AANEM Poster **268** | MGFA Scientific Session Poster **89**

Individual item-level analyses of myasthenia gravis-specific outcomes during zilucoplan treatment in RAISE and RAISE-XT

Joseph Post[1], Miriam Freimer[2], Channa Hewamadduma[3,4], M. Isabel Leite[5], Angelina Maniaol[6], Kimiaki Utsugisawa[7], Michael D. Weiss[8], Jos Bloemers[9], Babak Boroojerdi[10], Paul Mahoney[11], Natasa Savic[12], James F. Howard Jr.[13] on behalf of the RAISE and RAISE-XT study teams

[1]UCB, Charlotte, NC, USA; [2]Department of Neurology, The Ohio State University Wexner Medical Center, Columbus, OH, USA; [3]NIHR BRC Neuroscience Centre, Academic Neuromuscular Unit, Sheffield Teaching Hospitals NHS Foundation Trust, Sheffield, UK; [4]Sheffield Institute for Translational Neuroscience (SITraN), School of Medicine and Population Health, University of Sheffield, Sheffield, UK; [5]Nuffield Department of Clinical Neurosciences, University of Oxford, Oxford, UK; [6]Department of Neurology, Oslo University Hospital, Oslo, Norway; [7]Department of Neurology, Hanamaki General Hospital, Hanamaki, Japan; [8]Department of Neurology, University of Washington Medical Center, Seattle, WA, USA; [9]UCB, Breda, The Netherlands; [10]UCB, Monheim, Germany; [11]UCB, Slough, UK; [12]UCB, Bulle, Switzerland; [13]Department of Neurology, The University of North Carolina at Chapel Hill, Chapel Hill, NC, USA

AANEM & MGFA Scientific Session 2026, Orlando, FL, USA; September 29–October 2, 2026

Introduction

- Patients with gMG experience fluctuating, fatigable muscle weakness that may affect ocular, bulbar, respiratory and proximal muscles, leading to a diverse range of symptoms that can impact health-related quality of life[1,2]
- Zilucoplan is a small (15-amino acid) potent macrocyclic peptide complement C5 inhibitor approved for the treatment of adults with anti-AChR Ab+ gMG[3,4]
- In the Phase 3 RAISE study (NCT04115293), zilucoplan showed statistically significant and clinically meaningful improvements in MG-specific outcomes in patients with anti-AChR Ab+ gMG[4]
 - These improvements were sustained over 120 weeks in the Phase 3 OLE study, RAISE-XT (NCT04225871)[5]
- **This *post hoc* analysis assessed change in individual item scores for MG-ADL, QMG and MG-QoL15r**

Methods

- In RAISE, adults with MGFA Clinical Class II–IV anti-AChR Ab+ gMG were randomized to self-administer once-daily subcutaneous zilucoplan 0.3 mg/kg or placebo[4]
- Adults who completed RAISE could enter RAISE-XT to receive once-daily zilucoplan 0.3 mg/kg[5]
 - The primary outcome of RAISE-XT was incidence of TEAEs
- Analyses of MG-ADL, QMG and MG-QoL15r from the RAISE and RAISE-XT (interim data cutoff: November 11, 2023) studies included:
 - The proportion of patients showing no symptoms, improvement, and no change from baseline to Week 12 (RAISE), and from double-blind baseline to Week 120 (RAISE-XT)
 - Change from double-blind baseline (CFB) to Week 120

Results

- Overall, 200 patients enrolled in RAISE-XT; 183 received either zilucoplan 0.3 mg/kg or placebo in the double-blind studies[5]
- In RAISE, treatment with zilucoplan resulted in a greater proportion of patients showing improvement or no symptoms versus placebo at Week 12 across all MG-ADL and QMG items assessed (**Table 1**)
 - Similar results were observed for MG-QoL15r
- In RAISE-XT, the proportion of patients demonstrating improvements or no symptoms further increased and was sustained through to Week 120
- The items with the highest proportion of patients showing improvement or no symptoms at Week 120 were (**Figure 1**):
 - MG-ADL items: chewing and talking (both 93.2%); swallowing and brush hair/teeth (both 87.7%)
 - QMG items: swallowing 4 oz. water (91.4%), facial muscles (85.7%) and speech following counting from 1 to 50 (94.3%)
 - MG-QoL15r items: Trouble grooming and loss of personal independence (both 76.5%); trouble eating (88.2%) and difficulty speaking (86.8%)
- TEAEs occurred in 97.0% (n=194/200, RAISE-XT) of patients
 - The two most frequently occurring TEAEs were COVID-19 (35.5% [71/200]) and MG worsening (29.5% [59/200])

