Concomitant use of glucagon-like peptide-1 receptor agonists in patients with psoriasis or hidradenitis suppurativa receiving bimekizumab: Cross-sectional analysis of US claims data

Martina L. Porter,[1] Ana Filipa Macedo,[2,*] Janek Rudnik,[2] Aijing Shang,[3] Evelina van Ree,[4] Chao Song,[5] Jeremy Schafer,[6] Pablo de la Cueva[7]

[1]Beth Israel Deaconess Medical Center, Department of Dermatology, Harvard Medical School, Boston, Massachusetts, United States; [2]UCB, Brussels, Belgium; [3]UCB, Basel, Switzerland; [4]UCB, Breda, The Netherlands; [5]UCB, Smyrna, Georgia, United States; [6]UCB, Braine l'Alleud, Belgium; [7]Department of Dermatology, Hospital General Universitario Gregorio Marañón, Madrid, Spain.

*Affiliation at the time of study

Objective

To assess the prevalence of concomitant glucagon-like peptide-1 receptor agonist (GLP-1RA) use in bimekizumab (BKZ)-treated patients with psoriasis or hidradenitis suppurativa (HS) and to report the characteristics of those receiving both treatments in a cross-sectional analysis of United States (US) claims data.

Synopsis

- Patients with inflammatory skin diseases, such as psoriasis and HS, have higher rates of comorbid obesity and type 2 diabetes mellitus (T2DM) than the general population.[1,2]
- GLP-1RAs are indicated for the treatment of obesity and T2DM.[3]
- BKZ, a humanized IgG1 monoclonal antibody that selectively inhibits interleukin (IL)-17F in addition to IL-17A,[4] is approved for the treatment of moderate to severe plaque psoriasis and HS.[5,6]
- Evaluating the extent of concomitant GLP-1RA/BKZ use in routine practice, and characterizing patients receiving both treatments, can inform the management of patients with psoriasis or HS and comorbid obesity or T2DM.

Methods

- This observational, cross-sectional analysis using data from the US Komodo Health database included open and closed claims from Medicare and Commercial insurances; the study selection period was between November 1, 2023 and December 7, 2025.
 - Some open claims may not have been adjudicated and so prescriptions may not have been filled; closed claims confirm that the prescription was filled and medication dispensed.
 - BKZ approval for psoriasis in the US was October 2023;[7] BKZ approval for HS in the US was November 2024.[8]
- Eligible patients were ≥18 years, with a psoriasis or HS diagnosis and ≥1 BKZ claim during the selection period.
- Concomitant GLP-1RA/BKZ use was defined as any drug supply overlap within the selection period and index date was the first day of overlap (**Figure 1**).
- The primary endpoint was the prevalence of concomitant GLP-1RA/BKZ use through the whole selection period and by year, evaluated for each indication; data are reported as observed with 95% confidence intervals (CI).
 - Subgroup analyses were performed by sex, age group, and comorbidity.
 - A sensitivity analysis was performed using closed claims only, which was less prone to missing data and gaps in the patient journey.
- The secondary endpoint was patient characteristics at baseline (index date) for those receiving concomitant GLP-1RA/BKZ treatment, summarized descriptively.

Results

- During the selection period, 18,665 patients with psoriasis (60.9% had obesity; 29.4% had T2DM) and 5,082 patients with HS (72.4% had obesity; 31.7% had T2DM) received BKZ treatment, respectively.
- Among these patients, the prevalence of concomitant GLP-1RA use was >20% for both psoriasis and HS (**Figure 2**).
 - The sensitivity analysis produced similar prevalence results that supported the overall estimates.
 - GLP-1RA use was higher in female patients and those aged ≥46–≤65 years; patients with both obesity and T2DM had higher prevalence of GLP-1RA use versus patients with neither (**Figure 3**).
- Baseline characteristics of patients receiving concomitant GLP-1RA/BKZ treatment with psoriasis (N=3,754) and HS (N=1,170) are outlined in **Table 1**.
 - Most patients were biologic-experienced and >95% had a cardiometabolic condition.
- For the psoriasis and HS patient groups, 76.7% and 82.5% initiated BKZ while receiving GLP-1RA treatment, respectively.
- Semaglutide and tirzepatide accounted for ~40% and ~50% of prescribed GLP-1RAs in all BKZ-treated patients, respectively (**Figure 4**).
 - Dermatologists were often the prescribers of BKZ for patients with psoriasis (39.8%) and HS (50.9%) but were rarely the prescribers of GLP-1RAs (psoriasis: 0.4%; HS: 1.3%).

Conclusions

In routine clinical practice between November 1, 2023 and December 7, 2025, concomitant GLP-1RA use in bimekizumab-treated patients with psoriasis or HS increased to >20%. This, along with the high rates of comorbidities in patients receiving concomitant treatment, reflects a complex and multimorbid population.

Most patients with psoriasis or HS initiated bimekizumab while already receiving GLP-1RA treatment, suggesting a continued need for effective treatment of these inflammatory diseases alongside the management of metabolic comorbidities.

