AANEM Poster **289** | MGFA Scientific Session Poster **90**

Achievement of minimal symptom expression in patients with generalized myasthenia gravis receiving zilucoplan: A 120-week *post hoc* analysis of RAISE-XT

James F. Howard Jr.[1], Saskia Bresch[2], Miriam Freimer[3], M. Isabel Leite[4], Angelina Maniaol[5], Kimiaki Utsugisawa[6], Tuan Vu[7], Michael D. Weiss[8], Joel Baldwin[9], Babak Boroojerdi[10], Natasa Savic[11], Channa Hewamadduma[12,13], on behalf of the RAISE-XT study team

[1]Department of Neurology, The University of North Carolina at Chapel Hill, Chapel Hill, NC, USA; [2]Service de Neurologie, Hospital Pasteur, Centre Hospitalier Universitaire de Nice, Nice, France; [3]Department of Neurology, The Ohio State University Wexner Medical Center, Columbus, OH, USA; [4]Nuffield Department of Clinical Neurosciences, University of Oxford, Oxford, UK; [5]Department of Neurology, Oslo University Hospital, Oslo, Norway; [6]Department of Neurology, Hanamaki General Hospital, Hanamaki, Japan; [7]Department of Neurology, University of South Florida Morsani College of Medicine, Tampa, FL, USA; [8]Department of Neurology, University of Washington Medical Center, Seattle, WA, USA; [9]UCB, Slough, UK; [10]UCB, Monheim, Germany; [11]UCB, Bulle, Switzerland; [12]Academic Neuromuscular Unit, Sheffield Teaching Hospitals NHS Foundation Trust, Sheffield, UK; [13]Sheffield Institute for Translational Neuroscience (SITraN), School of Medicine and Population Health, University of Sheffield, Sheffield, UK

AANEM & MGFA Scientific Session 2026, Orlando, FL, USA; September 29–October 2, 2026

Introduction

- Zilucoplan is a small (15-amino acid) potent macrocyclic peptide complement C5 inhibitor approved for the treatment of adults with anti-AChR Ab+ gMG[1,2]
- Zilucoplan showed a rapid (Week 1) and complete complement inhibition (>95%), which was sustained long-term with daily dosing.[3,4] Additionally, approximately 90% complement inhibition was achieved within 3 hours after the first administration of zilucoplan[1]
- Zilucoplan mean plasma terminal elimination half-life was approximately 7–8 days,[3] which allows for a fast reversible effect if intact complement activity is urgently required
- The efficacy and safety of zilucoplan have been previously demonstrated in two randomized, placebo-controlled, double-blind studies (Phase 2 [NCT03315130]; Phase 3 [RAISE, NCT04115293]), and were further explored in RAISE-XT (NCT04225871), a Phase 3 OLE study[2,4]
- Achievement of MSE, defined as an MG-ADL score of 0 or 1, is being increasingly recognized as a treatment goal for patients with gMG[5]
- **This *post hoc* analysis of the Phase 2, RAISE and RAISE-XT studies assessed MSE response rates in patients being treated with zilucoplan, analyzed up to 120 weeks**

Methods

- In RAISE, adults with MGFA Disease Class II–IV anti-AChR Ab+ gMG were randomized 1:1 to once-daily subcutaneous zilucoplan 0.3 mg/kg or placebo for 12 weeks
- Patients completing RAISE, or the qualifying Phase 2 study, could enter RAISE-XT to self-administer once-daily subcutaneous injections of zilucoplan 0.3 mg/kg
 - The primary outcome of RAISE-XT was incidence of TEAEs
- In this *post hoc* analysis, data were pooled from patients who received zilucoplan 0.3 mg/kg or placebo in the double-blind studies. The following parameters were assessed:
 - Double-blind baseline MG-ADL scores in patients stratified by MSE achievement (MG-ADL score of 0 or 1, without rescue therapy) while on zilucoplan up to Week 120 (data cutoff: November 11, 2023)
 - Time to MSE without rescue therapy after initiating zilucoplan treatment among all patients considering 120 weeks of on-treatment follow-up (Kaplan–Meier analysis), and among patients who achieved MSE at any point, up to Week 120 (individual patient observed data)

