Poster **322**

Living with ocular symptoms of myasthenia gravis and patient priorities for clinical trials

Helen Schönborn, PhD[1], Anna Kole[2]

[1]admedicum GmbH & Co KG, Cologne, Germany; [2]UCB Paris, France

AANEM 2026, Orlando, FL, USA; September 29–October 2, 2026

Summary

Ocular symptoms of MG impose a substantial burden
Participants reported wide-ranging effects on daily functioning, independence, safety, work, social participation and emotional wellbeing

Patient priorities can inform trial design and participation
Patients confirmed that the MyVision/MyVisionXT studies measure outcomes relevant to their experience and highlighted priorities including improved symptom control, steroid-sparing options, fewer relapses, prevention of progression to gMG and reduced participation burden

Accessible, patient-centered trial communication is essential
Findings informed MyVision/MyVisionXT patient-facing materials, including visually adapted content, clear site and contact information, logistical support and a dedicated point of contact

Patient organization engagement can support ethical participation
Patient organizations can share clear, accessible and neutral information to support informed discussions between people living with MG and their clinicians

Introduction

Ocular symptoms of MG, including diplopia and ptosis, can impose a substantial burden on daily functioning, independence and emotional wellbeing.[1,2]

Progression from oMG to gMG is an important clinical concern for people living with ocular symptoms.[3]

Understanding what matters to people living with oMG, or gMG with ocular symptoms, can help ensure that clinical research is more relevant, accessible and feasible for participants.[4–6]

However, there is often underrepresentation of patients' experiences and patient organizations' perspectives in clinical trial design and recruitment planning.[4–6]

Objective: To understand the lived experience, unmet needs and trial-related preferences of people living with oMG or gMG with ocular symptoms, and to inform patient-centered, compliant recruitment strategies for the Phase 3 MyVision (NCT07463521)/MyVisionXT (NCT07465289) trials.

Abbreviations: gMG, generalized myasthenia gravis; MG, myasthenia gravis; oMG, ocular myasthenia gravis.

Acknowledgments: This study was funded by UCB. The authors thank Veronica Porkess, of UCB, Slough, UK, for publication and editorial support and Michael Wilbur of MW Advocacy Solutions, Paris, France, for writing support.

Author disclosures: Helen Schönborn is employed by admedicum GmbH & Co KG, who have received consulting fees from UCB for research on patient experience of living with MG. Anna Kole is an employee of UCB.

References: 1. Nair AG, et al. Indian J Ophthalmol. 2014;62(10):985–991. 2. Behbehani R. Eye Brain. 2023;15:1–13. 3. Fang CE, et al. BMJ Open. 2022;12(9):e060259. 4. U.S. Food and Drug Administration. Patient-focused drug development: Methods to identify what is important to patients. https://www.fda.gov/regulatory-information/search-fda-guidance-documents/patient-focused-drug-development-methods-identify-what-important-patients. Accessed August 2026. 5. Morgan TL, et al. Res Involv Engagem. 2025;11:142. 6. Crocker JC, et al. BMJ. 2018;363:k4738. 7. Braun V, et al. Qual Res Psychol. 2006;3(2):77–101.

Please use this QR code to download a PDF of the poster.

Methods

This mixed-methods study included:

1. **An online questionnaire assessing symptom burden, diagnostic journey and expectations regarding trial participation.** (9 responses from MG/oMG patient experts representing recognized patient organizations from **9 countries across Europe, US and Asia**).
2. **A semi-structured focus group, including 10 MG/oMG patient experts, 9 of whom completed the online questionnaire, representing recognized patient organizations from 10 countries across Europe, US and Asia.**

