Poster 66

Vitaccess Real™ Myasthenia Gravis studies protocol: Registry-based mixed-methods studies on treatment outcomes in patients initiated on zilucoplan or rozanolixizumab in the real world

Zabeen Mahuwala[1], Kerina Bonar[2], Natasa Savic[3], Thaïs Tarancón[4], Jos Bloemers[5], Saiful Islam[2], Olivia Knowles[6], Carmen Petitjean[6], Hanna Skrobanski[6], Saiju Jacob[7]

1 University of Kentucky Department of Neurology, Lexington, Kentucky, United States; 2 UCB, Slough, United Kingdom; 3 UCB, Bulle, Switzerland; 4 UCB, Madrid, Spain; 5 UCB, Breda, The Netherlands; 6 Vitaccess, London, United Kingdom; 7 University Hospitals Birmingham, Birmingham, United Kingdom

Background

- Zilucoplan (complement C5 inhibitor) and rozanolixizumab (neonatal Fc receptor blocker) are targeted therapies that have demonstrated efficacy in phase 3 trials in gMG.[1-4]
- However, real-world evidence remains limited and is needed to characterize treatment outcomes, utilization and patient experience in routine clinical practice.

Objective

To characterize real-world clinical outcomes and patient experience in adults with gMG initiating zilucoplan or rozanolixizumab.

Study design

The Vitaccess Real™ MG REALITY studies employ a registry-based **concurrent embedded mixed-methods** study design with quantitative and qualitative components (Figure 1).

Data are collected from multiple sources within the registry, including:

PROs via the VRMG platform | *Clinician-reported eCRFs* | *EMRs*

Qualitative interview data will complement quantitative outcomes, providing richer insight into patients' experiences, treatment satisfaction, and the real-world impacts of these therapies. See Table 1 for study outcomes.

Study population

Quantitative cohort
- Adults (≥18 years)
- With AChR or MuSK positive gMG
- Enrolled in the VRMG registry
- Residing in North America or Europe (Figure 2)
- Initiated zilucoplan or rozanolixizumab

Qualitative cohort
A subset of treated participants will be eligible for qualitative interviews.

Participants are recruited into the VRMG registry via:

clinical sites | *community neurology practices* | *direct-to-patient pathways*

Figure 2. Site locations currently recruiting for the VRMG registry used in the REALITY studies (subject to expansion)

