Poster **95**

Rozanolixizumab for ocular myasthenia gravis: Study design of a Phase 3, randomized, double-blind, placebo-controlled trial (MyVision) and open-label extension study (MyVisionXT)

Suraj Muley[1], Ibironke Addy[2], Jos Bloemers[3], Sarah Cluck[4], Denis Flemm[2], Yamina Merazga[5], Sebastian Zamorano Villablanca[6], Mark Kupersmith[7], Pushpa Narayanaswami[8]

[1]John Shufeldt School of Medicine and Medical Engineering, Arizona State University, Phoenix, AZ, USA; [2]UCB, Monheim, Germany; [3]UCB, Breda, The Netherlands; [4]UCB, Slough, UK; [5]UCB, New York, NY, USA; [6]UCB, Bulle, Switzerland; [7]Department of Ophthalmology, Icahn School of Medicine at Mount Sinai, New York Eye and Ear Infirmary and Union Square Eye Care, New York, NY, USA; [8]Department of Neurology, Beth Israel Deaconess Medical Center/Harvard Medical School, Boston, MA, USA

MGFA Scientific Session at AANEM 2026, Orlando, FL, USA; September 29, 2026

Introduction

- oMG is a clinical subtype of MG that affects only the ocular muscles, causing ptosis and/or diplopia[1]
 - Such ocular symptoms can impede daily activities and significantly reduce QoL[2]
- Rozanolixizumab, a humanized IgG4 mAb that targets and blocks FcRn with high affinity, is approved for the treatment of adults with anti-AChR Ab+ or anti-MuSK Ab+ gMG[3,4]
- In the Phase 3 MycarinG study (NCT03971422), patients with gMG receiving one 6-week cycle of rozanolixizumab (7 mg/kg or 10 mg/kg) demonstrated greater improvements in ocular symptoms across MG-specific outcomes than with placebo[5]
- These improvements were maintained over repeated 6-week cycles in the OLE studies MG0004 (NCT04124965) and MG0007 (NCT04650854)[6]
- **Here, we describe the study designs of the Phase 3 MyVision study (MG0038/NCT07463521) and its OLE, MyVisionXT (MG0039/NCT07465289), which will assess rozanolixizumab treatment in patients with oMG**

MyVision study design

MyVision is a Phase 3, multicenter, randomized, parallel-group, placebo-controlled, double-blind study to evaluate the efficacy and safety of one 6-week cycle of rozanolixizumab in adults with anti-AChR Ab+, anti-MuSK Ab+ or seronegative oMG

