AANEM Poster **389** | MGFA Scientific Session Poster **93**

Effect of rozanolixizumab on ocular symptoms in generalized myasthenia gravis: Analysis of Phase 3 studies

Zabeen K. Mahuwala[1], Elena Cortés Vicente[2], Ali A. Habib[3], Robert M. Pascuzzi[4], John Vissing[5], Tuan Vu[6], Jos Bloemers[7], Paul Mahoney[8], Thaïs Tarancón[9], Vera Bril[10*]

[1]Department of Neuromuscular Medicine, Epilepsy and Clinical Neurophysiology, University of Kentucky, Lexington, KY, USA; [2]Neuromuscular Diseases Unit, Hospital de la Santa Creu i Sant Pau, Barcelona, Spain; [3]MDA ALS & Neuromuscular Center, Department of Neurology, University of California, Irvine, Orange, CA, USA; [4]Neurology Department, Indiana University School of Medicine, Indiana University Health, Indianapolis, IN, USA; [5]Copenhagen Neuromuscular Center, Department of Neurology, Rigshospitalet, University of Copenhagen, Copenhagen, Denmark; [6]Department of Neurology, University of South Florida Morsani College of Medicine, Tampa, FL, USA; [7]UCB, Breda, The Netherlands; [8]UCB, Slough, UK; [9]UCB, Madrid, Spain; [10]Ellen and Martin Prosserman Centre for Neuromuscular Diseases, Toronto General Hospital, University of Toronto, Toronto, Ontario, Canada

AANEM & MGFA Scientific Session 2026, Orlando, FL, USA; September 29–October 2, 2026

Introduction

- Patients with gMG may experience disabling ocular symptoms, such as diplopia and ptosis, due to ocular muscle weakness and fatigability[1–3]
 - Such ocular symptoms impede daily activities and significantly reduce QoL[4]
- Rozanolixizumab, a humanized IgG4 mAb that targets and blocks FcRn with high affinity, is approved for the treatment of adults with anti-AChR Ab+ or anti-MuSK Ab+ gMG[4]
- In the Phase 3 MycarinG study (NCT03971422), one 6-week cycle of rozanolixizumab demonstrated greater improvements in ocular item scores across MG-specific outcomes than placebo in patients with gMG[5]
- **This *post hoc* analysis investigated the effect of repeated rozanolixizumab treatment cycles on ocular symptoms in patients with gMG**

Methods

- In MycarinG, patients aged ≥18 years with MGFA Disease Class II–IVa anti-AChR Ab+ or anti-MuSK Ab+ gMG with an MG-ADL score ≥3 (for non-ocular symptoms) and a QMG score ≥11 were enrolled
- Following MycarinG, patients could enroll in OLE studies MG0004 (NCT04124965) and then MG0007 (NCT04650854), or MG0007 directly
 - MG0004: once-weekly rozanolixizumab 7 mg/kg or 10 mg/kg for ≤52 weeks
 - MG0007: an initial 6-week rozanolixizumab 7 mg/kg or 10 mg/kg cycle, with further cycles administered upon symptom worsening at the investigator's discretion
- Final efficacy data were pooled across MycarinG, MG0004 (first 6 weeks) and MG0007 for patients with ≥2 symptom-driven rozanolixizumab cycles
- Mean item scores (weighted by sample size, per cycle) were evaluated at baseline and Day 43 across Cycles 1–13 for ocular items within the MG-ADL, QMG, MG Symptoms PRO and MG-QoL15r scales

Results

- Overall, 129 patients received ≥2 symptom-driven cycles
- Baseline demographics and disease characteristics are shown in **Table 1**
- Across 13 cycles, weighted mean ocular item-level scores improved between baseline and Day 43 for MG-ADL, QMG, MG Symptoms PRO and MG-QoL15r (**Figure 1**)
- Across Cycles 1–13 (weighted mean), the percentages of patients who improved to have no symptoms in each ocular item-level score across all four measures ranged from 17.7% (MG Symptoms PRO; Difficulty moving eyes side to side)) to 31.1% (MG-ADL; eyelid droop) (**Figure 2**)
- In total, 93.1% (175/188) of patients with ≥1 treatment cycle across MycarinG and MG0007 experienced a TEAE; most were mild or moderate

Table 1 Baseline demographics and disease characteristics were indicative of a broad population of patients with gMG

