Bimekizumab improves draining tunnel outcomes in patients with moderate to severe HS regardless of prior biologic use: 3-year data from BE HEARD EXT

Antonio Martorell,[1–3] Jennifer Hsiao,[4] Falk G. Bechara,[5,6] Christos C. Zouboulis,[2,7] Melinda Gooderham,[8,9] Takuya Miyagawa,[10] Bartosz Lukowski,[11] Christina Crater,[12] Nicola Tilt,[13] Martina L. Porter[14]

[1]Department of Dermatology, Hospital de Manises, Valencia, Spain; [2]European Hidradenitis Suppurativa Foundation e.V., Dessau, Germany; [3]Translational Epigenomics in Disease and Longevity (EPITELONG), Health Research Institute La Fe (IIS La Fe), Valencia, Spain; [4]Department of Dermatology, University of Southern California, Los Angeles, California, USA; [5]Department of Dermatology, Venereology, and Allergology, St. Josef-Hospital, Ruhr-University Bochum, Bochum, Germany; [6]ICH – International Center for Hidradenitis Suppurativa / Acne Inversa, Ruhr-University Bochum, Bochum, Germany; [7]Department of Dermatology, Venereology and Immunology, Universitaetsklinikum Ruppin-Brandenburg, Brandenburg Medical School Theodor Fontane and Faculty of Health Sciences Brandenburg, Neuruppin, Germany; [8]SKiN Centre for Dermatology, Probity Medical Research, Peterborough, Ontario, Canada; [9]Queen's University, Kingston, Ontario, Canada; [10]Department of Dermatology, University of Tokyo Graduate School of Medicine, Tokyo, Japan; [11]Vedim/UCB, Warsaw, Poland; [12]UCB, Morrisville, North Carolina, USA; [13]UCB, Slough, UK; [14]Beth Israel Deaconess Medical Center, Department of Dermatology, Harvard Medical School, Boston, Massachusetts, USA.

Objective

To report the impact of bimekizumab on draining tunnel (DT) outcomes in patients with moderate to severe hidradenitis suppurativa (HS) over 3 years, including in prior biologic use patient subgroups.

Synopsis

- HS is a chronic, inflammatory skin disease characterized by painful lesions, including DTs.[1]
- DTs cause irreversible structural damage, negatively impact quality of life and are indicative of disease progression.[2,3]
- Delays in initiating biologic treatment beyond the therapeutic 'window of opportunity' may lead to an increase in DT counts, thereby exacerbating this progression.[4]
 - Use of a greater number of prior biologic therapies may also reduce biologic responsiveness.[5]
- Interleukin (IL)-17F and IL-17A are highly expressed in patients with HS, with IL-17F more abundant in HS serum and lesional skin than IL-17A.[6,7] IL-17F expression is significantly upregulated in lesions, with the highest expression in draining tunnels.[8]
- Bimekizumab is a humanized IgG1 monoclonal antibody that selectively inhibits interleukin (IL)-17F in addition to IL-17A.[9]

Methods

- Data were pooled from the phase 3 BE HEARD I&II (NCT04242446, NCT04242498) trials and their open-label extension, BE HEARD EXT (NCT04901195) for patients with moderate to severe HS.[10,11]
- Data are reported for patients randomized to bimekizumab 320 mg from baseline in BE HEARD I&II who subsequently entered BE HEARD EXT and continued to receive bimekizumab (BKZ Total).
 - Data are also stratified by prior biologic use for HS (biologic-naïve or biologic-experienced).
- The following outcomes are reported:
 - Mean (standard deviation [SD]) DT count for all patients and those with ≥1 baseline DT over 3 years (Week 148);
 - Patients with ≥1 DT at baseline achieving DT counts of 0, 1–2, 3–5, or >5 at baseline, Year 1 (Week 48) and Year 3;
 - Patients with 1–2, 3–5, or >5 DTs at baseline achieving a DT count of 0 (DT=0) at Year 1 and Year 3.
- Data are reported as observed case (OC).

Results

- Of the 1,014 patients in BE HEARD I&II, 868 were randomized to bimekizumab at baseline; 556 patients completed Year 1 and entered BE HEARD EXT.
 - Of those, 444 patients were biologic-naïve and 112 were biologic-experienced; 425 had ≥1 DT at baseline.
- Baseline characteristics were generally similar between subgroups, though biologic-experienced patients had longer disease duration and more severe HS than those who were biologic-naïve (**Table 1**).
- Mean DT counts were generally higher in biologic-experienced versus biologic-naïve patients over 3 years; this trend was consistent in all patients with ≥1 DT at baseline (**Figures 1A** and **1B**).
- In patients with ≥1 DT at baseline, the proportions achieving DT counts of 0, 1–2, 3–5, and >5 are presented in **Figure 2**.
 - At Year 3, over two thirds of biologic-naïve patients achieved DT=0, compared with approximately half of those who were biologic-experienced.
 - Similarly, the proportion of patients with >5 DTs reduced from baseline to Year 3, with greater reductions seen at Year 1 in biologic-naïve versus biologic-experienced patients.
- Achievement of DT=0 at Years 1 and 3 was highest in patients with 1–2 baseline DTs and lowest in those with >5 baseline DTs (**Figure 3**).
 - Across baseline DT counts, the proportions of patients achieving DT=0 at Year 3 were generally higher in biologic-naïve versus biologic-experienced patients.

Conclusions

Reductions in DT counts were maintained through 3 years of bimekizumab treatment for all subgroups, though greater achievement was observed in biologic-naïve versus biologic-experienced patients.

Lower baseline DT counts were associated with greater DT clearance over 3 years, irrespective of prior biologic use.

These results may support early bimekizumab initiation during the therapeutic 'window of opportunity', when prior biologic use is likely to be low and HS less severe, to prevent disease progression.

