Retrospective Observation of Liver Function Tests in Patients Receiving Biologic or Targeted Oral Systemic Treatments with Psoriasis, Psoriatic Arthritis, and/or Hidradenitis Suppurativa

Matthew G. Bowles,[1] Uche Ndefo,[2] Ashley Lauterbach,[3] Autumn D. Zuckerman,[1] Prajakta P. Masurkar,[2] Robert Low,[2] Nicolas Gargurevich,[4] Leena Choi,[4] Katie A. O'Connell,[5] Chelsea P. Renfro[1,*]

[1]Vanderbilt Specialty Pharmacy, Vanderbilt Health, Nashville, TN, USA; [2]UCB, Smyrna, GA, USA; [3]College of Pharmacy & Health Sciences, Belmont University, Nashville, TN, USA; [4]Department of Biostatistics, Vanderbilt University Medical Center, Nashville, TN, USA; [5]Department of Dermatology, Vanderbilt University Medical Center, Nashville, TN, USA.

*Affiliation at time of study

Objectives

To understand real-world liver function test (LFT) monitoring practices among HCPs clinically managing patients living with psoriasis (PSO), psoriatic arthritis (PsA), and/or hidradenitis suppurativa (HS) receiving biologic or targeted oral systemic treatments.

To assess the frequency of LFT abnormalities and elevations and whether these led to treatment modifications.

Synopsis

- Current American Academy of Dermatology – National Psoriasis Foundation (AAD–NPF) guidelines acknowledge the occurrence of rare elevated liver transaminases among patients with PSO receiving biologic or oral targeted systemic treatments.[1]
- However, LFT monitoring recommendations are inconsistent.

Methods

- Study design and included patients: Real-world retrospective cohort study of adult patients (age: ≥18 years) with PSO, PsA, and/or HS newly initiating biologic or targeted oral systemic treatments, as prescribed by dermatologists and rheumatologists, from January 2021–June 2023
- Data source: eStar, Vanderbilt University Medical Center version
- Excluded: Known pre-existing liver conditions, such as non-alcoholic fatty liver disease and hepatitis, and history of chronic alcohol or substance misuse
- Assessments: LFTs at baseline (within 6 months of prescription) and 12 months post initiation of a qualifying treatment, treatment modifications due to LFT elevations
- "Abnormal" LFTs were defined as outside the reference range, and "elevated" LFTs, indicating clinical significance, were defined as >3x the upper limit of normal (ULN).
- Analysis: Multivariable negative binomial regression to analyze the effect of different patient characteristics on the number of LFT assessments at follow-up

Results

Baseline characteristics

- In total, 393 patients were included in the analysis, most of whom had PSO (86.3%), followed by PsA (64.4%) and HS (10.9%).
- Over half of patients (59.8%) had >1 diagnosis (mostly PSO and PsA).
- Further baseline characteristics are provided in the **Table**.

LFT monitoring at baseline and follow-up

- In total, 78.6% of patients had baseline LFT monitoring, with a mean (standard deviation [SD]) of 1.1 (1.0) LFTs conducted during the baseline period (**Figure 2**).
- In the year following prescription, 67.7% of patients had ≥1 LFTs, with a mean (SD) of 1.3 (1.6) LFTs conducted (**Figure 2**).
- Of the patients with ≥1 LFTs during follow-up, 51.9% had ≥1 abnormal LFTs and 1.1% had ≥1 elevated LFTs (**Figure 2**).
- Of the 207 individual abnormal LFTs, 48.3% were marked as resolved within a median (interquartile range [IQR]) of 151.0 (89.0–204.0) days (**Figure 3**).
- Treatment modifications due to LFT elevations were rare, occurring in 1/393 patients.

Frequency of LFT monitoring by patient characteristics

- On average, patients receiving concurrent treatments (incidence rate ratio [IRR]: 1.67) had more LFTs than patients not receiving them (p<0.001, **Figure 4**).
- Patients with ≥1 baseline comorbidities (IRR: 1.25) had, on average, more LFTs than patients without any baseline comorbidities (**Figure 4**).
- LFTs in other patient subgroups, including each IRR, are shown in **Figure 4**.

Conclusions

Most patients initiating biologic or oral targeted systemic treatments had ≥1 LFTs in the first year of treatment.

Although abnormal LFTs were common, clinically significant elevations (>3× ULN) were rare and seldom prompted treatment modifications.

