AANEM Poster **421** | MGFA Scientific Session Poster **87**

Effect of zilucoplan stratified by age of myasthenia gravis onset: RAISE-XT 120-week *post hoc* analysis

Michael D. Weiss[1], Saskia Bresch[2], Miriam Freimer[3], Channa Hewamadduma[4,5], Raul Juntas-Morales[6], M. Isabel Leite[7], Angelina Maniaol[8], Masayuki Masuda[9], Babak Boroojerdi[10], Paul Mahoney[11], Natasa Savic[12], James F. Howard Jr.[13] on behalf of the RAISE-XT study team

[1]Department of Neurology, University of Washington Medical Center, Seattle, WA, USA; [2]Service de Neurologie, Hospital Pasteur, Centre Hospitalier Universitaire de Nice, Nice, France; [3]Department of Neurology, The Ohio State University Wexner Medical Center, Columbus, OH, USA; [4]Academic Neuromuscular Unit, Sheffield Teaching Hospitals NHS Foundation Trust, Sheffield, UK; [5]Sheffield Institute for Translational Neuroscience (SITraN), School of Medicine and Population Health, University of Sheffield, Sheffield, UK; [6]Department of Neurology, Vall d'Hebron University Hospital, Barcelona, Spain; [7]Nuffield Department of Clinical Neurosciences, University of Oxford, Oxford, UK; [8]Department of Neurology, Oslo University Hospital, Oslo, Norway; [9]Department of Neurology, Tokyo Medical University, Tokyo, Japan; [10]UCB, Monheim, Germany; [11]UCB, Slough, UK; [12]UCB, Bulle, Switzerland; [13]Department of Neurology, The University of North Carolina at Chapel Hill, Chapel Hill, NC, USA

AANEM & MGFA Scientific Session 2026, Orlando, FL, USA; September 29–October 2, 2026

Introduction

- RAISE-XT (NCT04225871), a Phase 3, multicenter, open-label extension study, showed clinically meaningful and sustained improvements in MG-specific outcomes in patients with gMG during treatment with zilucoplan, a potent complement C5 inhibitor, up to Week 120[1]
- Patients with late-onset MG have a greater comorbidity burden than those with early-onset disease[2]
 - Quality of life for patients with late-onset MG may also be worse[3]
- **Here, we evaluated the efficacy of zilucoplan in patients with anti-AChR Ab+ gMG in subgroups defined by age of gMG onset**

Methods

- Adults with anti-AChR Ab+ gMG who completed the Phase 2 (NCT03315130) or Phase 3 RCT (NCT04115293; RAISE) could enter RAISE-XT and self-administer daily subcutaneous zilucoplan 0.3 mg/kg injections[4]
- In this *post hoc* analysis, we report CFB to Week 120 in the early- (≤50 years) and late- (>50 years) onset subgroups for the following outcome scores:
 - MG-ADL
 - QMG
 - MG-QoL15r
 - Neuro-QoL Short Form Fatigue
- The primary outcome of RAISE-XT was incidence of TEAEs;[4] safety for each age-of-onset subgroup was also assessed
- The interim data cut-off date for RAISE-XT was November 11, 2023

Results

- Overall, 106 and 77 patients were included in the early- and late-onset subgroups, respectively (**Table 1**)
 - Severity of disease at baseline was similar between the two subgroups
 - A greater proportion of patients in the early-onset subgroup received CS and ISTs, and had prior MG crises, prior thymectomy and thymoma diagnosis at baseline compared with the late-onset subgroup
- Improvements observed in MG-ADL and QMG scores were sustained through to Week 120 in both early- and late-onset subgroups (**Figure 1**)
- Improvements were also observed for MG-QoL15r and Neuro-QoL Short Form Fatigue scores at Week 120 in both age-of-onset subgroups (**Figure 2**)
 - Mean CFB scores were higher in early-onset subgroup compared with the late-onset subgroup, which may be due to different impact on quality of life in patients with early- and late-onset MG[3]
 - There is no established clinically meaningful change threshold for MG-QoL15r
- In the RAISE-XT overall population, TEAEs occurred in 97.0% (n/N=194/200) of patients
 - The incidence of TEAEs was generally balanced between the two age-of-onset subgroups (**Table 2**)
 - Patients in the late-onset subgroup experienced more serious and severe TEAEs compared with the early-onset subgroup; however, treatment-related TEAEs were lower in late-onset subgroup compared with the early-onset subgroup

Summary and conclusions

In this *post hoc* analysis, we assessed the long-term effect of zilucoplan in subgroups defined by age of onset in patients with gMG in RAISE-XT

Improvements were sustained for MG-specific outcomes at Week 120 in both age-of-onset subgroups

Zilucoplan was generally well tolerated in both age-of-onset subgroups

These data highlight that zilucoplan demonstrated long-term efficacy, irrespective of age at gMG onset

Table 1 Disease severity was comparable between both age-of-onset subgroups at baseline

		Early onset (N=106)	Late onset (N=77)
	Age, years, mean (SD)	**43.9** (12.4)	**66.2** (5.6)
	Sex, male, n (%)	**30** (28.3)	**53** (68.8)
MGFA Clinical Class, n (%)	II	**26** (24.5)	**28** (36.4)
	III	**73** (68.9)	**44** (57.1)
	IV	**7** (6.6)	**5** (6.5)
	MG-ADL score, mean (SD)	**6.3** (4.5)	**6.7** (4.2)
	QMG score, mean (SD)	**14.0** (6.0)	**14.2** (5.8)
	Prior thymectomy, n (%)	**73** (68.9)	**15** (19.5)
	Prior MG crisis, n (%)	**39** (36.8)	**20** (26.0)
	Thymoma diagnosis, n (%)	**32** (30.2)	**11** (14.3)
	Duration of disease, years, mean (SD)	**13.1** (11.2)	**4.2** (3.8)
Baseline gMG-specific therapies, n (%)	CS	**99** (93.4)	**63** (81.8)
	ISTs	**86** (81.1)	**47** (61.0)
	Cholinesterase inhibitors	**104** (98.1)	**73** (94.8)

mITT population, which included all patients enrolled in RAISE-XT who received at least one dose of zilucoplan and had at least one post-dosing MG-ADL score. Categories with notable differences between subgroups have been shaded in grey.