Summary and conclusions

This *post hoc* analysis assessed change in item scores for MG-ADL, QMG and MG-QoL15r using data from RAISE and RAISE-XT

Treatment with zilucoplan led to improvements or no symptoms in a greater proportion of patients across individual items in MG-specific outcomes at Week 12 versus placebo

During the extension study, the proportion of patients demonstrating these improvements or no symptoms further increased and was sustained through to Week 120

These data show that long-term treatment with zilucoplan may provide benefit across a broad range of signs and symptoms of gMG

Table 1 Across all MG-ADL and QMG items assessed, zilucoplan treatment resulted in a greater proportion of patients showing improvement or no symptoms versus placebo at Week 12 (RAISE)

Percentage of patients showing improvement or no symptoms

0 to <20% | 20 to <40% | 40 to <60% | 60 to <80% | 80 to 100%

(a) MG-ADL

MG-ADL item, % (n/N)	Placebo N=88	Zilucoplan 0.3 mg/kg N=86
Chewing	**49.4** (42/85)	**71.4** (60/84)
Swallowing	**55.3** (47/85)	**71.4** (60/84)
Talking	**57.6** (49/85)	**73.8** (62/84)
Breathing	**38.8** (33/85)	**52.4** (44/84)
Double vision	**50.6** (43/85)	**66.7** (56/84)
Eyelid droop	**48.2** (41/85)	**65.5** (55/84)
Brush hair/teeth	**52.9** (45/85)	**58.3** (49/84)
Rise from chair	**43.5** (37/85)	**63.1** (53/84)

(b) QMG

QMG item, % (n/N)	Placebo N=88	Zilucoplan 0.3 mg/kg N=86
Double vision	**35.7** (30/84)	**51.8** (43/83)
Ptosis	**50.0** (42/84)	**65.1** (54/83)
Facial muscles	**58.3** (49/84)	**66.3** (55/83)
Swallowing 4 oz. water	**76.2** (64/84)	**83.1** (69/83)
Speech following counting	**72.6** (61/84)	**79.5** (66/83)
Forced vital capacity	**63.1** (53/84)	**74.7** (62/83)
R arm outstretched	**26.2** (22/84)	**43.4** (36/83)
L arm outstretched	**25.0** (21/84)	**41.0** (34/83)
R hand grip (male)	**19.0** (8/42)	**27.3** (9/33)
R hand grip (female)	**19.0** (8/42)	**26.0** (13/50)
L hand grip (male)	**40.5** (17/42)	**54.5** (18/33)
L hand grip (female)	**31.0** (13/42)	**46.0** (23/50)
R leg outstretched	**22.6** (19/84)	**45.8** (38/83)
L leg outstretched	**22.6** (19/84)	**47.0** (39/83)
Head lifted	**34.5** (29/84)	**54.2** (45/83)

Figure 1 Most patients had improved MG-ADL, QMG and MG-QoL15r item scores at Week 120 (RAISE-XT)