These findings demonstrate that HCPs are routinely monitoring LFTs in patients on biologic and oral targeted systemic treatments, highlighting opportunities for clearer guidance on monitoring needs.

Summary

A **real-world retrospective cohort study** to understand **LFT monitoring practices** among patients with PSO, PsA, and/or HS receiving **biologic or targeted oral treatments**

During the 6-month baseline period prior to prescription: **79%** of patients **had ≥1 LFTs**

In the 12 months following prescription: **68%** of patients **had ≥1 LFTs**

and of these patients:
- **52% had ≥1 abnormal LFTs** (outside the reference range)
- **1% had ≥1 elevated LFTs** (>3x ULN)
- Approximately **half** of individual abnormal LFTs were resolved within a median of **151 days**

Most patients initiating a treatment **had ≥1 LFT assessments in the first year**, and while abnormal LFTs were common, **clinically significant elevations** (>3x ULN) and **treatment modifications** were **rare**

Figure 1 Study design

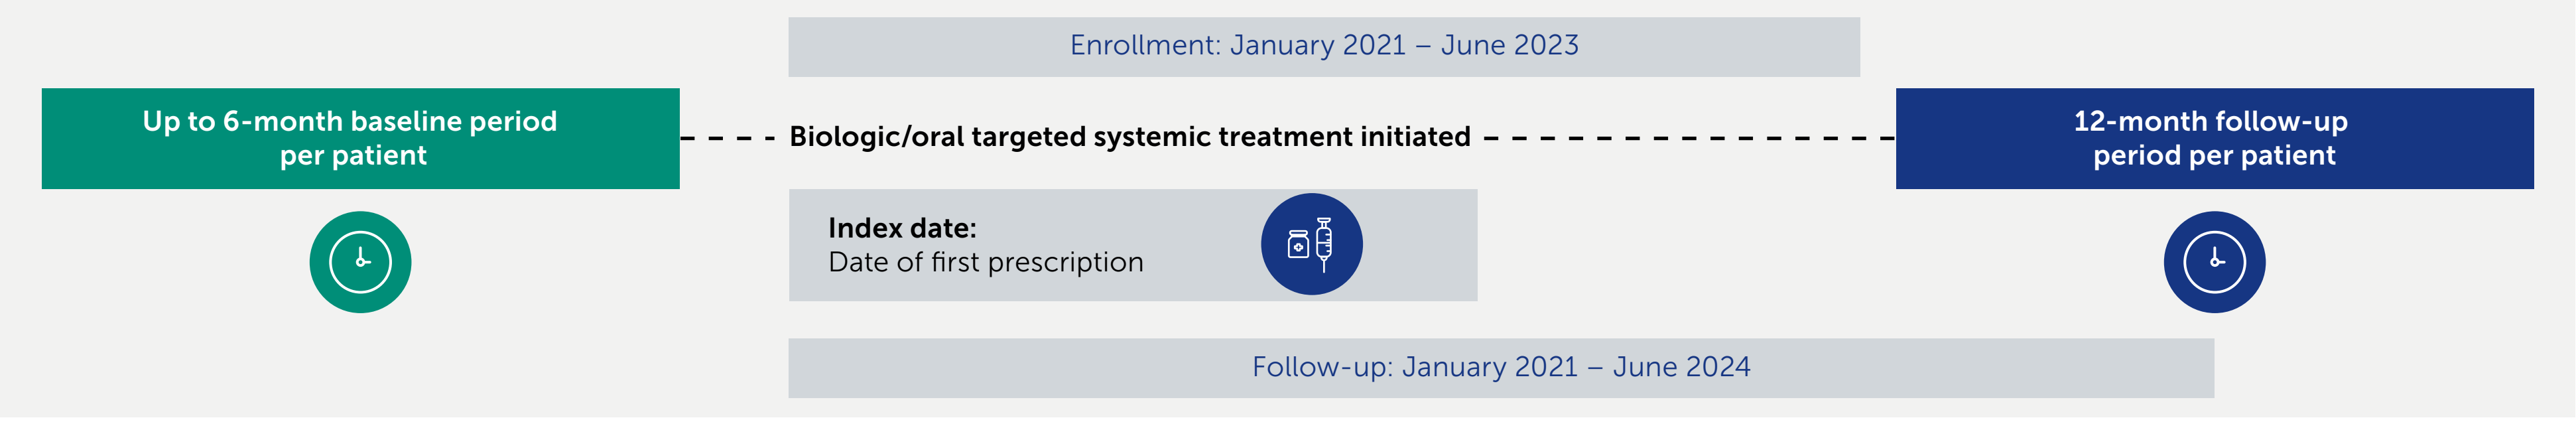
Enrollment: January 2021 – June 2023
Up to 6-month baseline period per patient
Biologic/oral targeted systemic treatment initiated
12-month follow-up period per patient
Index date:
Date of first prescription
Follow-up: January 2021 – June 2024