Table 2 Incidence of TEAEs was similar between early- and late-onset subgroups

	Early onset (N=106)	Late onset (N=77)
Any TEAE, n (%)	**102** (96.2)	**75** (97.4)
Serious TEAE, n (%)	**36** (34.0)	**38** (49.4)
Treatment-related serious TEAE, n (%)	**3** (2.8)	**1** (1.3)
TEAE resulting in permanent withdrawal from zilucoplan, n (%)	**13** (12.3)	**8** (10.4)
Treatment-related TEAE, n (%)	**41** (38.7)	**26** (33.8)
Severe TEAE, n (%)	**31** (29.2)	**32** (41.6)
Deaths, n (%)	**1** (0.9)	**3** (3.9)

Safety set, which included all patients who received at least one dose of zilucoplan in RAISE-XT. None of the deaths were related to zilucoplan.

Figure 1 Improvements in MG-ADL and QMG scores were sustained through to Week 120 for both age-of-onset subgroups

(a) MG-ADL

Early onset

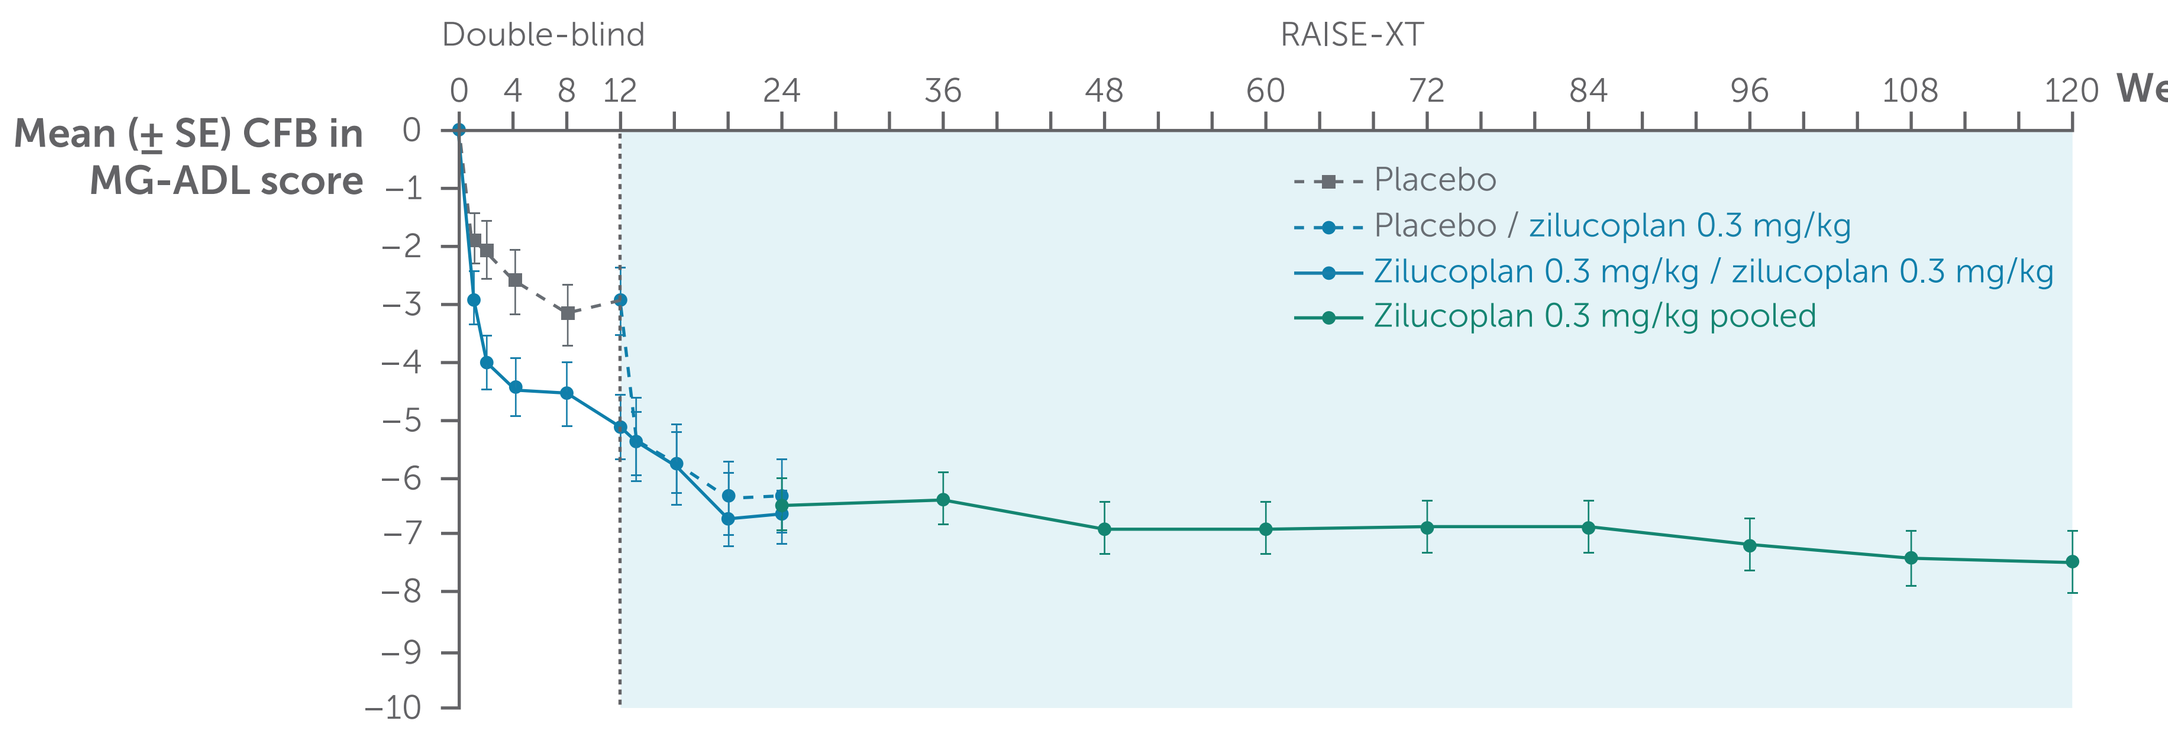
Double-blind
RAISE-XT
Week
Mean (± SE) CFB in MG-ADL score
Placebo
Placebo / zilucoplan 0.3 mg/kg
Zilucoplan 0.3 mg/kg / zilucoplan 0.3 mg/kg
Zilucoplan 0.3 mg/kg pooled

