Poster **100**

Clinical determinants of new rozanolixizumab treatment cycles in patients with generalized myasthenia gravis: A *post hoc* analysis of the Phase 3 MycarinG and open-label extension studies

Zabeen K. Mahuwala[1], Ali A. Habib[2], Robert M. Pascuzzi[3], Sabrina Sacconi[4], John Vissing[5], Tuan Vu[6], Bethany George[7], Thaïs Tarancón[8]

[1]Department of Neuromuscular Medicine, Epilepsy and Clinical Neurophysiology, University of Kentucky, Lexington, KY, USA; [2]MDA ALS & Neuromuscular Center, Department of Neurology, University of California, Irvine, Orange, CA, USA; [3]Neurology Department, Indiana University School of Medicine, Indiana University Health, Indianapolis, IN, USA; [4]Université Côte d'Azur, Peripheral Nervous System & Muscle Department, Pasteur 2 Hospital, Centre Hospitalier Universitaire de Nice, Nice, France; [5]Copenhagen Neuromuscular Center, Department of Neurology, Rigshospitalet, University of Copenhagen, Copenhagen, Denmark; [6]Department of Neurology, University of South Florida Morsani College of Medicine, Tampa, FL, USA; [7]UCB, Slough, UK; [8]UCB, Madrid, Spain

MGFA Scientific Session at AANEM 2026, Orlando, FL, USA; September 29, 2026

Introduction

- Rozanolixizumab is a humanized IgG4 mAb that targets and blocks FcRn with high affinity, approved for the treatment of adult patients with anti-AChR Ab+ and anti-MuSK Ab+ gMG[1,2]
- In the Phase 3 MycarinG study (NCT03971422), one 6-week cycle of rozanolixizumab (7 mg/kg or 10 mg/kg) significantly improved MG-ADL and QMG scores versus placebo[3]
- After MycarinG, patients could enroll in OLE studies MG0004 (NCT04124965) then MG0007 (NCT04650854), or MG0007 directly
- In MG0007, after an initial 6-week treatment cycle, subsequent rozanolixizumab cycles were initiated at the investigator's discretion, with advisory protocol guidance to consider a new cycle upon ≥2-point MG-ADL or ≥3-point QMG score increase
- **The objective of this *post hoc* analysis was to understand the factors leading to the initiation of a new rozanolixizumab cycle during long-term rozanolixizumab treatment**

Methods

- In MycarinG, patients were aged ≥18 years with MGFA Clinical Class II–IVa anti-AChR Ab+ or anti-MuSK Ab+ gMG, with an MG-ADL score ≥3 (for non-ocular symptoms) and a QMG score ≥11
- Patients who completed MycarinG or whose disease severity worsened during the treatment-free observation period were re-randomized to receive rozanolixizumab 7 mg/kg or 10 mg/kg, given once weekly in MG0004 or as symptom-driven cycles in MG0007
- Final data from patients with available MG-ADL and QMG data who had received or were awaiting a symptom-driven cycle were pooled across MycarinG, MG0004 (first 6 weeks, representing the only symptom-driven cycle in this study) and MG0007
- MG-ADL and QMG score changes during the treatment-free period from Day 43 in the previous cycle to the baseline of the new cycle, up to Cycle 13, were analyzed *post hoc* in patients receiving or awaiting a new cycle after initial rozanolixizumab treatment

Results

- Overall, 167 patients had received rozanolixizumab treatment and had initiated or were awaiting a symptom-driven treatment cycle (**Table 1**), of whom 147 had available MG-ADL and/or QMG data
- Among patients with available MG-ADL and/or QMG data, a total of 908 cycles were analyzed
- Initiation of a new rozanolixizumab cycle was generally preceded by worsening in at least one assessment, defined as a ≥2-point MG-ADL or ≥3-point QMG score increase (**Figure 1**):
 - In total, 619/908 (68.2%) and 557/908 (61.3%) cycles were initiated having met the definition of MG-ADL and QMG score worsening, respectively
 - Initiation of new cycles followed worsening on either or both scales in 694/908 (76.4%) cycles, and on neither scale in 140/908 (15.4%) cycles. Both QMG and MG-ADL data were missing for 74/908 (8.1%) cycles
- New rozanolixizumab treatment cycles were initiated following a mean (SD) worsening of 3.8 (3.3) and 4.9 (4.6) points in MG-ADL and QMG, respectively
- Changes in MG-ADL and QMG scores from the end of one cycle (Day 43) to the start of the next were generally consistent across cycles. Small increases in score changes between cycles were observed over the study, however these were not statistically significant (**Figure 2**)
- Overall, 214 cycles were initiated without following the protocol guidance for worsening on MG-ADL or QMG (i.e., scores had not met the threshold for worsening or had not been recorded). In a subset of these cycles, worsening was captured by other measures:
 - Worsening of MG Symptoms PRO Physical Fatigue scores was observed in 36.9% (n/N=79/214) of cycles, which was above the clinically meaningful threshold of 20.0 points in 4.2% (n/N=9/214) of cycles. Fatigue is a particularly important symptom to patients
 - Worsening of MG-QoL15r scores was observed in 25.2% (n/N=54/214) of cycles