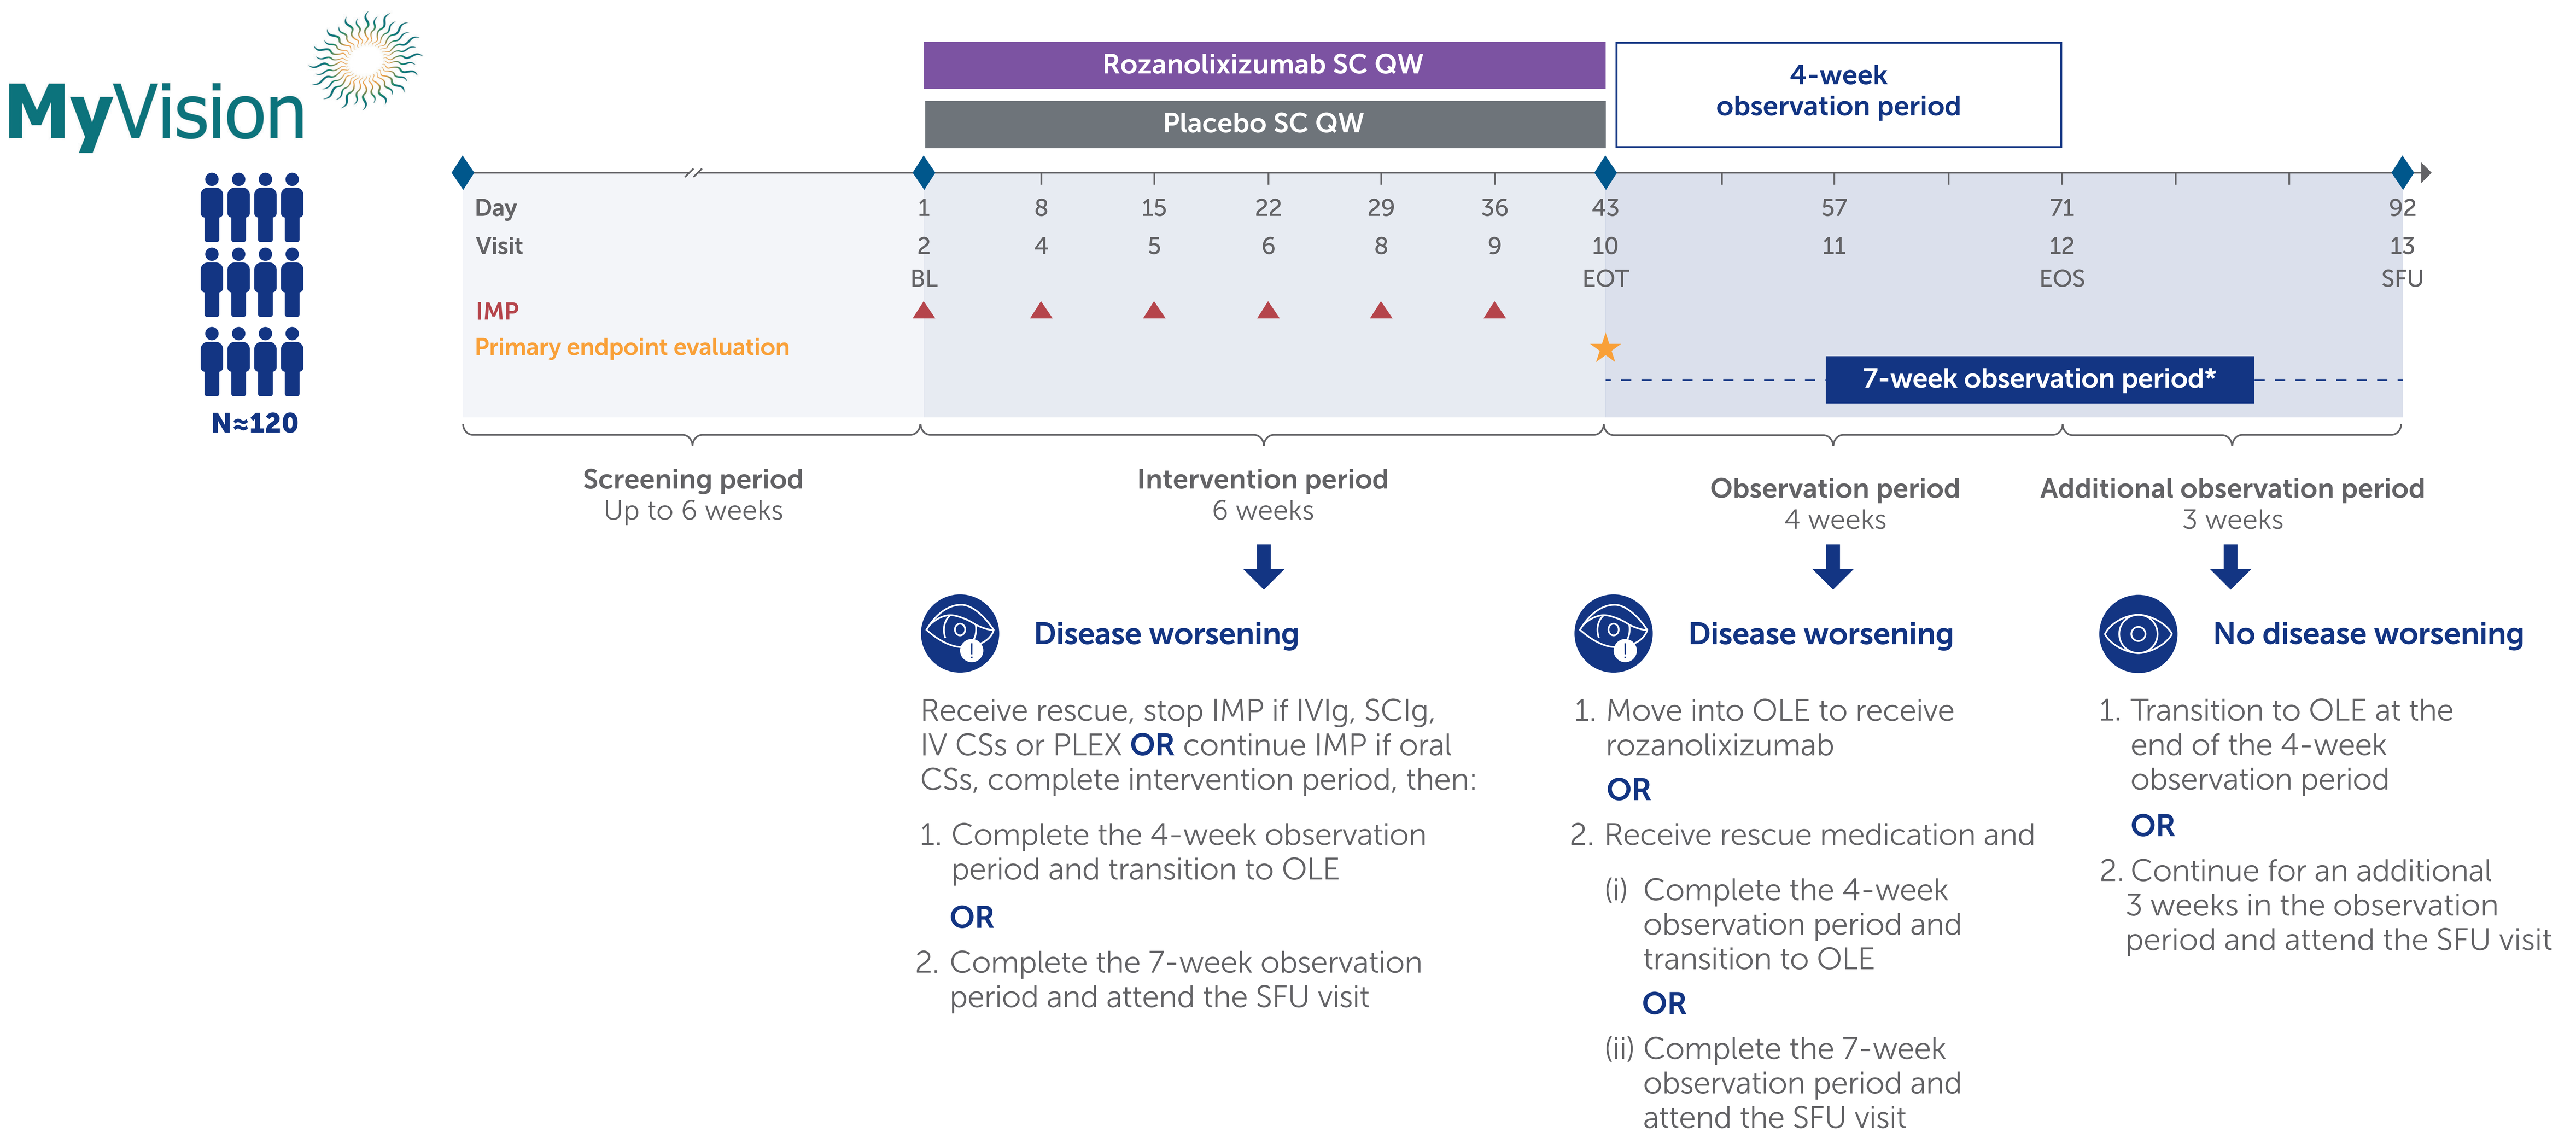
MyVision
N≈120
Rozanolixizumab SC QW
Placebo SC QW
4-week observation period
Day 1 8 15 22 29 36 43 57 71 92
Visit 2 4 5 6 8 9 10 11 12 13
BL EOT EOS SFU
IMP
Primary endpoint evaluation
7-week observation period*
Screening period
Up to 6 weeks
Intervention period
6 weeks
Observation period
4 weeks
Additional observation period
3 weeks
Disease worsening
Receive rescue, stop IMP if IVIg, SCIg, IV CSs or PLEX OR continue IMP if oral CSs, complete intervention period, then:
1. Complete the 4-week observation period and transition to OLE
OR
2. Complete the 7-week observation period and attend the SFU visit
Disease worsening
1. Move into OLE to receive rozanolixizumab
OR
2. Receive rescue medication and
(i) Complete the 4-week observation period and transition to OLE
OR
(ii) Complete the 7-week observation period and attend the SFU visit
No disease worsening
1. Transition to OLE at the end of the 4-week observation period
OR
2. Continue for an additional 3 weeks in the observation period and attend the SFU visit