Number of patients (n)											
Placebo (n=50)	50	50	–	–	–	–	–	–	–	–	–
Zilucoplan 0.3 mg/kg (n=56)	56	56	–	–	–	–	–	–	–	–	–
Zilucoplan 0.3 mg/kg (n=106)	–	–	102	101	96	94	88	87	84	72	51

Late onset

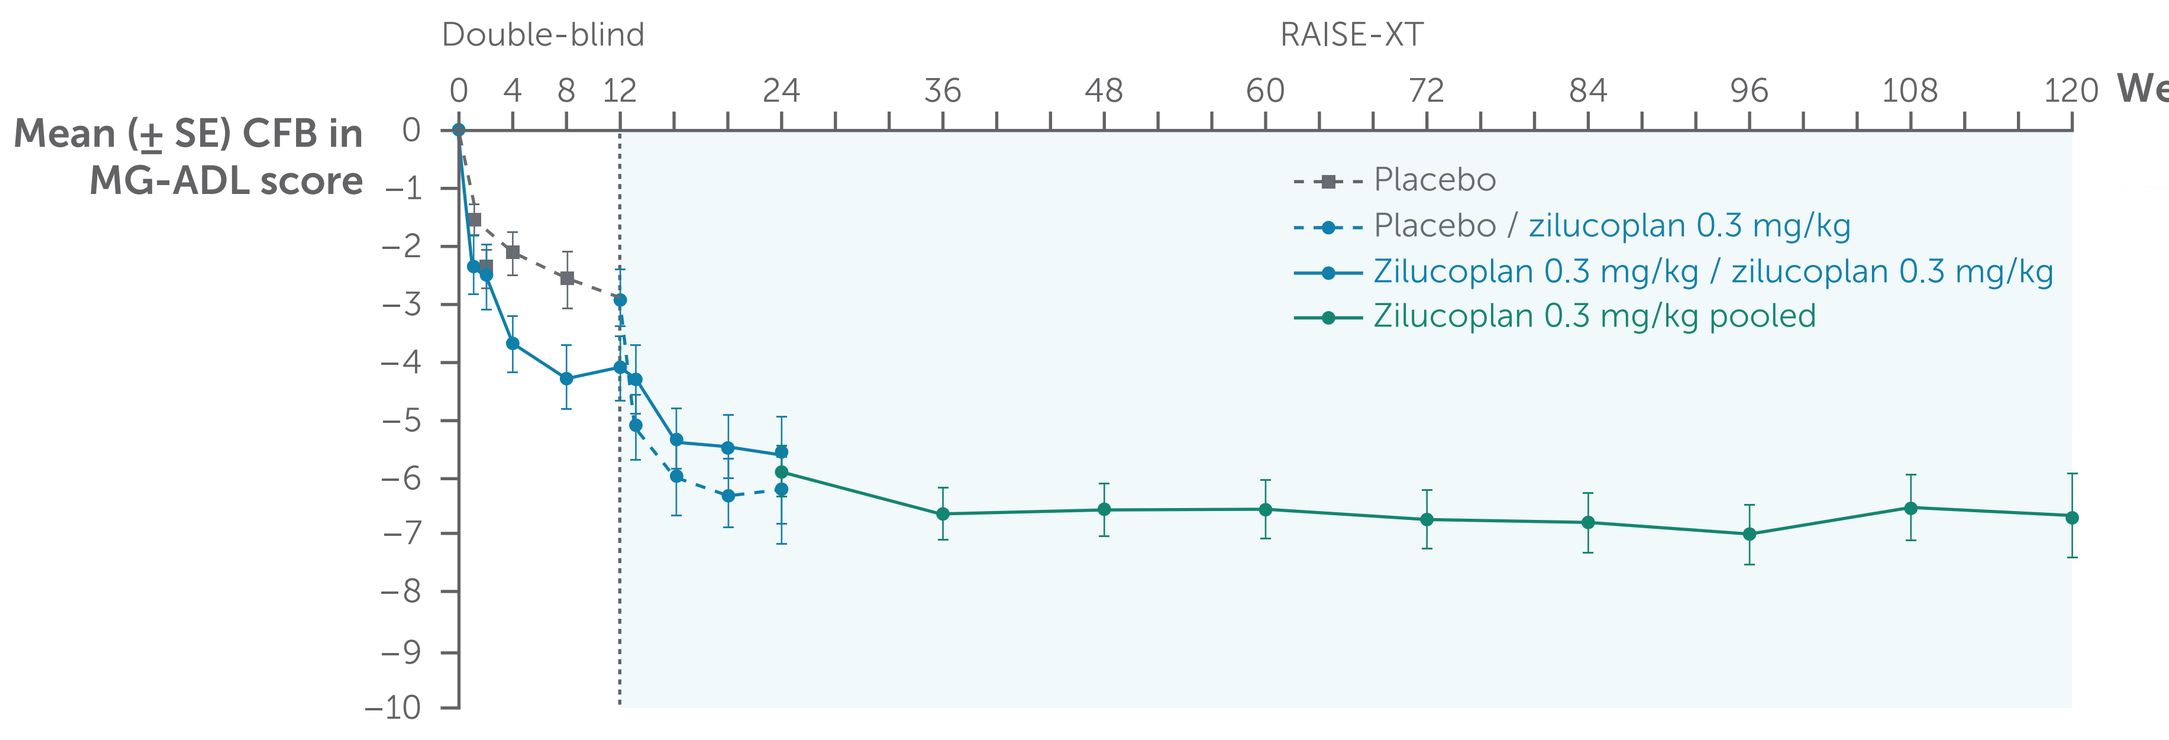
Double-blind
RAISE-XT
Week
Mean (± SE) CFB in MG-ADL score
Placebo
Placebo / zilucoplan 0.3 mg/kg
Zilucoplan 0.3 mg/kg / zilucoplan 0.3 mg/kg
Zilucoplan 0.3 mg/kg pooled

