Bimekizumab and other biologics for moderate to severe psoriasis: Systematic literature review and meta-analysis of real-world PASI 100 response over time

Diego Orsini,[1] Yukie Yamaguchi,[2] Lucy Carty,[3] Aijing Shang,[4] Balint Szilagyi,[5] George Han[6]

[1]Departmental Faculty of Medicine UniCamillus, "Saint Camillus, International University of Health and Medical Sciences", Rome, Italy; [2]Department of Environmental Immuno-Dermatology, Yokohama City University Graduate School of Medicine, Yokohama, Japan; [3]UCB, Brussels, Belgium; [4]UCB, Basel, Switzerland; [5]UCB, Monheim am Rhein, Germany; [6]Department of Dermatology, Icahn School of Medicine at Mount Sinai, New York, NY, USA.

Objective

To synthesize and generate standardized PASI 100 response profiles over time for bimekizumab and IL-17A or IL-23 biologic therapies using a systematic literature review (SLR) and meta-analysis of real-world evidence.

Synopsis

- Complete skin clearance is increasingly achievable for individuals with moderate to severe psoriasis treated with biologic therapies.[1]
- Randomized clinical trials and network meta-analyses have previously demonstrated the superior efficacy of bimekizumab versus several biologics for psoriasis.[2–5]
- Real-world evidence provides insights into biologic outcomes in routine clinical care settings, which include broader patient populations than clinical trials. However, as most available evidence comes from individual observational studies, uncertainty remains regarding the achievement of skin responses, including complete skin clearance, in routine clinical practice.

Methods

- An SLR of real-world evidence studies evaluating therapies for adults with psoriasis published 2020–2025 was conducted using MEDLINE and Embase.
- Publications with restricted populations (e.g., studies limited to bio-naïve patients) were excluded, as were publications restricted by patient characteristics (e.g., elderly patients) or site-specific psoriasis (e.g., nail psoriasis).
- Real-world PASI 100 response data were analyzed for adults with psoriasis treated with bimekizumab, guselkumab, ixekizumab, secukinumab, risankizumab, and tildrakizumab.
- Reported proportions of patients achieving PASI 100 at Weeks 4–52, according to available data, were analyzed using a longitudinal logistic regression meta-analysis approach.
 - Analyses were conducted separately for each biologic.
 - Study was included as a random intercept to account for between-study differences and summarized using the estimated random-effects standard deviation (τ).
- Model-estimated PASI 100 response rates with 95% confidence intervals (CI) were generated from Weeks 4–52 for each biologic.

Results

- Overall, 165 real-world publications were identified; following review, 77 real-world publications from >35 countries across Asia-Pacific, Europe, Latin America, the Middle East and North America were included in the analysis, with the largest contributions originating from Europe and China.
- As some publications included more than one drug, a total of 94 biologic datasets were analyzed; the number of available publications for each biologic ranged 8–28.
 - Differences in market availability and duration of clinical use may have influenced the amount of available evidence for individual biologics.
 - The majority of publications described retrospective, multi-center cohort studies.
- Patient characteristics pooled across all included publications are presented in **Table 1**.
- Reported PASI 100 response rates demonstrated substantial variability and heterogeneity between studies within each biologic (**Figure 1**).
- The highest modelled responses were observed for bimekizumab across all analyzed time points (**Table 2**).
 - The relative order of estimated PASI 100 responses for other biologics varied at different time points.
 - Guselkumab had the lowest model-estimated response rate at Week 4, while secukinumab had the lowest model-estimated response rate at Week 52.
- At Week 4, estimated PASI 100 responses ranged from 5.6% (95% CI: 3.7%, 8.5%) to 29.4% (24.9%, 34.3%) across biologics (**Figure 2**).
 - At Week 16, estimates ranged from 28.1% (22.2%, 34.8%) to 65.9% (60.8%, 70.6%).
 - By Week 52, PASI 100 estimates ranged from 46.8% (40.8%, 52.9%) to 79.9% (73.2%, 85.3%).

