Poster **92**

Initiating rozanolixizumab in the United States: Treatment utilization among patients with myasthenia gravis

Mohita Kumar[1], Thaïs Tarancón[2], Jianing Yi[3], Rosa Falotico[4], Minjee Park[5], Sofia Zappacosta[6]

[1]UCB, Atlanta, GA, USA; [2]UCB, Madrid, Spain; [3]Alira Health, Framingham, MA, USA; [4]Alira Health, Milan, Italy; [5]Alira Health, Basel, Switzerland; [6]UCB, Bulle, Switzerland

MGFA Scientific Session at AANEM 2026, Orlando, FL, USA; September 29, 2026

Introduction

- Rozanolixizumab is a humanized IgG4 monoclonal antibody that blocks FcRn with high affinity[1]
- Based on clinical trial data,[2,3] rozanolixizumab was approved in the US in June 2023 for the treatment of anti-AChR Ab+ or -MuSK Ab+ generalized myasthenia gravis (MG)[4]
- **The objective of this real-world study was to describe patient demographics, comorbidities, rozanolixizumab treatment patterns, corticosteroid use and HCRU in adults with MG who initiated rozanolixizumab in the US**

Methods

- This was a retrospective non-interventional cohort study of adult patients (≥18 years) with MG in the US, using de-identified, patient-level data from medical and pharmacy claims from The Komodo Healthcare Map®
- Data were included from patients who received ≥1 medical or paid pharmacy claim for rozanolixizumab, had ≥1 ICD code for MG before the index date and had continuous plan coverage for 12 months before the index date (first recorded treatment date with rozanolixizumab in either pharmacy or medical claims)
 - Patients were included between July 1, 2022, and October 31, 2025. The baseline period was 12 months prior to the index date and the follow-up period was from the index date until the end of continuous enrollment or end of data availability, whichever occurred first

Results

- A total of 1,026 patients were included in the analysis
 - Of those, 52.9% were female, with a median (IQR) age of 67 (22) years, and approximately half were insured by Medicare (**Figure 1**)
- The most common comorbidities were hypertension (55.7%), dyslipidemia (50.1%) and infections (48.5%; **Figure 2**)
- For rozanolixizumab initiators:
 - The mean (SD) number of cycles was 2.4 (1.9) across all follow-up (N=1,026)
 - The mean (SD) number of cycles was 3.0 (2.2) among patients with ≥12 months' follow-up (n=586)
- For patients with ≥2 cycles and therefore eligible for cycle interval assessment:
 - The mean (SD) time between cycles was 8.5 (8.2) weeks during the follow-up period (n=571)
 - The mean (SD) time between cycles was 9.2 (9.7) weeks among patients with ≥12 months' follow-up (n=390)
- The proportion of patients with ≥1 biologic therapy claim during baseline was 32.9% (338/1026) and the mean (SD) number of biologic therapies used was 1.2 (0.5) during that period
 - 52.7% (178/338) of patients received efgartigimod IV, 24.3% (82/338) received ravulizumab and 16.3% (55/338) received efgartigimod SC during the baseline period
- There were 37.1% (381/1,026) of patients with ≥12 months' follow-up who had a corticosteroid claim during the baseline period
 - Approximately a third of these patients (136/381; 35.7%) had no corticosteroid claims after rozanolixizumab initiation, and 27.3% (104/381) had fewer corticosteroid claims (**Figure 3**)
 - Among patients with ≥1 corticosteroid claim during the baseline period, the dose of corticosteroid decreased across intervals during the follow-up period (**Figure 4**)
- The percentage of patients with ≥1 claim of NSIST and IVIg/SCIg/PLEX was 37.4% (384/1,026) and 30.5% (313/1,026) during the baseline period, respectively. There was a reduction in the use of these background therapies during follow-up:
 - NSIST use was 31.9% (187/586) 7–12 months post-rozanolixizumab initiation
 - IVIg/SCIg/PLEX use was 12.1% (71/586) 7–12 months post-rozanolixizumab initiation
- All-cause and MG-related HCRU incidence rates, MG exacerbations and MG crisis incidence rates decreased from baseline to follow-up (**Figure 5**)

Summary and conclusions

This was a retrospective, non-interventional cohort study of patient-level data from medical and pharmacy claims in adult patients with MG initiating rozanolixizumab in the US between 2022 and 2025

The aim of the study was to describe patient demographics, comorbidities, rozanolixizumab treatment patterns, corticosteroid use and HCRU in adults in the US who initiated rozanolixizumab treatment for MG

Following rozanolixizumab initiation, reductions in corticosteroid claims were observed, including reduced doses of corticosteroids over time

Reductions in all-cause and MG-related HCRU were observed after the initiation of rozanolixizumab

Figure 1 Approximately half of patients were aged 61–80 years (a), approximately half were from the South (b) and approximately half were insured by Medicare (c)

