Sex-specific and age-adjusted prevalence of hidradenitis suppurativa: Results from Komodo US claims data suggest sex parity of HS symptoms

Hadar Lev-Tov,[1] John R. Ingram,[2] Jennifer Hsiao,[3] Maria A. Aleshin,[4] Barry M. McGrath,[5] Shawn Rajan,[6] Nersi Alaeddin,[7] 'Matladi N. Ndlovu,[8] Amit Garg[9]

[1]Dr. Phillip Frost Department of Dermatology and Cutaneous Surgery, University of Miami Miller School of Medicine, Miami, FL, USA; [2]Division of Infection and Immunity, Cardiff University, Cardiff, UK; [3]Department of Dermatology, University of Southern California, Los Angeles, CA, USA; [4]Department of Dermatology, Stanford University School of Medicine, Stanford, CA, USA; [5]HS Ireland, Hidradenitis Suppurativa Association, County Clare, Ireland and patient author; [6]UCB, Smyrna, GA, USA; [7]UCB, Monheim am Rhein, Germany; [8]UCB, Brussels, Belgium; [9]Northwell, New Hyde Park, NY, USA

Objective

To report the sex-specific, age-adjusted annual prevalence of diagnosed hidradenitis suppurativa (HS) compared with HS-related symptom-defined disease using Komodo Health US claims data.

Background

- HS is a chronic, inflammatory skin disease which has a profound negative impact on patients' health-related quality of life.[1]
- On average, patients with HS experience significant diagnostic delays of 7–10 years from symptom onset, meaning there is a large undiagnosed population.[2]
- HS is traditionally reported to be approximately three times more prevalent in females than males, based primarily on studies in North America and Europe.[3]
- Improving HS prevalence estimates may support better treatment outcomes.[4]

Methods

- This study is a retrospective, real-world analysis of US secondary healthcare claims data from the Komodo Health database with >300 million patients selected from October 2016 to December 2024 (**Figure 1**).[5]
- Age-adjusted sex-specific annual prevalence data are reported from 2016 to 2024 for two mutually exclusive groups: patients with an ICD-10-based HS diagnosis and those with HS-related symptom-defined disease, coded on or after October 1, 2016.
- HS diagnosis was defined as presence of ICD code L73.2.[6]
- As a proxy for undiagnosed patients, ICD-10 codes for HS symptoms including abscesses, furuncles and pilonidal cysts were used, with the presence of the above-defined claims required on ≥5 distinct dates over the study duration.
 - ICD codes for abscess and furuncle were specifically for HS-prone regions.
 - Patients with ≥5 claims of solely pilonidal cyst were excluded.
 - Patients who received an HS diagnosis during the study period were excluded from the HS-related symptom-defined disease cohort.
 - Age was assessed at the date of the first diagnosis or fifth HS-related symptom claim.
- All analyses are descriptive.

Results

- A total of approximately 2.4 million patients were included in the analysis.
 - Of these, ~1.6 million had a diagnosis of HS and ~0.8 million had HS-related symptom-defined disease (**Figure 2**).
- Between 2017 and 2024, the overall annual prevalence of diagnosed HS ranged between 101 and 166 cases per 100,000 population.

Diagnosed HS

- The prevalence of diagnosed HS was markedly higher among females, and was generally consistent with prevalence estimates reported in the literature (**Figure 3**).[3]
- In 2017, the prevalence was 62 and 148 cases per 100,000 population among males and females, respectively.
- This female predominance persisted through 2024, reaching 97 and 253 cases per 100,000 population among males and females, respectively.

HS symptom-defined disease

- In contrast, HS-related symptom-defined disease (according to predefined ICD-10 codes) exhibited a more balanced sex distribution, with a slight male predominance (**Figure 4**).
- The prevalence of HS symptom-defined disease was 43 and 37 cases per 100,000 population in 2017 among males and females, respectively.
- Of note, sex distribution remained similar to 2024, with 67 and 54 cases per 100,000 population among males and females, respectively.

Conclusions

A similar prevalence of HS-related symptoms in the absence of a formal HS diagnosis was observed between sexes, contrary to current knowledge that HS prevalence is markedly higher among females than males.

These findings may suggest underdiagnosis in males and highlight the need for greater awareness within the healthcare community to reduce diagnostic disparities between male and female patients.

Summary

The **sex-specific, age-adjusted** annual **prevalence** of **diagnosed HS** and **HS-related symptom-defined disease** were reported using Komodo Health US claims data

Diagnosed HS (ICD-10 code L73.2)		HS symptom-defined disease (according to predefined ICD-10 codes)	
97 cases per 100,000	**253** cases per 100,000	**67** cases per 100,000	**54** cases per 100,000

A **similar prevalence** of HS symptom-defined disease was observed in males and females, suggesting that HS may be **underdiagnosed in males**

Data presented are from 2024.