Summary and conclusions

In the MycarinG and OLE studies, initiation of a new cycle of rozanolixizumab treatment was at the investigator's discretion, with advisory protocol guidance to consider a new cycle when MG-ADL or QMG scores worsened

In this *post hoc* analysis, we found that new cycles of rozanolixizumab were generally initiated following deterioration in MG-ADL and/or QMG scores

Some patients received new rozanolixizumab cycles without prior worsening in MG-ADL or QMG scores. Some of these patients experienced worsening on other measures, including the MG Symptoms PRO Physical Fatigue scale and the MG-QoL15r

Initiation of additional rozanolixizumab treatment cycles can be individualized based on patients' symptoms, including fatigue or impaired quality of life

Table 1 Baseline demographics and disease characteristics were indicative of a broad population of patients with gMG

		RLZ total (N=167)
	Age, mean (SD)	**52.7** (16.3)
	Female, n (%)	**96** (57.5)
MGFA Clinical Class, n (%)	II	**69** (41.3)
	III	**93** (55.7)
	IV	**5** (3.0)
	Duration of disease, years, mean (SD)*	**8.4** (8.5)
	Anti-AChR Ab+, n (%)	**152** (91.0)
	Anti-MuSK Ab+, n (%)	**16** (9.6)
	MG-ADL score, mean (SD)	**8.4** (3.5)
	QMG score, mean (SD)	**15.7** (3.6)

*From diagnosis.

Figure 1 New cycles were generally initiated following a worsening in MG-ADL and/or QMG score