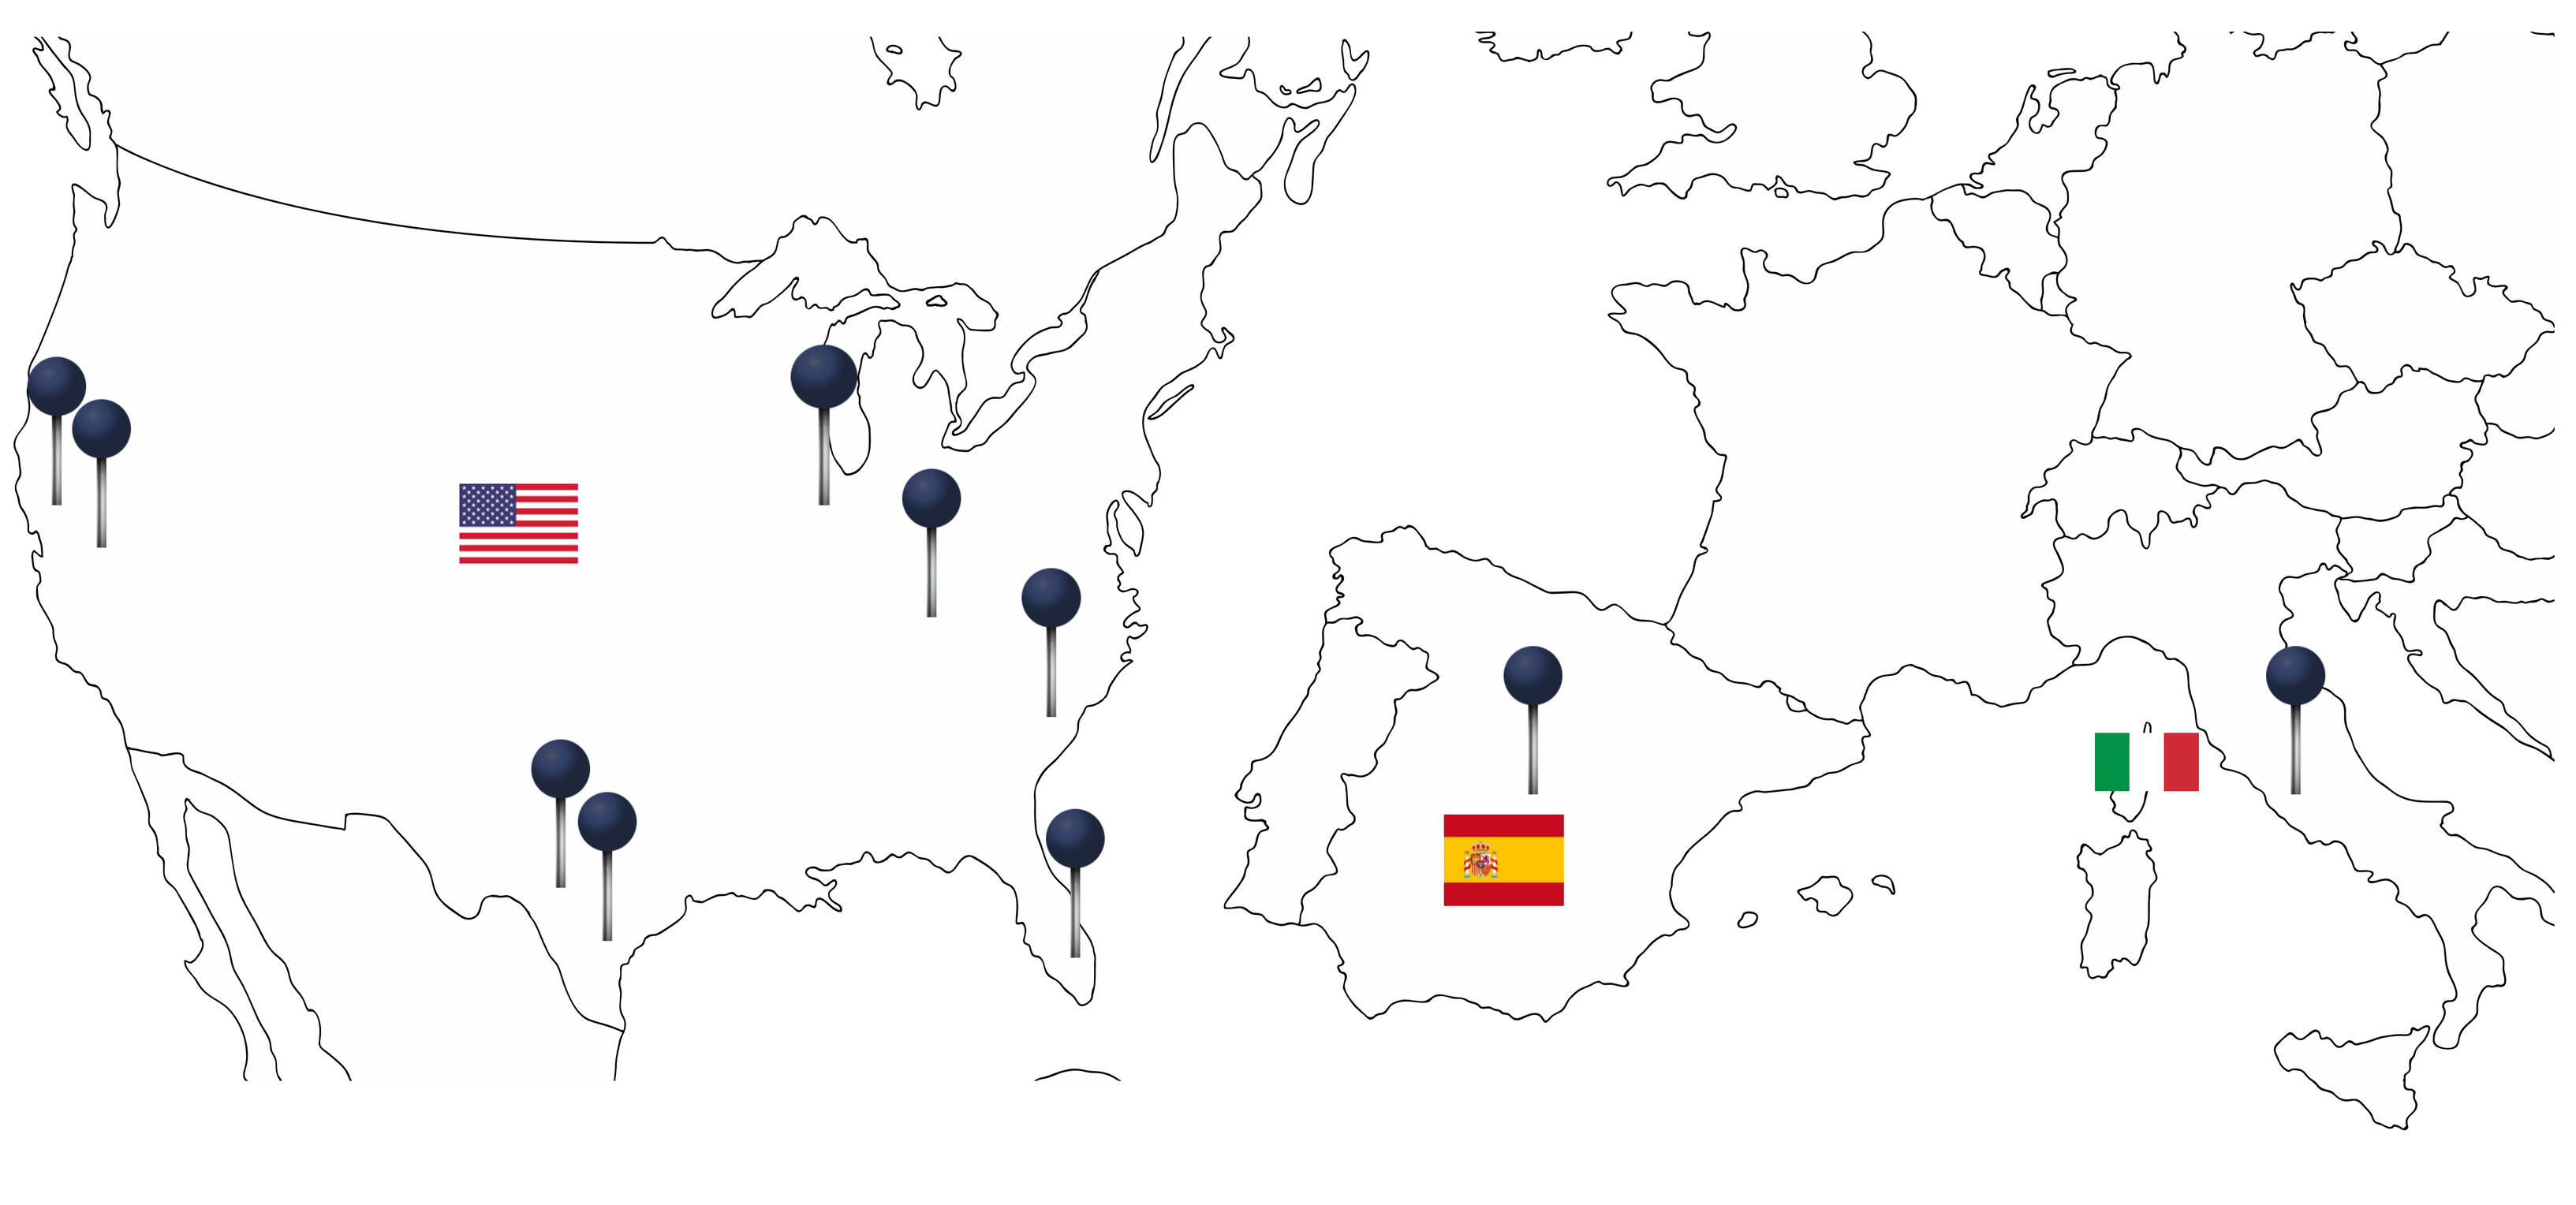

Data analysis

Quantitative component
- Descriptive statistics for baseline characteristics.
- Longitudinal analyses from baseline over 12 months using mixed models for repeated measures of change, adjusted for key baseline covariates.
- Treatment utilization, medication use and HCRU summaries at 6 and 12 months.
- Subgroup and sensitivity analyses where feasible.

Qualitative component
- Thematic analysis using Braun and Clarke's framework.[5]

Integration
- Joint displays to assess convergence, complementarity and divergence.