Number of patients (n)											
Placebo (n=40)	40	40	–	–	–	–	–	–	–	–	–
Zilucoplan 0.3 mg/kg (n=37)	37	37	–	–	–	–	–	–	–	–	–
Zilucoplan 0.3 mg/kg (n=77)	–	–	73	69	66	62	61	61	58	51	35

(b) QMG

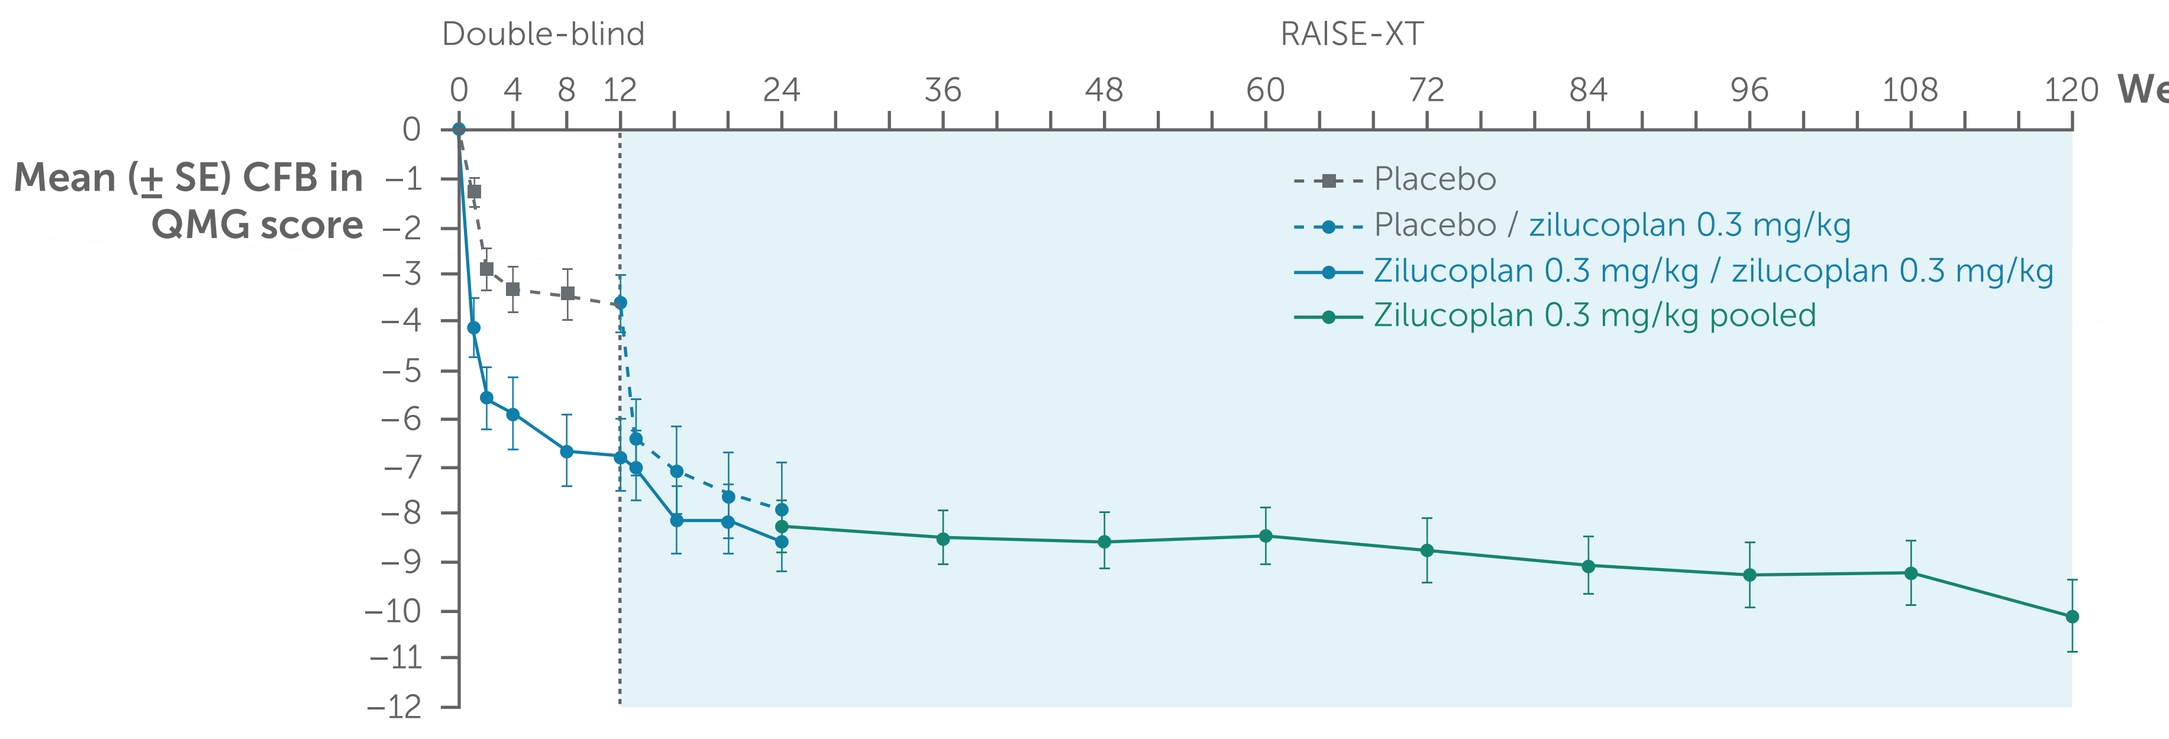
Double-blind
RAISE-XT
Week
Mean (± SE) CFB in QMG score
Placebo
Placebo / zilucoplan 0.3 mg/kg
Zilucoplan 0.3 mg/kg / zilucoplan 0.3 mg/kg
Zilucoplan 0.3 mg/kg pooled

