Bimekizumab durability of response through 4 years in patients with plaque psoriasis who stopped and re-started treatment

Christopher E. M. Griffiths,[1] Antonio Costanzo,[2] Kim A. Papp,[3,4] David Rosmarin,[5] Luis Puig,[6] Andrew Blauvelt,[7] Krista Wixted,[8] José M. López Pinto,[9] Jérémy Lambert,[10] Rhys Warham,[11,12] George Han[13]

[1]Department of Dermatology, King's College Hospital, King's College London, London, UK; [2]Dermatology, Humanitas Clinical and Research Centre, IRCCS, Rozzano, Milan, Italy; [3]Probity Medical Research and Alliance Clinical Trials, Waterloo, ON, Canada; [4]Division of Dermatology, Temerty Faculty of Medicine, University of Toronto, Toronto, ON, Canada; [5]Indiana University School of Medicine, Indianapolis, IN, USA; [6]Department of Dermatology, Hospital de la Santa Creu i Sant Pau, Universitat Autònoma de Barcelona, Barcelona, Spain; [7]Blauvelt Consulting LLC, Annapolis, MD, USA; [8]UCB, Morrisville, NC, USA; [9]UCB, Madrid, Spain; [10]UCB, Colombes, France; [11]UCB, Slough, UK; [12]Veramed, London, UK; [13]Department of Dermatology, Icahn School of Medicine at Mount Sinai, New York, NY, USA.

Objective

To report the long-term durability of treatment responses over an extended follow-up period (up to 4 years) in patients with moderate to severe plaque psoriasis who stopped and then restarted bimekizumab (BKZ).

Synopsis

- Real-world psoriasis treatment may be interrupted due to factors including, but not limited to, surgery, hospitalization, financial considerations, and travel.
- While BKZ has demonstrated durable responses through 2 years' retreatment following interruption,[1] it is important to continue assessing how well responses are recaptured or maintained over longer periods upon retreatment.

Methods

- Data are reported from the BE READY phase 3 trial and its open-label extension (OLE) BE BRIGHT.[2,3] Included patients were randomized to BKZ 320 mg every 4 weeks (Q4W), achieved ≥90% improvement from baseline in Psoriasis Area and Severity Index (PASI 90) at Week 16, then were rerandomized to placebo (PBO) for a 40-week randomized-withdrawal period before OLE entry (**Figure 1**).
- Data are presented for two patient subgroups who stopped and restarted BKZ (**Figure 2**):
 - **Week 16–56 PBO Group**: patients who maintained ≥PASI 75 at every visit from Week 16 to 56, and continued PBO to Week 56, and switched to BKZ Q4W in the OLE;
 - **Escape Group**: patients who relapsed (<PASI 75 during Week 20–56) and entered a 12-week open-label BKZ Q4W arm before OLE entry (required ≥PASI 50).
- PASI 90, PASI 100, and DLQI 0/1 responses are reported through OLE Week 144 (Year 4) as observed cases (OC).

Results

- Baseline demographics and disease characteristics are presented in **Table 1**.
- Of 105 patients initially randomized to BKZ Q4W who achieved PASI 90 and were rerandomized to PBO at Week 16, 31.4% (33/105) maintained ≥PASI 75 to Week 56 and were included in the **Week 16–56 PBO Group**.
 - Of the patients rerandomized to PBO, 62.9% (66/105) relapsed during the randomized-withdrawal period and entered the escape arm to receive BKZ Q4W before entering the OLE (**Escape Group**; mean time to relapse [SD]: 158.8 [47.7] days/22.7 [6.8] weeks).
- Among the **Week 16–56 PBO Group**, 51.5% (17/33) and 33.3% (11/33) achieved PASI 90 and PASI 100, respectively, at Week 56 (**Figure 3A**). At Year 4, 96.3% (26/27) and 88.9% (24/27) achieved PASI 90 and PASI 100, respectively (6 patients missing).
 - DLQI 0/1 rates were high through Year 4 in the **Week 16–56 PBO Group** (**Figure 3B**).
- Among the **Escape Group**, 90.8% (59/65) and 63.1% (41/65) achieved PASI 90 and PASI 100, respectively, after 12 weeks of BKZ retreatment (**Figure 4A**). At Year 4, 92.7% (51/55) and 83.6% (46/55) achieved PASI 90 and PASI 100, respectively (11 patients missing).
 - Similarly, DLQI 0/1 rates were high through Year 4 in the **Escape Group** (**Figure 4B**).

Conclusions

Among patients who maintained ≥PASI 75 during bimekizumab withdrawal, 100% (32/32) achieved PASI 90 following 12 weeks of bimekizumab retreatment.

Among patients who relapsed after bimekizumab withdrawal, high proportions achieved complete/near-complete skin clearance and DLQI 0/1 after 12 weeks' retreatment.

In both groups, high response levels were durable through ≥3 years' bimekizumab retreatment, suggesting that pausing bimekizumab treatment did not meaningfully impact long-term disease control.

Figure 1 BE READY/BE BRIGHT study design[a]