Table Characteristics of included patients

n (%), unless otherwise specified	N=393
Age, years, median (IQR)	50.1 (36.7–60.9)
Sex, female	245 (62.3)
Racial group	
White	327 (83.2)
Black or African American	29 (7.4)
Asian	6 (1.5)
Hispanic/Latinx	5 (1.3)
Other/Unknown	26 (6.6)
Diagnosis[a]	
PSO only	104 (26.5)
PSA only	27 (6.9)
HS only	27 (6.9)
PSO + PsA	219 (55.7)
PSO + HS	9 (2.3)
PSO + PsA + HS	7 (1.8)
Comorbidities of interest	
Obesity (BMI: ≥30 kg/m^2)	188 (47.8)
Dyslipidemia	100 (25.4)
Diabetes	77 (19.6)
None	157 (39.9)
Biologic/oral targeted systemic treatment class	
TNFi	179 (45.5)
PDE4i	109 (27.7)
IL-23i	40 (10.2)
IL-17A/Fi	38 (9.7)
IL-12/23i	18 (4.6)
JAKi	9 (2.3)
Concurrent treatments	
Oral corticosteroid	113 (28.8)
Methotrexate	50 (12.7)
Other[b]	2 (0.5)
None	251 (63.9)

[a] Patients could have multiple diagnoses: 219 patients (55.7%) had PSO and PsA, 9 patients (2.3%) had PSO and HS, and 7 patients (1.8%) had PSO, PsA and HS. **[b]** One patient each was receiving mycophenolate and acitretin.

