Bimekizumab efficacy & safety versus risankizumab to 24 weeks in patients with psoriatic arthritis and concomitant moderate-to-severe psoriasis: An exploratory analysis of the BE BOLD study

Richard G. Langley,[1] Alice B. Gottlieb,[2] Mark Lebwohl,[3] E. James Song,[4] Diamant Thaçi,[5] Akimichi Morita,[6] Philip J. Mease,[7] Iain B. McInnes,[8] Laure Gossec,[9] Yoshiya Tanaka,[10] Barbara Ink,[11] Alexander Marten,[12] Jason Coarse,[13] Myriam Manente,[14] Balint Szilagyi,[12] Joseph F. Merola[15]

[1]Division of Clinical Dermatology & Cutaneous Science, Department of Medicine, Dalhousie University, Halifax, NS, Canada; [2]Department of Dermatology, University of Texas Southwestern Medical Center, Dallas TX, USA; [3]Department of Dermatology, Icahn School of Medicine at Mount Sinai, New York, NY, USA; [4]Frontier Dermatology, Mill Creek, WA, USA; [5]Institute and Comprehensive Center for Inflammation Medicine, University of Lübeck, Lübeck, Germany; [6]Department of Geriatric and Environmental Dermatology, Nagoya City University Graduate School of Medical Sciences, Nagoya, Japan; [7]Department of Rheumatology, Providence-Swedish Medical Center and University of Washington, Seattle, WA, USA; [8]College of Medical Veterinary and Life Sciences, University of Glasgow, Glasgow, UK; [9]Sorbonne Universite and Pitie-Salpetriere Hospital, Paris, France; [10]Department of Molecular Targeted Therapeutics, University of Occupational and Environmental Health, Kitakyushu, Japan; [11]UCB, Slough, UK; [12]UCB, Monheim am Rhein, Germany; [13]UCB, Morrisville, NC, USA; [14]UCB, Braine-l'Alleud, Belgium; [15]Department of Dermatology and Department of Medicine, Division of Rheumatology, UT Southwestern Medical Center, Dallas, TX, USA.

Objective

To report head-to-head efficacy and safety of bimekizumab (BKZ) and risankizumab (RZB) at approved doses in patients with active psoriatic arthritis (PsA) and concomitant moderate-to-severe psoriasis (PSO) to 24 weeks.

Synopsis

- BKZ, a selective inhibitor of interleukin (IL)-17A and IL-17F, and RZB, an IL-23 inhibitor, are both approved treatments that have demonstrated efficacy and tolerability in PsA and PSO.[1–4]
- BKZ showed superiority for the joint-focused primary endpoint (≥50% improvement from baseline in American College of Rheumatology response criteria [ACR50]) compared with RZB at Week 16 (49.1% vs 38.0%, p=0.0059), and comparable safety in patients with PsA in the whole study analysis of BE BOLD (including patients of all PSO severities).[5]
- Patients with PsA and both joint and significant skin involvement present with higher disease burden and worsened quality of life than those with joint and no PSO symptoms.[6]

Methods

- BE BOLD (NCT06624228) was a multicenter, phase 3b, randomized, double-blinded, active comparator-controlled, parallel group, 24-week study in patients aged ≥18 with active PsA who were biologic disease-modifying antirheumatic drug-naïve or had prior inadequate response or intolerance to a maximum of one tumor necrosis factor inhibitor.
- Patients were randomized 1:1 to receive subcutaneous BKZ or RZB to 24 weeks at approved doses, per the European Medicines Agency/United States Food and Drug Administration labels.[7–10]
- In this exploratory analysis, data for the subgroup of patients with moderate-to-severe PSO (body surface area [BSA] ≥10% and Investigator's Global Assessment [IGA] ≥3 and Psoriasis Area and Severity Index [PASI] ≥12) are reported.
 - These patients received BKZ 320 mg every 4 weeks (Q4W) to Week 16 (final dose in this study; patients then received 320 mg every 8 weeks [Q8W]).
 - RZB was dosed 150 mg and administered at baseline, Week 4, and Week 16, regardless of psoriasis severity.
- The exploratory outcomes reported here are: ACR50, ≥90/100% improvement from baseline in PASI (PASI90/100), ACR50+PASI100, ACR50+PASI90, and PASI percentage change from baseline (CfB).
- Missing data were imputed using non-responder imputation (NRI; binary outcomes) or multiple imputation (MI; continuous oucomes).
- Safety data to the completion of study are reported, including the 12-week safety follow-up period. Treatment-emergent adverse events (TEAEs) were classified using the Medical Dictionary for Regulatory Activities v27.0.