Summary

Biologic-naïve patients displayed **higher achievement** of **DT outcomes** with **bimekizumab** at Year 3 compared with biologic-experienced patients, though **improvements** were **maintained** through 3 years, **regardless of prior biologic use**

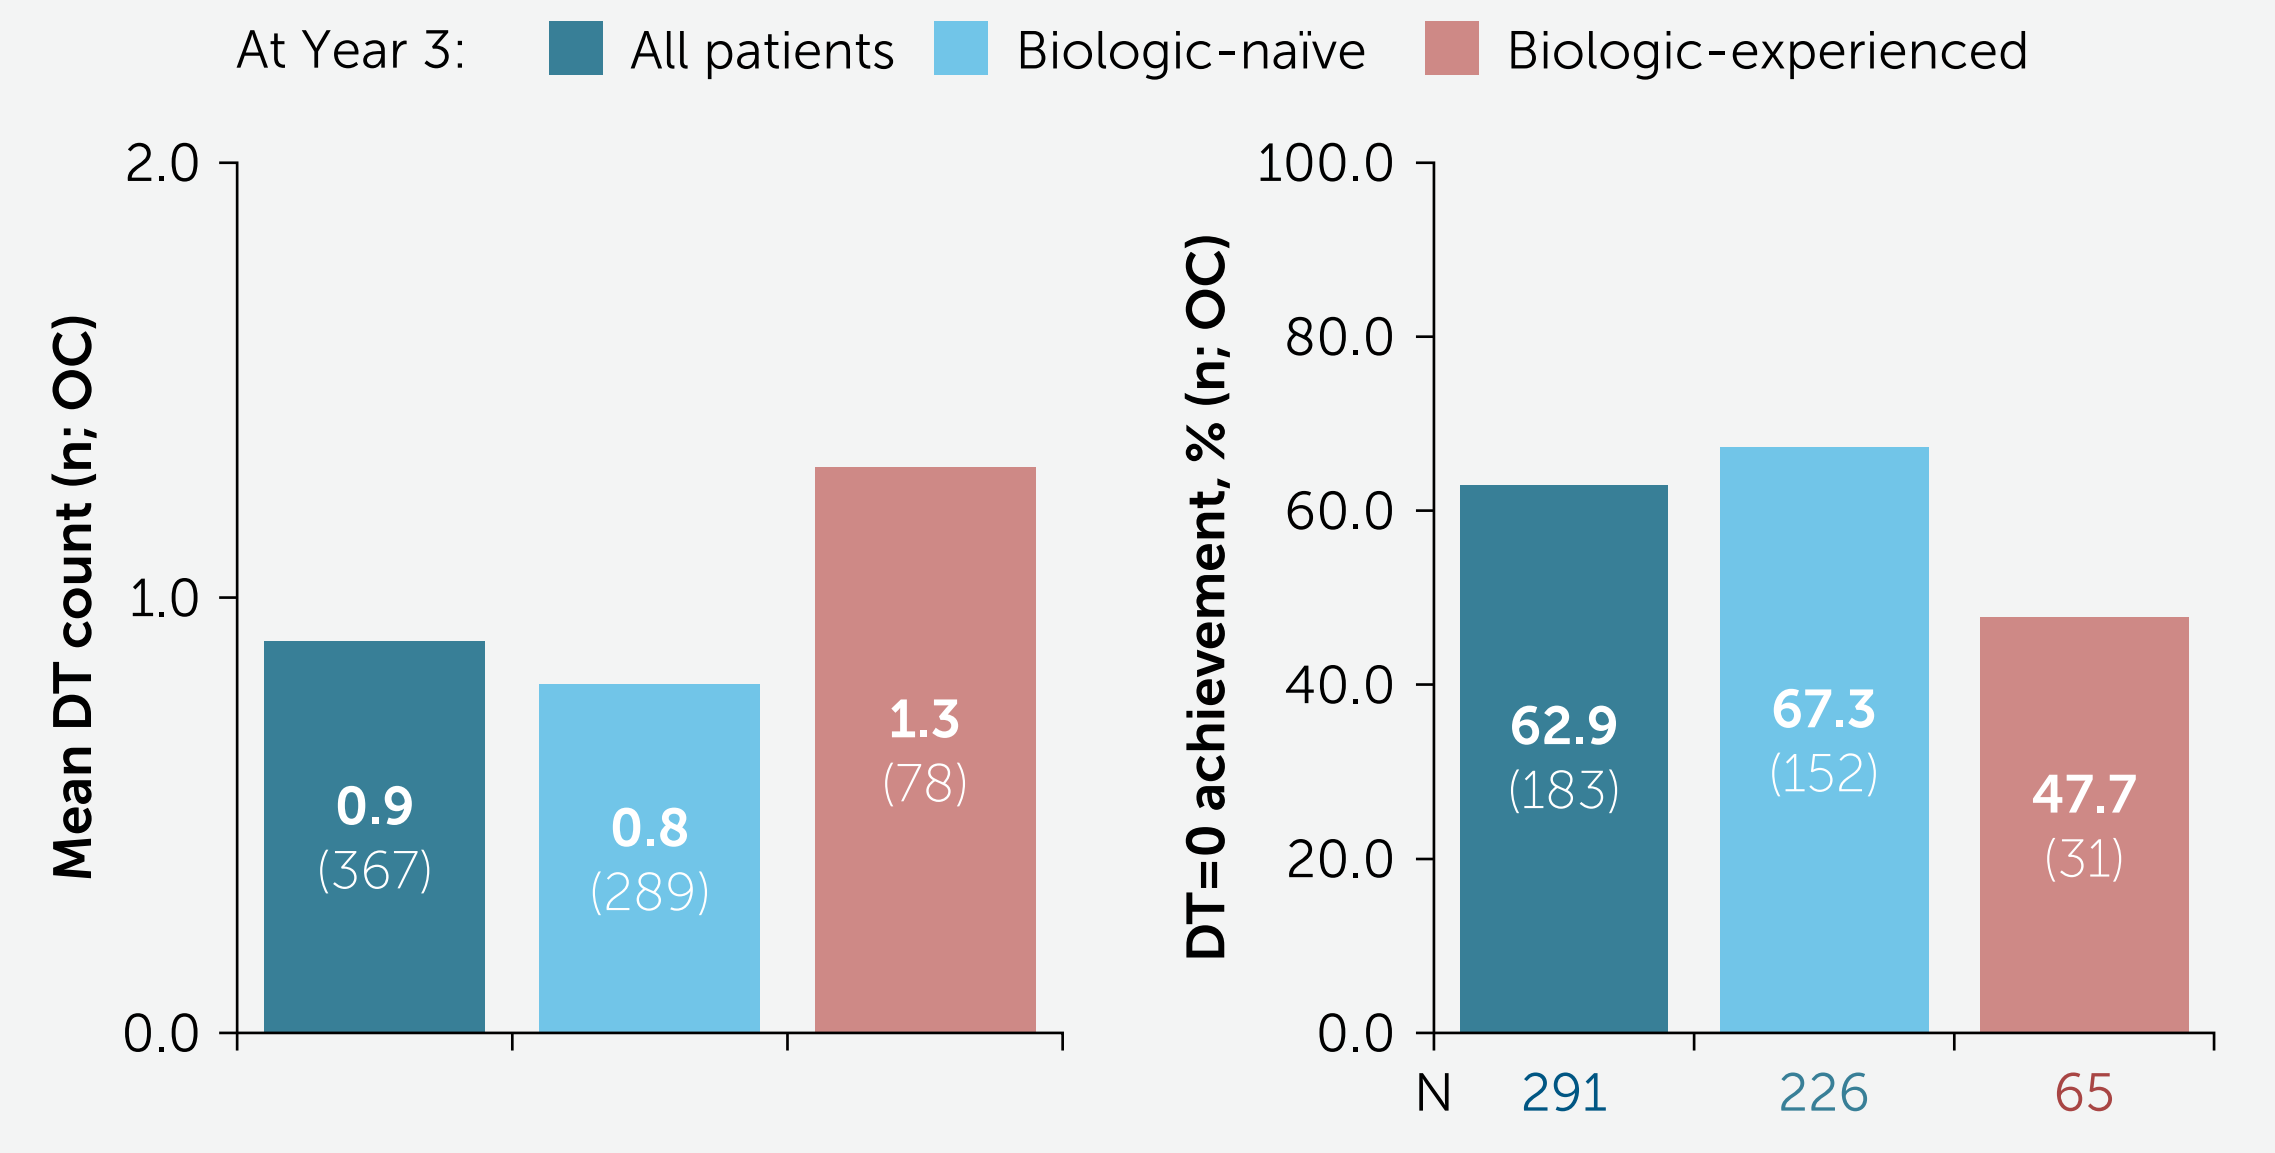
At Year 3: All patients Biologic-naïve Biologic-experienced
Mean DT count (n; OC)
0.9 (367)
0.8 (289)
1.3 (78)
DT=0 achievement, % (n; OC)
62.9 (183)
67.3 (152)
47.7 (31)
N 291 226 65