		RLZ total (N=129)
	Age, years, mean (SD)	**50.9** (16.3)
	Sex, female, n (%)	**77** (59.7)
	Age at initial gMG diagnosis, years, mean (SD)	**43.1** (17.8)
	Duration of disease from diagnosis, years, mean (SD)	**8.1** (8.5)
MGFA Disease Class, n (%)	II	**52** (40.3)
	III	**72** (55.8)
	IV	**5** (3.9)
	Anti-AChR Ab+, n (%)	**117** (90.7)
	Anti-MuSK Ab+, n (%)	**12** (9.3)
	MG-ADL score, mean (SD)	**8.7** (3.4)
	MG-ADL ocular subdomain score, mean (SD)	**2.8** (1.7)
	QMG score, mean (SD)	**16.0** (3.8)
	QMG ocular subdomain score, mean (SD)	**4.0** (1.9)
Baseline gMG medication, n (%)	CS for systemic use	**80** (62.0)
	Immunosuppressants	**66** (51.2)
	Parasympathomimetics	**113** (87.6)

Summary and conclusions

This *post hoc* analysis of pooled data from the Phase 3 MycarinG study and its OLEs evaluated the effect of long-term, cyclic rozanolixizumab treatment on the ocular signs and symptoms experienced by patients with gMG

Long-term rozanolixizumab treatment cycles led to improvements across ocular items in MG-specific outcome measures, suggesting a benefit for patients with gMG experiencing ocular signs and symptoms

Following these results, rozanolixizumab is being investigated in the MyVision study (NCT07463521) in patients with ocular MG

Figure 1 Across 13 cycles, weighted mean ocular item-level scores improved between baseline and Day 43 for (a) MG-ADL, (b) QMG, (c) MG Symptoms PRO and (d) MG-QoL15r measures