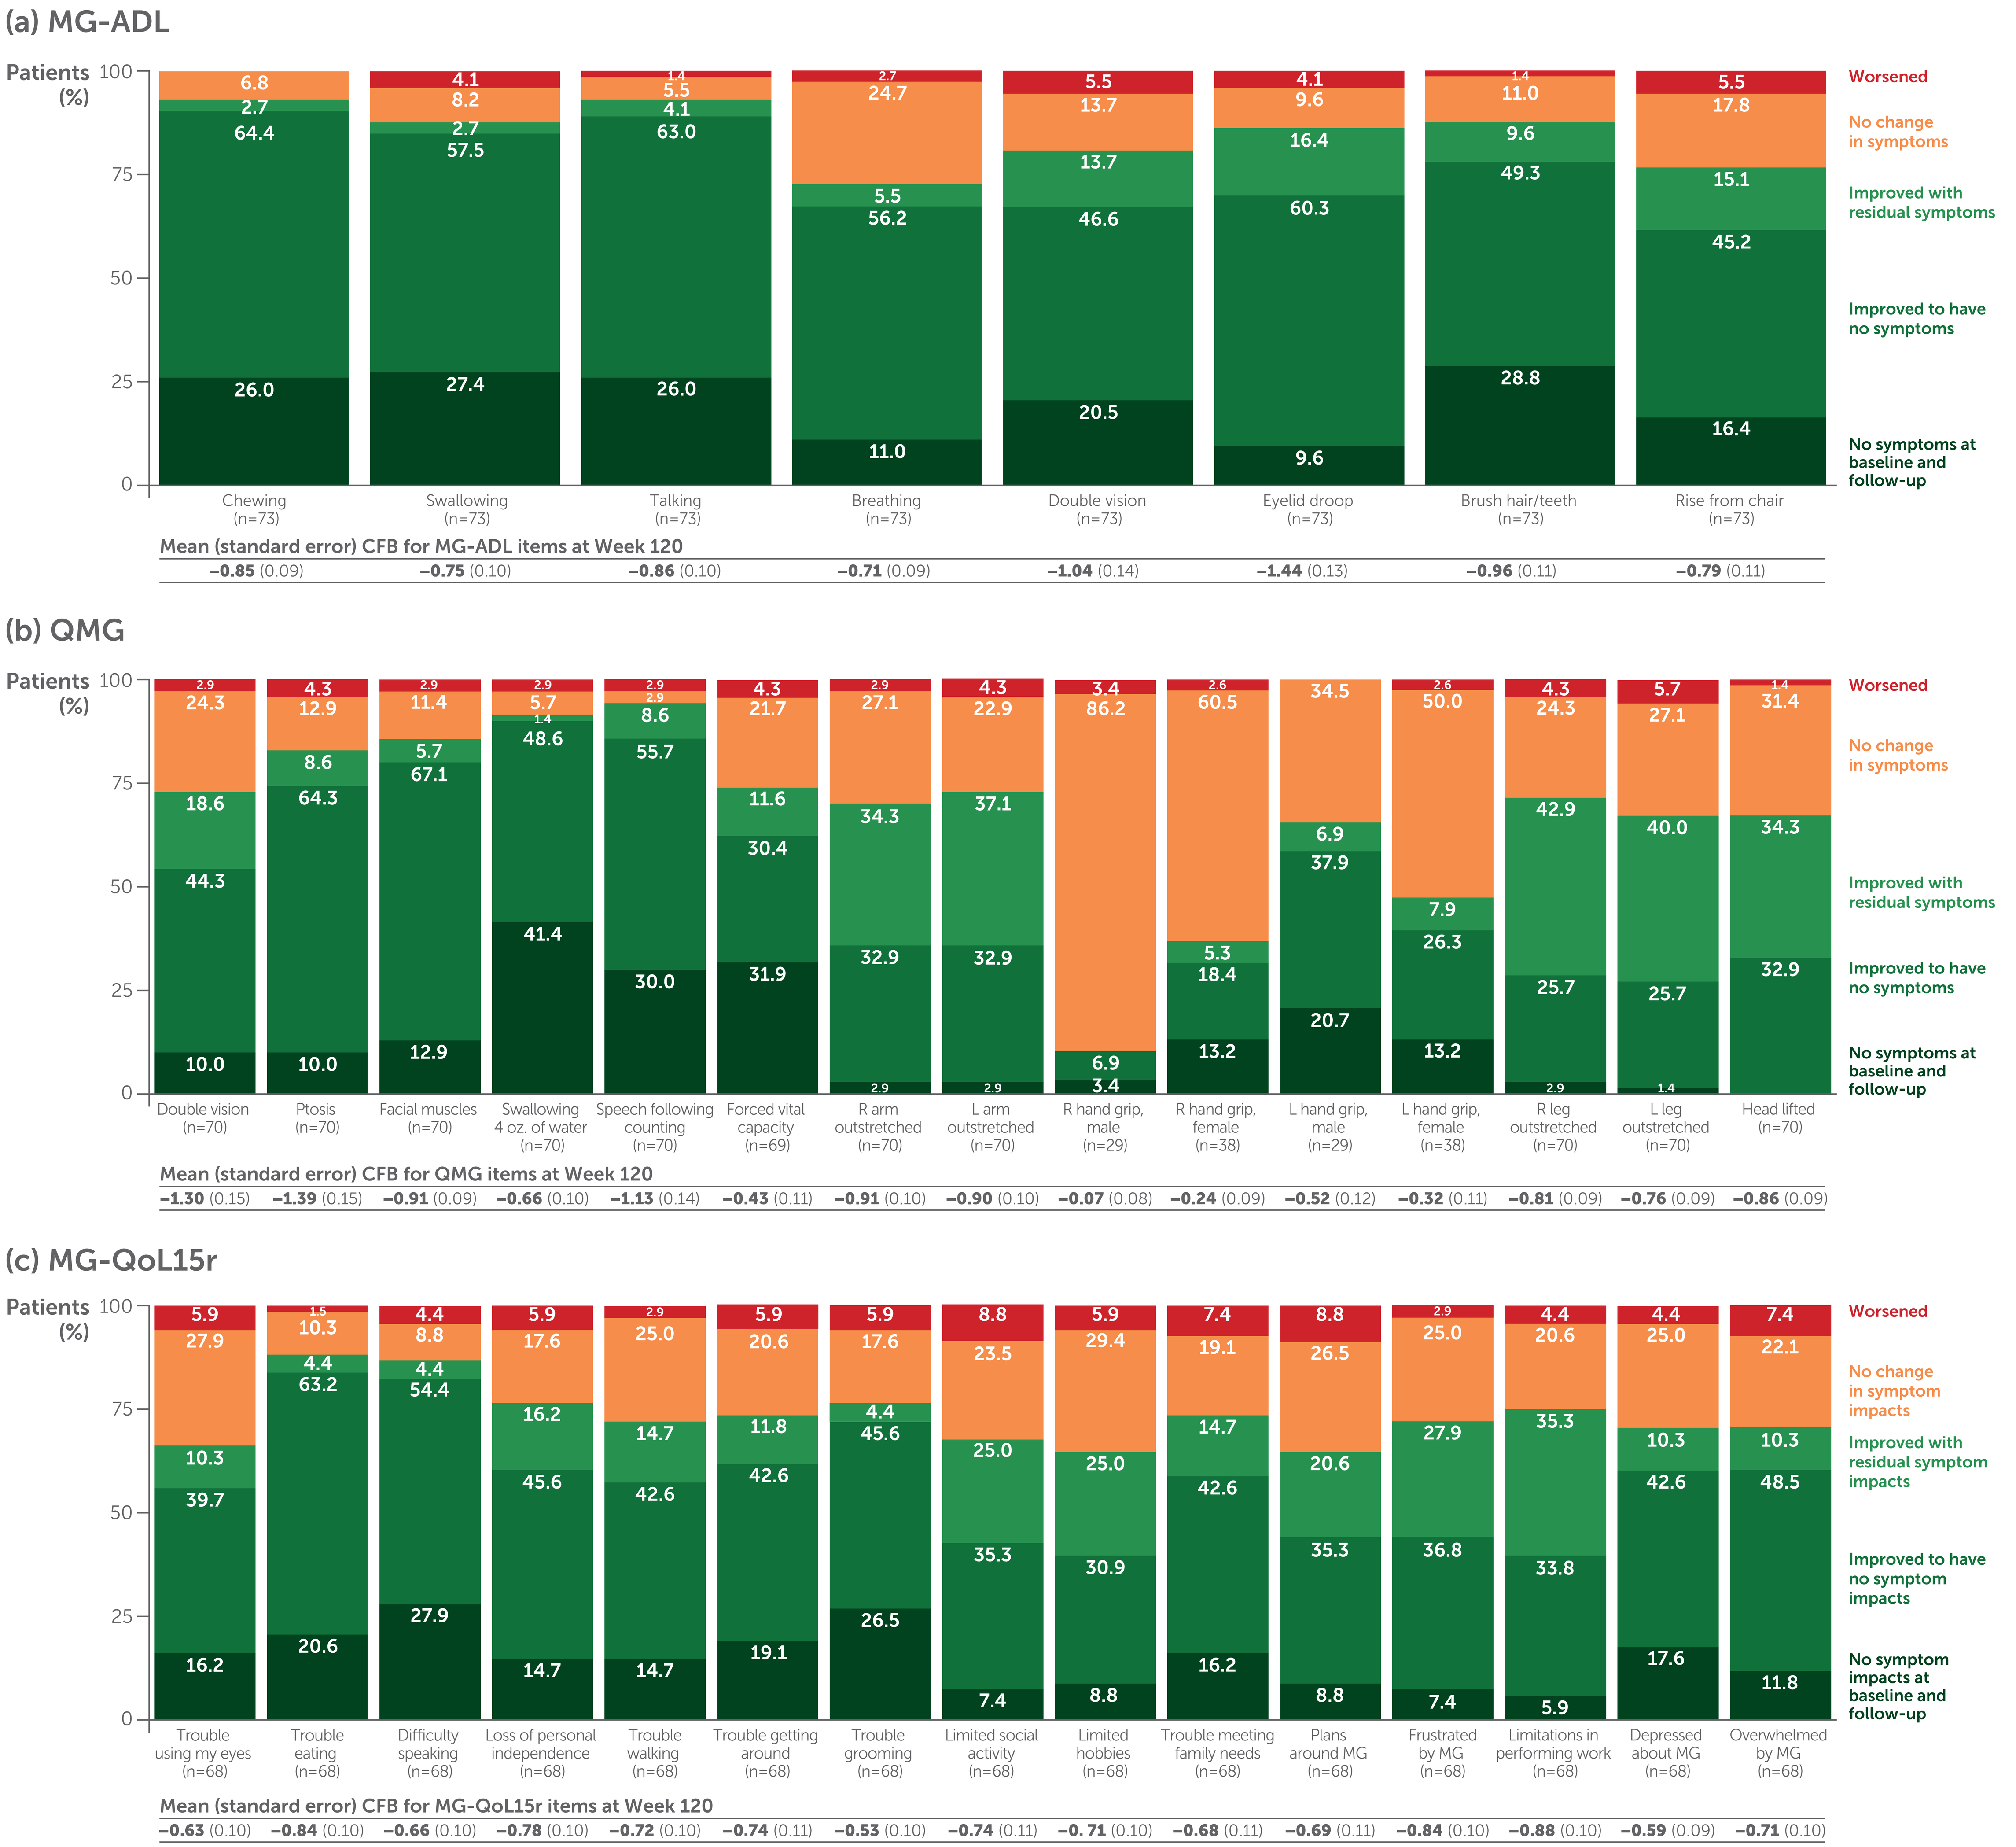
(a) MG-ADL
Patients (%)
Chewing (n=73)
Swallowing (n=73)
Talking (n=73)
Breathing (n=73)
Double vision (n=73)
Eyelid droop (n=73)
Brush hair/teeth (n=73)
Rise from chair (n=73)
Worsened
No change in symptoms
Improved with residual symptoms
Improved to have no symptoms
No symptoms at baseline and follow-up
Mean (standard error) CFB for MG-ADL items at Week 120
−0.85 (0.09) −0.75 (0.10) −0.86 (0.10) −0.71 (0.09) −1.04 (0.14) −1.44 (0.13) −0.96 (0.11) −0.79 (0.11)
(b) QMG
Patients (%)