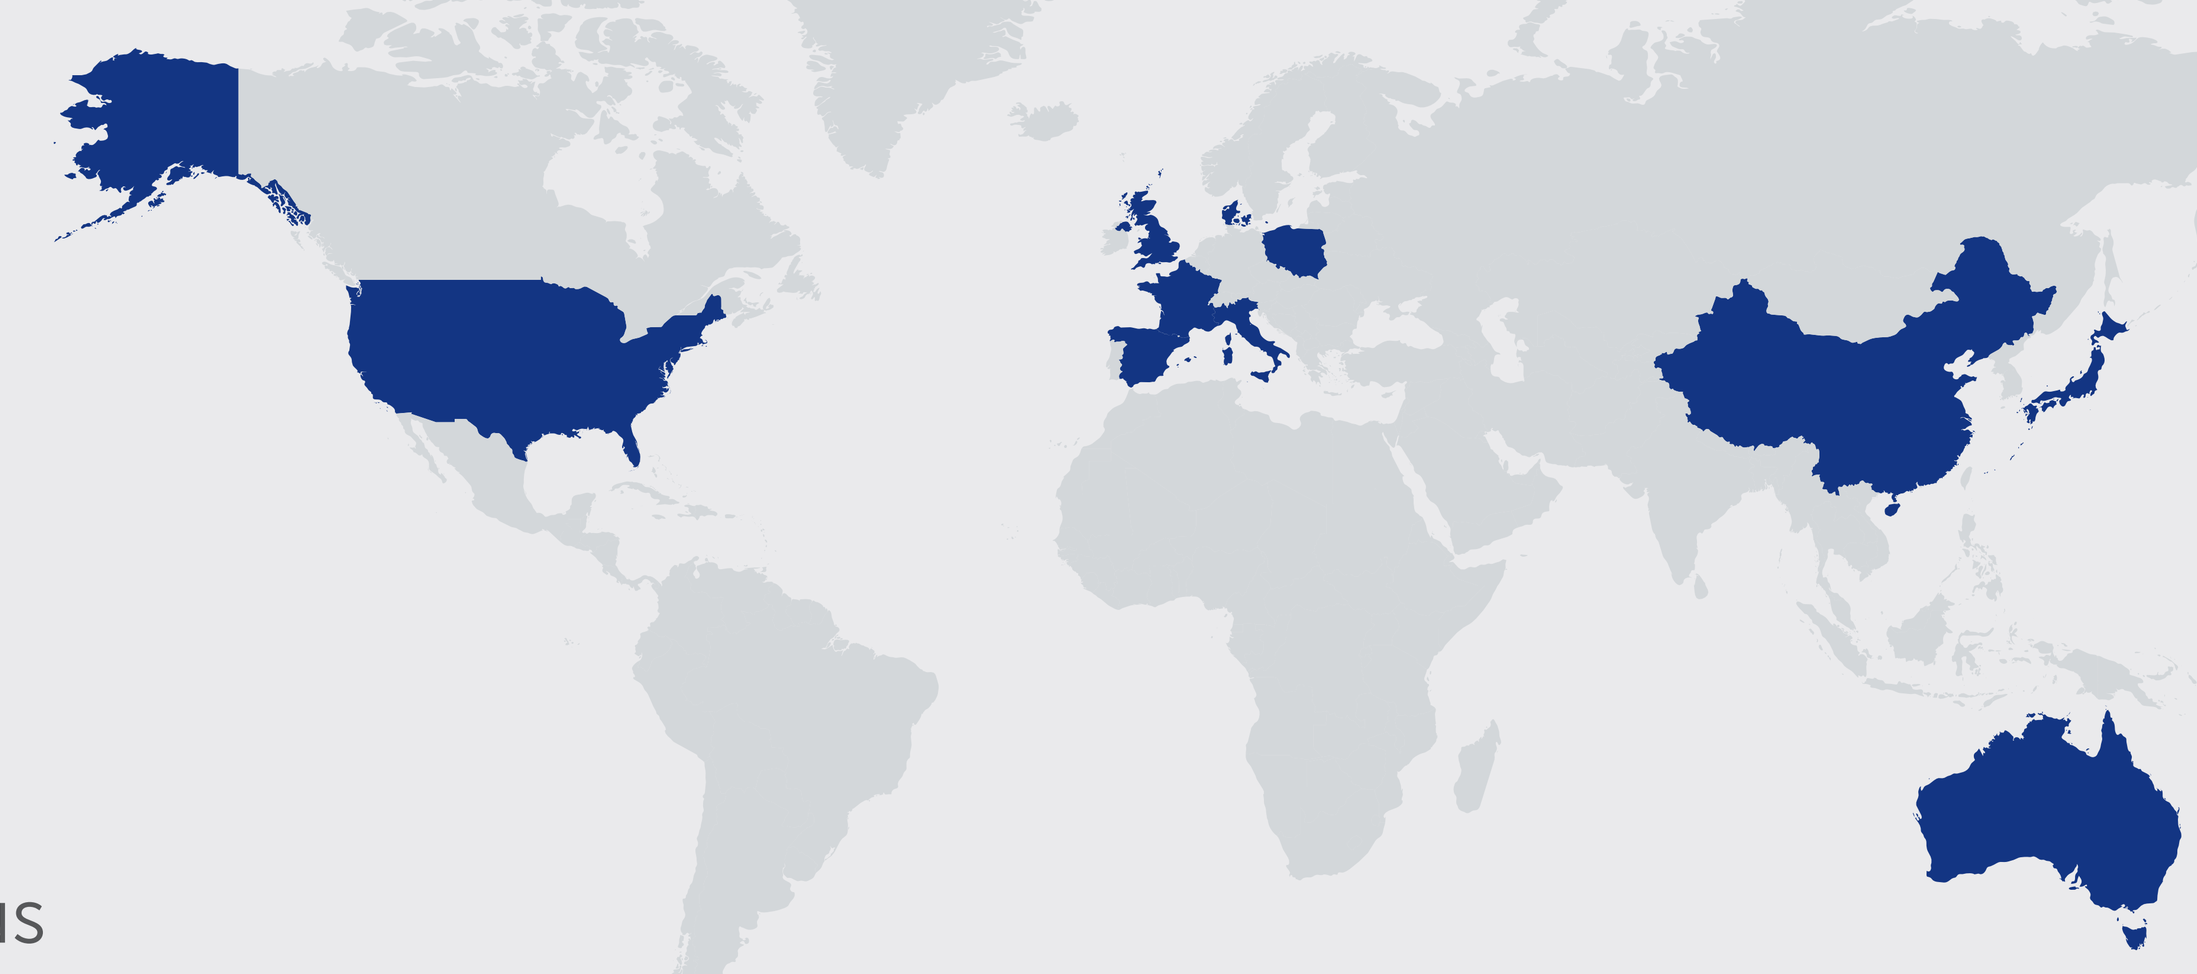

Questionnaire responses were analyzed descriptively and focus group insights thematically to identify key challenges, unmet needs and priorities relevant to clinical research design and recruitment.[7]

Results

During the **semi-structured focus groups**, people living with ocular symptoms of MG described a substantial and multifaceted burden affecting independence, safety, work, social participation and emotional wellbeing, highlighting the need for clinical trials and recruitment approaches that better reflect patient priorities.

1. Ocular symptoms affected daily functioning and independence

Participants described visual symptoms as disruptive to routine activities and personal autonomy.

Reported impacts included difficulties with:
- **Driving**
- **Reading and digital device use**
- **Work and productivity**
- **Household tasks, including cooking safely**
- **Childcare**
- **Moving safely around the home and community**

These limitations contributed to **reduced independence and increased reliance on others.**

2. The burden extended beyond physical symptoms

Participants described emotional and social consequences associated with ocular symptoms.

Key impacts included:
- **Social isolation**
- **Loss of confidence**
- **Frustration linked to fluctuating symptoms**
- **Anxiety around safety and disease progression**
- **Reduced ability to participate fully in work, family and social life**

These findings suggest that **the burden of ocular MG may not be fully captured** by clinical assessment alone.

3. Access to specialized and multidisciplinary care remained uneven

Participants reported challenges accessing appropriate care, particularly in rural settings.

Unmet needs included:
- **More consistent access to specialized MG expertise**
- **Better multidisciplinary support**
- **Greater understanding of ocular symptoms among healthcare professionals**
- **Clearer information and support following diagnosis**

Limited access to care may **exacerbate the functional and emotional burden** of ocular symptoms.

4. Participants expressed clear treatment priorities

Participants strongly favored treatment approaches that could reduce long-term treatment burden and improve day-to-day functioning.

Priority outcomes included:
- **Improved control of ocular symptoms**
- **Steroid-sparing treatment options**
- **Reduced fluctuation in symptoms**