Figure 1 Komodo Health US claims data analysis study design

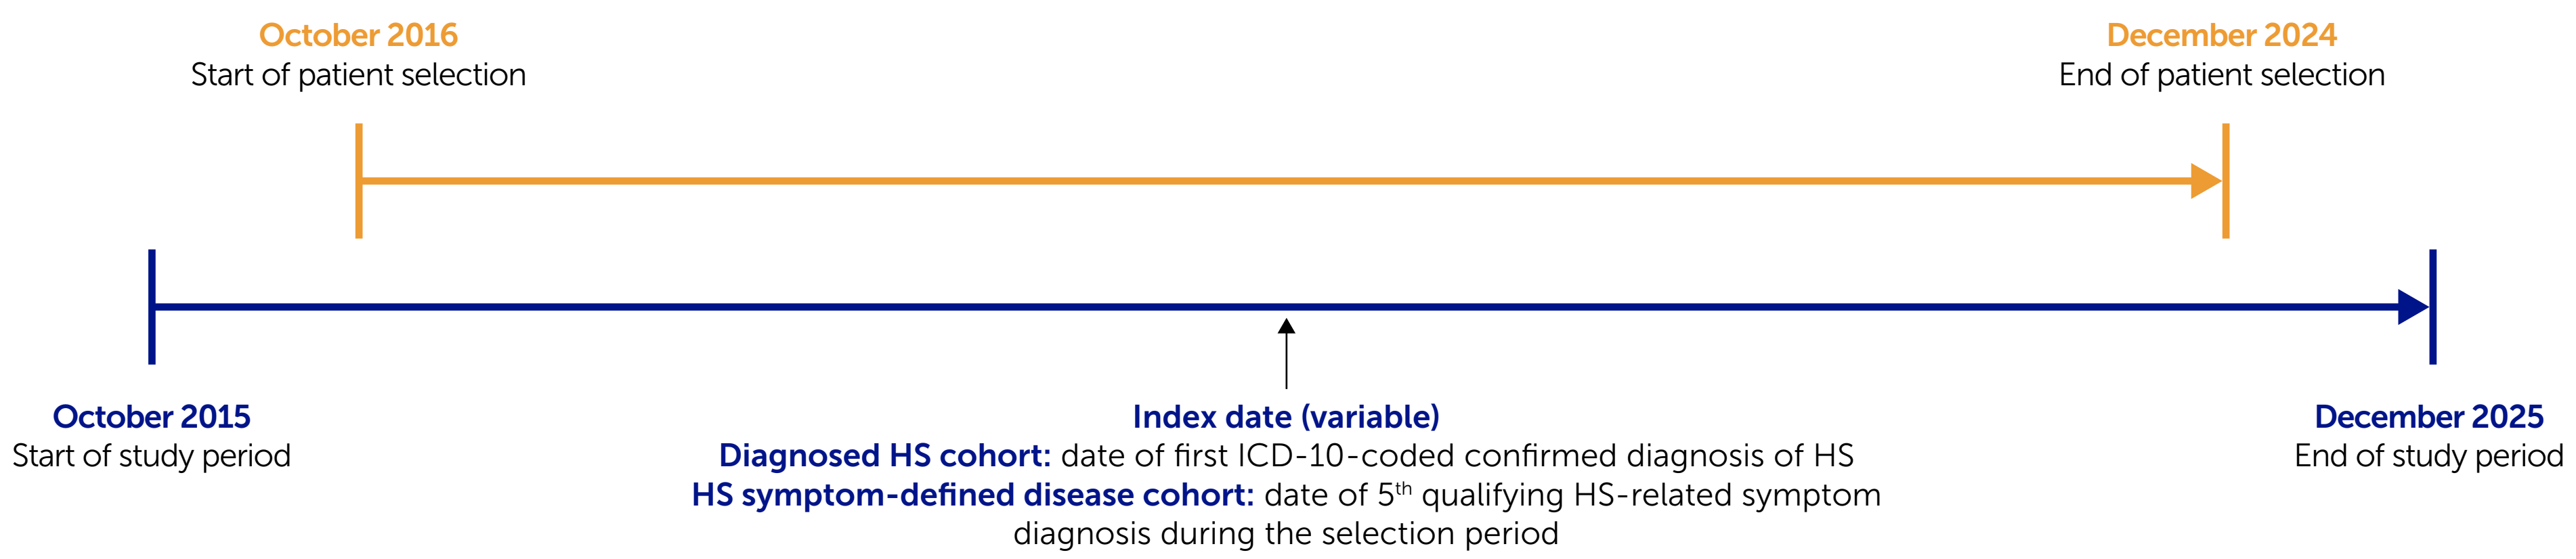
October 2016
Start of patient selection
December 2024
End of patient selection
October 2015
Start of study period
Index date (variable)
Diagnosed HS cohort: date of first ICD-10-coded confirmed diagnosis of HS
HS symptom-defined disease cohort: date of 5th qualifying HS-related symptom diagnosis during the selection period
December 2025
End of study period