Inclusion criteria
- Aged ≥18 years
- MGFA Clinical Class I MG with anti-AChR Ab+, anti-MuSK Ab or seronegative status
- MGII ocular score (PRO part) ≥6 (with ≥2 ocular items with a score ≥2 at screening and baseline)
- Ocular onset <3 years before screening, or ≥3 years provided they have demonstrated response to treatment† in the past year

Exclusion criteria
- Any clinically significant medical or psychiatric condition, active serious infection or history of serious infection, recent or planned major surgery and/or significant laboratory abnormality
- Other relevant ocular, autoimmune, neurological diseases or active neoplastic disease
- Previous treatment with FcRn inhibitors or prohibited medications‡
- Live vaccination within 4 weeks prior to baseline visit, or planned during the study or within 8 weeks after the final study dose

Primary endpoint:
- CFB in MGII ocular score (PRO part) at Day 43

Secondary endpoints:
- CFB to Day 43 in:
 - MGSPRO Ocular Muscle Weakness score
 - MG-ADL ocular subdomain score
 - MGII ocular score (physical examination part)
 - MG-QoL15r total score

Secondary safety endpoint:
- Incidence of TEAEs

Visit 3 and Visit 7 are PK assessments. *Study participants who will not transition to the OLE study must complete a 7-week observation period from Day 43 and conclude at the SFU Visit (Day 92), corresponding to 8 weeks following the final dose on Day 36 of study intervention. †IVIg, PLEX, SCIg, pyridostigmine, and/or CSs. ‡Prohibited medications include immunosuppressants (pimecrolimus within 4 weeks or vinca alkaloids within 12 weeks of screening), C5 inhibitors (eculizumab, ravulizumab or zilucoplan) within 6 weeks of screening and B-cell depleters (including rituximab and inebilizumab) within 6 months of screening.

MyVisionXT study design

MyVisionXT is an OLE study to evaluate the long-term safety, tolerability and efficacy of repeated symptom-driven treatment cycles of rozanolixizumab in eligible patients from MyVision