Results

Baseline characteristics

- Of 553 patients randomized at baseline, 277 received BKZ and 276 received RZB.
 - 29/277 (10.5%) BKZ-treated and 31/276 (11.2%) RZB-treated patients had concomitant moderate-to-severe PSO; of these, 26/29 (89.7%) and 26/31 (83.9%), respectively, completed 24 weeks of double-blind treatment.
- Baseline characteristics were broadly comparable between treatment groups, although RZB-treated patients were more likely to be male, had higher proportions of baseline enthesitis, elevated high sensitivity C-reactive protein (hs-CRP), and concomitant conventional synthetic disease-modifying antirheumatic drug (csDMARD) use, compared with BKZ-treated patients (**Table 1**).

Efficacy outcomes

- A higher proportion of patients with PsA and moderate-to-severe PSO receiving BKZ compared with RZB achieved ACR50 as rapidly as Week 4, increasing further to Week 16 and sustained to Week 24 (**Figure**).
- PASI100 response rates were higher for patients treated with BKZ than with RZB at Week 4 and Week 24; higher PASI90 response rates and mean percentage CfB in PASI were observed with BKZ than with RZB across all timepoints (**Figure**).
- ACR50+PASI100 and ACR50+PASI90 responses were generally comparable between BKZ and RZB at Week 4; however, by Week 24, response rates were higher with BKZ than with RZB (**Figure**).

Safety

- 26/29 (89.7%) BKZ-treated and 27/31 (87.1%) RZB-treated patients completed the 12-week safety follow-up.
- In this exploratory subgroup analysis, safety was comparable between treatment arms and no new safety signals were identified for BKZ (**Table 2**).
 - Study discontinuations due to TEAEs were low (BKZ: n=1; RZB: n=0).
 - Of the safety topics of interest, cases of fungal infections (BKZ: n=2; RZB: n=0), neutropenia (BKZ: n=1; RZB: n=0), and hepatic events (BKZ: n=2; RZB: n=2) were low across treatment arms.
 - There were no deaths or cases of serious infections, *Candida* infections, active tuberculosis, malignancy, anaphylaxis, hypersensitivity, inflammatory bowel disease, suicidal ideation and behavior, or cardiovascular events for both treatment arms.

Conclusions

In this exploratory subgroup analysis, greater response rates were achieved with bimekizumab than risankizumab in patients with psoriatic arthritis and concomitant moderate-to-severe psoriasis as rapidly as Week 4, increasing further to Week 16 and sustained to Week 24. Safety was comparable between treatment arms and no new safety signals were identified for bimekizumab.

These findings suggest bimekizumab may provide more comprehensive control of inflammation than IL-23 inhibition with risankizumab in patients with psoriatic arthritis and significant skin involvement, potentially due to the inhibition of both IL-23-dependent and IL-23-independent IL-17A/F pathways.

Summary

In the **BE BOLD** study, **bimekizumab (dual IL-17A/IL-17F inhibitor)** was **superior** to **risankizumab (IL-23 inhibitor)** for the **primary joint** efficacy endpoint, **ACR50**, at **Week 16** for **all** patients with **psoriatic arthritis**.[5]

In patients with PsA and concomitant moderate-to-severe psoriasis:[a]

Greater improvements were observed for **bimekizumab-treated[b]** versus **risankizumab-treated[c]** patients as **rapidly** as **Week 4** across all outcomes.

Numerically higher response rates were observed for **bimekizumab-treated[b]** versus **risankizumab-treated[c]** patients at Week 24.

	BKZ	RZB
ACR50	65.5%	41.9%
PASI100	58.6%	45.2%
ACR50+PASI100	44.8%	25.8%
ACR50+PASI90	62.1%	38.7%
PASI90	79.3%	71.0%

Safety profiles were **comparable** between treatment arms, with **no new safety signals identified** for **bimekizumab**

These findings suggest that **bimekizumab** may provide **more comprehensive control of inflammation** than IL-23 inhibition with risankizumab in patients with PsA and **significant skin involvement**, potentially due to the inhibition of both **IL-23-dependent** and **IL-23-independent IL-17A/F pathways**.

NRI data reported. **[a]** Defined as BSA ≥10% and IGA ≥3 and PASI ≥12; **[b]** BKZ 320 mg Q4W/Q8W; **[c]** RZB 150 mg at baseline, Week 4, and Week 16.