Figure 2 Patient selection

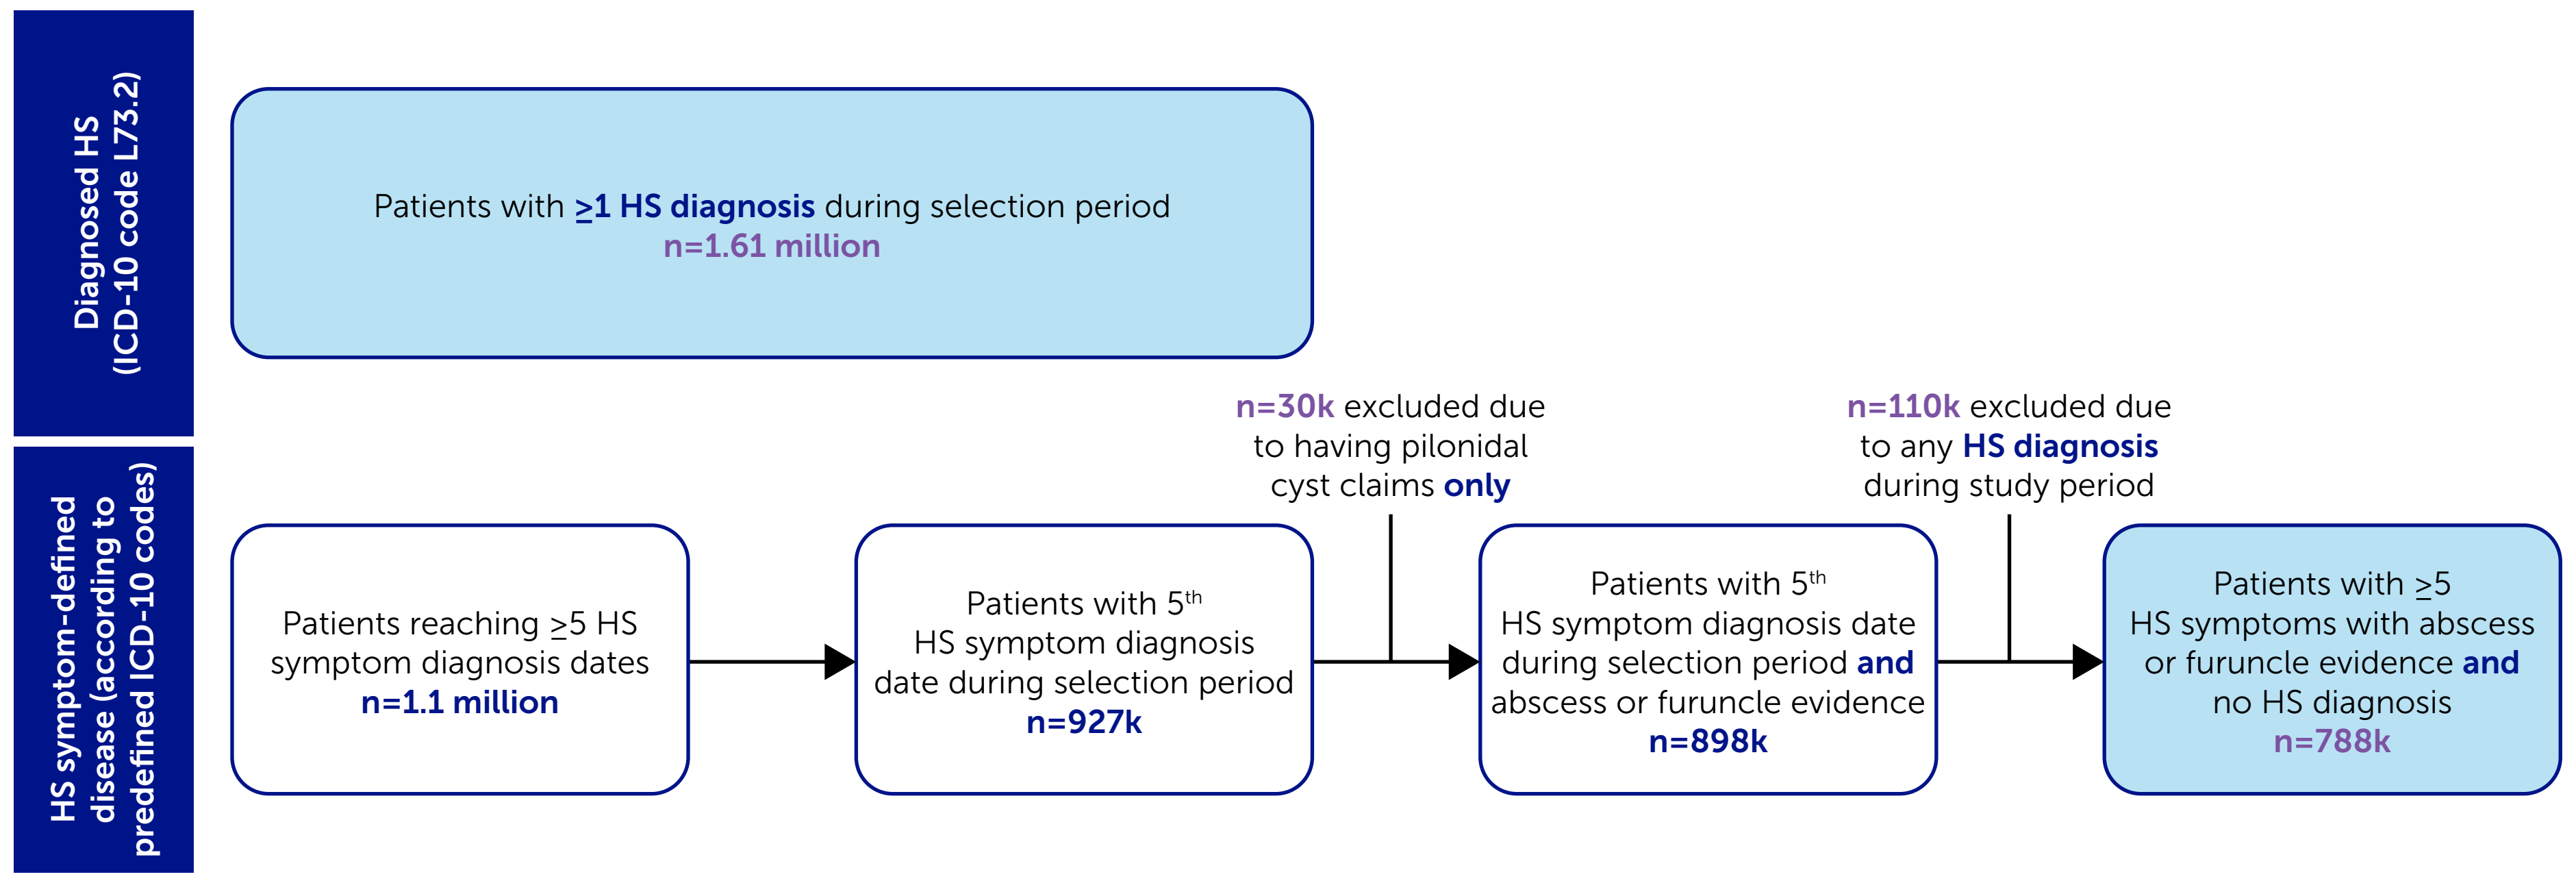
Diagnosed HS (ICD-10 code L73.2)
Patients with ≥1 HS diagnosis during selection period n=1.61 million
HS symptom-defined disease (according to predefined ICD-10 codes)
Patients reaching ≥5 HS symptom diagnosis dates n=1.1 million
Patients with 5th HS symptom diagnosis date during selection period n=927k
n=30k excluded due to having pilonidal cyst claims only
Patients with 5th HS symptom diagnosis date during selection period and abscess or furuncle evidence n=898k
n=110k excluded due to any HS diagnosis during study period
Patients with ≥5 HS symptoms with abscess or furuncle evidence and no HS diagnosis n=788k