Conclusions

This analysis synthesized real-world PASI 100 responses for biologic therapies in moderate to severe psoriasis. While there was variation between biologics, numerically, bimekizumab showed the highest estimated PASI 100 response rates from the earliest time point (Week 4) through to Week 52.

While formal statistical comparisons were not conducted, descriptively, this suggests bimekizumab had earlier and higher-level responses than other biologics over time. These results establish a foundation for future comparative effectiveness real-world evidence studies to adequately adjust for relevant confounding factors.

Summary

A **systematic literature review** of **real-world publications** evaluating biologic therapies for adults with psoriasis published 2020–2025 was conducted

Published **real-world PASI 100 responses** were used to model **treatment response over time** for **six biologics**

Across all evaluated timepoints (Weeks 4–52), **bimekizumab** had the highest model-estimated **PASI 100 response rate**

PASI 100	Week 4	Week 16	Week 52
Highest	Bimekizumab 29.4%	Bimekizumab 65.9%	Bimekizumab 79.9%
Lowest	Guselkumab 5.6%	Tildrakizumab 28.1%	Secukinumab 46.8%

Table 1 Baseline characteristics of pooled patients identified in all publications

	BKZ	GUS	IXE	RIS	SEC	TIL
Studies, N	8	18	14	16	28	10
Patients, N	1,895	4,225	4,333	6,379	7,639	1,023
Age (years), mean (SD)	49.0 (14.7)	49.7 (14.9)	47.1 (14.2)	49.8 (15.1)	46.9 (14.1)	51.7 (16.2)
Sex, male, n (%)	1,170 (64.2)	2,558 (63.2)	2,258 (63.7)	3,354 (63.7)	4,040 (61.9)	614 (60.0)
BMI, mean (SD)	27.4 (6.1)	28.8 (6.3)	27.3 (5.8)	28.1 (6.2)	27.8 (5.8)	28.0 (6.0)
Disease duration (years), mean (SD)	14.5 (11.0)	20.1 (13.3)	15.6 (13.3)	18.4 (12.9)	17.2 (12.7)	19.3 (12.7)
PASI, mean (SD)	15.8 (8.8)	14.1 (9.0)	15.5 (9.3)	14.6 (8.3)	17.1 (10.1)	13.6 (7.6)
Biologic-naïve, n (%)	832 (45.6)	1,698 (41.9)	2,130 (66.8)	2,627 (49.9)	4,366 (68.9)	619 (60.5)
Psoriatic arthritis, n (%)	360 (21.3)	744 (19.7)	688 (24.5)	725 (14.4)	1,442 (23.4)	110 (16.1)

The number of contributing studies and patients varies by characteristic depending on available data.

Table 2 Pooled model-estimated PASI 100 responses at key timepoints

	Week 4	Week 8	Week 16	Week 24	Week 52
Model-estimated PASI 100 response, % (95% CI)					
BKZ	**29.4 (24.9, 34.3)**	**42.8 (37.8, 48.0)**	**65.9 (60.8, 70.6)**	**74.7 (69.9, 78.9)**	**79.9 (73.2, 85.3)**
GUS	**5.6 (3.7, 8.5)**	**11.0 (8.1, 14.7)**	29.0 (23.4, 35.2)	41.1 (34.4, 48.2)	50.8 (43.6, 57.9)
IXE	15.7 (10.9, 22.0)	30.3 (22.9, 38.9)	55.6 (46.2, 64.6)	60.1 (50.8, 68.8)	59.5 (50.0, 68.3)
RIS	13.1 (9.9, 17.2)	21.1 (17.2, 25.6)	41.1 (35.9, 46.4)	53.9 (48.5, 59.2)	68.0 (62.9, 72.7)
SEC	9.5 (7.3, 12.3)	21.3 (17.4, 25.7)	46.3 (40.4, 52.2)	47.5 (41.6, 53.5)	**46.8 (40.8, 52.9)**
TIL	9.6 (6.0, 15.0)	14.5 (10.4, 19.9)	**28.1 (22.2, 34.8)**	**40.0 (33.0, 47.5)**	48.8 (40.4, 57.2)

Green bolding shows highest model-estimated PASI 100 response rate at each timepoint, red bolding shows lowest response rate at each timepoint.