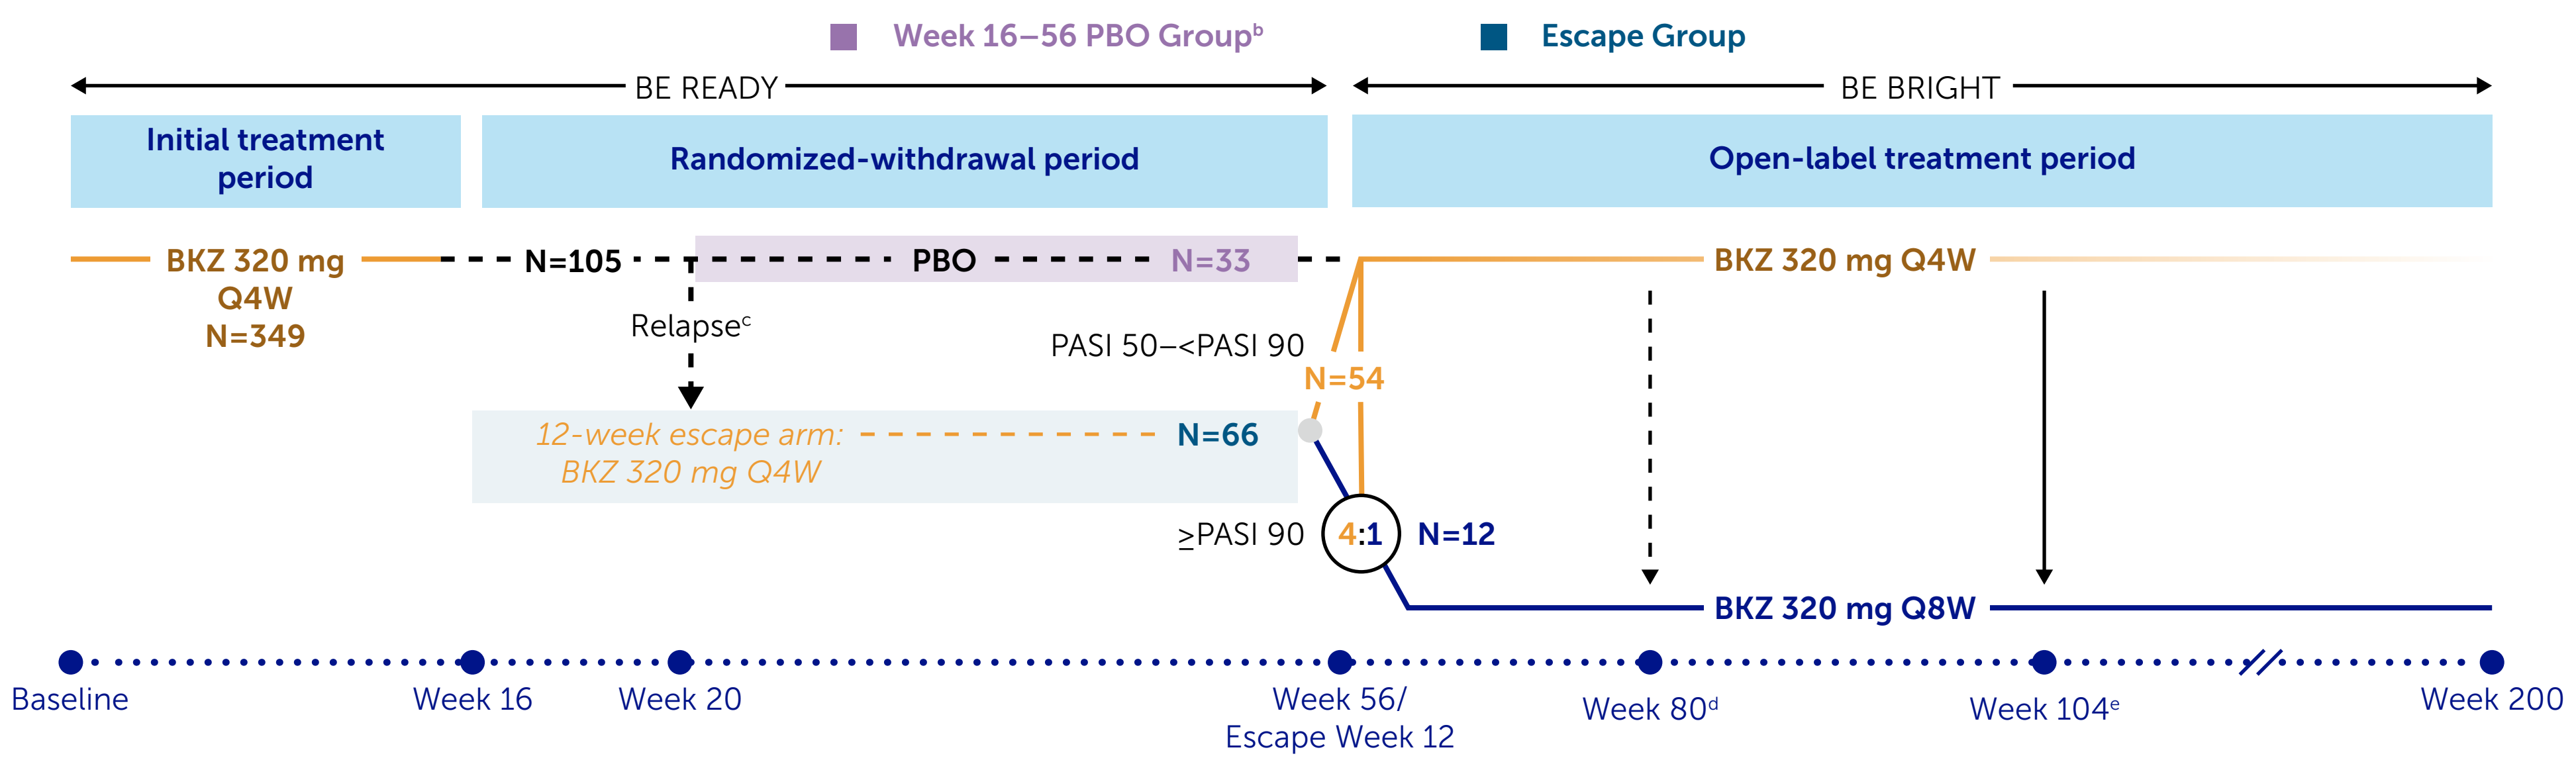

Week 16–56 PBO Group[b]
Escape Group
BE READY
BE BRIGHT
Initial treatment period
Randomized-withdrawal period
Open-label treatment period
BKZ 320 mg Q4W N=349
N=105
PBO
N=33
Relapse[c]
12-week escape arm: BKZ 320 mg Q4W
N=66
PASI 50–<PASI 90
N=54
≥PASI 90
4:1
N=12
BKZ 320 mg Q4W
BKZ 320 mg Q8W
Baseline
Week 16
Week 20
Week 56/ Escape Week 12
Week 80[d]
Week 104[e]
Week 200

[a] Full study designs have been reported previously;[1–3] [b] Excludes patients who escaped PBO treatment without meeting the criteria to do so; of the remaining 105 patients who achieved PASI 90 at Week 16 and were rerandomized to PBO, 99 entered the OLE (5 patients discontinued from BE READY [3 due to adverse events, 2 lost to follow-up] and 1 was not treated in the OLE);[1,2] [c] 'Relapse' was defined as <PASI 75 response at any visit between Week 20–56; median time to relapse from last BKZ dose (Week 12) in Week 16 PASI 90 responders was 32 weeks;[2] median time to loss of PASI 90 was 28 weeks;[4] [d] Patients receiving BKZ 320 mg Q4W who had achieved PASI 90 could be switched to receive BKZ 320 mg Q8W at the investigator's discretion (7 patients in the Week 16–56 PBO Group; 9 patients in the Escape Group); [e] At Week 104 (OLE Week 48), or the next scheduled visit, all patients who had remained on BKZ 320 mg Q4W were re-assigned to BKZ 320 mg Q8W.