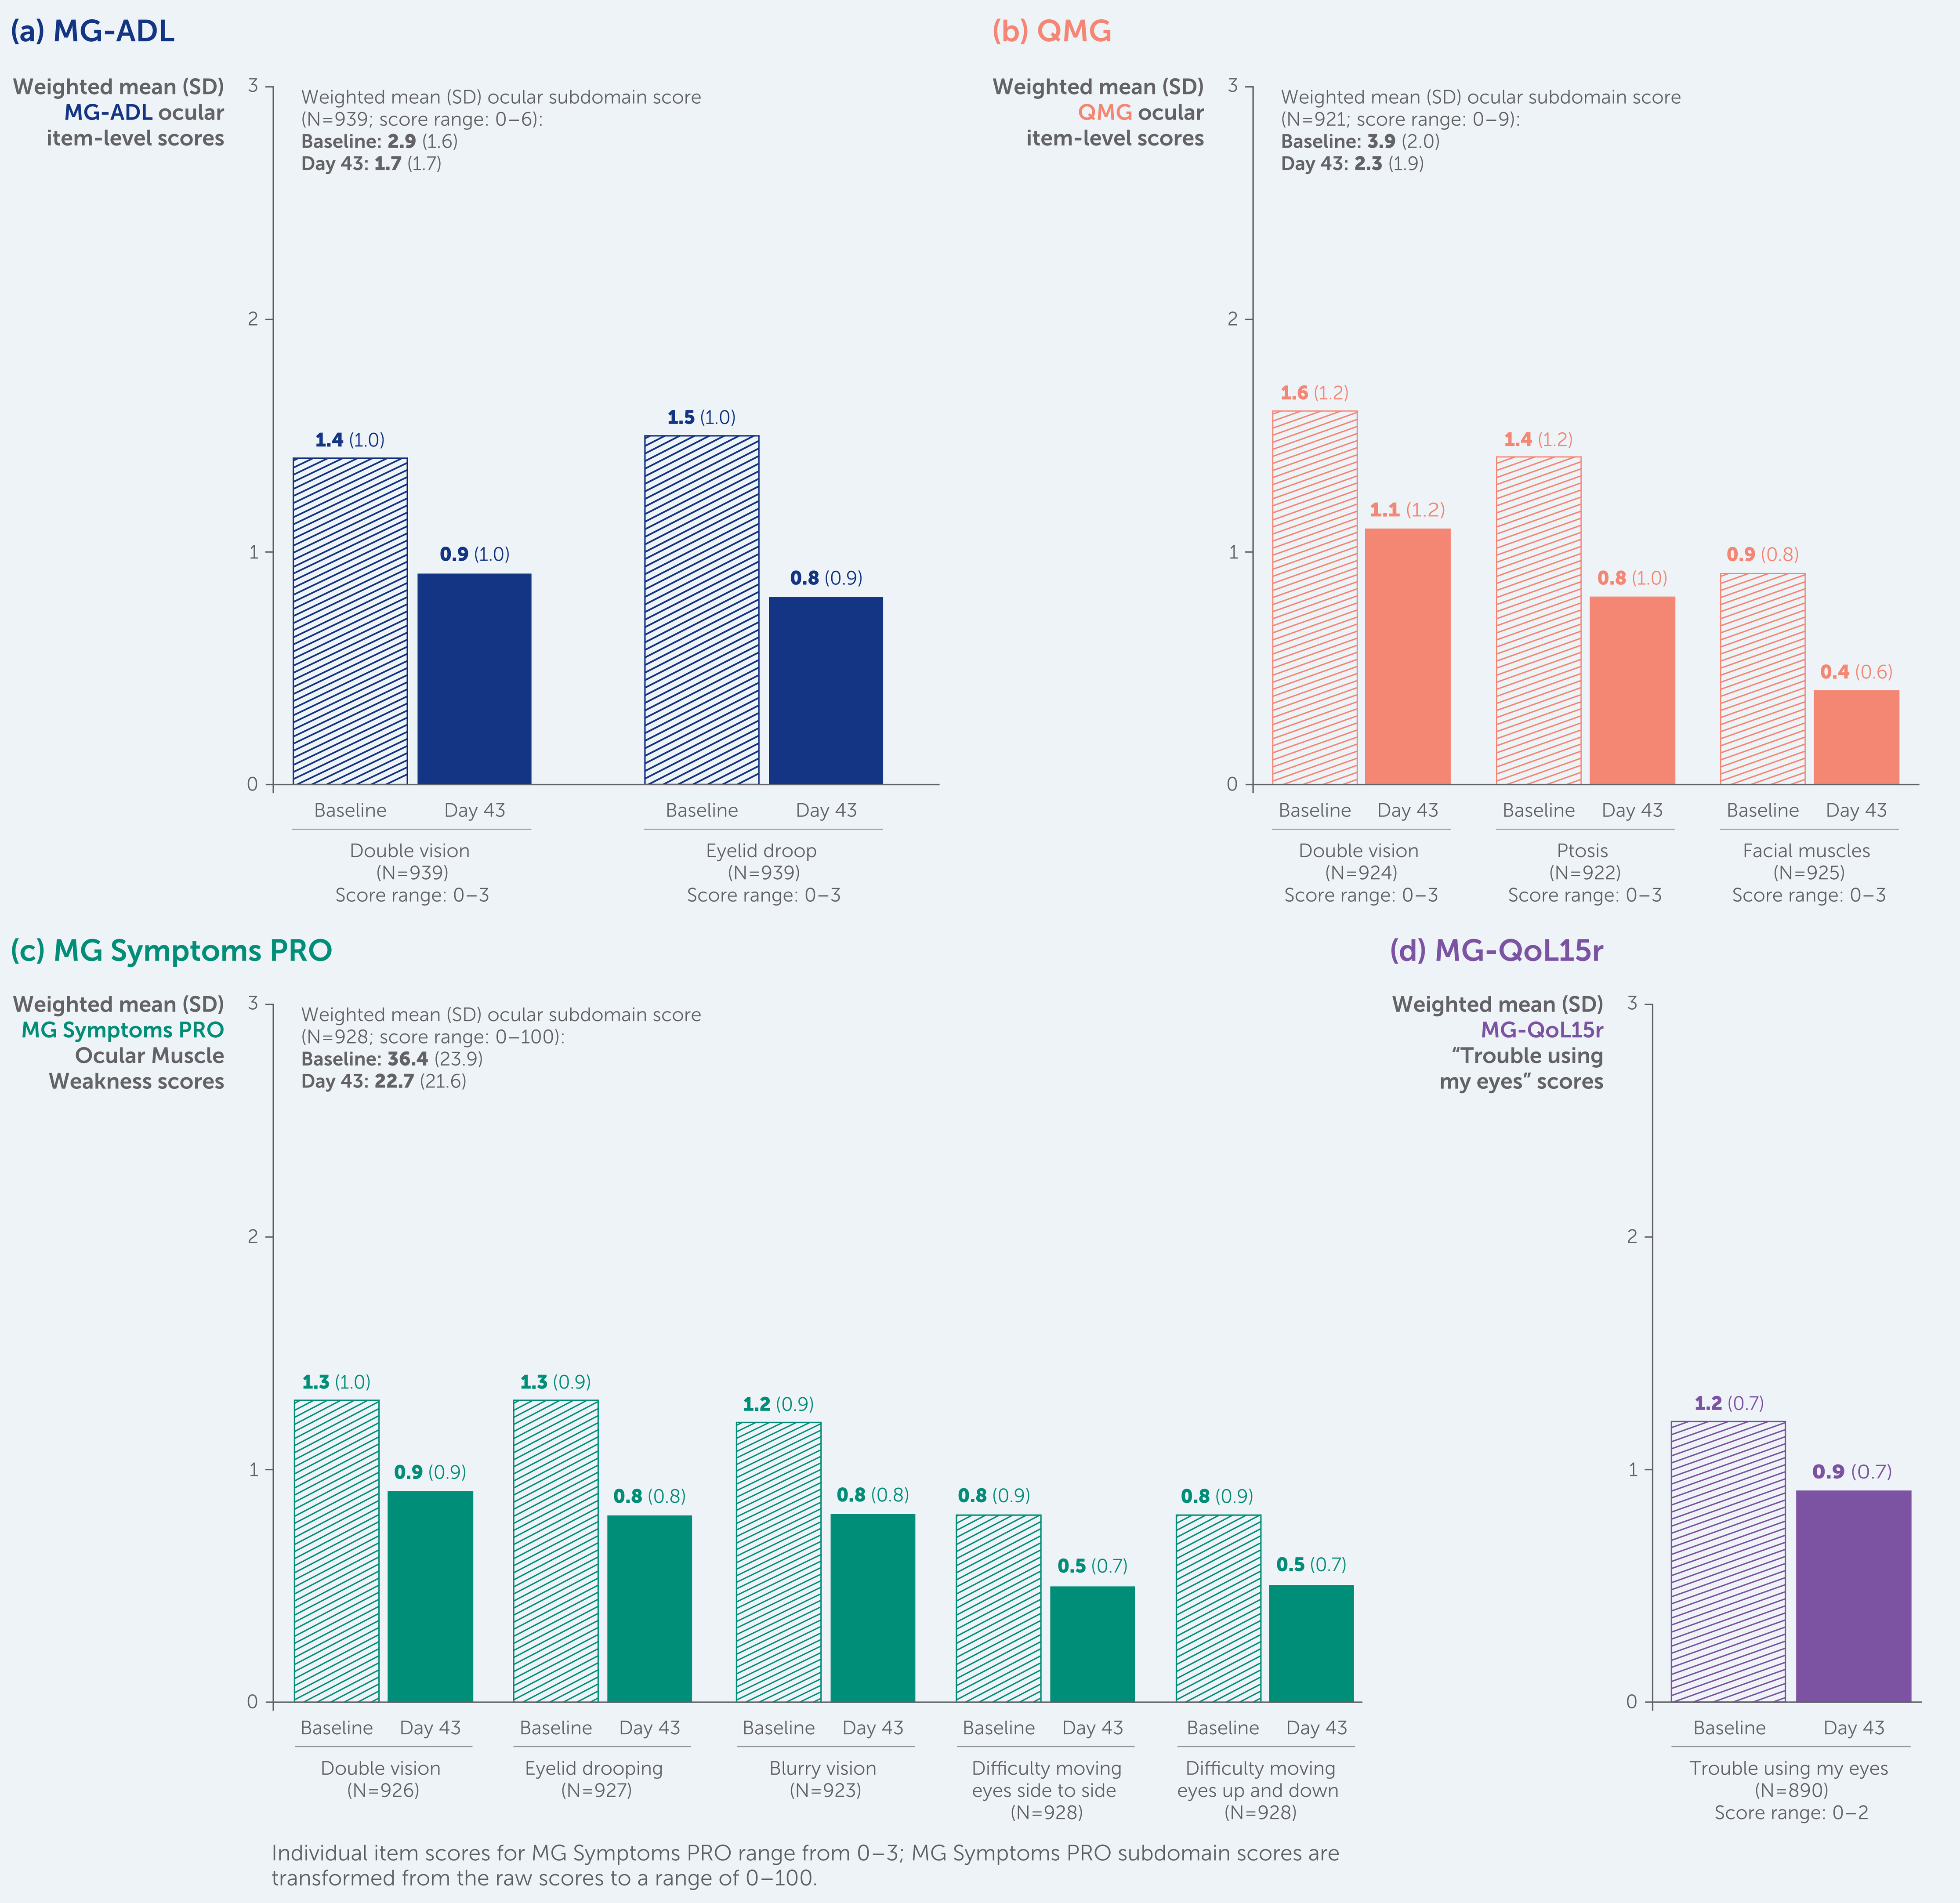
(a) MG-ADL
Weighted mean (SD) MG-ADL ocular item-level scores
Weighted mean (SD) ocular subdomain score (N=939; score range: 0–6): Baseline: 2.9 (1.6) Day 43: 1.7 (1.7)
1.4 (1.0)
0.9 (1.0)
1.5 (1.0)
0.8 (0.9)
Baseline
Day 43
Double vision (N=939) Score range: 0–3
Eyelid droop (N=939) Score range: 0–3
(b) QMG
Weighted mean (SD) QMG ocular item-level scores
Weighted mean (SD) ocular subdomain score (N=921; score range: 0–9): Baseline: 3.9 (2.0) Day 43: 2.3 (1.9)