Figure 2 Patients with ≥1 LFT assessments during baseline and follow-up

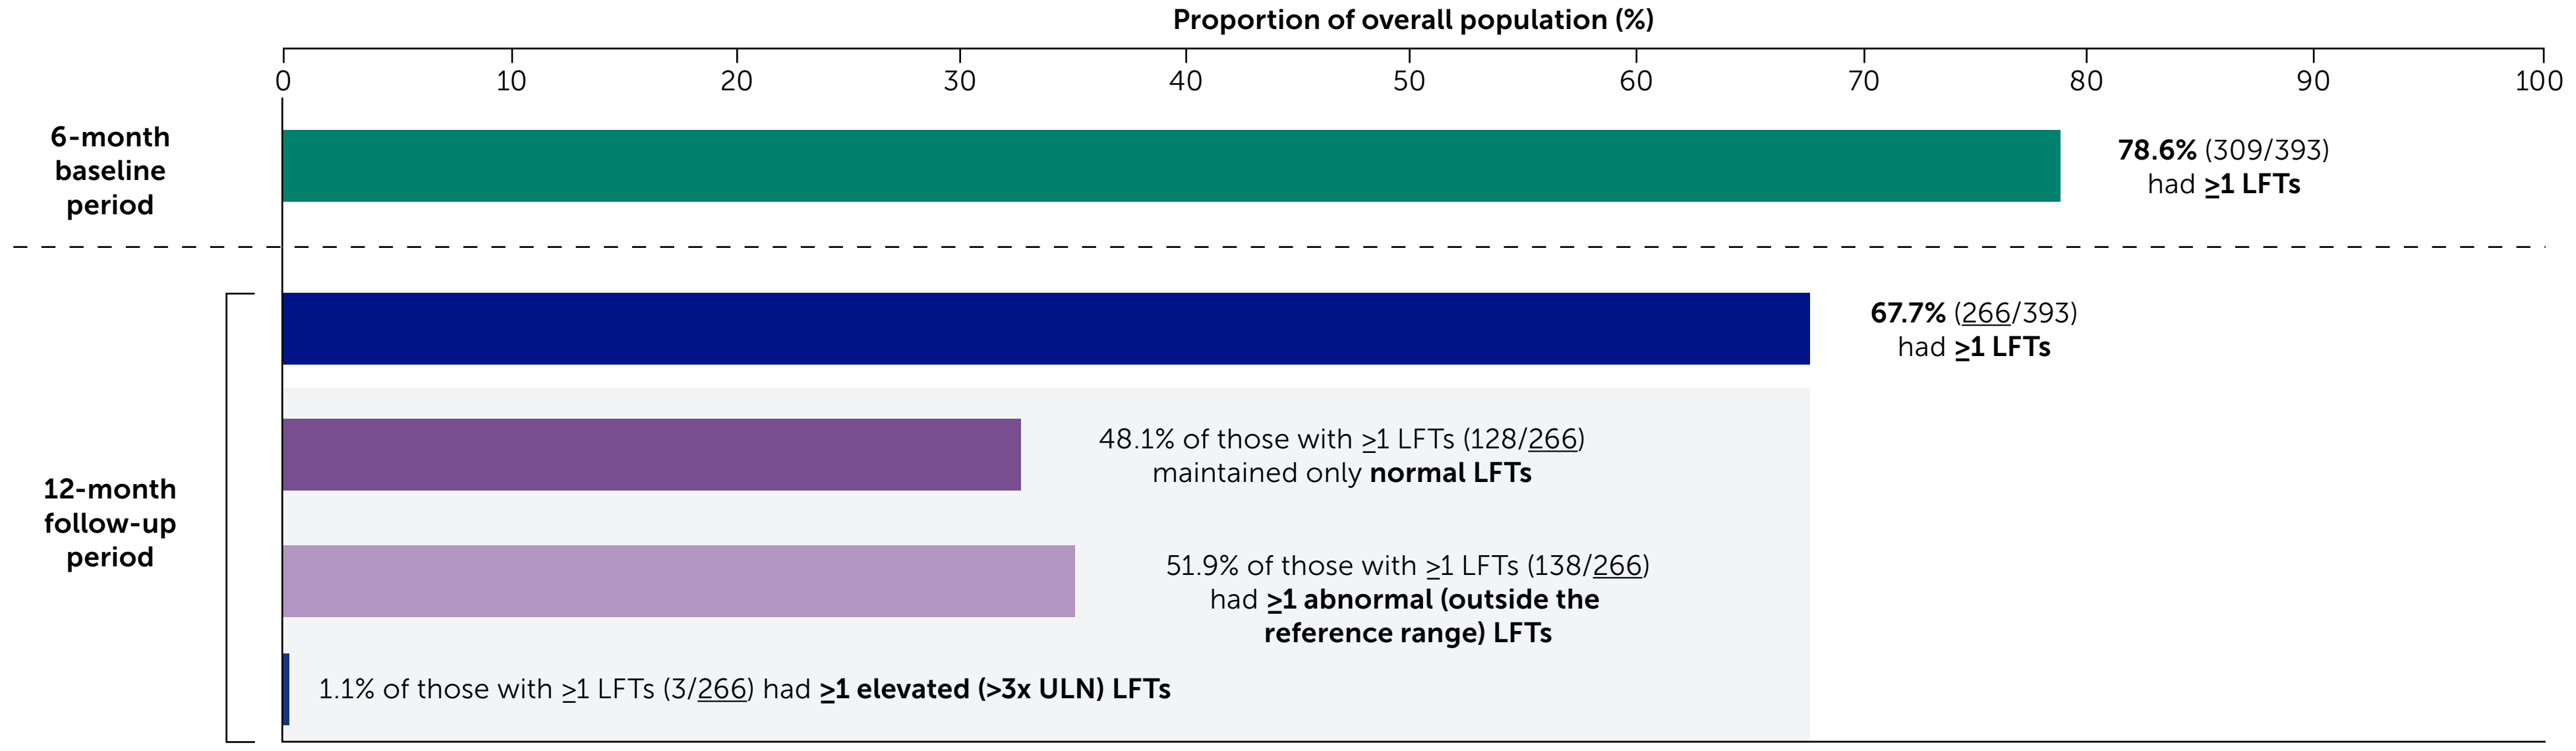
Proportion of overall population (%)
0
10
20
30
40
50
60
70
80
90
100
6-month baseline period
78.6% (309/393) had ≥1 LFTs
12-month follow-up period
67.7% (266/393) had ≥1 LFTs
48.1% of those with ≥1 LFTs (128/266) maintained only normal LFTs
51.9% of those with ≥1 LFTs (138/266) had ≥1 abnormal (outside the reference range) LFTs
1.1% of those with ≥1 LFTs (3/266) had ≥1 elevated (>3x ULN) LFTs