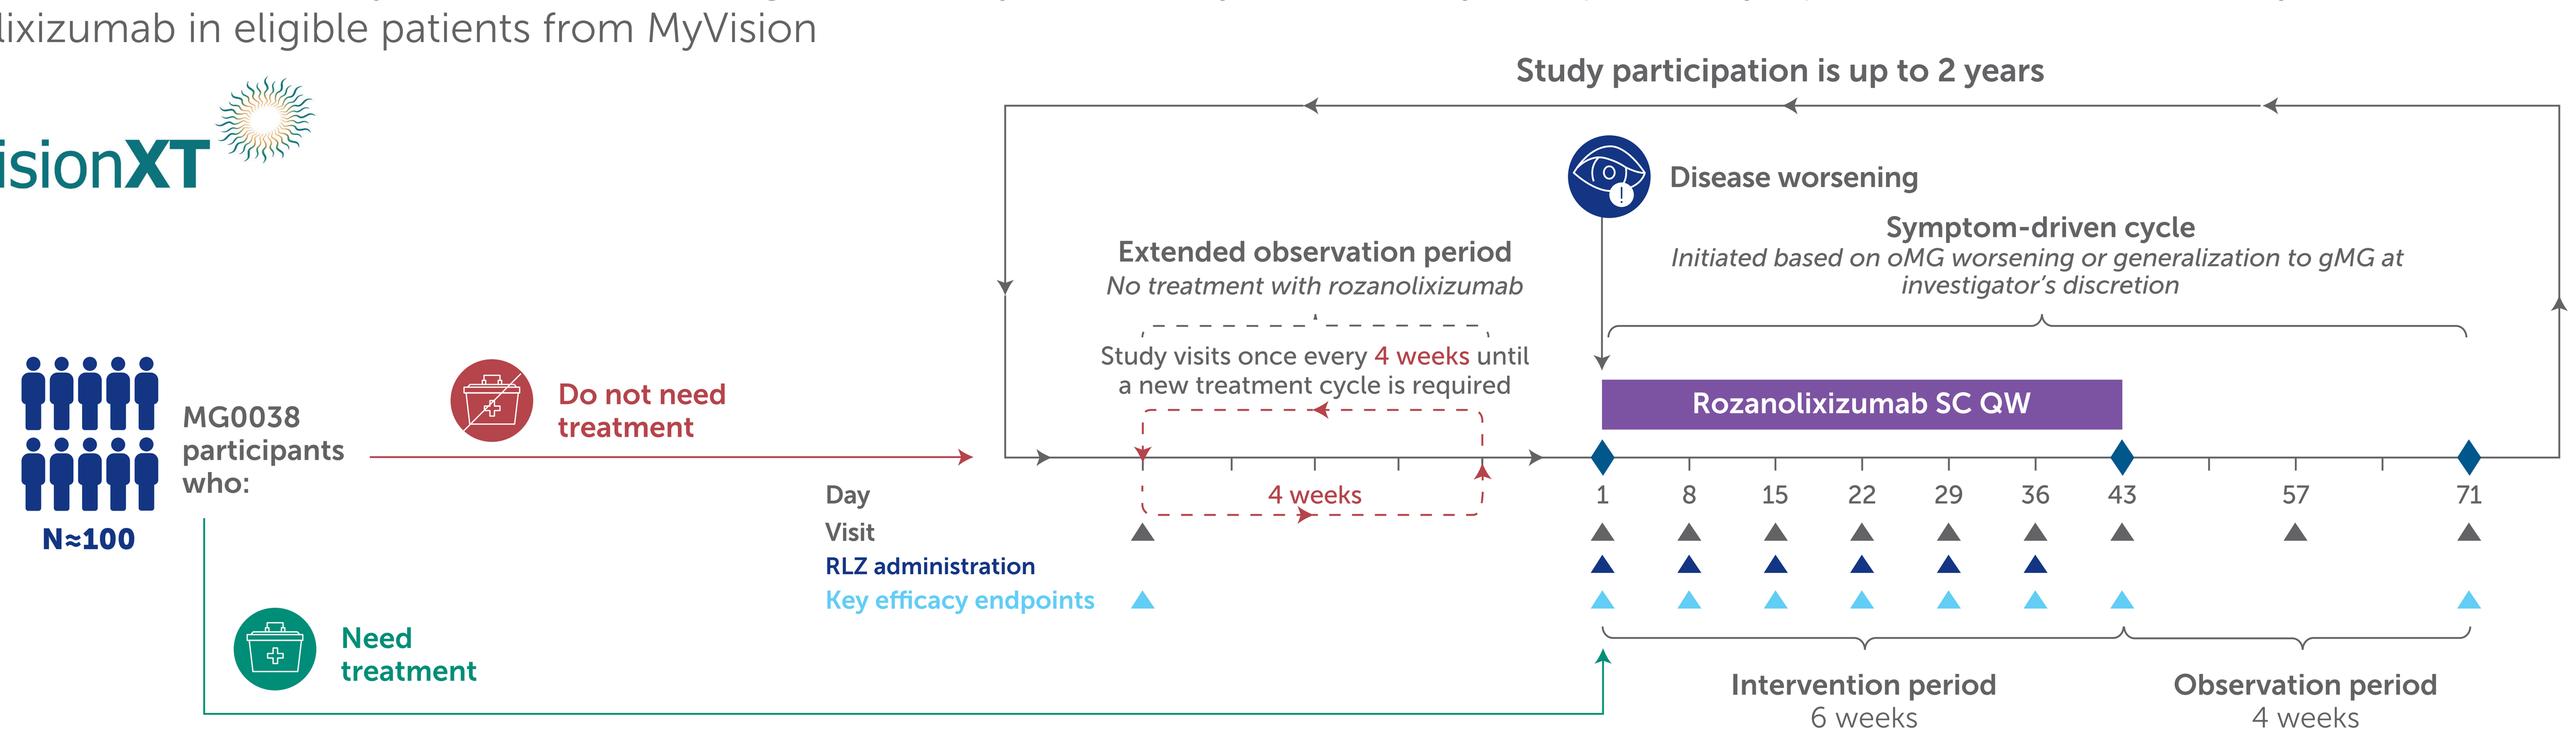
MyVisionXT
N≈100
MG0038 participants who:
Do not need treatment
Need treatment
Study participation is up to 2 years
Extended observation period
No treatment with rozanolixizumab
Study visits once every 4 weeks until a new treatment cycle is required
4 weeks
Disease worsening
Symptom-driven cycle
Initiated based on oMG worsening or generalization to gMG at investigator's discretion
Rozanolixizumab SC QW
Day 1 8 15 22 29 36 43 57 71
Visit
RLZ administration
Key efficacy endpoints
Intervention period
6 weeks
Observation period
4 weeks