1.6 (1.2)
1.1 (1.2)
1.4 (1.2)
0.8 (1.0)
0.9 (0.8)
0.4 (0.6)
Double vision (N=924) Score range: 0–3
Ptosis (N=922) Score range: 0–3
Facial muscles (N=925) Score range: 0–3
(c) MG Symptoms PRO
Weighted mean (SD) MG Symptoms PRO Ocular Muscle Weakness scores
Weighted mean (SD) ocular subdomain score (N=928; score range: 0–100): Baseline: 36.4 (23.9) Day 43: 22.7 (21.6)
1.3 (1.0)
0.9 (0.9)
1.3 (0.9)
0.8 (0.8)
1.2 (0.9)
0.8 (0.8)
0.8 (0.9)
0.5 (0.7)
0.8 (0.9)
0.5 (0.7)
Double vision (N=926)
Eyelid drooping (N=927)
Blurry vision (N=923)
Difficulty moving eyes side to side (N=928)
Difficulty moving eyes up and down (N=928)
Individual item scores for MG Symptoms PRO range from 0–3; MG Symptoms PRO subdomain scores are transformed from the raw scores to a range of 0–100.
(d) MG-QoL15r
Weighted mean (SD) MG-QoL15r "Trouble using my eyes" scores
1.2 (0.7)
0.9 (0.7)
Trouble using my eyes (N=890) Score range: 0–2