Figure 3 Outcomes of individual LFT assessments during follow-up

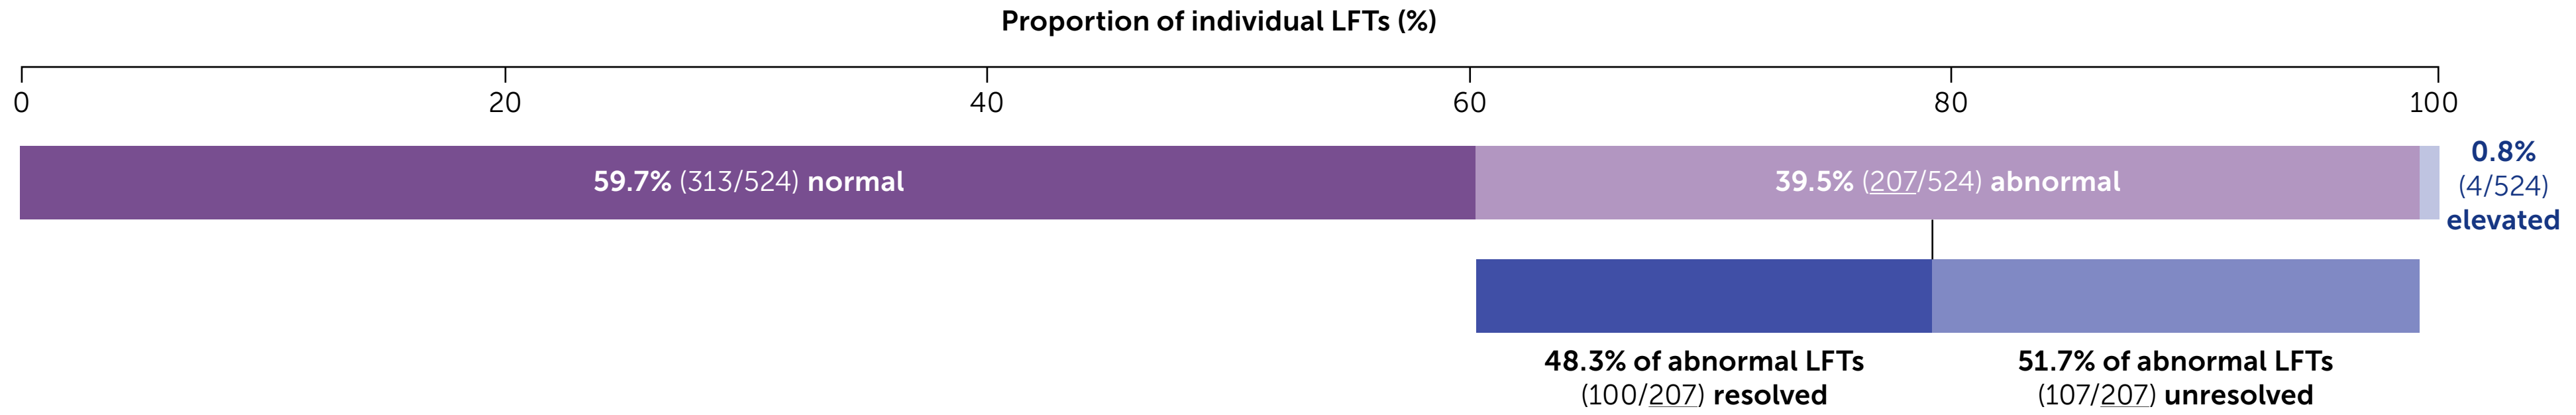
Proportion of individual LFTs (%)
0
20
40
60
80
100
59.7% (313/524) normal
39.5% (207/524) abnormal
0.8% (4/524) elevated
48.3% of abnormal LFTs (100/207) resolved
51.7% of abnormal LFTs (107/207) unresolved

Patients may have had ≥1 LFTs in the 12-month follow-up period.