Inclusion criteria
- Received ≥1 IMP dose in MyVision and opted to receive rozanolixizumab in MyVisionXT

Primary endpoint:
- Incidence of TEAEs

Secondary endpoints:
- For the first three symptom-driven cycles, CFB to Day 43 in:
 - MGII ocular score (PRO part)
 - MGSPRO Ocular Muscle Weakness score
 - MG-ADL ocular subdomain score
 - MG-QoL15r total score
- Study patient who generalizes to MGFA Class II or above over time
- CFB in MGII total and generalized score over time
- CFB in MG-ADL total score over time

Symptom-driven cycles (defined as cycles initiated based on oMG worsening) are initiated at the investigator's discretion either immediately or following symptom worsening during the extended observation period. For any patient who receives IVIg, SCIg or PLEX rescue therapy during the intervention period or the 4-week observation period in MyVision, a ≥2-week interval must occur before the initiation of a symptom-driven rozanolixizumab cycle in MyVisionXT. The dosing strategies for MyVision and MyVisionXT align with the approved regional labels for rozanolixizumab in gMG.

Summary and conclusions

The clinical course of oMG carries a significant patient burden, characterized by impaired quality of life and daily living due to ocular symptoms, diagnostic challenges and suboptimal clinical outcomes

Rozanolixizumab has demonstrated efficacy in MG-specific ocular outcomes in *post hoc* analyses of patients with gMG

MyVision is an ongoing, Phase 3, randomized, double-blind, placebo-controlled study evaluating the efficacy and safety of rozanolixizumab in adults with oMG (currently recruiting)

MyVisionXT, an open-label extension (OLE) study, will assess the long-term safety, tolerability and efficacy of repeated, symptom-driven rozanolixizumab treatment cycles

Together, these studies will provide critical data on the short- and long-term therapeutic potential of rozanolixizumab for patients with oMG