- **Prevention of progression from oMG to gMG**
- **Improved ability to work, drive and manage daily activities**

These priorities highlight the **importance of aligning clinical research outcomes with what matters most to patients.**[5–7]

5. Patient organizations can support ethical and compliant trial awareness

Patient organizations' preferred contribution to clinical trial recruitment focused on dissemination of neutral, accessible information to help people living with MG have informed discussions with clinicians.

Important requirements included:
- **Clear, visually adapted patient-facing materials**
- **Plain-language trial information**
- **Transparent site and contact information**
- **Logistical support for participation**
- **Culturally appropriate content**
- **Careful distinction between awareness-raising and recruitment pressure**

These observations demonstrate how **patient groups can support trial awareness in ways that will resonate with the community.**

Patient expert survey findings: from lived experience to trial priorities

Survey responses from 9 MG/oMG patient experts supported the qualitative findings and helped validate the relevance of a study proposition focused on ocular symptom burden, treatment needs and accessible trial participation.

What **patients** confirmed

88.9%	agreed or strongly agreed that diplopia is typically the most burdensome symptom for people with oMG
88.9%	agreed or strongly agreed that fluctuating symptoms increase disease burden
88.9%	agreed or strongly agreed that oMG affects patients' social relationships and emotional health
77.8%	identified reading or looking at a screen as among the daily activities most affected by oMG symptoms
66.7%	identified driving as among the daily activities most affected by oMG symptoms

What **trials** need to address

100%	indicated that patients need information on the safety profile of the study medication before deciding whether to participate
100%	indicated that patients need information on monitoring and medical consultation during and after the trial
100%	agreed or strongly agreed that patients would like a dedicated contact person before, during and after the trial
88.9%	identified support and reimbursement for travel as important information for trial decision-making
77.8%	agreed or strongly agreed that people with oMG would be interested in trial participation if the study addressed their specific symptoms