BKZ: bimekizumab; CI: confidence interval; GUS: guselkumab; IXE: ixekizumab; PASI 100: 100% improvement in Psoriasis Area and Severity Index; RIS: risankizumab; SD: standard deviation; SEC: secukinumab; SLR: systematic literature review; TIL: tildrakizumab.

Figure 1 Published real-world PASI 100 responses over time by biologic

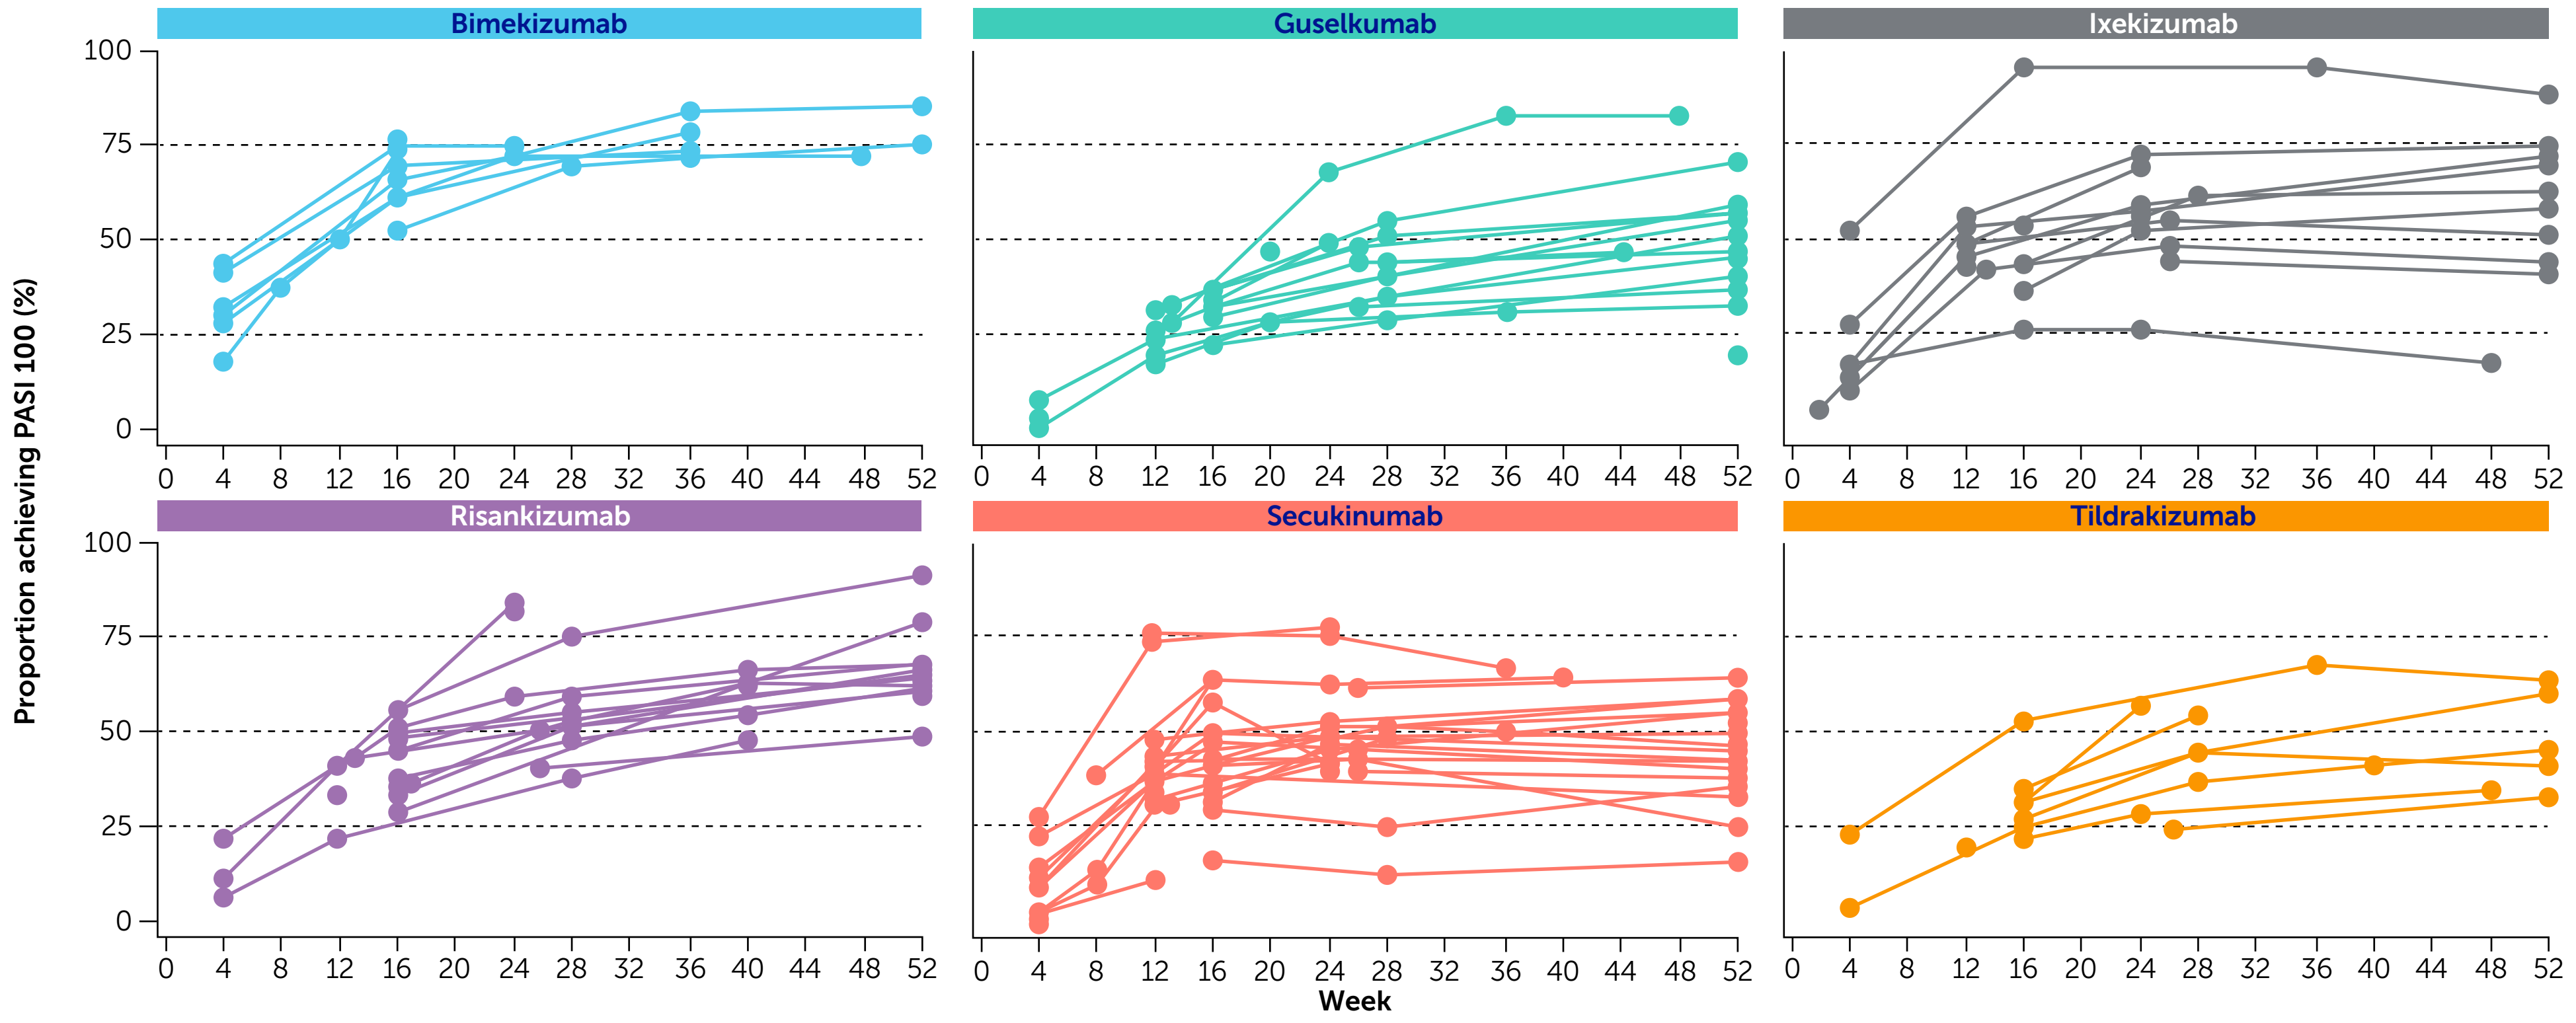
Bimekizumab
Guselkumab
Ixekizumab
Risankizumab
Secukinumab
Tildrakizumab
Proportion achieving PASI 100 (%)
Week