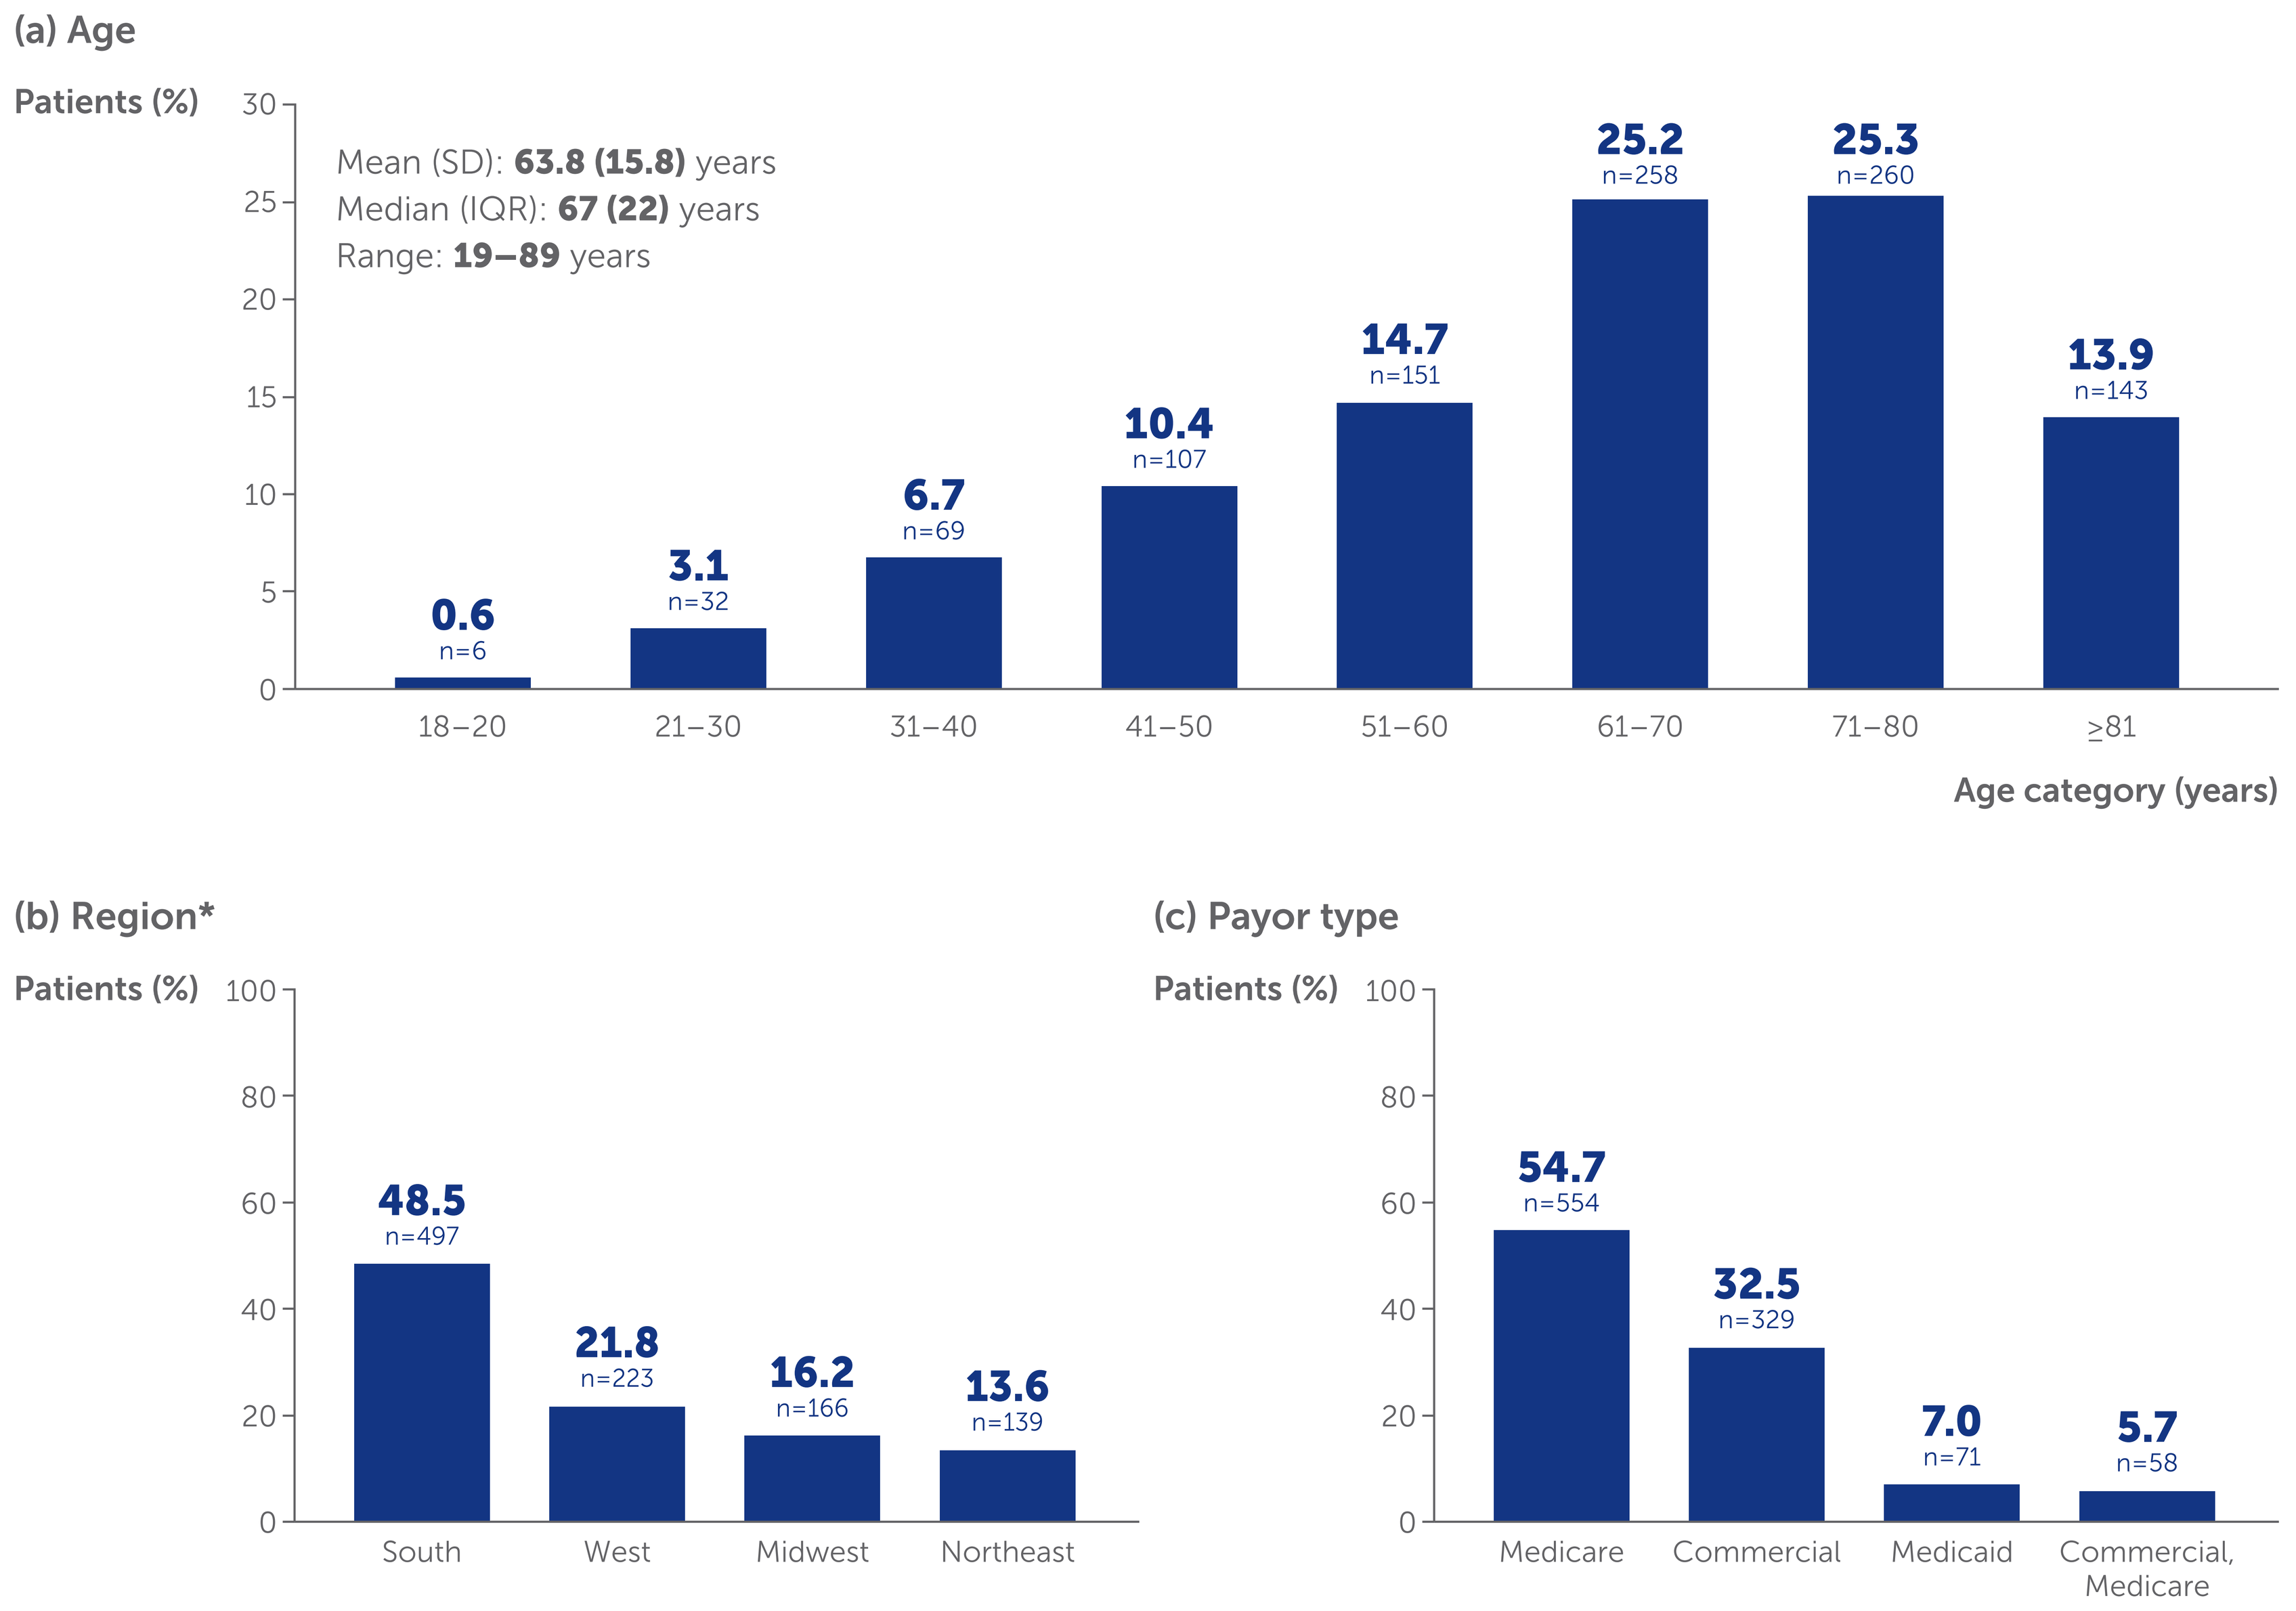
(a) Age
Patients (%)
Mean (SD): 63.8 (15.8) years
Median (IQR): 67 (22) years
Range: 19–89 years
0.6 n=6
3.1 n=32
6.7 n=69
10.4 n=107
14.7 n=151
25.2 n=258
25.3 n=260
13.9 n=143
18–20
21–30
31–40
41–50
51–60
61–70
71–80
≥81
Age category (years)
(b) Region*
Patients (%)
48.5 n=497
21.8 n=223
16.2 n=166
13.6 n=139
South
West
Midwest
Northeast
(c) Payor type
Patients (%)
54.7 n=554
32.5 n=329
7.0 n=71
5.7 n=58
Medicare
Commercial
Medicaid
Commercial, Medicare

