg, subcutaneous immunoglobulin; SD, standard deviation.

Acknowledgments: This study was funded by UCB. The authors acknowledge Julia Stevens, PhD, of Ogilvy Health, for editorial assistance, which was funded by UCB. The authors thank Kate Weatherall, PhD, CMPP, of UCB, for publication and editorial support. The authors thank the participants and their caregivers, in addition to the investigators and their teams who contributed to this study.

Author disclosures: Chioma Ezenduka, Kristina Bardenheuer, Thaïs Tarancón and Diogo Pata are employees and shareholders of UCB. Rosa Falotico, Jianing Yi and Minjee Park are employees of Alira Health.

References: 1. RYSTIGGO® US PI. https://www.ucb-usa.com/RYSTIGGO-prescribing-information.pdf. Accessed August 2026. 2. Sirina J, et al. Cellular Immunol. 2026; 426:105128. 3. Binks SNM, et al. J Neurol. 2025;272(3):226.

Please use this QR code to download a PDF of the poster.