Table 1 Baseline characteristics

	All patients rerandomized to PBO (N=105)	Week 16–56 PBO Group (N=33)	Escape Group (N=66)
Age (years), mean (SD)	42.3 (11.6)	41.2 (10.7)	43.2 (11.8)
Sex, male, n (%)	77 (73.3)	22 (66.7)	50 (75.8)
Racial group, white, n (%)	95 (90.5)	32 (97.0)	59 (89.4)
Weight (kg), mean (SD)	87.6 (19.2)	81.8 (20.9)	90.8 (18.3)
Duration of psoriasis (years), mean (SD)	18.9 (12.5)	14.6 (8.3)	20.6 (13.0)
PASI, mean (SD)	19.4 (6.8)	18.2 (4.8)	19.7 (7.5)
BSA (%), mean (SD)	22.9 (14.5)	18.9 (10.4)	24.6 (16.0)
IGA, n (%)			
3: moderate	73 (69.5)	24 (72.7)	45 (68.2)
4: severe	32 (30.5)	9 (27.3)	21 (31.8)
DLQI total, mean (SD)	9.4 (5.7)	9.4 (6.2)	9.6 (5.7)
Any prior systemic therapy, n (%)	77 (73.3)	19 (57.6)	54 (81.8)
Any prior biologic therapy, n (%)	40 (38.1)	6 (18.2)	33 (50.0)
Medical history of PsA, n (%)	18 (17.1)	6 (18.2)	12 (18.2)