*South: Delaware, District of Columbia, Florida, Georgia, Maryland, North Carolina, South Carolina, Virginia, West Virginia, Alabama, Kentucky, Mississippi, Tennessee, Arkansas, Louisiana, Oklahoma, Texas. West: Arizona, Colorado, Idaho, New Mexico, Montana, Utah, Nevada, Wyoming, Alaska, California, Hawaii, Oregon, Washington. Midwest: Indiana, Illinois, Michigan, Ohio, Wisconsin, Iowa, Kansas, Minnesota, Missouri, Nebraska, North Dakota, South Dakota. Northeast: Connecticut, Maine, Massachusetts, New Hampshire, Rhode Island, Vermont, New Jersey, New York, Pennsylvania.

Figure 2 Comorbid conditions were common at baseline, and the most frequently reported were hypertension, dyslipidemia and infections

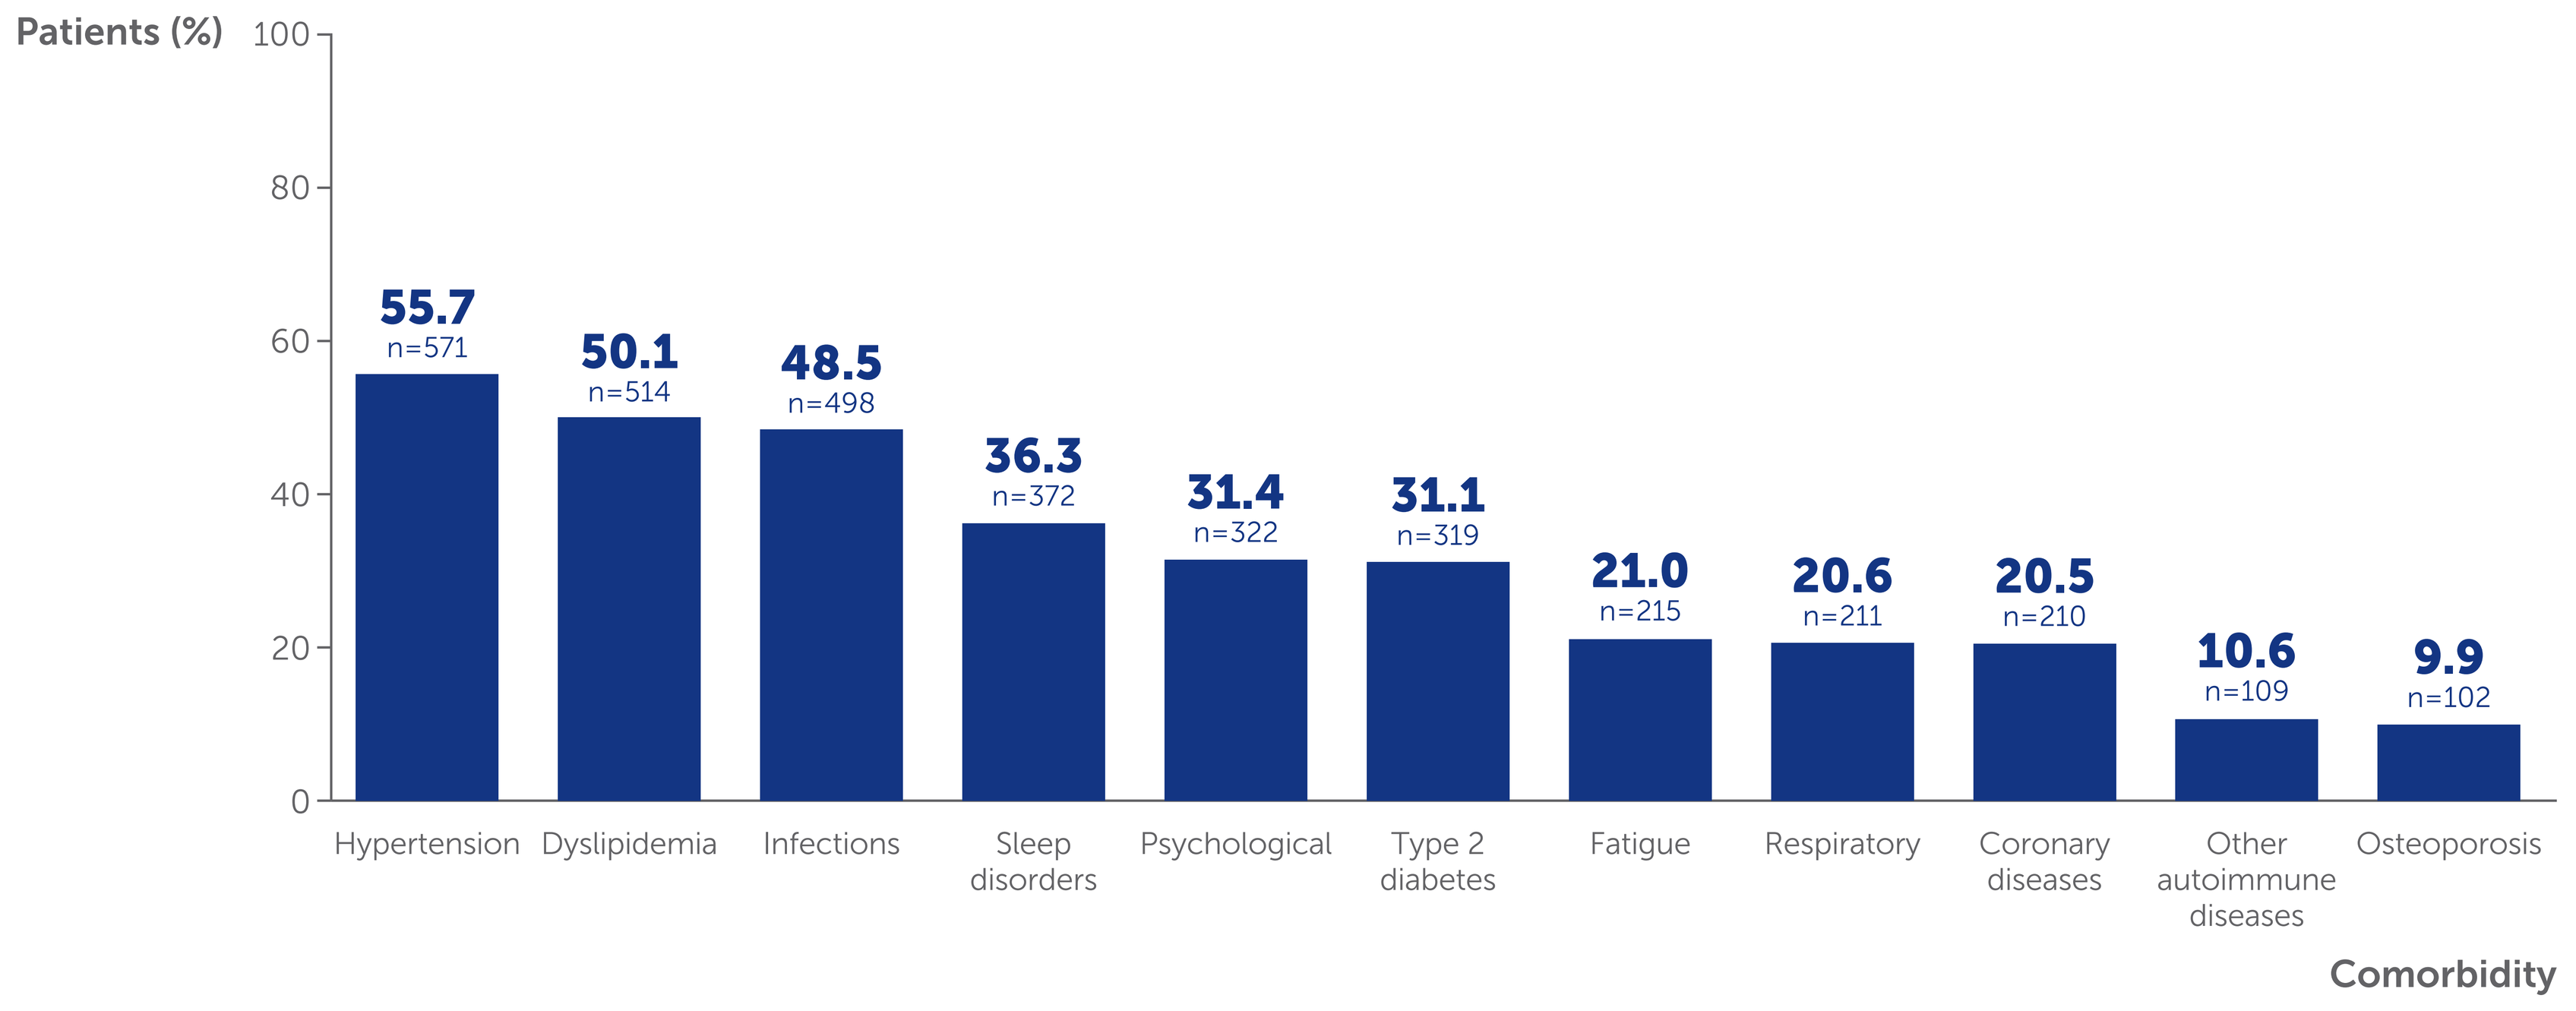
Patients (%)
55.7 n=571
50.1 n=514
48.5 n=498
36.3 n=372
31.4 n=322
31.1 n=319
21.0 n=215
20.6 n=211
20.5 n=210
10.6 n=109
9.9 n=102
Hypertension
Dyslipidemia
Infections
Sleep disorders
Psychological
Type 2 diabetes
Fatigue
Respiratory
Coronary diseases
Other autoimmune diseases
Osteoporosis
Comorbidity