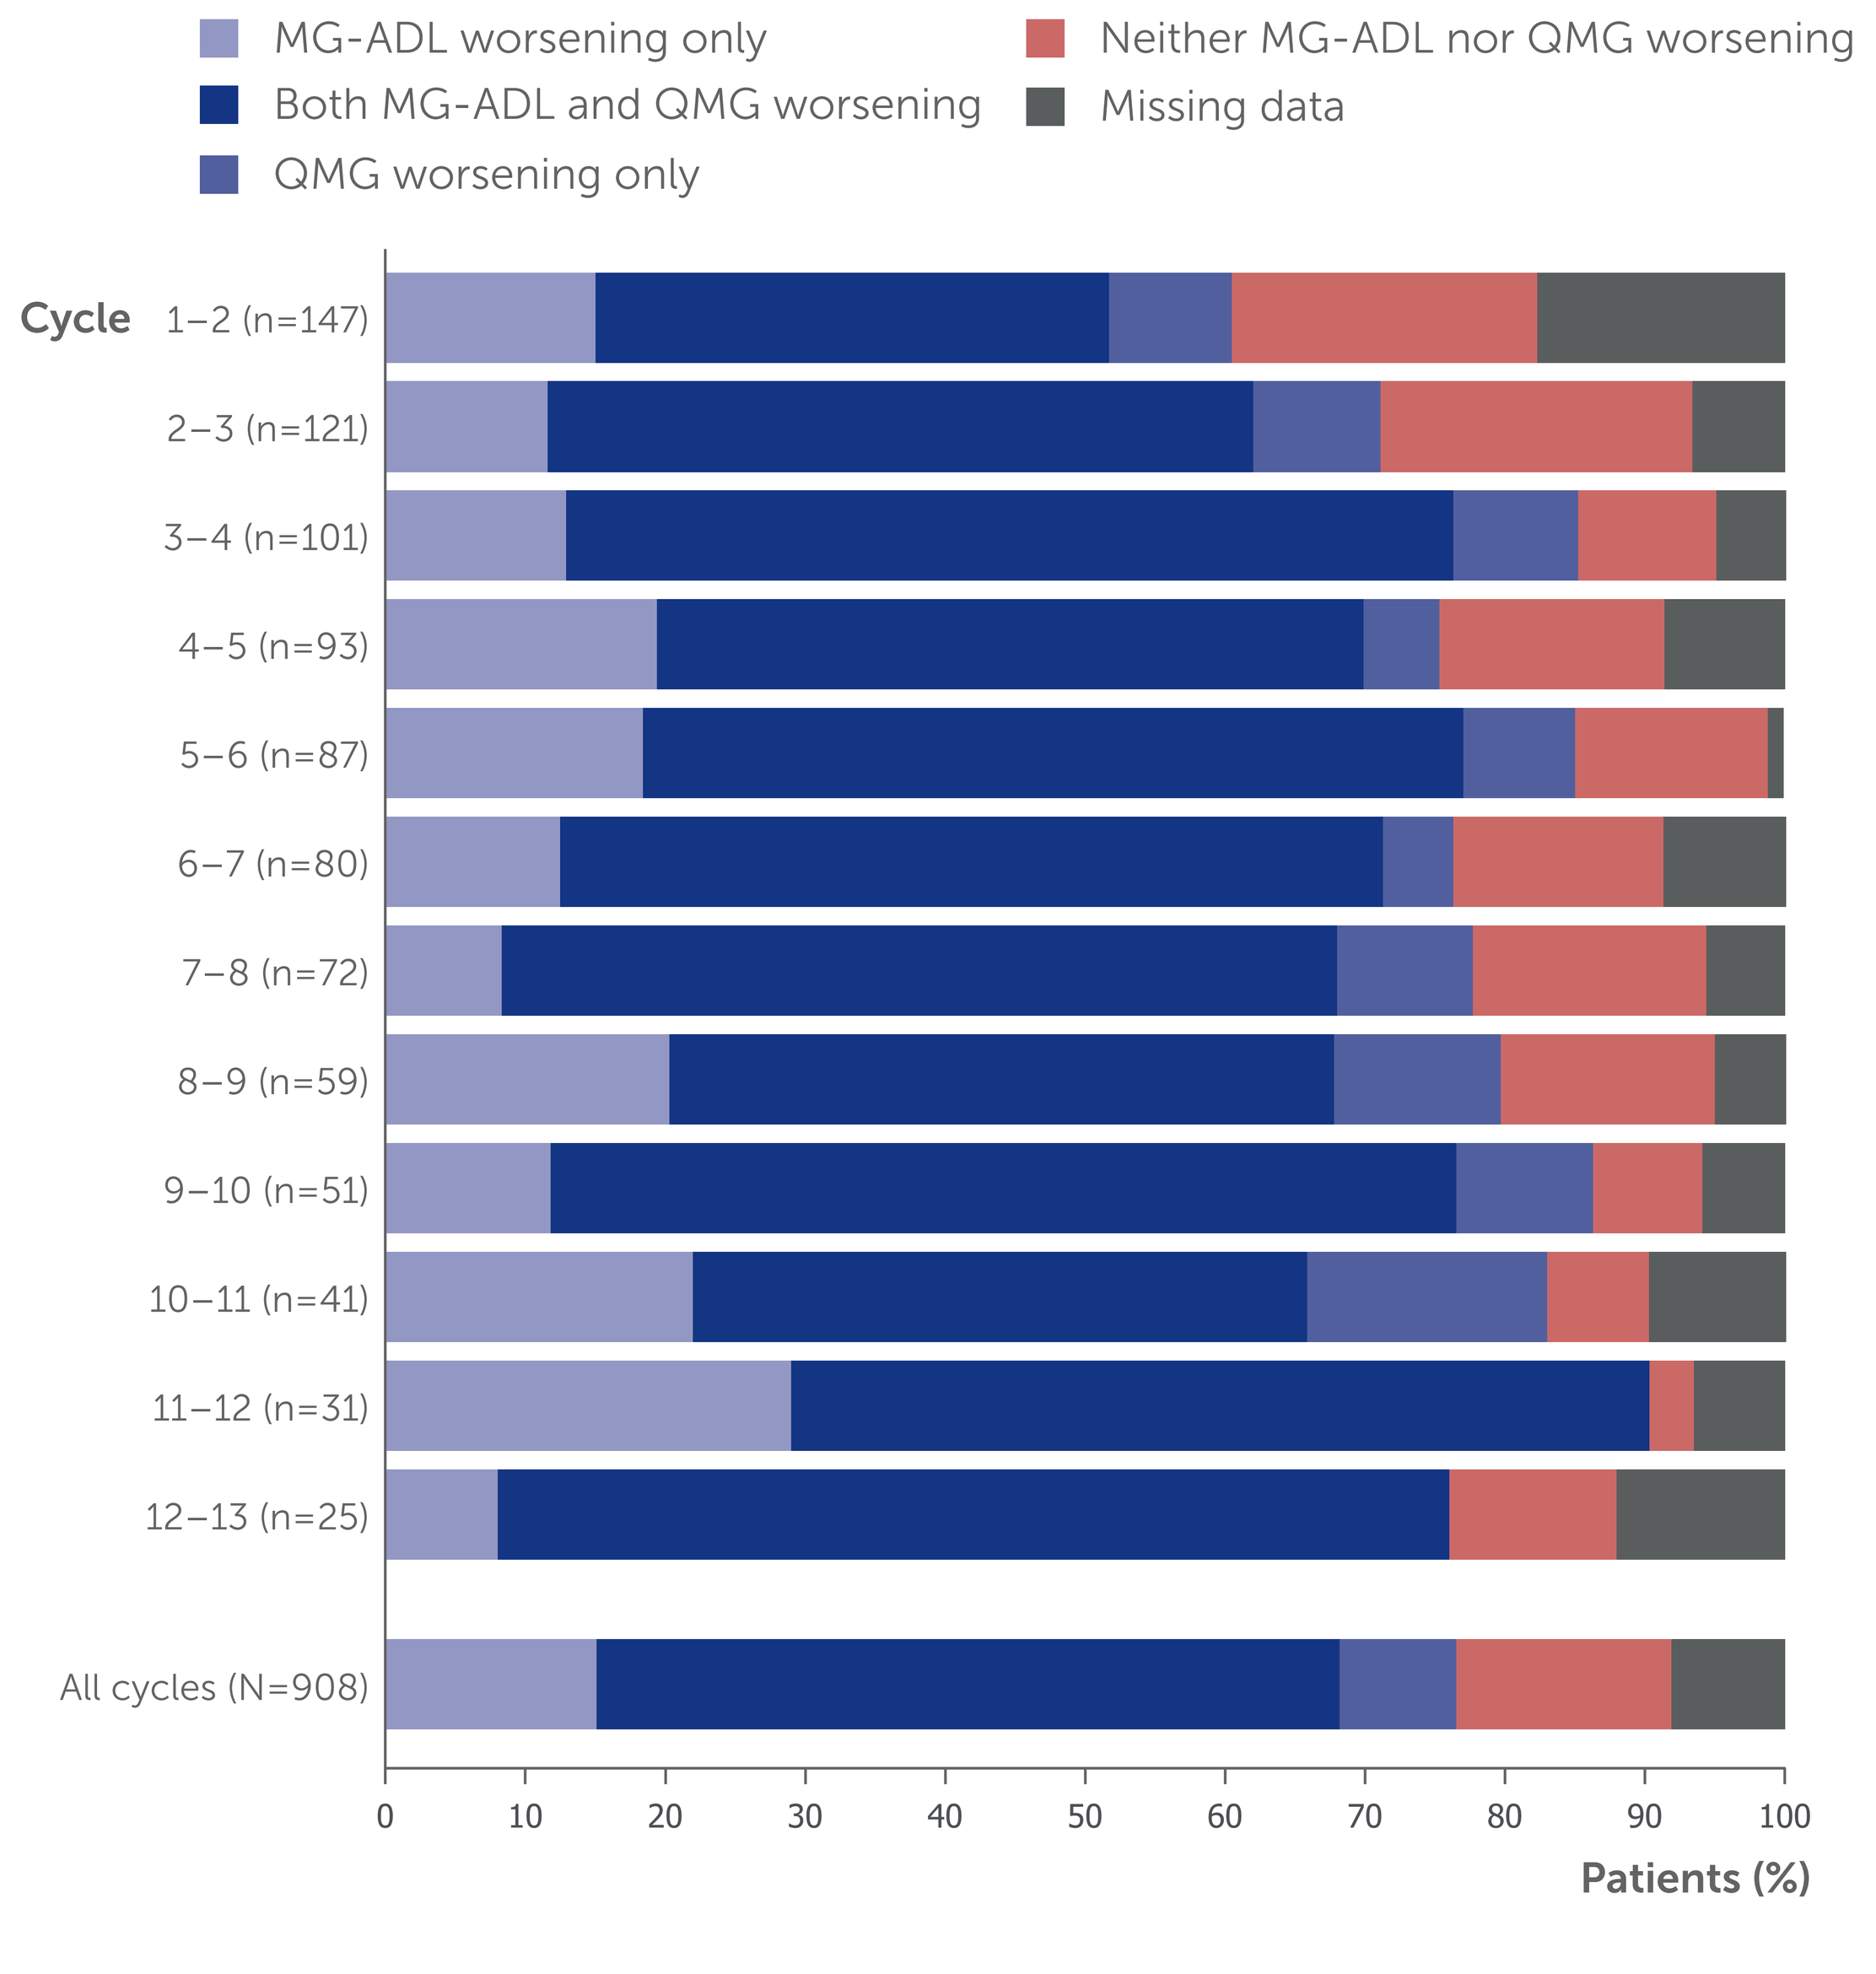
MG-ADL worsening only
Both MG-ADL and QMG worsening
QMG worsening only
Neither MG-ADL nor QMG worsening
Missing data
Cycle
1–2 (n=147)
2–3 (n=121)
3–4 (n=101)
4–5 (n=93)
5–6 (n=87)
6–7 (n=80)
7–8 (n=72)
8–9 (n=59)
9–10 (n=51)
10–11 (n=41)
11–12 (n=31)
12–13 (n=25)
All cycles (N=908)
0 10 20 30 40 50 60 70 80 90 100
Patients (%)