Figure 2 Patient subgroups

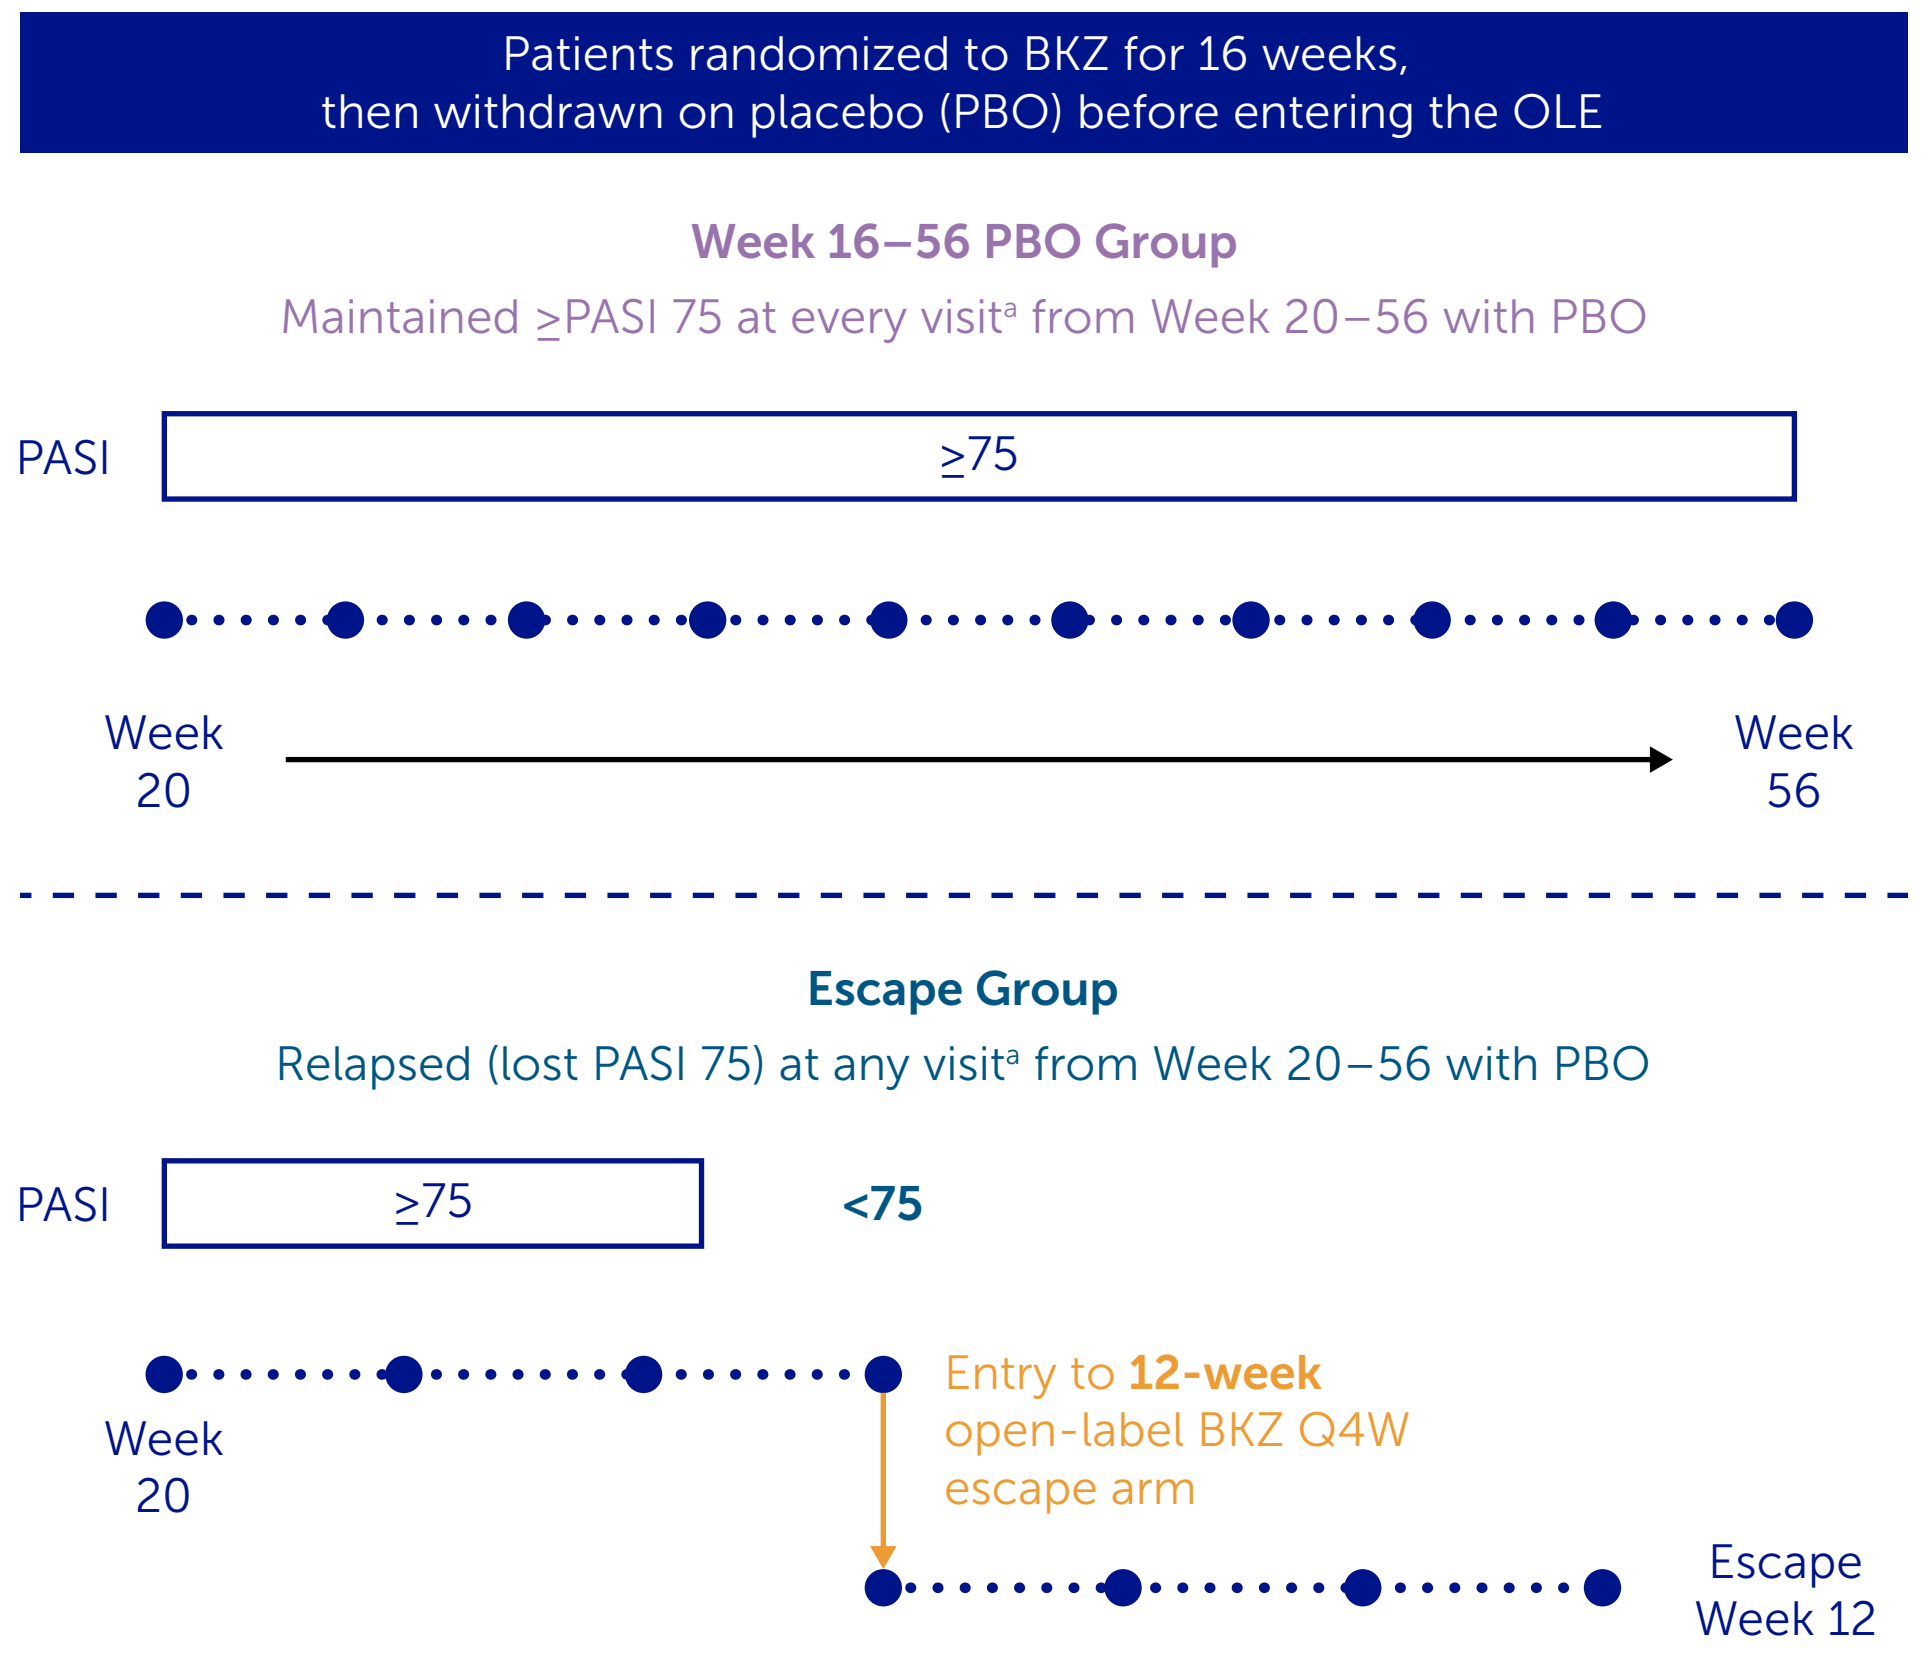

Patients randomized to BKZ for 16 weeks, then withdrawn on placebo (PBO) before entering the OLE
Week 16–56 PBO Group
Maintained ≥PASI 75 at every visit[a] from Week 20–56 with PBO
PASI
≥75
Week 20
Week 56
Escape Group
Relapsed (lost PASI 75) at any visit[a] from Week 20–56 with PBO
PASI
≥75
<75
Week 20
Entry to 12-week open-label BKZ Q4W escape arm
Escape Week 12

[a] Blue dots represent clinical trial visits every 4 weeks.

Figure 3 PASI 90, PASI 100, and DLQI 0/1 response rates through 4 years in the Week 16–56 PBO Group (OC)