Figure 3 For patients with a corticosteroid claim at baseline, approximately a third discontinued after initiation of rozanolixizumab

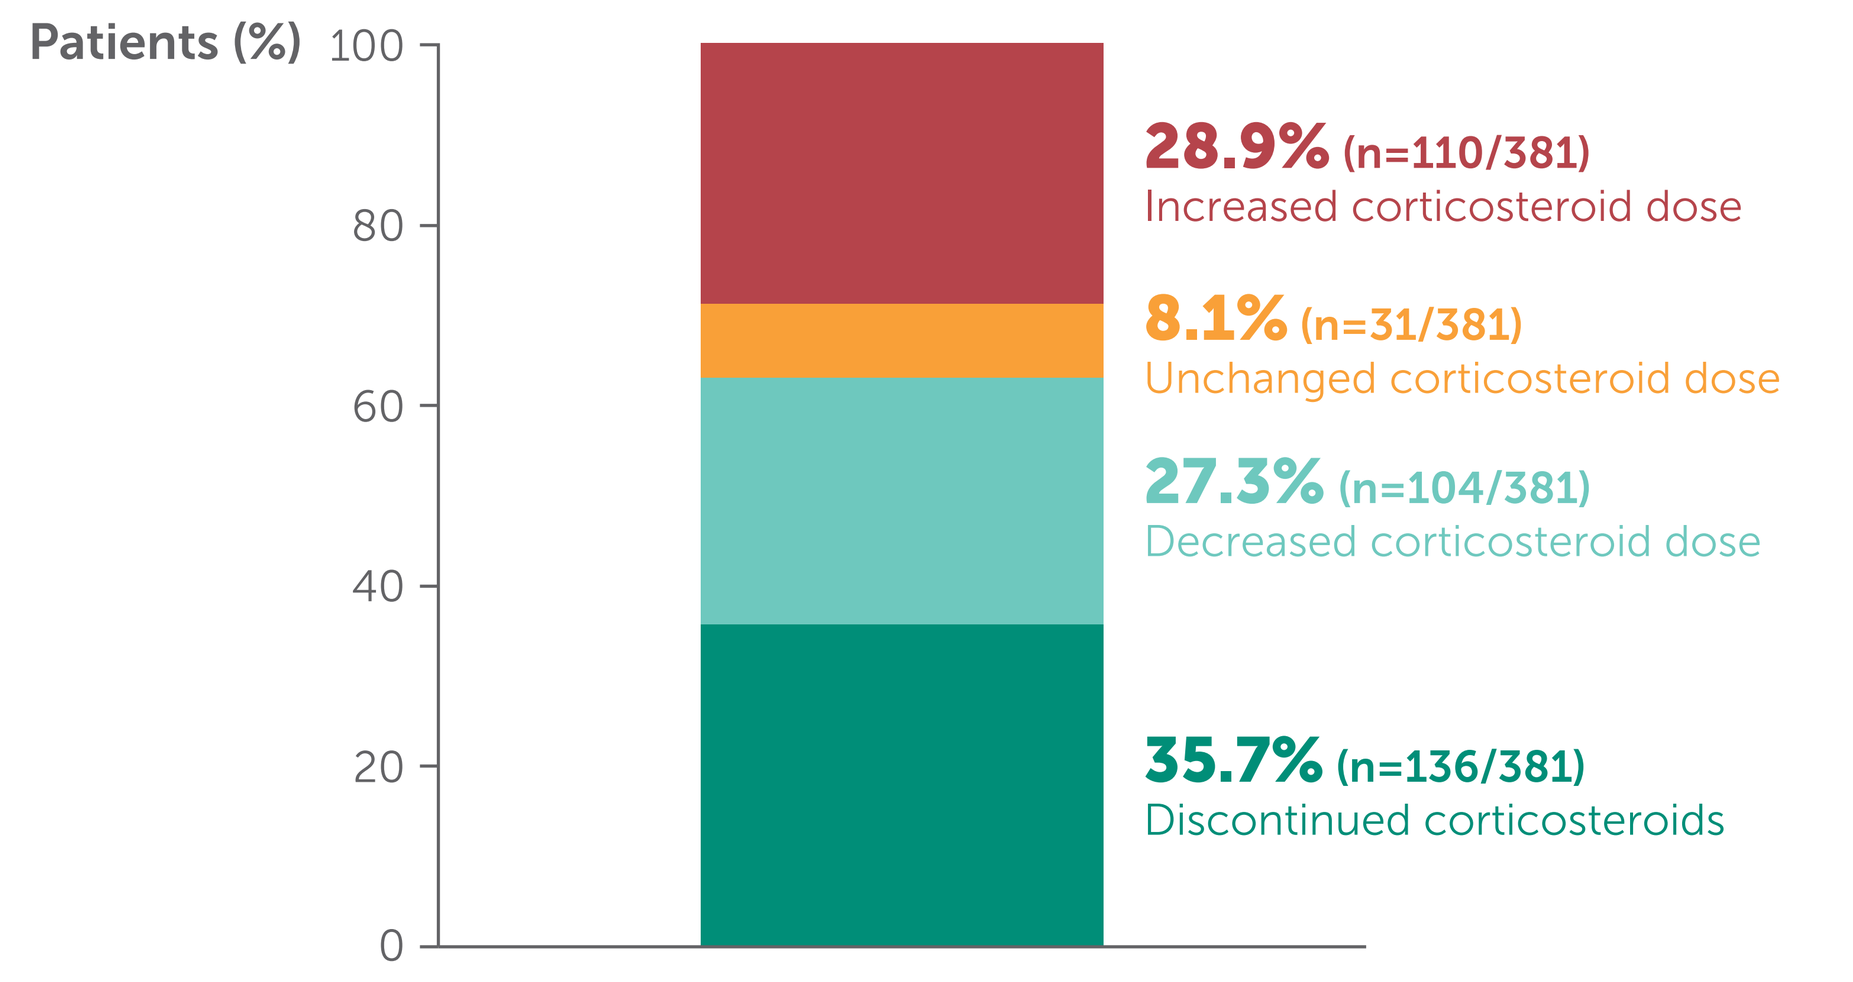
Patients (%)
28.9% (n=110/381)
Increased corticosteroid dose
8.1% (n=31/381)
Unchanged corticosteroid dose
27.3% (n=104/381)
Decreased corticosteroid dose
35.7% (n=136/381)
Discontinued corticosteroids

The denominator is the number of patients with ≥12 months' follow-up who had a corticosteroid claim during the baseline period (n=381).