Summary

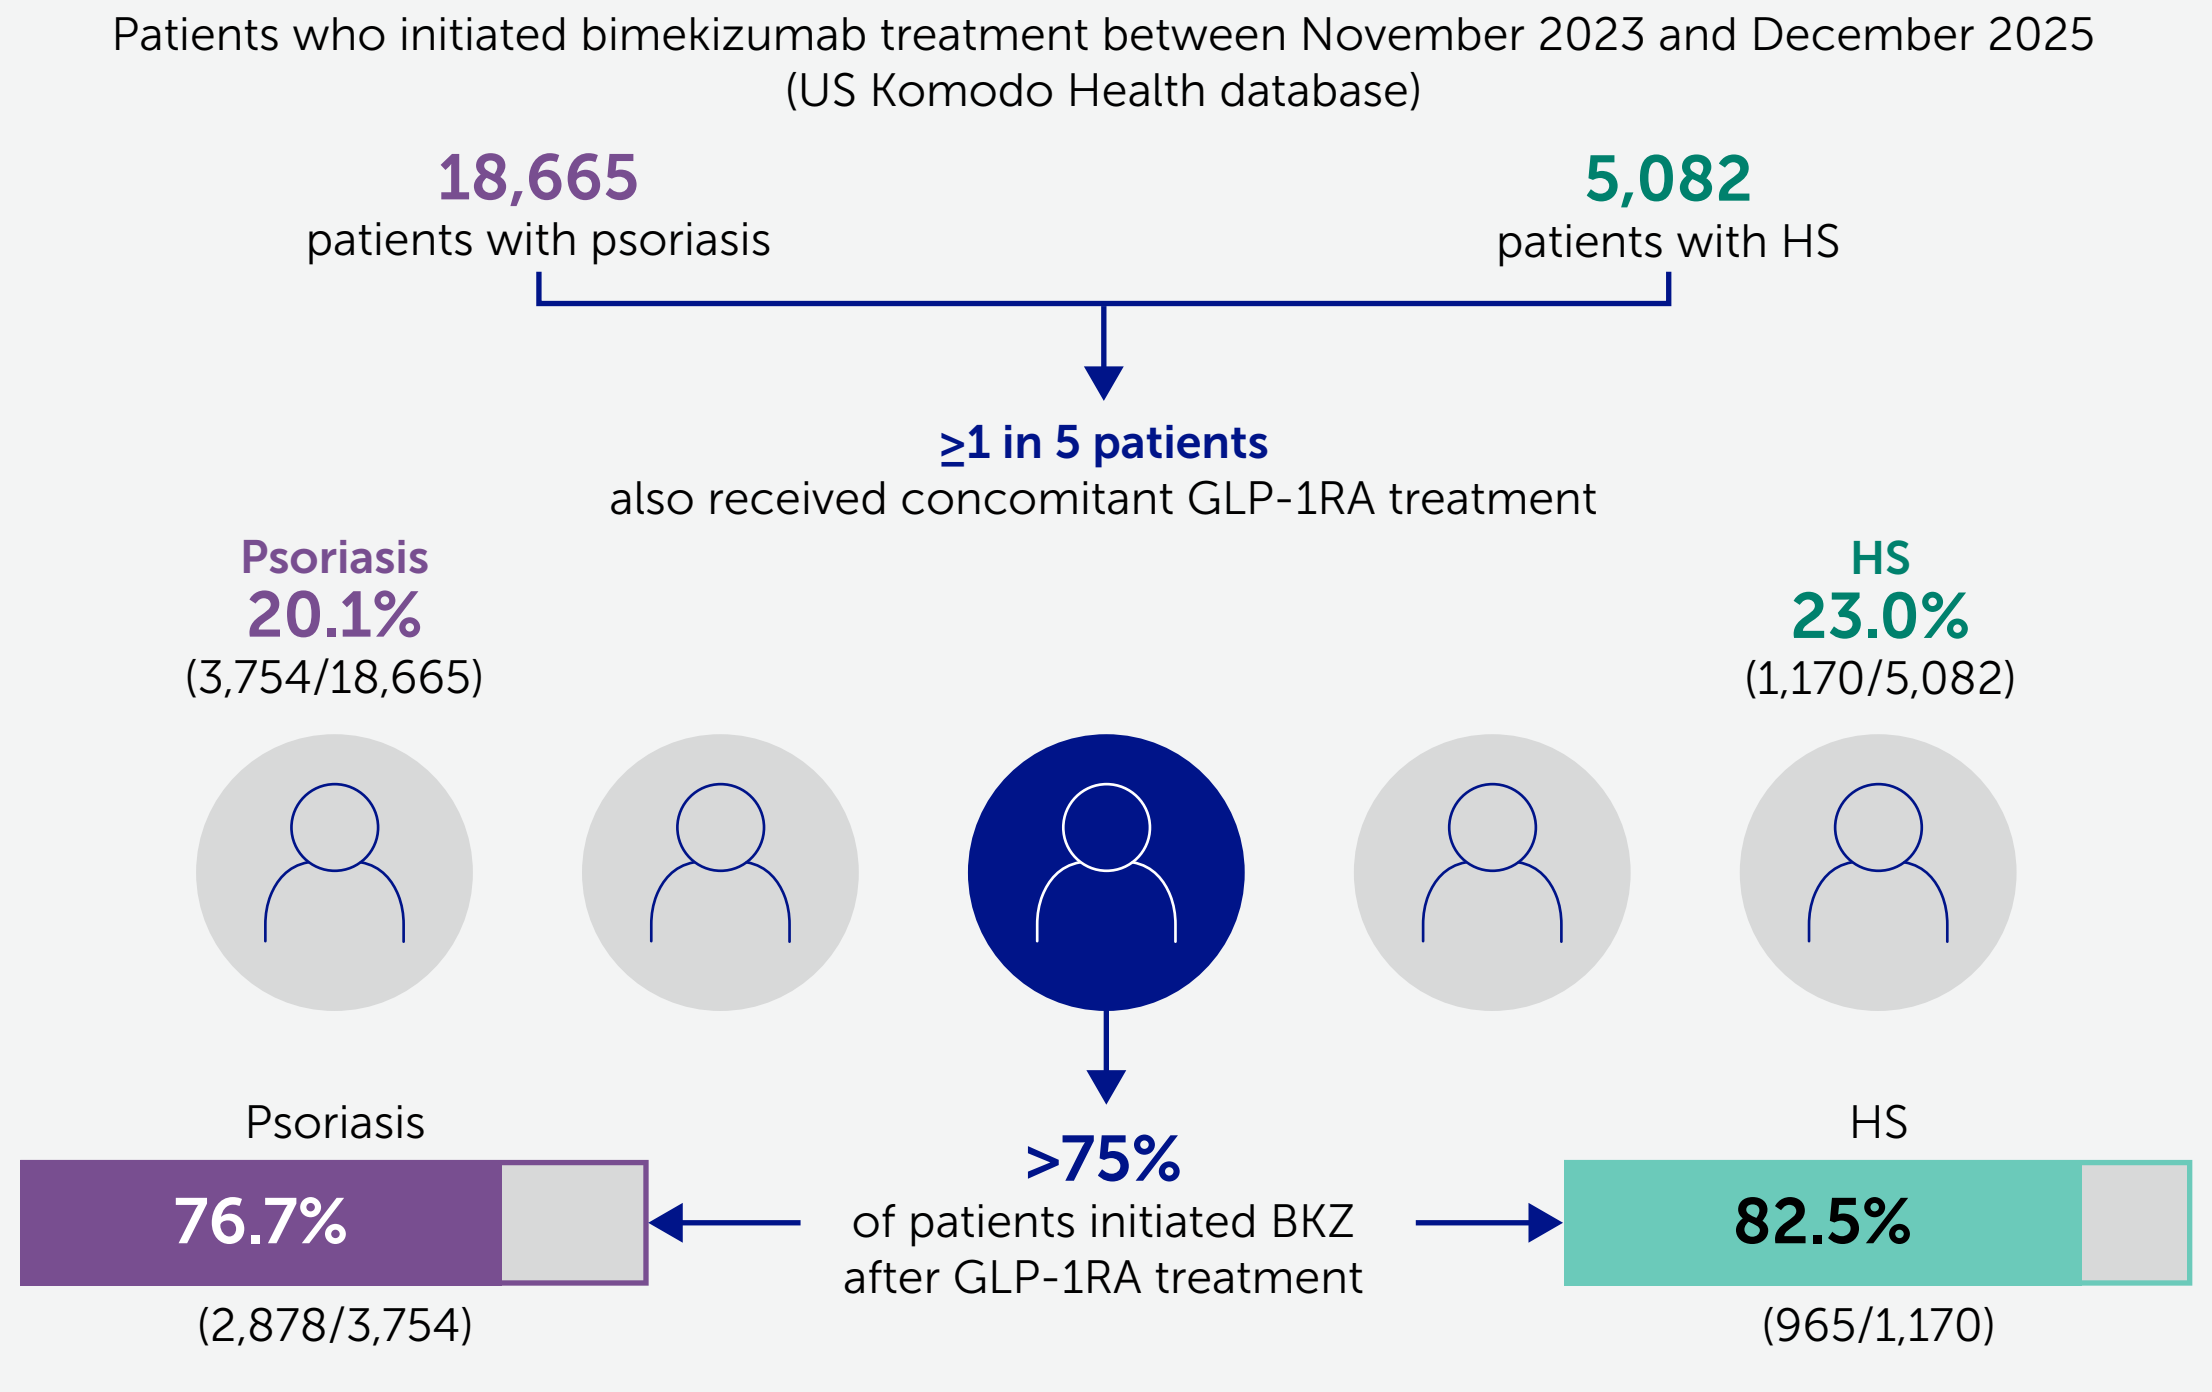
Patients who initiated bimekizumab treatment between November 2023 and December 2025
(US Komodo Health database)
18,665
patients with psoriasis
5,082
patients with HS
≥1 in 5 patients
also received concomitant GLP-1RA treatment
Psoriasis
20.1%
(3,754/18,665)
HS
23.0%
(1,170/5,082)
Psoriasis
76.7%
(2,878/3,754)
>75%
of patients initiated BKZ after GLP-1RA treatment
HS
82.5%
(965/1,170)

Most patients initiated bimekizumab **while already receiving** GLP-1RA treatment, suggesting a **continued need** for effective treatment of these inflammatory diseases **alongside** the management of metabolic comorbidities.