Results

- Of 200 patients enrolled in RAISE-XT, 183 received zilucoplan 0.3 mg/kg or placebo in the double-blind studies
- Baseline demographics and disease characteristics were indicative of a broad population of patients with mild-to-severe gMG (**Table 1**)
- The cumulative proportion of patients achieving MSE over 120 weeks of follow up after zilucoplan initiation was 62.7% (**Figure 1**; Kaplan-Meier analysis)
- Patients who achieved MSE had MG-ADL scores of 2–18 at double-blind baseline (mean [SD]: 9.7 [3.0], n=102) and patients who did not achieve MSE had baseline scores of 6–19 (11.0 [2.9], n=81; **Figure 2**)
- Among patients who achieved MSE, MSE was first attained as early as 1 week after initiating zilucoplan treatment (**Figure 3**)
 - Within 12 weeks of initiating treatment, 58.8% (n/N=60/102) of these patients had achieved MSE
 - More than three-quarters of patients (78.4% [n/N=80/102]) achieved MSE within 24 weeks
- Overall, 730 observations of MSE were recorded among all patients who achieved MSE, looking up to Week 120; 61.2% (n/N=447/730) of observed events had no residual symptoms reported (MG-ADL score of 0)
 - An analysis of MSE events with an MG-ADL score of 1 showed that ocular items were the most common residual symptoms (33.6%, comprising 13.8% with residual double vision and 19.8% with residual eyelid droop)
- TEAEs occurred in 97.0% (n/N=194/200) of patients and most were mild or moderate

Summary and conclusions

This *post hoc* analysis of pooled Phase 2, RAISE and RAISE-XT data assessed MSE response in patients with anti-AChR Ab+ gMG treated with zilucoplan and analyzed up to Week 120

MG-ADL scores at double-blind baseline were similar between patients who did and did not achieve MSE. Patients with a broad range of scores, up to 18, achieved MSE during zilucoplan treatment

Among patients who achieved MSE, some attained MSE as early as 1 week after zilucoplan initiation and the majority (58.8%) did so within 12 weeks; more than three-quarters of patients (78.4%) achieved MSE within 24 weeks

These data demonstrate that while most patients had a rapid MSE response to zilucoplan, some patients achieved MSE later, supporting the long-term use of zilucoplan

Figure 1 MSE was attained as rapidly as 1 week after initiating zilucoplan treatment