Abbreviations: Ab+, antibody positive; AChR, acetylcholine receptor; BL, baseline; C5, component 5; CFB, change from baseline; CS, corticosteroid; EOS, end of study; EOT, end of treatment; FcRn, neonatal fragment crystallizable receptor; (g)MG, (generalized) myasthenia gravis; IgG4, immunoglobulin G subunit 4; IMP, investigational medicinal product; IV, intravenous; IVIg, intravenous immunoglobulin; mAb, monoclonal antibody; MG-ADL, Myasthenia Gravis Activities of Daily Living; MGFA, Myasthenia Gravis Foundation of America; MGII, Myasthenia Gravis Impairment Index; MGSPRO, Myasthenia Gravis Symptoms Patient-Reported Outcome; MG-QoL15r, Myasthenia Gravis Quality of Life 15-item revised; MuSK, muscle-specific tyrosine kinase; OLE, open-label extension; oMG, ocular myasthenia gravis; PK, pharmacokinetics; PLEX, plasma exchange; PRO, patient-reported outcome; QoL, quality of life; QW, once weekly; RLZ, rozanolixizumab; SC, subcutaneous; SCIg, subcutaneous immunoglobulin; SFU, safety follow-up; TEAE, treatment-emergent adverse event.

Acknowledgments: This study was funded by UCB. The authors acknowledge Qudsia Ehsan, BSc, of Ogilvy Health, London, UK, for editorial assistance, which was funded by UCB. The authors thank Veronica Porkess, PhD, CMPP, of UCB for publication and editorial support. The authors thank the patients and their caregivers, in addition to the investigators and their teams who contributed to this study.

Author disclosures: Suraj Muley has received grants from argenx, Dianthus, Immunovant, Novartis, Regeneron, Sanofi, Takeda and UCB. Dr Muley has also received royalties or licenses from GBS and UpToDate CIDP, and has served on a speaker bureau for Alexion, Amgen, CSL Behring, Grifols, Johnson & Johnson, Takeda and UCB. Dr Muley has received honoraria for consulting services from Alexion, Amgen, Catalyst, CSL Behring, Grifols, Immunovant, Johnson & Johnson, Takeda, UCB and Vor. Ibironke Addy, Jos Bloemers, Sarah Cluck, Denis Flemm, Yamina Merazga and Sebastian Zamorano are employees and shareholders of UCB. Mark Kupersmith is a paid Consultant for Dandelion, Dompe, Life Biosciences, Oculus, Serenity and UCB. Pushpa Narayanaswami has received grant support from the Agency for Healthcare Research and Quality (AHRQ), Alexion Pharmaceuticals, argenx, Cabaletta Bio, Dianthus, National Institutes of Health (NIH) and Patient-Centered Outcomes Research Institute (PCORI), and served on advisory boards for Alexion Pharmaceuticals, Amgen, argenx, Cabaletta Bio, Cartesian, Dianthus Therapeutics, ImmunAbs, Immunovant, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Merck Serono, Sarepta, UCB, Viridian Therapeutics and Vor Biopharma; she was a member of the Data and Safety Monitoring Board for Sanofi and is a member of the Data and Safety Monitoring Board for argenx and NMD Pharma. She has received royalties from Springer Nature.

References: 1. O'Hare M, Doughty C. Semin Neurol. 2019;39(6):749–760. 2. Meisel A, et al. Eur J Neurol. 2024;31(7):e16280. 3. RYSTIGGO® US PI. https://www.accessdata.fda.gov/drugsatfda_docs/label/2025/761286s007lbl.pdf. Accessed August 2026. 4. Smith B, et al. MAbs. 2018;10(7):1111–1130. 5. Mahuwala ZK, et al. Neurology. 2025;104(7 Suppl 1):1866. 6. Cortés Vicente E, et al. J Neuroumusc Dis. 2026;13(2_suppl): PP01.186.

Please use this QR code to download a PDF of the poster.