Figure 1. Registry-based mixed-methods study design

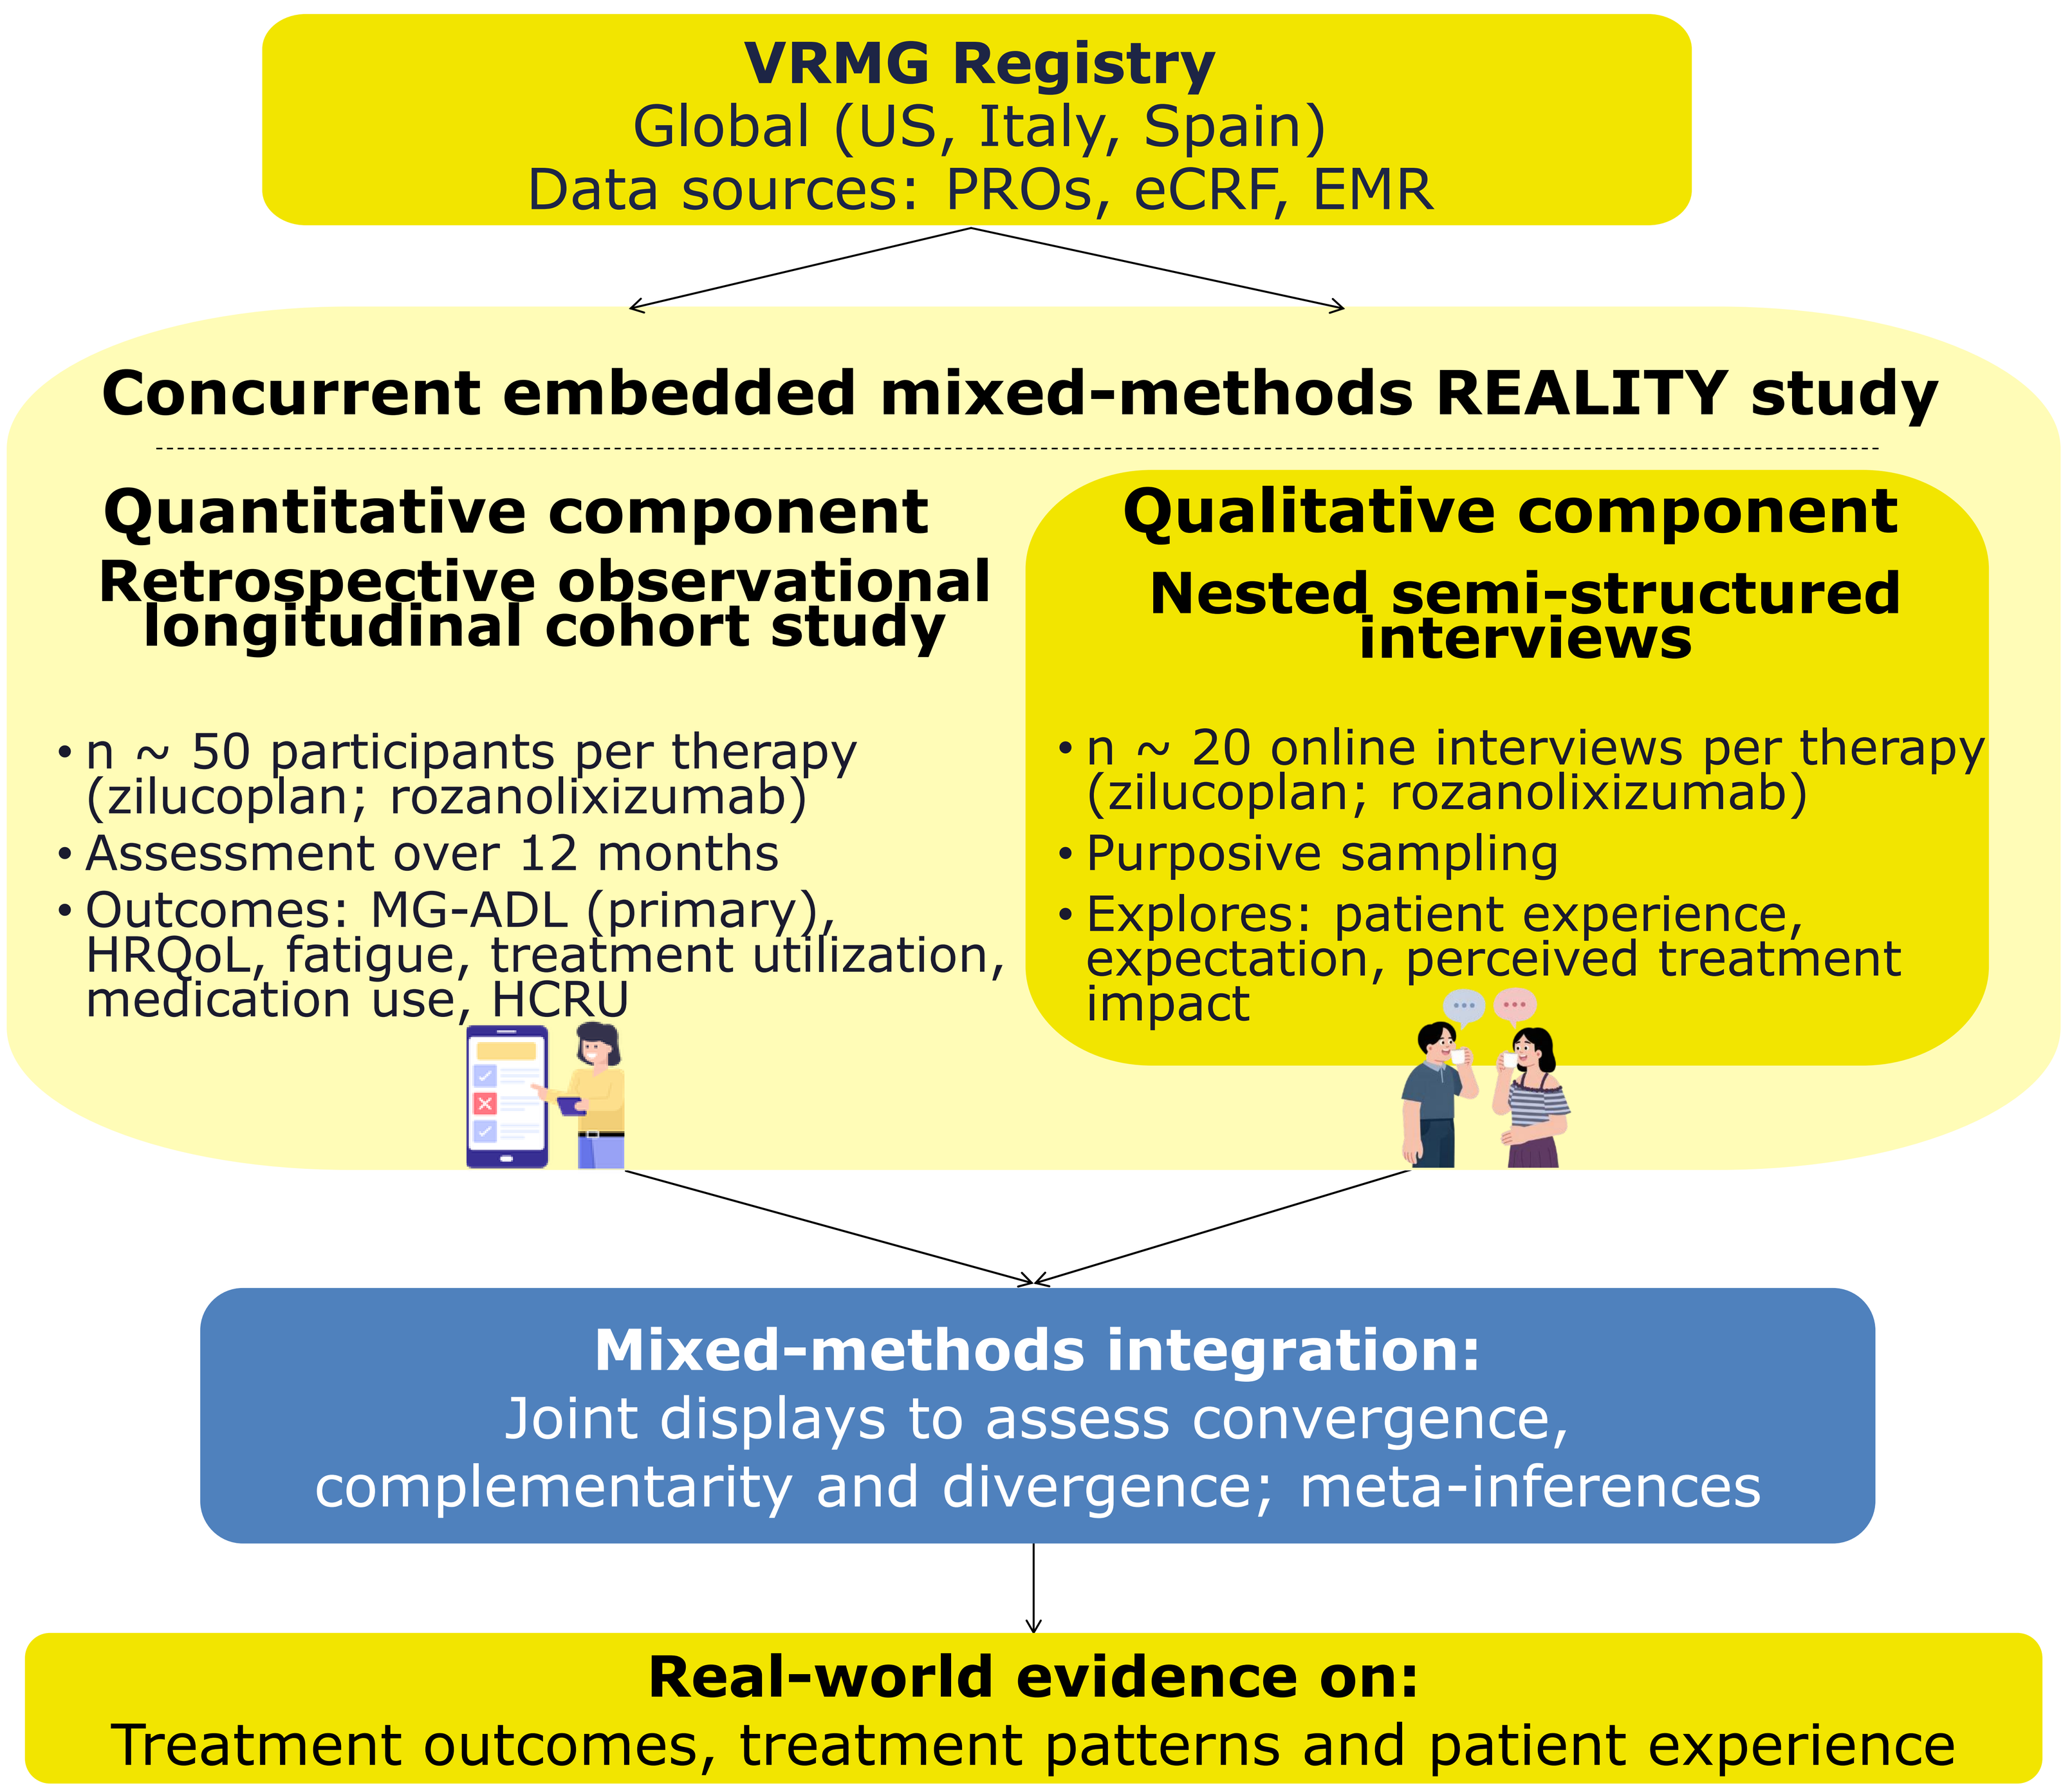
VRMG Registry
Global (US, Italy, Spain)
Data sources: PROs, eCRF, EMR
Concurrent embedded mixed-methods REALITY study
Quantitative component
Retrospective observational longitudinal cohort study
• n ~ 50 participants per therapy (zilucoplan; rozanolixizumab)
• Assessment over 12 months
• Outcomes: MG-ADL (primary), HRQoL, fatigue, treatment utilization, medication use, HCRU
Qualitative component
Nested semi-structured interviews
• n ~ 20 online interviews per therapy (zilucoplan; rozanolixizumab)
• Purposive sampling
• Explores: patient experience, expectation, perceived treatment impact
Mixed-methods integration:
Joint displays to assess convergence, complementarity and divergence; meta-inferences
Real-world evidence on:
Treatment outcomes, treatment patterns and patient experience