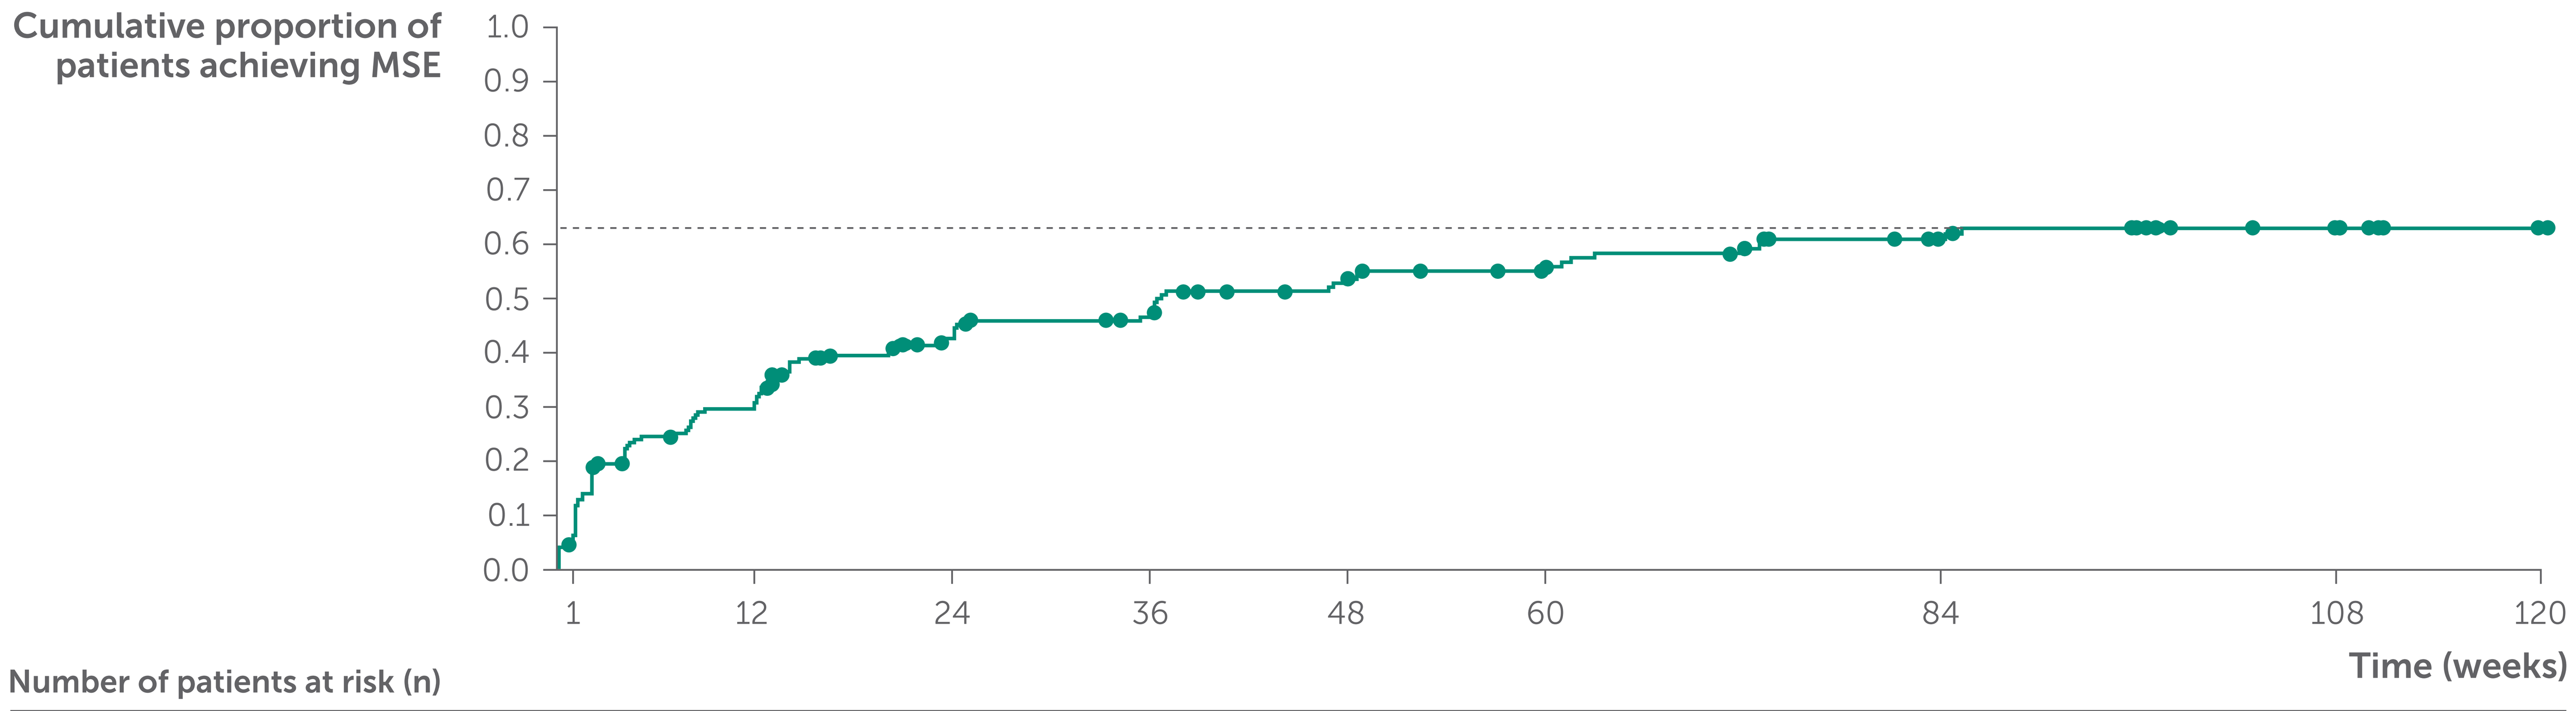
Cumulative proportion of patients achieving MSE
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
1
12
24
36
48
60
84
108
120
Time (weeks)

Number of patients at risk (n)									
Zilucoplan 0.3 mg/kg pooled (N=183)	174	125	89	79	64	56	40	25	–

mITT population, which included all patients enrolled in RAISE-XT who received at least one dose of zilucoplan and had at least one post-dosing MG-ADL score. Time to MSE was defined as the time from the start of zilucoplan treatment (double-blind baseline for patients treated with zilucoplan in the double-blind studies, and RAISE-XT baseline for patients treated with placebo in the double-blind studies).

Table 1 Baseline demographics and disease characteristics were indicative of a broad population of patients with mild-to-severe gMG

		Zilucoplan 0.3 mg/kg pooled (N=183)
	Age, years, mean (SD)	**52.9** (15.0)
	Sex, male, n (%)	**83** (45.4)
MGFA Clinical Class, n (%)	II	**54** (29.5)
	III	**117** (63.9)
	IV	**12** (6.6)
	MG-ADL score, mean (SD)	**10.3** (3.0)
	QMG score, mean (SD)	**19.0** (4.1)
	Prior thymectomy, n (%)	**88** (48.1)
	Prior MG crisis, n (%)	**59** (32.2)
	Duration of disease from diagnosis, years, mean (SD)	**9.1** (9.9)
gMG-specific therapies, n (%)	Cholinesterase inhibitors	**150** (82.0)
	Corticosteroids	**120** (65.6)
	Immunosuppressants	**94** (51.4)

mITT population, which included all patients enrolled in RAISE-XT who received at least one dose of zilucoplan and had at least one post-dosing MG-ADL score. Data obtained from double-blind baseline.