Table 1 Baseline characteristics of patients receiving concomitant GLP-1RA/BKZ treatment

	Patients with psoriasis N=3,754	Patients with HS N=1,170
Age (years), mean (SD)	53.6 (11.0)	46.5 (11.5)
Sex, n (%)		
Female	2,334 (62.2)	869 (74.3)
Male	1,375 (36.6)	292 (25.0)
Missing	45 (1.2)	9 (0.8)
Comorbidities, n (%)		
Any cardiometabolic condition	3,619 (96.4)	1,140 (97.4)
Any cardiovascular condition	1,792 (47.7)	585 (50.0)
Obesity[a]	3,189 (85.0)	1,066 (91.1)
T2DM	2,428 (64.7)	748 (63.9)
Both obesity and T2DM	2,122 (56.5)	696 (59.5)
Biologic-experienced, n (%)	3,363 (89.6)	990 (84.6)
1 prior biologic therapy	852 (22.7)	374 (32.0)
2 prior biologic therapies	908 (24.2)	412 (35.2)
≥3 prior biologic therapies	1,603 (42.7)	204 (17.4)

Baseline characteristics at index date. Dates of psoriasis or HS diagnosis were not available prior to the patient having data in the Komodo database; therefore, mean time since diagnosis could not be calculated. **[a]** Determined using International Classification of Diseases codes that describe a diagnosis of obesity.

Figure 1 Study design

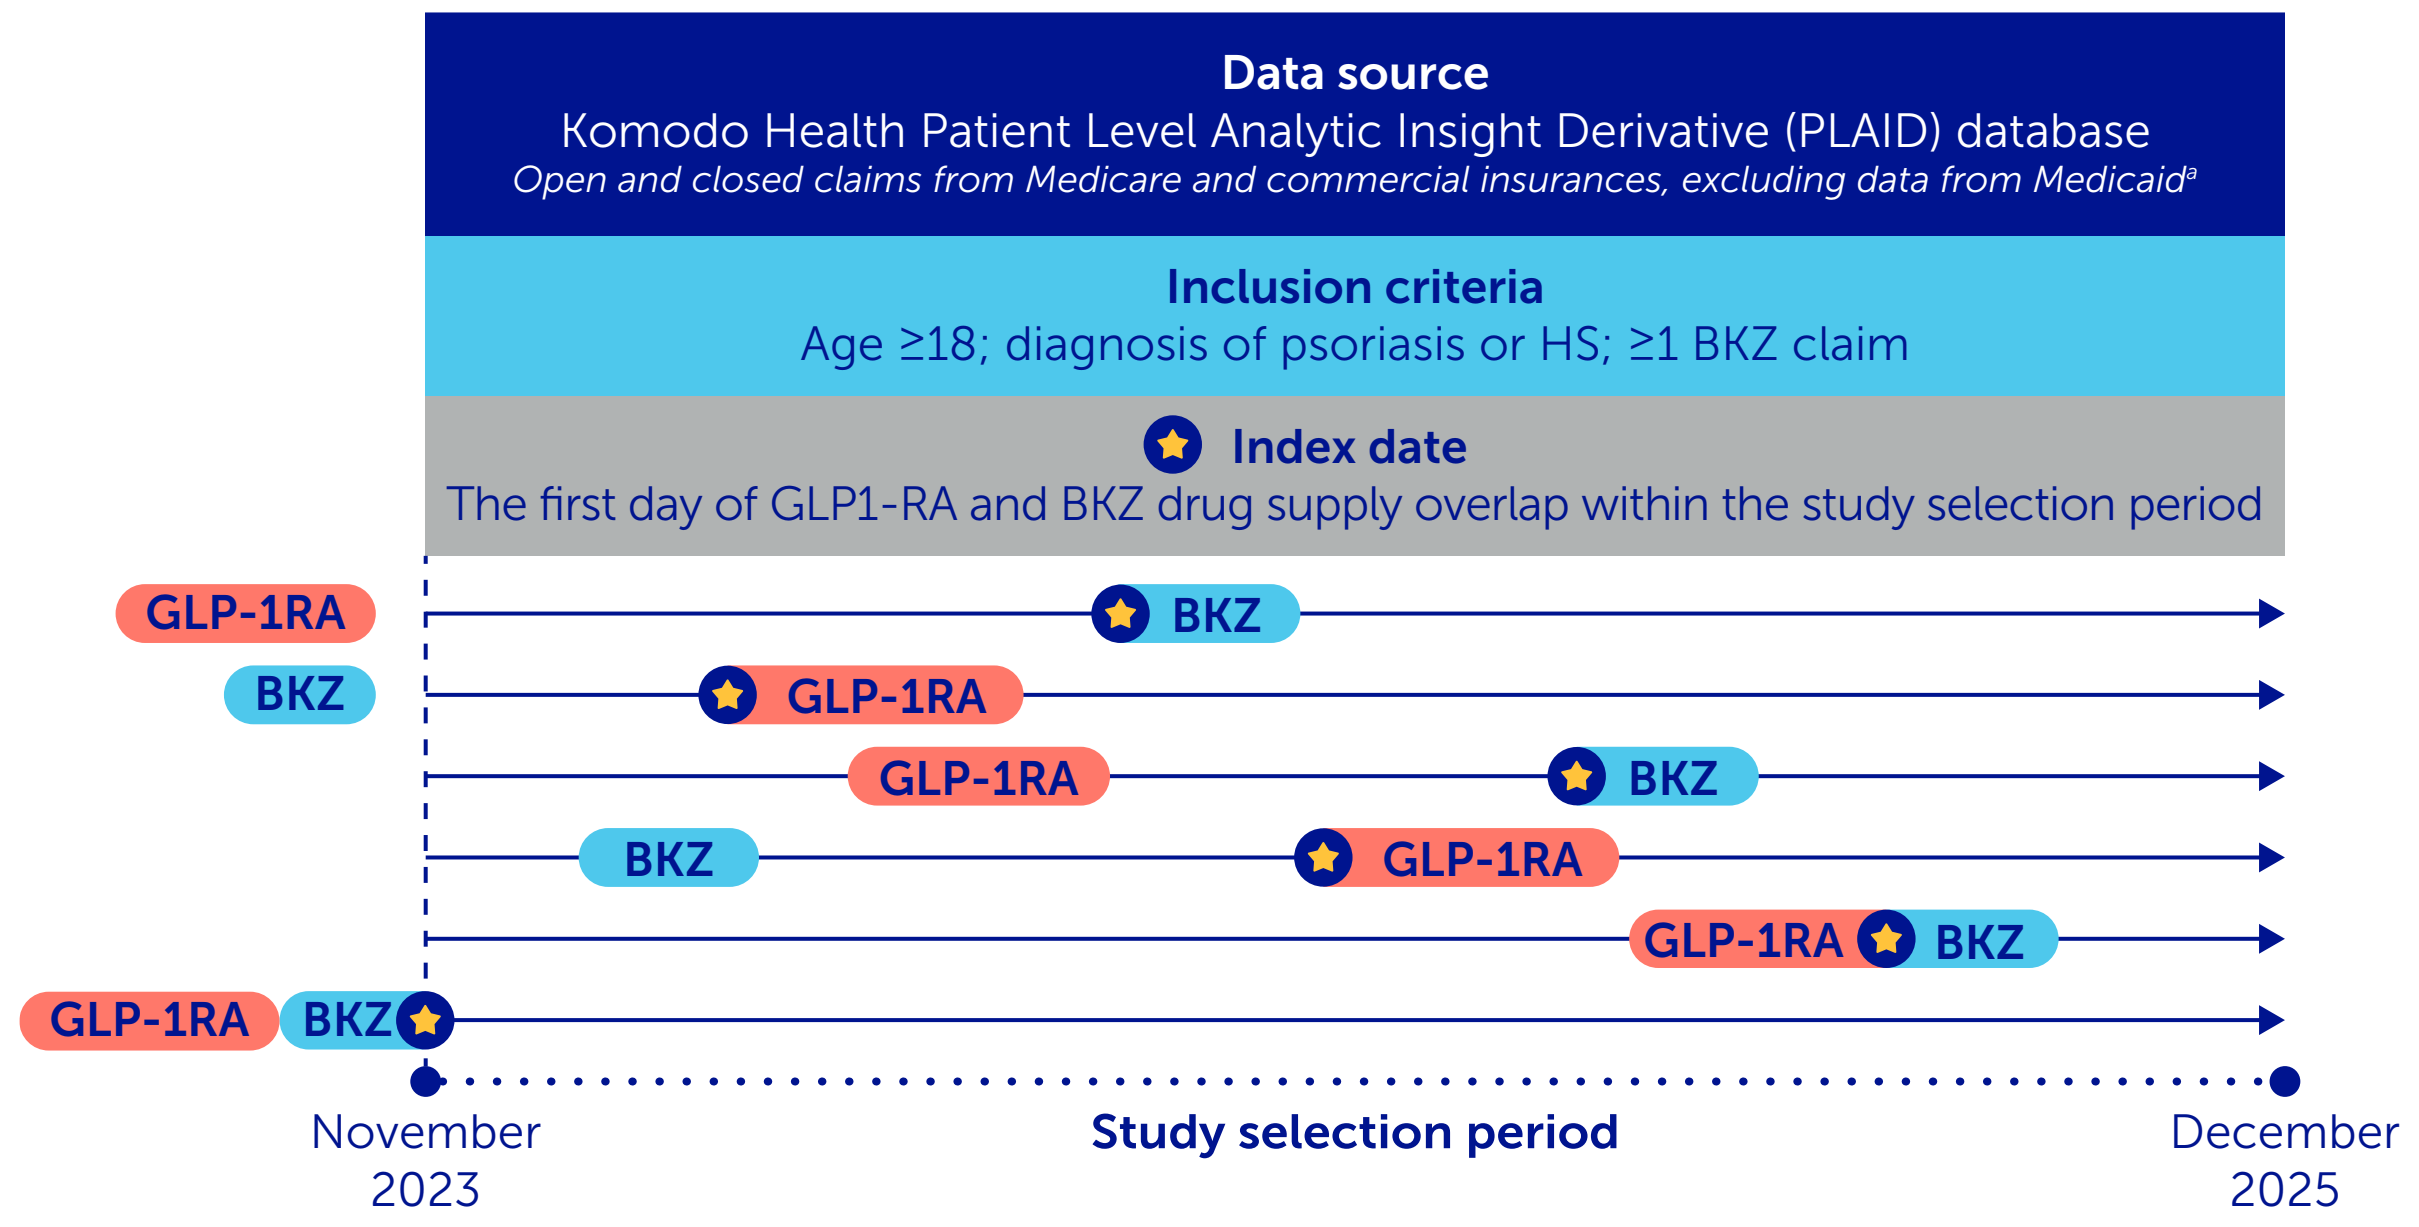
Data source
Komodo Health Patient Level Analytic Insight Derivative (PLAID) database
Open and closed claims from Medicare and commercial insurances, excluding data from Medicaid[a]
Inclusion criteria
Age ≥18; diagnosis of psoriasis or HS; ≥1 BKZ claim
Index date
The first day of GLP1-RA and BKZ drug supply overlap within the study selection period
GLP-1RA
BKZ
November 2023
Study selection period
December 2025