Figure 4 The average daily dose of corticosteroids was substantially reduced during the follow-up period compared with 3 months prior to rozanolixizumab initiation

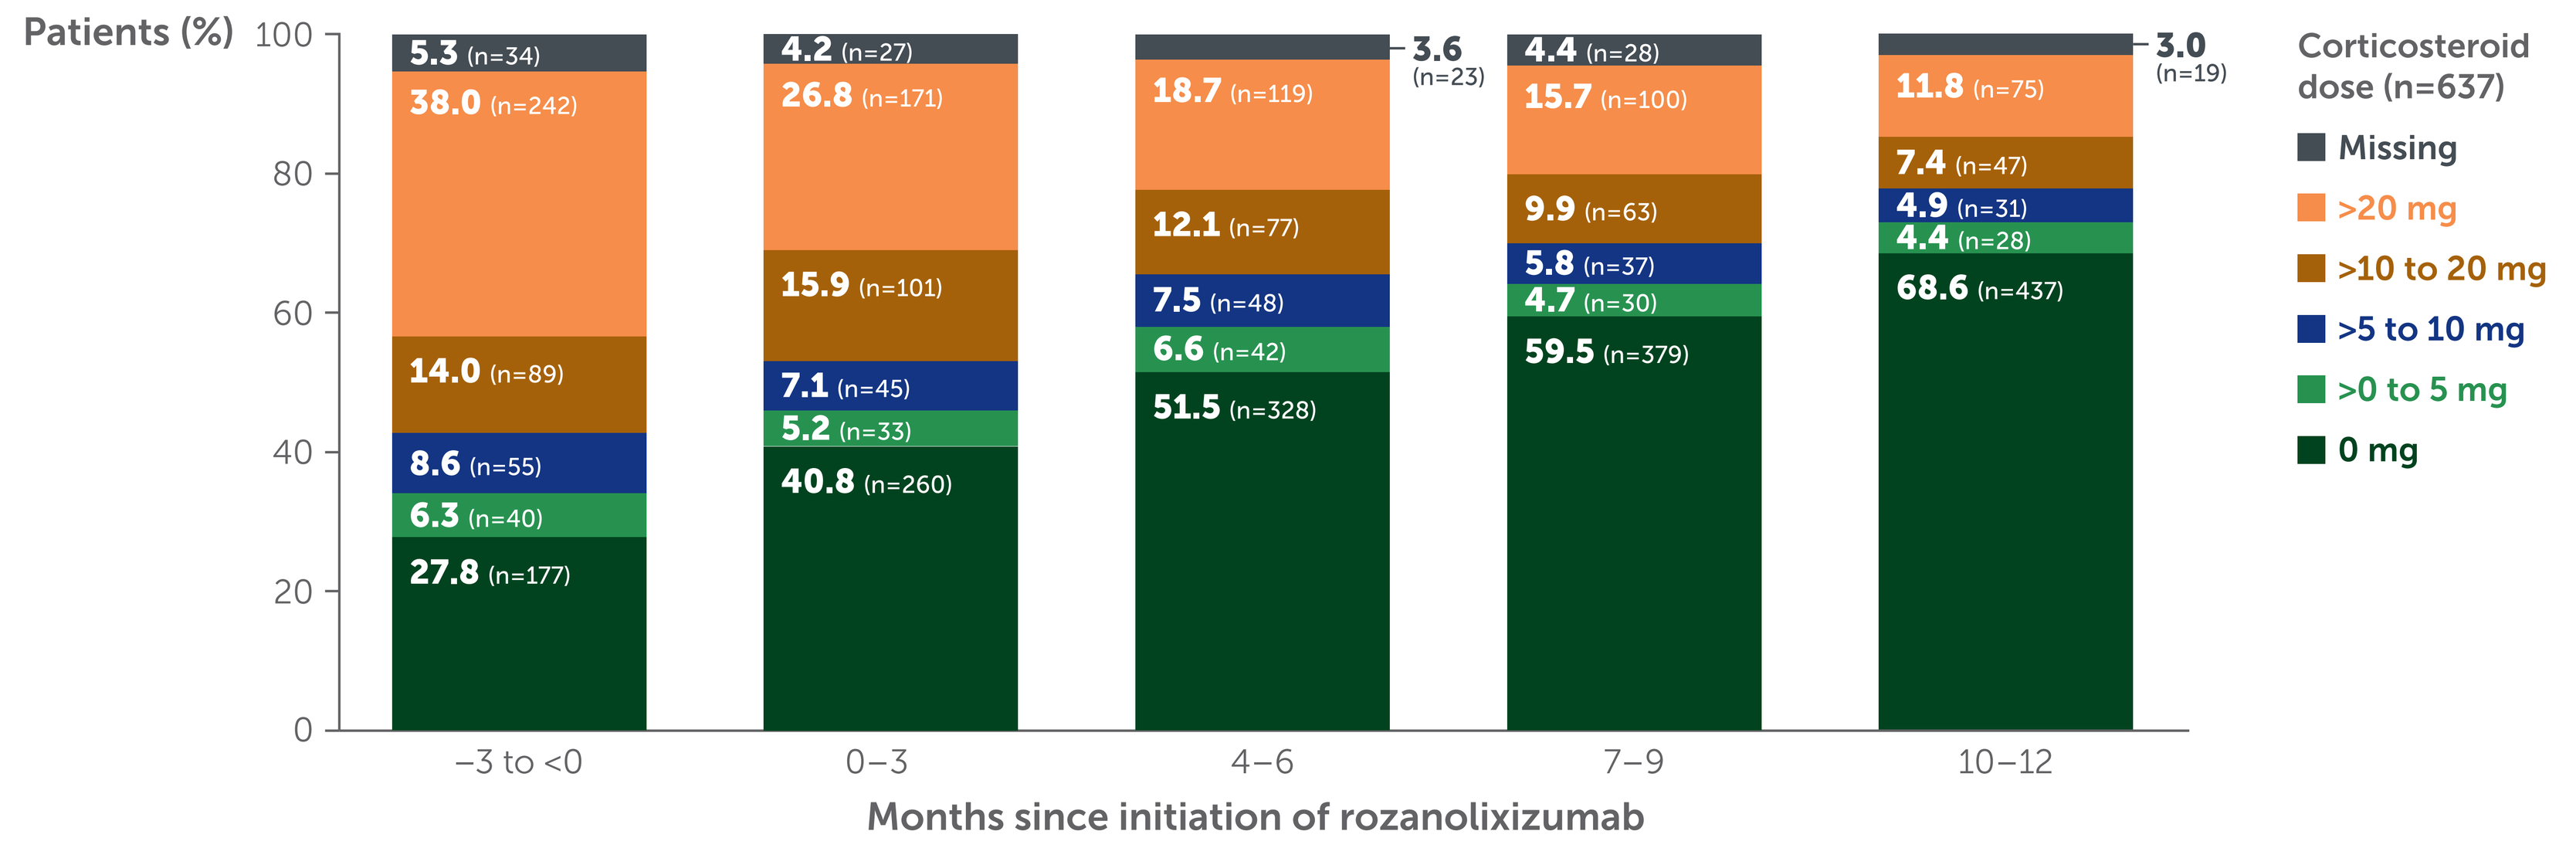
Patients (%)
5.3 (n=34)
38.0 (n=242)
14.0 (n=89)
8.6 (n=55)
6.3 (n=40)
27.8 (n=177)
4.2 (n=27)
26.8 (n=171)
15.9 (n=101)
7.1 (n=45)
5.2 (n=33)
40.8 (n=260)
3.6 (n=23)
18.7 (n=119)
12.1 (n=77)
7.5 (n=48)
6.6 (n=42)
51.5 (n=328)
4.4 (n=28)
15.7 (n=100)
9.9 (n=63)
5.8 (n=37)
4.7 (n=30)
59.5 (n=379)
3.0 (n=19)
11.8 (n=75)
7.4 (n=47)
4.9 (n=31)
4.4 (n=28)
68.6 (n=437)
−3 to <0
0–3
4–6
7–9
10–12
Months since initiation of rozanolixizumab
Corticosteroid dose (n=637)
Missing
>20 mg
>10 to 20 mg
>5 to 10 mg
>0 to 5 mg
0 mg