Figure 2 MG-ADL scores at double-blind baseline were broadly similar between patients who achieved MSE and those who did not

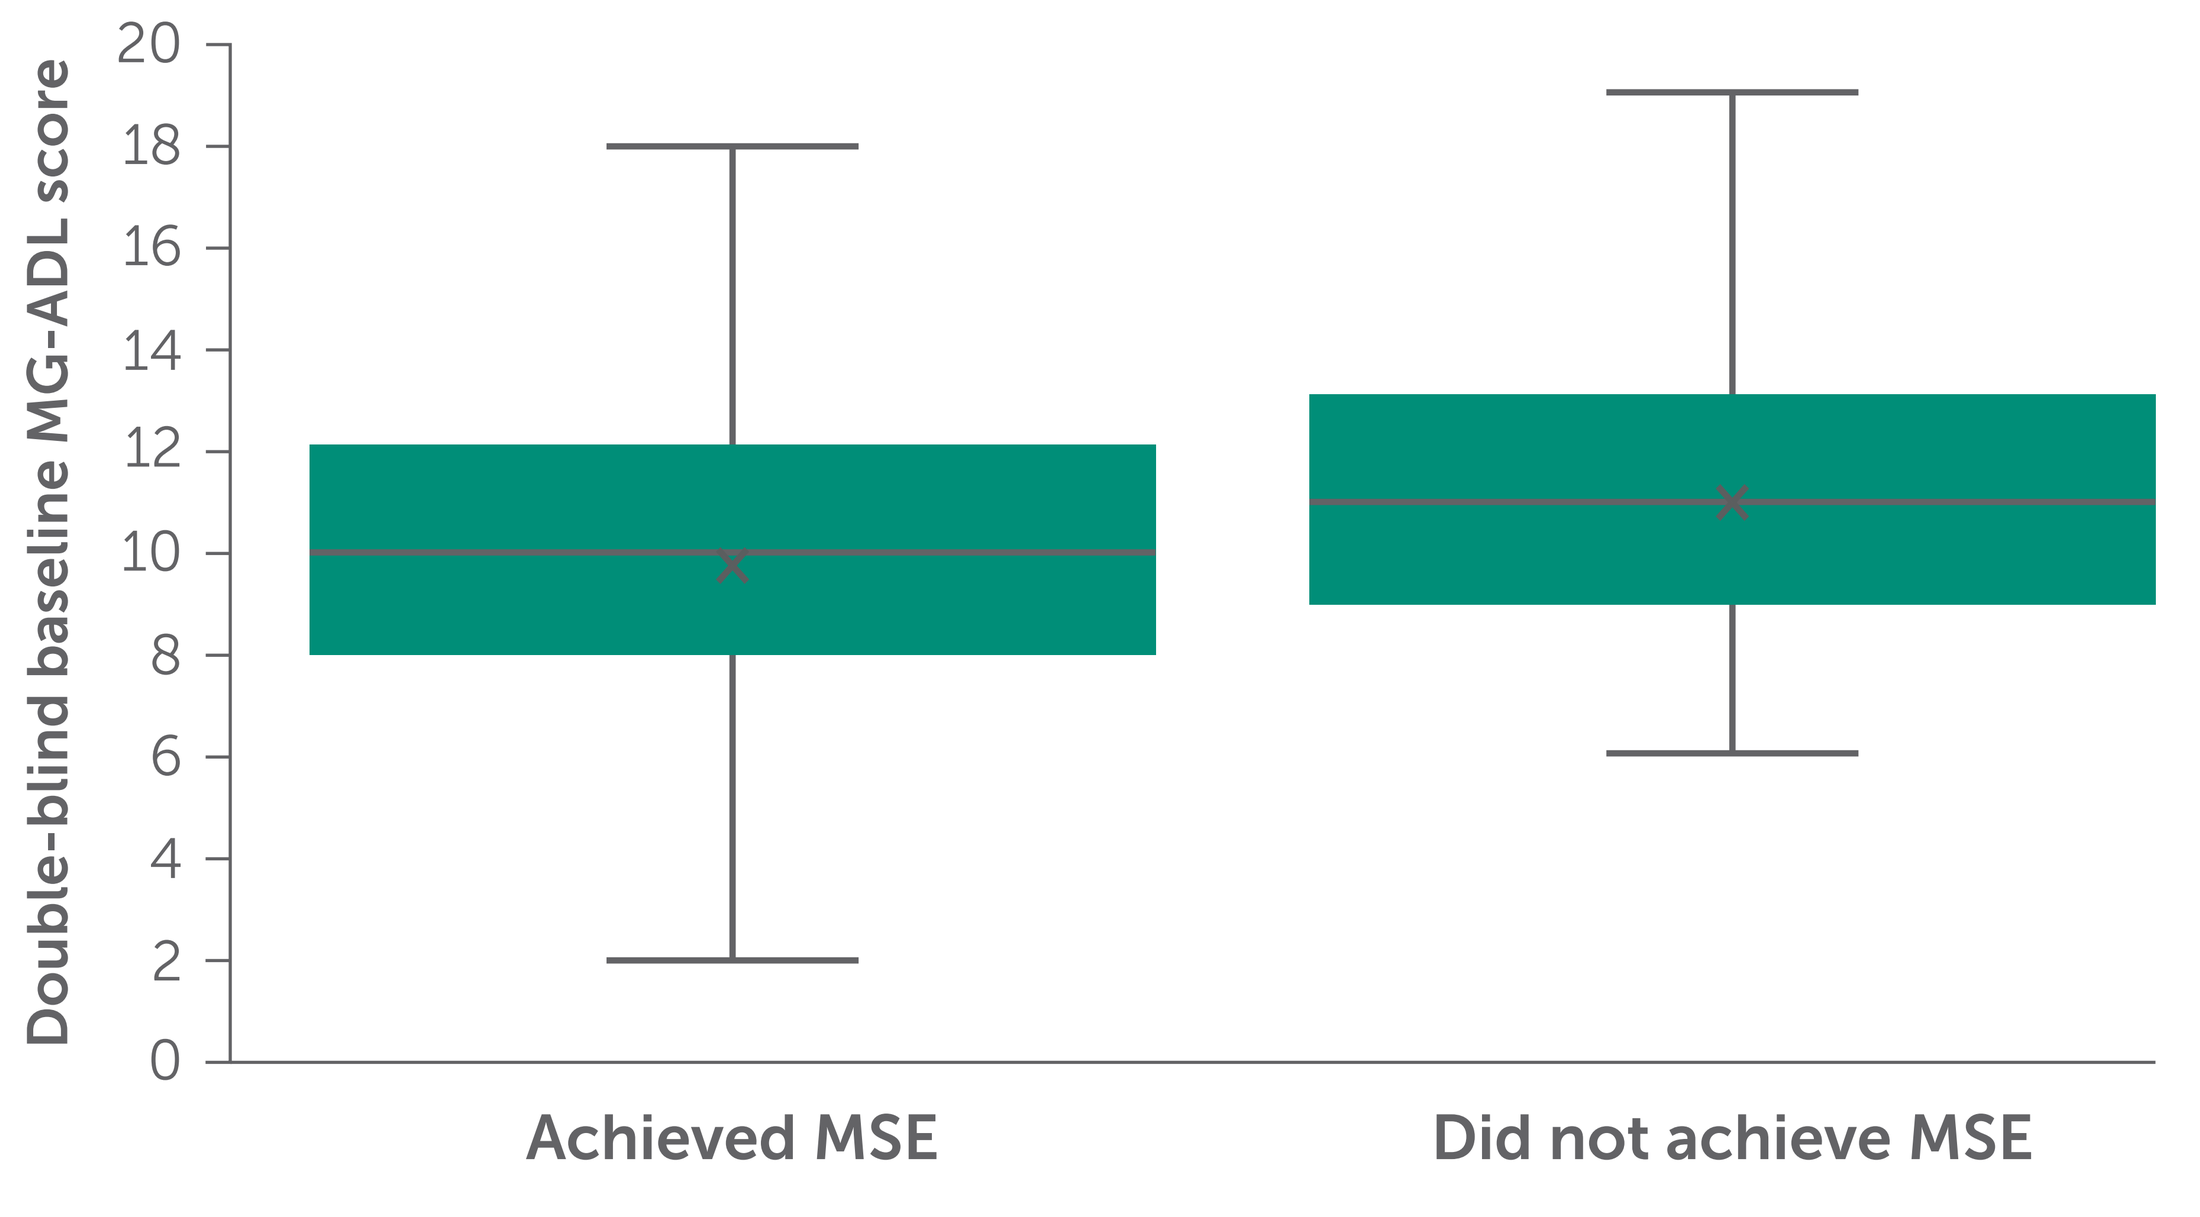
Double-blind baseline MG-ADL score
20
18
16
14
12
10
8
6
4
2
0
Achieved MSE
Did not achieve MSE

mITT population, which included all patients enrolled in RAISE-XT who received at least one dose of zilucoplan and had at least one post-dosing MG-ADL score. The cross symbol indicates the mean. The horizontal line within the box indicates the median, and the box boundaries represent the upper and lower quartiles. The upper and lower error bars indicate the maximum and minimum, respectively.