Table 1 Baseline characteristics in patients with PsA and concomitant moderate-to-severe PSO

	BKZ 320 mg[a] n=29	RZB 150 mg[b] n=31
Age, years, mean (SD)	49.0 (13.8)	47.4 (11.1)
Sex, male, n (%)	17 (58.6)	23 (74.2)
BMI, kg/m², mean (SD)	29.6 (6.1)	29.7 (5.1)
Tender joint count, mean (SD)	21.8 (13.4)	17.6 (12.5)
Swollen joint count, mean (SD)	12.8 (5.8)	11.2 (8.1)
Patient Global Assessment-PsA, mean (SD)	65.5 (19.6)	64.8 (19.6)
PASI, mean (SD)	21.8 (9.7)	19.4 (9.4)
hs-CRP ≥6 mg/L, n (%)	9 (31.0)	16 (51.6)
Any csDMARD at baseline,[c] n (%)	14 (48.3)	22 (71.0)
Enthesitis, Leeds Enthesitis Index >0, n (%)	11 (37.9)	20 (64.5)
Dactylitis, Leeds Dactylitis Index >0, n (%)	5 (17.2)	4 (12.9)

Randomized set. Moderate-to-severe PSO defined as BSA ≥10% and IGA ≥3, and PASI ≥12. **[a]** BKZ 320 mg Q4W/Q8W. Final dose administered at Week 16; **[b]** Dosed at baseline, Week 4, and Week 16; **[c]** Includes methotrexate, sulfasalazine, and leflunomide.

Figure Efficacy outcomes to Week 24 in patients with PsA and concomitant moderate-to-severe PSO (NRI, MI)