HS: hidradenitis suppurativa; ICD: International Classification of Diseases; US: United States.

Figure 3 Sex-specific age-adjusted annual prevalence of diagnosed HS

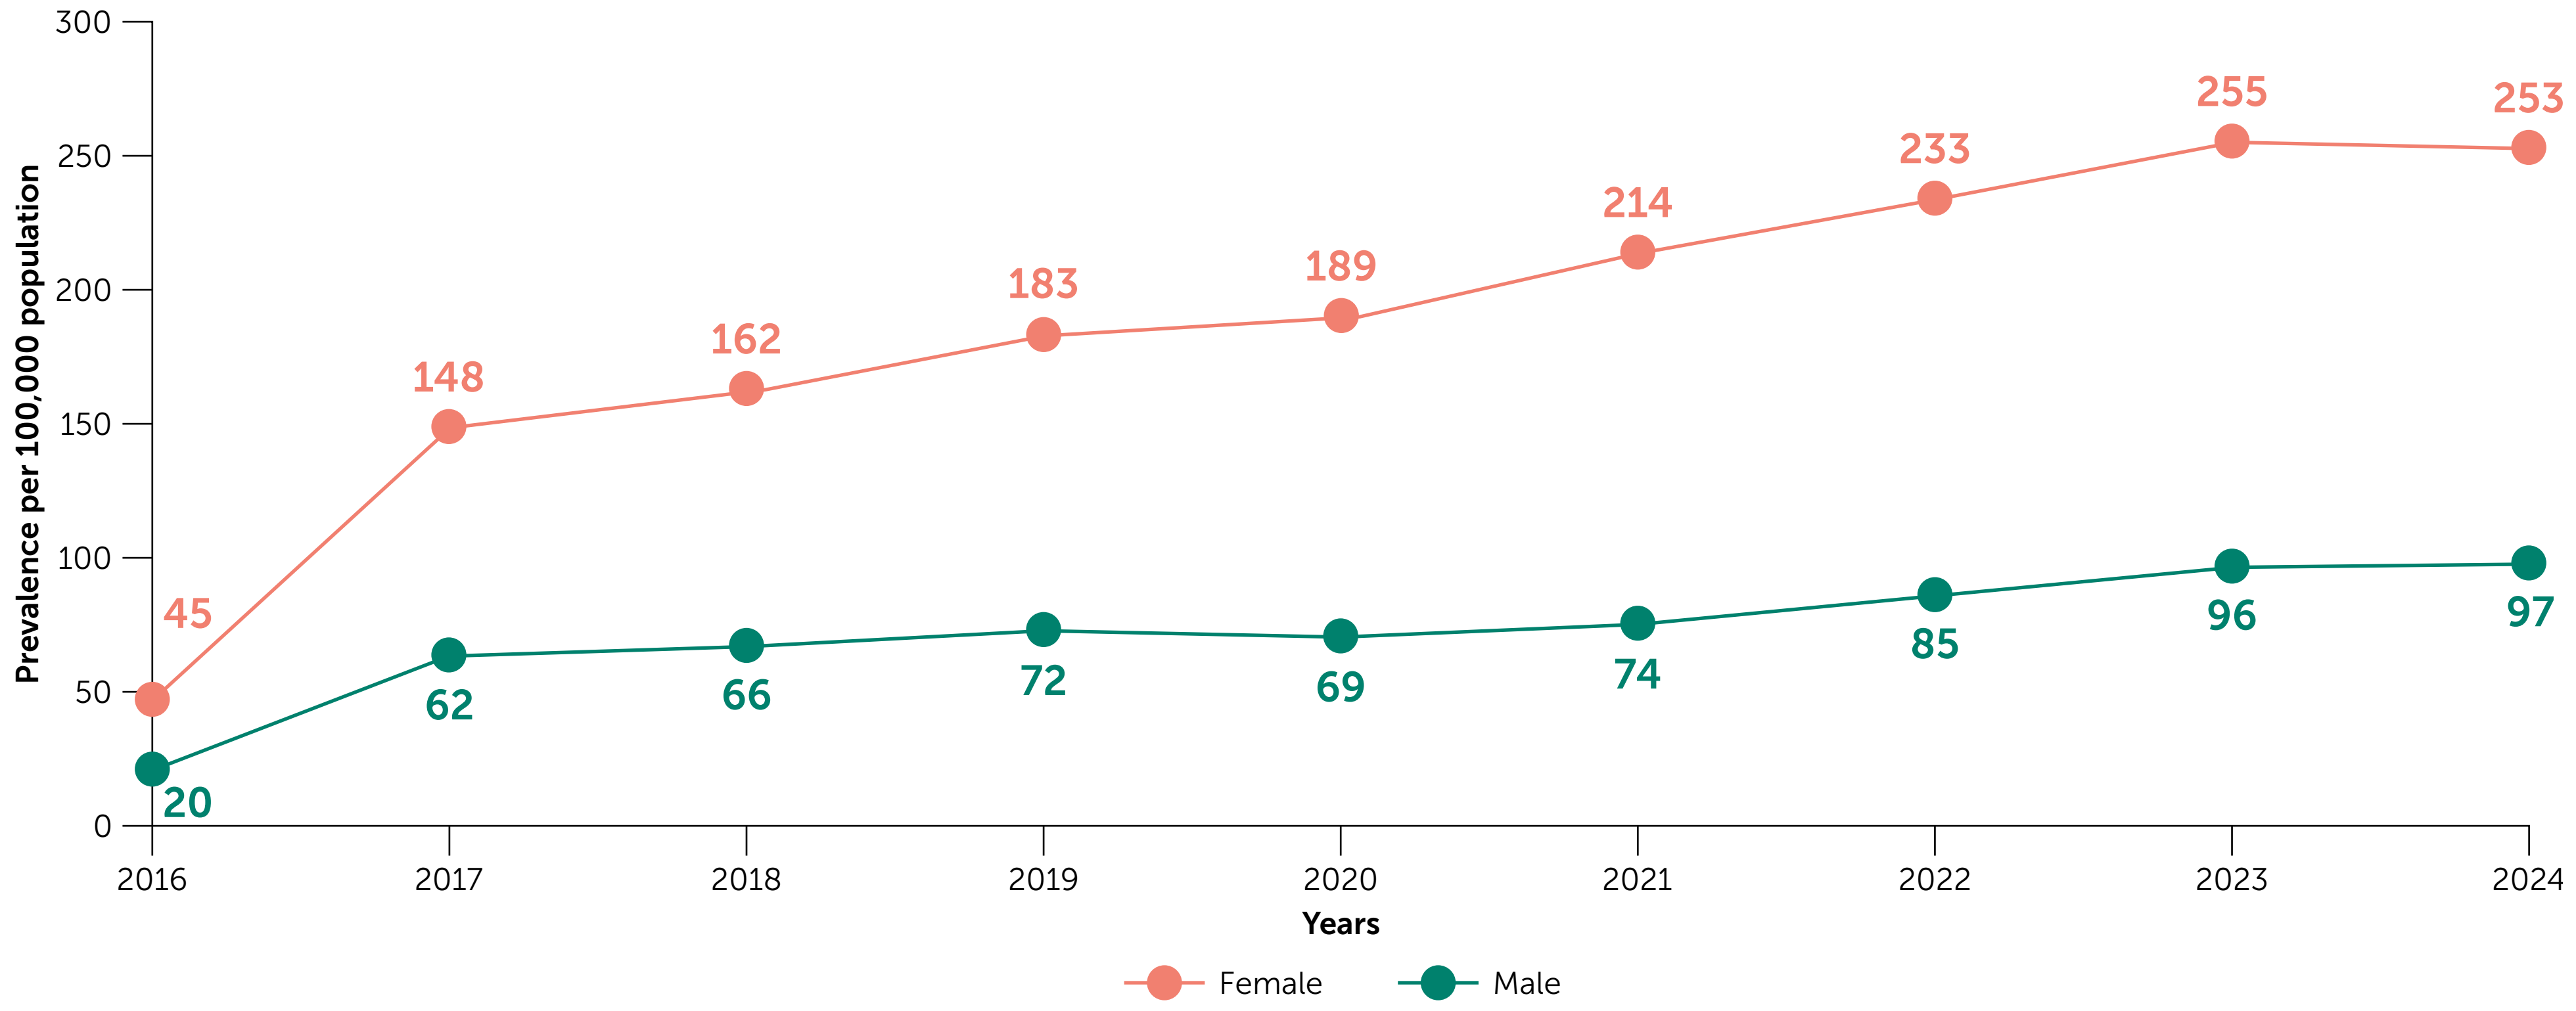
Prevalence per 100,000 population
300
250
200
150
100
50
0
45
148
162
183
189
214
233
255
253
20
62
66
72
69
74
85
96
97
2016
2017
2018
2019
2020
2021
2022
2023
2024
Years
Female
Male