Double vision (n=70)
Ptosis (n=70)
Facial muscles (n=70)
Swallowing 4 oz. of water (n=70)
Speech following counting (n=70)
Forced vital capacity (n=69)
R arm outstretched (n=70)
L arm outstretched (n=70)
R hand grip, male (n=29)
R hand grip, female (n=38)
L hand grip, male (n=29)
L hand grip, female (n=38)
R leg outstretched (n=70)
L leg outstretched (n=70)
Head lifted (n=70)
Worsened
No change in symptoms
Improved with residual symptoms
Improved to have no symptoms
No symptoms at baseline and follow-up
Mean (standard error) CFB for QMG items at Week 120
−1.30 (0.15) −1.39 (0.15) −0.91 (0.09) −0.66 (0.10) −1.13 (0.14) −0.43 (0.11) −0.91 (0.10) −0.90 (0.10) −0.07 (0.08) −0.24 (0.09) −0.52 (0.12) −0.32 (0.11) −0.81 (0.09) −0.76 (0.09) −0.86 (0.09)
(c) MG-QoL15r
Patients (%)
Trouble using my eyes (n=68)
Trouble eating (n=68)
Difficulty speaking (n=68)
Loss of personal independence (n=68)
Trouble walking (n=68)
Trouble getting around (n=68)
Trouble grooming (n=68)
Limited social activity (n=68)
Limited hobbies (n=68)
Trouble meeting family needs (n=68)
Plans around MG (n=68)
Frustrated by MG (n=68)
Limitations in performing work (n=68)
Depressed about MG (n=68)
Overwhelmed by MG (n=68)
Worsened
No change in symptom impacts
Improved with residual symptom impacts
Improved to have no symptom impacts
No symptom impacts at baseline and follow-up
Mean (standard error) CFB for MG-QoL15r items at Week 120
−0.63 (0.10) −0.84 (0.10) −0.66 (0.10) −0.78 (0.10) −0.72 (0.10) −0.74 (0.11) −0.53 (0.10) −0.74 (0.11) −0. 71 (0.10) −0.68 (0.11) −0.69 (0.11) −0.84 (0.10) −0.88 (0.10) −0.59 (0.09) −0.71 (0.10)