Figure 3 The majority of patients who achieved MSE did so within 12 weeks of zilucoplan initiation, with some patients achieving MSE as early as 1 week after initiating treatment

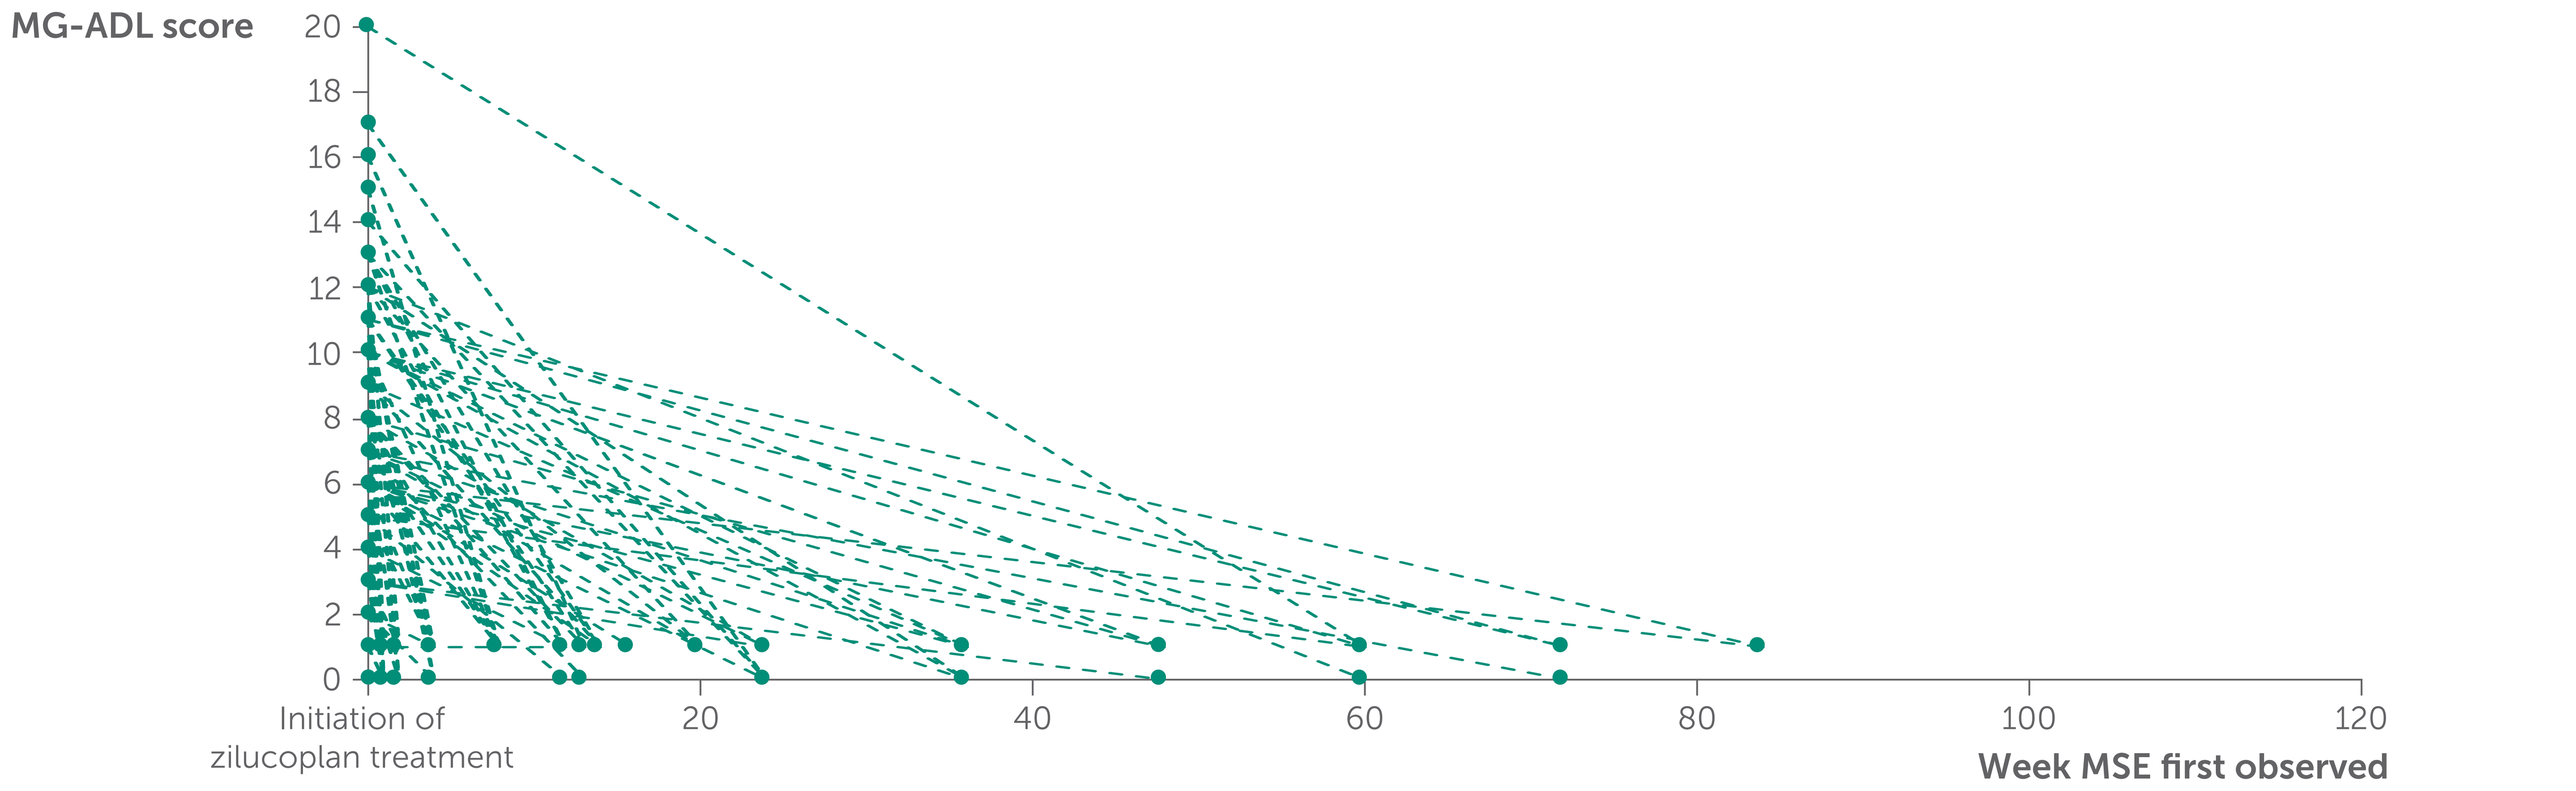
MG-ADL score
20
18
16
14
12
10
8
6
4
2
0
Initiation of zilucoplan treatment
20
40
60
80
100
120
Week MSE first observed