A) PASI 90/PASI 100 B) DLQI 0/1

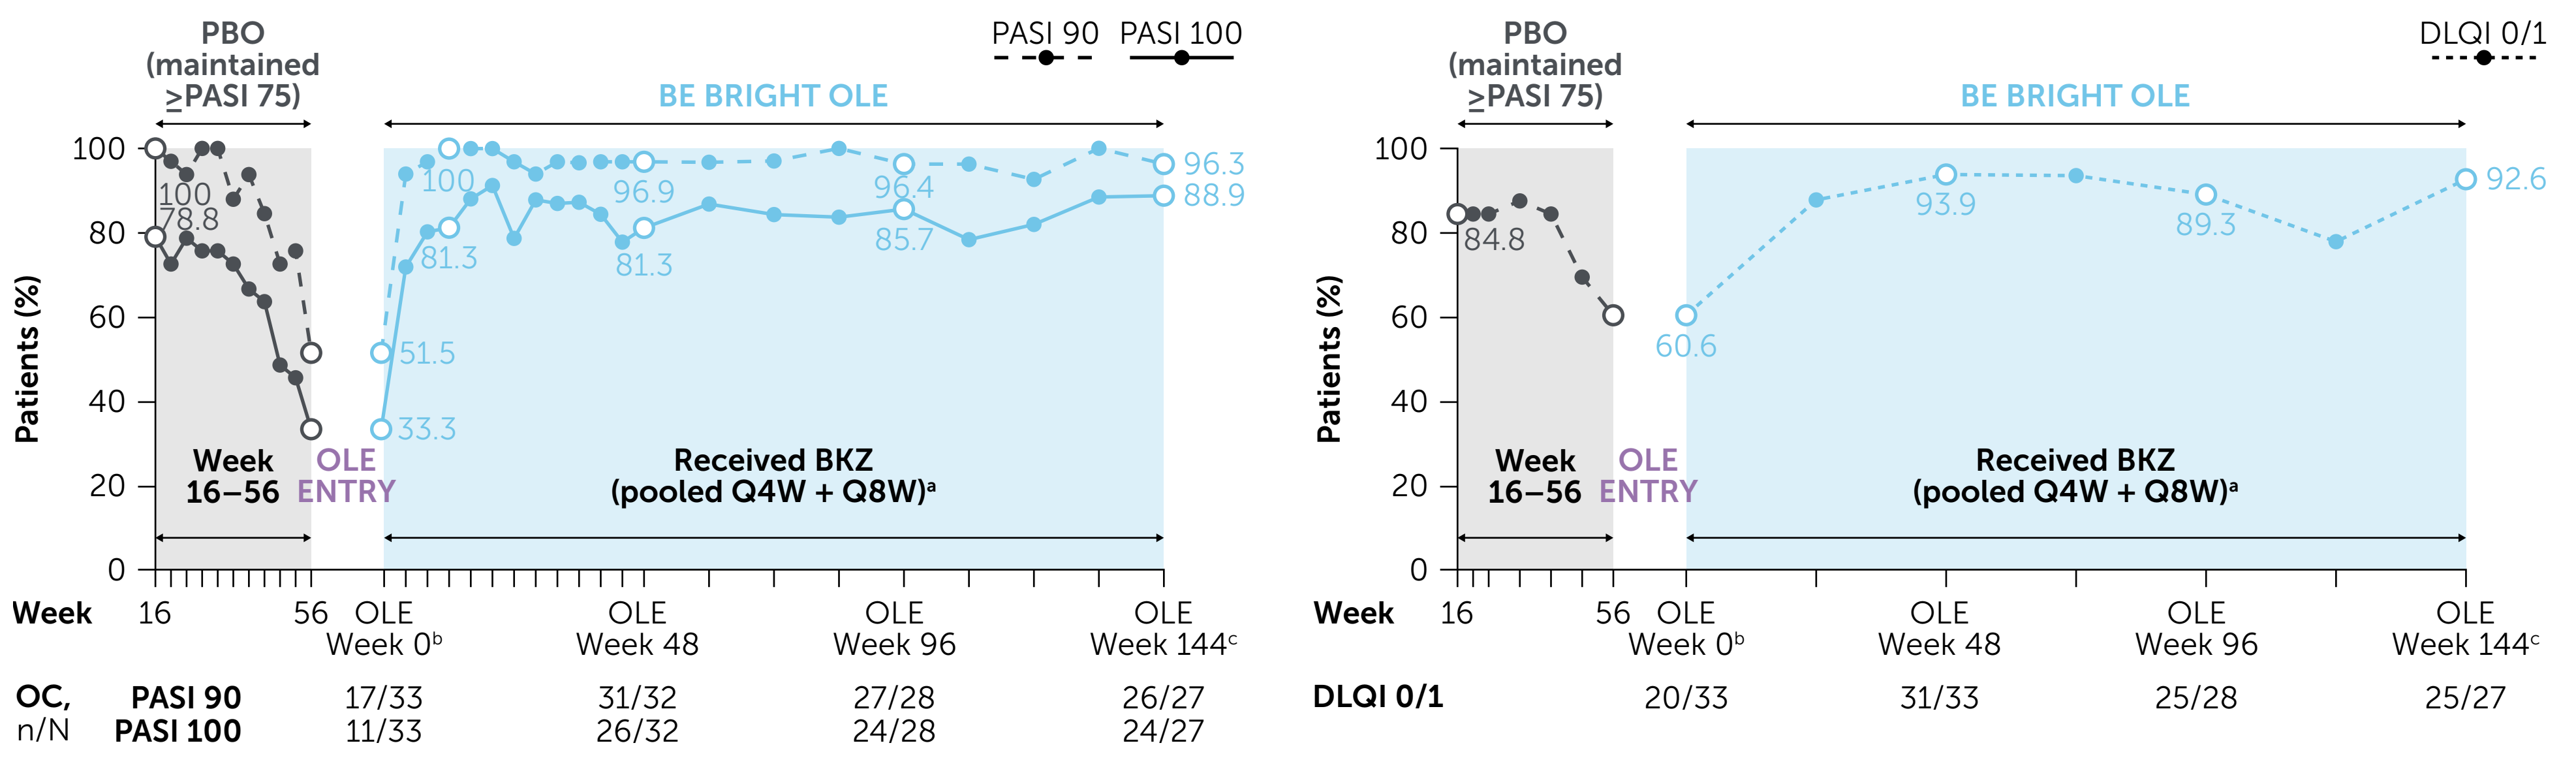

PBO (maintained ≥PASI 75)
PASI 90
PASI 100
BE BRIGHT OLE
Patients (%)
100
80
60
40
20
0
100
78.8
51.5
33.3
100
81.3
96.9
81.3
96.4
85.7
96.3
88.9
Week 16–56
OLE ENTRY
Received BKZ (pooled Q4W + Q8W)[a]
Week 16 56 OLE Week 0[b] OLE Week 48 OLE Week 96 OLE Week 144[c]
OC, n/N PASI 90 17/33 31/32 27/28 26/27
PASI 100 11/33 26/32 24/28 24/27
DLQI 0/1
84.8
60.6
93.9
89.3
92.6
DLQI 0/1 20/33 31/33 25/28 25/27

[a] Data reported from the BE BRIGHT OLE are pooled for patients who received BKZ 320 mg Q4W and Q8W; [b] Patients in the Week 16–56 PBO Group had their OLE Week 0 study assessments at the end of the 40-week randomized-withdrawal period (Week 56), having maintained ≥PASI 75 at every visit prior to that point; [c] At Year 4, 6 patients were missing from the Week 16–56 PBO Group having discontinued during the OLE (2 due to adverse events, 1 due to lack of efficacy, 1 due to protocol violation, 1 lost to follow-up, 1 due to consent being withdrawn).

Figure 4 PASI 90, PASI 100, and DLQI 0/1 response rates through 4 years in the Escape Group (OC)[a,b]