Number of patients (n)											
Placebo (n=50)	50	49	–	–	–	–	–	–	–	–	–
Zilucoplan 0.3 mg/kg (n=56)	56	55	–	–	–	–	–	–	–	–	–
Zilucoplan 0.3 mg/kg (n=106)	–	–	100	101	96	91	88	87	78	70	50

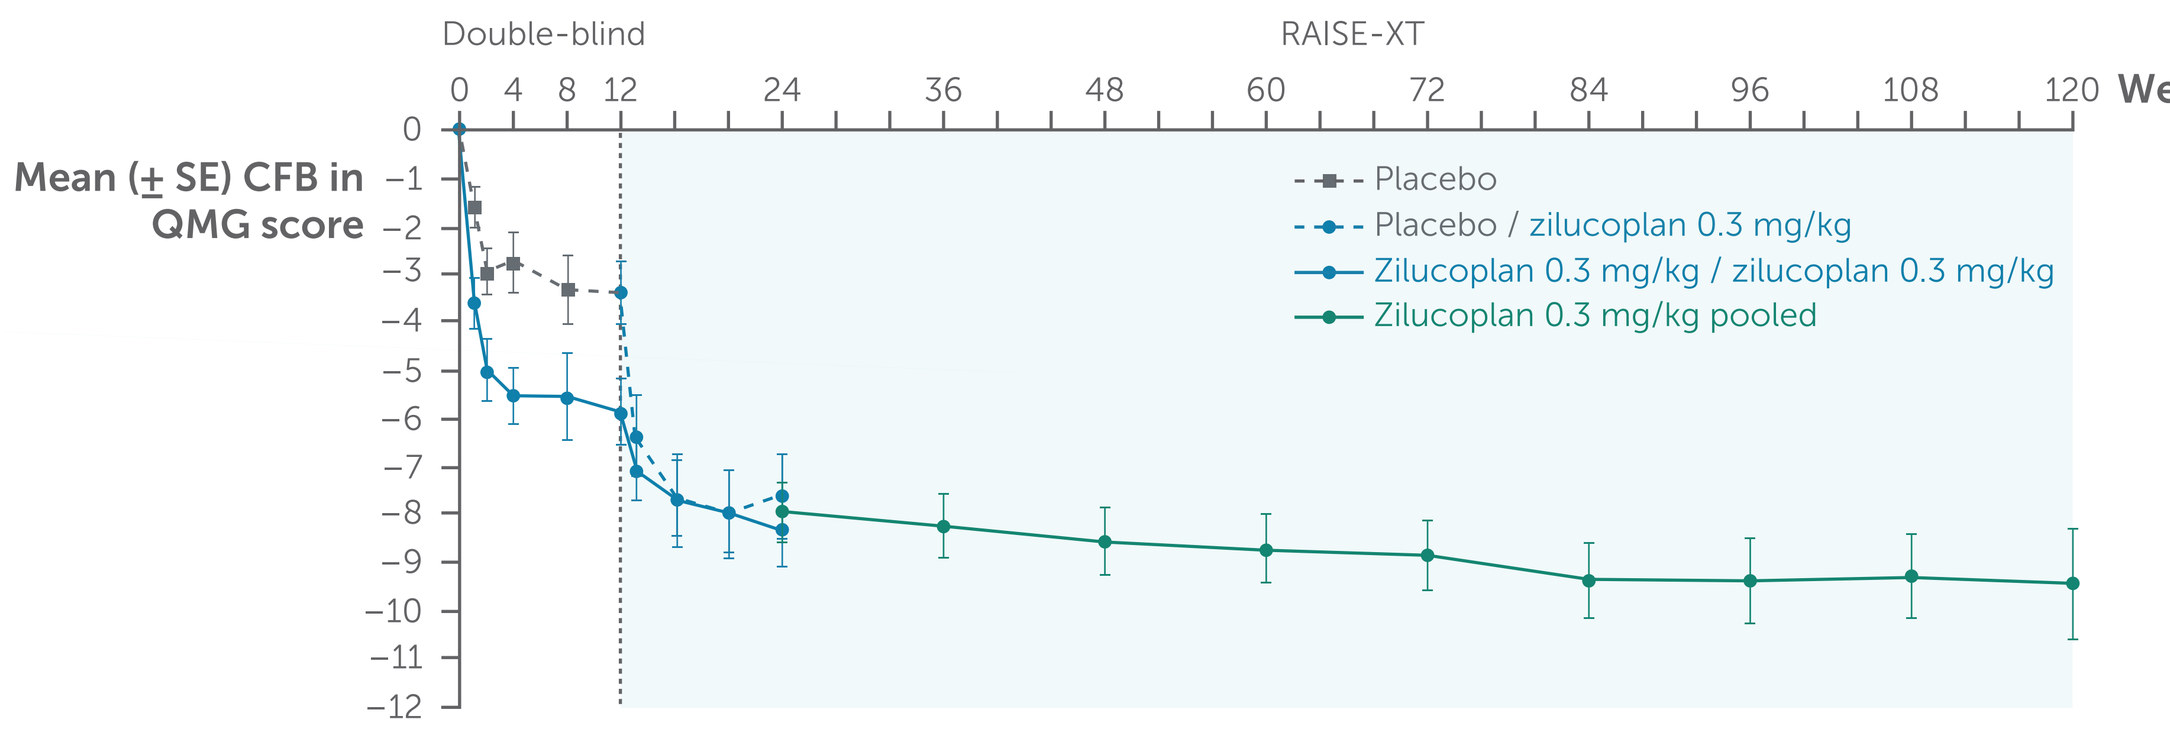
Double-blind
RAISE-XT
Week
Mean (± SE) CFB in QMG score
Placebo
Placebo / zilucoplan 0.3 mg/kg
Zilucoplan 0.3 mg/kg / zilucoplan 0.3 mg/kg
Zilucoplan 0.3 mg/kg pooled