Outcomes

Table 1. Registry-based mixed-methods study outcomes

Category	Outcomes	Assessment timing
Baseline characteristics	Demographics, diagnostic and clinical characteristics, medical history and prior treatment history	Baseline
Primary outcome	Change from baseline in MG-ADL total score	Baseline over 12 months
Other PROs	Change from baseline: • MG-QoL-15r total score • MG Symptoms PRO Physical Fatigue Scale total score • NeuroQoL Fatigue SF total score	Baseline over 12 months
Treatment utilization patterns	• Treatment utilization (duration, discontinuation) • Administration (method [rozanolixizumab only], frequency)	Baseline, 6 and 12 months
Medication use	• Oral steroids • Non-steroidal immunosuppressants • Rescue therapies	Baseline, 6 and 12 months
HCRU	Healthcare visits and hospitalizations by setting (clinic or outpatient; emergency visits; follow up and results; home visits; hospital inpatient; telemedicine appointments), including gMG exacerbation-related utilization, and duration and length of stay	Baseline, 6 and 12 months
Qualitative outcomes	Lived experience, perceived changes since treatment initiation, expectations, satisfaction and burden	June to November 2026

Results

- Recruitment began in August 2024. Registry progress (August 2026):
 - 11 clinical sites recruiting, 250 patients enrolled.
 - Currently 10 eligible participants either receiving zilucoplan or rozanolixizumab.
- **Quantitative component**: Interim analysis planned following achievement of approximately 50% of the target sample size.
- **Qualitative component:** Qualitative interviews are planned between June and November 2026, with findings expected to be integrated with the quantitative results in the final study reporting.

Conclusion

- This registry-based mixed-methods design will provide a patient-centric understanding of real-world treatment outcomes in adults with gMG initiating zilucoplan or rozanolixizumab.
- By integrating longitudinal quantitative outcomes with in-depth qualitative patient experience data, the study will capture treatment utilization patterns, medication use, HCRU, PROs and patients lived experiences of, and perspectives on, the treatment.
- This approach is particularly valuable in rare diseases such as gMG, where real-world evidence can complement clinical trial data and support more informed, patient-centered care and decision-making.

Abbreviations:
AChR, acetylcholine receptor; eCRFs, electronic case report forms; EMRs, electronic medical records; gMG, generalized myasthenia gravis; HCRU, healthcare resource utilization; HRQoL, health-related quality of life; MG-ADL, Myasthenia Gravis Activities of Daily Living; MG-QoL-15r, Myasthenia Gravis Quality of Life 15-item Revised; MG Symptoms PRO Fatigue Scale, Myasthenia Gravis Symptoms Patient-Reported Outcome Physical Fatigue Scale; MuSK, muscle-specific kinase; NeuroQoL Fatigue SF, Quality of Life in Neurological Disorders Short Form – Fatigue; NS-IST, non-steroidal immunosuppressive therapy; PRO, patient-reported outcome; VRMG, Vitaccess Real™ Myasthenia Gravis

References

1: Howard JF et al. JAMA Neurol 2020;77(5):582;
2: Bril V et al. Neurology 2021;96(6):e853–e865;
3: Howard JF et al. Lancet Neurol 2023;22(5):395–406;
4: Bril V et al. Lancet Neurol 2023;22(5):383–394;
5: Braun V et al. Qual Res Psychol 2006;3(2):77–101.

Vitaccess Ltd. is the Sponsor of the Vitaccess Real™ MG Registry. UCB is the first subscriber of the Vitaccess Real™ MG Registry and has funded its setup and the REALITY studies.

Scan QR code for digital poster

Acknowledgements: The authors thank all patients and their caregivers for participating in this study, as well as all investigators and site staff for their contributions and acknowledge Patrik Öhagen for his contributions to this study.

Disclosures: KB, NS, TT, JB, and SI are employees of UCB and may hold shares in the company. OK, CP and HS are employees of Vitaccess, which served as a paid consultant to UCB in connection with the design of this study and the development of this publication. ZM has received compensation for advisory board participation from Alexion Pharmaceuticals and Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine) and has served as a panellist for Academic CME. She has received research funding for clinical trials paid to the University of Kentucky from Alexion Pharmaceuticals, argenx, Immunovant, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), RemeGen Biosciences and UCB. She has received consulting fees and reimbursement of expenses from Vitaccess. SJ has served as an international advisory board member for Alexion, Alnylam, argenx, Immunovant, Johnson and Johnson, Merck, Novartis, Regeneron and UCB; is currently an expert panel member of the Myasthenia Gravis consortium for argenx and UCB; and has received speaker fees from Terumo BCT and Eisai Pharmaceuticals. He has received consulting fees and reimbursement of expenses from Vitaccess.