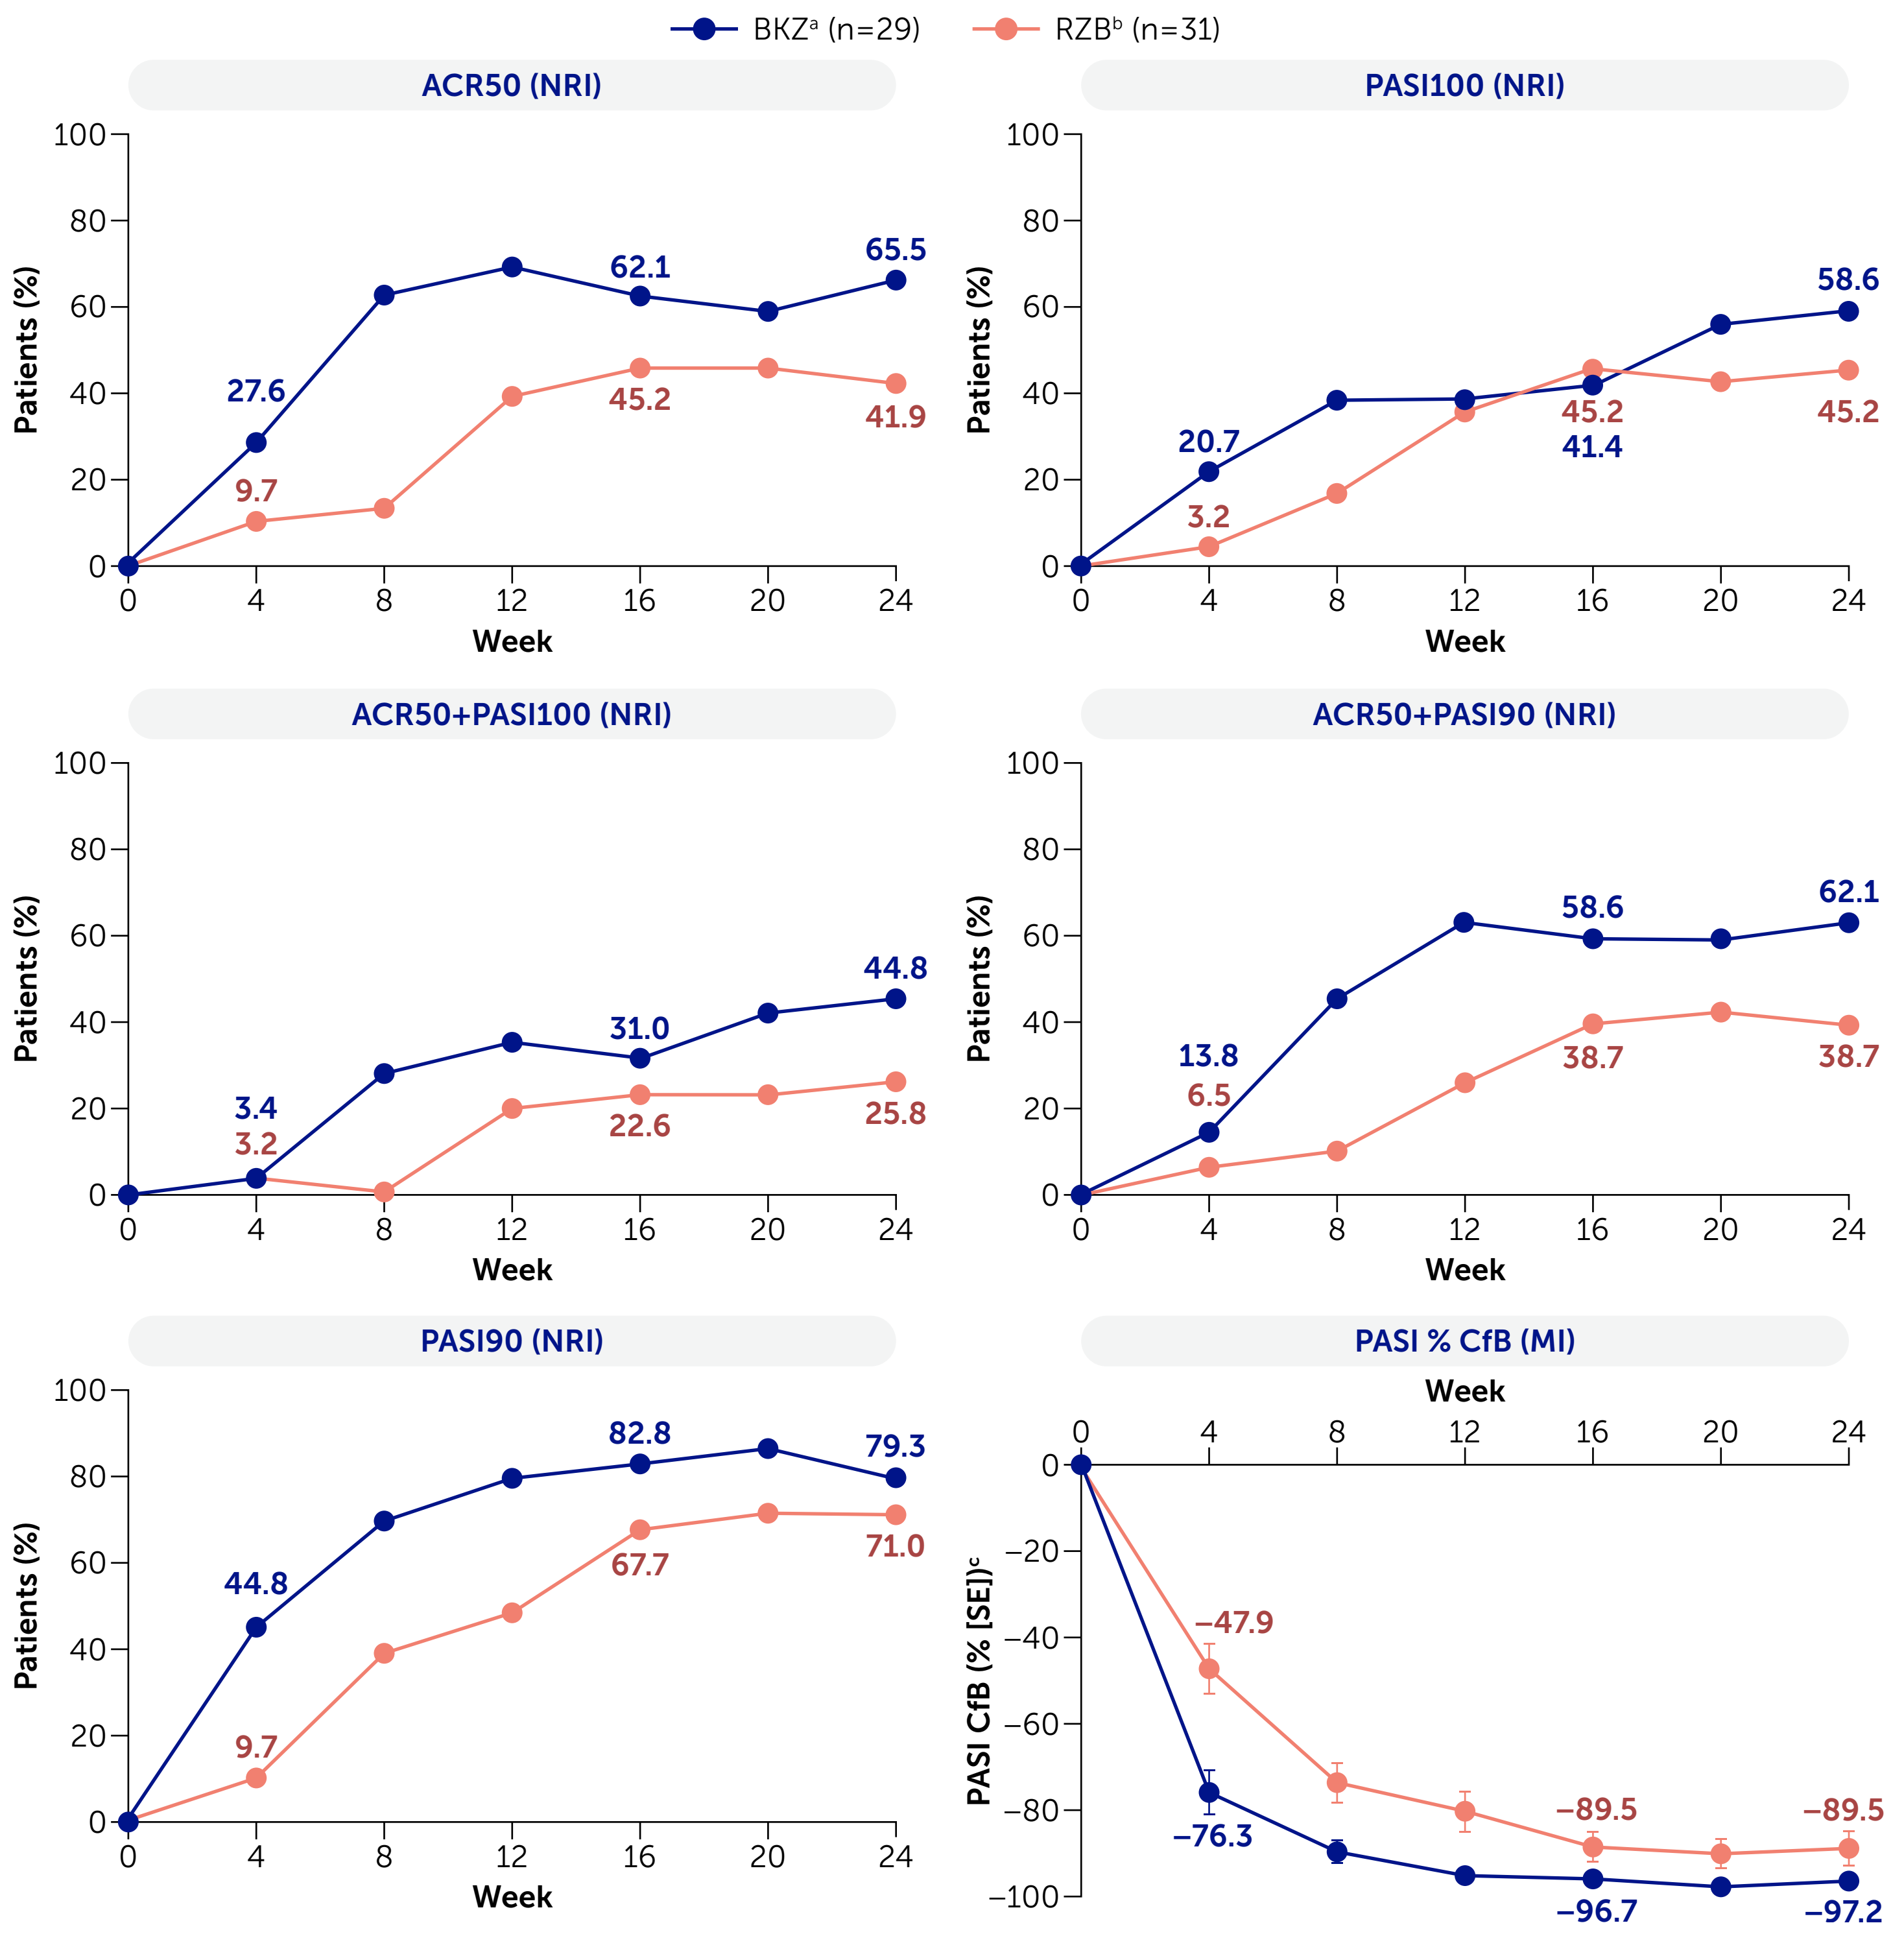
BKZ[a] (n=29)
RZB[b] (n=31)
ACR50 (NRI)
PASI100 (NRI)
ACR50+PASI100 (NRI)
ACR50+PASI90 (NRI)
PASI90 (NRI)
PASI % CfB (MI)
Patients (%)
PASI CfB (% [SE])[c]
Week
0 4 8 12 16 20 24
27.6
9.7
62.1
45.2
65.5
41.9
20.7
3.2
45.2
41.4
58.6
45.2
3.4
3.2
31.0
22.6
44.8
25.8
13.8
6.5
58.6
38.7
62.1
38.7
44.8
9.7
82.8
67.7
79.3
71.0
−47.9
−76.3
−89.5
−96.7
−89.5
−97.2