Figure 2 Changes in MG-ADL and QMG scores during the treatment-free period from Day 43 of one rozanolixizumab cycle to the start of the next cycle were generally consistent across cycles

Making sense of this information
y= The change in scores during the treatment-free period between cycles
x= Density of patients
The widest part of the shape represents the most common change in score between cycles
Interquartile range <Q1/>Q3

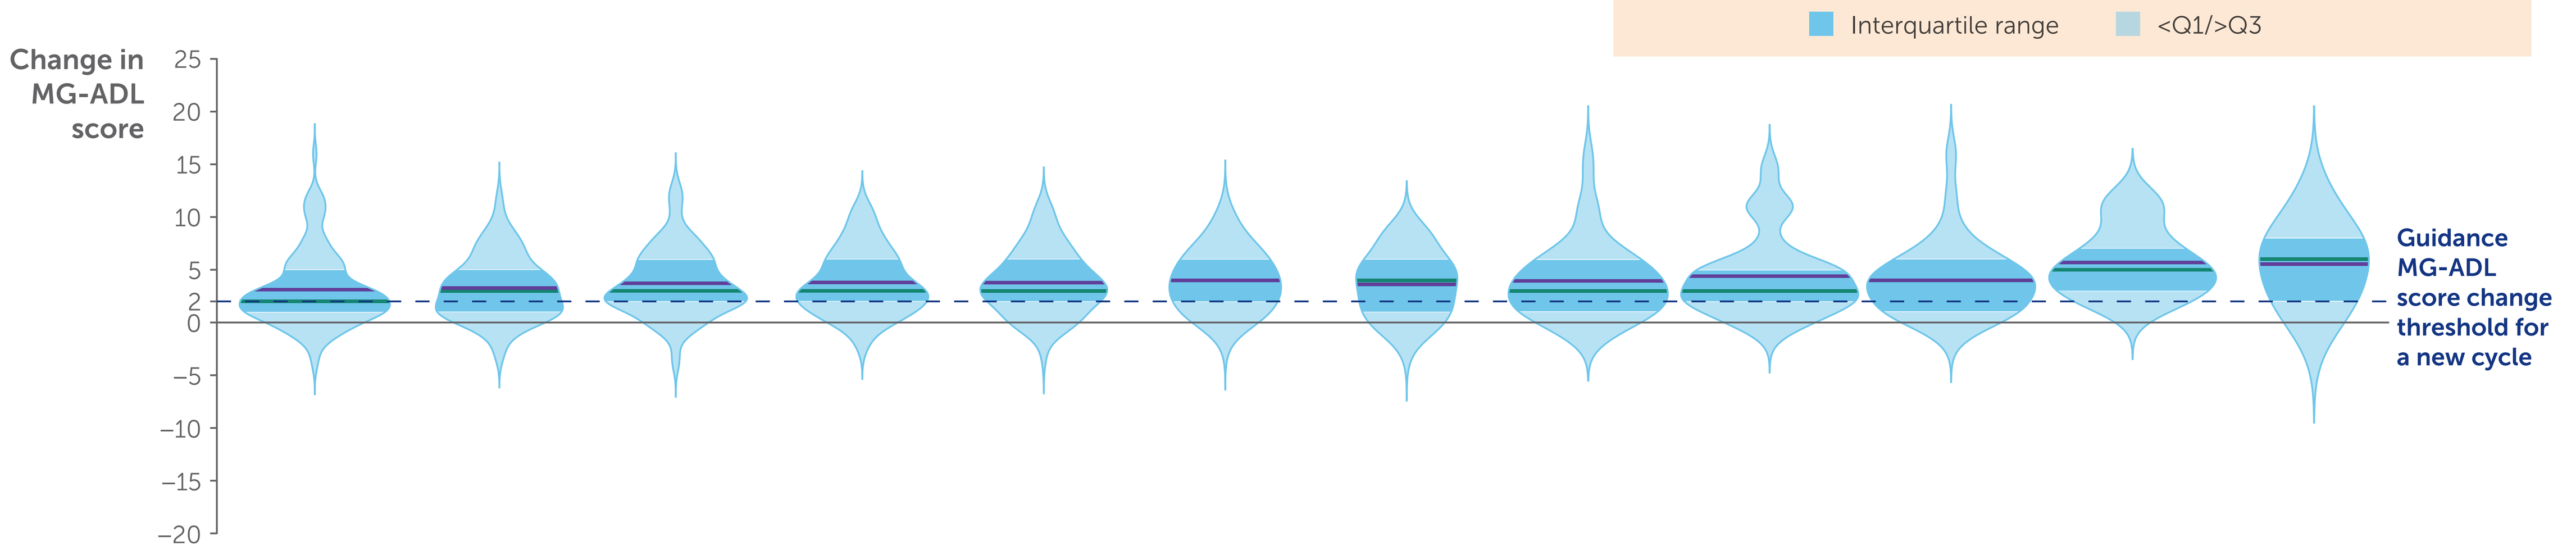
Change in MG-ADL score
25
20
15
10
5
2
0
−5
−10
−15
−20
Guidance MG-ADL score change threshold for a new cycle

Cycles	1–2	2–3	3–4	4–5	5–6	6–7	7–8	8–9	9–10	10–11	11–12	12–13
N	147	121	101	93	87	80	72	59	51	41	31	25
Mean	3.1	3.3	3.7	3.8	3.8	4.0	3.6	3.9	4.4	4.0	5.7	5.5
Median	2	3	3	3	3	4	4	3	3	4	5	6