Early initiation of **bimekizumab** during the 'window of opportunity' may **prevent disease progression**

Table 1 Baseline characteristics

	All BKZ Total N=556	Biologic-naïve N=444	Biologic-experienced N=112
Age (years), mean (SD)	36.3 (12.2)	36.0 (12.0)	37.5 (12.6)
Sex (female), n (%)	299 (53.8)	244 (55.0)	55 (49.1)
Racial group, n (%)			
White	448 (80.6)	351 (79.1)	97 (86.6)
Black or African American	55 (9.9)	50 (11.3)	5 (4.5)
BMI (kg/m²), mean (SD)	32.5 (7.8)	32.5 (7.9)	32.4 (7.6)
Duration of disease (years), mean (SD)	7.4 (7.1)	7.2 (7.2)	8.3 (6.9)
IHS4 score, mean (SD)	35.6 (31.5)	33.3 (28.3)	44.7 (40.9)
AN count, mean (SD)	16.9 (18.5)	16.7 (15.6)	17.6 (27.3)
Hurley stage,[a] n (%)			
II	303 (54.5)	263 (59.2)	40 (35.7)
III	253 (45.5)	181 (40.8)	72 (64.3)
Baseline antibiotic use, n (%)	54 (9.7)	47 (10.6)	7 (6.3)
DLQI total score, mean (SD)	11.0 (6.8)	10.5 (6.6)	13.2 (7.1)
HiSQOL total score, mean (SD)	24.6 (12.8)	23.8 (12.7)	28.0 (12.6)

[a] This refers to the worst overall Hurley stage derived from the Hurley stages recorded across all anatomical regions.