The denominator is the number of patients who had a corticosteroid claim during the baseline period (n=637). Patients with a null corticosteroid claim and a null ADD value during a given period were classified as having an ADD of 0 mg for that period. Conversely, patients with a valid corticosteroid claim but a null ADD value during a given period were classified as having missing ADD data for that period.

Figure 5 The incidence rates for all-cause HCRU outcomes decreased after rozanolixizumab initiation compared with the baseline period

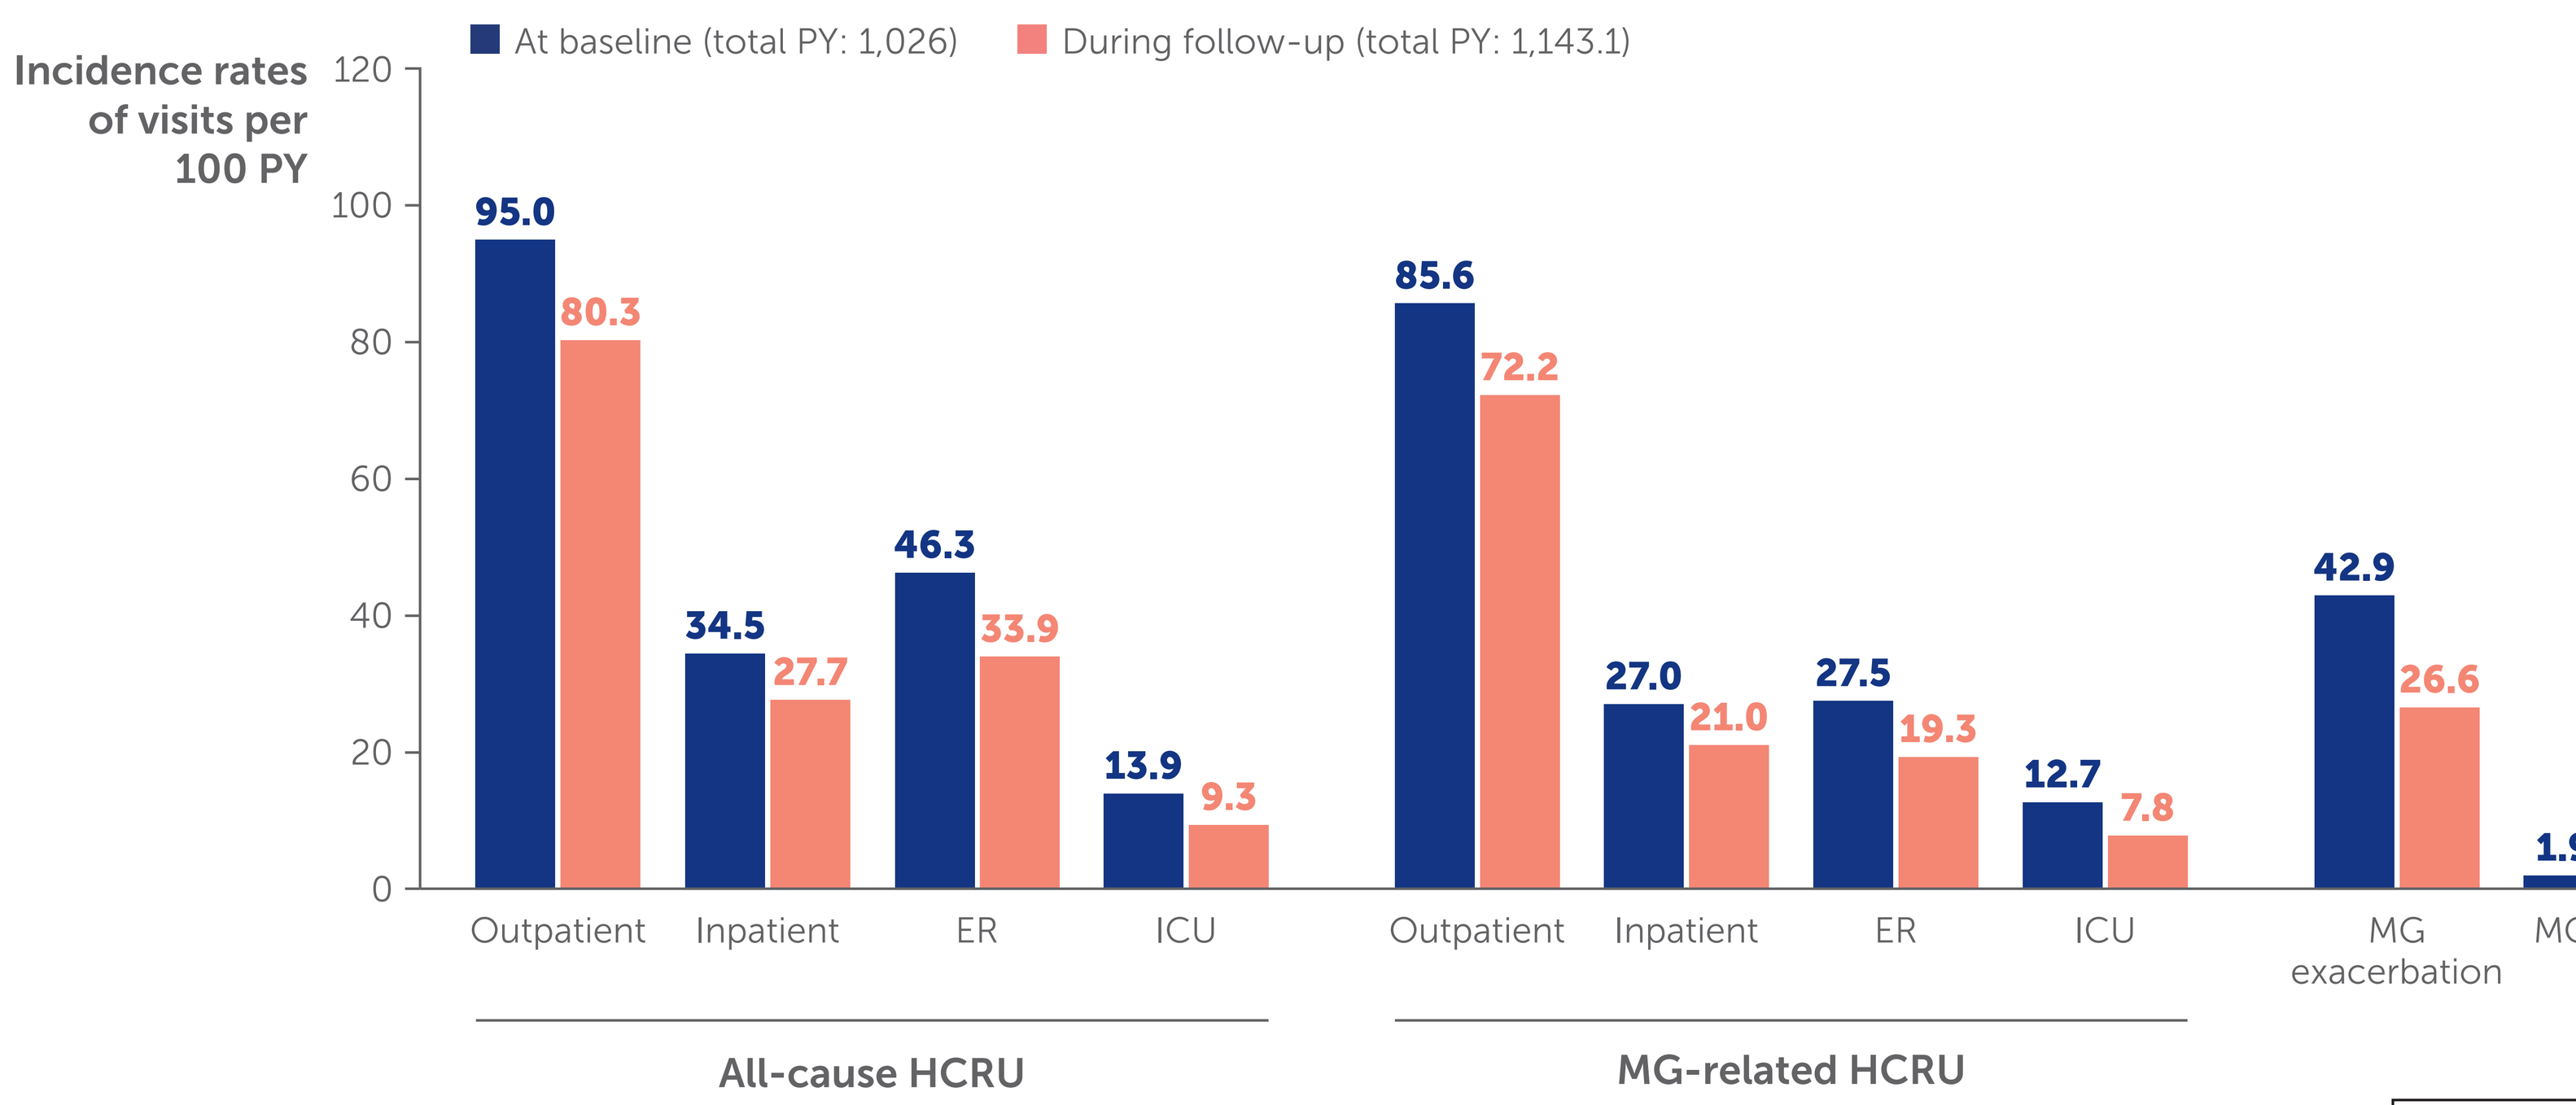
At baseline (total PY: 1,026)
During follow-up (total PY: 1,143.1)
Incidence rates of visits per 100 PY
95.0
80.3
34.5
27.7
46.3
33.9
13.9
9.3
85.6
72.2
27.0
21.0
27.5
19.3
12.7
7.8
42.9
26.6
1.9
1.0
Outpatient
Inpatient
ER
ICU
Outpatient
Inpatient
ER
ICU
MG exacerbation
MG crisis
All-cause HCRU
MG-related HCRU

Abbreviations: Ab+, antibody positive; AChR, acetylcholine receptor; ADD, average daily dose; ER, emergency room; FcRn, neonatal fragment crystallizable receptor; gMG, generalized myasthenia gravis; HCRU, healthcare resource utilization; ICD, International Classification of Diseases; ICU, intensive care unit; IgG4, immunoglobulin G subunit 