mITT population, which included all patients enrolled in RAISE-XT who received at least one dose of zilucoplan and had at least one post-dosing MG-ADL score. In patients who received zilucoplan in the double-blind studies, zilucoplan was initiated at double-blind baseline. In patients who received placebo in the double-blind studies, zilucoplan was initiated at RAISE-XT baseline. MG-ADL score could differ between double-blind baseline and initiation of zilucoplan. The week MSE was first observed is relative to the appropriate baseline.

Abbreviations: Ab+, antibody positive; AChR, acetylcholine receptor; C5, component 5; gMG, generalized myasthenia gravis; MG, myasthenia gravis; MG-ADL, Myasthenia Gravis Activities of Daily Living; MGFA, Myasthenia Gravis Foundation of America; mITT, modified intent-to-treat; MSE, minimal symptom expression; OLE, open-label extension; QMG, Quantitative Myasthenia Gravis; SD, standard deviation; TEAE, treatment-emergent adverse event.

Acknowledgments: This study was funded by UCB. The authors acknowledge Annabel Dimmock, MBiol, of Ogilvy Health, London, UK, for editorial assistance, which was funded by UCB. The authors thank Veronica Porkess, PhD, CMPP, of UCB for publication and editorial support. The authors thank the patients and their caregivers, in addition to the investigators and their teams who contributed to this study.