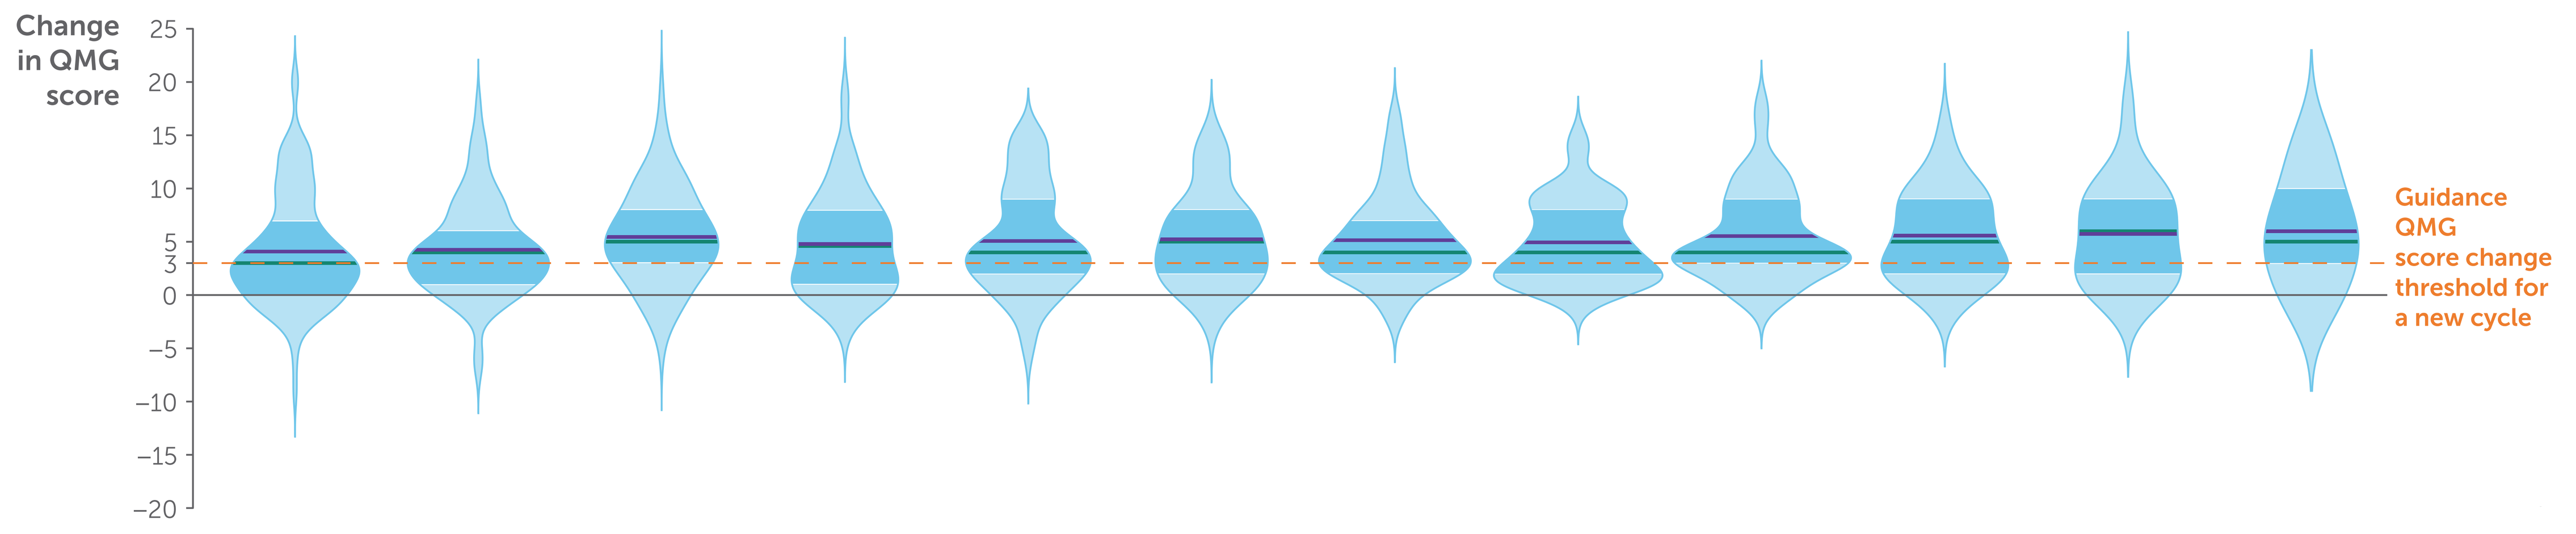
Change in QMG score
25
20
15
10
5
3
0
−5
−10
−15
−20
Guidance QMG score change threshold for a new cycle

Cycles	1–2	2–3	3–4	4–5	5–6	6–7	7–8	8–9	9–10	10–11	11–12	12–13
N	147	121	101	93	87	80	72	59	51	41	31	25
Mean	4.1	4.2	5.5	4.8	5.1	5.2	5.1	4.9	5.5	5.6	5.7	6.0
Median	3	4	5	5	4	5	4	4	4	5	6	5

Abbreviations: Ab+, antibody positive; AChR, acetylcholine receptor; FcRn, neonatal fragment crystallizable receptor; gMG, generalized myasthenia gravis; IgG4, immunoglobulin G subunit 4; mAb, monoclonal antibody; MG-ADL, Myasthenia Gravis Activities of Daily Living; MGFA, Myasthenia Gravis Foundation of America; MG Symptoms PRO, Myasthenia Gravis Symptoms Patient-Reported Outcomes; MG-QoL15r, Myasthenia Gravis Quality of Life 15-item revised; MuSK, muscle-specific tyrosine kinase; OLE, open-label extension; Q, quartile; QMG, Quantitative Myasthenia Gravis; RLZ, rozanolixizumab; SD, standard deviation.