Figure 1 Mean DT count over 3 years

A) Mean DT count over 3 years

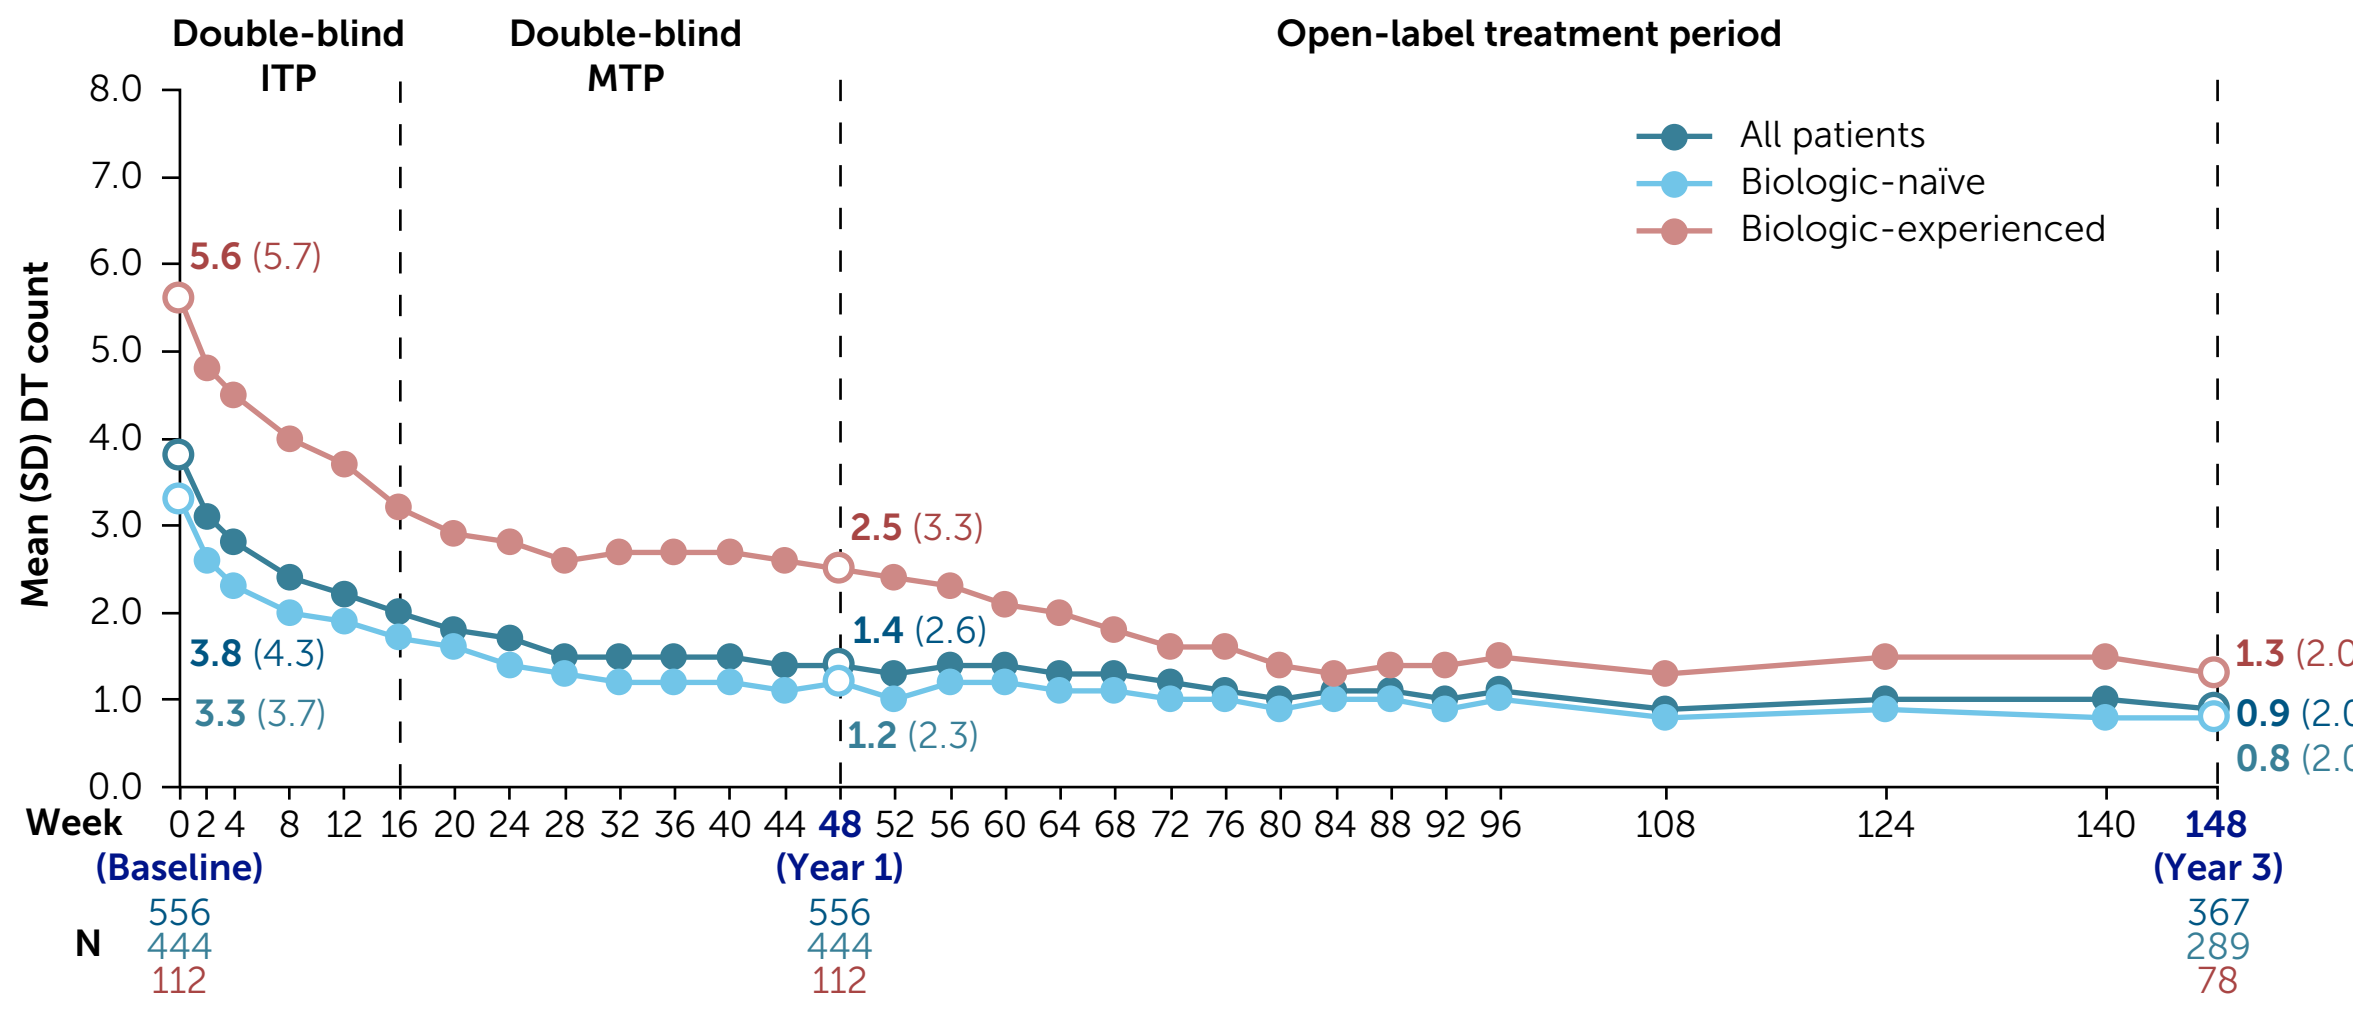
Double-blind ITP
Double-blind MTP
Open-label treatment period
All patients
Biologic-naïve
Biologic-experienced
Mean (SD) DT count
5.6 (5.7)
3.8 (4.3)
3.3 (3.7)
2.5 (3.3)
1.4 (2.6)
1.2 (2.3)
1.3 (2.0)
0.9 (2.0)
0.8 (2.0)
Week 0 2 4 8 12 16 20 24 28 32 36 40 44 48 52 56 60 64 68 72 76 80 84 88 92 96 108 124 140 148
(Baseline) (Year 1) (Year 3)
N 556 444 112 556 444 112 367 289 78