The low prevalence in 2016 is due to the patient selection beginning in October 2016, limiting patient capture during that calendar year.

Figure 4 Sex-specific age-adjusted annual prevalence of HS symptom-defined disease

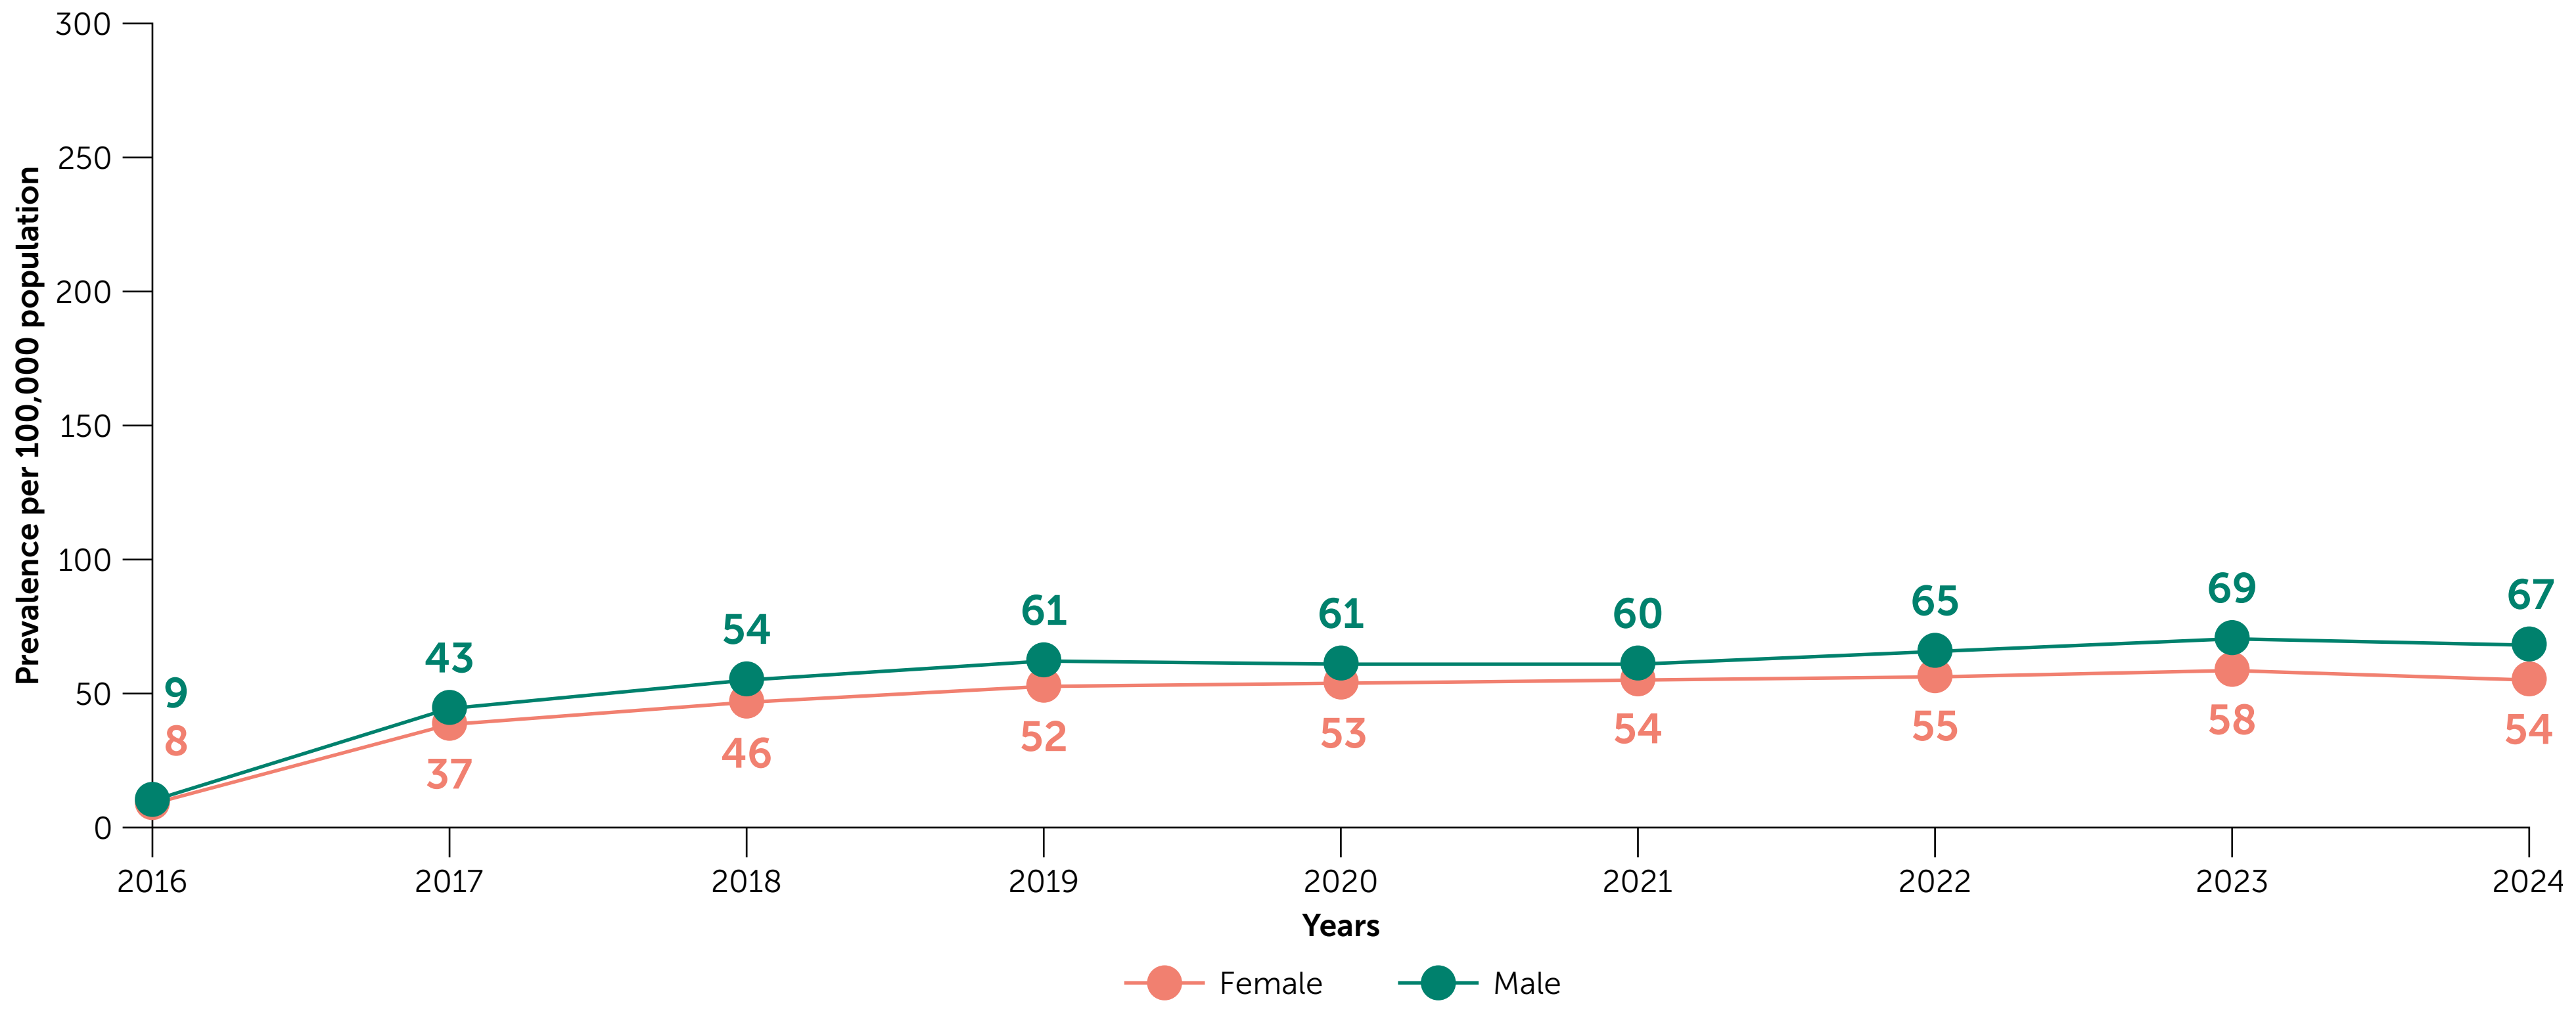
Prevalence per 100,000 population
300
250
200
150
100
50
0
9
43
54
61
61
60
65
69
67
8
37
46
52
53
54
55
58
54
2016
2017
2018
2019
2020
2021
2022
2023
2024
Years
Female
Male