Circles represent individual data points from each publication.

Figure 2 Model-estimated PASI 100 responses over time with 95% confidence intervals

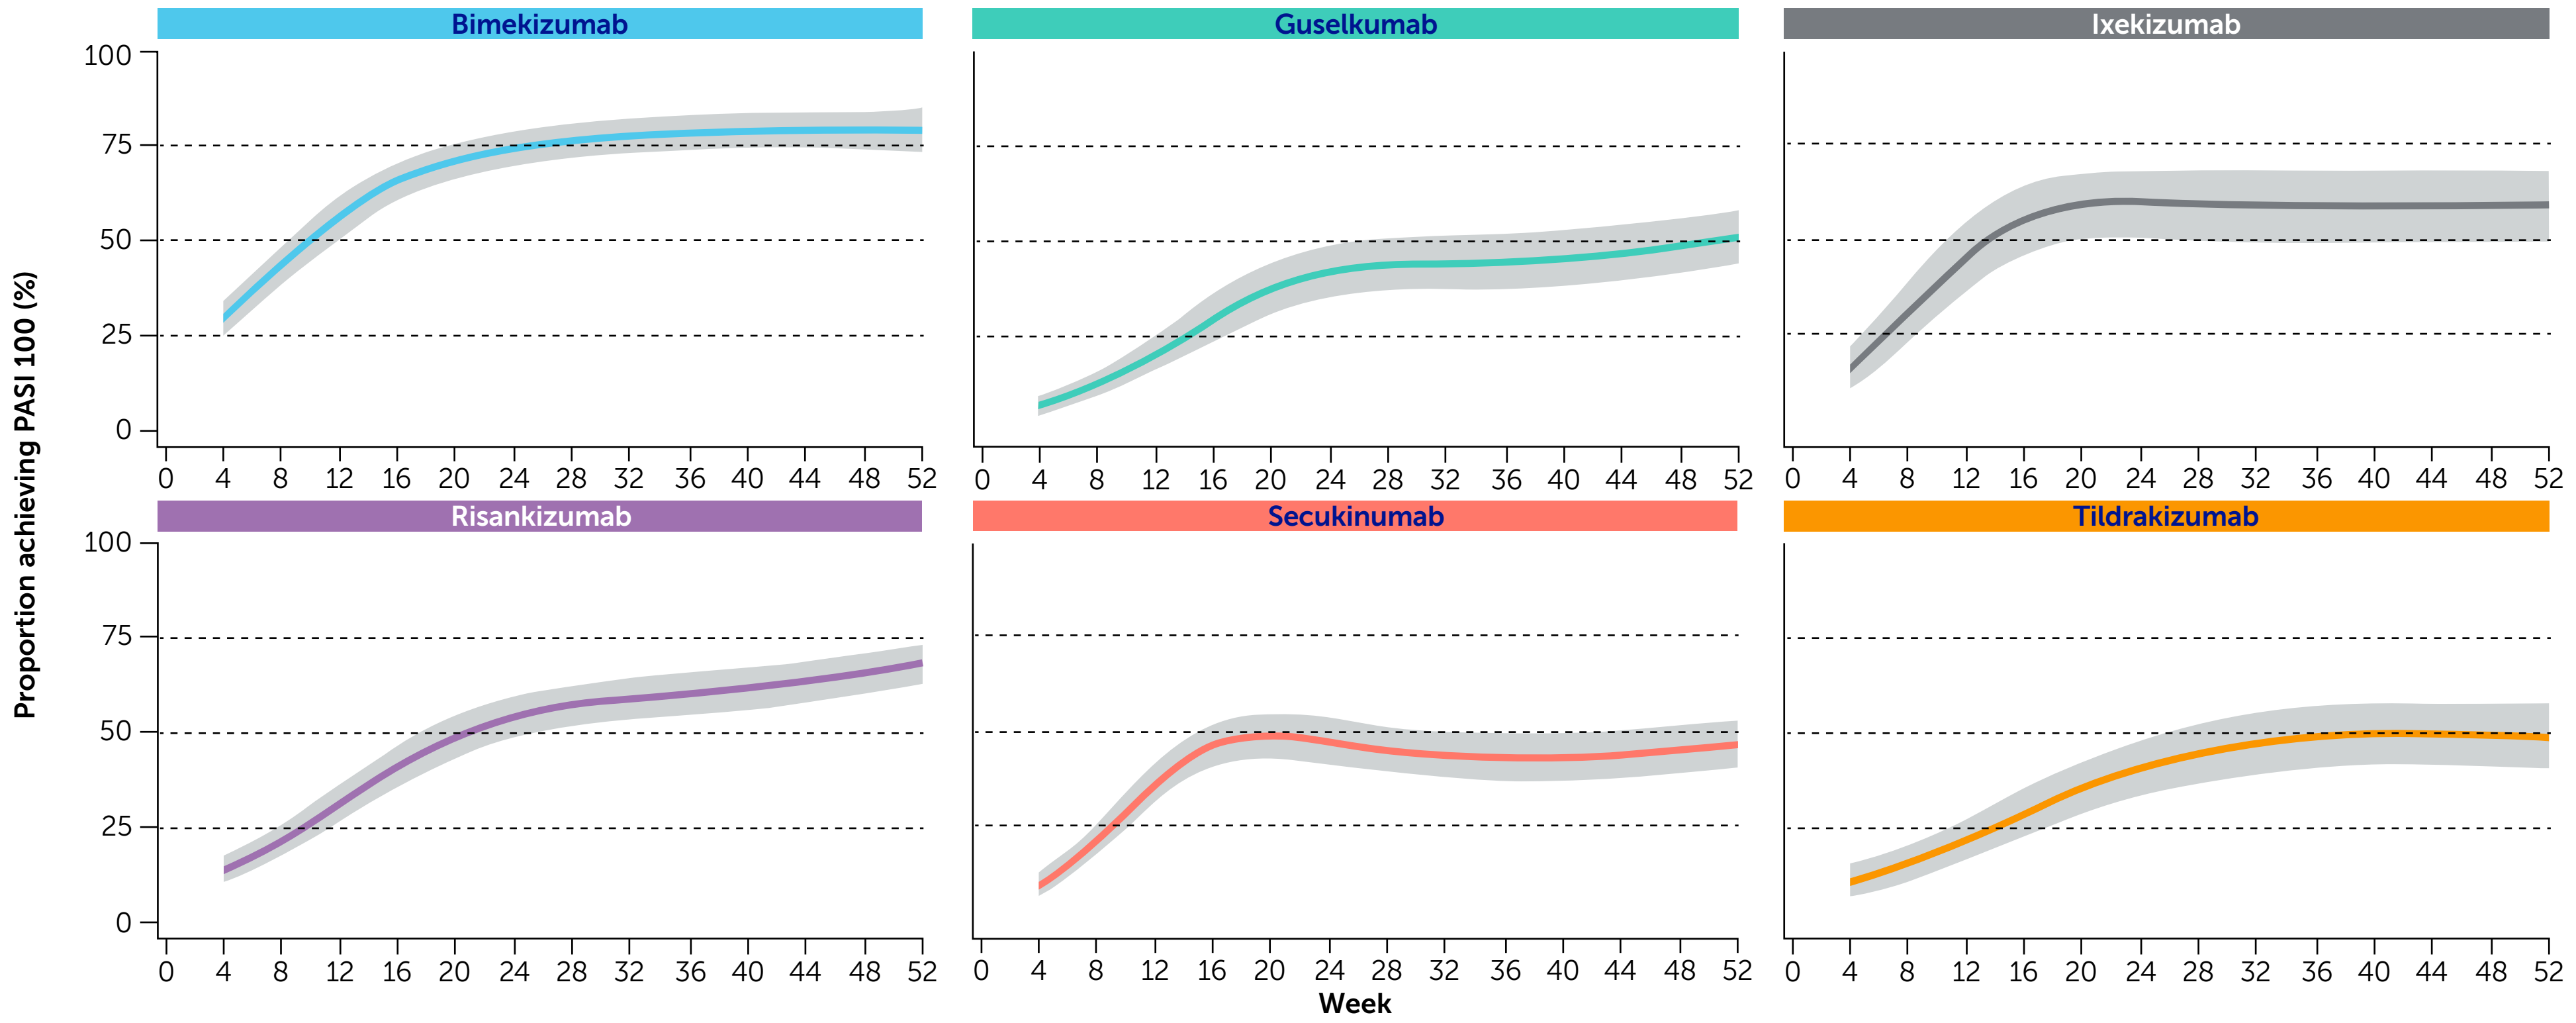
Bimekizumab
Guselkumab
Ixekizumab
Risankizumab
Secukinumab
Tildrakizumab
Proportion achieving PASI 100 (%)
Week