Author disclosures: James F. Howard Jr. has received research funding (paid to his institution) from Ad Scientiam, Alexion/AstraZeneca Rare Disease, argenx, Cartesian Therapeutics, Centers for Disease Control and Prevention, Merck EMD Serono, MGFA, Muscular Dystrophy Association, NIH, NMD Pharma, UCB and Vor Biopharma; honoraria/consulting fees from AcademicCME, Alexion/AstraZeneca Rare Disease, Amgen, argenx, Biohaven Ltd, Cartesian Therapeutics, CheckRare CME, CorEvitas, H. Lundbeck A/S, Japan Tobacco Company, Kyverna Therapeutics, MedLive, Merck EMD Serono, NMD Pharma, Novartis, PeerView CME, Physicians' Education Resource (PER, MJH) CME, Regeneron Pharmaceuticals, Seismic Therapeutics, Toleranzia AB, UCB, Vertex Pharmaceuticals and Vor Biopharma; non-financial support from Alexion/AstraZeneca Rare Disease, argenx, Biohaven Ltd, Cartesian Therapeutics, Toleranzia AB, UCB and Vor Biopharma. Saskia Bresch has served as a paid Consultant for Alexion Pharmaceuticals, argenx, Biogen, Bristol Myers Squibb, Merck, Roche, Sandoz, Sanofi Genzyme (now Sanofi) and UCB. Miriam Freimer has served as a paid Consultant for Arcellx, argenx and UCB. She receives research support from Abcuro, Alnylam Pharmaceuticals, argenx, Avidity Biosciences, COUR Pharmaceuticals, Dianthus Therapeutics, Fulcrum Therapeutics, Johnson & Johnson Innovative Medicine, the NIH, RemeGen Biosciences and UCB. M. Isabel Leite is funded by the NHS (Myasthenia and Related Disorders Service and National Specialised Commissioning Group for Neuromyelitis Optica, UK) and by the University of Oxford, UK. She has been awarded research grants from UK associations for patients with myasthenia and with muscular disorders (Myaware and Muscular Dystrophy UK, respectively) and the University of Oxford. She has received speaker honoraria or travel grants from Biogen, the Guthy-Jackson Charitable Foundation, Novartis and UCB. She serves on scientific or educational advisory boards for argenx, Horizon Therapeutics (now Amgen) and UCB. Angelina Maniaol has received payment for travel, meeting attendance, consulting honoraria or advisory board participation from Alexion Pharmaceuticals, argenx, Biogen, Johnson & Johnson and UCB. Kimiaki Utsugisawa has served as a paid Consultant for argenx, Chugai Pharmaceutical, HanAll Biopharma, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Merck, Mitsubishi Tanabe Pharma, UCB and Viela Bio (now Amgen); he has received speaker honoraria from Alexion Pharmaceuticals, argenx, the Japan Blood Products Organization and UCB. Tuan Vu is the USF Site Principal Investigator for MG clinical trials sponsored by Alexion/AstraZeneca Rare Disease, Amgen, argenx, Cartesian Therapeutics, COUR Pharmaceuticals, Dianthus Therapeutics, ImmunAbs, Immunovant, Johnson & Johnson, NMD Pharma, Regeneron Pharmaceuticals, UCB and Vor, and has served as a speaker for Alexion/AstraZeneca Rare Disease, Amgen, argenx and CSL Behring. He has performed consulting work for Alexion/AstraZeneca Rare Disease, argenx, Dianthus Therapeutics, NMD Pharma, Regeneron and Vor. Michael D. Weiss has received honoraria for serving on scientific advisory boards for Alexion Pharmaceuticals, Amgen, Amylyx Pharmaceuticals, argenx, Biogen, Immunovant, Mitsubishi Tanabe Pharma and Ra Pharmaceuticals (now UCB), consulting honoraria from CSL Behring and Cytokinetics, and speaker honoraria from Amgen, Soleo Health, and UCB. He has been a paid Medical Monitor for a NeuroNEXT study. Joel Baldwin is an employee of Cogent Skills Limited working for UCB. Babak Boroojerdi and Natasa Savic are employees and shareholders of UCB. Channa Hewamadduma is Principal Investigator/NCI in clinical trials in various neuromuscular disorders sponsored by Alexion Pharmaceuticals, argenx, Avidity, Cartesian, Dyne, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Novartis, PepGen, Regeneron Pharmaceuticals and UCB. He has received funding for consultancy on scientific or educational advisory boards for argenx, Biogen, Lupin, Roche and UCB, and has received an investigator-led research grant from UCB. His study activities were supported by a Sheffield NIHR BRC UK centre grant. He is a trustee of the myasthenia gravis patient organization Myaware.

References: 1. ZILBRYSQ® US PI. https://www.accessdata.fda.gov/drugsatfda_docs/label/2024/216834s001lbl.pdf. Accessed September 2026. 2. Howard JF Jr., et al. Lancet Neurol. 2023;22(5):395–406. 3. ZILBRYSQ® EU SmPC. https://www.ema.europa.eu/en/documents/product-information/zilbrysq-epar-product-information_en.pdf. Accessed September 2026. 4. Howard JF Jr., et al. Ther Adv Neurol Disord. 2024;17:17562864241243186. 5. Uzawa A, et al. Acta Neurol Belg. 2023;123(3):979–982.

Please use this QR code to download a PDF of the poster.