Randomized set. Moderate-to-severe PSO defined as BSA ≥10% and IGA ≥3 and PASI ≥12. **[a]** BKZ 320 mg Q4W/Q8W. Final dose administered at Week 16; **[b]** Dosed at baseline, Week 4, and Week 16; **[c]** SE data for BKZ treatment were negligible between Weeks 12 and 24.

Table 2 Safety in patients with PsA and concomitant moderate-to-severe PSO

n (%)	BKZ 320 mg[a] n=29; 16.8 PY	RZB 150 mg[b] n=31; 17.2 PY
Any TEAE	15 (51.7)	13 (41.9)
Serious TEAEs	0	1 (3.2)[c]
Study discontinuations due to TEAEs	1 (3.4)[d]	0
Severe TEAEs	1 (3.4)[e]	0
Deaths	0	0
Most common TEAEs[f]		
Nasopharyngitis	3 (10.3)	3 (9.7)
Upper respiratory tract infection	2 (6.9)	1 (3.2)
Alanine aminotransferase increased	1 (3.4)	2 (6.5)
Aspartate aminotransferase increased	1 (3.4)	2 (6.5)
Gamma-glutamyltransferase increased	1 (3.4)	2 (6.5)
Blood triglycerides increased	2 (6.9)	0
Diarrhea	0	2 (6.5)
Osteoarthritis	0	2 (6.5)
Safety topics of interest		
Serious infections	0	0
Opportunistic infections	0	0
Fungal infections	2 (6.9)[g]	0
Candida infections	0	0
Oral candidiasis	0	0
Malignancies	0	0
Neutropenia	1 (3.4)	0
Anaphylactic reaction	0	0
Hypersensitivity	0	0
Injection site reactions	0	0
Inflammatory bowel disease[h]	0	0
Suicidal ideation and behavior[i]	0	0
Cardiovascular events[j]	0	0
Hepatic events	2 (6.9)	2 (6.5)

Safety set. Safety is reported to study completion, including the 12-week safety follow-up period; as participants were permitted to use any other medications once 28 days had passed from their final study dose, participants may have continued or discontinued the same treatment or switched to another treatment within the safety follow-up period. TEAEs were classified using the Medical Dictionary for Regulatory Activities v27.0. Moderate-to-severe PSO defined as BSA ≥10% and IGA ≥3 and PASI ≥12. **[a]** BKZ 320 mg Q4W/Q8W. Final dose administered at Week 16; **[b]** Dosed at baseline, Week 4, and Week 16; **[c]** One reported case of osteoarthropathies, deemed not related to RZB treatment; **[d]** Patient discontinued BKZ due to erythema; **[e]** One reported case of erythema; **[f]** TEAEs occurring in >5% of either group, ordered from most to least common; **[g]** One case of oral fungal infection and one case of tongue fungal infection; **[h]** No cases of adjudicated inflammatory bowel disease; **[i]** No cases of adjudicated suicidal ideation and behavior; **[j]** No cases of adjudicated MACE.

ACR50: ≥50% improvement from baseline in American College of Rheumatology response criteria; **BKZ:** bimekizumab; **BL:** baseline; **BMI:** body mass index; **BSA:** body surface area; **CfB:** change from baseline; **csDMARD:** conventional synthetic disease-modifying antirheumatic drug; **hs-CRP:** high-sensitivity C-reactive protein; **IGA:** Investigator's Global Assessment; **IL:** interleukin; **MACE:** major adverse cardiovascular event; **MI:** multiple imputation; **NRI:** non-responder imputation; **PASI:** Psoriasis Area and Severity Index; **PASI90/100:** ≥90/100% improvement from baseline in Psoriasis Area and Severity Index; **PsA:** psoriatic arthritis; **PSO:** psoriasis; **PY:** patient-years; **Q4W/Q8W:** every 4/8 weeks; **RZB:** risankizumab; **SD:** standard deviation; **SE:** standard error; **TEAE:** treatment-emergent adverse event.