For patients with multiple episodes of concomitant GLP-1RA/BKZ use, the first drug supply overlap was the index date. For patients with drug supply overlap before the start of the selection period, the index date was November 1, 2023. **[a]** Data from Medicaid were excluded since this insurance does not cover GLP-1RA use for weight loss in most US states.

Figure 2 Prevalence of concomitant GLP-1RA use in BKZ-treated patients with psoriasis or HS (primary endpoint)

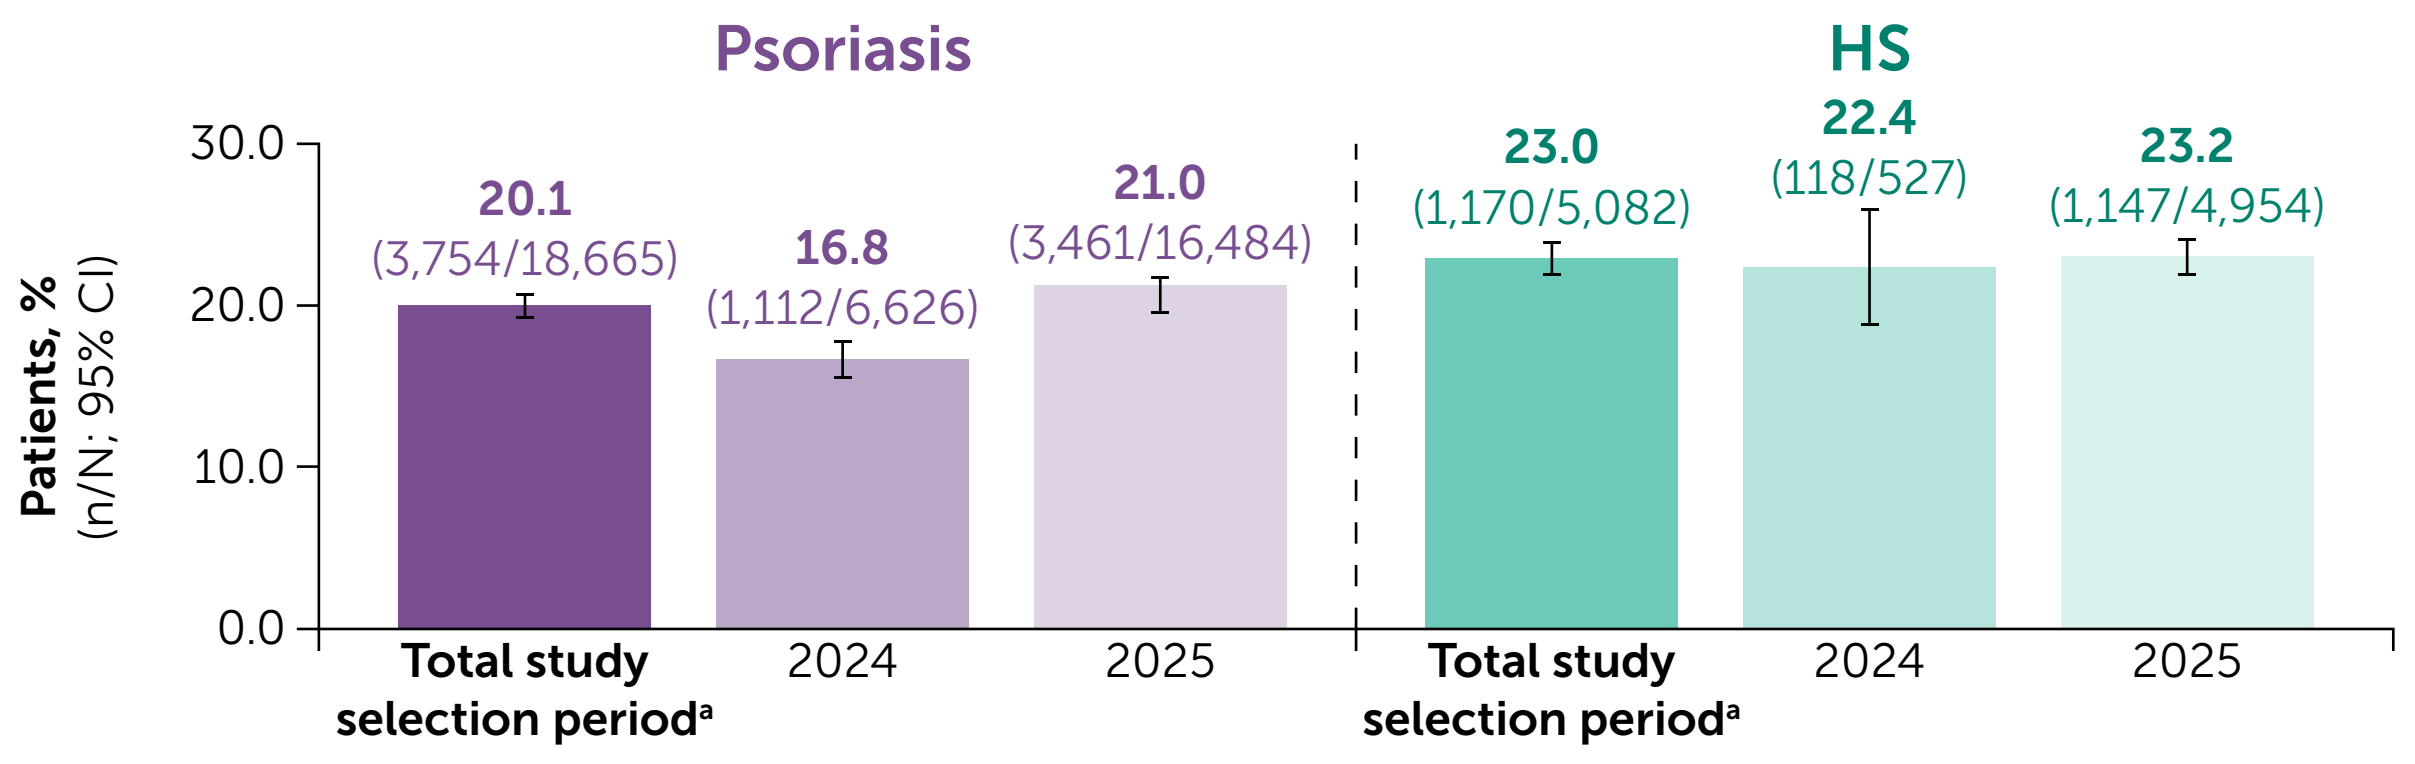
Psoriasis
HS
Patients, % (n/N; 95% CI)
30.0
20.0
10.0
0.0
20.1
(3,754/18,665)
16.8
(1,112/6,626)
21.0
(3,461/16,484)
23.0
(1,170/5,082)
22.4
(118/527)
23.2
(1,147/4,954)
Total study selection period[a]
2024
2025
Total study selection period[a]
2024
2025

Sensitivity analysis (total study selection period; closed claims only)	Psoriasis N=9,080	HS N=2,397
Patients, % (95% CI)	20.7 (19.9, 21.6)	24.9 (23.1, 26.6)

Bimekizumab was approved in the US for the treatment of psoriasis in October 2023, and HS in November 2024.[5] **[a]** Study selection period: November 1, 2023–December 7, 2025; due to small patient numbers in the HS group in 2023 (off-label use or use for psoriasis as a comorbidity), 2023 data are not presented separately in this figure (psoriasis: 12.2% [34/279]; HS: 29.4% [5/17]).

Figure 3 Prevalence of concomitant GLP-1RA use in BKZ-treated patients with psoriasis or HS **by subgroup**