The low prevalence in 2016 is due to the patient selection beginning in October 2016, limiting patient capture during that calendar year.

References: [1]Kimball AB et al. Dermatol Ther (Heidelb) 2024;14:83–98; [2]Murray N et al. Dermatol Ther (Heidelb) 2024;14:3211–27; [3]Ingram JR. Br J Dermatol 2020;183:990–8; [4]Ingram JR et al. Dermatol Ther (Heidelb) 2024;14:2859–76; [5]Komodo Health database: https://www.komodohealth.com/product/healthcare-map/; [6]Smiley C et al. Dermatol 2023;239:273–6.
Author Contributions: Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **HLT**, **JRI**, **JH**, **MAA**, **BMM**, **SR**, **NA**, **MNN** and **AG**; Drafting of the publication, or reviewing it critically for important intellectual content: **HLT**, **JRI**, **JH**, **MAA**, **BMM**, **SR**, **NA**, **MNN** and **AG**; Final approval of the publication: **HLT**, **JRI**, **JH**, **MAA**, **BMM**, **SR**, **NA**, **MNN** and **AG**. **Author Disclosures: HLT:** Consultant for Incyte, Insmed, MoonLake Immunotherapeutics, Novartis, and UCB. Received a stipend as immediate past-Editor-in-Chief of the British Journal of Dermatology and an authorship honorarium from UpToDate. Consultant for AbbVie, Bluefin Biomedicine, Boehringer Ingelheim, Cantargia, ChemoCentryx, Citryll, Elasmogen, Engitix, Incyte, Indero, InflaRx, Insmed, Kymera Therapeutics, MoonLake Immunotherapeutics, Navigator Medicines, Novartis, Spyre Therapeutics, Sun Pharma, T-Therapeutics, UCB, UNION Therapeutics and Viela Bio. Co-copyright holder of HiSQOL©, HIDE©, HS-IGA©, HS-LTC©, I-GLASS© and Patient Global Assessment© instruments for HS. Department receives income from copyright of the Dermatology Life Quality Index (DLQI©) and related instruments. **JH:** On the Board of Directors for the Hidradenitis Suppurativa Foundation (unpaid); served as an advisor, investigator, and/or speaker for AbbVie, Aclaris, Amgen, Arcutis, AstraZeneca, Avalo, Boehringer Ingelheim, Galderma, Incyte, Insmed, LEO Pharma, MoonLake Immunotherapeutics, Navigator Medicines, Novartis, Oruka Therapeutics, Pfizer, Sanofi, Spyre, Regeneron, and UCB. **MAA:** Consulting fees from AbbVie, Santa Ana Bio, Mirador Therapeutics; advisory board for Novartis; research funding from UCB. **BMM:** Received disease-related consultancy fees, honoraria, and support for attending meetings from Novartis and UCB; consultancy fees from Almirall, Incyte, and MoonLake Immunotherapeutics; **SR**, **NA**, **MNN:** Employees of UCB. **AG:** Advisor for AbbVie, Almirall, Bluefin Biomedicine, Boehringer Ingelheim, Immunitas Therapeutics, Incyte, Insmed, Navigator Medicines, Novartis, Oruka Therapeutics, Otsuka Pharmaceutical, Pfizer, Priovant, Sun Pharma, Takeda, UCB, and Zura Bio and receives honoraria; receives research grants from AbbVie, CHORD COUSIN Collaboration (C3), and UCB; co-copyright holder of the HIdradenitis suppurativa DrainagE (HIDE©), Hidradenitis Suppurativa Investigator Global Assessment (HS-IGA©), and Hidradenitis Suppurativa Quality of Life (HiSQOL©) instruments. **Acknowledgements:** This study was funded by UCB. We thank the patients and their caregivers in addition to the investigators and their teams who contributed to this study. The authors also thank Susana Lobo Berastegui, MSci, UCB, Madrid, Spain for editorial review during poster development and publication coordination, Brindley Brooks, HS Connect, Washington, USA for review, Isabel Merrien, PgDip, Costello Medical, London, UK for medical writing and editorial assistance and the Costello Medical Creative team for design support. All costs associated with development of this poster were funded by UCB.

To receive a copy of this poster, scan the QR code.

Link expiration: 09 Jan 2027

Presented at Fall Clinical Dermatology Conference 2026 | October 08–11 | Las Vegas, NV