References: [1]Gossec L. Rheumatology (Oxford) 2026:keag118; [2]Kristensen LE. Rheumatol Ther 2024;11:617–32; [3]Östör A. Rheumatol Ther 2024;11:633–48; [4]Armstrong A. Dermatol Ther (Heidelb) 2022;12:1777–92; [5]McInnes IB. Presented at EADV 2026 (P2807); [6]Walsh JA. Joint Bone Spine 2023;90:105534; [7]EMA Summary of Product Characteristics. Bimzelx. Available from: https://www.ema.europa.eu/en/documents/product-information/bimzelx-epar-product-information_en.pdf. Accessed: September 2026; [8]EMA Summary of Product Characteristics. Skyrizi. Available from: https://www.ema.europa.eu/en/documents/product-information/skyrizi-epar-product-information_en.pdf. Accessed: September 2026; [9]Prescribing Information. BIMZELX®. Available from: https://www.accessdata.fda.gov/drugsatfda_docs/label/2023/761151s000lbl.pdf. Accessed: September 2026; [10]FDA Prescribing Information. SKYRIZI®. Available from: https://www.accessdata.fda.gov/drugsatfda_docs/label/2022/761262s000lbl.pdf. Accessed: September 2026.
Author Contributions: Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **RGL**, **ABG**, **ML**, **EJS**, **DT**, **AMo**, **PJM**, **IBM**, **LG**, **YT**, **BI**, **AMa**, **JC**, **MM**, **BS**, **JFM**; Drafting of the publication, or reviewing it critically for important intellectual content: **RGL**, **ABG**, **ML**, **EJS**, **DT**, **AMo**, **PJM**, **IBM**, **LG**, **YT**, **BI**, **AMa**, **JC**, **MM**, **BS**, **JFM**; Final approval of the publication: **RGL**, **ABG**, **ML**, **EJS**, **DT**, **AMo**, **PJM**, **IBM**, **LG**, **YT**, **BI**, **AMa**, **JC**, **MM**, **BS**, **JFM**. **Author Disclosures: RGL:** Principal investigator for AbbVie, Amgen, Boehringer Ingelheim, Celgene, Eli Lilly and Company, LEO Pharma, Merck, Novartis, Pfizer, and UCB; served on scientific advisory boards for AbbVie, Amgen, Boehringer Ingelheim, Celgene, Eli Lilly and Company, LEO Pharma, Merck, Novartis, Pfizer, and UCB; provided lectures for AbbVie, Amgen, Celgene, Eli Lilly and Company, LEO Pharma, Merck, Novartis, and Pfizer. **ABG:** Received research/educational grants from BMS, Janssen, Moonlake, and UCB (all paid to Mount Sinai School of Medicine until May 1, 2025); sub I at UTSW on studies from BMS and Janssen; PI on a BMS study; received honoraria as an advisory board member and consultant for Amgen, AbbVie (terminated July 2026), AstraZeneca, Biogen, BMS, Eli Lilly, Janssen, Novartis, Oruka, Sanofi, SunPharma, Takeda, Teva, and UCB. **ML:** Employee of Mount Sinai; receives research funds from AbbVie, Arcutis, Avotres, Boehringer Ingelheim, Cara Therapeutics, Clexio, Dermavant, Eli Lilly and Company, Incyte, Inozyme, Johnson & Johnson, Pfizer, Sanofi-Regeneron, and UCB; consultant for Aikium, Almirall, AltruBio, Amgen, Apogee, Arcutis, AstraZeneca, Atomwise, Avotres Therapeutics, Boehringer Ingelheim, BMS, Castle Biosciences, Celltrion, CorEvitas, Dermavant, Dermsquared, Evommune, Facilitation of International Dermatology Education, Forte Biosciences, Galderma, Genentech, Incyte, LEO Pharma, Mayne Pharmaceuticals, Meiji Seika Pharma, Mindera, Mirium Pharmaceuticals, Oruka, Pfizer, Sanofi-Regeneron, Revolo, Seanergy, Strata, Sun Pharma, Takeda, Trevi, and Verrica. **EJS:** Consultant/promotional speaker for AbbVie, Alumis, Alphyn, Amgen, Apogee, Arcutis, BMS, Boehringer Ingelheim, Dermavant/Organon, DermBiont, Galderma, Incyte, Johnson & Johnson, LEO Pharma, Novartis, Ortho Dermatologics, Pfizer, Phothera, Sanofi, Regeneron, Sun Pharma, Takeda, and