Number of patients (n)											
Placebo (n=40)	40	40	–	–	–	–	–	–	–	–	–
Zilucoplan 0.3 mg/kg (n=37)	37	37	–	–	–	–	–	–	–	–	–
Zilucoplan 0.3 mg/kg (n=77)	–	–	72	68	65	62	61	59	54	49	33

mITT population.

Figure 2 At Week 120, improvement from baseline was observed across MG-QoL15r and Neuro-QoL Short Form Fatigue scores in both age-of-onset subgroups

MG-QoL15r

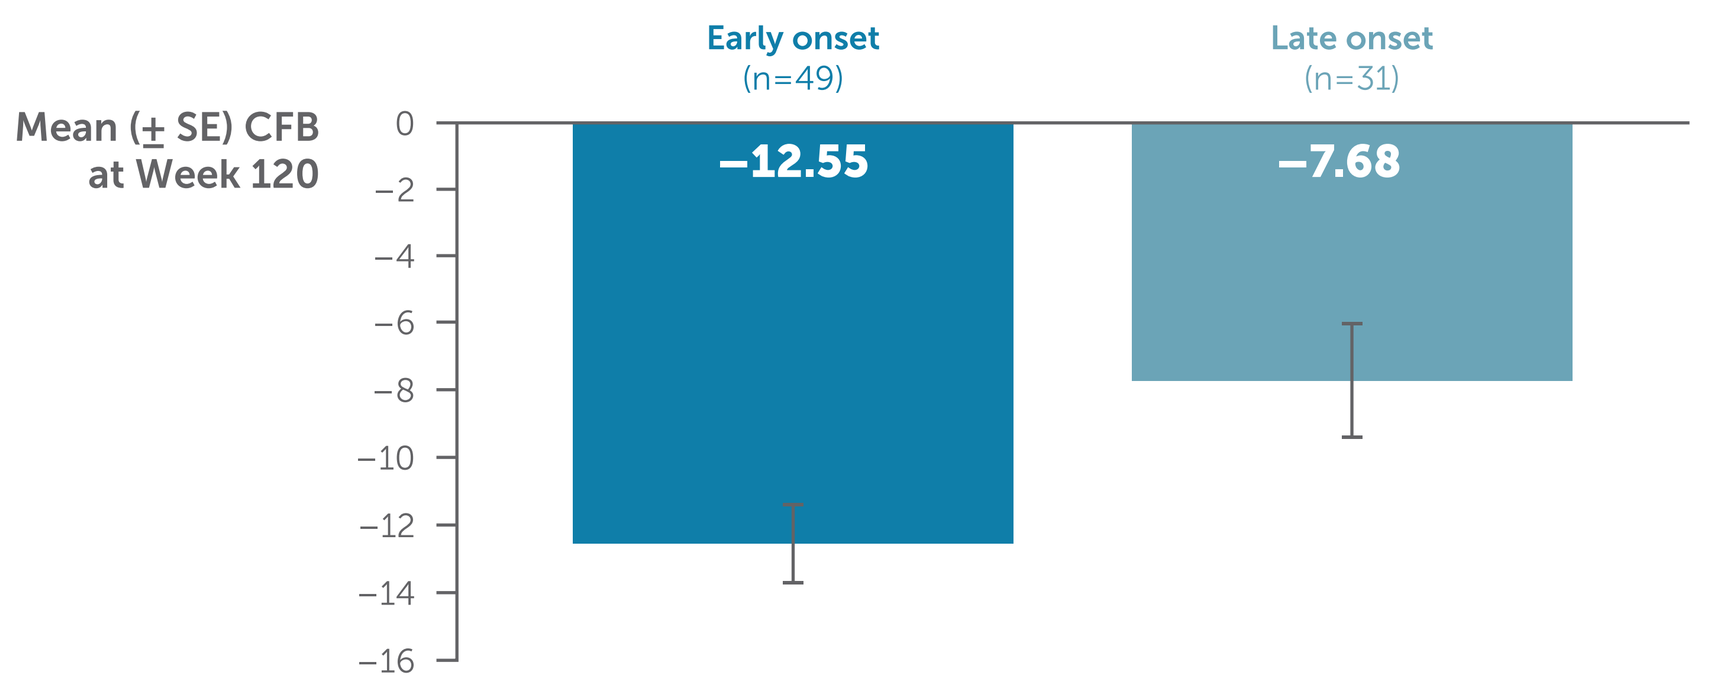
Early onset (n=49)
Late onset (n=31)
Mean (± SE) CFB at Week 120
−12.55
−7.68