A) Psoriasis

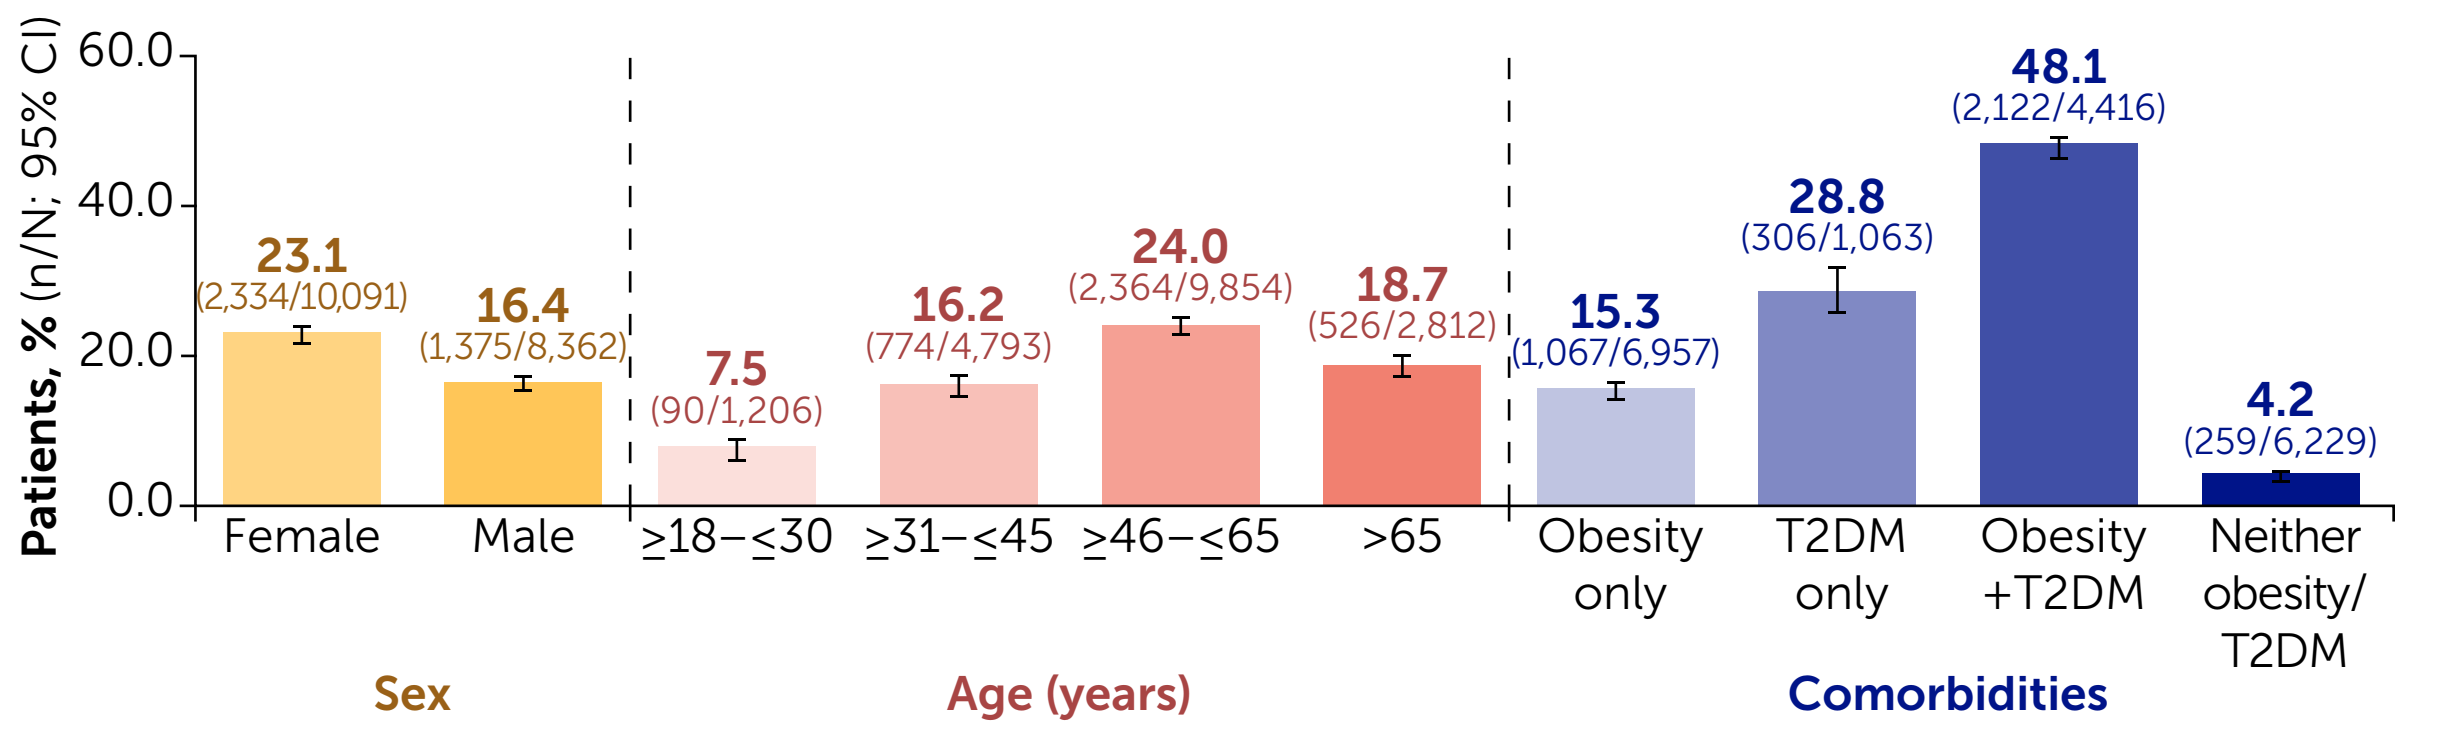
Patients, % (n/N; 95% CI)
60.0
40.0
20.0
0.0
23.1
(2,334/10,091)
16.4
(1,375/8,362)
7.5
(90/1,206)
16.2
(774/4,793)
24.0
(2,364/9,854)
18.7
(526/2,812)
15.3
(1,067/6,957)
28.8
(306/1,063)
48.1
(2,122/4,416)
4.2
(259/6,229)
Female
Male
≥18–≤30
≥31–≤45
≥46–≤65
>65
Obesity only
T2DM only
Obesity +T2DM
Neither obesity/ T2DM
Sex
Age (years)
Comorbidities