B) Mean DT count in patients with ≥1 DT at baseline over 3 years

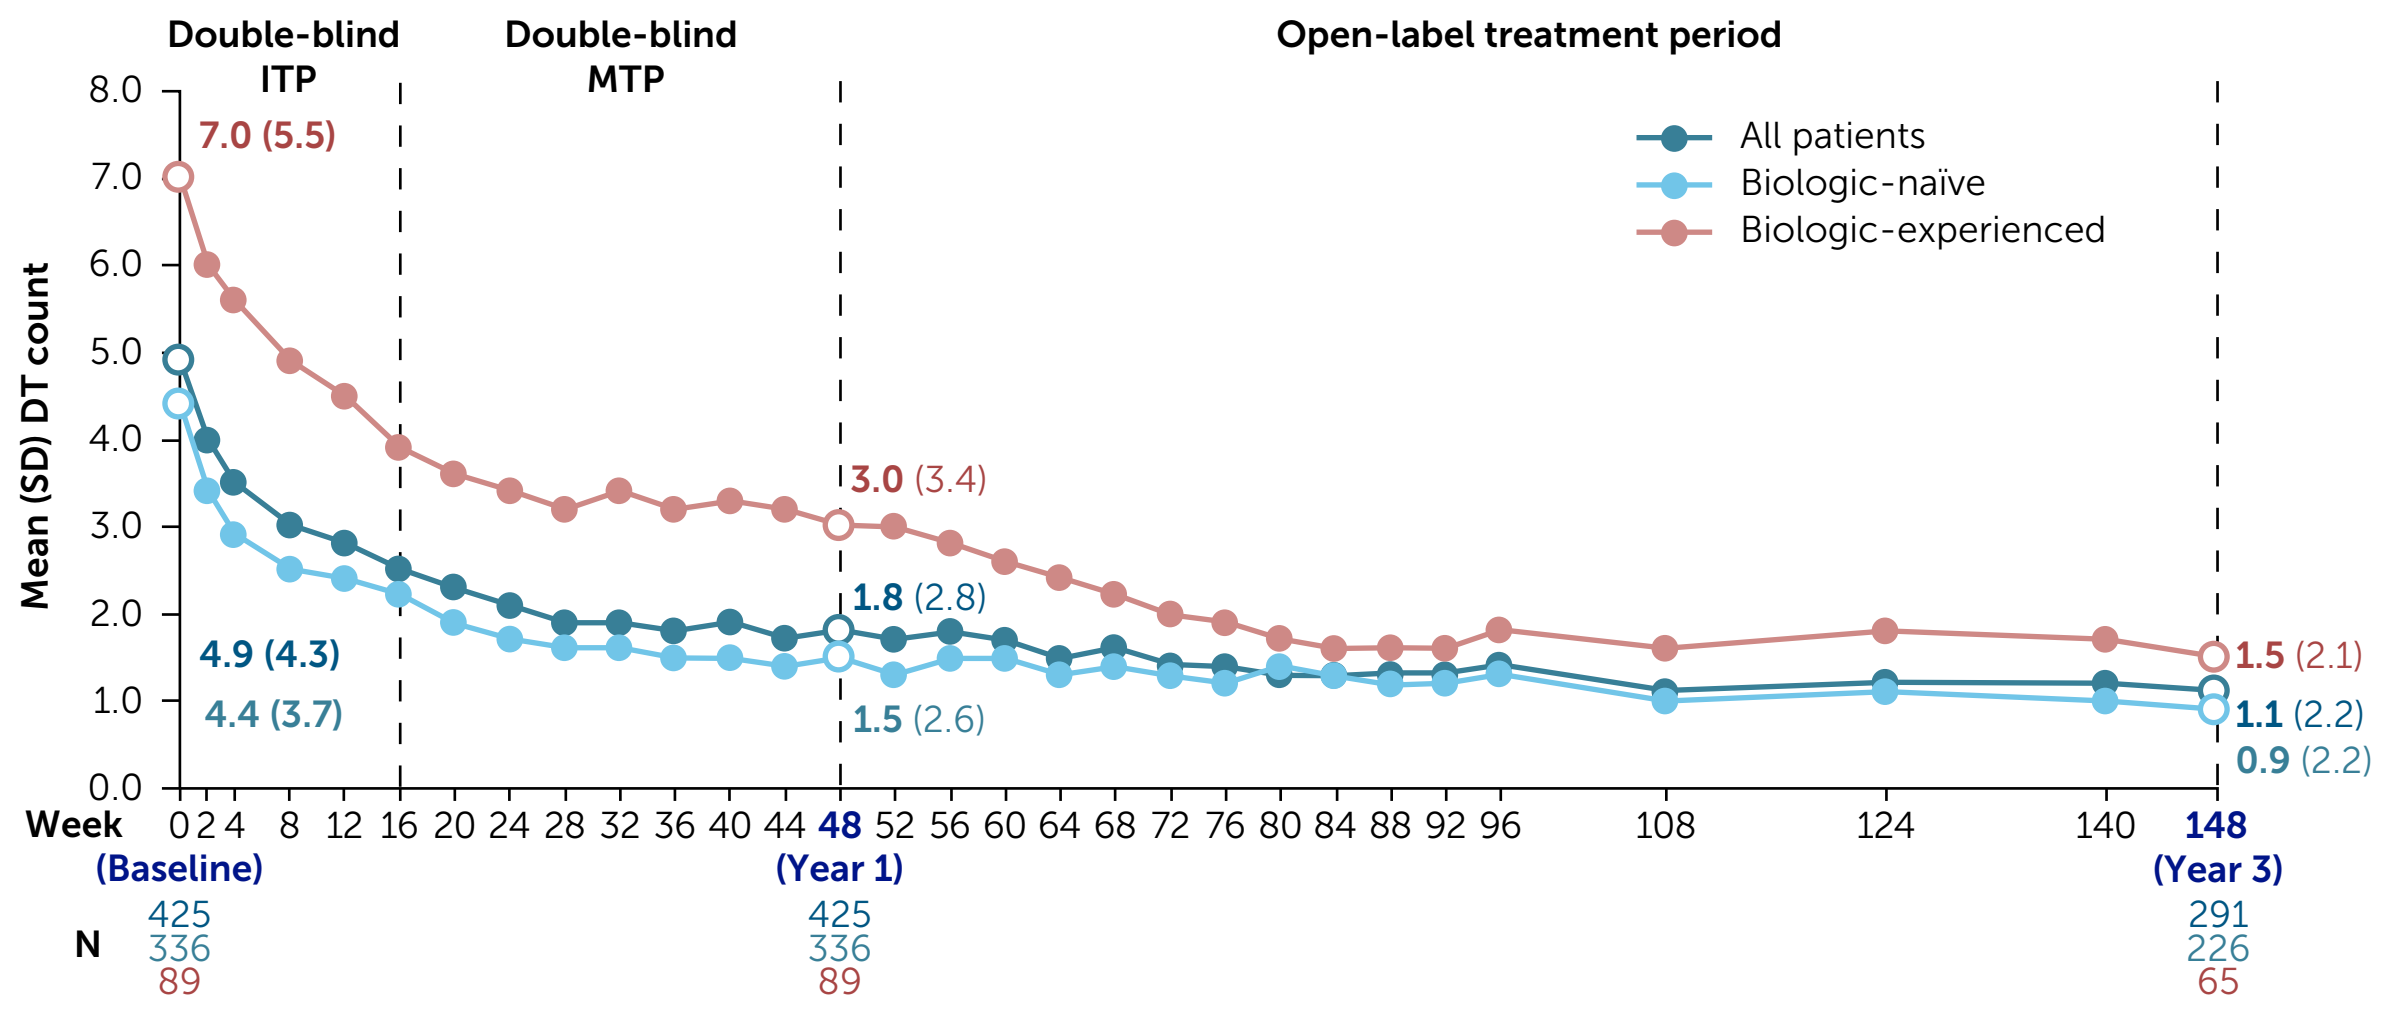
Double-blind ITP
Double-blind MTP
Open-label treatment period
All patients
Biologic-naïve
Biologic-experienced
Mean (SD) DT count
7.0 (5.5)
4.9 (4.3)
4.4 (3.7)
3.0 (3.4)
1.8 (2.8)
1.5 (2.6)
1.5 (2.1)
1.1 (2.2)
0.9 (2.2)
Week 0 2 4 8 12 16 20 24 28 32 36 40 44 48 52 56 60 64 68 72 76 80 84 88 92 96 108 124 140 148
(Baseline) (Year 1) (Year 3)
N 425 336 89 425 336 89 291 226 65