No patients had worsened scores for chewing (MG-ADL) and L hand grip (male; QMG). MG-ADL and QMG item severity scores range from 0 to 3 and MG-QoL15r items range from 0 to 2. Higher scores correspond to more severe impairment. A decrease from baseline indicates improvement.

Abbreviations: anti-AChR Ab+, anti-acetylcholine receptor antibody-positive; C5, component 5; CFB, change from baseline; gMG, generalized myasthenia gravis; MG, myasthenia gravis; MG-ADL, Myasthenia Gravis Activities of Daily Living; MGFA, Myasthenia Gravis Foundation of America; MG-QoL15r, Myasthenia Gravis Quality of Life 15-item revised; OLE, open-label extension; QMG, Quantitative Myasthenia Gravis; TEAE, treatment-emergent adverse event.

Acknowledgments: This study was funded by UCB. The authors acknowledge Qudsia Ehsan, BSc, of Ogilvy Health, London, UK, for editorial assistance, which was funded by UCB. The authors thank Veronica Porkess, PhD, of UCB for publication and editorial support. The authors thank the patients and their caregivers, in addition to the investigators and their teams who contributed to this study.

Author disclosures: Joseph Post is an employee and shareholder of UCB. Miriam Freimer has served as a paid Consultant for Arcellx, argenx and UCB. She receives research support from Abcuro, Alnylam Pharmaceuticals, argenx, Avidity Biosciences, COUR Pharmaceuticals, Dianthus Therapeutics, Fulcrum Therapeutics, Johnson & Johnson Innovative Medicine, the NIH, RemeGen Biosciences and UCB. Channa Hewamadduma is Principal Investigator/NCI in clinical trials in various neuromuscular disorders sponsored by Alexion Pharmaceuticals, argenx, Avidity, Cartesian, Dyne, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Novartis, PepGen, Regeneron Pharmaceuticals and UCB. He has received funding for consultancy on scientific or educational advisory boards for argenx, Biogen, Lupin, Roche and UCB, and has received an investigator-led research grant from UCB. His study activities were supported by a Sheffield NIHR BRC UK centre grant. He is a trustee of the myasthenia gravis patient organisation Myaware. M. Isabel Leite is funded by the NHS (Myasthenia and Related Disorders Service and National Specialised Commissioning Group for Neuromyelitis Optica, UK) and by the University of Oxford, UK. She has been awarded research grants from UK associations for patients with myasthenia and with muscular disorders (Myaware and Muscular Dystrophy UK, respectively) and the University of Oxford. She has received speaker honoraria or travel grants from Biogen, the Guthy-Jackson Charitable Foundation, Novartis and UCB. She serves on scientific or educational advisory boards for argenx, Horizon Therapeutics (now Amgen) and UCB. Angelina Maniaol has received payment for travel, meeting attendance, consulting honoraria or advisory board participation from Alexion Pharmaceuticals, argenx, Biogen, Johnson & Johnson and UCB. Kimiaki Utsugisawa has served as a paid Consultant for argenx, Chugai Pharmaceutical, HanAll Biopharma, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Merck, Mitsubishi Tanabe Pharma, UCB and Viela Bio (now Amgen); he has received speaker honoraria from Alexion Pharmaceuticals, argenx, the Japan Blood Products Organization and UCB. Michael D. Weiss has received honoraria for serving on scientific advisory boards for Alexion Pharmaceuticals, Amgen, Amylyx Pharmaceuticals, argenx, Biogen, Immunovant, Mitsubishi Tanabe Pharma and Ra Pharmaceuticals (now UCB), consulting honoraria from CSL Behring and Cytokinetics, and speaker honoraria from Amgen, Soleo Health, and UCB. He has been a paid Medical Monitor for a NeuroNEXT study. Jos Bloemers, Babak Boroojerdi, Paul Mahoney and Natasa Savic are employees and shareholders of UCB. James F. Howard Jr. has received research funding (paid to his institution) from Ad Scientiam, Alexion/AstraZeneca Rare Disease, argenx, Cartesian Therapeutics, Centers for Disease Control and Prevention, Merck EMD Serono, MGFA, Muscular Dystrophy Association, NIH, NMD Pharma, UCB and Vor Biopharma; honoraria/consulting fees from AcademicCME, Alexion/AstraZeneca Rare Disease, Amgen, argenx, Biohaven Ltd, Cartesian Therapeutics, CheckRare CME, CorEvitas, H. Lundbeck A/S, Japan Tobacco Company, Kyverna Therapeutics, MedLive, Merck EMD Serono, NMD Pharma, Novartis, PeerView CME, Physicians' Education Resource (PER, MJH) CME, Regeneron Pharmaceuticals, Seismic Therapeutics, Toleranzia AB, UCB, Vertex Pharmaceuticals and Vor Biopharma; non-financial support from Alexion/AstraZeneca Rare Disease, argenx, Biohaven Ltd, Cartesian Therapeutics, Toleranzia AB, UCB and Vor Biopharma.

References: 1. Jackson K, et al. Neurol Ther. 2023;12(1):107–128. 2. Gilhus NE, et al. Nat Rev Dis Primers. 2019;5(1):30. 3. ZILBRYSQ® EU SmPC. https://www.ema.europa.eu/en/documents/product-information/zilbrysq-epar-product-information_en.pdf. Accessed August 2026. 4. Howard JF Jr., et al. Lancet Neurol. 2023;22(5):395–406. 5. Howard JF Jr., et al. Long-term safety and efficacy of zilucoplan in generalized myasthenia gravis: 120-week interim analysis of RAISE-XT [poster]. AANEM 2024. Poster 192.

Please use this QR code to download a PDF of the poster.

These data were previously presented at ICNMD 2026, Florence, Italy; July 7–11, 2026