B) HS

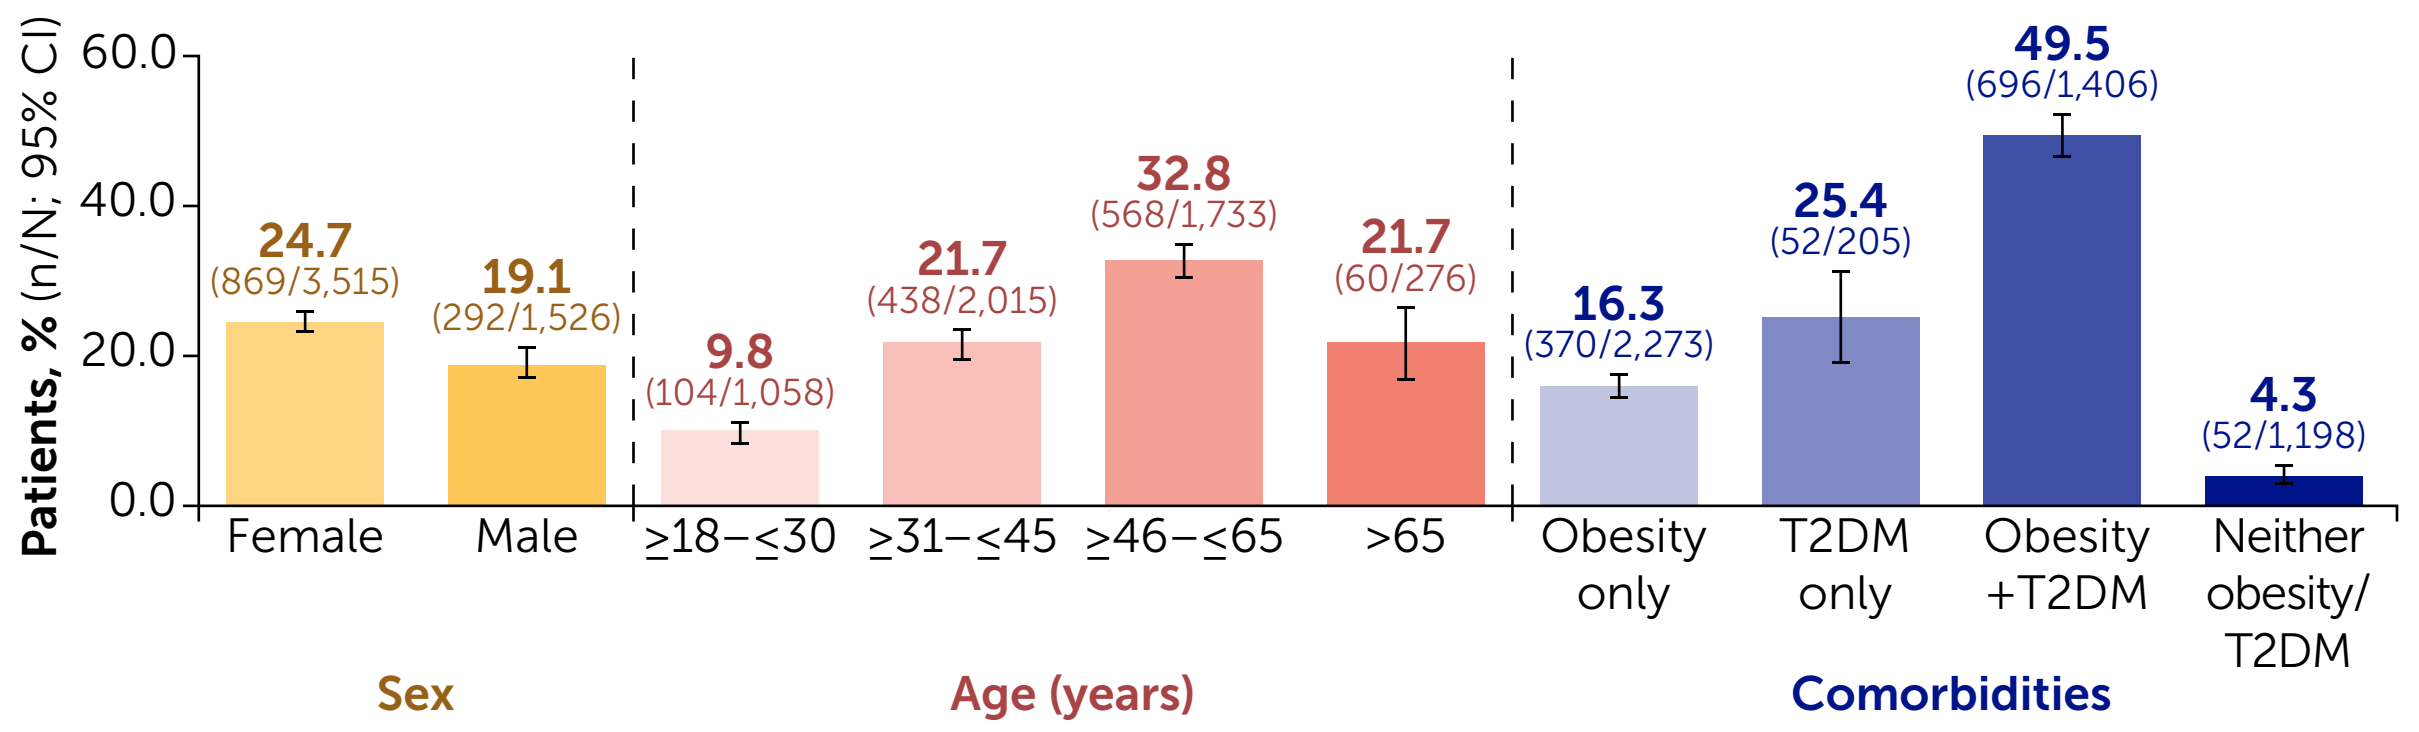
Patients, % (n/N; 95% CI)
60.0
40.0
20.0
0.0
24.7
(869/3,515)
19.1
(292/1,526)
9.8
(104/1,058)
21.7
(438/2,015)
32.8
(568/1,733)
21.7
(60/276)
16.3
(370/2,273)
25.4
(52/205)
49.5
(696/1,406)
4.3
(52/1,198)
Female
Male
≥18–≤30
≥31–≤45
≥46–≤65
>65
Obesity only
T2DM only
Obesity +T2DM
Neither obesity/ T2DM
Sex
Age (years)
Comorbidities