Figure 2 Most patients' ocular item-level scores improved across Cycles 1–13

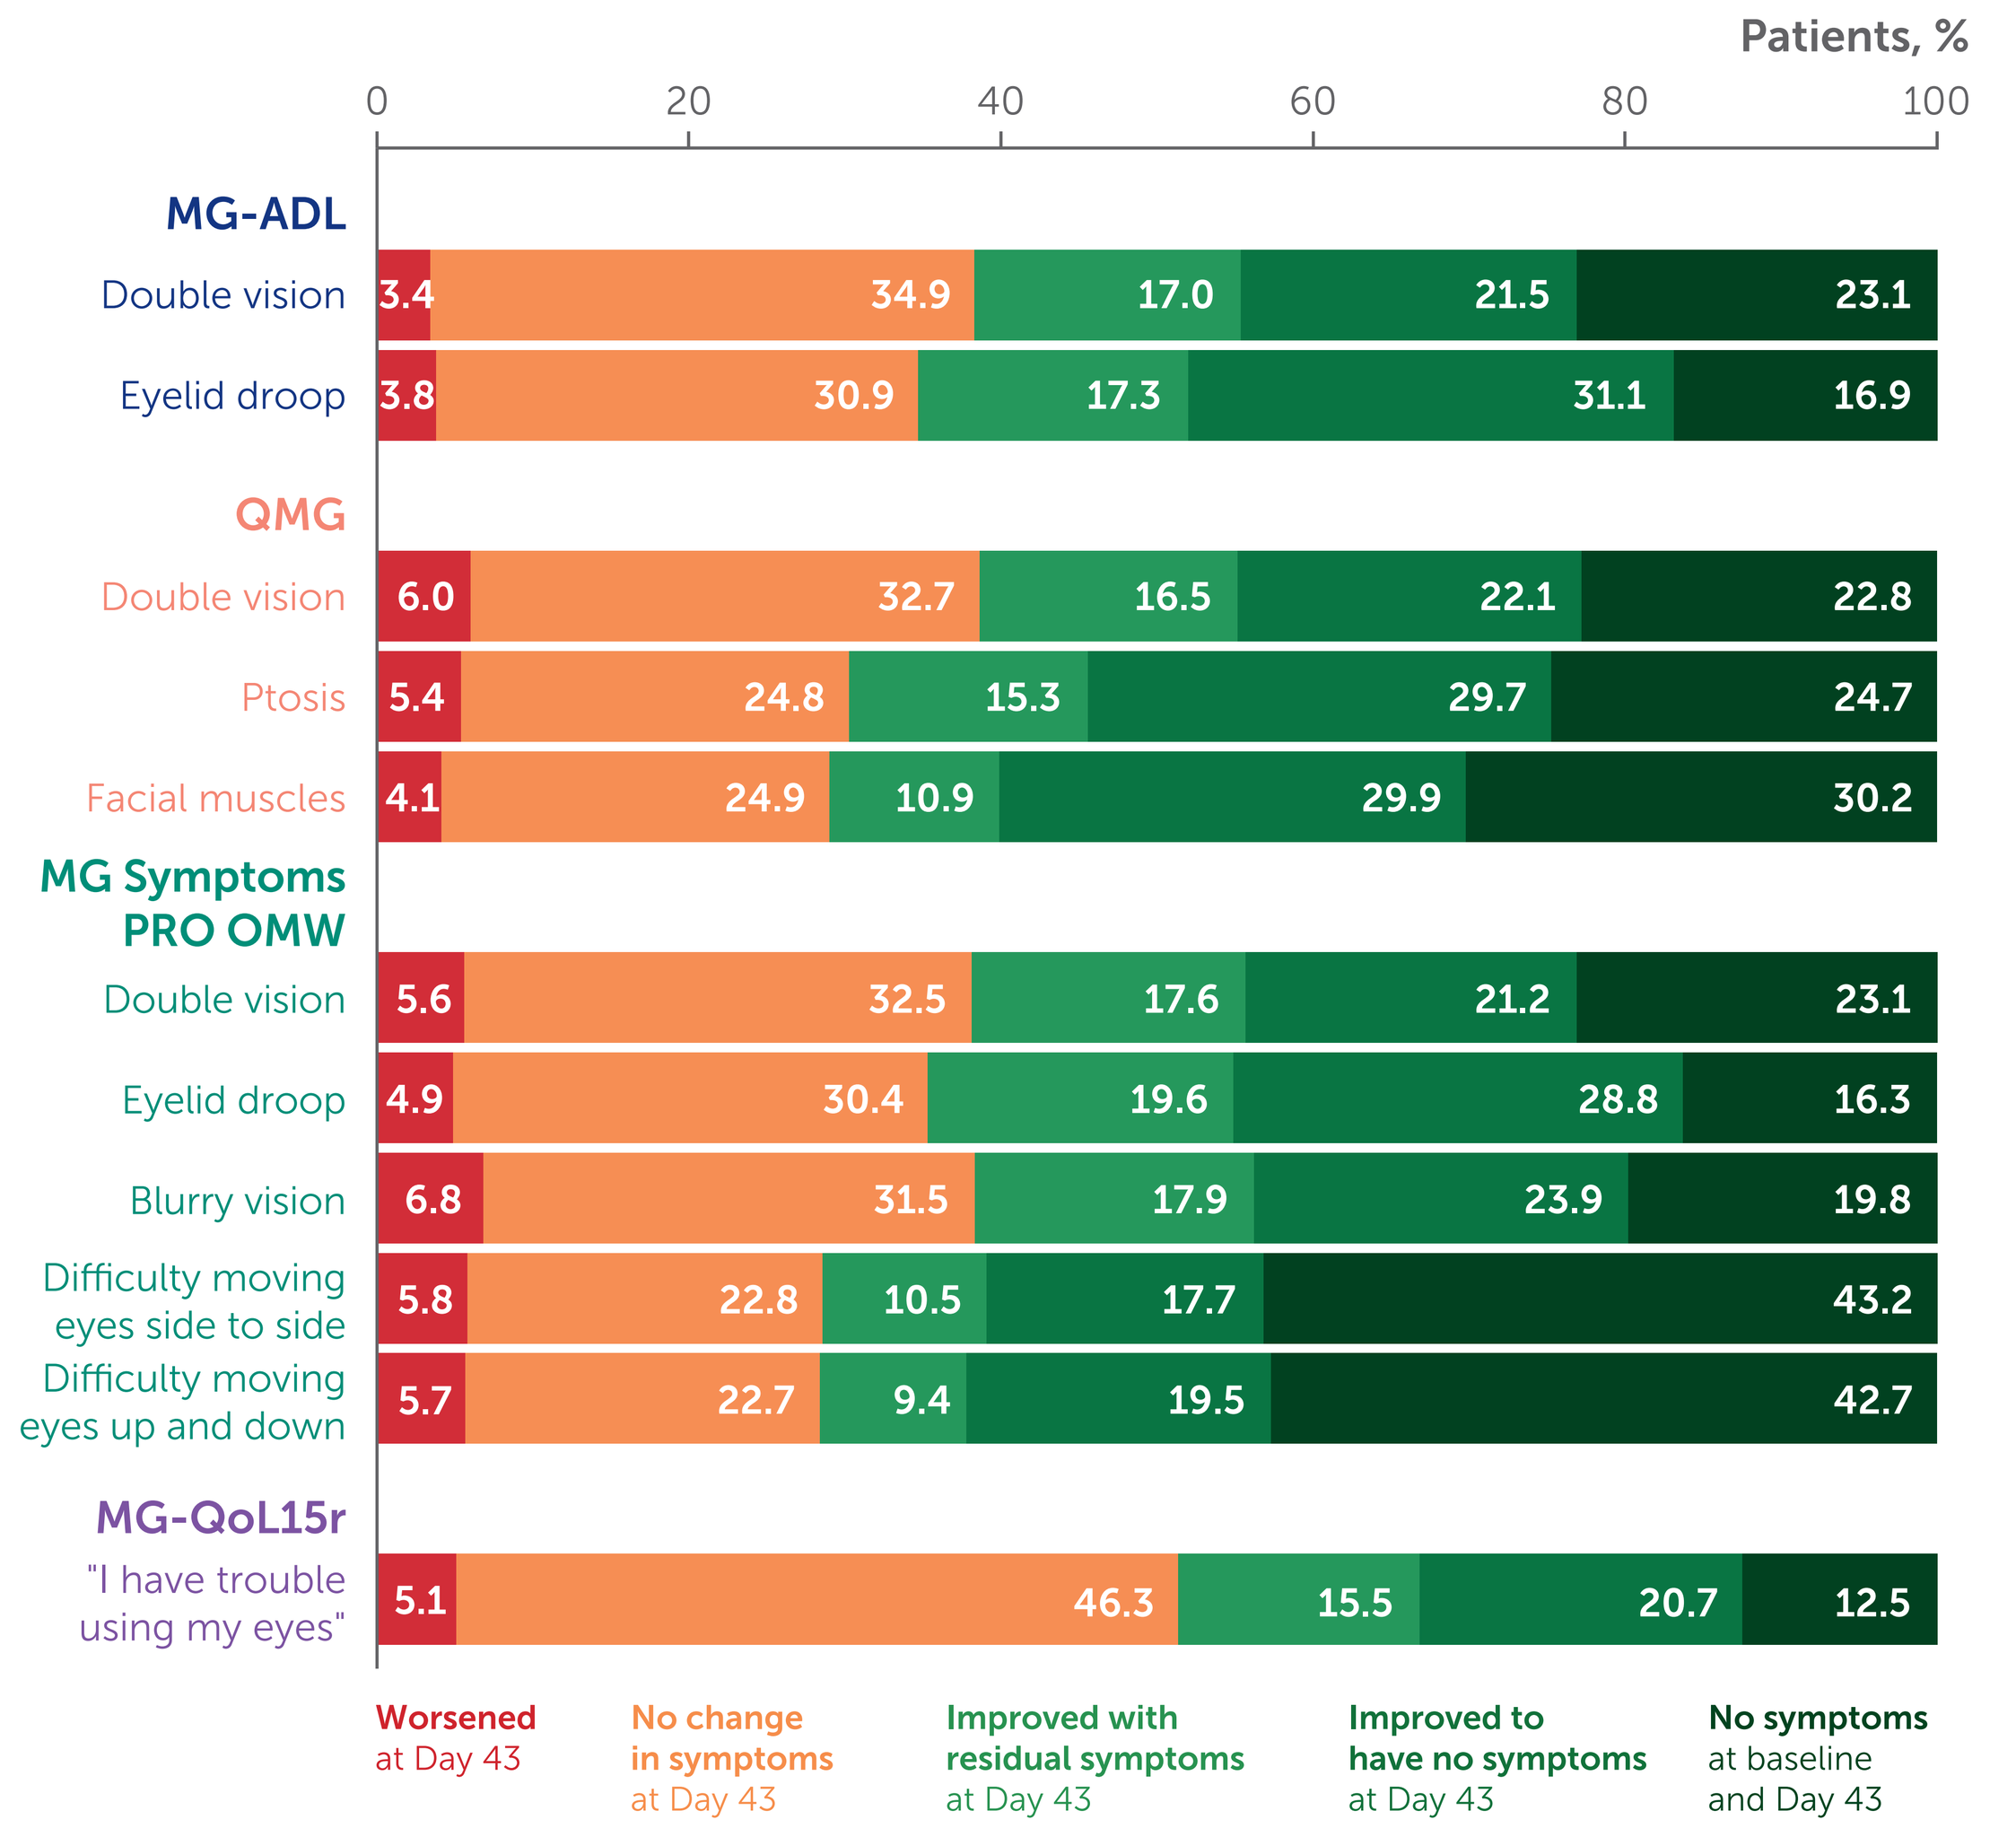
Patients, %
0
20
40
60
80
100
MG-ADL
Double vision 3.4 34.9 17.0 21.5 23.1
Eyelid droop 3.8 30.9 17.3 31.1 16.9
QMG
Double vision 6.0 32.7 16.5 22.1 22.8
Ptosis 5.4 24.8 15.3 29.7 24.7
Facial muscles 4.1 24.9 10.9 29.9 30.2
MG Symptoms PRO OMW
Double vision 5.6 32.5 17.6 21.2 23.1
Eyelid droop 4.9 30.4 19.6 28.8 16.3
Blurry vision 6.8 31.5 17.9 23.9 19.8
Difficulty moving eyes side to side 5.8 22.8 10.5 17.7 43.2
Difficulty moving eyes up and down 5.7 22.7 9.4 19.5 42.7
MG-QoL15r
"I have trouble using my eyes" 5.1 46.3 15.5 20.7 12.5
Worsened at Day 43
No change in symptoms at Day 43
Improved with residual symptoms at Day 43
Improved to have no symptoms at Day 43
No symptoms at baseline and Day 43