A) PASI 90/PASI 100 B) DLQI 0/1

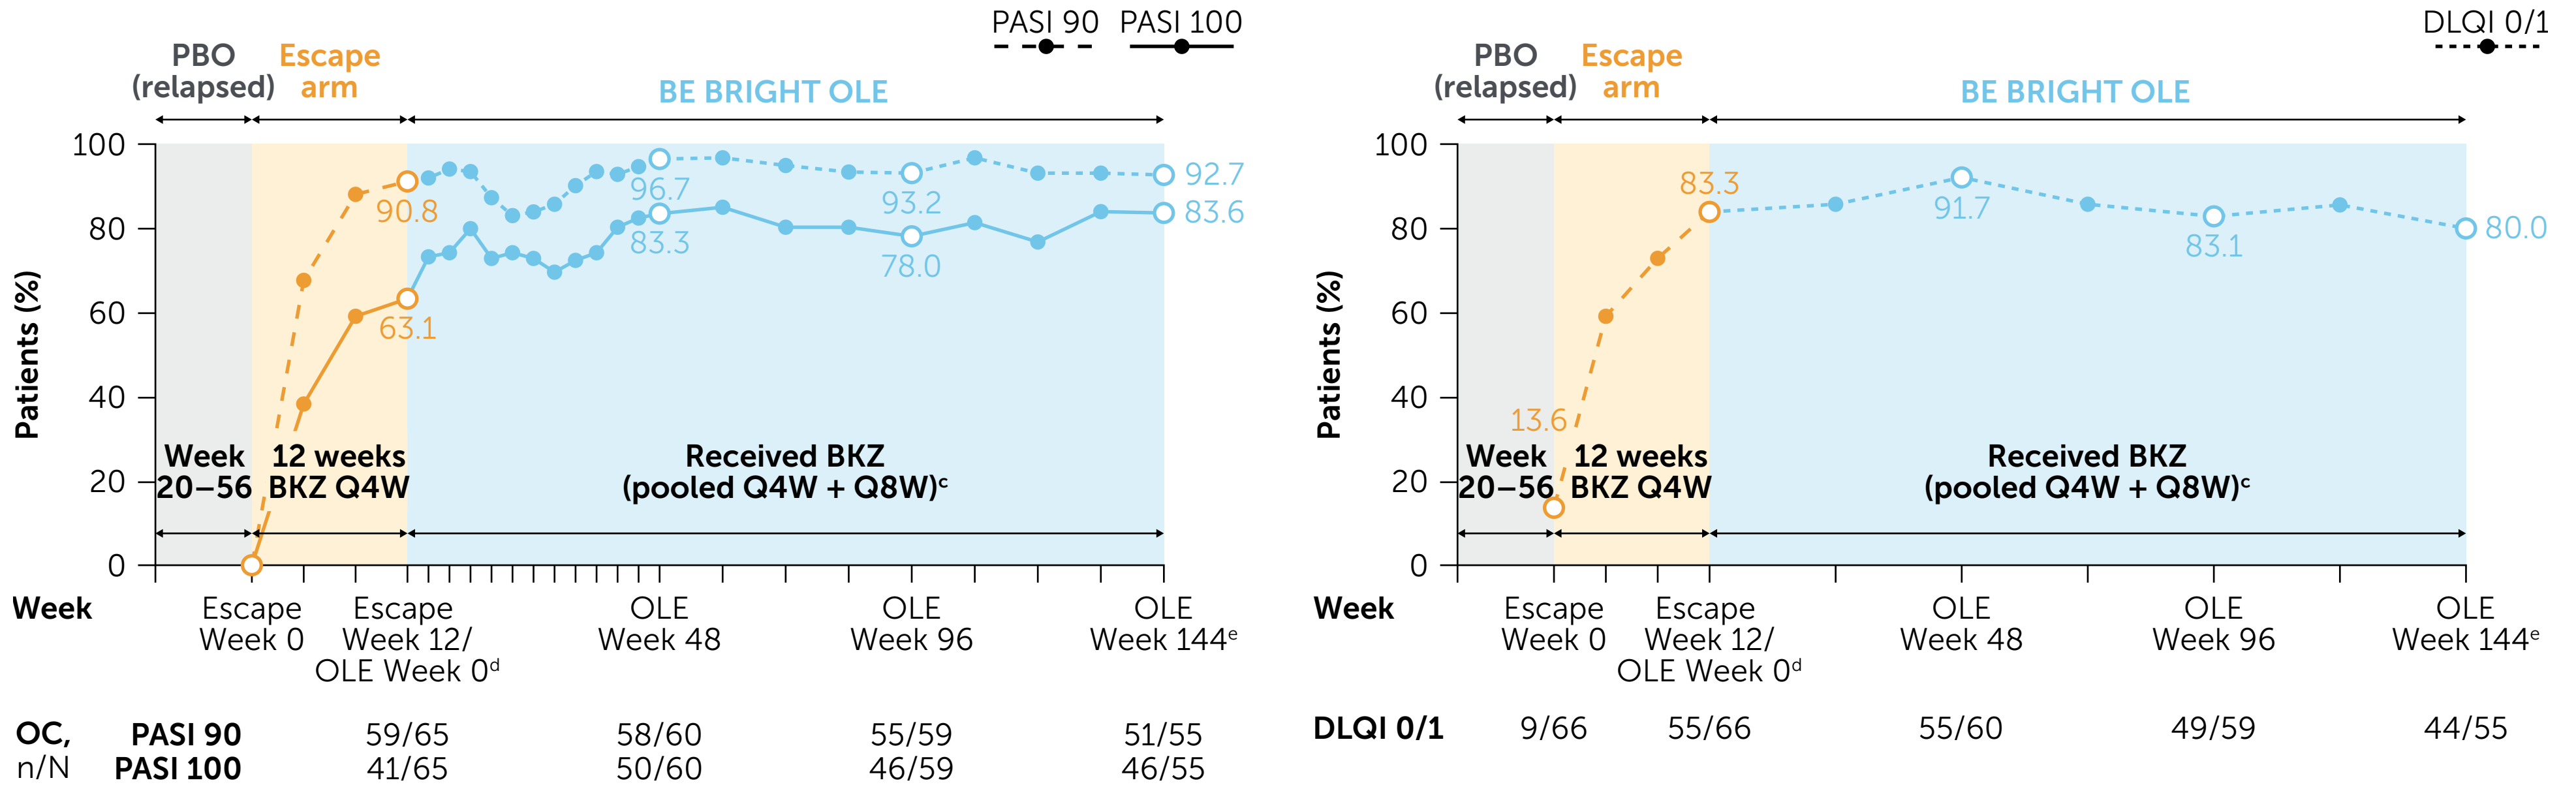

PBO (relapsed)
Escape arm
BE BRIGHT OLE
PASI 90
PASI 100
Patients (%)
100
80
60
40
20
0
90.8
63.1
96.7
83.3
93.2
78.0
92.7
83.6
Week 20–56
12 weeks BKZ Q4W
Received BKZ (pooled Q4W + Q8W)[c]
Week Escape Week 0 Escape Week 12/ OLE Week 0[d] OLE Week 48 OLE Week 96 OLE Week 144[e]
OC, n/N PASI 90 59/65 58/60 55/59 51/55
PASI 100 41/65 50/60 46/59 46/55
DLQI 0/1
13.6
83.3
91.7
83.1
80.0
DLQI 0/1 9/66 55/66 55/60 49/59 44/55

[a] 'Relapse' was defined as <PASI 75 response at any visit between Week 20–56; [b] Generally, patients with higher weight (>100 kg [n=25] vs ≤100 kg [n=74]; OR: 2.22 [95% CI: 0.78, 6.30]; nominal p=0.1249), longer disease duration (≥median disease duration [17.17 years; n=50] vs <median disease duration [17.17 years; n=49]; OR: 4.55 [95% CI: 1.78, 11.65]; nominal p=0.0013), and with prior biologic exposure (prior biologic exposure [n=39] vs no prior biologic exposure [n=60]; OR: 7.27 [95% CI: 2.38, 22.15]; nominal p<0.001) had higher relapse rates in the BE READY randomized-withdrawal period. ORs (95% CIs) and p-values are from a stratified CMH test with region as the stratification variable; p-values are nominal and intended for exploratory insight, with an OR >1 indicating a higher relapse rate; [c] Data reported from the BE BRIGHT OLE are pooled for patients who received BKZ 320 mg Q4W and Q8W; [d] Patients in the Escape Group had their OLE Week 0 study assessments at the end of the 12-week escape arm, having achieved PASI 50 at the end of the 12 weeks; 65/66 patients had a PASI measurement recorded at Escape Week 12/OLE Week 0, as 1 patient missed this visit; [e] At Year 4, 11 patients were missing from the Escape Group having discontinued during the OLE (6 due to adverse events, 1 due to lack of efficacy, 1 lost to follow-up, 3 due to consent being withdrawn).