Data reported for total study selection period: November 1, 2023–December 7, 2025.

Figure 4 Types of prescribed GLP-1RAs and prescriber/insurance types in BKZ-treated patients

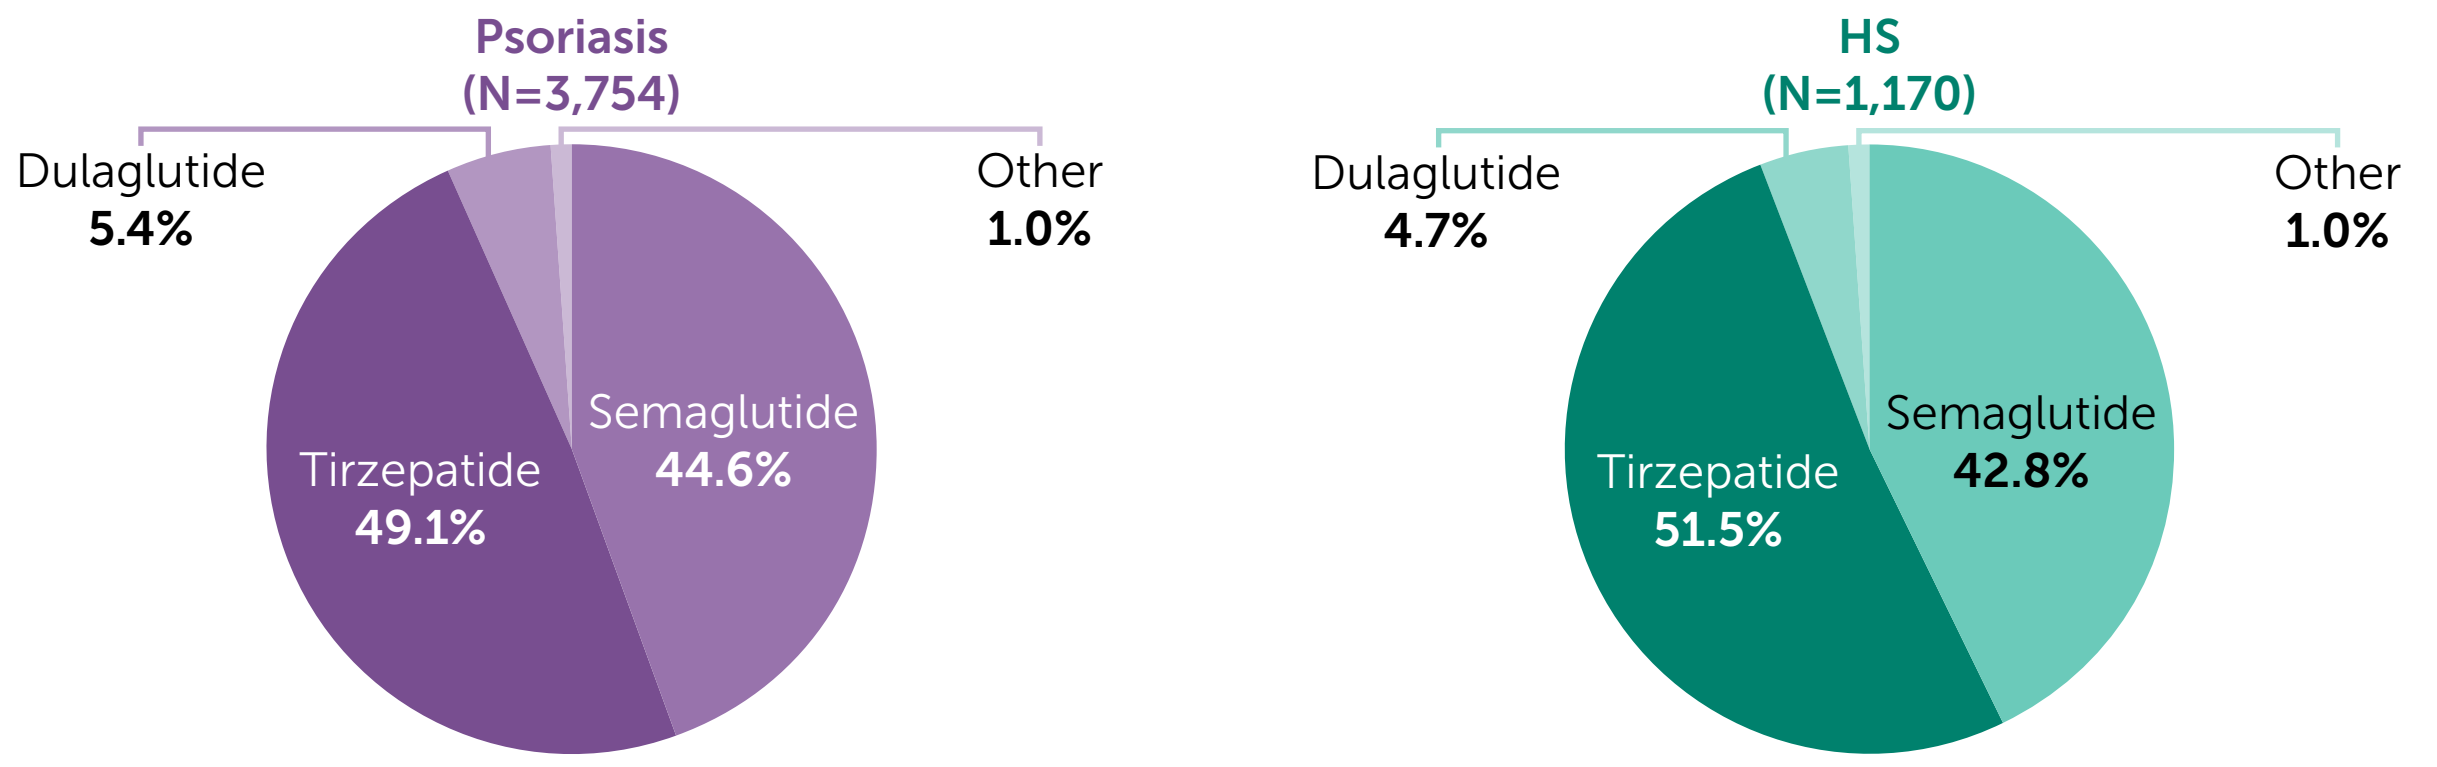
Psoriasis
(N=3,754)
Dulaglutide
5.4%
Other
1.0%
Semaglutide
44.6%
Tirzepatide
49.1%
HS
(N=1,170)
Dulaglutide
4.7%
Other
1.0%
Semaglutide
42.8%
Tirzepatide
51.5%