Baseline	Early onset	Late onset
N	**106**	**77**
Mean (SE) score	**18.8** (0.7)	**17.8** (0.8)

Neuro-QoL Short Form Fatigue T-score

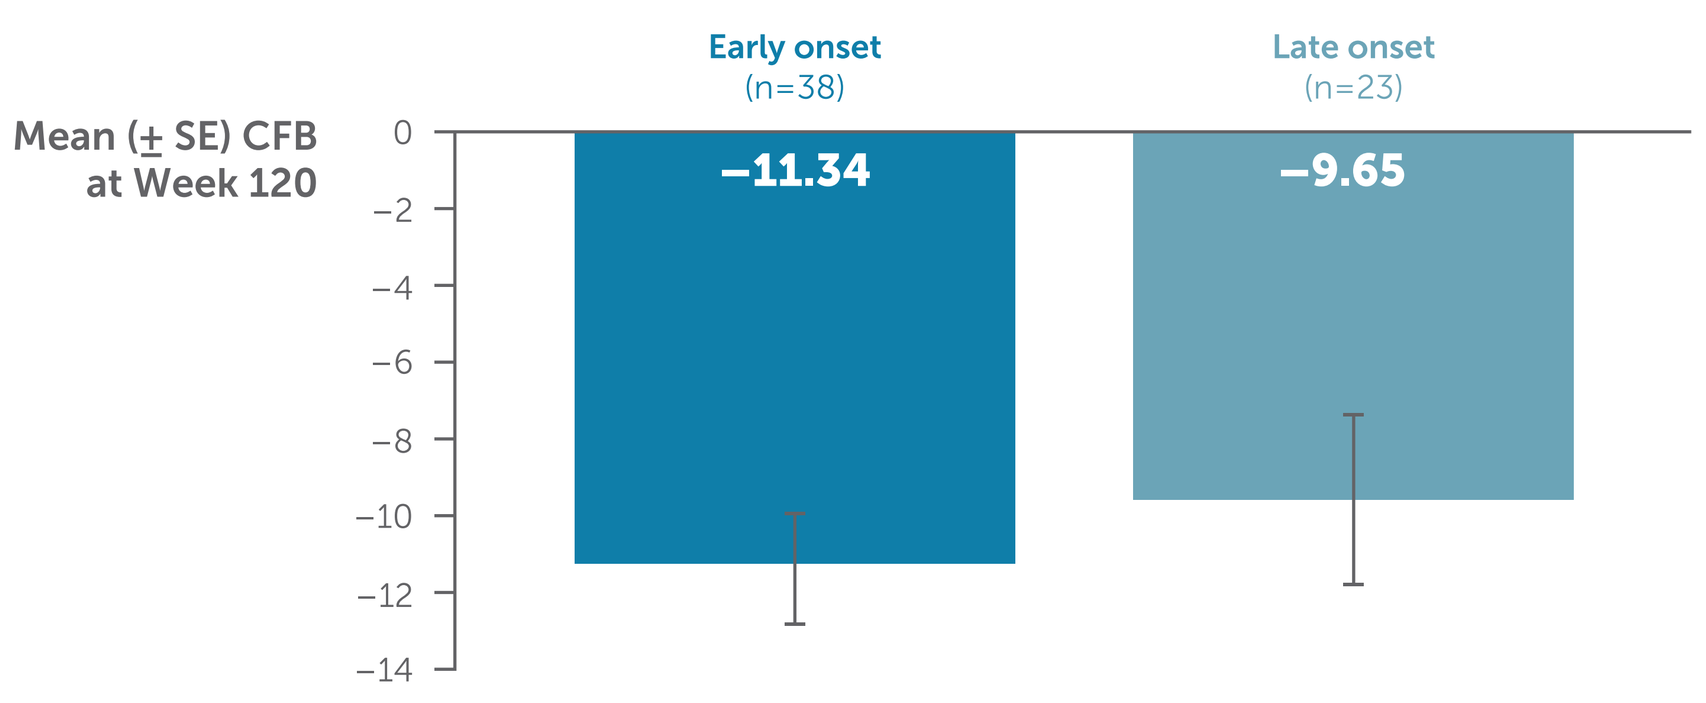
Early onset (n=38)
Late onset (n=23)
Mean (± SE) CFB at Week 120
−11.34
−9.65

Baseline	Early onset	Late onset
N	**90**	**65**
Mean (SE) score	**29.9** (0.7)	**28.8** (0.9)

mITT population.