BKZ: bimekizumab; **BSA:** body surface area; **CI:** confidence interval; **CMH:** Cochran-Mantel-Haenszel; **DLQI:** Dermatology Life Quality Index; **IGA:** Investigator's Global Assessment; **OC:** observed case; **OLE:** open-label extension; **OR:** odds ratio; **PASI 50/75/90/100:** ≥50%/≥75%/≥90%/100% improvement from baseline in Psoriasis Area and Severity Index; **PBO:** placebo; **PsA:** psoriatic arthritis; **Q4W:** every 4 weeks; **Q8W:** every 8 weeks; **SD:** standard deviation.

References: [1]Costanzo A et al. Presented at EADV 2023; Poster P2511; [2]Gordon KB et al. Lancet 2021;397:475–86 (NCT03410992); [3]Blauvelt A et al. J Am Acad Dermatol 2025;93:644–53 (NCT03598790); [4]Blauvelt A et al. Presented at AAD 2021; Poster 27380.

Current Affiliations: AB is now affiliated with Department of Dermatology, University of Maryland School of Medicine, Baltimore, MD. **Author Contributions:** Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **CEMG**, **AC**, **KAP**, **DR**, **LP**, **AB**, **KW**, **JMLP**, **JL**, **RW**, **GH**; Drafting of the publication, or reviewing it critically for important intellectual content: **CEMG**, **AC**, **KAP**, **DR**, **LP**, **AB**, **KW**, **JMLP**, **JL**, **RW**, **GH**; Final approval of the publication: **CEMG**, **AC**, **KAP**, **DR**, **LP**, **AB**, **KW**, **JMLP**, **JL**, **RW**, **GH**. **Disclosures: CEMG:** Received honoraria and/or research grants from AbbVie, Almirall, Anaptysbio, Boehringer Ingelheim, Bristol Myers Squibb, Celgene, Dermavant, Eli Lilly and Company, Evelo Bioscience, Galderma, GSK, Inmagene, Janssen, Kyowa Kirin, Novartis, ONO Pharmaceuticals, Sun Pharma, and UCB. **AC:** Received investigator and/or speaker, and/or advisor fees for AbbVie, Almirall, Amgen, Boehringer Ingelheim, Celgene, Eli Lilly and Company, Galderma, Janssen, LEO Pharma, Novartis, Pfizer, Regeneron Pharmaceuticals, Sandoz, Sanofi, and UCB. **KAP:** Received honoraria and/or grants from AbbVie, Acelyrin, Akros, Alumis, Amgen, Arcutis, Bausch Health/Valeant, Boehringer Ingelheim, Bristol Myers Squibb, Can-Fite Biopharma, Celltrion, Concert Pharmaceuticals, Dermavant, Dermira, DiCE Therapeutics, Eli Lilly and Company, Evelo Biosciences, Forbion, Galderma, Horizon Therapeutics, Incyte, Johnson & Johnson, Kymab, Kyowa Hakko Kirin, LEO Pharma, Meiji Seika Pharma, Mitsubishi Pharma, Nimbus Therapeutics, Novartis, Pfizer, Reistone, Sandoz, Sanofi-Aventis/Genzyme, Sun Pharma, Takeda, Tarsus, UCB, and Zai Lab. **DR:** Received honoraria as a consultant for AbbVie, Abcuro, AltruBio, Boehringer Ingelheim, Bristol Myers Squibb, Celgene, Concert, Dermavant, Dermira, Eli Lilly and Company, Incyte, Janssen, Kyowa Kirin, Novartis, Pfizer, Regeneron, Sanofi, Sun Pharma, UCB, and VielaBio; has received research support from AbbVie, Amgen, Bristol Myers Squibb, Celgene, Dermira, Eli Lilly and Company, Galderma, Incyte, Janssen, Merck, Novartis, Pfizer, and Regeneron; has served as a paid speaker for AbbVie, Amgen, Celgene, Eli Lilly and Company, Janssen, Novartis, Pfizer, Regeneron, and Sanofi. **LP:** Received consultancy/speaker's honoraria from and/or participated in trials sponsored by AbbVie, Almirall, Amgen, Biogen, Boehringer Ingelheim, Bristol Myers Squibb, Eli Lilly and Company, Fresenius Kabi, Horizon, Janssen, LEO Pharma, Novartis, Pfizer, Sandoz, STADA, Sun Pharma, and UCB. **AB:** Served as a speaker and received honoraria for Eli Lilly and Company, Janssen, and UCB; served as a scientific adviser and received honoraria for AbbVie, Almirall, Alumis, Amgen, Anaptysbio, Apogee, Arcutis, Bausch Health, Eli Lilly and Company, Incyte, ImageneBio, Janssen, Khanda Therapeutics, LEO Pharma, Lifordi, Mirador, Novartis, Oruka, Pfizer, Protagonist, Re-AIM, Recludix, Regeneron, Sanofi, Sun Pharma, Takeda, UCB, and Zai Lab; owns stock in Oruka. **KW**, **JMLP**, **JL:** Employees and shareholders of UCB. **RW:** Veramed statistical consultant for UCB. **GH:** Received investigator, consultant, advisor, or speaker fees for AbbVie, Amgen, Athenex, Boehringer Ingelheim, Bond Avillion, Bristol Myers Squibb, Castle Biosciences, Celgene, Dermavant, Dermtech, Eli Lilly and Company, Incyte, Janssen, LEO Pharma, MC2, Ortho Dermatologics, Pfizer, Regeneron, Sanofi Genzyme, Sun Pharma, and UCB. **Acknowledgments:** These studies were funded by UCB. We would like to thank the patients and their caregivers in addition to all the investigators and their teams who contributed to these studies. The authors acknowledge Inés Dueñas Pousa, UCB, Madrid, Spain, for publication coordination, Esme Nias, BSc, Costello Medical, London, UK for medical writing support and editorial assistance, Camille Callais, MSc, Costello Medical, London, UK for editorial assistance and the Costello Medical Creative team for design support. All costs associated with development of this presentation were funded by UCB.

To receive a copy of this poster, scan the QR code.

Link expiration: 09 Jan 2027

Presented at Fall Clinical Dermatology Conference 2026 | October 08–11 | Las Vegas, NV

Previously presented at AAD 2026