Acknowledgments: This study was funded by UCB. The authors acknowledge Annabel Dimmock, MBiol, of Ogilvy Health, London, UK, for editorial assistance, which was funded by UCB. The authors thank Veronica Porkess, PhD, CMPP, of UCB for publication and editorial support. The authors thank the patients and their caregivers, in addition to the investigators and their teams who contributed to this study.

Author disclosures: Zabeen K. Mahuwala has received compensation for advisory board participation from Alexion Pharmaceuticals and Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine) and has served as a panellist for Academic CME. She has received research funding for clinical trials paid to the University of Kentucky from Alexion Pharmaceuticals, argenx, Immunovant, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), RemeGen Biosciences and UCB. Ali A. Habib has received research support and/or honoraria from Alexion/AstraZeneca Rare Disease, Amgen, Arcellx, argenx, Cabaletta Bio, Cartesian Therapeutics, COUR Pharmaceuticals, Genentech/Roche, Grifols, Immunovant, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Kyverna, Merck, MGNet, Nkarta, NMD Pharma, Novartis, Regeneron Pharmaceuticals and UCB. Robert M. Pascuzzi is Professor Emeritus of Neurology at Indiana University and receives compensation for his professional work from Indiana University Health. He has no financial relationship with any pharmaceutical company and receives no compensation from any pharmaceutical company (present or past). Robert M. Pascuzzi speaks at educational seminars on a broad variety of general neurology topics for primary care physicians through the organisation Medical Education Resources (an educational organisation with no links or ties to any pharmaceutical or healthcare business company). Therefore, Robert M. Pascuzzi has no conflicts of interest related to this research, manuscript, presentation or publication. Sabrina Sacconi has nothing to disclose. John Vissing has been a Consultant on advisory boards for Amicus Therapeutics, Biogen, Edgewise Therapeutics, Fulcrum Therapeutics, Genethon, Horizon Therapeutics (now Amgen), Lupin, ML Biopharma, Novartis, Regeneron Pharmaceuticals, Roche, Sanofi Genzyme (now Sanofi), Sarepta Therapeutics and UCB. He has received research and travel support and/or speaker honoraria from Alexion Pharmaceuticals, argenx, Biogen, Edgewise Therapeutics, Fulcrum Therapeutics, Lupin, Sanofi Genzyme (now Sanofi) and UCB. He is a Principal Investigator in clinical trials for Alexion Pharmaceuticals, argenx, Atamyo Therapeutics, Genethon, Horizon Therapeutics (now Amgen), Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), ML Biopharma, Novartis, Regeneron Pharmaceuticals, Roche, Sanofi Genzyme (now Sanofi) and UCB. Tuan Vu is the USF Site Principal Investigator for MG clinical trials sponsored by Alexion/AstraZeneca Rare Disease, Amgen, argenx, Cartesian Therapeutics, COUR Pharmaceuticals, Dianthus Therapeutics, ImmunAbs, Immunovant, Johnson & Johnson, NMD Pharma, Regeneron Pharmaceuticals, UCB and Vor, and has served as a speaker for Alexion/AstraZeneca Rare Disease, Amgen, argenx and CSL Behring. He has performed consulting work for Alexion/AstraZeneca Rare Disease, argenx, Dianthus Therapeutics, NMD Pharma, Regeneron and Vor. Bethany George is an employee of Cogent Skills Limited working for UCB. Thaïs Tarancón is an employee and shareholder of UCB.

References: 1. RYSTIGGO® US PI. https://www.accessdata.fda.gov/drugsatfda_docs/label/2025/761286s007lbl.pdf. Accessed September 2026. 2. Smith, B, et al. MAbs. 2018; 10(7):1111–1130. 3. Bril V, et al. Lancet Neurol. 2023;22(5):383–394.

Please use this QR code to download a PDF of the poster.

These data were previously presented at ICNMD 2026, Florence, Italy; July 7–11, 2026