OC, N: N represents number of patients with a non-missing draining tunnel count assessment in the given week.

Figure 2 Patients with ≥1 DT at baseline with 0, 1–2, 3–5, or >5 DTs at baseline, Year 1, and Year 3

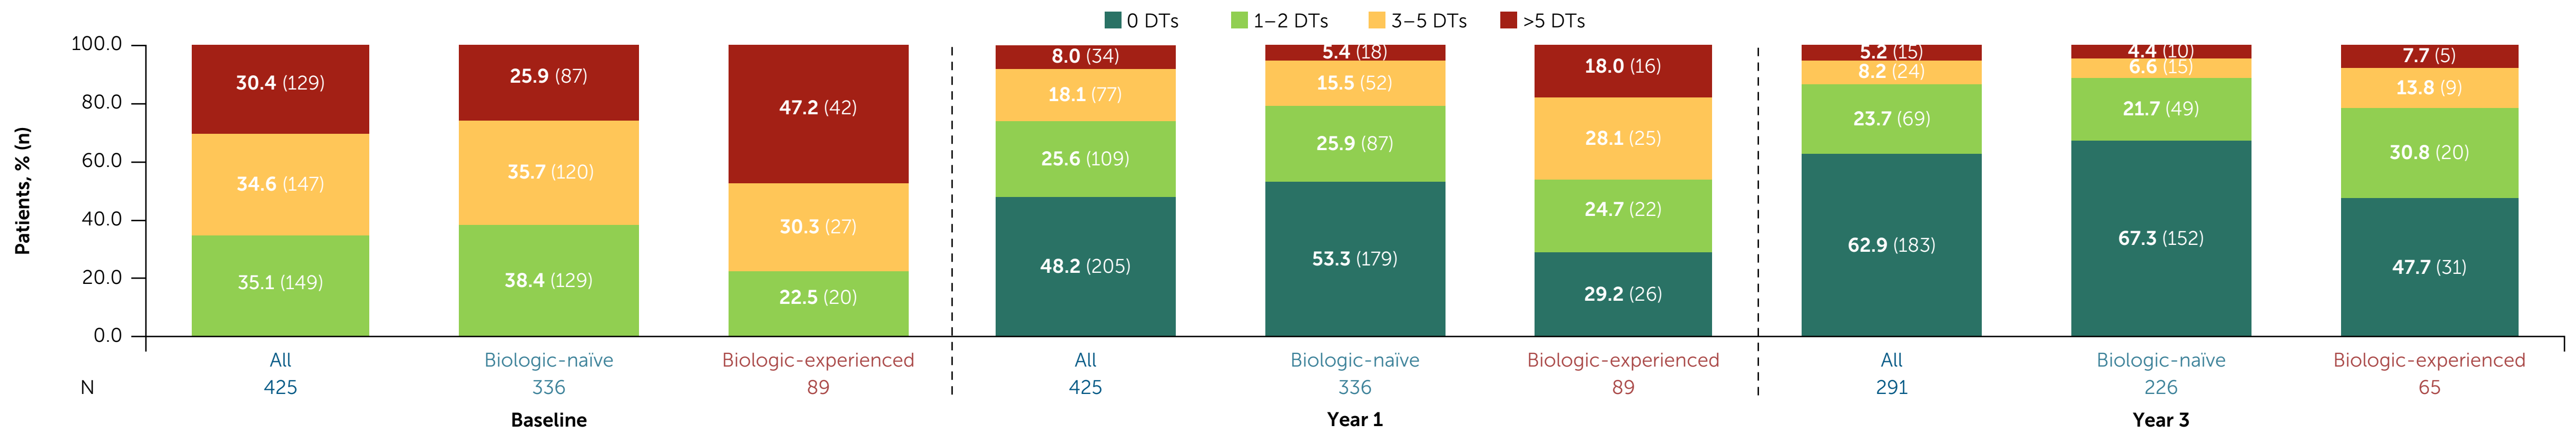
0 DTs 1–2 DTs 3–5 DTs >5 DTs
Patients, % (n)
Baseline: All 425: 30.4 (129), 34.6 (147), 35.1 (149)
Biologic-naïve 336: 25.9 (87), 35.7 (120), 38.4 (129)
Biologic-experienced 89: 47.2 (42), 30.3 (27), 22.5 (20)
Year 1: All 425: 8.0 (34), 18.1 (77), 25.6 (109), 48.2 (205)
Biologic-naïve 336: 5.4 (18), 15.5 (52), 25.9 (87), 53.3 (179)
Biologic-experienced 89: 18.0 (16), 28.1 (25), 24.7 (22), 29.2 (26)
Year 3: All 291: 5.2 (15), 8.2 (24), 23.7 (69), 62.9 (183)
Biologic-naïve 226: 4.4 (10), 6.6 (15), 21.7 (49), 67.3 (152)
Biologic-experienced 65: 7.7 (5), 13.8 (9), 30.8 (20), 47.7 (31)

OC, N: N represents number of patients with a non-missing draining tunnel count assessment in the given week, and percentages are calculated accordingly.