4; IQR, interquartile range; IV, intravenous; IVIg, intravenous immunoglobulin; MG, myasthenia gravis; MuSK, muscle-specific tyrosine kinase; NSIST, nonsteroidal immunosuppressant therapy; PLEX, plasma exchange; PY, person-year; SC, subcutaneous; SCIg, subcutaneous immunoglobulin; SD, standard deviation.

Acknowledgments: This study was funded by UCB. The authors acknowledge Annick Moon, PhD, of Ogilvy Health, London, UK, for editorial assistance, which was funded by UCB. The authors thank Patrik Öhagen, formerly of UCB, for statistical analysis support, and Kate Weatherall, PhD, of UCB, for publication and editorial support. The authors thank the patients whose data were included in the Komodo Healthcare Map®.

Author disclosures: Mohita Kumar, Thaïs Tarancón, and Sofia Zappacosta are employees and shareholders of UCB. Jianing Yi, Rosa Falotico and Minjee Park are employees of Alira Health.

References: 1. Smith B, et al. MAbs. 2018;10(7):1111–1130. 2. Bril V, et al. Lancet Neurol. 2023;22(5):383–394. 3. Bril V, et al. J Neuromuscul Dis. 2025;12(2):218–230. 4. RYSTIGGO® US PI. https://www.accessdata.fda.gov/drugsatfda_docs/label/2023/761286s000lbl.pdf. Accessed August 2026.

Please use this QR code to download a PDF of the poster.