	Psoriasis N=3,754	HS N=1,170
GLP-1RA prescriber type, n (%)		
Internal medicine	1,233 (32.8)	352 (30.1)
Family medicine	1,084 (28.9)	311 (26.6)
Nurse practitioner	890 (23.7)	310 (26.5)
Dermatologist	16 (0.4)	15 (1.3)
Other	533 (14.2)	182 (15.6)
Insurance type,[a] n (%)		
Commercial	2,822 (75.2)	870 (74.4)
Medicare	702 (18.7)	232 (19.8)
Commercial/Medicare	116 (3.1)	23 (2.0)
Missing	114 (3.0)	45 (3.9)

[a] Data from Medicaid were excluded since this insurance does not cover GLP-1RA use for weight loss in most US states.

BKZ: bimekizumab; **CI:** confidence interval; **GLP-1RA:** glucagon-like peptide-1 receptor agonist; **HS:** hidradenitis suppurativa; **IgG:** Immunoglobulin; **IL:** interleukin; **SD:** standard deviation; **T2DM:** type 2 diabetes mellitus; **US:** United States.

References: [1]Reich K et al. Arch Dermatol Res 2015;307:875–83; [2]Elzawawi KE et al. Int Wound J 2024;21:e70035; [3]Nasreen A et al. eClinicalMedicine 2024;75:102782; [4]Adams R et al. Front Immunol 2020;11:1894; [5]FDA. BIMZELX® Highlights of Prescribing Information. Available at: https://www.accessdata.fda.gov/drugsatfda_docs/label/2024/761151s010lbl.pdf [Accessed September 2026]; [6]EMA. BIMZELX® SUMMARY OF PRODUCT CHARACTERISTICS. Available at: https://www.ema.europa.eu/en/documents/product-information/bimzelx-epar-product-information_en.pdf [Accessed September 2026]; [7]UCB News. BIMZELX® FDA Psoriasis Approval 2023. Available at: https://www.ucb.com/sites/default/files/press_files/93a57a7451a70708.pdf [Accessed September 2026]; [8]UCB News. BIMZELX® FDA Hidradenitis Suppurativa Approval 2024. Available at: https://www.ucb.com/sites/default/files/press_files/a9cdd0e5f6ae72fc.pdf [Accessed September 2026]. **Author Contributions:** Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **MLP**, **AFM**, **JR**, **AS**, **EvR**, **CS**, **JS**, **PdlC**; Drafting of the publication, or reviewing it critically for important intellectual content: **MLP**, **AFM**, **JR**, **AS**, **EvR**, **CS**, **JS**, **PdlC**; Final approval of the publication: **MLP**, **AFM**, **JR**, **AS**, **EvR**, **CS**, **JS**, **PdlC**. **Author Disclosures: MLP:** Consultant and investigator for AbbVie, Arcutis, Aristea Therapeutics, Avalo Therapeutics, Eli Lilly and Company, Incyte, Janssen, Merck, MoonLake Immunotherapeutics, Navigator Biosciences, Novartis, Pfizer, Prometheus, Sanofi, Sonoma Biotherapeutics, UCB, Ventyx and Zura Bio; consultant for Almirall, FIDE, Insmed, Oruka, Otsuka, Propeller Biosciences, Spyre, and Trifecta Clinical/WCG; investigator for AnaptysBio, Bayer, Bristol Myers Squibb, OASIS Pharmaceuticals, and Regeneron; received royalties from Beth Israel Deaconess Medical Center; received fellowship funding to institution from AbbVie. **AFM:** Consultant for UCB. **JR:** Contractor for UCB. **AS**, **EvR**, **CS**, **JS:** Employees and shareholders of UCB. **PdlC:** Served as a consultant, speaker and/or investigator for AbbVie, Almirall, Boehringer Ingelheim, Bristol Myers Squibb, Celgene, Eli Lilly and Company, Johnson & Johnson, LEO Pharma, MSD, Novartis, Pfizer, Roche, Sanofi, and UCB. **Acknowledgements:** This study was funded by UCB. The authors thank all patients included in the database used in this study. The authors acknowledge Inés Dueñas Pousa, PhD, of UCB, for editorial review during poster development and publication coordination, Oscar Jenkyn-Jones, MSc, of Costello Medical, United Kingdom, for medical writing and editorial assistance and the Costello Medical Creative team for design support. All costs associated with development of this poster were funded by UCB.

To receive a copy of this poster, scan the QR code.

Link expiration: January 9, 2027

Presented at Fall Clinical Dermatology Conference 2026 | October 08–11 | Las Vegas, NV

Previously presented at EADV 2026