Figure 3 Patients with 1–2, 3–5, or >5 DTs at baseline achieving DT=0 at Year 1 and Year 3

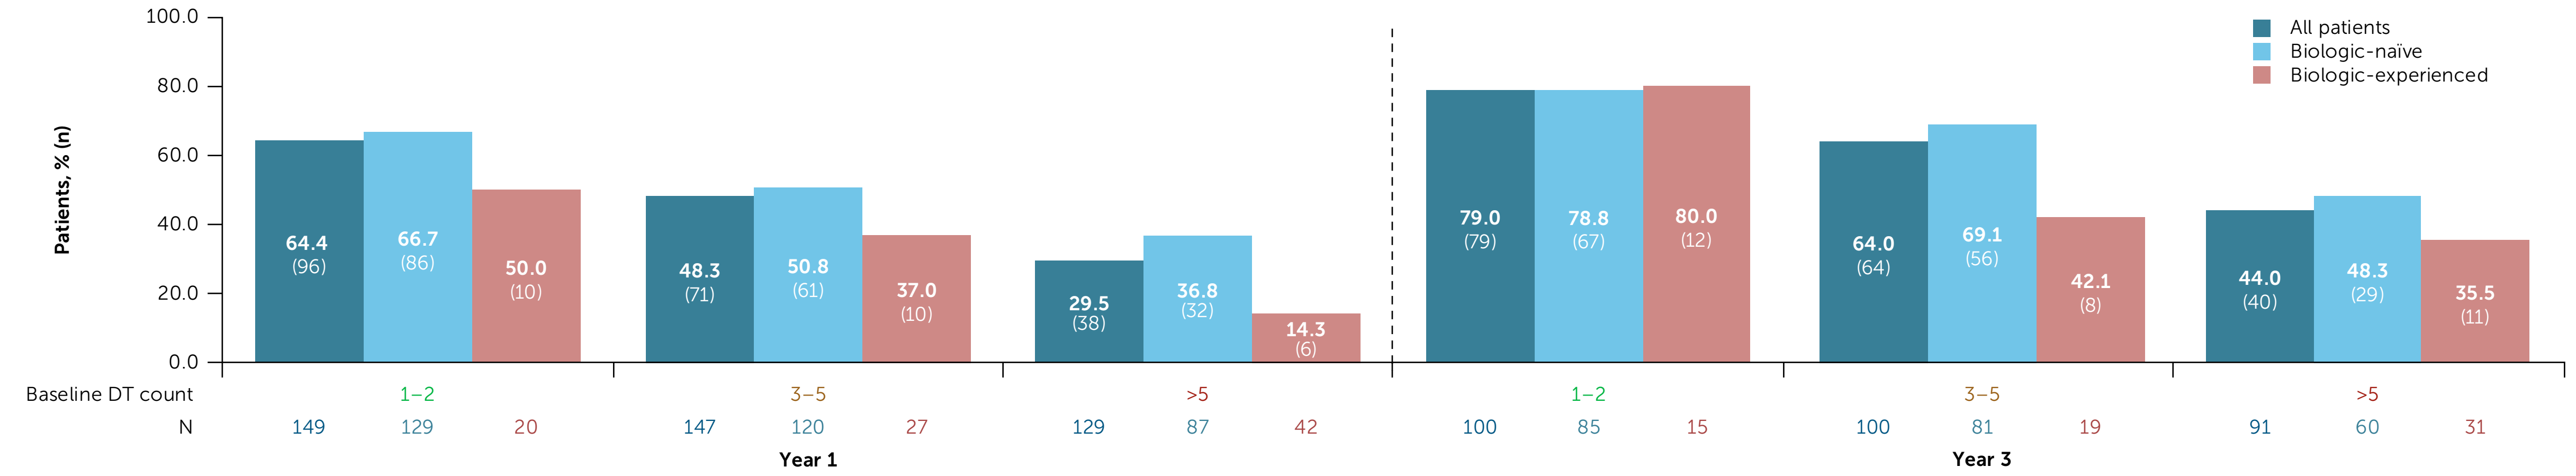
All patients
Biologic-naïve
Biologic-experienced
Patients, % (n)
Year 1, Baseline DT count 1–2: 64.4 (96), 66.7 (86), 50.0 (10); N 149 129 20
3–5: 48.3 (71), 50.8 (61), 37.0 (10); N 147 120 27
>5: 29.5 (38), 36.8 (32), 14.3 (6); N 129 87 42
Year 3, 1–2: 79.0 (79), 78.8 (67), 80.0 (12); N 100 85 15
3–5: 64.0 (64), 69.1 (56), 42.1 (8); N 100 81 19
>5: 44.0 (40), 48.3 (29), 35.5 (11); N 91 60 31

OC, N: N represents number of patients with a non-missing draining tunnel count assessment in the given week, and percentages are calculated accordingly.

AN: abscess and inflammatory nodule; **BKZ:** bimekizumab; **BMI:** body mass index; **DLQI:** Dermatology Life Quality Index; **DT:** draining tunnel; **HiSQOL:** Hidradenitis Suppurativa Quality of Life; **HS:** hidradenitis suppurativa; **IHS4:** International Hidradenitis Suppurativa Severity Score System; **IL:** interleukin; **ITP:** initial treatment period; **MTP:** maintenance treatment period; **OC:** observed case; **SD:** standard deviation.