Abbreviations: Ab+, antibody positive; AChR, acetylcholine receptor; CS, corticosteroid; FcRn, neonatal fragment crystallizable receptor; (g)MG, (generalized) myasthenia gravis; IgG4, immunoglobulin G4; mAb, monoclonal antibody; MG-ADL, Myasthenia Gravis Activities of Daily Living; MGFA, Myasthenia Gravis Foundation of America; MG-QoL15r, Myasthenia Gravis Quality of Life 15-item revised; MuSK, muscle-specific tyrosine kinase; OLE, open-label extension; OMW, Ocular Muscle Weakness; PRO, patient-reported outcome; QMG, Quantitative Myasthenia Gravis; QoL, quality of life; RLZ, rozanolixizumab; SD, standard deviation; TEAE, treatment-emergent adverse event.

Acknowledgments: In memoriam of Prof. Vera Bril, whose invaluable contribution to this work is gratefully acknowledged. This study was funded by UCB. The authors acknowledge Bea Perks, PhD, for Ogilvy Health, London, UK, for editorial assistance, which was funded by UCB. The authors thank Veronica Porkess, PhD, of UCB for publication and editorial support. The authors thank the patients and their caregivers, in addition to the investigators and their teams who contributed to this study.

Author disclosures: Zabeen K. Mahuwala has received compensation for advisory board participation from Alexion Pharmaceuticals and Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine) and has served as a panellist for Academic CME. She has received research funding for clinical trials paid to the University of Kentucky from Alexion Pharmaceuticals, argenx, Immunovant, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), RemeGen Biosciences and UCB. Elena Cortés Vicente receives public speaking honoraria and compensation for advisory boards and/or consultation fees from Alexion Pharmaceuticals, argenx, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine) and UCB. Ali A. Habib has received research support and/or honoraria from Alexion/AstraZeneca Rare Disease, Amgen, Arcellx, argenx, Cabaletta Bio, Cartesian Therapeutics, COUR Pharmaceuticals, Genentech/Roche, Grifols, Immunovant, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Kyverna, Merck, MGNet, Nkarta, NMD Pharma, Novartis, Regeneron Pharmaceuticals and UCB. Robert M. Pascuzzi speaks at educational seminars on a broad variety of general neurology topics for primary care physicians through the organisation Medical Education Resources (an educational organisation with no links or ties to any pharmaceutical or healthcare business company). Therefore, Robert M. Pascuzzi has no conflicts of interest related to this research, manuscript, presentation or publication. John Vissing has been a Consultant on advisory boards for Amicus Therapeutics, Biogen, Edgewise Therapeutics, Fulcrum Therapeutics, Genethon, Horizon Therapeutics (now Amgen), Lupin, ML Biopharma, Novartis, Regeneron Pharmaceuticals, Roche, Sanofi Genzyme (now Sanofi), Sarepta Therapeutics and UCB. He has received research and travel support and/or speaker honoraria from Alexion Pharmaceuticals, argenx, Biogen, Edgewise Therapeutics, Fulcrum Therapeutics, Lupin, Sanofi Genzyme (now Sanofi) and UCB. He is a Principal Investigator in clinical trials for Alexion Pharmaceuticals, argenx, Atamyo Therapeutics, Genethon, Horizon Therapeutics (now Amgen), Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), ML Biopharma, Novartis, Regeneron Pharmaceuticals, Roche, Sanofi Genzyme (now Sanofi) and UCB. Tuan Vu is the USF Site Principal Investigator for MG clinical trials sponsored by Alexion/AstraZeneca Rare Disease, Amgen, argenx, Cartesian Therapeutics, COUR Pharmaceuticals, Dianthus Therapeutics, ImmunAbs, Immunovant, Johnson & Johnson, NMD Pharma, Regeneron Pharmaceuticals, UCB and Vor, and has served as a speaker for Alexion/AstraZeneca Rare Disease, Amgen, argenx and CSL Behring. He has performed consulting work for Alexion/AstraZeneca Rare Disease, argenx, Dianthus Therapeutics, NMD Pharma, Regeneron and Vor. Jos Bloemers, Paul Mahoney and Thaïs Tarancón are employees and shareholders of UCB. Vera Bril was a Consultant for Akcea, Alexion Pharmaceuticals, Alnylam, argenx, CSL, Grifols, Immunovant, Ionis, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Momenta (now Johnson & Johnson), Novo Nordisk, Octapharma, Pfizer, Powell Mansfield, Roche, Sanofi, Takeda Pharmaceuticals and UCB. She received research support from Akcea, Alexion Pharmaceuticals, argenx, CSL, Grifols, Immunovant, Ionis, Momenta (now Johnson & Johnson), Octapharma, Takeda Pharmaceuticals, UCB and Viela Bio (now Amgen).

References: 1. Gilhus NE, et al. Nat Rev Dis Primers. 2019;5(1):30. 2. Cornblath WT. Asia Pac J Ophthalmol (Phila). 2018;7(4):257–259. 3. Jacob S. Eye (Lond). 2024;38(12): 2422–2436. 4. RYSTIGGO® US PI. https://www.ucb-usa.com/RYSTIGGO-prescribing-information.pdf. Accessed August 2026. 5. Mahuwala ZK, et al. Neurology. 2025;104(7 Suppl 1):1866.

Please use this QR code to download a PDF of the poster.

These data were previously presented at ICNMD 2026, Florence, Italy; July 7–11, 2026