Colored lines shows meta-analysis treatment trajectories. Shaded area shows 95% confidence intervals.

References: [1]Augustin M Dermatol Ther (Heidelb) 2024;14:2841–57; [2]Warren RB N Engl J Med 2021;385:130–41 (NCT03412747); [3]Reich K Lancet 2021;397:487–98 (NCT03370133); [4]Reich K N Engl J Med 2021;385:142–52 (NCT03536884); [5]Armstrong M Dermatol Ther (Heidlb) 2022;12:1777–92. **Author Contributions:** Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **DO**, **YY**, **LC**, **AS**, **BS**, **GH**; Drafting of the publication, or reviewing it critically for important intellectual content: **DO**, **YY**, **LC**, **AS**, **BS**, **GH**; Final approval of the publication: **DO**, **YY**, **LC**, **AS**, **BS**, **GH**. **Author Disclosures:** **DO:** Speaker and/or consultant for AbbVie, Almirall, Boehringer Ingelheim, Bristol Meyers Squibb, Eli Lilly and Company, LEO Pharma, and UCB. **YY:** Research grants, consulting fees, and/or speaker fees from AbbVie, Amgen, Boehringer Ingelheim, Bristol Myers Squibb, Eli Lilly and Company, Janssen, Kyowa Kirin, LEO Pharma, Maruho Pharmaceutical, Novartis, Sun Pharma, Torii Pharmaceutical and UCB. **LC:** External contractor to UCB Pharma. No other conflicts of interest to declare. **AS, BS:** Employees and shareholders of UCB. **GH:** Investigator, consultant, advisor, or speaker for AbbVie, Amgen, Athenex, Boehringer Ingelheim, Bond Avillion, Bristol Myers Squibb, Castle Biosciences, Celgene, Dermavant, Dermtech, Eli Lilly and Company, Incyte, Janssen, LEO Pharma, MC2, Ortho Dermatologics, Pfizer, Regeneron, Sanofi Genzyme, Sun Pharma, and UCB. **Acknowledgements:** This study was funded by UCB. We thank the investigators and their teams who contributed to this study. The authors also acknowledge Inés Dueñas Pousa, PhD, CMPP, of UCB, for editorial review during poster development and publication coordination and Meg Smith, PhD of Costello Medical, Manchester, UK for medical writing and editorial assistance and the Costello Medical Creative team for graphic design assistance. All costs associated with development of this poster were funded by UCB.

To receive a copy of this poster, scan the QR code.

Link expiration:
09 January 2027

Presented at Fall Clinical Dermatology Conference 2026 | October 08–11 | Las Vegas, NV