References: [1]Margesson LJ. Best Pract Res Clin Obstet Gynaecol 2014;28:1013–27; [2]Ingram JR et al. J Eur Acad Dermatol Venereol 2025;39:1431–41; [3]Navarazhina K et al. J Allergy Clin Immunol 2021;147:2213–24; [4]Haselgruber S et al. Dermatol Ther 2024;14:1875–90; [5]Fernandez-Crehuet P et al. Dermatol Ther (Heidelb) 2023;13:1029–38; [6]Rastrick J et al. Br J Dermatol 2025;192:660–71; [7]Rumberger BE et al. Inflamm Res 2020;69:967–73; [8]Ben Abdallah H et al. Front Immunol 2026;16:1715474; [9]Adams R et al. Front Immunol 2020;11:1894; [10]Kimball AB et al. Lancet 2024;403:2504–19; [11]Sayed CJ et al. J Am Acad Dermatol 2026;94:867–78.
Author Contributions: Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **AM**, **JH**, **FGB**, **CCZ**, **MG**, **TM**, **BL**, **CC**, **NT**, **MLP**; drafting of the publication, or reviewing it critically for important intellectual content: **AM**, **JH**, **FGB**, **CCZ**, **MG**, **TM**, **BL**, **CC**, **NT**, **MLP**; final approval of the publication: **AM**, **JH**, **FGB**, **CCZ**, **MG**, **TM**, **BL**, **CC**, **NT**, **MLP**. **Author Disclosures: AM:** Received honoraria and/or travel grants and/or acted as an advisory board member for AbbVie, Almirall, Amgen, Boehringer Ingelheim, Bristol Myers Squibb, Eli Lilly and Company, Janssen, Legit Health, LEO Pharma, L'Oréal, Novartis, Otsuka, Sanofi, and UCB; worked as a principal investigator in clinical trials supported by AbbVie, Alumis, Bristol Myers Squibb, Citryll, Eli Lilly and Company, Galderma, Indero, Janssen, Kymera Therapeutics, Legit Health, MoonLake Immunotherapeutics, MSD, Novartis, Pfizer, Priovant, Sanofi, Sun Pharma, Takeda, UCB, and Zura. **JH:** On the Board of Directors for the Hidradenitis Suppurativa Foundation (unpaid); served as an advisor, investigator and/or speaker for AbbVie, Aclaris, Amgen, Arcutis, AstraZeneca, Avalo, Boehringer Ingelheim, Galderma, Incyte, Insmed, LEO Pharma, MoonLake Immunotherapeutics, Navigator Medicines, Novartis, Oruka Therapeutics, Pfizer, Spyre, Sanofi, Regeneron, and UCB. **FGB:** Received honoraria for participation in advisory boards, in clinical trials, and/or as a speaker from AbbVie Inc., AbbVie Deutschland GmbH & Co. KG, Acelyrin, Almirall, Avalo, Beiersdorf, Boehringer Ingelheim, Pharma GmbH & Co. KG, Celltrion, Citryll, Eli Lilly and Company, Incyte Corporation, Inflax, JanssenCilag GmbH, Johnson & Johnson, Merck, Mölnlycke, MoonLake Immunotherapeutics, Navigator Medicines, Novartis Pharma GmbH, Sanofi, Sitala, Takeda, and UCB. **CCZ:** Received disease-relevant honoraria as a consultant for Almirall, Boehringer Ingelheim, Eli Lilly and Company, Engitix, Incyte, Indero, LEO, MSD, Novartis, Paratus, Pfizer, Sanofi, Takeda, UCB, and ZuraBio and lecture fees from BMS, Novartis, Pfizer, Streamed-Up, and UCB. Received research grants from Brandenburg Medical School Theodor Fontane, EADV, European Union and German Federal Ministry of Education and Research; President of the European Hidradenitis Suppurativa Foundation (EHSF) e.V. and the Deutsches Register Morbus Adamantiades-Behçet e.V.; coordinator of the Acquired immunological LOw prevalence and Complex AdulT diseases of thE Skin (ALLOCATE Skin) Skin group of the ERN European Reference Network (ERN) Skin; chair of the Acne, Rosacea and Hidradenitis Suppurativa (ARHS) Task Force group of the EADV and board member of the International Society for Behçet's Disease; Editor of the EADV News; co-copyright holder of IHS4 on behalf of the EHSF e.V. **MG:** Investigator, speaker, consultant or advisory board member for AbbVie, Acelyrin, Akros, Alumis, Amgen, AnaptysBio, Arcutis, Aristea, Bausch Health, Boehringer Ingelheim, Bristol Myers Squibb, Dermavant, Dermira, Eli Lilly and Company, Expedition Therapeutics Inc, Galderma, GSK, Incyte, JAMP Pharma, Janssen, Kyowa Kirin, L'Oréal, MedImmune, Meiji, MoonLake Immunotherapeutics, Nektar Therapeutics, Nimbus, Novartis, Oruka, Pfizer, Q32 Bio, Regeneron, Reistone, Sanofi, Sun Pharma, Takeda, Tarsus, UCB, Union, Ventyx, and Vyne. **TM:** Advisor, consultant, speaker and/or investigator for AbbVie, Boehringer Ingelheim, Bristol Myers Squibb, Novartis, ONO Pharma, and UCB. **BL**, **CC**, **NT**: Employees and shareholders of UCB. **MLP:** Consultant and investigator for AbbVie, Arcutis, Aristea Therapeutics, Avalo Therapeutics, Eli Lilly and Company, Incyte, Janssen, Merck, MoonLake Immunotherapeutics, Navigator Biosciences, Novartis, Pfizer, Prometheus, Sanofi, Sonoma Biotherapeutics, UCB, Ventyx, and Zura Bio; consultant for Almirall, FIDE, Insmed, Oruka, Otsuka, Propeller Biosciences, Spyre, and Trifecta Clinical/WCG; investigator for AnaptysBio, Bayer, Bristol Myers Squibb, OASIS Pharmaceuticals, and Regeneron; received royalties from Beth Israel Deaconess Medical Center; received fellowship funding to institution from AbbVie. **Acknowledgements:** These studies were funded by UCB. We thank the patients and their caregivers in addition to the investigators and their teams who contributed to these studies. The authors also thank Susana Lobo Berastegui, MSci, of UCB, for editorial review during poster development and publication coordination, Lauren Carlberg, PhD, and Sarah Johnson, MSci, Costello Medical, Manchester, UK, for medical writing and editorial assistance and the Costello Medical Creative team for design support. All costs associated with development of this poster were funded by UCB.

To receive a copy of this poster and the study design figure, scan the QR code.

Link expiration: January 9, 2027

Presented at Fall Clinical Dermatology Conference 2026 | October 08–11 | Las Vegas, NV

Previously presented at EADV 2026