Figure 4 Regression analysis for number of LFT assessments, categorized by patient subgroups, at follow-up[a]

Patient subgroup	IRR (95% CI)	p value
Receiving concurrent treatments	1.67 (1.35–2.06)	<0.001
With ≥1 baseline comorbidities	1.25 (1.00–1.57)	0.0503
With ≥1 new diagnoses during study	1.21 (0.87–1.68)	0.254
With ≥1 abnormal LFTs at baseline	1.16 (0.94–1.44)	0.155
Male	1.08 (0.87–1.34)	0.469
With >1 dermatology diagnoses	1.08 (0.87–1.34)	0.502
Age (per 1-year increment)	1.00 (0.99–1.01)	0.897
Receiving PDE4i	0.77 (0.60–0.98)	0.037

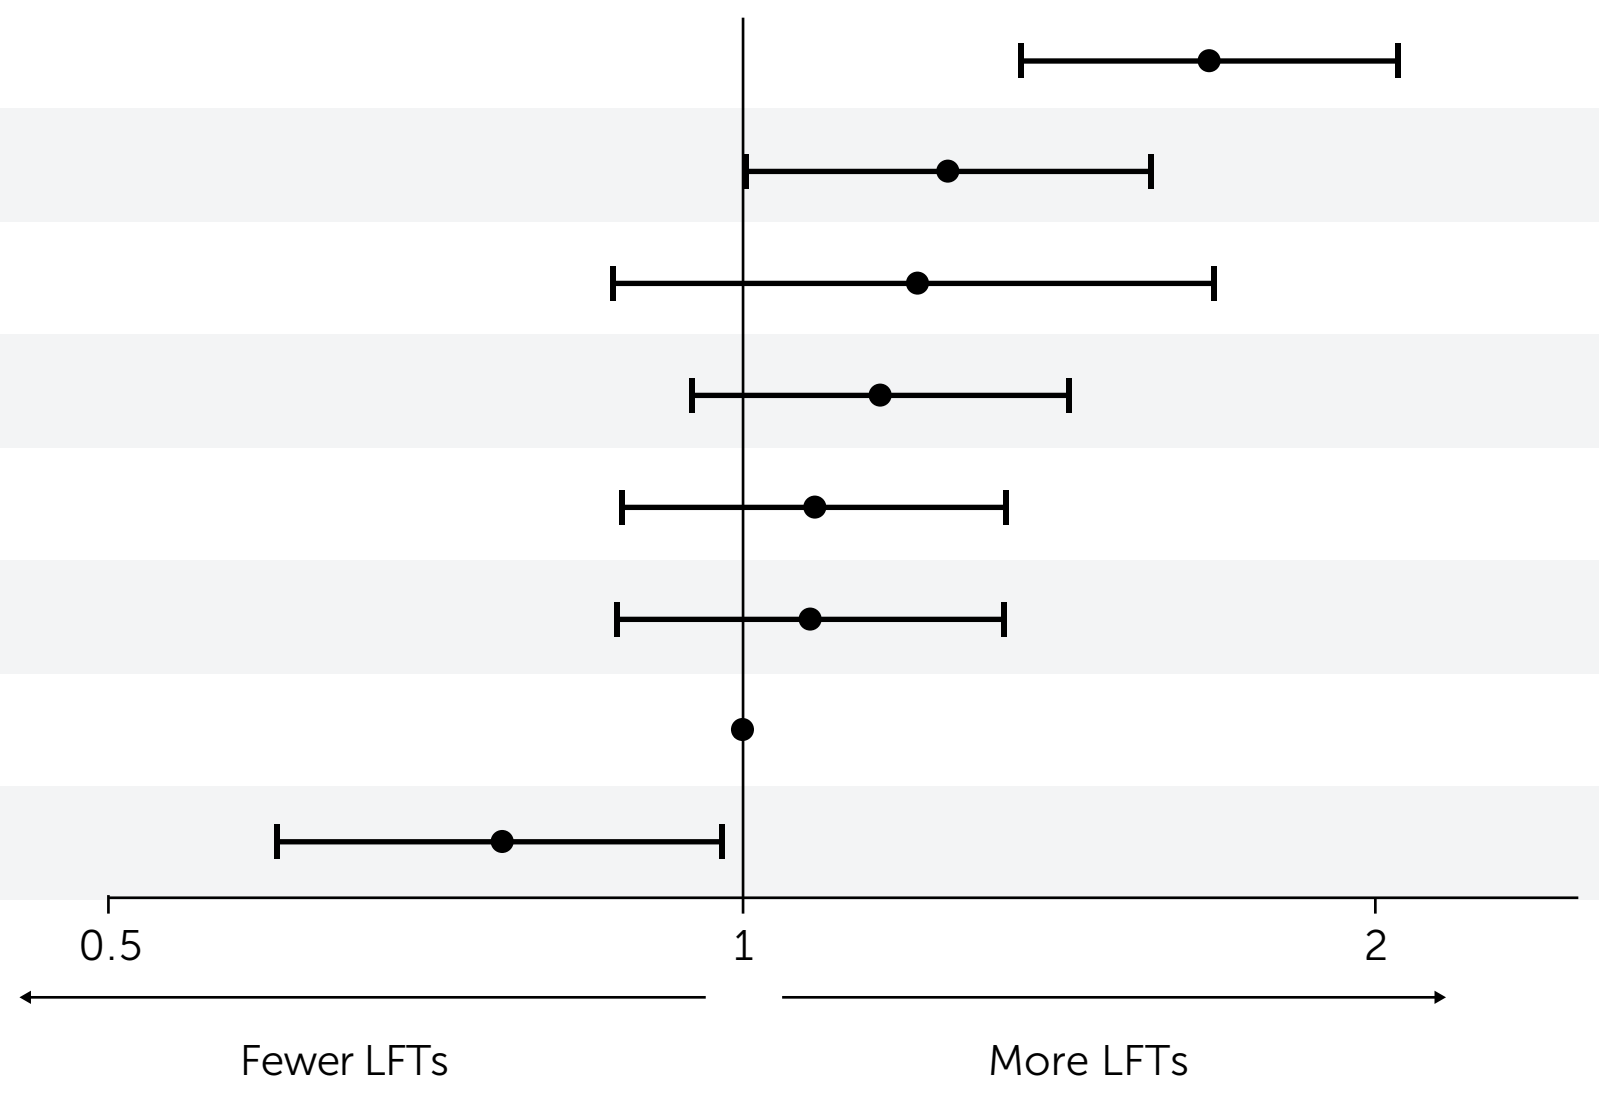
0.5
1
2
Fewer LFTs
More LFTs

[a] This analysis was conducted using multivariable negative binomial regression.

AAD–NPF: American Academy of Dermatology – National Psoriasis Foundation; **BMI:** body mass index; **HS:** hidradenitis suppurativa; **IL-12/23i:** interleukin-12/23 inhibitor; **IL-17A/Fi:** interleukin-17A/F inhibitor; **IL-23i:** interleukin-23 inhibitor; **IQR:** interquartile range; **IRR:** incidence rate ratio; **JAKi:** janus kinase inhibitor; **LFT:** liver function test; **PDE4i:** phosphodiesterase-4 inhibitor; **PsA:** psoriatic arthritis; **PSO:** psoriasis; **TNFi:** tumor necrosis factor inhibitor; **ULN:** upper limit of normal.

References: [1]Menter A et al. J Am Acad Dermatol. 2019;80(4):1029–72.