Abbreviations: Ab+, antibody positive; AChR, acetylcholine receptor; C5, component 5; CFB, change from baseline; CS, corticosteroids; gMG, generalized myasthenia gravis; ISTs, immunosuppressants; MG, myasthenia gravis; MG-ADL, Myasthenia Gravis Activities of Daily Living; MGFA, Myasthenia Gravis Foundation of America; MG-QoL15r, Myasthenia Gravis Quality of Life 15-item revised; mITT, modified intention to treat; Neuro-QoL, Quality of Life in Neurological Disorders; QMG, Quantitative Myasthenia Gravis; RCT, randomized controlled trial; SD, standard deviation; SE, standard error; TEAE, treatment-emergent adverse event.

Acknowledgments: This study was funded by UCB. The authors acknowledge Nishtha Chandra, PhD, of Ogilvy Health, London, UK, for editorial assistance, which was funded by UCB. The authors thank Veronica Porkess, PhD, of UCB for publication and editorial support. The authors thank the patients and their caregivers, in addition to the investigators and their teams who contributed to this study.

Author disclosures: Michael D. Weiss has received honoraria for serving on scientific advisory boards for Alexion Pharmaceuticals, Amgen, Amylyx Pharmaceuticals, argenx, Biogen, Immunovant, Mitsubishi Tanabe Pharma and Ra Pharmaceuticals (now UCB), consulting honoraria from CSL Behring and Cytokinetics, and speaker honoraria from Amgen, Soleo Health, and UCB. He has been a paid Medical Monitor for a NeuroNEXT study. Saskia Bresch has served as a paid Consultant for Alexion Pharmaceuticals, argenx, Biogen, Bristol Myers Squibb, Merck, Roche, Sandoz, Sanofi Genzyme (now Sanofi) and UCB. Miriam Freimer has served as a paid Consultant for Arcellx, argenx and UCB. She receives research support from Abcuro, Alnylam Pharmaceuticals, argenx, Avidity Biosciences, COUR Pharmaceuticals, Dianthus Therapeutics, Fulcrum Therapeutics, Johnson & Johnson Innovative Medicine, the NIH, RemeGen Biosciences and UCB. Channa Hewamadduma is Principal Investigator/NCI in clinical trials in various neuromuscular disorders sponsored by Alexion Pharmaceuticals, argenx, Avidity, Cartesian, Dyne, Janssen Pharmaceuticals (now Johnson & Johnson Innovative Medicine), Novartis, PepGen, Regeneron Pharmaceuticals and UCB. He has received funding for consultancy on scientific or educational advisory boards for argenx, Biogen, Lupin, Roche and UCB, and has received an investigator-led research grant from UCB. His study activities were supported by a Sheffield NIHR BRC UK centre grant. He is a trustee of the myasthenia gravis patient organisation Myaware. Raul Juntas-Morales has nothing to disclose. M. Isabel Leite is funded by the NHS (Myasthenia and Related Disorders Service and National Specialised Commissioning Group for Neuromyelitis Optica, UK) and by the University of Oxford, UK. She has been awarded research grants from UK associations for patients with myasthenia and with muscular disorders (Myaware and Muscular Dystrophy UK, respectively) and the University of Oxford. She has received speaker honoraria or travel grants from Biogen, the Guthy-Jackson Charitable Foundation, Novartis and UCB. She serves on scientific or educational advisory boards for argenx, Horizon Therapeutics (now Amgen) and UCB. Angelina Maniaol has received payment for travel, meeting attendance, consulting honoraria or advisory board participation from Alexion Pharmaceuticals, argenx, Biogen, Johnson & Johnson and UCB. Masayuki Masuda has served as a paid Consultant for Alexion Pharmaceuticals and argenx, and has received speaker honoraria from Alexion Pharmaceuticals, argenx, Asahi Kasei Medical, Chugai Pharmaceutical, the Japan Blood Products Organization, Takeda Pharmaceuticals and UCB. Babak Boroojerdi, Paul Mahoney and Natasa Savic are employees and shareholders of UCB. James F. Howard Jr. has received research funding (paid to his institution) from Ad Scientiam, Alexion/AstraZeneca Rare Disease, argenx, Cartesian Therapeutics, Centers for Disease Control and Prevention, Merck EMD Serono, MGFA, Muscular Dystrophy Association, NIH, NMD Pharma and UCB; honoraria/consulting fees from AcademicCME, Alexion/AstraZeneca Rare Disease, Amgen, argenx, Biohaven Ltd, Cartesian Therapeutics, CheckRare CME, CorEvitas, Curie.Bio, H. Lundbeck A/S, Japan Tobacco Company, Kyverna Therapeutics, Merck EMD Serono, NMD Pharma, Novartis, PeerView CME, Physicians' Education Resource (PER, MJH) CME, Regeneron Pharmaceuticals, Seismic Therapeutics, Toleranzia AB, UCB and Vor Biopharma; non-financial support from Alexion/AstraZeneca Rare Disease, argenx, Biohaven Ltd, Cartesian Therapeutics, Toleranzia AB, UCB and Vor Biopharma.

References: 1. Howard JF Jr., et al. Long-term safety and efficacy of zilucoplan in generalized myasthenia gravis: 120-week interim analysis of RAISE-XT [poster]. AANEM 2024. Poster 192. 2. Huang X, et al. Neurology. 2025;105(12):e214428. 3. Szczudlik P, et al. Front Neurol. 2020;11:553626. 4. Howard JF Jr., et al. Ther Adv Neurol Disord. 2024;17:17562864241243186.

Please use this QR code to download a PDF of the poster.

These data were previously presented at the Association of British Neurologists (ABN) 2026, Birmingham, UK; May 5–8, 2026