UCB; investigator for AbbVie, Alumis, Amgen, Apogee, Arcutis, BMS, DermBiont, Incyte, Johnson & Johnson, LEO Pharma, MoonLake Immunotherapeutics, Pfizer, Sanofi, Sun Pharma, TIMBER Pharmaceuticals, Veradermics, and Zura Bio; board member for International Psoriasis Council and National Psoriasis Foundation Medical Advisory Board; senior editor for the Journal of Psoriasis and Psoriatic Arthritis. **DT:** Investigator and/or consultant/advisor for AbbVie, Almirall, Amgen, BMS, Boehringer Ingelheim, Celltrion, Eli Lilly and Company, Galderma, Incyte, Johnson & Johnson, Kyowa Kirin, LEO Pharma, L'Oreal, New Bridge, Novartis, Pfizer, Regeneron, Samsung, Sanofi, Stada, Sun Pharma, Takeda, Target-RWE, UCB, and Vichy; received grants from AbbVie, LEO Pharma, and Novartis. **AMo:** Research grants, consulting fees, and/or speaker's fees from AbbVie, Boehringer Ingelheim, BMS, Celgene, Eisai, Eli Lilly and Company, Janssen, Kyowa Hakko Kirin, LEO Pharma, Maruho, Mitsubishi Tanabe Pharma, Nichi-Iko, Nippon Kayaku, Novartis, Pfizer, Sun Pharma, Taiho Pharmaceutical, Torii Pharmaceutical, UCB, and Ushio. **PJM:** Research grants from AbbVie, Acelyrin, Amgen, BMS, Eli Lilly and Company, Johnson & Johnson Innovative Medicine, Novartis, Pfizer Inc., Sana, and UCB; consulting fees from AbbVie, Acelyrin, Amgen, BMS, Century, Cullinan, Eli Lilly and Company, Inmagene, Johnson & Johnson Innovative Medicine, MoonLake Immunotherapeutics, Novartis, Pfizer Inc., Spyre, Takeda, and UCB; speakers bureau fees from AbbVie, Amgen, Eli Lilly and Company, Johnson & Johnson Innovative Medicine, Novartis, Pfizer Inc., and UCB. **IBM:** Consulting fees and honoraria from AbbVie, AstraZeneca, Boehringer Ingelheim, BMS, Cabaletta, Causeway Therapeutics, Celgene, Eli Lilly and Company, Janssen, MoonLake Immunotherapeutics, Montai, Novartis, Pioneering Medicines, and UCB; research support from Boehringer Ingelheim, BMS, Celgene, Janssen, Novartis, and UCB. **LG:** Research grants: AbbVie, Lilly, Novartis, and UCB; consulting fees from AbbVie, AlfaSigma, Amgen, BMS, Celltrion, J&J, Lilly, Moonlake, Novartis, Nordic Pharma, Oruka, Pfizer, Stada, Takeda, and UCB; non-financial support from AbbVie, Amgen, Biogen, BMS, Celltrion, J&J, Lilly, Pfizer, and UCB. **YT:** Speaker fees and/or honoraria from AbbVie, AstraZeneca, Biogen, Boehringer Ingelheim, BMS, Chugai, Eisai, Eli-Lilly, Gilead, GSK, Novartis, Otsuka, Sandoz, Sanofi, Taisho, Tanabe, and UCB; research grants from Asahi Kasei. **BI:** Shareholder of AbbVie, GSK, and UCB. Employee of UCB. **AMa**, **JC**, **MM**, **BS:** Shareholders and employees of UCB. **JFM:** Consultant and/or investigator for AbbVie, Amgen, AstraZeneca, Biogen, BMS, Boehringer Ingelheim, Eli Lilly, Galderma, Janssen, MoonLake, Novartis, Oruka, Pfizer, Regeneron, Sanofi, Sun Pharma, and UCB. **Acknowledgments:** This study was funded by UCB. We thank the patients and their caregivers in addition to the investigators and their teams who contributed to this study. The authors acknowledge Lyes Derouiche, PhD, CMPP, of UCB, for publication coordination, Zahraa Omran, MSci, of Costello Medical, Bristol, for medical writing and editorial assistance, and the Costello Medical Creative team for graphic design assistance. All costs associated with development of this poster were funded by UCB.

To receive a copy of this poster, scan the QR code.

Link expiration: January 09 2027

Presented at Fall Clinical Dermatology Conference 2026 | October 8–11 | Las Vegas, NV