Author Contributions: All authors had substantial contributions to the study conception/design or acquisition/analysis/interpretation of data, drafted the publication or reviewed it critically for important intellectual content, and approved the final publication. **Author Disclosures:** **MGB:** Advisory board participant for Eli Lilly and Company. **ADZ:** Research support paid to institution from BeiGene, BridgeBio, Sanofi, Takeda, and UCB. **UN**, **PPM**, **RL:** Employees and shareholders of UCB. **AL**, **NG**, **LC**, **KAO:** None. **CPR:** Employee of Loopback Health; research support paid to institution from BeiGene, BridgeBio, Takeda, and UCB. **Acknowledgments:** The authors acknowledge Tiffany Park, PharmD, of UCB, Smyrna, GA, USA, for editorial review during manuscript development and publication coordination, Kaity McCafferty Layte, BSc, of Mac Medical Communications, London, UK, and Lucy Berry, MBBS, CMPP, of Joinery Medical Communications, Biggleswade, UK, for medical writing support and editorial assistance, and the Costello Medical Creative Team for design support. This study was funded by UCB. All costs associated with development of this poster were funded by UCB.

To receive a copy of this poster, scan the QR code.

Link expiration: 09 January 2027

Presented at Fall Clinical Dermatology Conference 2026 | October 08–11 | Las Vegas, NV